Understanding drivers of treatment preferences in locally advanced or metastatic urothelial carcinoma: a qualitative interview study with patients, caregivers, and physicians

Andrea B Apolo,[1] Nicholas Simon,[1] Mallory Farrar,[2] Simrun Grewal,[2] Zsolt Hepp,[2] Lisa Mucha,[3] Christine Michaels-Igbokwe,[4] Sebastian Heidenreich,[5] Katelyn Cutts,[4] John L Gore[6]

[1]Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA; [2]Seagen Inc, Bothell, WA, USA; [3]Astellas Pharma Global Development, Inc, Northbrook, IL, USA; [4]Evidera, Montreal, Quebec, Canada; [5]Evidera, London, UK; [6]Department of Urology, University of Washington, Seattle, WA, USA

Background

- Locally advanced or metastatic urothelial carcinoma (la/mUC) is an aggressive malignancy with a 5-year OS of 7.7% in the USA.[1]
- The development and selection of treatments for la/mUC have historically focused on clinical outcomes, but recent FDA guidelines signify a shift towards more direct patient involvement in treatment decisions.
- Consequently, the FDA supports patient-preference research as part of their guidance for patient-focused drug development.[2,3]
- To facilitate the consideration of various perspectives, this study explored factors that may influence the preferences of patients, caregivers, and physicians around aspects of la/mUC treatments.
 - This study reports the initial, qualitative results of a larger study, and future publications will expand on these results with quantitative data from a larger patient sample.

Methods

- A literature review was conducted in line with PRISMA guidelines to inform the interview guides.
 - The review included quantitative and qualitative studies that elicited preferences for medical treatments from patients with la/mUC.
 - To ensure that only contemporary data were captured, manuscripts published before 2011, abstracts published before 2019, and publications reporting only secondary data were excluded.
- Interview guides were developed based on the key concepts identified during the literature review and were designed to elicit perspectives on disease impact (symptoms, HRQoL, and survival), unmet needs, and therapy goals (including ORR).
- Interview guides were used with participating patients, caregivers, and clinicians in qualitative telephone interviews, followed by a treatment choice task.
 - These data then were analyzed to determine the participants' preferences and perspectives.
- Patients with a diagnosis of la/mUC and their primary caregivers (providing ≥8 hours per week of care to the patient) were eligible for inclusion.
- Interviews were conducted with patients with la/mUC diagnosed ≤3 years prior to the study and who were undergoing treatment and with their paired caregivers.
 - Patients residing in hospice care were not eligible for the study.
- Interviews also were conducted with medical oncologists in the USA.
- Physicians were recruited independently from the patients and caregivers and required to have experience in treating patients with la/mUC and to have treated ≥4 patients with la/mUC in the previous 12 months.
- All participants were required to be USA based, aged ≥18 years at enrollment, provide electronic consent, communicate in English, and agree to being audio recorded.
- Qualitative interviews were conducted following the semistructured interview guides. Topics covered included symptom experience, treatment expectations, and treatment decision-making. These data were analyzed qualitatively to identify themes that provided insight into participant preferences for la/mUC treatments.
- Two structured choice tasks, each describing two hypothetical treatments, were developed based on the results of the literature review and on participant responses during interviews. They were conducted with all participants to facilitate discussion about the relative importance of treatment attributes.
- For each task, participants were asked to choose between two hypothetical treatments (A or B) that required tradeoffs between treatment attributes (OS, PFS, cancer pain, and the risk of SAEs).
- Choice task 1:
 - Treatment A: shorter OS and PFS, less severe pain, less chance of SAEs.
 - Treatment B: longer OS and PFS, more severe pain, more chance of SAEs.
- Choice task 2:
 - Treatment A: longer OS and PFS, more severe pain, and more chance of SAEs.
 - Treatment B: shorter OS and PFS, less severe pain, and less chance of SAEs.
- Categorical variables, including symptom frequency, were summarized using frequency statistics (n,%).

