Poster No. OT-03-03
San Antonio Breast Cancer Symposium®
December 8–11, 2020

SGNLVA-001: A PHASE 1 OPEN-LABEL DOSE ESCALATION AND EXPANSION STUDY OF SGN-LIV1A ADMINISTERED WEEKLY IN BREAST CANCER (TRIAL IN PROGRESS)

Heather Beckwith[1], Diana Medgyesy[2], Jame Abraham[3], Rita Nanda[4], Katherine H. R. Tkaczuk[5], Ian Krop[6], Lajos Pusztai[7], Shanu Modi[8], Monica Mita[9], Jennifer Specht[10], Sara Hurvitz[11], Hyo Han[12], Kevin Kalinsky[13], Sharon Wilks[14], Joyce O'Shaughnessy[15], Lowell Hart[16], Hope Rugo[17], Zahi Mitri[18], Phillip M. Garfin[19], Howard Burris[2]

[1]University of Minnesota, Minneapolis, MN; [2]Front Range Cancer Specialists, Fort Collins, CO; [3]National Surgical Adjuvant Breast and Bowel Project Foundation and Cleveland Clinic, Cleveland, OH; [4]The University of Chicago, Chicago, IL; [5]University of Maryland Greenebaum Comprehensive Cancer Center, Baltimore, MD; [6]Dana-Farber Cancer Institute, Boston, MA; [7]Yale University Cancer Center, New Haven, CT; [8]Memorial Sloan Kettering Cancer Center, New York, NY; [9]Cedars-Sinai Comprehensive Cancer Center, Los Angeles, CA; [10]University of Washington, Seattle, WA; [11]David Geffen School of Medicine at UCLA, Los Angeles, CA; [12]Moffitt Cancer Center, Tampa, FL; [13]Herbert Irving Comprehensive Cancer Center, New York, NY; [14]Texas Oncology, San Antonio, TX; [15]Baylor University Medical Center, Texas Oncology, US Oncology, Dallas, TX; [16]Florida Cancer Specialists and Research Institute, Fort Myers, FL; [17]University of California San Francisco Helen Diller Family Comprehensive Cancer Center, San Francisco, CA; [18]Oregon Health and Science University, Portland, OR; [19]Seagen Inc., Bothell, WA; [20]Sarah Cannon Research Institute and Tennessee Oncology, Nashville, TN

Breast Cancer Background

- Breast cancer is the most common malignancy in women in the US.
- Approximately one-third of these patients will eventually develop recurrent or metastatic disease.[4]
- The prognosis is poor in those with metastatic breast cancer (mBC), with a 5-year relative survival of less than 24%.[1]
- The current therapies for metastatic disease delay disease progression but are not curative.
- This is a phase 1 dose-escalation and dose-expansion study to define the safety and tolerability and establish a maximum tolerated dose (MTD) of once weekly (q1wk) ladiratuzumab vedotin (LV) in patients with hormone receptor-positive (HR+)/ human epidermal growth factor receptor 2 (HER2) negative mBC and metastatic triple negative breast cancer (mTNBC).
- LV is an investigational antibody-drug conjugate (ADC) that has been shown to be active and tolerable in mBC at a recommended dose of 2.5 mg/kg every 21 days.[3]
 - More frequent dosing has improved the activity and/or safety of other ADCs.[3]
- More than 200 patients in Parts A–D of this trial have received LV administered q3wk, allowing for accurate pharmacokinetic modeling.
 - PK modeling showed LV dosed weekly resulted in improved safety, efficacy, or both safety and efficacy.

LIV-1

- LIV-1
 - Transmembrane protein that has been found in breast cancer.[6]
 - Expression has been linked with malignant progression to metastasis and is associated with lymph node involvement in breast cancer.[2]

LV Proposed Mechanism of Action

- After binding LIV-1, LV is internalized and trafficked through the endocytic pathway to reach the lysosomes.
- Degradation of the drug linker in lysosomes releases monomethyl auristatin E (MMAE).
- LV-mediated delivery of MMAE drives antitumor activity through:
 - Microtubule disruption, cell cycle arrest, and apoptosis.
 - Inducing ICD.[5]

