Discovery of a novel auristatin antibody-drug conjugate drug linker with similar efficacy and reduced bone marrow toxicity compared to vedotin

Noah A. Bindman, Roma Yumul, Erica E. McKinney, Scott Blackburn, Nicole Stevens, Xinqun Zhang, Brendan Drouhard, Sarah Anderson, Calvin Neace, David Ortiz, KC Crowder, Nancy Everds, Chris Carosino, Forgivemore Magunda, Django Sussman, Esther Trueblood, Nicole M. Okeley, Peter Senter

[1]Seagen, Bothell, WA

Background

- Antibody-drug conjugates (ADCs) containing the vedotin drug linker (mc-vcMMAE) have been effective in the clinic across multiple indications, yet off-target release of monomethyl auristatin E (MMAE) can result in dose-limiting bone marrow toxicity and peripheral neuropathy.
- Widening the therapeutic window of vedotin ADCs by maintaining potency in cancer cells while mitigating bone marrow toxicity would be beneficial.
- One approach to decreasing bone marrow toxicity is to develop a peptide drug linker that is preferentially cleaved in cancer cells over bone marrow, thus releasing MMAE selectively in cancer cells.
- We developed an in vitro screen designed to approximate the ability of bone marrow or cancer cells to cleave a short peptide. From this screen we developed a drug linker that preferentially releases MMAE in cancer cells upon cleavage of the tripeptide DLeu-Ala-Glu.

Discovery of DLeu-Ala-Glu (DLAE) tripeptide

- The DLAE tripeptide was discovered via a fluorescence screen assessing proteolysis of tripeptides against cancer cells (HPAFII) and normal human bone marrow homogenates.

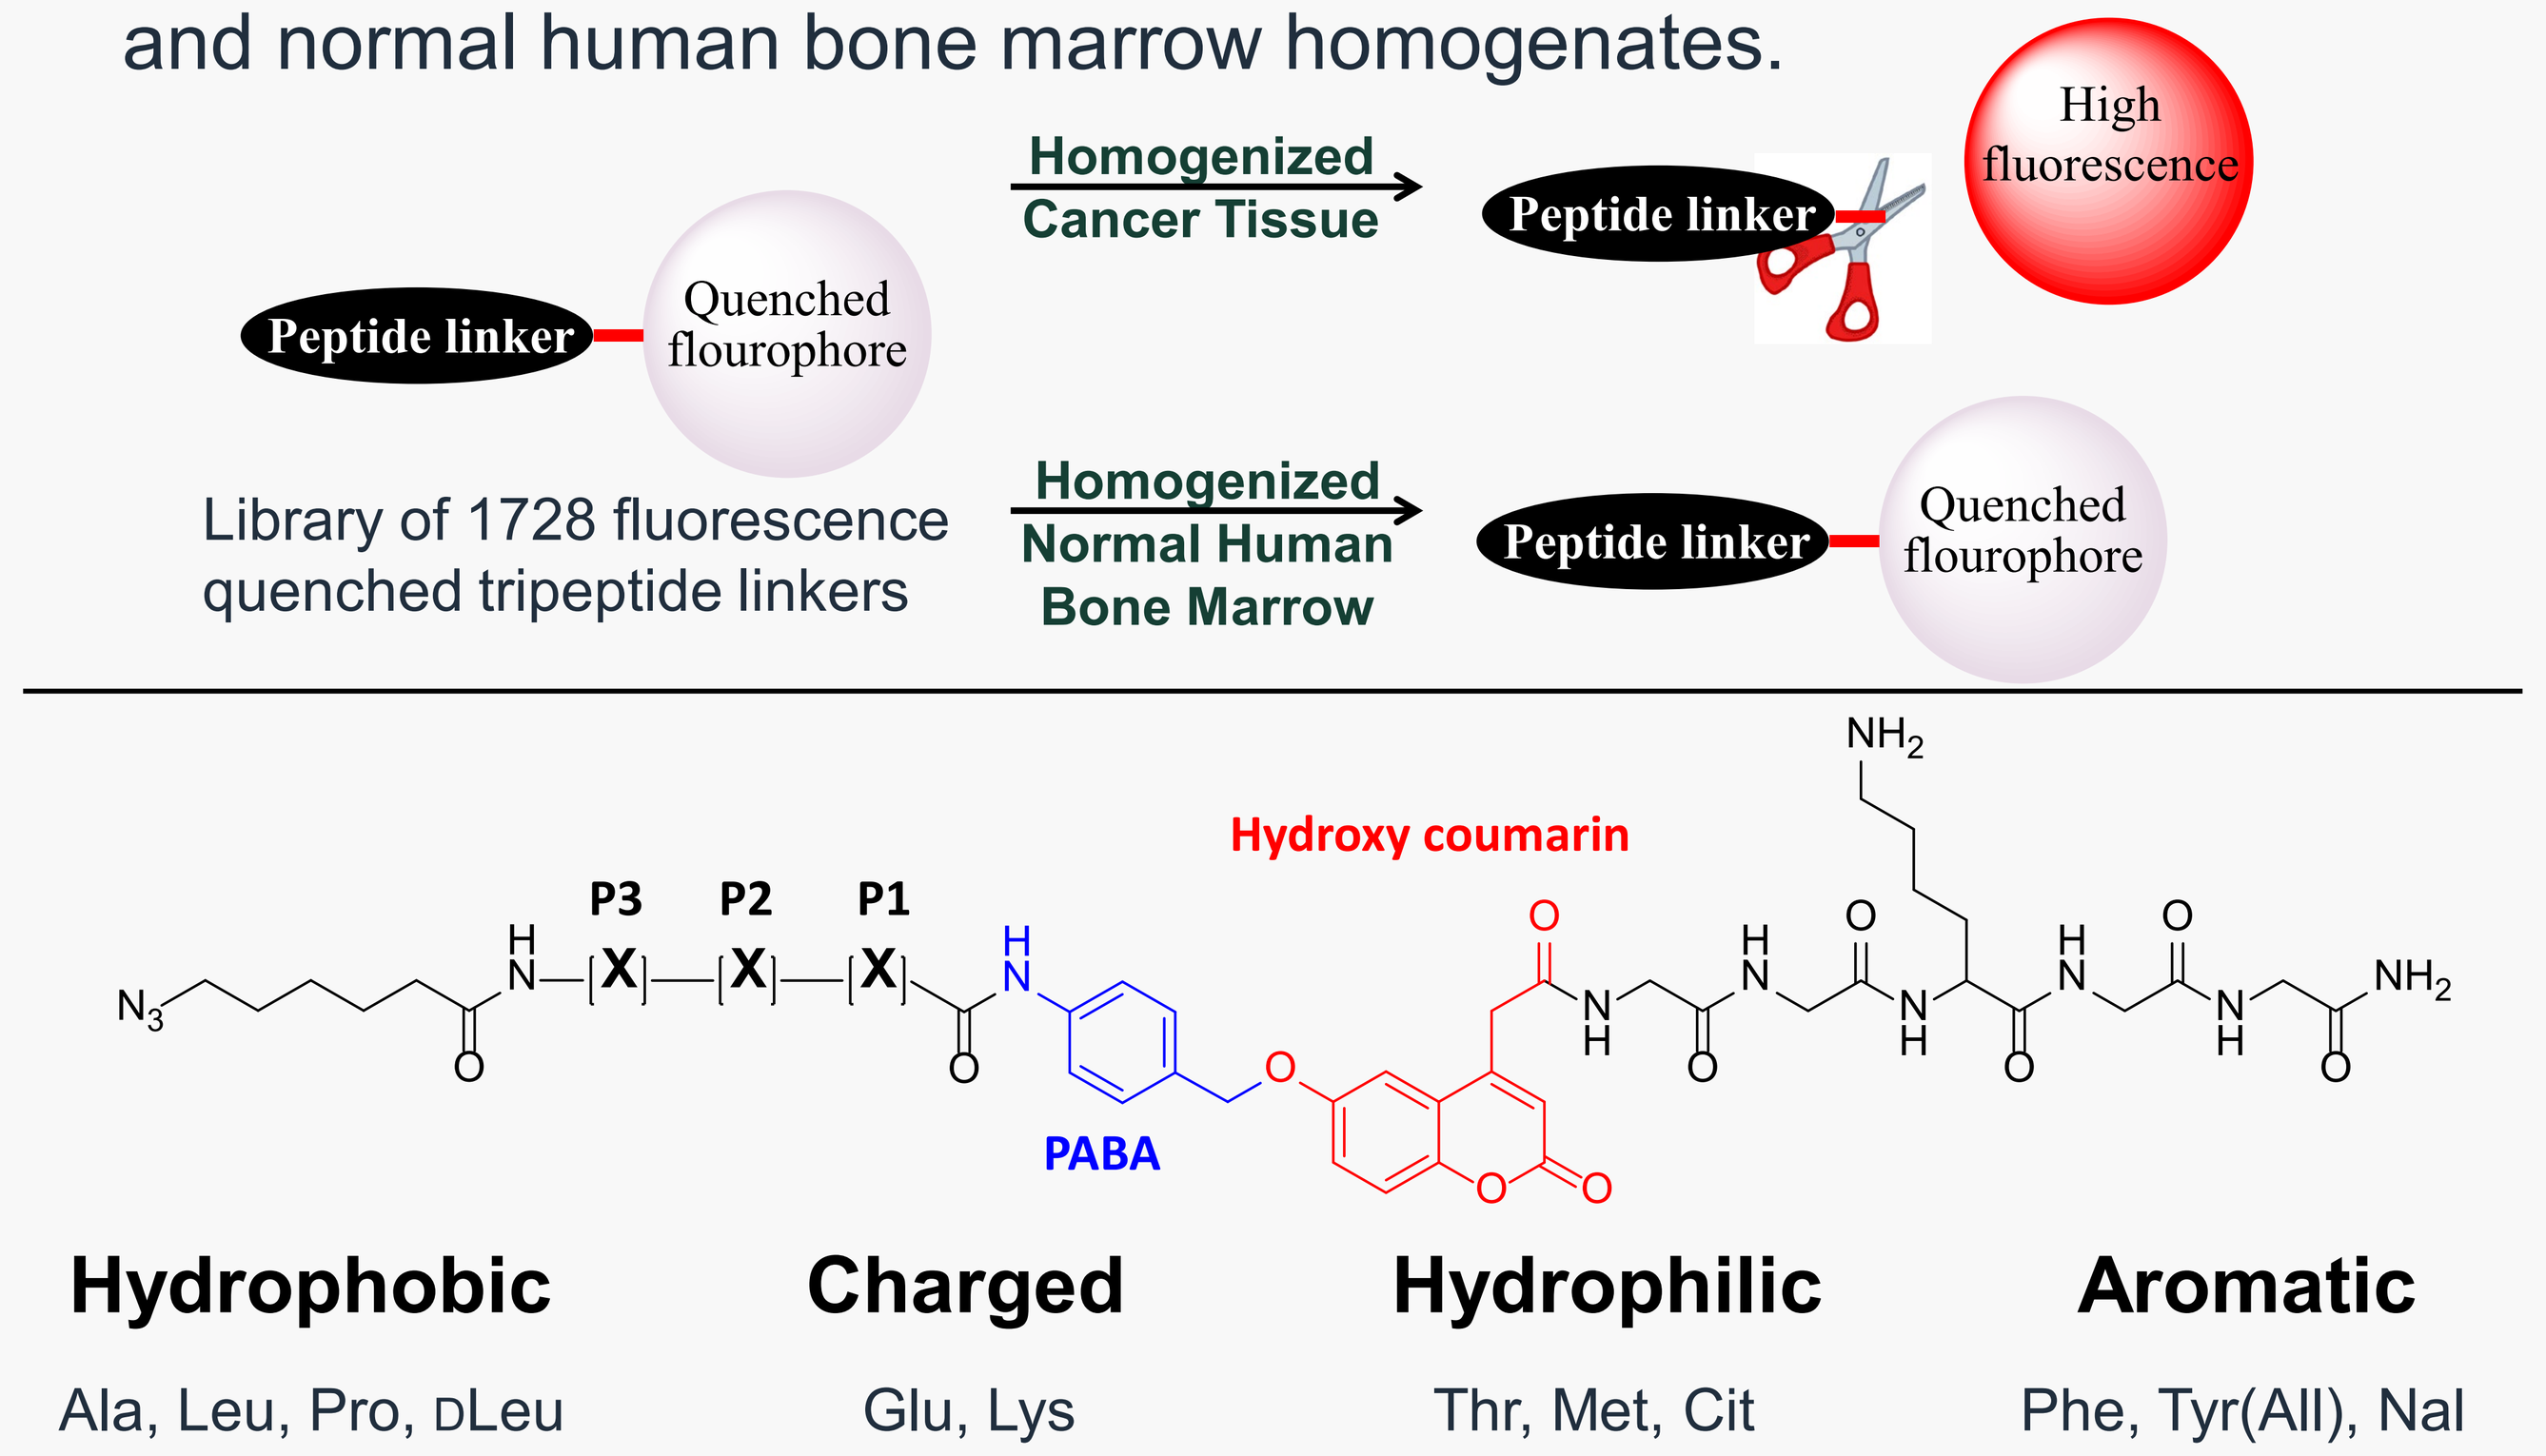
Peptide linker
Quenched flourophore
Library of 1728 fluorescence quenched tripeptide linkers
Homogenized Cancer Tissue
Peptide linker
High fluorescence
Homogenized Normal Human Bone Marrow
Peptide linker
Quenched flourophore
Hydroxy coumarin
P3
P2
P1
PABA
Hydrophobic
Charged
Hydrophilic
Aromatic
Ala, Leu, Pro, DLeu
Glu, Lys
Thr, Met, Cit
Phe, Tyr(All), Nal