Results

Survey participants

- 30 participants took part in the survey:
 - 10 patients: 60% were female; mean age was 58 years; 80% of patients had metastatic disease; 70% were <1 year since diagnosis (100% ≤3 years) and were currently undergoing treatment: previous first-line treatment was cisplatin (80% of patients), carboplatin (20% of patients), and checkpoint inhibitor/immunotherapy (60% of patients); 50% of patients were currently ineligible (physician confirmed) for cisplatin.
 - 10 caregivers: 70% were female; mean age was 50 years; 80% of caregivers were patient's spouse or other family member.
 - 10 physicians: 30% were female; 50% of physicians had an academic affiliation; physicians' mean experience treating la/mUC was 17 years (range: 7-25 years).
 - The most frequently reported symptoms at diagnosis were pain (by 90% of patients and 90% of caregivers) and blood in urine (by 100% of physicians) (**Figure 1**).
 - All three groups reported emotional impacts, with depression/sadness as the most common emotional impact (**Figure 1**).

Figure 1. Patient, caregiver, and physician perspectives on symptoms and QoL

Identified symptoms	Patients	Caregivers	Physicians
Pain	n=9 (90%)	n=9 (90%)	n=8 (80%)
Blood in urine	n=6 (60%)	n=6 (60%)	n=10 (100%)
Fatigue	n=5 (50%)	n=6 (60%)	n=7 (70%)
Frequent urination	n=4 (40%)	n=2 (20%)	n=3 (30%)

Symptom impacts

"A lot of just weakness, I just feel kind of… **depressed**,… **fatigue**… I just don't feel myself." -**Patient 1**

"It started with **back pain**, and she kind of blew it off as, you know, just getting old—lower back pain that would go on, gradually, getting a little bit worse…" -**Caregiver 2**

QoL-identified impacts

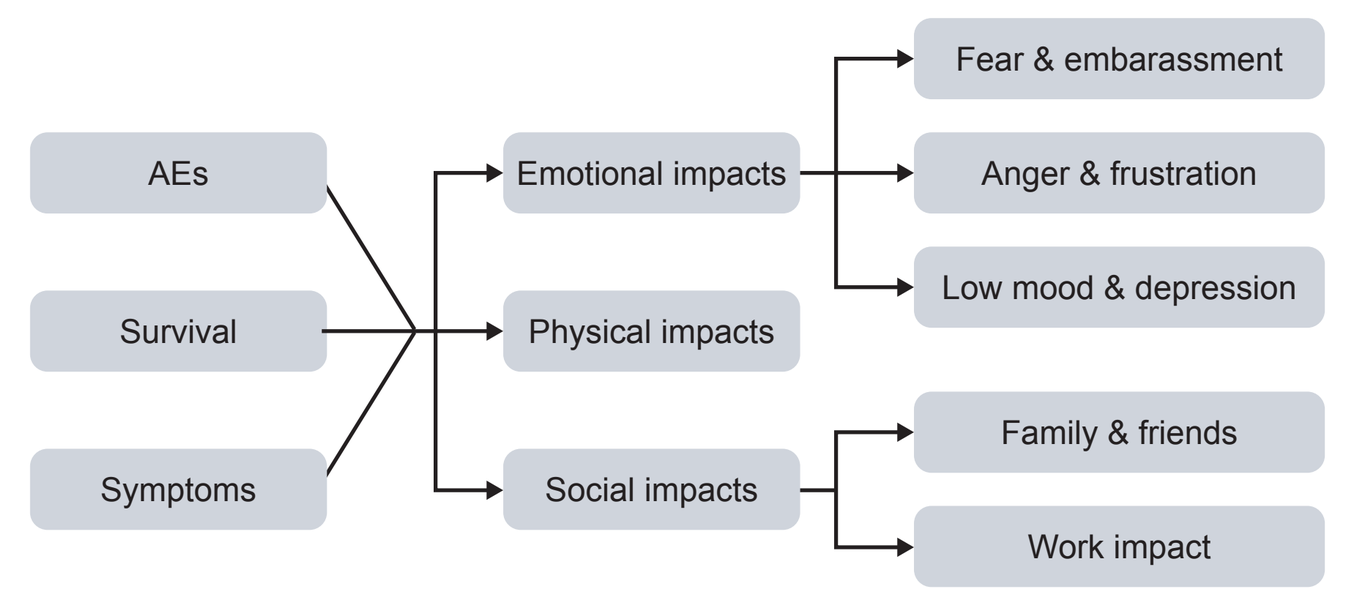
AEs
Survival
Symptoms
Emotional impacts
Physical impacts
Social impacts
Fear & embarassment
Anger & frustration
Low mood & depression
Family & friends
Work impact

