PHASE 1 DOSE-ESCALATION STUDY OF SEA-TGT MONOTHERAPY IN PATIENTS WITH ADVANCED MALIGNANCIES (SGNTGT-001)

Elena Garralda Cabanas,[1] Elisa Fontana,[2] Honey Kumar Oberoi,[1] Nataliya V. Uboha,[3] Ecaterina E. Dumbrava,[4] Emiliano Calvo,[5] Giuseppe Curigliano,[6] Diwakar Davar,[7] Bridget P. Keenan,[8] Vincent K. Lam,[9] Sudhir Manda,[10] Amitkumar Mehta,[11] Anna Minchom,[12] Alison Moskowitz,[13] Ravi Paluri,[14] Vincent Ribrag,[15] Donald Richards,[16] Lillian Siu,[17] Paul Swiecicki,[18] Trisha Wise-Draper,[19] Jasmine Zain,[20] Andres Forero-Torres,[21] Ping Xu,[21] Stephen Ansell[22]

[1]Hospital Universitario Vall d'Hebron, Barcelona, Spain; [2]Sarah Cannon Research Institute UK, London, UK; [3]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [4]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [5]START Madrid-CIOCC, Centro Integral Oncologico Clara Campal, Madrid, Spain; [6]European Institute of Oncology, IRCCS and University of Milano, Milan, Italy; [7]University of Pittsburgh Medical Center Hillman Cancer Center, Pittsburg, PA, USA; [8]University of California San Francisco, San Francisco, CA, USA; [9]Johns Hopkins Sidney Kimmel Comprehensive Cancer Center, Baltimore, MD, USA; [10]Arizona Oncology Associates PC HOPE, Tucson, AZ, USA; [11]University of Alabama at Birmingham, Birmingham, AL, USA; [12]The Royal Marsden Hospital (Surrey), London, UK; [13]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [14]Atrium Health Wake Forest Baptist Medical Center, Winston-Salem, NC, USA; [15]Institut Gustave Roussy, Villejuif, France; [16]Texas Oncology-Tyler, Tyler, TX, USA; [17]Princess Margaret Cancer Centre, Toronto, ON, Canada; [18]University of Michigan Comprehensive Cancer Center, Ann Arbor, MI, USA; [19]University of Cincinnati Cancer Institute, Cincinnati, OH, USA; [20]City of Hope National Medical Center, Duarte, CA, USA; [21]Seagen Inc., Bothell, WA, USA; [22]Mayo Clinic Rochester, Rochester, MN, USA

Background

- TIGIT is an inhibitory immune checkpoint receptor expressed on subsets of T-cells, and NK cells.
- TIGIT binds to 2 ligands; CD155 and CD112 and inhibits T-cell and NK-cell functions.
- SEA-TGT is an investigational human, nonfucosylated monoclonal antibody directed against TIGIT and blocks TIGIT's interaction with CD155 and CD112
 - The sugar-engineered Fc backbone has shown enhanced effector function in preclinical models on tumor cells
- SGNTGT-001 (NCT04254107) is a phase 1, open-label, multicenter study evaluating SEA-TGT in advanced solid tumor and lymphomas malignancies, consisting of dose escalation (Part A), dose expansion (Part B), and combination therapies (Parts C and D)
- Based on data from dose escalation, the optimal biological dose is presented here

