FPN 438TiP

MOUNTAINEER-03: PHASE 3 STUDY OF TUCATINIB, TRASTUZUMAB, AND MODIFIED FOLFOX6 AS FIRST-LINE TREATMENT IN HER2+ METASTATIC COLORECTAL CANCER (TRIAL IN PROGRESS)

Thierry Andre[1], Tanios Bekaii-Saab[2], Josep Tabernero[3], Salvatore Siena[4], Takayuki Yoshino[5], David Adelberg[6], James Ward[7], Shan Yang[7], John Strickler[8], Eric Van Cutsem[9]

[1]Sorbonne Université et INSERM 'Instabilité des Microsatellites et Cancer, Hôpital Saint Antoine, Paris, France; [2]Mayo Clinic, Scottsdale, AZ, USA; [3]Vall d'Hebron University Hospital, Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [4]Università degli Studi di Milano and Grande Ospedale Metropolitano Niguarda, Milan, Italy; [5]National Cancer Center Hospital East, Kashiwa, Japan; [6]Merck & Co., Inc., Rahway, NJ, USA; [7]Seagen Inc., Bothell, WA, USA; [8]Duke University Medical Center, Durham, NC, USA; [9]University Hospital Gasthuisberg-University of Leuven, Leuven, Belgium

Background and Rationale

- Colorectal cancer (CRC) is the third most common cancer and the second leading cause of cancer-related death worldwide[1]
- Approximately 20% of patients have metastatic disease at the time of diagnosis, and up to 40% of patients initially diagnosed with locoregional cancer will develop recurrent disease[2,3]
- Current standard of care for treatment of metastatic colorectal cancer (mCRC) is multi-agent chemotherapy, with or without a VEGF- or EGFR-inhibitor; treatments are not curative and survival outcomes remain poor[4,5]
- HER2 amplification is present in approximately 3%–5% of patients with mCRC and approximately 5%–14% of patients with RAS/BRAF WT mCRC[6–10]
- HER2 alterations have been identified as potential mechanisms of primary resistance and poor response to anti-EGFR therapies[9,11–14]
 - Recent developments have increased interest in HER2 as a therapeutic target for mCRC and HER2-directed therapies as a mechanism to better tailor treatment for some patients
 - There are currently no FDA- or EMA-approved therapies for HER2+ mCR
- Tucatinib is a highly selective HER2-directed TKI
 - Preclinical data across tumor types, including CRC, show strong evidence supporting the increased activity of tucatinib in combination with trastuzumab relative to other agents
 - Tumor regression rates in tucatinib and trastuzumab monotherapy were 26% and 15%, respectively; tumor regression rates were 66% when tucatinib and trastuzumab were administered in combination[15]
 - Tucatinib is currently approved in multiple regions for patients with HER2+ metastatic breast cancer who have received one or more prior anti-HER2 therapies in the metastatic setting[16]
 - Tucatinib is currently being investigated in gastrointestinal cancers
 - Results from the primary endpoint analysis from the single-arm phase 2 MOUNTAINEER trial (NCT03043313) showed durable and clinically meaningful antitumor activity of tucatinib and trastuzumab in patients with previously treated RAS wild-type, HER2+ mCRC
 - cORR of 38.1%, median DOR per BICR of 12.4 months[17]
 - Median duration of follow-up of 16.3 months
 - Tucatinib plus trastuzumab was well tolerated with a low discontinuation rate (5.8%) and no deaths due to AEs
 - Most common AE was diarrhoea, which was predominately low grade (Grade 3 events, 3.5% and no Grade 4 events)