QoL impact

"Anything to help prolong, but, for me, **prolonging has to equal quality of life**. You can't… we'll give you an extra year but be miserable with the treatment." -**Caregiver 1**

"It depends on the side effects that they have. It may be that they have a mild-to-moderate symptom, but sometimes in the grand scheme of things, even if **it affects their quality of life**, then that tends to be a problem." -**Clinician 8**

- Patients (n=7) and caregivers (n=7) relied on physicians for decision-making but felt that alternative treatments were not discussed (patients: n=5; caregivers: n=3) (**Figure 2**).

Figure 2. Patient, caregiver, and physician perspectives on treatment expectations, selection, considerations, and interaction with physicians

Treatment expectations

"I expect that the **cancer will not progress**, and … she can hopefully live a more comfortable, happy, and positive life." -**Caregiver 5**

"The improvement I expect from my treatment is… I definitely know it's not going to cure my cancer, but I'm hoping that it will **keep it from spreading**." -**Patient 9**

Treatment selection and considerations

"If you cannot use cisplatin, you are going to go to another agent, either carboplatin or immunotherapy. So you are **losing one line of treatment** that is very effective." -**Clinician 1**

"I still think… **treatment is relatively limited** whether you're cisplatin-eligible or ineligible." -**Clinician 7**

Physician, patient, and caregiver interaction

"…**it wasn't laid out like here's a bunch of options and you can pick; it was more this is what's happening**, this is what your husband needs to do." -**Caregiver 1**

"To be honest, I don't recall. I just remember him stating the chemotherapy, he **recommended for me was the best one**, I don't recall asking anything else." -**Patient 9**

- All three groups described PFS and treatment response as most important or very important for them, and all described the expectation that treatment would improve symptoms and/or improve patient HRQoL.
 - While caregivers and physicians also emphasized OS, patients did not mention it as frequently.
- Physicians focused treatment discussions on AEs (n=8) and ORR (n=6) and, to a lesser extent, survival (n=3).

Perspectives on treatment response rate

- 8 patients and 6 caregivers reported understanding treatment response as important.
- 5 patients and 7 caregivers linked treatment response to the value of a treatment.
- 2 patients and 2 caregivers described response to treatment as stopping the progression of cancer, and 3 caregivers described it as shrinking the tumor.

Hypothetical choice tasks

- In the two hypothetical choice tasks, no single attribute (OS, PFS, pain reduction, and risk of SAEs) was considered the most important by all groups.
- In choice task 1, similar proportions of participants in each group selected each hypothetical choice with no attribute the single most important (**Figure 3**).
- In choice task 2, patients (70%) and physicians (78%) were more likely to select Treatment A, while caregivers were equally likely to select either hypothetical choice (**Figure 4**).
- Overall, each group made attribute trade-offs consistent with an approach that weighs benefits and risks in treatment selection.
- All groups were willing to accept some level of risk of experiencing an AE, but the accepted risk of SAEs varied (patients' accepted risk ranged 0–50%; caregivers' 5–100%; and physicians' 3–30%).

Limitations

- These data are subject to limitations due to the relatively small sample of patients, caregivers, and physicians enrolled in the study.
 - The patient population was predominantly <60 years old and female (60%), which may not reflect the predominantly older male patient population seen in clinical practice.
- Sample sizes also were impacted by some participants not completing all survey instruments; however, this is a common occurrence in qualitative research.