Proposed Mechanism of Action of SEA-TGT

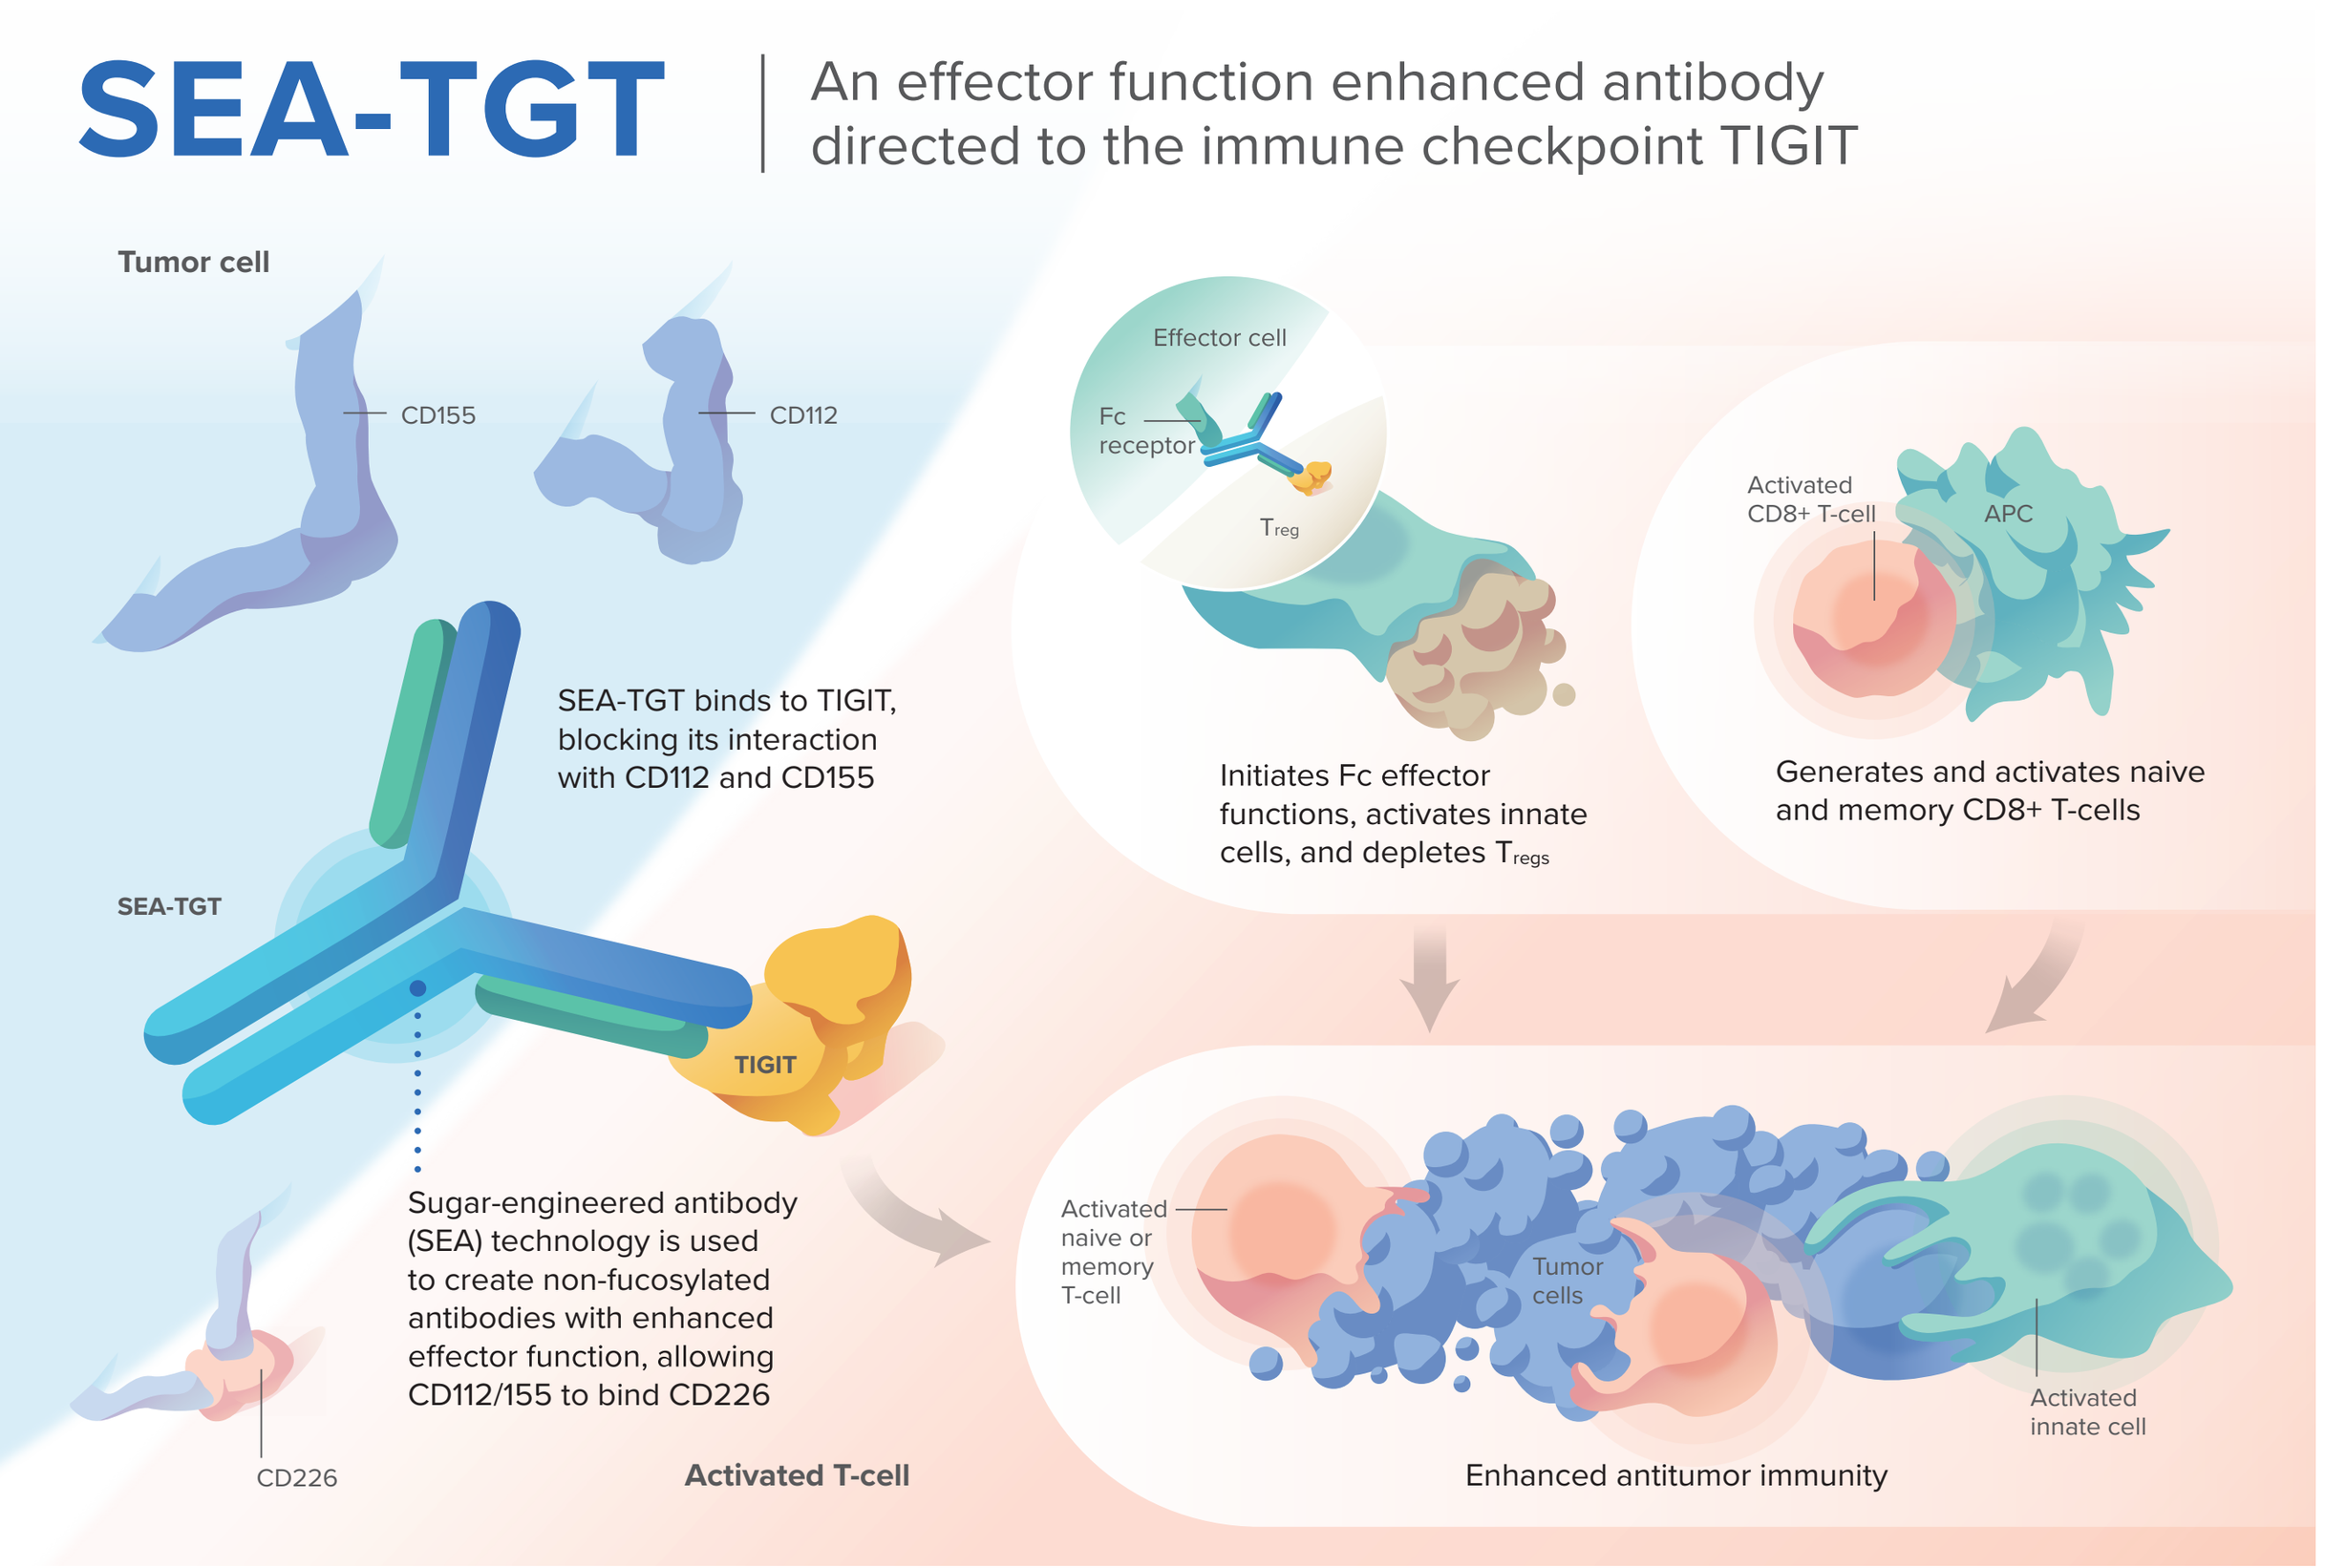

SEA-TGT
An effector function enhanced antibody directed to the immune checkpoint TIGIT
Tumor cell
CD155
CD112
SEA-TGT binds to TIGIT, blocking its interaction with CD112 and CD155
SEA-TGT
TIGIT
Sugar-engineered antibody (SEA) technology is used to create non-fucosylated antibodies with enhanced effector function, allowing CD112/155 to bind CD226
CD226
Activated T-cell
Effector cell
Fc receptor
Treg
Initiates Fc effector functions, activates innate cells, and depletes Tregs
Activated CD8+ T-cell
APC
Generates and activates naive and memory CD8+ T-cells
Activated naive or memory T-cell
Tumor cells
Activated innate cell
Enhanced antitumor immunity

SEA-TGT is an investigational agent, and its safety and efficacy have not been established. Proposed mechanism of action based on preclinical data.

© 2023 Seagen Inc., Bothell WA 98021. All rights reserved. USM/TGT/2020/0002(1)