Figure 1. **Top**, Schematic of fluorescence assay designed to identify tripeptides preferentially cleaved in cancer cells over bone marrow cells. **Bottom,** Structure of fluorescence quenched tripeptide library. Hydroxy coumarin becomes fluorescent upon proteolysis between P1 and PABA position. Unnatural amino acids are D-Leucine (DLeu), Citrulline (Cit), O-Allyl-Tyrosine (Tyr(All)), and 2-Naphthylalanine (Nal).

Tripeptide Seq[2]	Relative Fluorescence[1] HPAF-II	Relative Fluorescence[1] Human BM	HPAF-II/Human BM Ratio	Reduced BM toxicity in rat as MMAE ADC compared to Vedotin[3]
Ala-Val-Cit	**1.9**	**1.6**	**1.2**	**No**
DLeu-Ala-Glu	**1.9**	**0.9**	**2.0**	**Yes**
DLeu-Leu-Lys	1.5	1.0	1.4	No
DLeu-Leu-Met(O)	1.3	0.8	1.6	No
DLeu-Leu-Cit	2.9	1.4	2.0	No
Cit-Nal-Thr	0.4	0.6	0.6	No
Pro-Nal-Lys	1.9	0.7	1.9	No

Table 1. Fluorescence fold change and rat bone marrow toxicity of several tripeptide coumarin and MMAE conjugates. [1] Fluorescence fold change over average peptide in screen. [2] Natural amino acids are abbreviated with their traditional three letter codes (Cit = citrulline, Met(O) = methionine sulfoxide, Nal = 2-naphthylalanine. [3] Vedotin-like linkers were synthesized with the Val-Cit dipeptide substituted for a tripeptide and conjugated to a non-targeting hIgG1 antibody as a mixed 4-load. Bone marrow toxicity was assessed after a single dose in rats at 10 mg/kg.