Figure 3. Hypothetical choice task #1

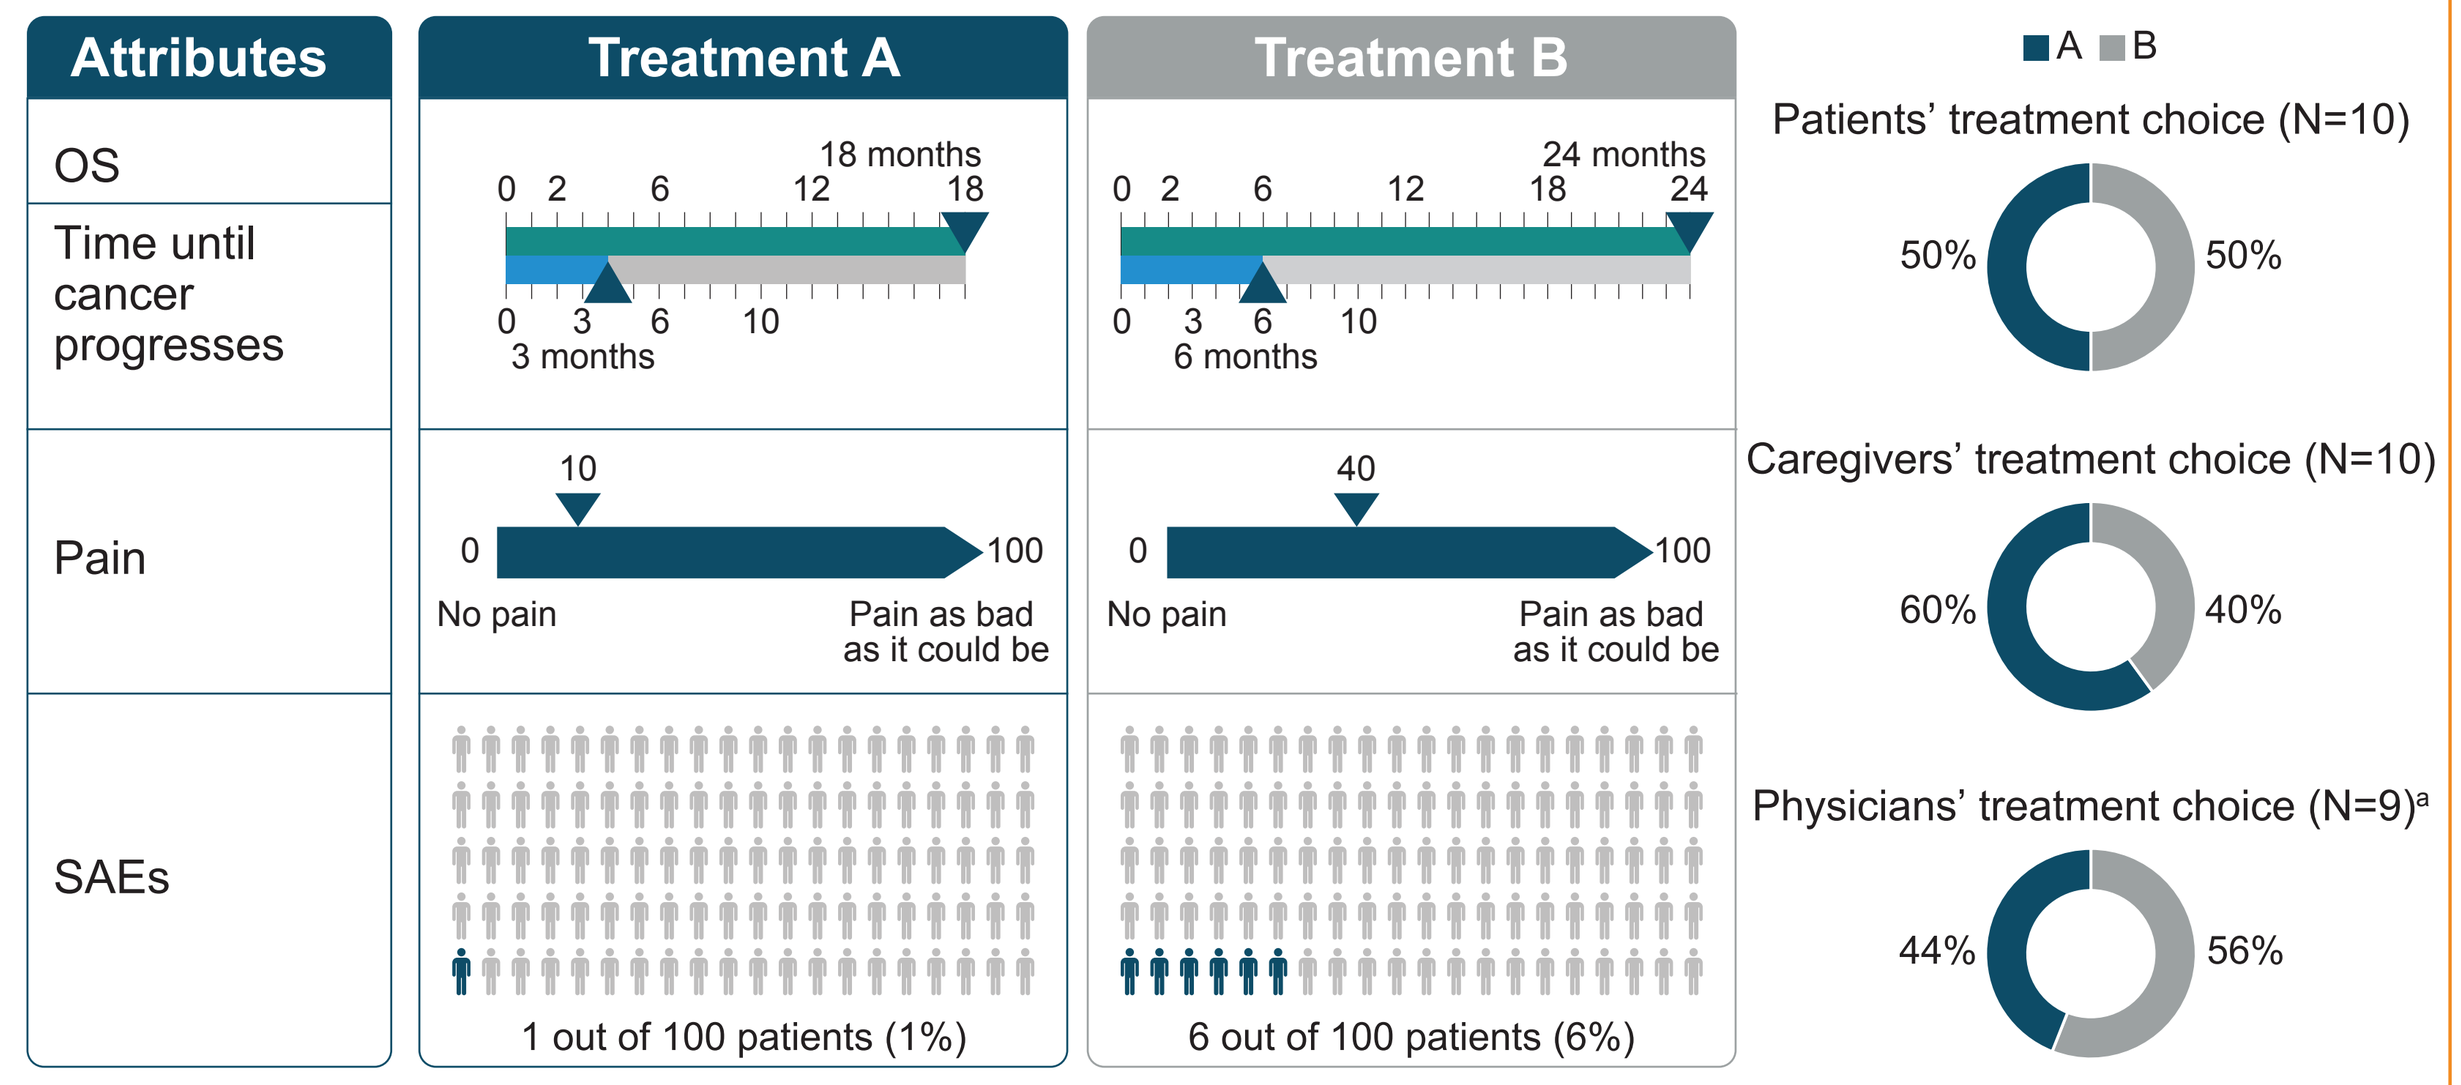
Attributes
Treatment A
Treatment B
OS
Time until cancer progresses
Pain
SAEs
18 months
3 months
24 months
6 months
No pain
Pain as bad as it could be
1 out of 100 patients (1%)
6 out of 100 patients (6%)
A B
Patients' treatment choice (N=10)
50% 50%
Caregivers' treatment choice (N=10)
60% 40%
Physicians' treatment choice (N=9)a
44% 56%