Study Design - Part A Dose Escalation

Study Design

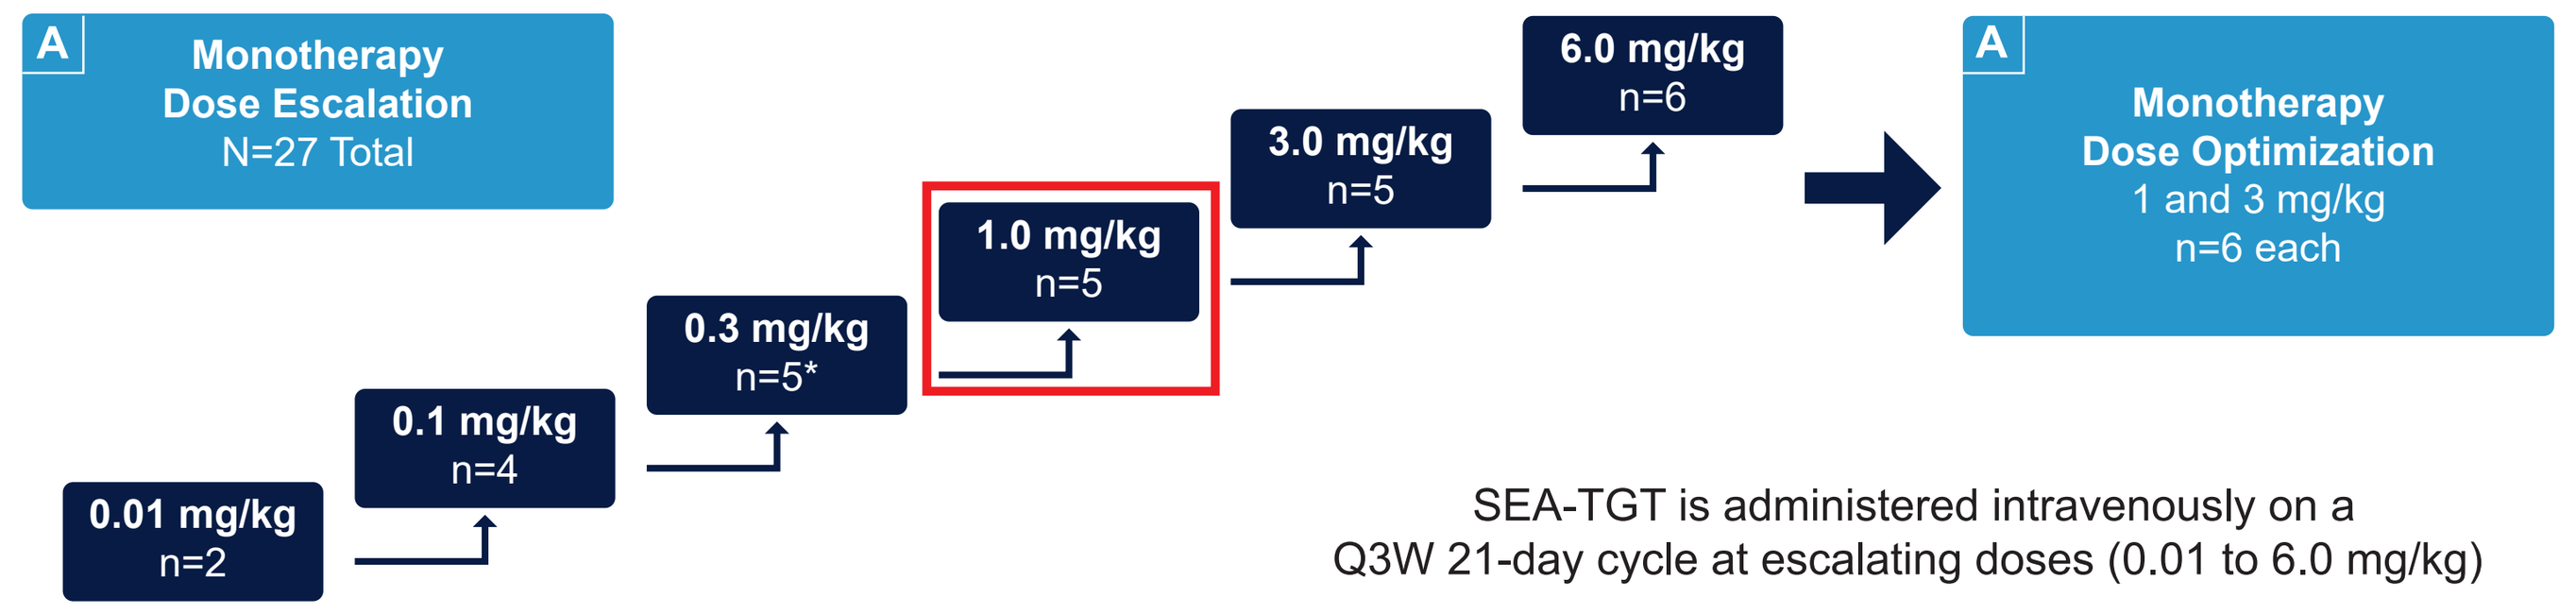

A Monotherapy Dose Escalation N=27 Total
0.01 mg/kg n=2
0.1 mg/kg n=4
0.3 mg/kg n=5*
1.0 mg/kg n=5
3.0 mg/kg n=5
6.0 mg/kg n=6
A Monotherapy Dose Optimization 1 and 3 mg/kg n=6 each

SEA-TGT is administered intravenously on a Q3W 21-day cycle at escalating doses (0.01 to 6.0 mg/kg)

Data cut off: 05 October 2022

*One patient was enrolled at 0.3 mg/kg and was treated at this dose for Cycles 1-4 before switching to 3.0 mg/kg

Study Objectives

- Primary
 - Evaluate the safety and tolerability of SEA-TGT
 - Identify the MTD, MAD, or recommended dose for SEA-TGT
- Secondary
 - Assess antitumor activity of SEA-TGT
 - Assess the PK and immunogenicity of SEA-TGT
- Optimal biological dose was defined by using the CUI (See 2023 AACR Abstract #5668; Poster 17, Section 44)
- Antitumor activity was based on RECIST v1.1 or Lugano classification with LYRIC

Key Eligibility Criteria

- Patients (≥18 years of age) with histologically or cytologically confirmed advanced or metastatic malignancies that have relapsed, are refractory, or had progression
- Eligible diagnoses: Solid Tumors (NSCLC, GE junction carcinoma, cutaneous melanoma, HNSCC, bladder cancer, cervical cancer, ovarian cancer, TNBC) and selected lymphomas (cHL, DLBCL, and PTCL-NOS)

Demographics and Characteristics at Baseline

	0.01 mg/kg (n=2)	0.1 mg/kg (n=4)	0.3 mg/kg (n=5)	1.0 mg/kg (n=11)	3.0 mg/kg (n=11)	6.0 mg/kg (n=6)	Total (N=39)
Age (years), median (range)	56.0 (54, 58)	59.5 (24, 79)	52.0 (20, 75)	59.0 (21, 80)	52.0 (37, 71)	49.0 (25, 78)	54.0 (20, 80)
Sex n (%)							
Male	2 (100)	3 (75)	4 (80)	8 (73)	7 (64)	6 (100)	30 (77)
Female	0	1 (25)	1 (20)	3 (27)	4 (36)	0	9 (23)
ECOG Performance Status, n (%)							
0	2 (100)	2 (50)	2 (40)	7 (64)	6 (55)	3 (50)	22 (56)
1	0	2 (50)	3 (60)	4 (36)	5 (45)	3 (50)	17 (44)
Number of prior therapies, median (range)	3.5 (3, 4)	6.5 (5, 8)	7.0 (3, 9)	5.0 (2, 7)	4.0 (2, 8)	3.5 (2, 8)	5.0 (2, 9)

Safety Results

Treatment-Emergent AEs

	0.01 mg/kg (n=2)	0.1 mg/kg (n=4)	0.3 mg/kg[a] (n=5)	1.0 mg/kg (n=11)	3.0 mg/kg (n=11)	6.0 mg/kg (n=6)	Total (n=39)
Patient with any TEAEs[a]	2 (100)	4 (100)	5 (100)	11 (100)	11 (100)	6 (100)	39 (100)
Treatment-related[b] TEAEs	2 (100)	3 (75.0)	2 (40.0)	7 (63.6)	7 (63.6)	6 (100)	27 (69.2)
Patient with ≥ Grade 3 TEAEs	0	1 (25.0)	3 (60.0)	6 (54.5)	8 (72.7)	3 (50.0)	21 (53.8)
≥ Grade 3 treatment-related TEAEs	0	0	0	2 (18.2)	3 (27.3)	2 (33.3)	7 (17.9)
Patients with any TE SAEs	0	3 (75.0)	1 (20.0)	6 (54.5)	4 (36.4)	1 (16.7)	15 (38.5)
Treatment-related TE SAEs	0	1 (25.0)	0	0	1 (9.1)	0	2 (5.1)

a Treatment-emergent adverse events are newly occurring adverse events (not present at baseline) or adverse events that worsen after first dose of investigational product.
b Related to treatment with investigational product as assessed by investigator.