Tucatinib Proposed Mechanism of Action

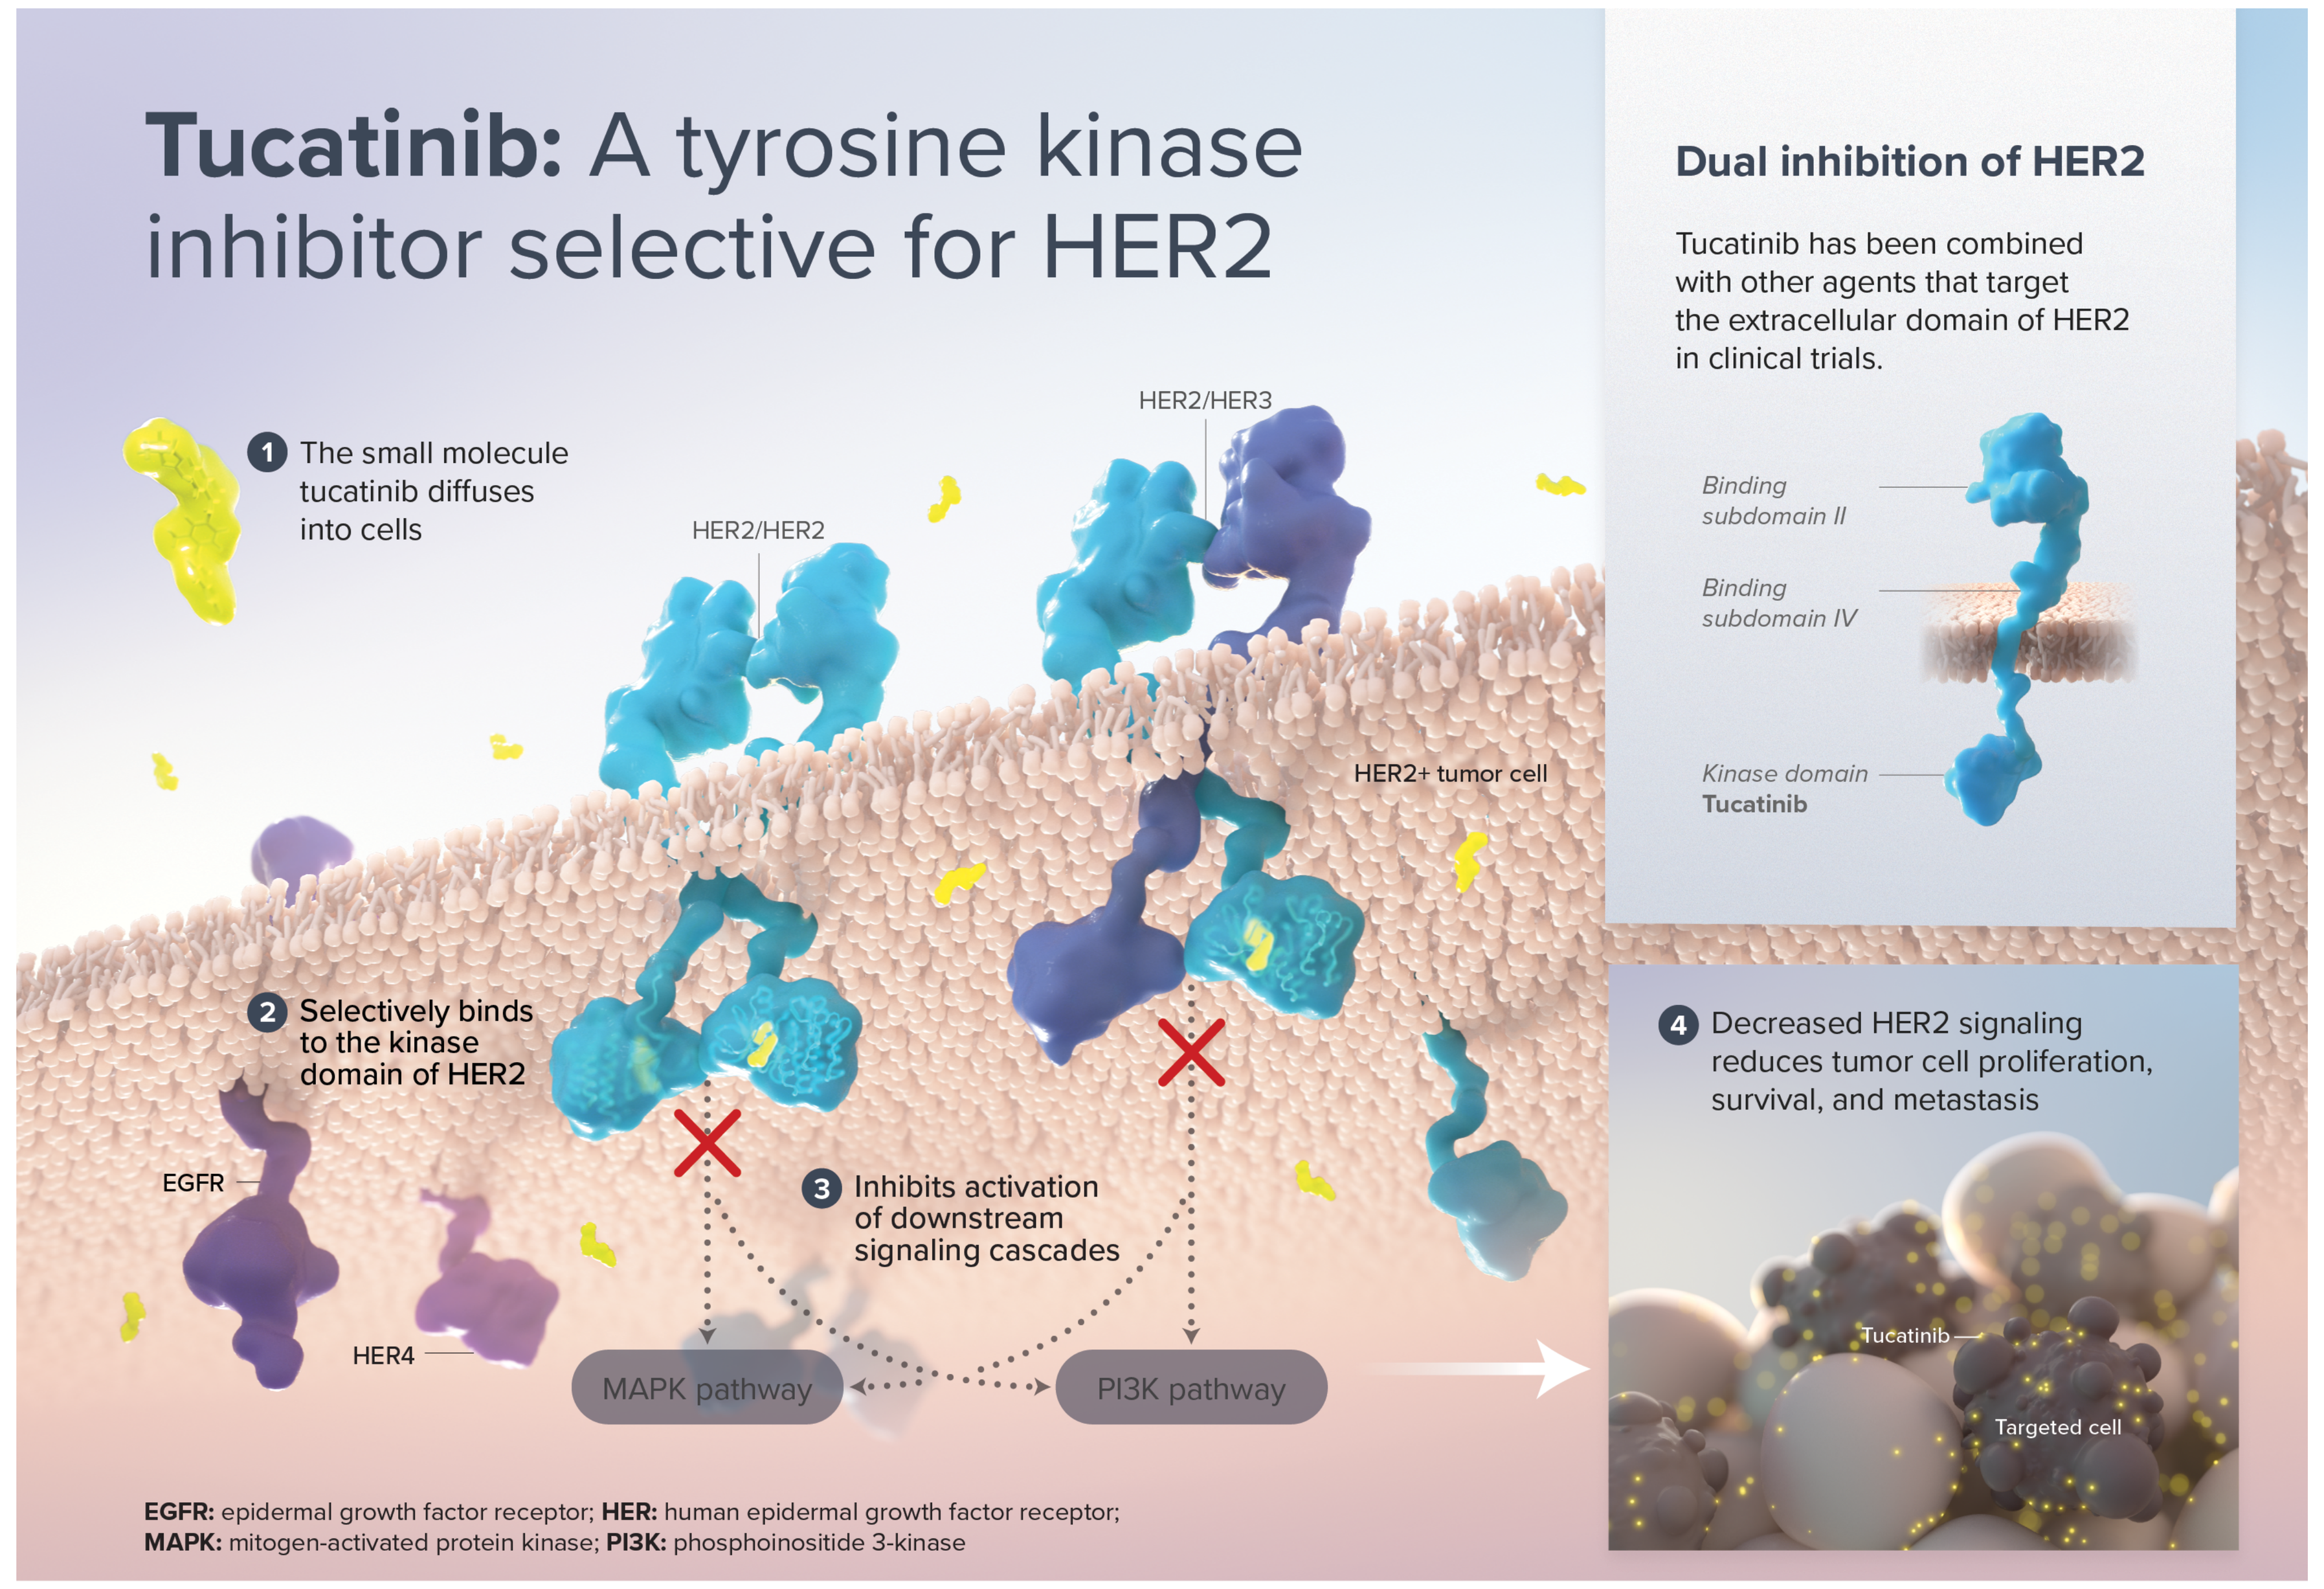
Tucatinib: A tyrosine kinase inhibitor selective for HER2
1 The small molecule tucatinib diffuses into cells
HER2/HER2
HER2/HER3
HER2+ tumor cell
2 Selectively binds to the kinase domain of HER2
EGFR
HER4
3 Inhibits activation of downstream signaling cascades
MAPK pathway
PI3K pathway
Dual inhibition of HER2
Tucatinib has been combined with other agents that target the extracellular domain of HER2 in clinical trials.
Binding subdomain II
Binding subdomain IV
Kinase domain
Tucatinib
4 Decreased HER2 signaling reduces tumor cell proliferation, survival, and metastasis
Tucatinib
Targeted cell
EGFR: epidermal growth factor receptor; HER: human epidermal growth factor receptor; MAPK: mitogen-activated protein kinase; PI3K: phosphoinositide 3-kinase