Attribute[b]	Most important to patients[c] (n=8)	Most important to caregivers[c] (n=10)	Most important to physicians[c] (n=8)	Least important to patients (n=6)	Least important to caregivers (n=5)	Least important to physicians (n=9)
Pain	4	5	3	1	1	2
OS	4	3	6	1	0	2
SAEs	3	3	3	2	3	0
PFS	1	2	3	0	1	3
None were least important	—	—	—	2	0	2

[a]One physician said that treatment choice would be dependent on patient age and was unable to make a decision.
[b]As some participants did not choose their most/least important treatment attributes, the N for some attributes totals <10.
[c]Some respondents selected more than one attribute as most important/least important.

Figure 4. Hypothetical choice task #2

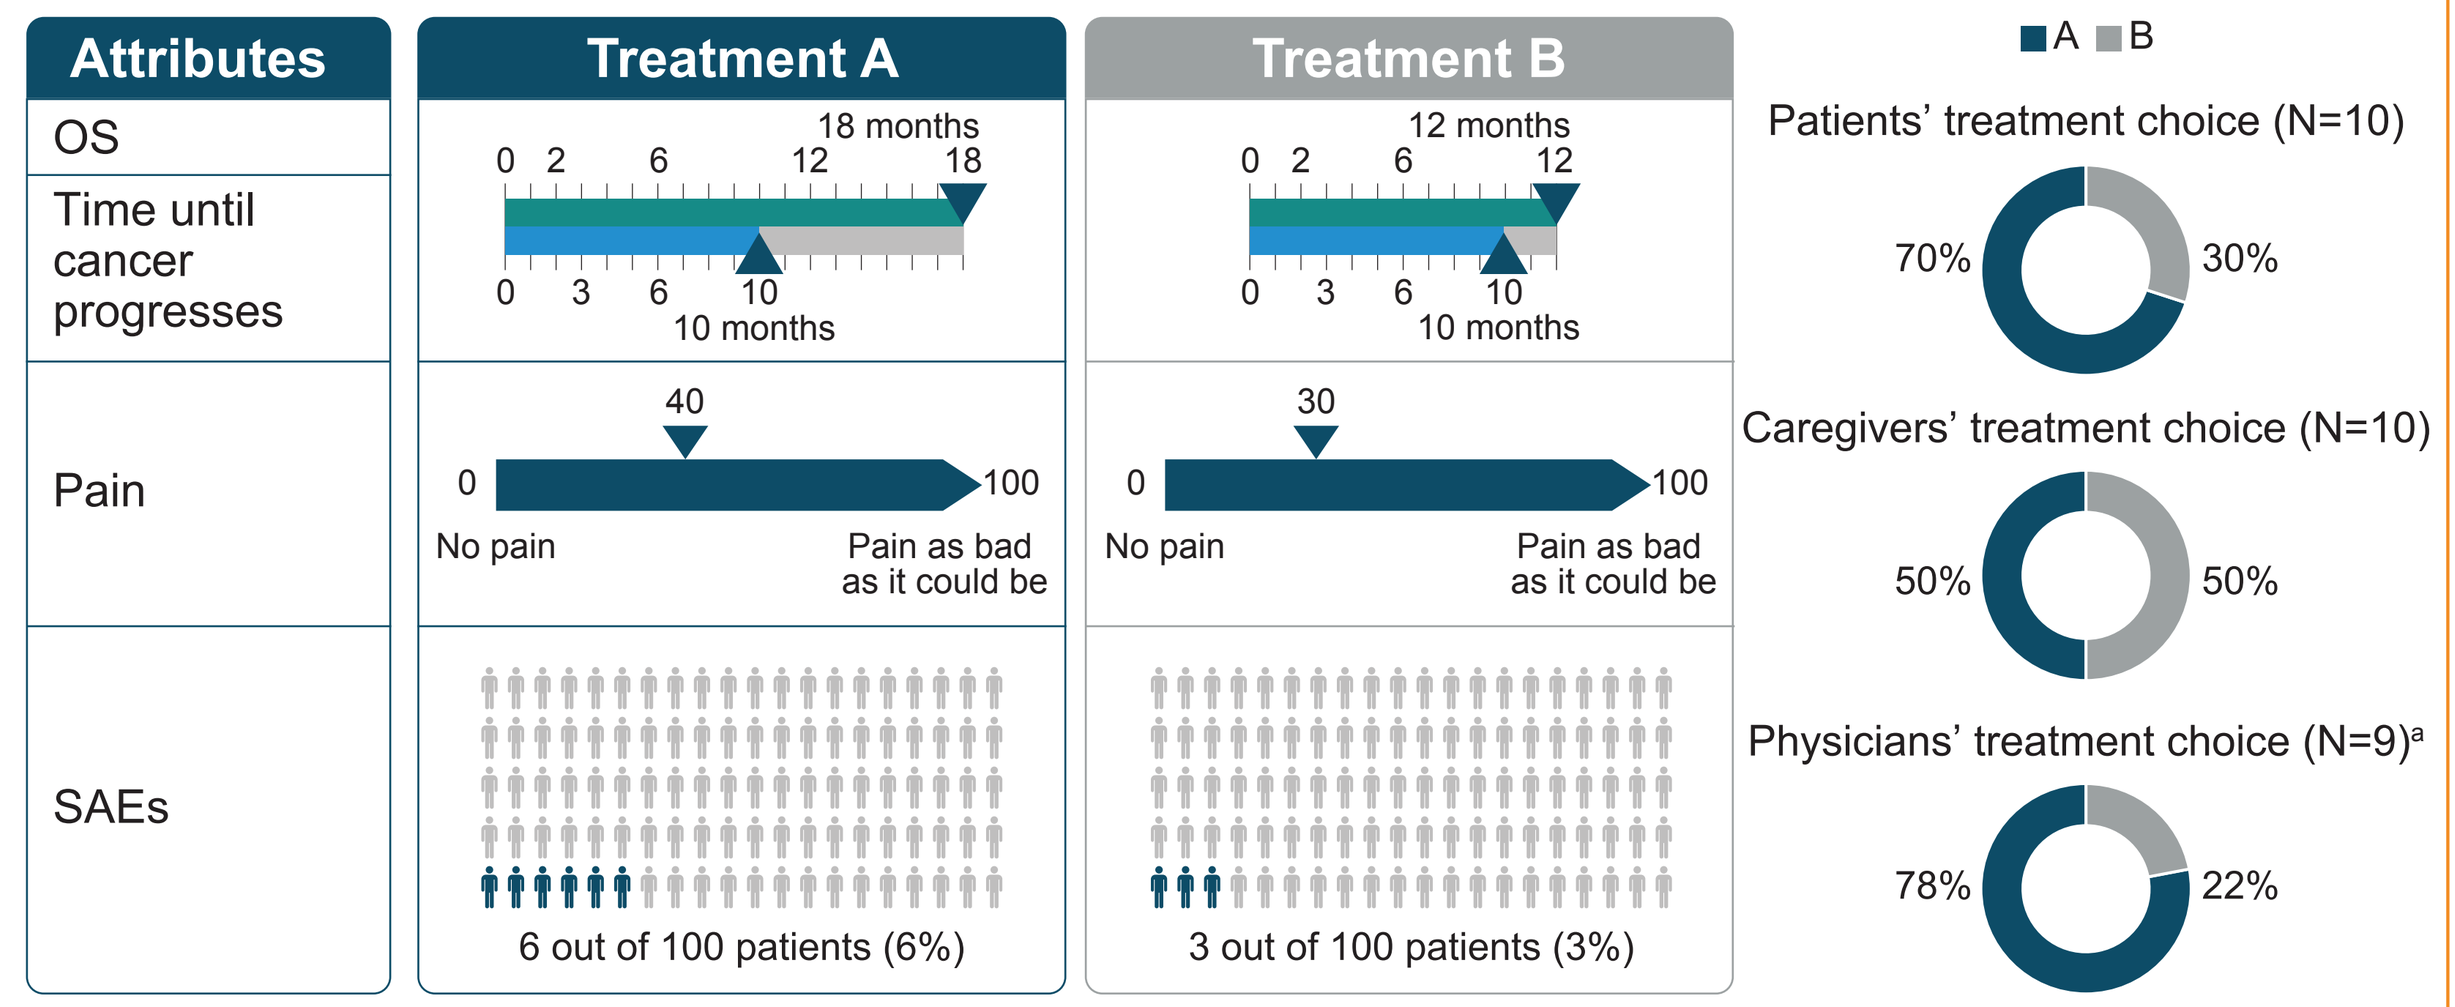
Attributes
Treatment A
Treatment B
OS
Time until cancer progresses
Pain
SAEs
18 months
10 months
12 months
No pain
Pain as bad as it could be
6 out of 100 patients (6%)
3 out of 100 patients (3%)
A B
Patients' treatment choice (N=10)
70% 30%
Caregivers' treatment choice (N=10)
50% 50%
Physicians' treatment choice (N=9)a
78% 22%