- TEAEs related to SEA-TGT most frequently reported (≥10% of all patients)
 - IRR, chills, pyrexia, fatigue, rash maculopapular, and rash
- Grade 3 or higher TEAEs related to SEA-TGT reported by patients
 - Rash, anemia, hyperlipasaemia, lymphopenia, pancreatitis, rash macular, rash pruritic
- TE SAE related to SEA-TGT reported by patients
 - Chills, hypotension, hypoxia, and pancreatitis
- No Grade 4 or 5 TEAEs related to SEA-TGT were reported by patients
- One patient experienced a DLT of rash pruritic at 6.0 mg/kg dose level

TEAEs related to SEA-TGT by Preferred Term occurring in ≥ 10% of patients with at least 1 event

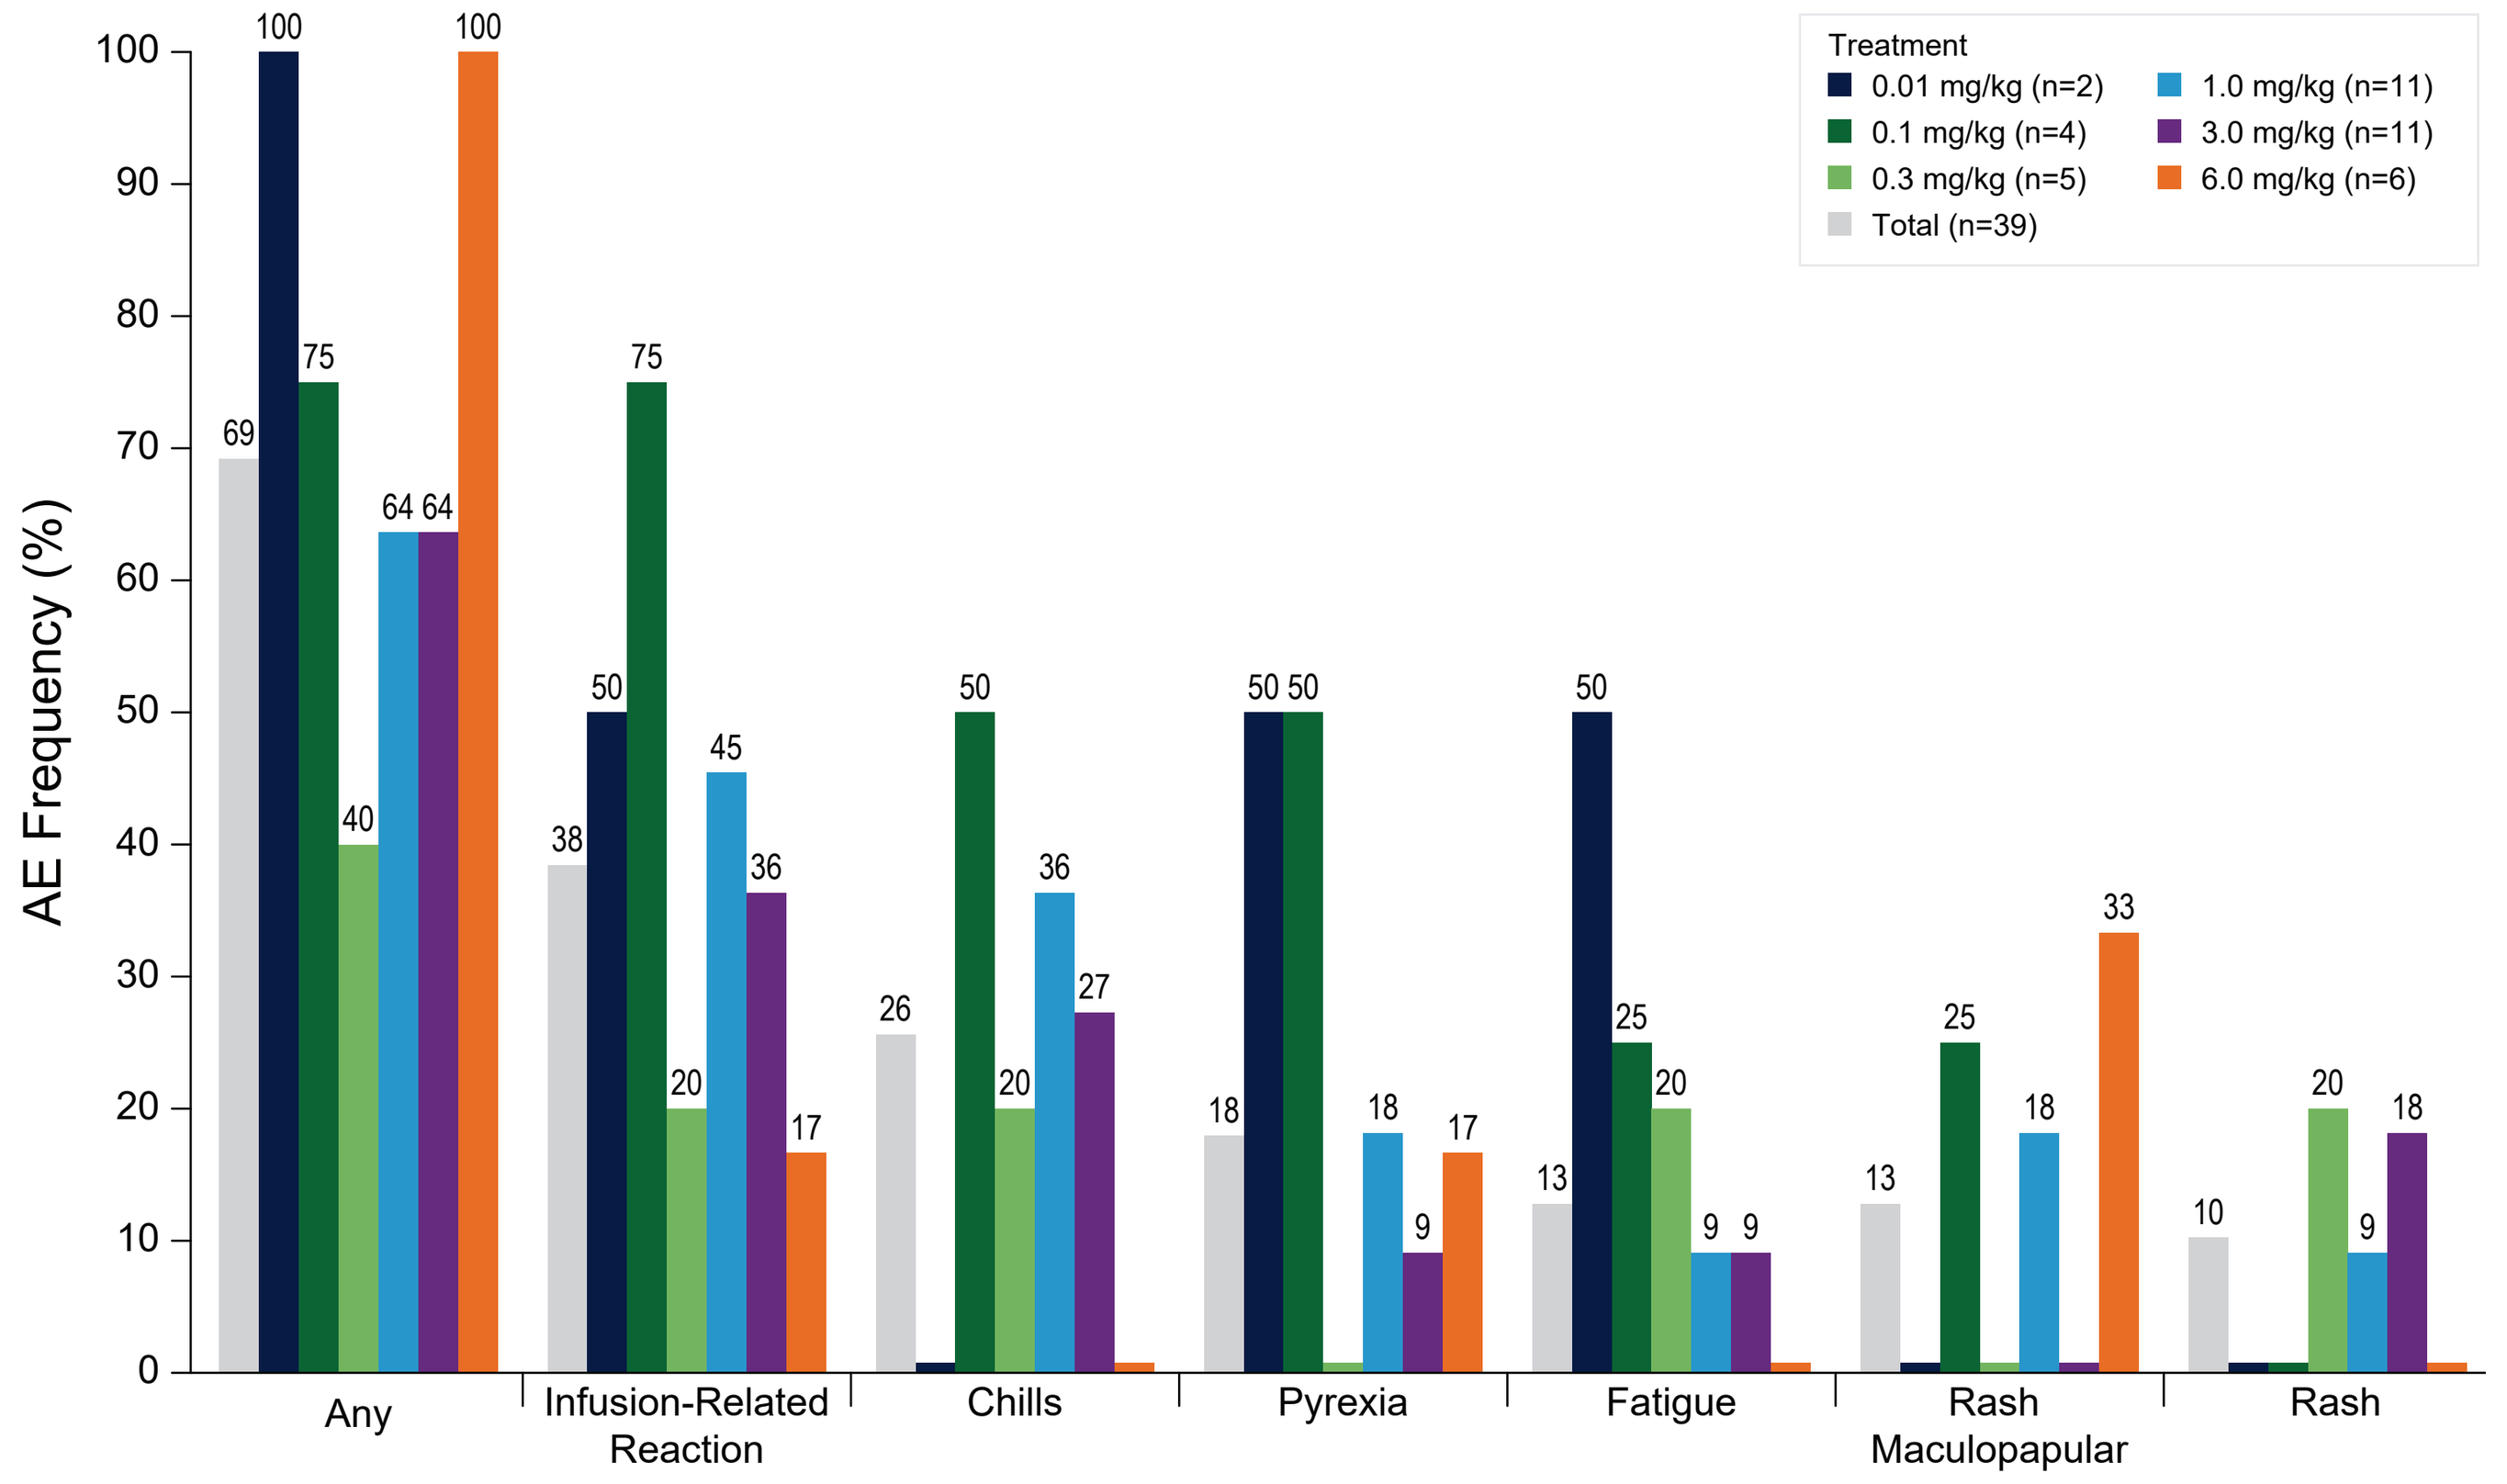

AE Frequency (%)
Treatment
0.01 mg/kg (n=2)
0.1 mg/kg (n=4)
0.3 mg/kg (n=5)
Total (n=39)
1.0 mg/kg (n=11)
3.0 mg/kg (n=11)
6.0 mg/kg (n=6)
Any
Infusion-Related Reaction
Chills
Pyrexia
Fatigue
Rash Maculopapular
Rash