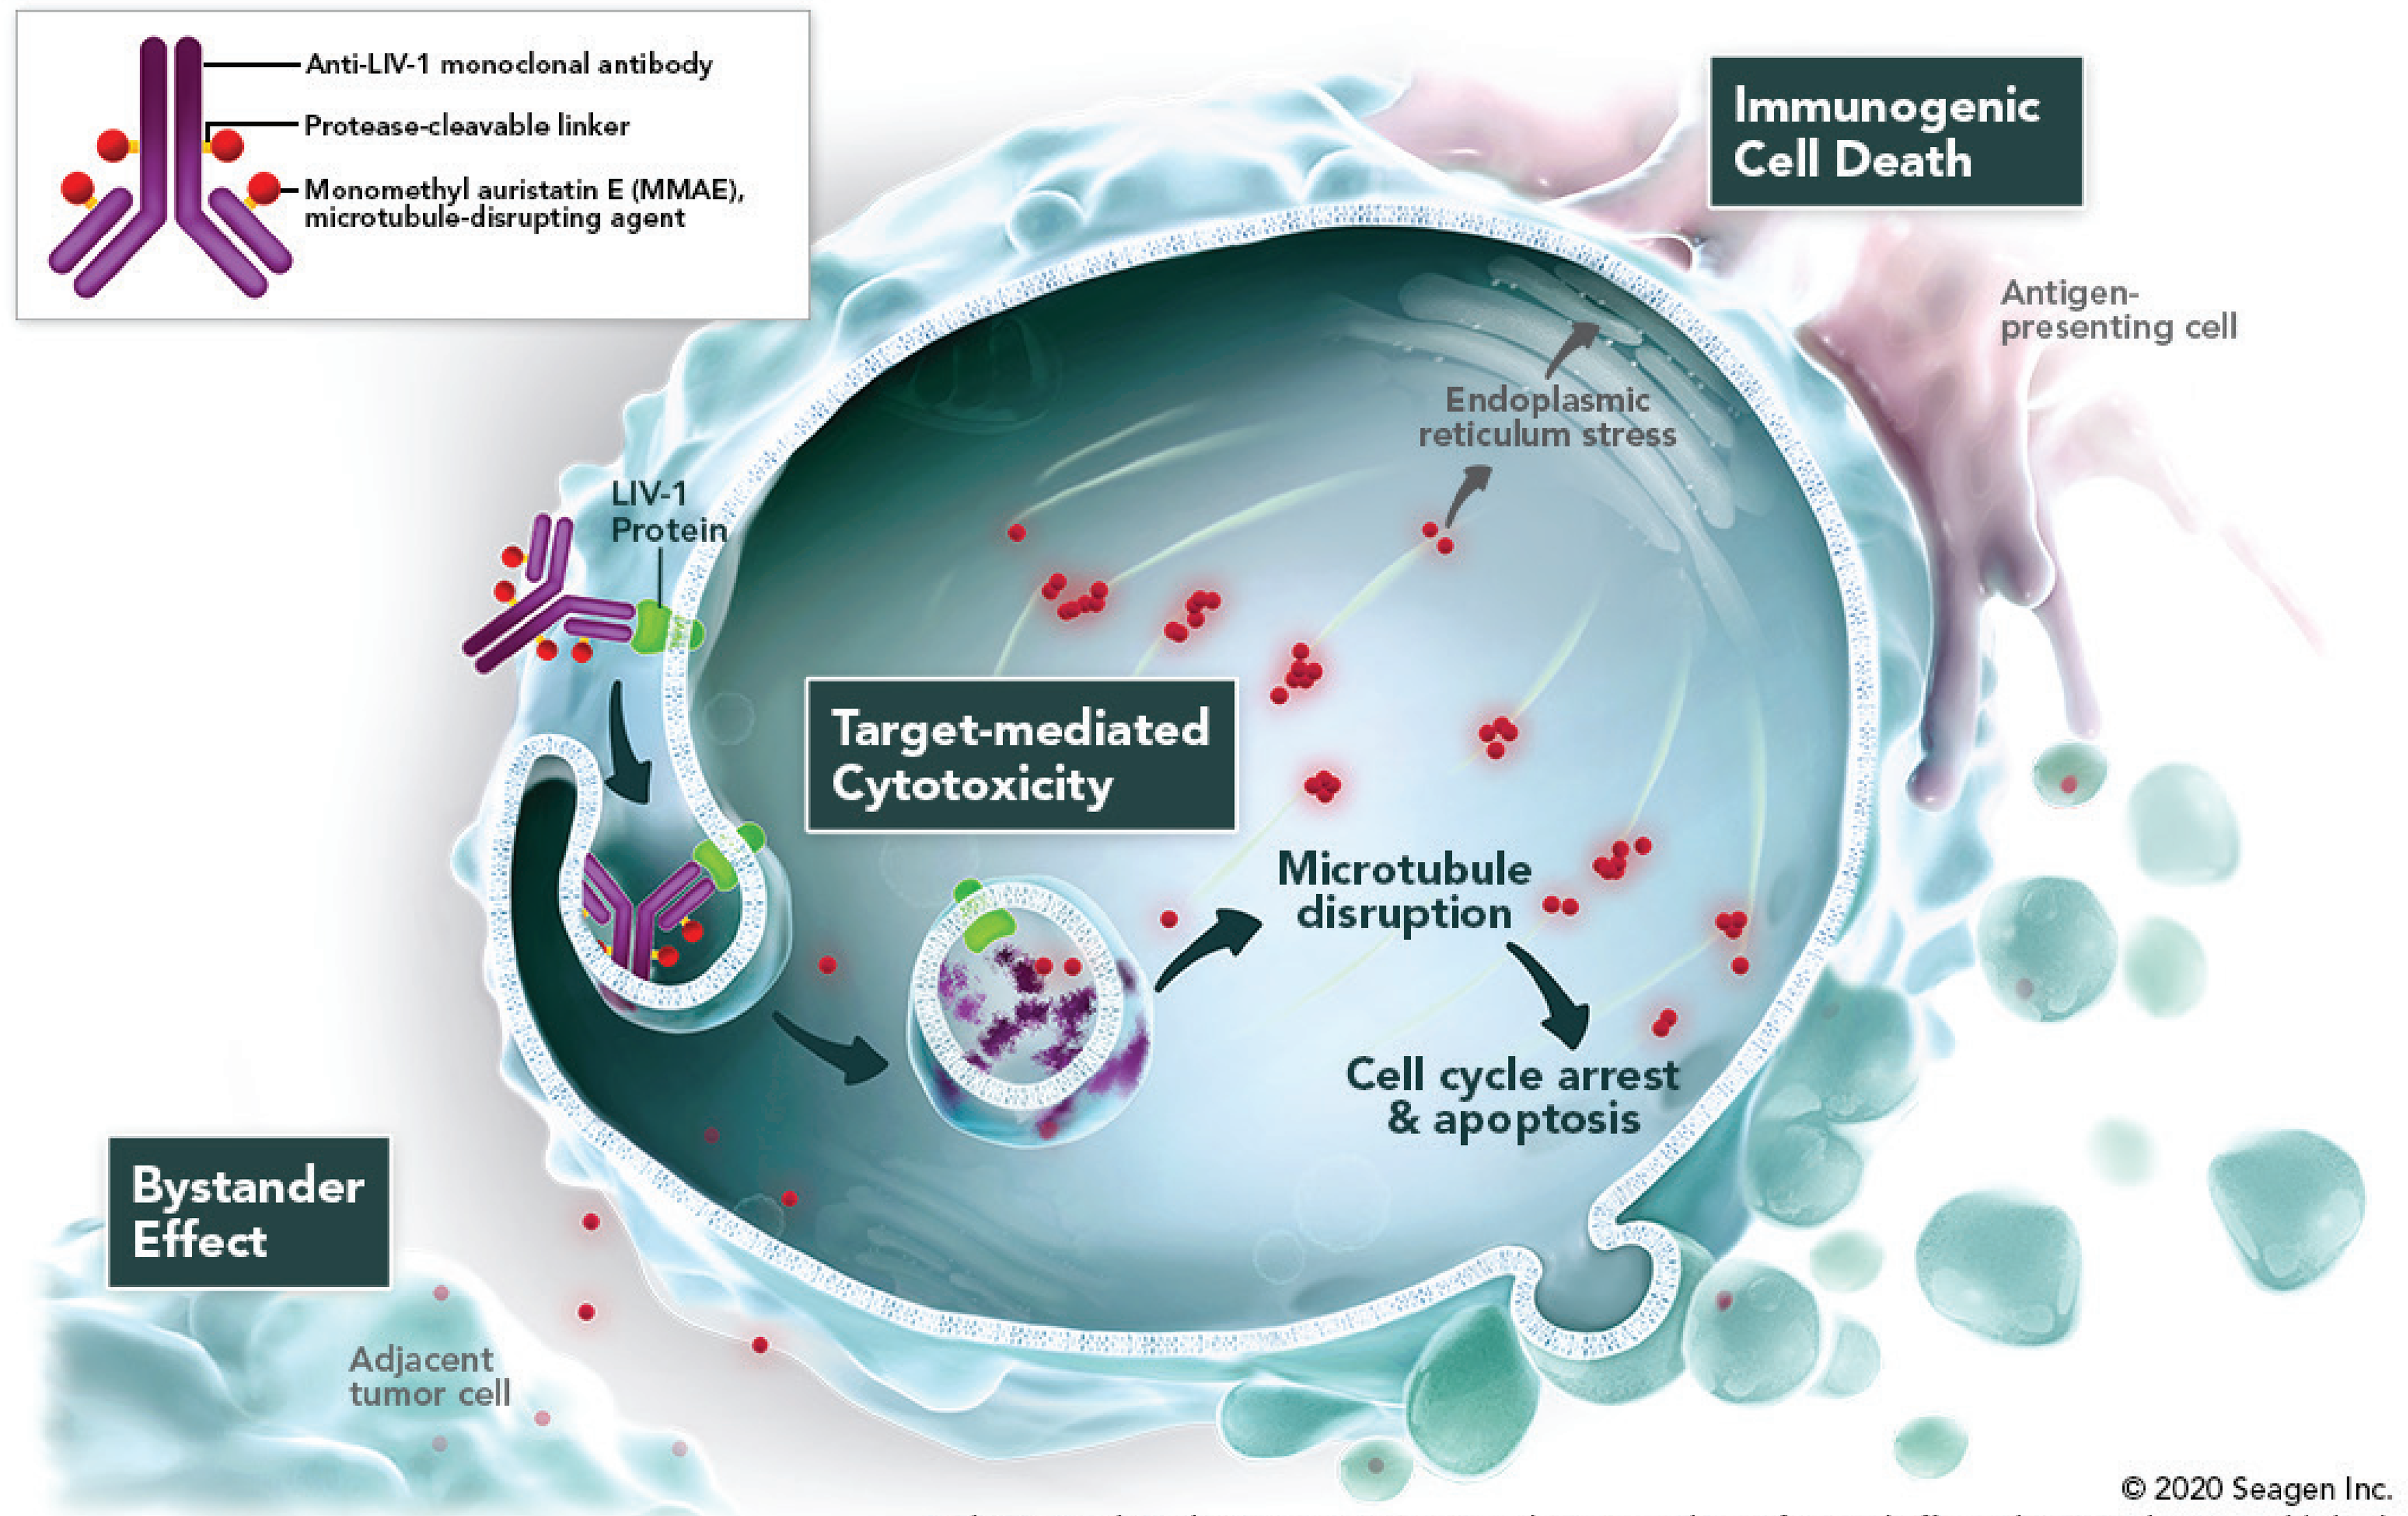
Anti-LIV-1 monoclonal antibody
Protease-cleavable linker
Monomethyl auristatin E (MMAE), microtubule-disrupting agent
Immunogenic Cell Death
Antigen-presenting cell
Endoplasmic reticulum stress
LIV-1 Protein
Target-mediated Cytotoxicity
Microtubule disruption
Cell cycle arrest & apoptosis
Bystander Effect
Adjacent tumor cell