Tucatinib is an investigational agent, and its safety and efficacy have not been established. There is no guarantee that tucatinib will receive regulatory approval and become commercially available for uses being investigated.

© 2022 Seagen Inc. Bothell WA 98021. All rights reserved. USM/TUC/2019/0018

Study Design

- MOUNTAINEER-03 (NCT05253651) is a global, open-label, randomized, phase 3 study of tucatinib with trastuzumab and mFOLFOX6 versus standard of care for the first-line treatment of HER2+ and RAS wild-type mCRC

Study Population

- Measurable disease per RECIST v1.1
- ECOG PS 0–1
- HER2+, RAS WT, mCRC
- No prior treatment in metastatic setting
- May have received adjuvant treatment if completed >6 months prior to enrollment

N≈400* → Randomize 1:1

Treatment

Tucatinib experimental arm
Tucatinib 300 mg PO BID
Trastuzumab 8 mg/kg loading dose, then 6 mg/kg IV (Q3W)
mFOLFOX6 (Q2W)

Standard-of-care control arm
mFOLFOX6 (Q2W), or
mFOLFOX6 (Q2W) + bevacizumab (Q2W), or
mFOLFOX6 (Q2W) + cetuximab (QW)

Endpoints

Primary
- PFS per RECIST 1.1 by BICR assessment

Secondary
- OS
- cORR
- PFS
- DOR
- PFS2
- Safety
- PK
- PROs

*Stratification by both primary tumor location (left-sided versus all other) and liver metastases (presence or absence)

Eligibility

Key Inclusion Criteria

- Histologically and/or cytologically documented adenocarcinoma of the colon or rectum, which is metastatic and/or unresectable
- HER2+ disease, determined centrally using a tissue-based HER2 assay
- *RAS* wild-type mCRC, determined locally or centrally
- ≥18 years
- Measurable disease per RECIST v1.1
- ECOG PS of 0 or 1
- Life expectancy of ≥3 months
- Adequate baseline hematologic, hepatic, renal, coagulation, and cardiac function
- Based on MRI or CT with contrast, previously treated, asymptomatic brain metastases permitted

Key Exclusion Criteria

- Received prior treatment in the metastatic setting or completed adjuvant treatment ≤6 months prior to enrollment
- Received radiation therapy within 14 days prior to enrollment (or within 7 days of stereotactic radiosurgery)
- Previous treatment with anti-HER2 therapy
- Clinically significant cardiopulmonary disease
- Ongoing ≥Grade 2 diarrhea of any etiology at screening

Key Study Assessments

Efficacy

- Radiological disease assessments (CT and/or MRI) will be performed at baseline, during study treatment (every 6 weeks ±7 days for the first 72 weeks then every 12 weeks ±7 days), and at the end of treatment visit

Pharmacokinetics

- Plasma concentrations of tucatinib (blood samples collected on Day 1 of Cycles 2 to 6)
- Pharmacodynamic and Biomarker Assessments

Safety

- Laboratory Assessments will be performed locally

Patient-reported Outcomes

- The EORTC QLQ-C30 and EQ-5D-5L instruments will be used prior to any study treatment and then at protocol-specified time points

References

1. Baidoun F, Elshiwy K, Elkeraie Y, et al. Curr Drug Targets. 2021;22(9)998-1009.
2. National Cancer Institute: Cancer Stat Facts: Colorectal Cancer. 2019. https://seer.cancer.gov/statfacts/html/colorect.html (accessed August 11, 2022).
3. Kahi CJ, Boland CR, Dominitz JA, et al. Colonoscopy surveillance after colorectal cancer resection: Recommendations of the US Multi-Society Task Force on Colorectal Cancer. Gastroenterology 2016; 150: 758–768.e11.
4. Heinemann V, Singh M, Hardstock F, et al. Clin Colorectal Cancer. 2022;21(2):122-131.
5. Van Cutsem E, Cervantes A, Adam R, et al. Ann Oncol. 2016;27(8):1386-1422.
6. Benson AB, Venook AP, Al-Hawary MM, et al. J Natl Compr Canc Netw. 2021;19(3):329-359.
7. Kang A, Bloudek L, Mordi U, et al. J Manag Care Spec Pharm. 2021;S20-21.
8. Patel JN, Fong MK, Jagosky M. J Pers Med. 2019;9(1):3.
9. Sartore-Bianchi A, Amatu A, Porcu L, et al. The Oncologist. 2019;24(10):1395-1402.
10. Strickler J, Ng K, Cercek A, et al. J Clin Oncol. 2021;39(3 Suppl):Abstract TPS153.
11. Bertotti A, Papp E, Jones S, et al. Nature. 2015;526:263-267.
12. Jeong JH, Kim J, Hong YS, et al. Clin Colorectal Cancer. 2017;16(3):e147-152.
13. Raghav K, Loree JM, Morris JS, et al. JCO Precis Oncol. 2019;3:1-13.
14. Sawada K, Nakamura Y, Yamanaka T, et al. Clin Colorectal Cancer. 2018;17(3):198-205.
15. Kulukian A, Lee P, Taylor J, et al. Mol Cancer Ther. 2020;19(4):976-987.
16. TUKYSA [prescribing information]. Bothell, WA: Seagen, Inc.; 2021.
17. Strickler J, Cercek A, Siena S, et al. Ann Oncol. 2022;33(Suppl 4):S375-376.