Safety Results, cont'd

TEAEs of Special Interest

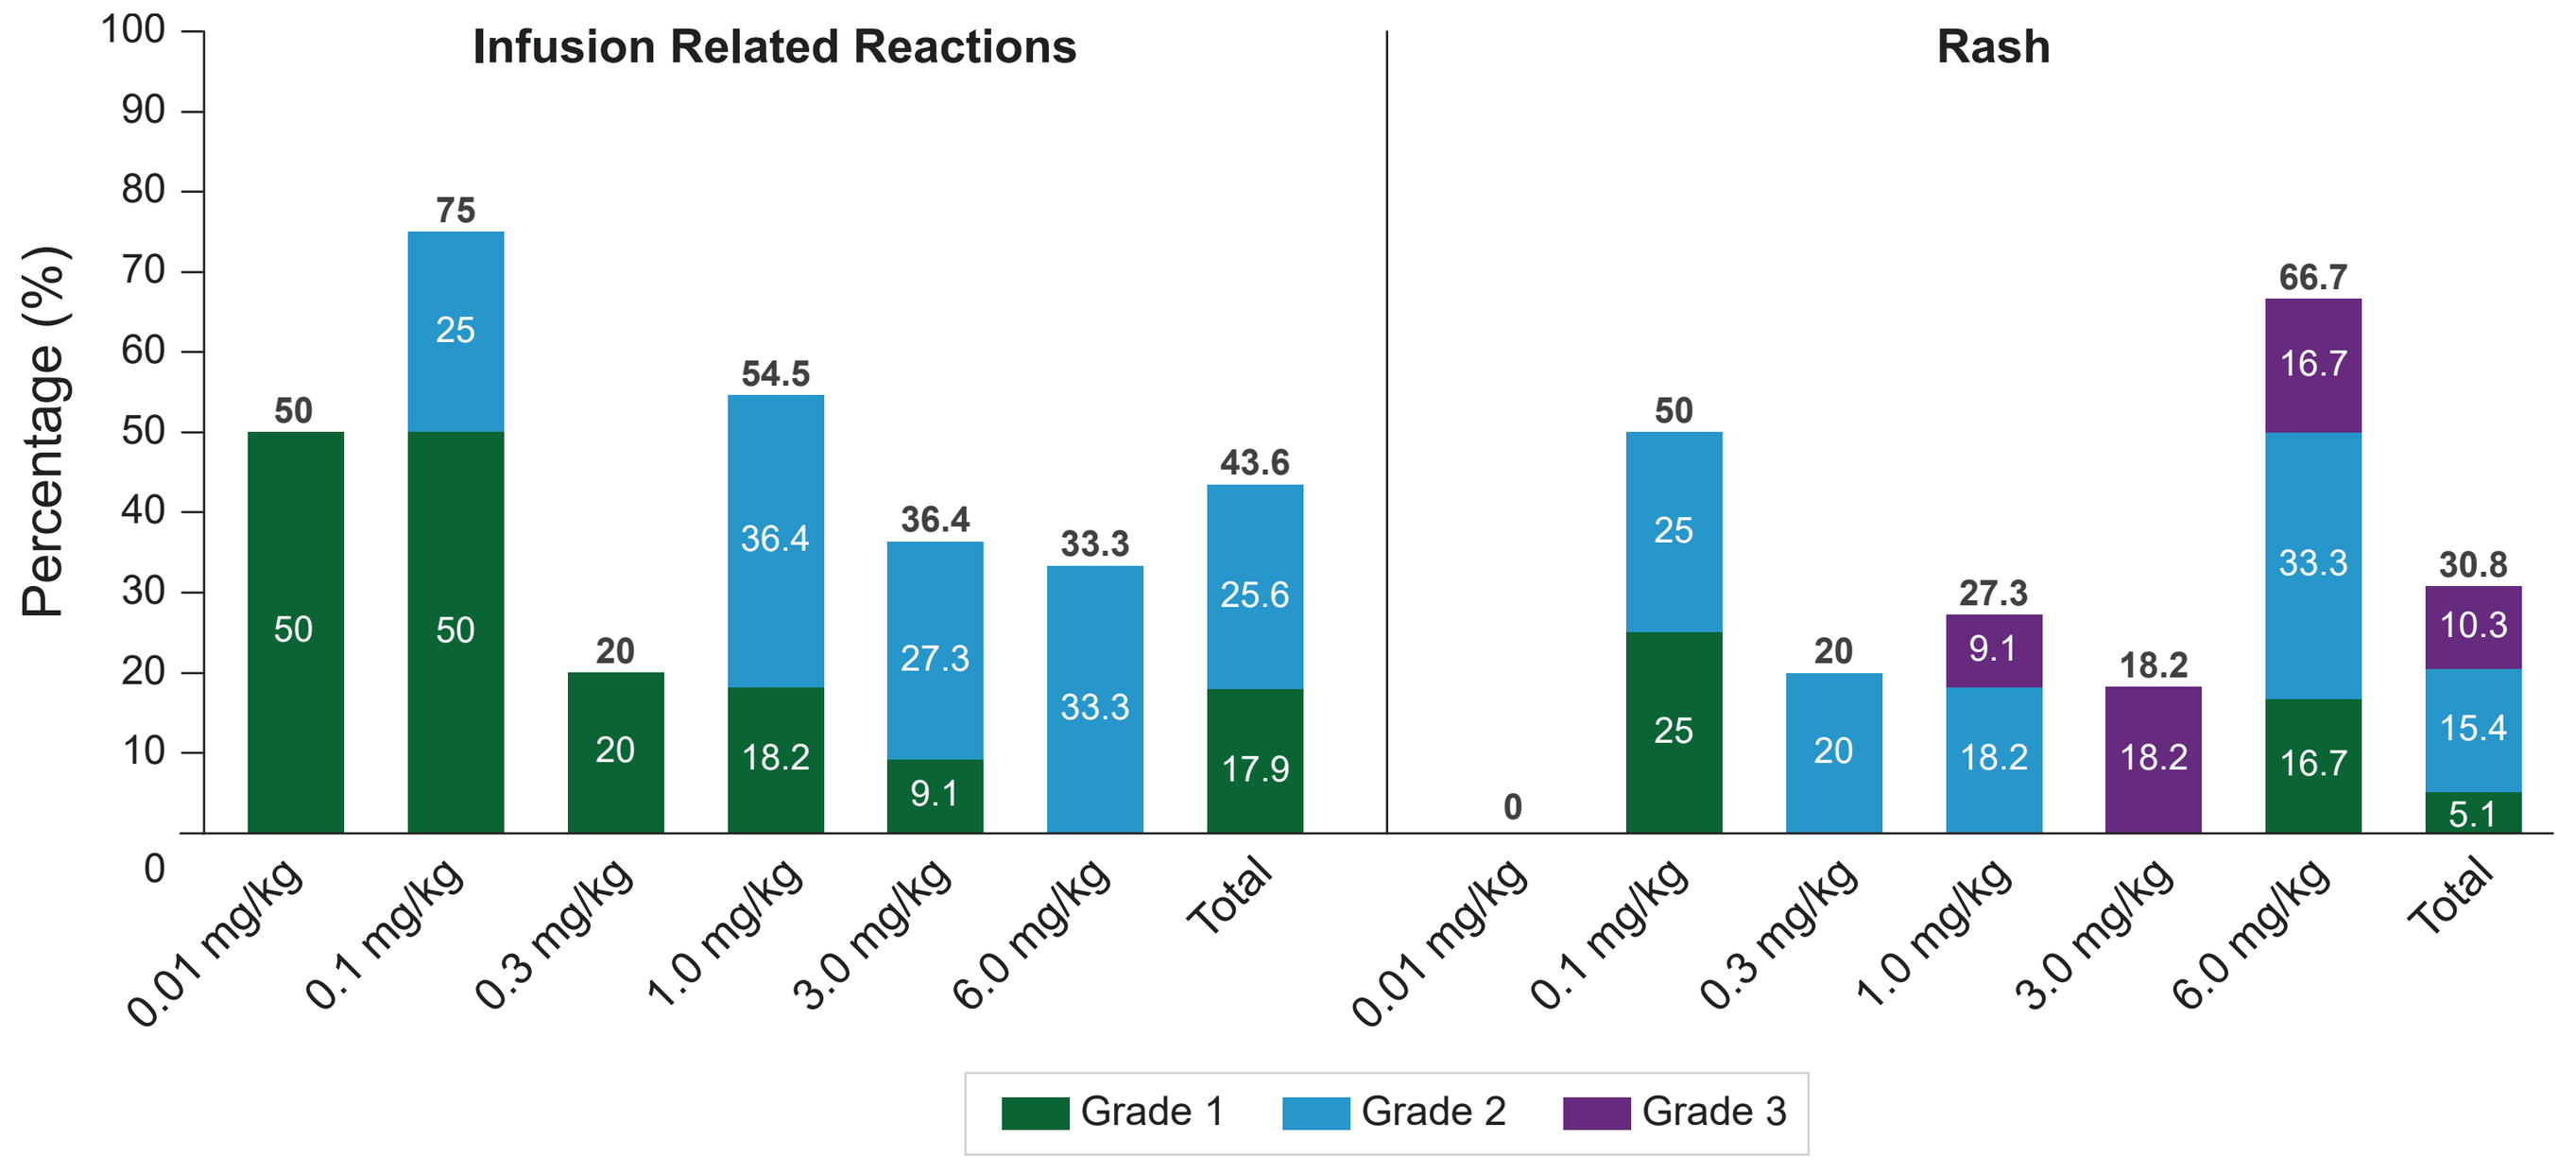

Infusion Related Reactions
Rash
Percentage (%)
0.01 mg/kg
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
6.0 mg/kg
Total
Grade 1
Grade 2
Grade 3

Treatment-emergent adverse events are newly occurring adverse events (not present at baseline) or adverse events that worsen after first dose of investigational product.
At each preferred term, multiple occurrences of events within a patient are counted only once at the highest toxicity grade.
Coding and classification criteria: MedDRA v25.0 and CTCAE v5.0

Preferred Terms Included in TEAEs of Special Interest

- Infusion-Related Reactions*: IRR, chills, pyrexia, nausea, arthralgia, hypertension, hyperventilation, hypophosphatemia, hypotension, hypoxia, tachycardia, vomiting
- Rash*: rash maculopapular, rash, rash erythematous, rash macular, rash pruritic