© 2020 Seagen Inc.
Ladiratuzumab Vedotin is an investigational agent, and its safety and efficacy have not been established.

New dose escalation and dose expansion cohorts have opened in Study SGNLVA-001. Patients with endocrine therapy-resistant HR+/HER2-negative metastatic breast cancer and metastatic triple-negative breast cancer may be eligible to receive LV dosed once weekly.

Study Design

- SGNLVA-001 is an ongoing Phase 1 study of LV monotherapy in breast cancer.
- This multipart study is currently enrolling in Part E.
- Part E is a dose-escalation and expansion study of LV monotherapy delivered weekly:

Dose Escalation | HR+/HER2-negative

Dose Level -1	←	Dose Level 0	→	Dose Level 1	→	Dose Level 2	→	Dose Level 3
0.75 mg/kg		1.00 mg/kg		1.25 mg/kg		1.50 mg/kg		1.75 mg/kg

↓ ↓ ↓ ↓ ↓

Dose Level -1	←	Dose Level 0	→	Dose Level 1	→	Dose Level 2	→	Dose Level 3
0.75 mg/kg		1.00 mg/kg		1.25 mg/kg		1.50 mg/kg		1.75 mg/kg

Dose Expansion | HR+/HER2-negative and mTNBC

- The study will accrue up to 82 subjects:
 - 42 HR+/HER2 negative
 - 40 mTNBC

Objectives

Primary Objectives

- To evaluate the safety and tolerability of q1wk dosing of LV in patients with mTNBC and endocrine-resistant ER+ or PR+ (HR+)/HER2-negative mBC.
- To identify the MTD of LV.