Objectives and Associated Endpoints

Primary Objective	Primary Endpoint
Compare PFS per RECIST v1.1 by BICR assessment between treatment arms	PFS per RECIST v1.1 by BICR assessment
Key Secondary Objectives	**Key Secondary Endpoints**
Evaluate activity in treatment arms	OS, cORR per RECIST v1.1 by BICR and INV assessments, PFS per RECIST v1.1 by INV assessment, DOR per BICR and INV assessments
Evaluate time from randomization to disease progression on next-line treatment or death from any cause (PFS2)	Time from randomization to disease progression on next-line treatment or death from any cause (PFS2)
Assess the overall safety profiles by treatment arm	Safety, measured by adverse events, abnormalities in vital signs and laboratory values, and frequency of dose holds and dose reductions
Evaluate the PK of tucatinib	PK of tucatinib
Assess key PROs	PROs as assessed by EORTC QLQ-C30

Summary

- MOUNTAINEER-03 is investigating the efficacy and safety of tucatinib in combination with trastuzumab and mFOLFOX6 in comparison to mFOLFOX6 given with or without either bevacizumab or cetuximab as first-line treatment in patients with HER2+ mCRC
- Enrollment began in 2022 and is ongoing in the United States with sites planned globally

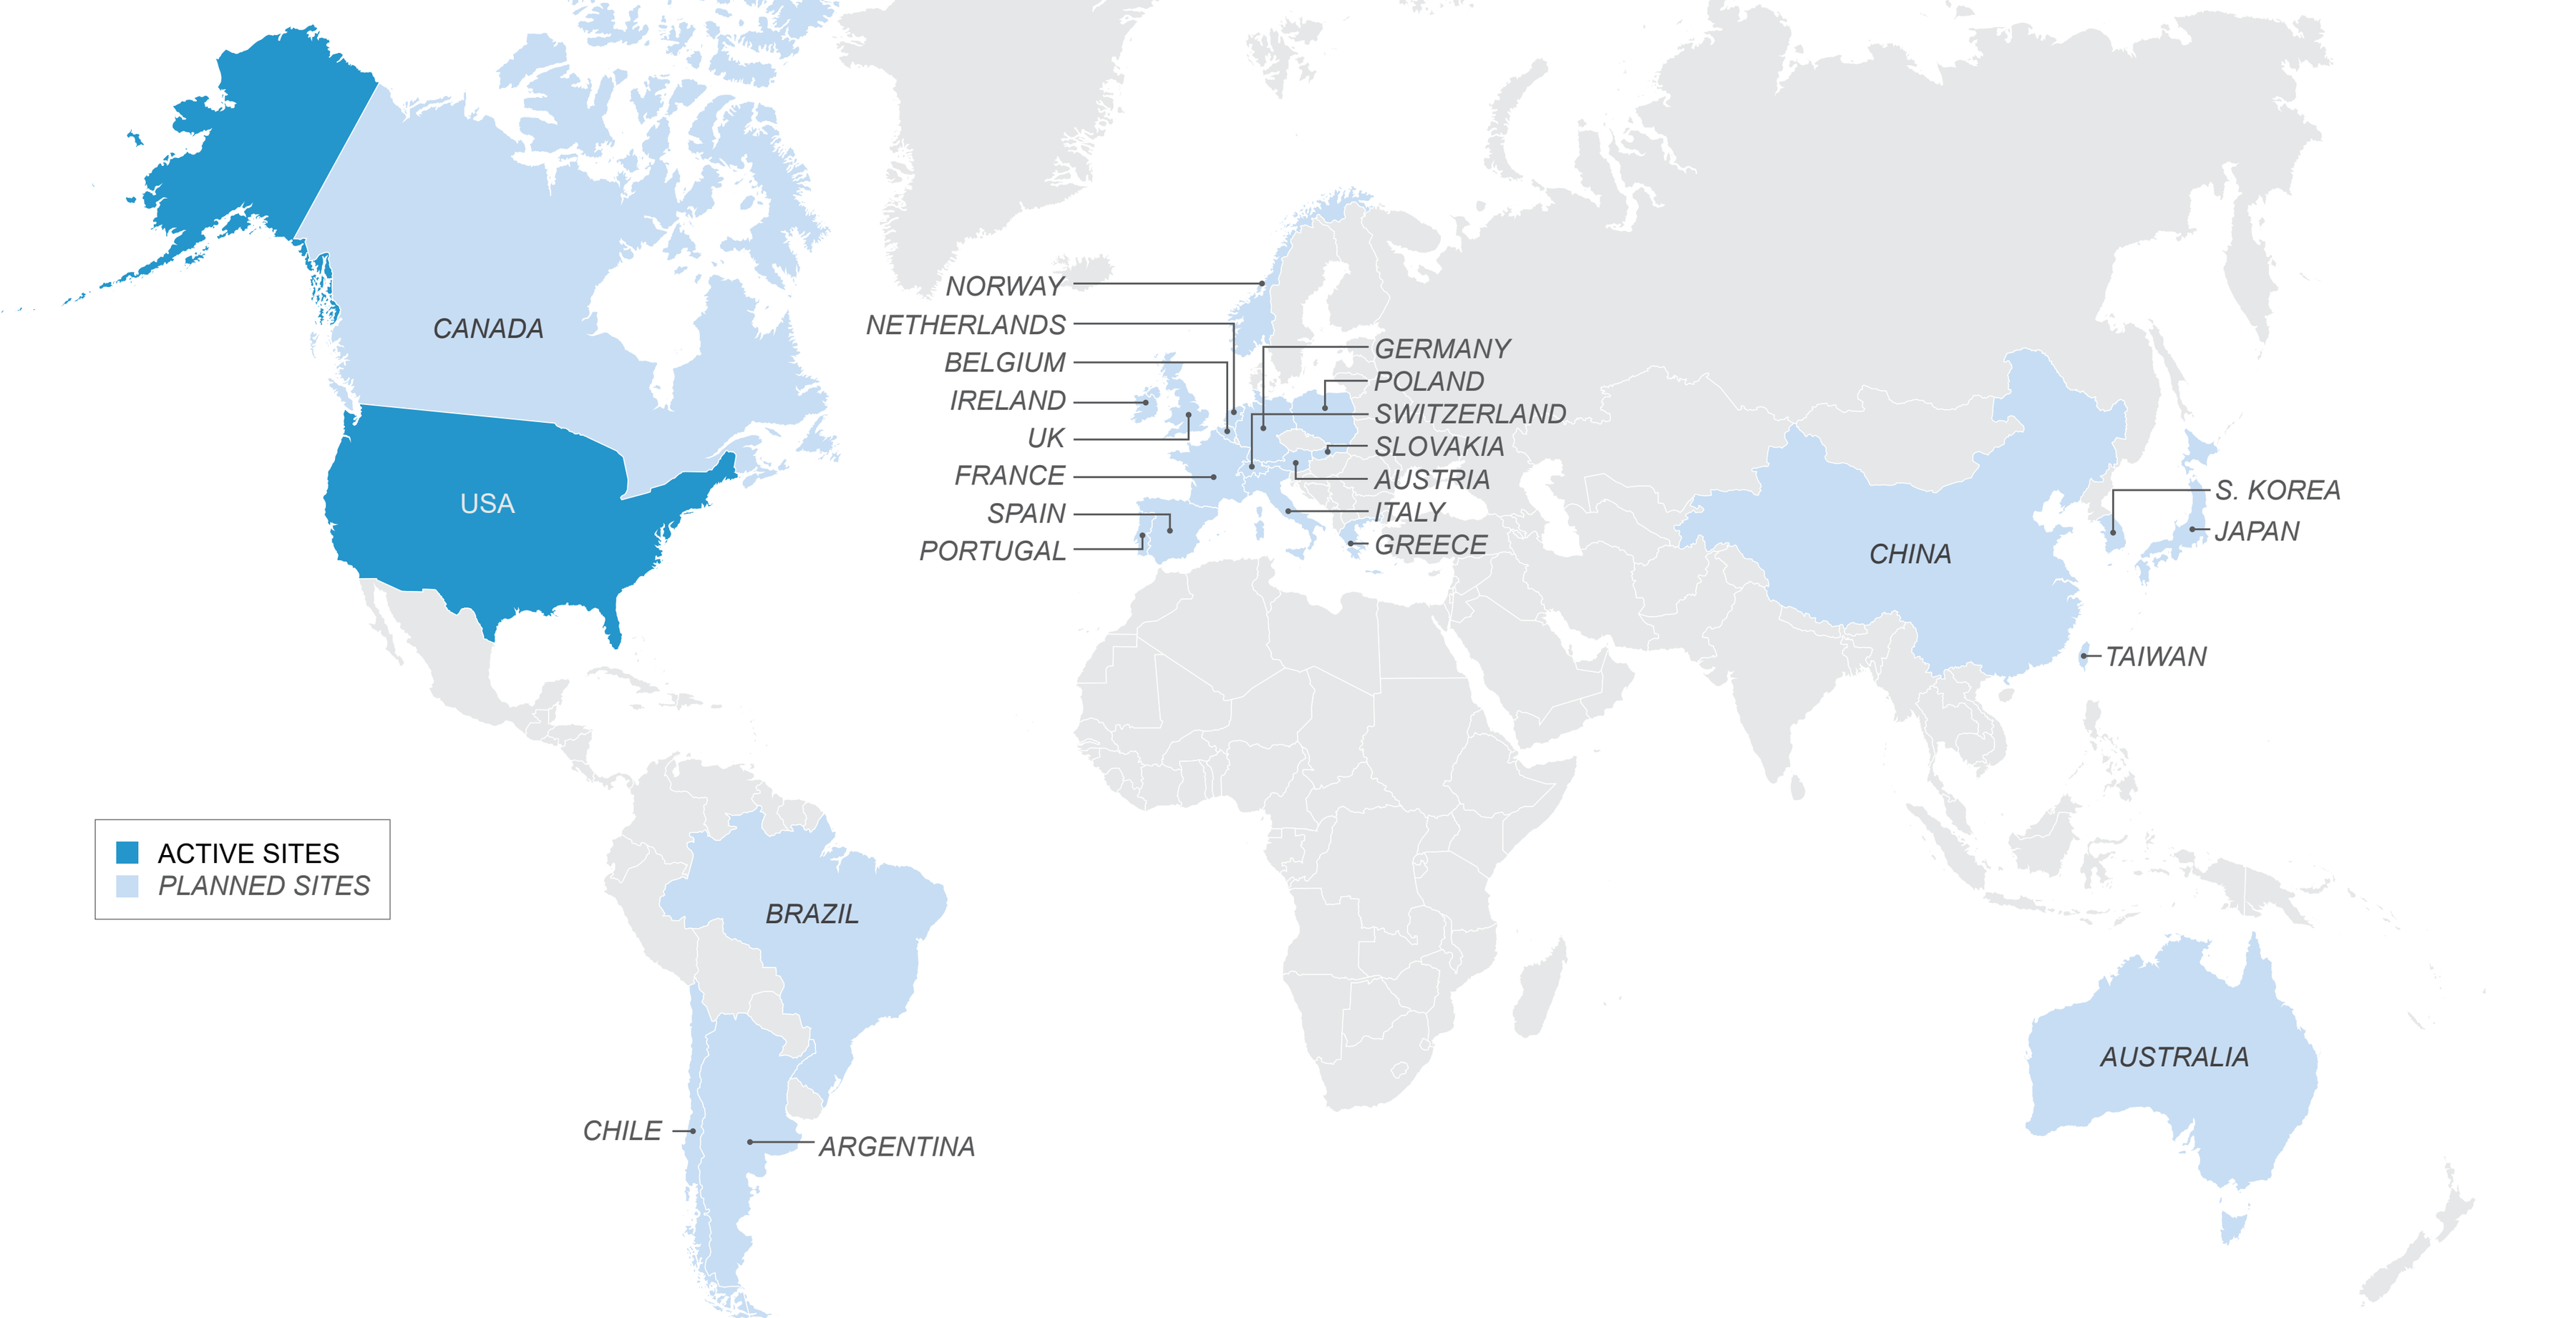
CANADA
USA
NORWAY
NETHERLANDS
BELGIUM
IRELAND
UK
FRANCE
SPAIN
PORTUGAL
GERMANY
POLAND
SWITZERLAND
SLOVAKIA
AUSTRIA
ITALY
GREECE
CHINA
S. KOREA
JAPAN
TAIWAN
BRAZIL
CHILE
ARGENTINA
AUSTRALIA
ACTIVE SITES
PLANNED SITES