Preclinical comparison of DLAE-MMAE and vedotin ADCs

DLAE-MMAE exhibits similar cytotoxicity to vedotin against cancer cells

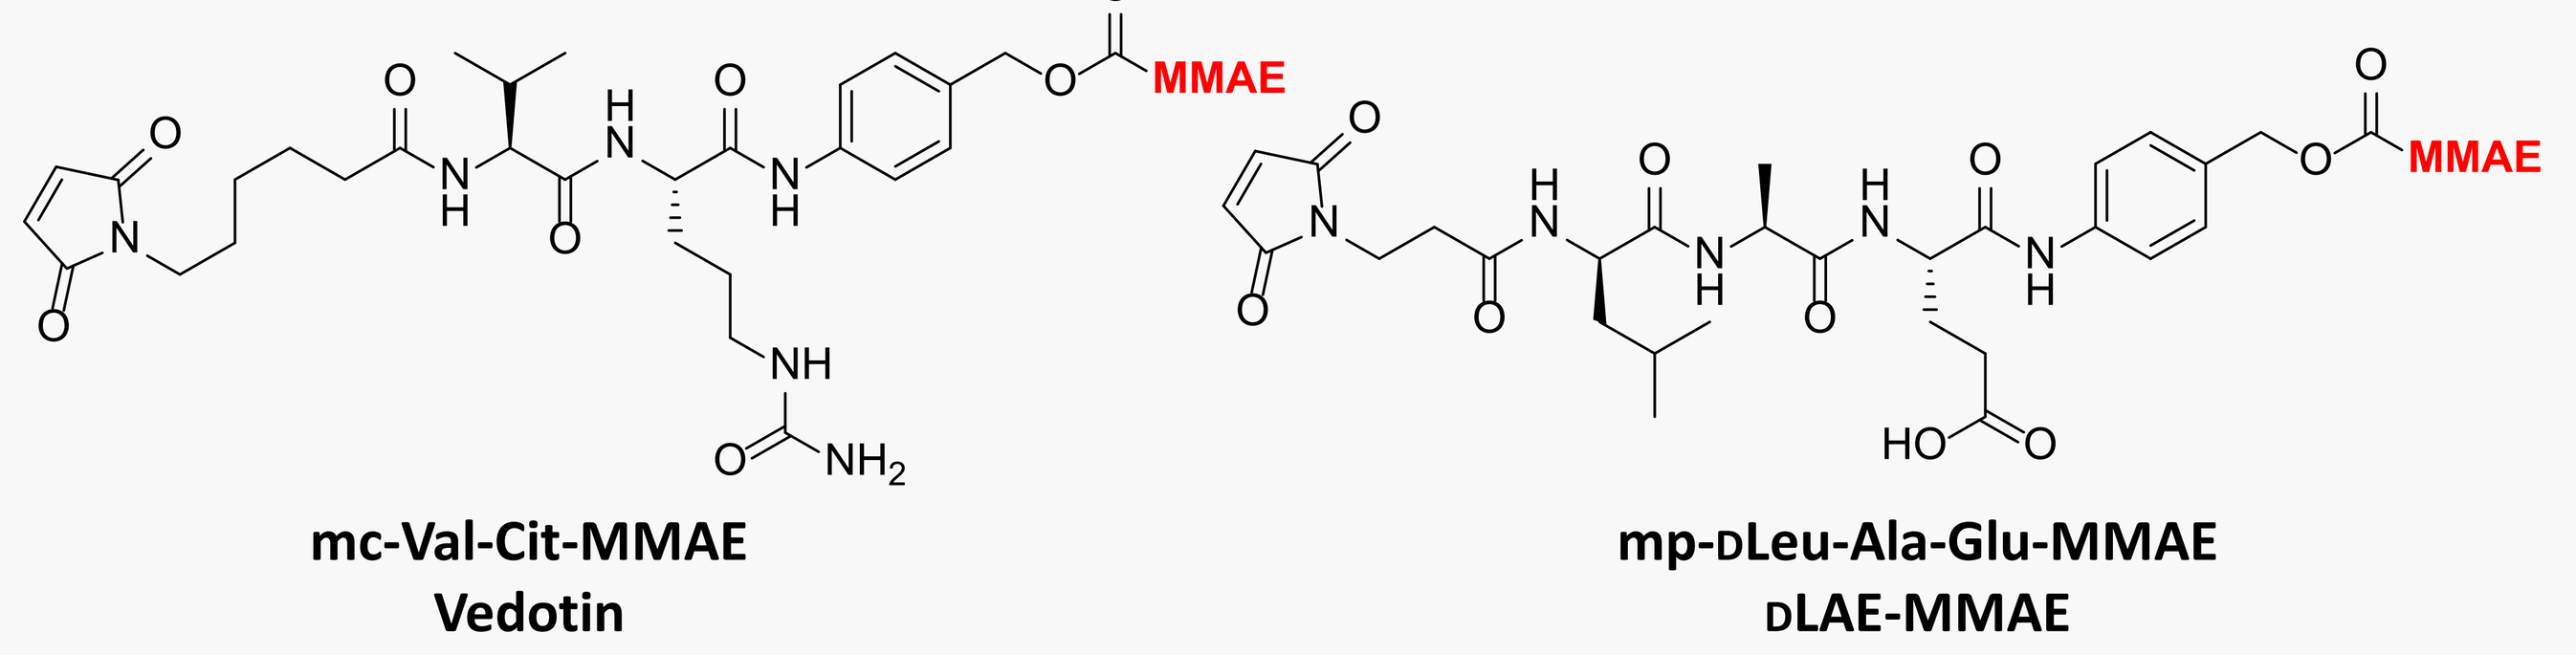
MMAE
MMAE
mc-Val-Cit-MMAE
Vedotin
mp-DLeu-Ala-Glu-MMAE
DLAE-MMAE

ADC	BxPC3[1]		HL-60		L540cy		MM.1R		MOLM-13		SK-MEL-5		SU-DHL-4	
cOKT9-vedotin	15	8	72	0	19	9	6	1	96	1	18	14	4	5
cOKT9-DLAE-MMAE	15	10	84	3	12	8	6	2	163	0	11	18	3	2

Table 2: Cytotoxicity, against several cancer cell lines, of DAR4 DLAE-MMAE and vedotin ADCs conjugated to the anti-CD71 antibody cOKT9. [1]Cytotoxicity values in blue denote IC50s (ng/mL); Values in white denote % live cells remaining at maximum concentration of ADC. Cell lines encompass pancreatic adenocarcinoma, AML, lymphoma, multiple myeloma, and melanoma.

Compared to vedotin, DLAE-MMAE has reduced proteolysis by bone marrow proteases, a potential mechanism for its reduced bone marrow toxicity