Attribute[b]	Most important to patients[c] (n=6)	Most important to caregivers[c] (n=6)	Most important to physicians[c] (n=7)	Least important to patients (n=6)	Least important to caregivers (n=6)	Least important to physicians (n=6)
OS	5	4	6	0	0	0
Pain	2	2	1	2	4	3
SAEs	2	1	2	3	1	0
PFS	1	1	2	0	1	4
None were least important	—	—	—	1	0	0

[a]One physician said that treatment choice would be dependent on patient age and was unable to make a decision.
[b]As some participants did not choose their most/least important treatment attributes, the N for some attributes totals <10.
[c]Some respondents selected more than one attribute as most important/least important.

Conclusions

- HRQoL for patients with la/mUC was found to be impaired by disease-related symptoms as well as emotional, physical, and social impacts.
- In qualitative interviews, physicians, patients, and caregivers all cited PFS and ORR as being most or very important to them.
- In hypothetical choice tasks, caregivers were more likely to prioritize treatments associated with lower pain and lower risk of SAEs than did patients and physicians.
- Factors that influenced treatment choice in la/mUC were heterogeneous and dependent on individual valuation of different treatment aspects, with no single variable equally important to physicians, patients, and caregivers.
- While this study included a relatively small number of patients, future quantitative research informed by these results will expand on these data and further identify which treatment attributes are most important to patients.
 - The quantitative phase will incorporate multidimensional thresholding, with participants ranking potential treatment benefits and risks.
- Treatment selection was found to be driven by physicians, and patients would likely benefit from shared, informed decision-making to identify the treatment option that best aligns with their viewpoint on clinical outcomes, AEs, HRQoL, and pain control.

Abbreviations

AE, adverse event; FDA, US Food and Drug Administration; HRQoL, health-related quality of life; la/mUC, locally advanced or metastatic urothelial carcinoma; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; QoL, quality of life; SAE, severe adverse event.

References

1. National Cancer Institute. Surveillance, Epidemiology, and End Results Program. *Cancer Stat Facts: Bladder Cancer.* National Institutes of Health; 2021. Accessed July 22, 2022. https://seer.cancer.gov/statfacts/html/urinb.html.
2. Food and Drug Administration. *Patient-Focused Drug Development: Collecting Comprehensive and Representative Input: Guidance for Industry, Food and Drug Administration Staff, and Other Stakeholders*; Availability. Food and Drug Administration; 2020. FDA publication FDA-2018-D-1893.
3. Food and Drug Administration, *Patient-Focused Drug Development: Methods to Identify What Is Important to Patients; Guidance for Industry, Food and Drug Administration Staff, and Other Stakeholders.* Food and Drug Administration; 2020. FDA publication FDA-2019-D-4247.

DISCLOSURES: This work was supported by funding from Seagen Inc and Astellas, Inc.
MF, SG, and ZH are employees of, and hold stock in, Seagen Inc.
LM is an employee of Astellas Inc.
CM-I, SH, and KC are employees of Evidera, which received funding from Seagen Inc and Astellas Inc in connection with this study.
JLG has previously received consulting fees from Seagen Inc.
ABA and NS have no conflicts of interest to disclose.
ACKNOWLEDGMENTS: Medical writing support was provided by Philip Ruane of Curo Consulting, a division of Envision Pharma Group, and funded by Seagen Inc.

Corresponding author: Andrea B Apolo (andrea.apolo@nih.gov)

Please scan this Quick Response (QR) code with your smartphone app to view an electronic version of this poster.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

Presented at the American Society of Clinical Oncology (ASCO) Genitourinary Cancers Symposium, San Francisco, CA, USA, February 16–18, 2023
© Copyright 2023