Secondary Objectives

- To assess the antitumor activity of LV.
- To assess the pharmacokinetics of LV.
- To assess the immunogenicity of LV.

Endpoints

Safety Endpoints

- Type, incidence, severity, seriousness, and relatedness of adverse events (AEs).
- Type, incidence, and severity of laboratory abnormalities.
- Incidence of dose-limiting toxicity.

Efficacy Endpoints

- ORR, confirmed and unconfirmed.
- Duration of response (DOR).
- Progression-free survival (PFS) and PFS ratio relative to prior therapy.
- Overall Survival (OS).

Pharmacokinetics and Pharmacodynamics

- Markers of pharmacokinetics and pharmacodynamics will be assessed.

Eligibility

Key Inclusion Criteria

- General:
 - Females ≥18 years of age.
 - At least 1 measurable lesion per Response Evaluation Criteria for Solid Tumors (RECIST) v1.1.
 - Eastern Cooperative Oncology Group (ECOG) status of ≤1.
 - Patients with brain metastasis must have received definitive treatment of the lesions.
- HR+/HER2-negative patients:
 - Pathologically confirmed HR+ (ER and PR) disease (ASCO-CAP 2018) HER2-negative (ASCO-CAP 2017) disease.
 - Must have received all appropriate hormonal therapy and not be a candidate for further hormonal therapy.
 - Must have received no or 1 prior line of cytotoxic therapy in the locally advanced (LA)/mBC setting.
- mTNBC patients:
 - Must have pathologically confirmed triple negative disease.
 - Must have received 1 prior line of cytotoxic therapy in the LA/mBC setting.

Key Exclusion Criteria

- Pre-existing neuropathy of Grade ≥2.
- History of another primary invasive malignancy that has not been in remission for at least 3 years.
- Any active Grade ≥3 (per the National Cancer Institute Common Terminology Criteria for Adverse Events Version 4.03 [NCI CTCAE v4.03]) viral, bacterial, or fungal infection within 2 weeks prior to the first dose of LV.
- Positive for hepatitis B by surface antigen expression, active hepatitis C infection or a known history of being seropositive for HIV.
- Documented history of a cerebral vascular event.
- Prior treatment with LV or prior treatment with an MMAE-containing therapy.

Assessments

Safety Assessments

- All patients will be followed for safety.
- Safety will be monitored by the safety monitoring committee during dose escalation and will include:
 - Surveillance and recording of AEs including serious adverse events.
 - Recording of concomitant medication and measurements of protocol-specified physical examination findings and laboratory tests.

Response Assessments

- Response assessed every 2 cycles of treatment in the first ten cycles and every fourth cycle thereafter. Clinical response determined by the investigator at each assessment according to RECIST v1.1. (Clinical response may also be assessed by BICR according to RECIST v1.1.)
- For patients who discontinue treatment prior to disease progression, assessments will be conducted every 6 weeks (±1 week) starting from the most recent tumor assessment until progression or the initiation of a new anticancer treatment.
- All patients will be followed for survival until death or study closure, whichever occurs first. Patients will be followed every 12 weeks (±1 week) for the first year, and then every 24 weeks (±1 week) thereafter.

Study Sites and Completion Dates

- 34 sites across United States.
- Study start: October 22, 2013.
- Estimated study completion: March 31, 2022.

Acknowledgments

- Thank you to our patients and their families for their participation in the study, and to all research personnel for their support of this important trial.
- This study is funded by Seagen Inc.

References

1. Howlader N, et al. SEER Cancer Statistics Review, 1975-2010, National Cancer Institute. Bethesda, MD based on November 2012 SEER data submission, posted to the SEER web site, 2013.
2. Manning DL, et al. Eur J Cancer 1994; 30A(5): 675–8.
3. Modi S, et al. Proceedings of the 2017 San Antonio Breast Cancer Symposium: Abstract PD3-14. 2017.
4. O'Shaughnessy J. Oncologist 2005; 10(Suppl 3): 20-9.
5. Specht J, et al. Ann Oncol 2018; 29(Suppl 8): viii92.
6. Taylor KM, et al. Biochem J 2003; 375(Pt 1): 51-9.

This presentation is the intellectual property of the first author/presenter.
Contact them at **Heather Beckwith, einho003@umn.edu** for permission to reprint and/or distribute.

Seagen®