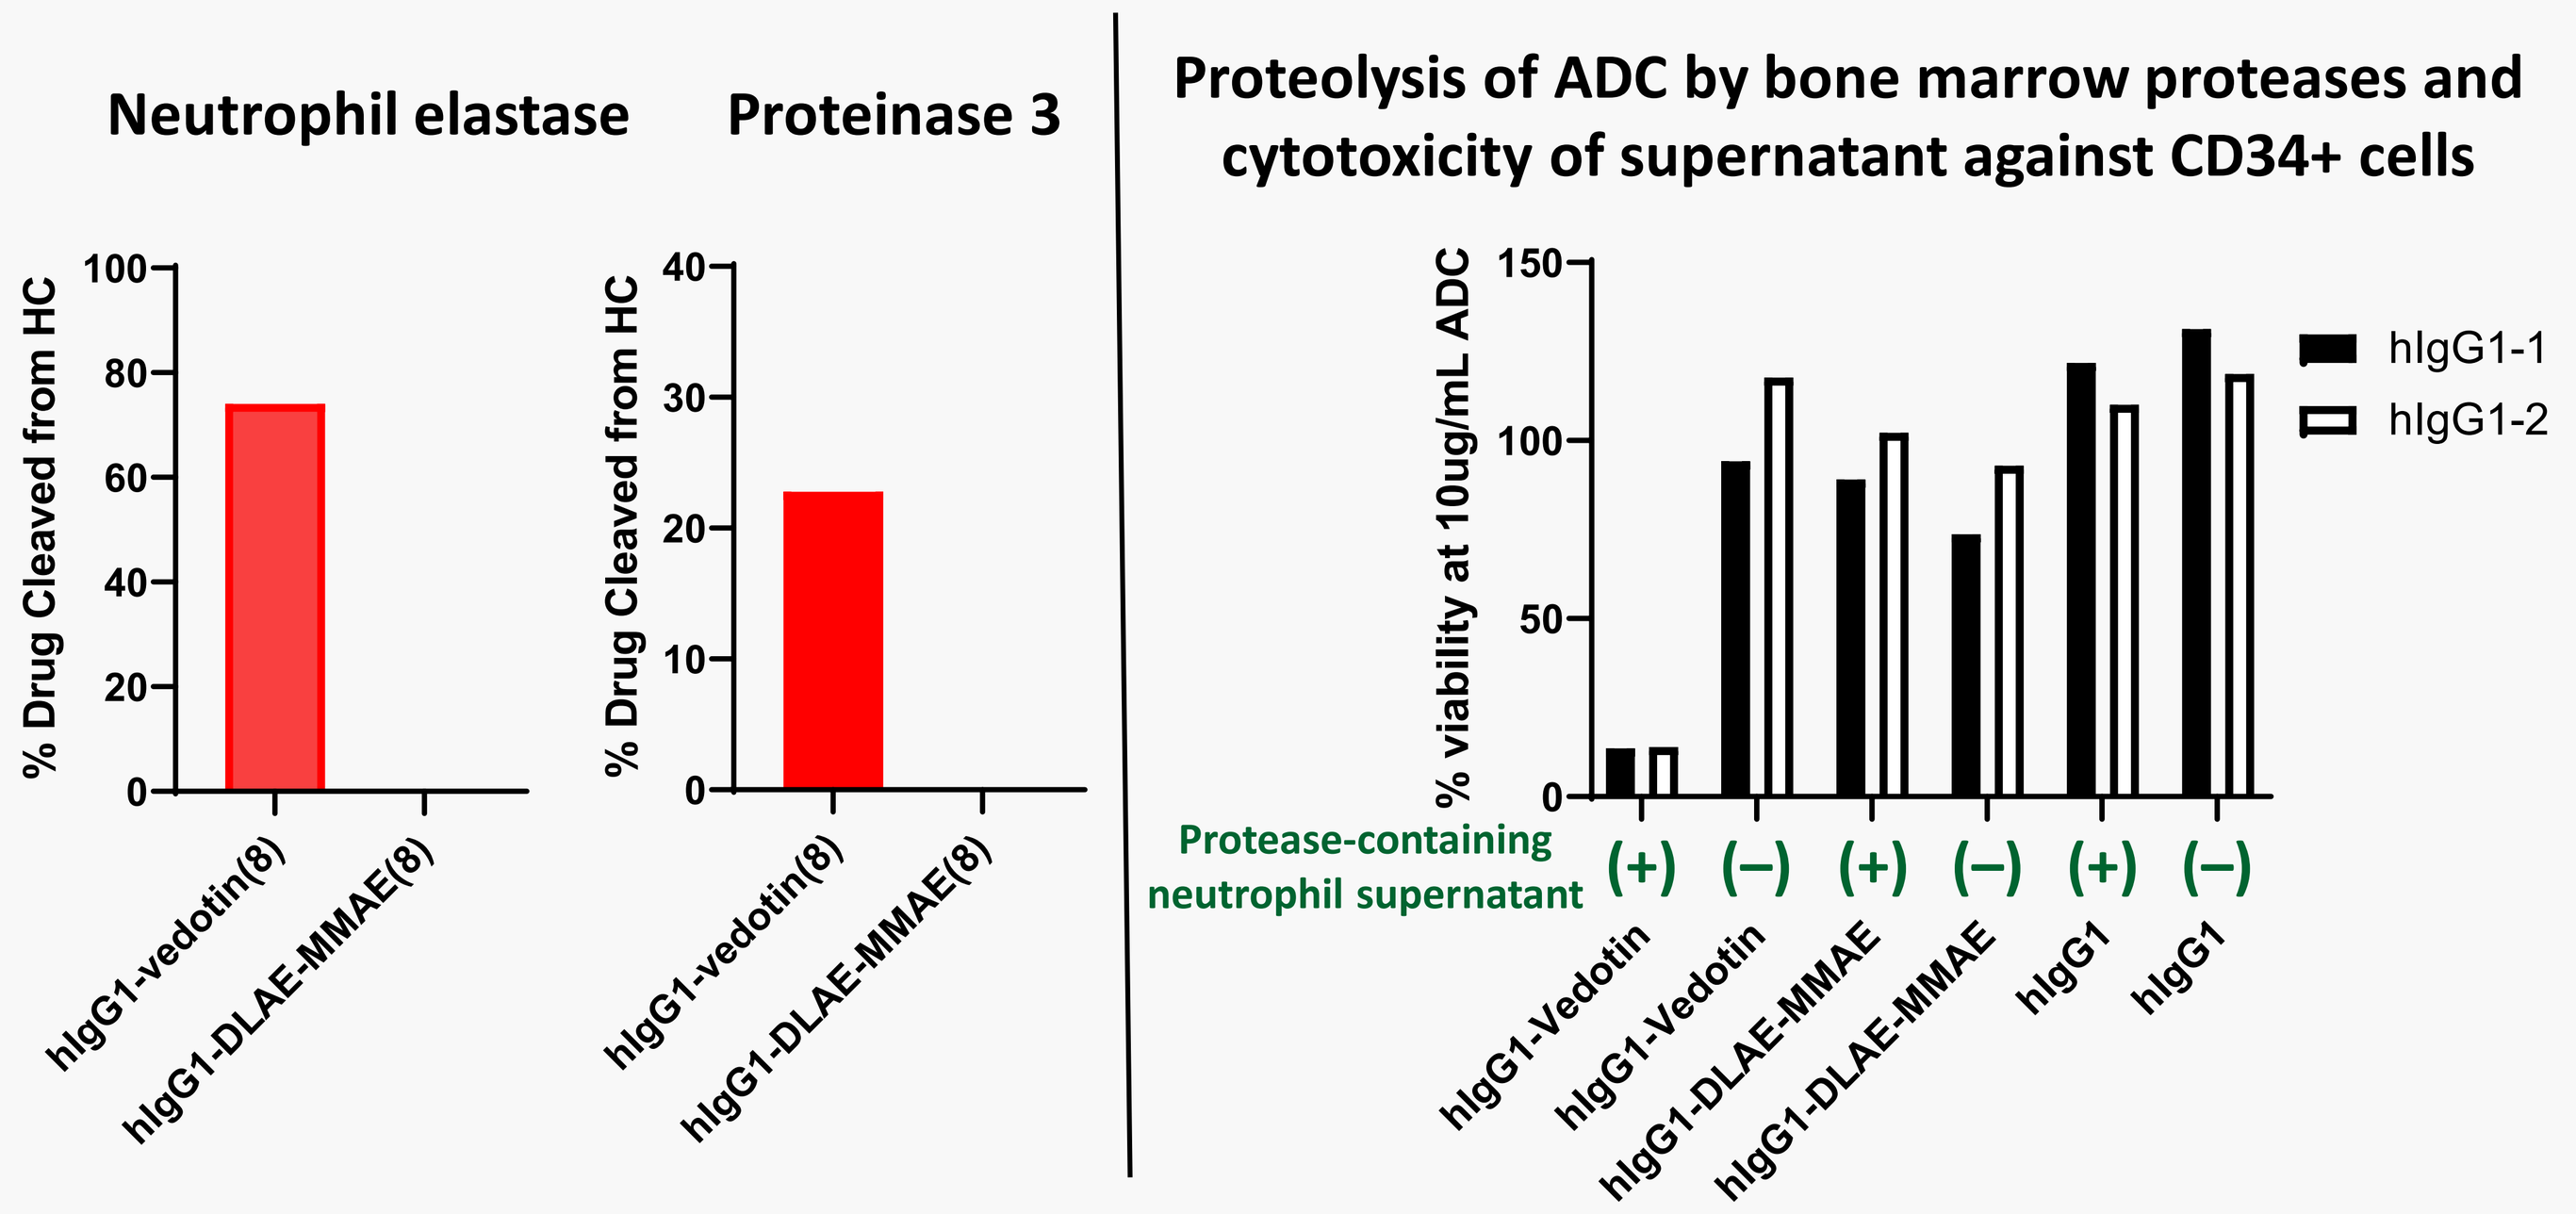
Neutrophil elastase
Proteinase 3
% Drug Cleaved from HC
hIgG1-vedotin(8)
hIgG1-DLAE-MMAE(8)
Proteolysis of ADC by bone marrow proteases and cytotoxicity of supernatant against CD34+ cells
% viability at 10ug/mL ADC
hIgG1-1
hIgG1-2
Protease-containing neutrophil supernatant
(+) (–) (+) (–) (+) (–)
hIgG1-Vedotin
hIgG1-Vedotin
hIgG1-DLAE-MMAE
hIgG1-DLAE-MMAE
hIgG1
hIgG1

Figure 2: **Left:** Proteolysis of vedotin and DLAE-MMAE ADCs by human neutrophil elastase and proteinase 3**.** ADCs (8-load; 10 ug) were incubated with protease (200 ng), human neutrophil elastase or proteinase 3, for 2 h at 37C. Proteolysis of drug linker from the heavy chain was assessed. **Right:** Cytotoxicity assay assessing drug release from DLAE-MMAE and vedotin ADCs by proteases secreted by differentiating granulocytes. CD34+ cells were differentiated into granulocytes over two weeks. Supernatant was collected and then either supernatant or blank media was combined with DLAE-MMAE or vedotin ADCs, conjugated to non-targeting antibodies hIgG1-1 and hIgG1-2, and incubated for 24 h at 37C. The ADC mixture was then added to CD34+ cells which had been expanded for one week and cytotoxicity was assessed.

DLAE-MMAE exhibits similar efficacy to vedotin in mouse efficacy studies

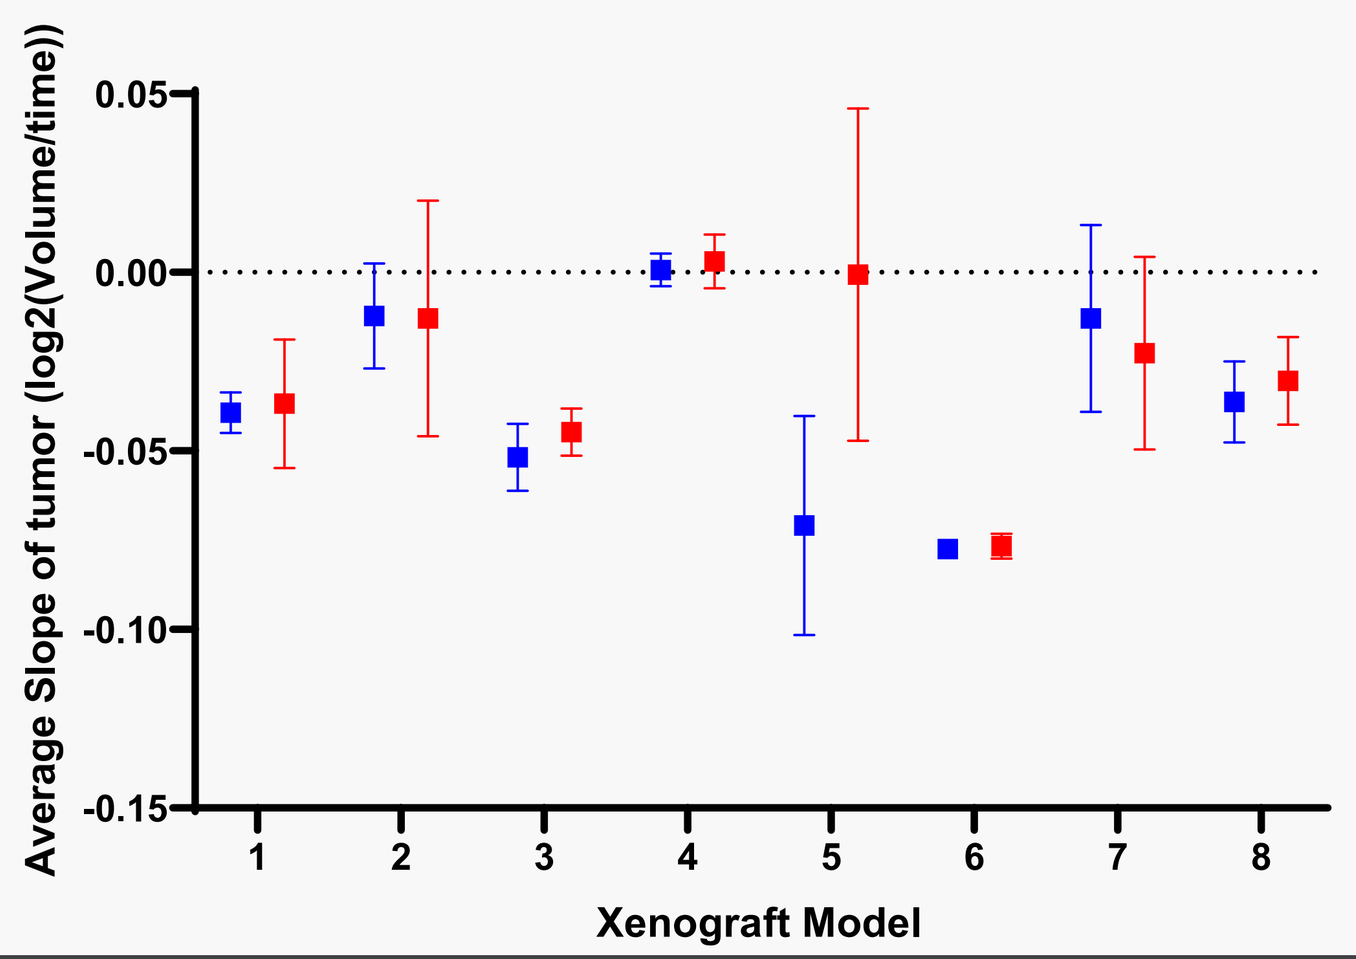
Average Slope of tumor (log2(Volume/time))
Xenograft Model