Abbreviations

BICR, blinded independent central review; BID, twice per day; BRAF, proto-oncogene B-Raf; cORR, confirmed objective response rate; CT, computed tomography; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance score; EGFR, epidermal growth factor receptor; EORTC QLQ-C30, European Organization for the Research and Treatment of Cancer Quality of Life Questionnaire; EQ-5D-5L, EuroQol 5 dimensions 5 levels; HER2, human epidermal growth factor receptor 2; INV, investigator; mFOLFOX 6, 5-fluorouracil, leucovorin, and oxaliplatin; MRI, magnetic resonance imaging; OS, overall survival; PFS, progression free survival; PFS2, time from randomization to disease progression on next-line treatment or death from any cause; PK, pharmacokinetics; PO, orally; PROs, patient-reported outcomes; Q2W, every 2 weeks; Q3W, every 3 weeks; RAS, rat sarcoma virus; RECIST, Response Evaluation Criteria in Solid Tumors; TKI, tyrosine kinase inhibitor; VEGF, vascular endothelial growth factor; WT, wild-type

Disclosures

This study is sponsored by Seagen Inc., Bothell, WA, USA in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. TA: Invited speaker for Amgen, AstraZeneca, BMS, Merck & Co., Inc, Roche/Ventana, Sanofi, Seagen, Servier; Speaker's Bureau for BMS, Merck & Co., Inc, Servier; Advisory Board for Amgen, Astellas Pharma, BMS, Gristone Oncology, HallioDx, Kaleido Biosciences, Merck & Co., Inc, Pierre Fabre, Seagen, Servier; Research Grant from BMS; Principal Investigator at BMS, GlaxoSmithKline, Merck & Co., Inc, Servier; Honoraria/Consulting or Advisory Role at Amgen, Astellas Pharma, AstraZeneca, BMS, Chugai, Clovis, Gristone Oncology, GlaxoSmithKline, Haliodx, Kaleido Biosciences, Merck & Co., Inc., Pierre Fabre, Roche/Ventana, Sanofi, Seagen, Servier. TBS: Research funding to institution: Agios, Arys, Arcus, Atreca, Boston Biomedical, Bayer, Eisai, Celgene, Lilly, Ipsen, Clovis, Seattle Genetics, Genentech, Novartis, Mirati, Merus, Abgenomics, Incyte, Pfizer, BMS; Consulting to institution: Ipsen, Arcus, Pfizer, Seattle Genetics, Bayer, Genentech, Incyte, Eisai and Merck; Consulting to self: Stemline, AbbVie, Boehringer Ingelheim, Janssen, Daichii Sankyo, Natera, TreosBio, Celularity, Exact Science, Sobi, Beigene, Kanaph, Astra Zeneca, Deciphera, MJH Life Sciences, Aptitude Health, Illumina and Foundation Medicine; IDMC/DSMB: Fibrogen, Suzhou Kintor, Astra Zeneca, Exelixis, Merck/Eisai, PanCan and 1Globe; Scientific Advisory Board: Imugene, Immuneering, Xilis, Replimune Artiva and Sun Biopharma; Royalties: UpToDate; Inventions/Patents: WO/2018/183488: HUMAN PD1 PEPTIDE VACCINES AND USES THEREOF – Licensed to ImugeneWO/2019/055687: METHODS AND COMPOSITIONS FOR THE TREATMENT OF CANCER CACHEXIA – Licensed to Recursion. JT: Scientific consultancy role for Array Biopharma, AstraZeneca, Bayer, Boehringer Ingelheim, Chugai, Daiichi Sankyo, F. Hoffmann-La Roche Ltd, Genentech Inc, HalioDX SAS, Hutchison MediPharma International, Ikena Oncology, Inspirna Inc, IQVIA, Lilly, Menarini, Merck Serono, Merus, MSD, Mirati, Neophore, Novartis, Ona Therapeutics, Orion Biotechnology, Peptomyc, Pfizer, Pierre Fabre, Samsung Bioepis, Sanofi, Scandion Oncology, Scorpion Therapeutics, Seagen, Servier, Sotio Biotech, Taiho, Tessa Therapeutics and TheraMyc; Stocks: Oniria Therapeutics; Educational collaboration with Imedex, Medscape Education, MJH Life Sciences, PeerView Institute for Medical Education and Physicians Education Resource (PER). SS: Consultancy: Amgen, Bayer, BMS, CheckmAb, Clovis Oncology, Daiichi Sankyo, Incyte, Merck, Novartis, Seagen, Roche/Genentech; Equity ownership: Guarddant, Myriad; Patents and Royalties for Amgen; Research funding/Grants: MSD Oncology; Travel Expenses: Amgen, Bayer, Roche. TY: Invited speaker for Bayer, Chugai, Merck Biopharma, MSD, Ono; Grant to Institution from Amgen, Boehringer Ingelheim, Chugai, Daiichi Sankyo, Genomedia, MSD, Ono, Sysmex, Taiho; Local PI, Institutional for Chugai, Daiichi Sankyo, Ono, Pfizer, Sanofi. KN: Employment and Travel Expenses: Merck. DA: Employment: Merck Research Laboratories. JW: Employment and Stock/Equity ownership: Seagen. SY: Employment and Stock/Equity ownership: Seagen. JS: Grants from Amgen, Bayer, Erasca, Seagen, Daiichi-Sankyo, Gossamer Bio, Astar D3, Sanofi, Roche/Genentech, Curgenix, Nektar, Abbvie, Silverback Therapeutics; Consulting fees from Abbvie, Takeda, AstraZeneca, Bayer, GSK, Invivata, Mereo Biopharma, Pfizer, Seagen, Silverback Therapeutics, Viatris; Honoraria from Bayer, Natera, Pfizer; Travel expenses from Seagen, Guardant Health, Monitoring/Advisory Board from Abbvie, Pinoyr Immunotherapeutics; Receipt of services from Guardant Health. EVC: Grants from Amgen, Bayer, Boehringer Ingelheim, BMS, Celgene, Ipsen, Lilly, Merck Sharp & Dohme, Merck KGaA, Novartis, Roche, Servier; Consulting from AbbVie, Array, Astellas, AstraZeneca, Bayer, Beigene, Biocartis, Boehringer Ingelheim, BMS, Celgene, Daiichi, Halozyme, GSK, Helsinn, Incyte, Ipsen, Janssen Research, Lilly, Merck Sharp & Dohme, Merck KGaA, Mirati, Novartis, Pierre Fabre, Roche, Seagen, Servier, Sirtex, Terumo, Taiho, TRIGR, Zymeworks.

Acknowledgements

Thank you to our patients and their families for their participation and to all research personnel for their support of this important trial. Under the guidance of the authors, assistance in medical writing was provided by Sarah Canestaro, MS (contract medical writer) and was funded by Seagen Inc. in accordance with Good Publication Practice (GPP3) guidelines.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors,
Thierry Andre, thierry.andre@aphp.fr

European Society of Medical Oncology; Paris, France; September 9–13, 2022