Figure 3: Efficacy of **DLAE-MMAE** and **vedotin** ADCs in 8 xenograft models at matched dose and schedule (Pancreatic adenocarcinoma (1 and 2), breast adenocarcinoma (3 and 4), Hodgkin lymphoma (5 and 6), pharyngeal cancer (7), and squamous cell carcinoma (8). Similar efficacies were observed across various DAR4 ADCs targeting different tumor antigens. Efficacy was calculated as average slope of tumor growth with a negative value representing a tumor regression.

In rat, DLAE-MMAE ADC has reduced bone marrow toxicity and similar exposure compared to vedotin ADC

- Bone marrow toxicity of DLAE-MMAE and vedotin conjugated to a non-targeted antibody was compared in rats. A third conjugate (mp-Val-Cit-MMAE) was also compared to assess the impact of an mp vs mc maleimide on bone marrow toxicity.

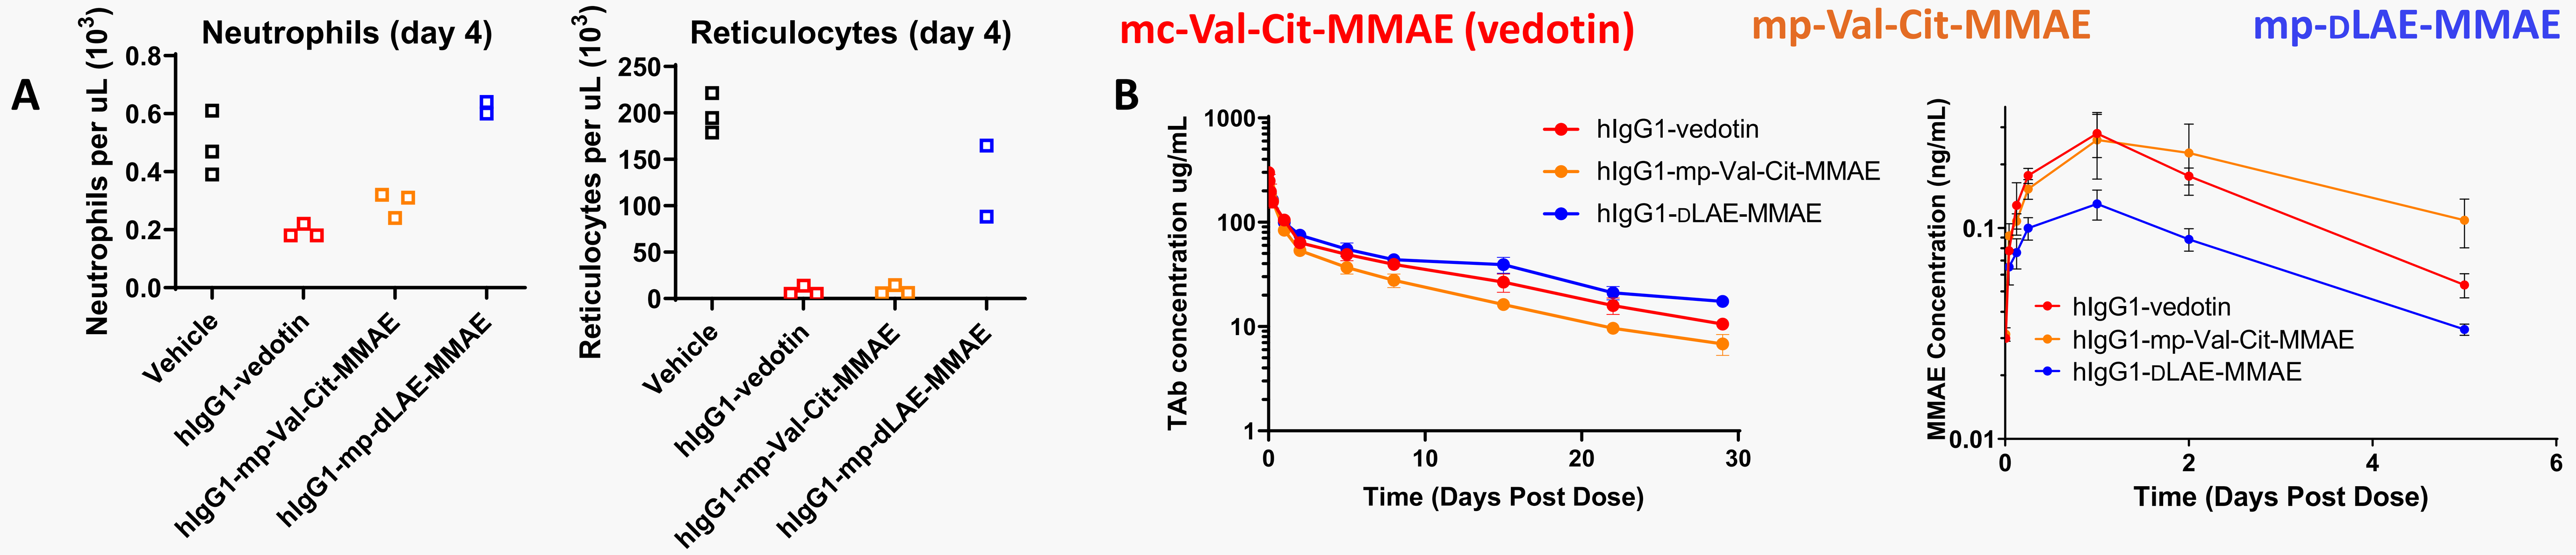
A
Neutrophils (day 4)
Neutrophils per uL (10^3)
Reticulocytes (day 4)
Reticulocytes per uL (10^3)
Vehicle
hIgG1-vedotin
hIgG1-mp-Val-Cit-MMAE
hIgG1-mp-dLAE-MMAE
mc-Val-Cit-MMAE (vedotin)
mp-Val-Cit-MMAE
mp-DLAE-MMAE
B
TAb concentration ug/mL
MMAE Concentration (ng/mL)
hIgG1-vedotin
hIgG1-mp-Val-Cit-MMAE
hIgG1-DLAE-MMAE
Time (Days Post Dose)

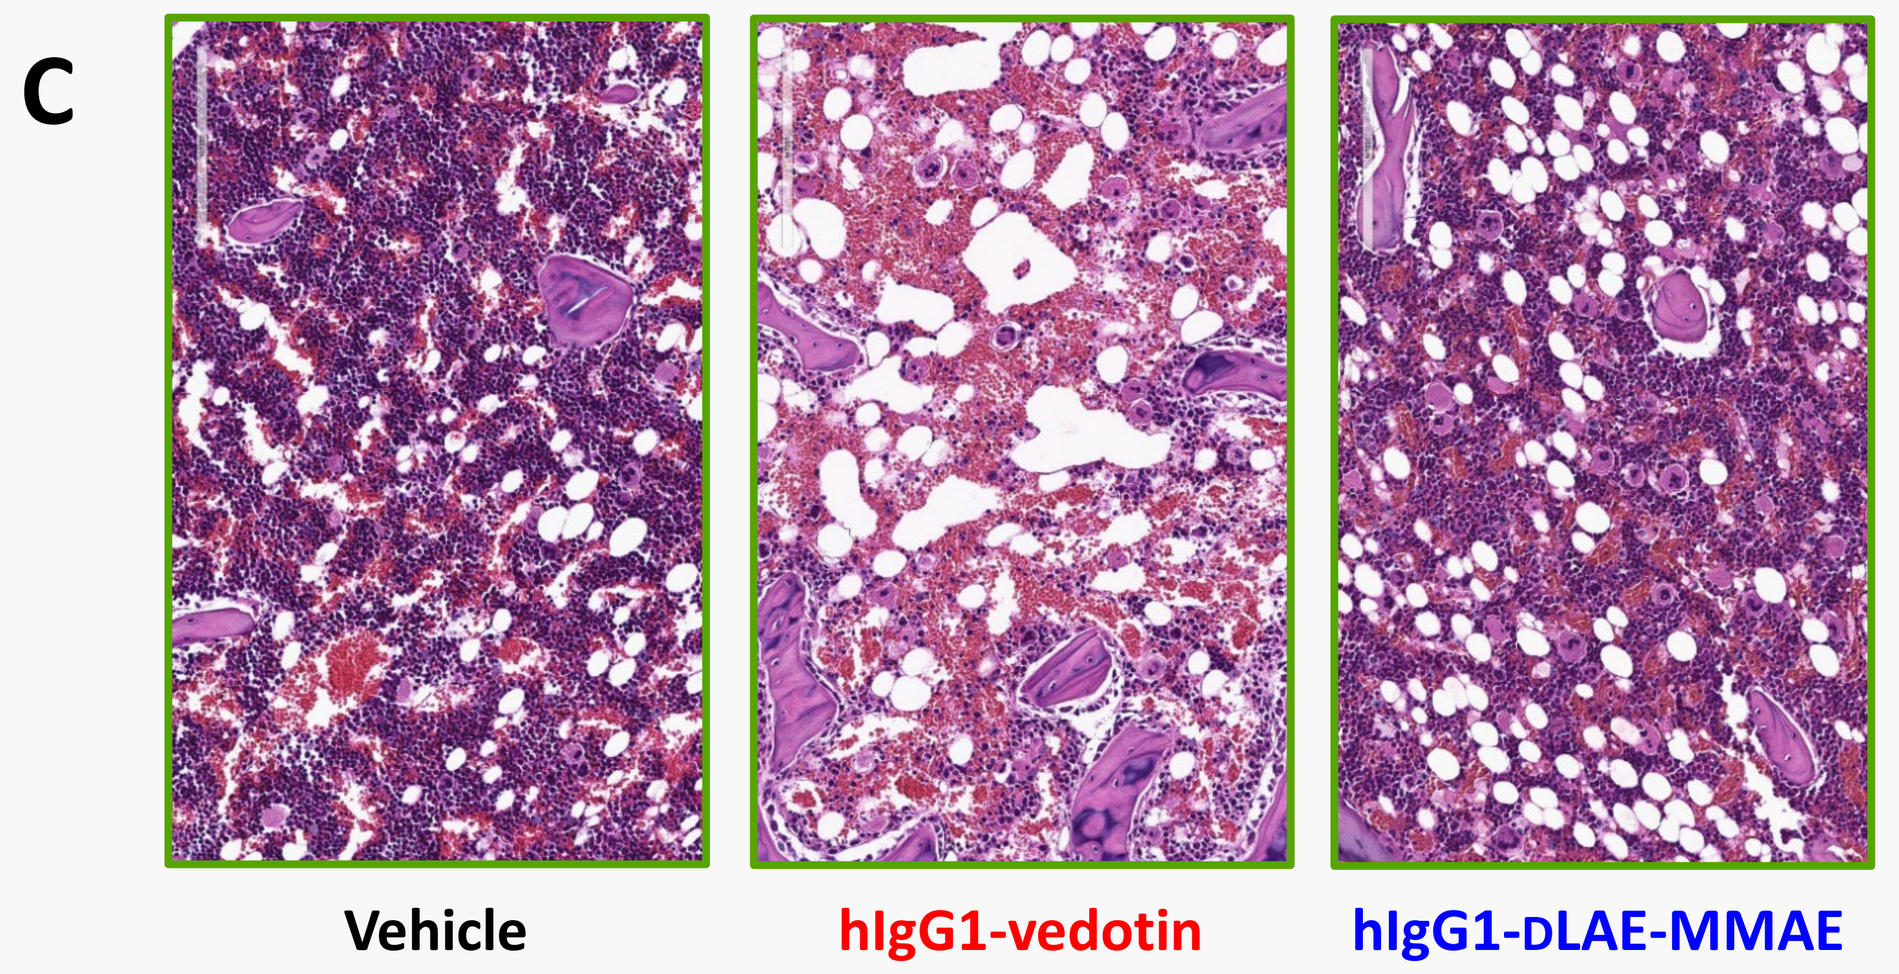
C
Vehicle
hIgG1-vedotin
hIgG1-DLAE-MMAE

Figure 4. Comparison of hIgG1-vedotin, DLAE-MMAE, and -mp-Val-Cit-MMAE in a rat toxicity model at 10 mg/kg. **A)** Neutrophil (left) and reticulocyte (right) counts on day 4. **B)** Serum concentrations of total antibody (left) and unconjugated MMAE (right). **C)** Histopathology of bone marrow on day 4. Dark blue stain represents nuclei from hematopoietic cells.

Rat toxicity conclusions

- Rats administered hIgG1-DLAE-MMAE had neutrophil and reticulocyte counts more like vehicle and significantly higher than hIGg1-vedotin.
- Rats administered hIgG1-DLAE-MMAE had a 2.7-fold greater MTD, similar antibody exposure, and reduced concentrations of unconjugated MMAE in plasma when compared to hIgG1-vedotin.
- There was no significant difference in toxicity when comparing hIgG1-vedotin (mc-maleimide) and hIgG1-mp-Val-Cit-MMAE, suggesting maleimide stability has little impact.

In NHP, DLAE-MMAE ADC has reduced bone marrow toxicity, 1.8-fold higher MTD, and similar exposure compared to vedotin ADC following a single dose

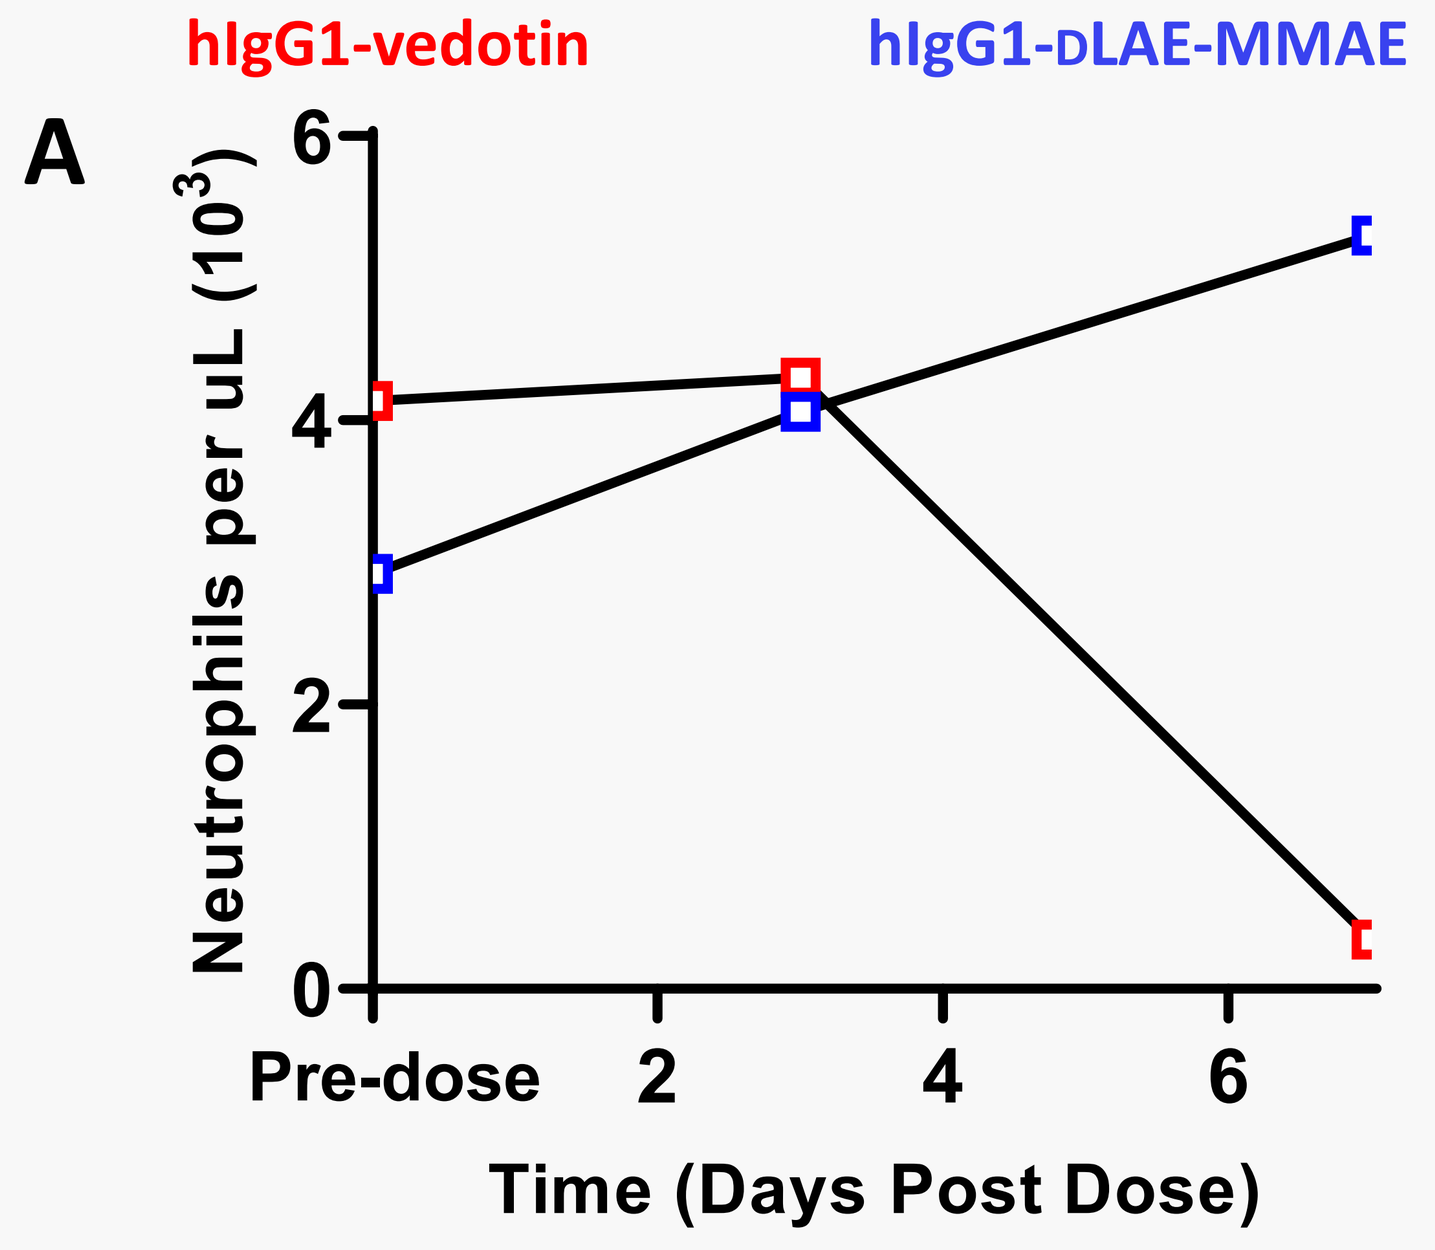
A
hIgG1-vedotin
hIgG1-DLAE-MMAE
Neutrophils per uL (10^3)
Pre-dose
Time (Days Post Dose)

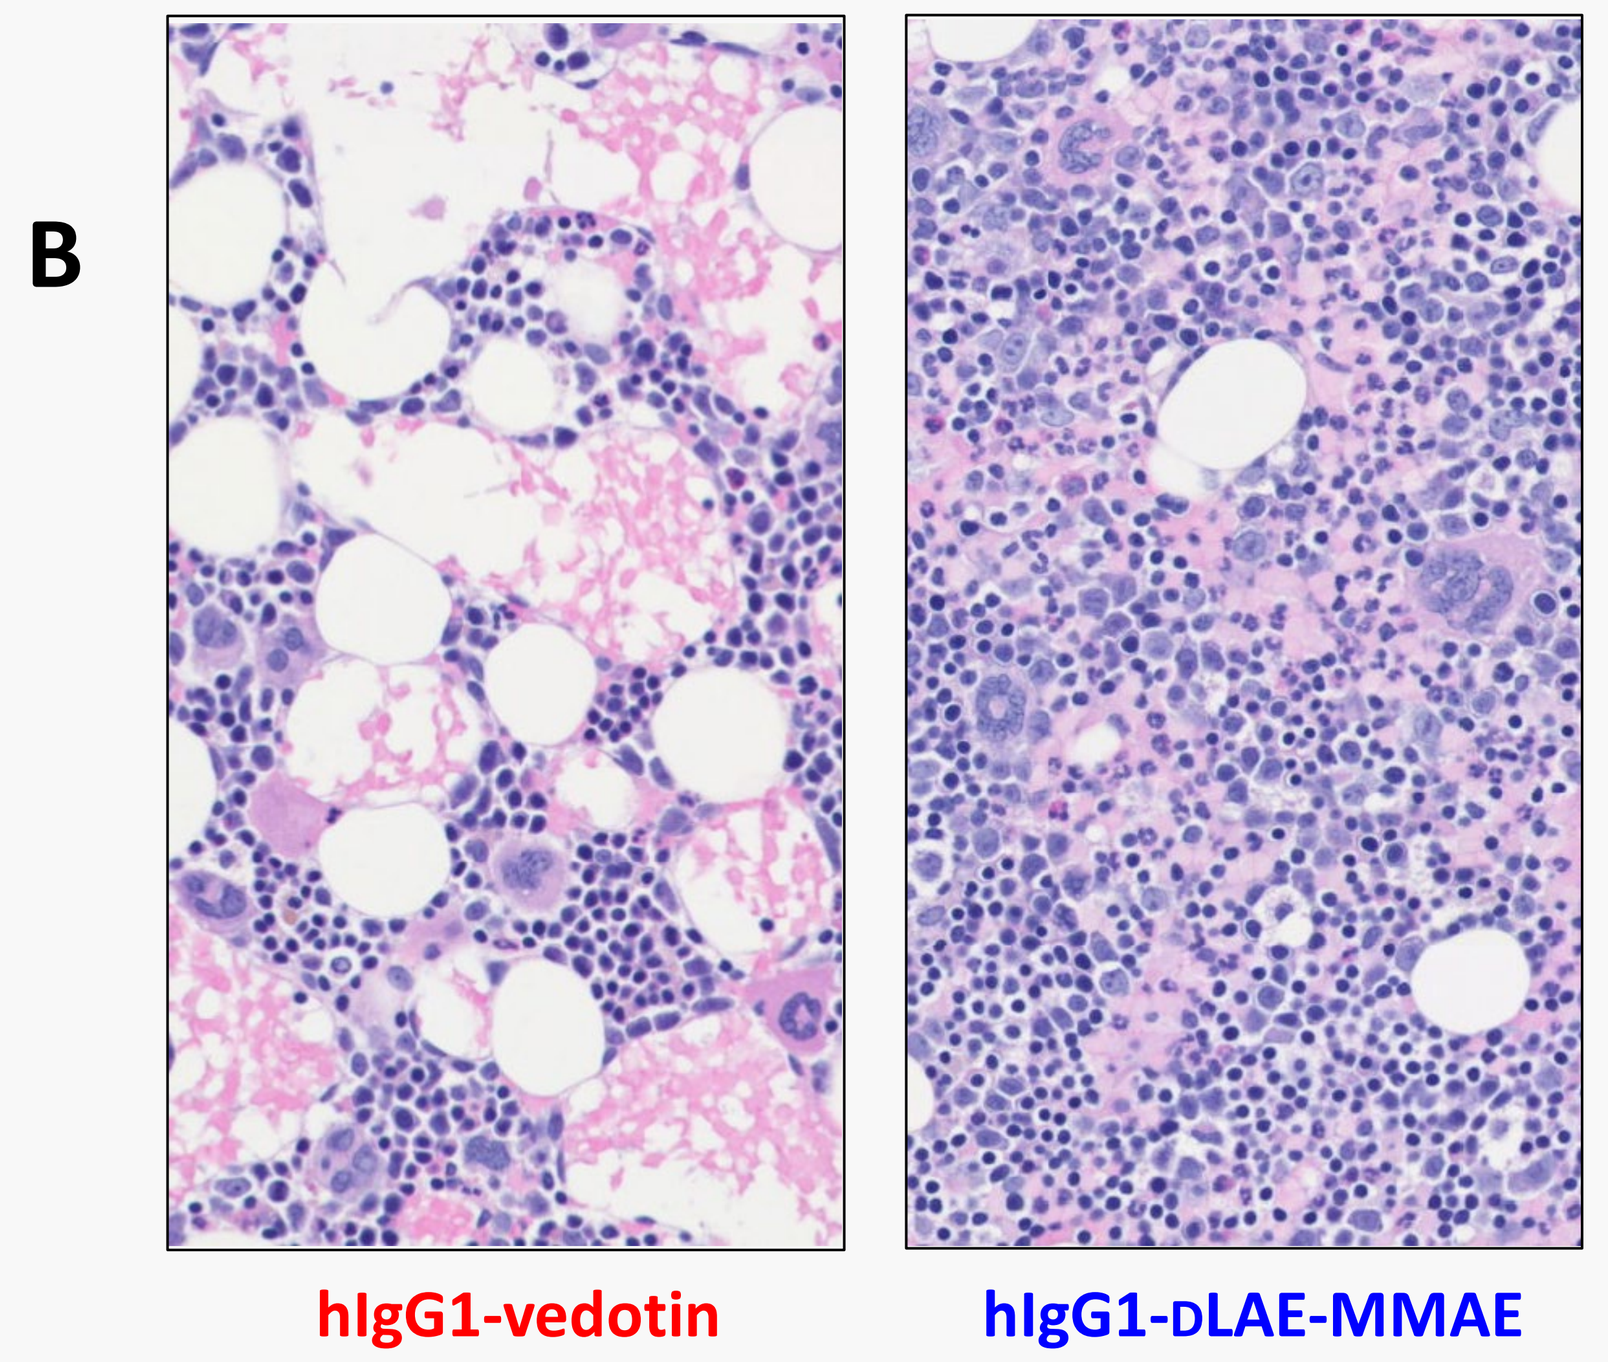
B
hIgG1-vedotin
hIgG1-DLAE-MMAE

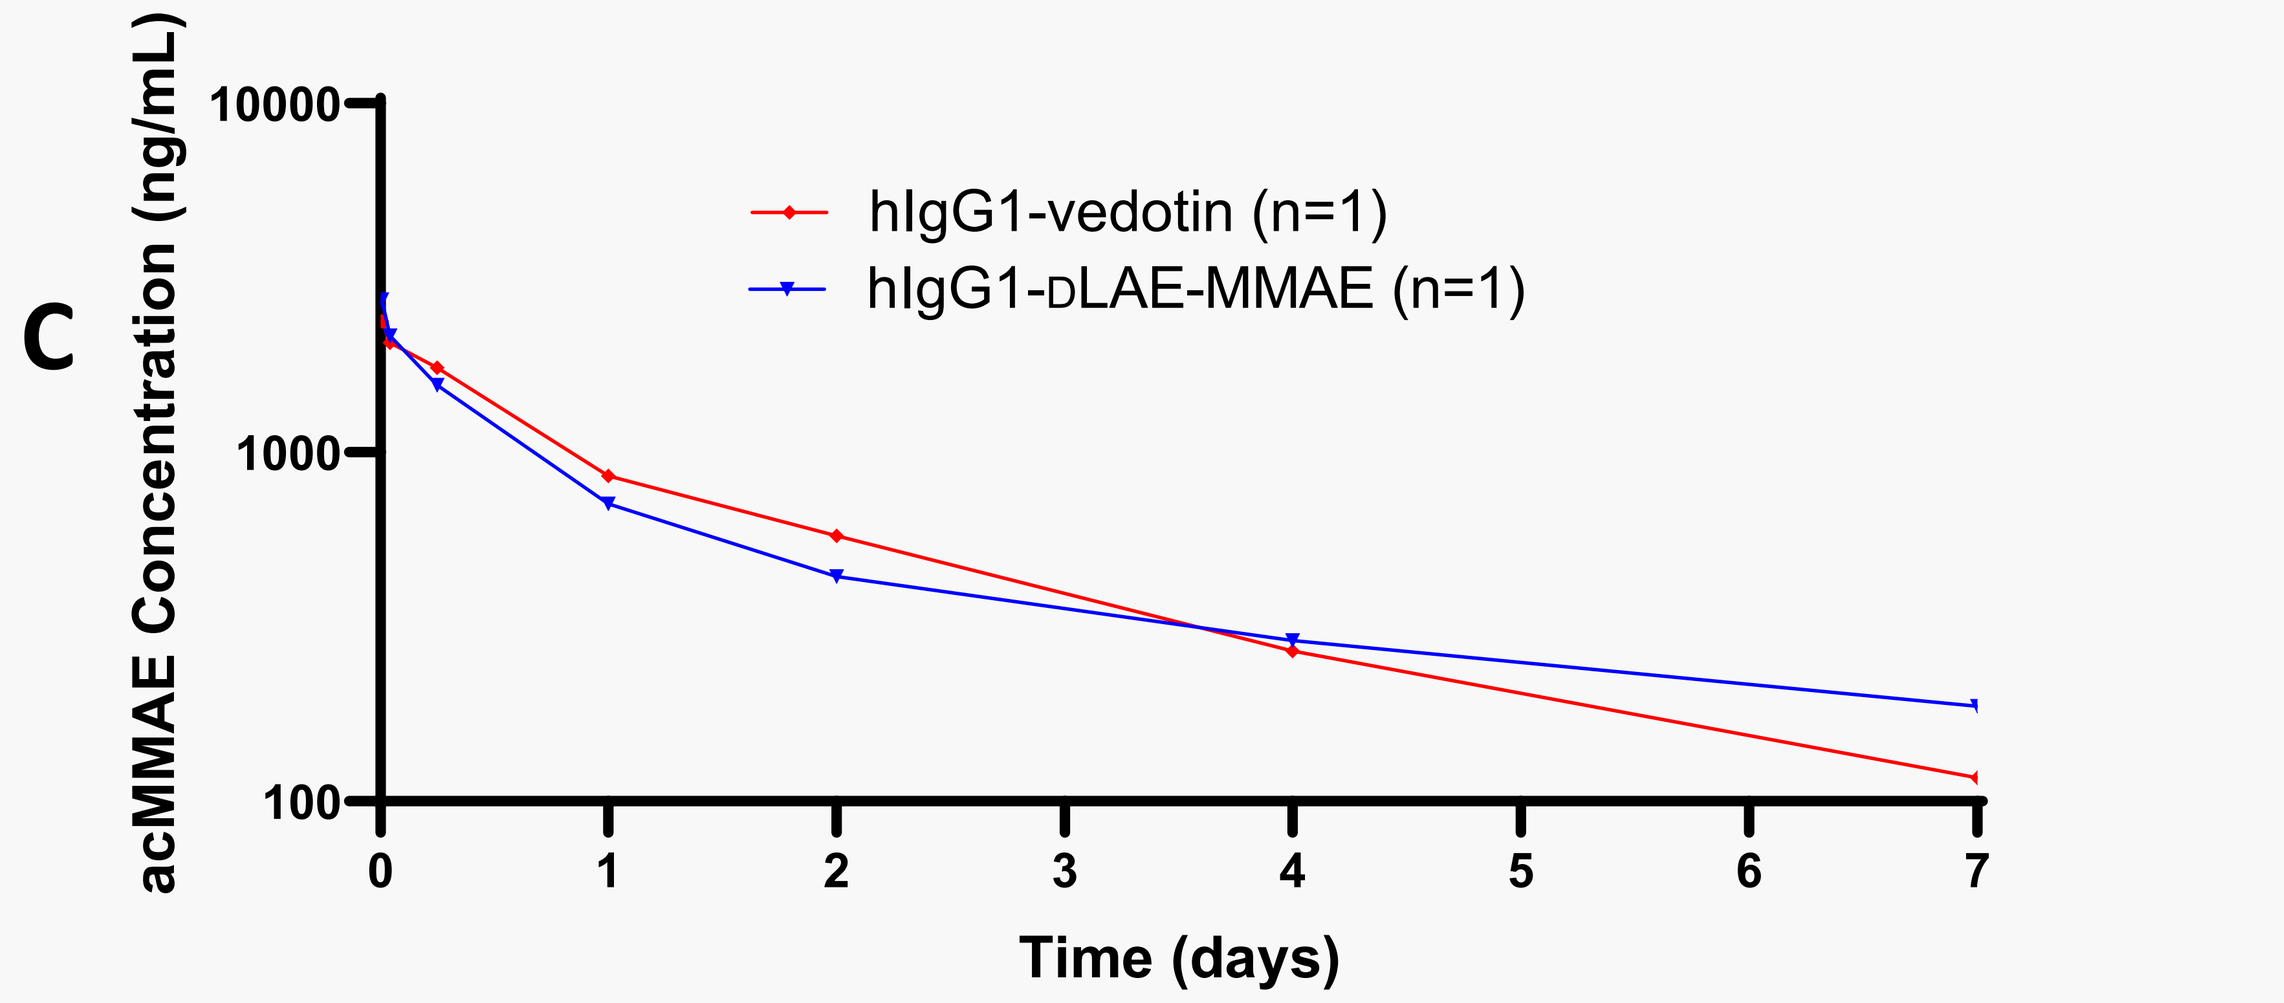
C
acMMAE Concentration (ng/mL)
hIgG1-vedotin (n=1)
hIgG1-DLAE-MMAE (n=1)
Time (days)

Figure 5. Comparison of hIgG1-vedotin and -DLAE-MMAE in a NHP toxicity model at 5 mg/kg. **A)** Neutrophil counts on day 3 and 7. **B)** Histopathology of bone marrow on day 7. Dark blue stain represents nuclei from hematopoietic cells. hIgG1-vedotin shows decreased erythroid and myeloid lineage bone marrow cellularity. hIgG1-DLAE-MMAE shows normal bone marrow cellularity. **C)** Antibody conjugated MMAE exposure after a single dose of ADC.

Conclusions

- ADCs with the DLAE-MMAE drug linker had similar efficacy to vedotin, but reduced bone marrow toxicity and a higher MTD in rat and NHP toxicity studies (2.7 and 1.8-fold higher MTD, respectively).
- Reduced proteolysis of the DLAE-MMAE drug linker by neutrophil proteases is one potential mechanism for its reduced bone marrow toxicity.
- Given the preclinical characteristics of the DLAE-MMAE drug linker, a Phase 1, first-in-human study is planned to evaluate the safety and antitumor activity of SGN-35T in lymphoid malignancies **(Poster No. C132)**.

Poster No. C113
EORTC-NCI-AACR International Conference; October 11-15, 2023

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from EORTC or the author of this poster, Noah Bindman (NbindmanSeagen.com)