* Some of the terms included under IRR and rash were also considered by the investigators as immune mediated. No other immune-mediated adverse events were seen

Dose Modification Associated with TEAEs

	0.01 mg/kg (N=2)	0.1 mg/kg (N=4)	0.3 mg/kg (N=5)	1.0 mg/kg (N=11)	3.0 mg/kg (N=11)	6.0 mg/kg (N=6)	Total (N=39)
Patients with any TEAEs[a] resulting in dose modification	0	1 (25.0)	1 (20.0)	7 (63.6)	4 (36.4)	3 (50.0)	16 (41.0)
Dose discontinued	0	0	0	0	0	1 (16.7)	1 (2.6)
Dose reduced	0	0	0	0	0	0	0
Dose interrupted	0	1 (25.0)	0	3 (27.3)	4 (36.4)	1 (16.7)	9 (23.1)
Dose eliminated	0	0	0	3 (27.3)	1 (9.1)	1 (16.7)	5 (12.8)
Dose delayed	0	1 (25.0)	1 (20.0)	3 (27.3)	0	0	5 (12.8)

Treatment-emergent adverse events are newly occurring adverse events (not present at baseline) or adverse events that worsen after first dose of investigational product.

Antitumor Activity in Lymphomas per Lugano Criteria with LYRIC

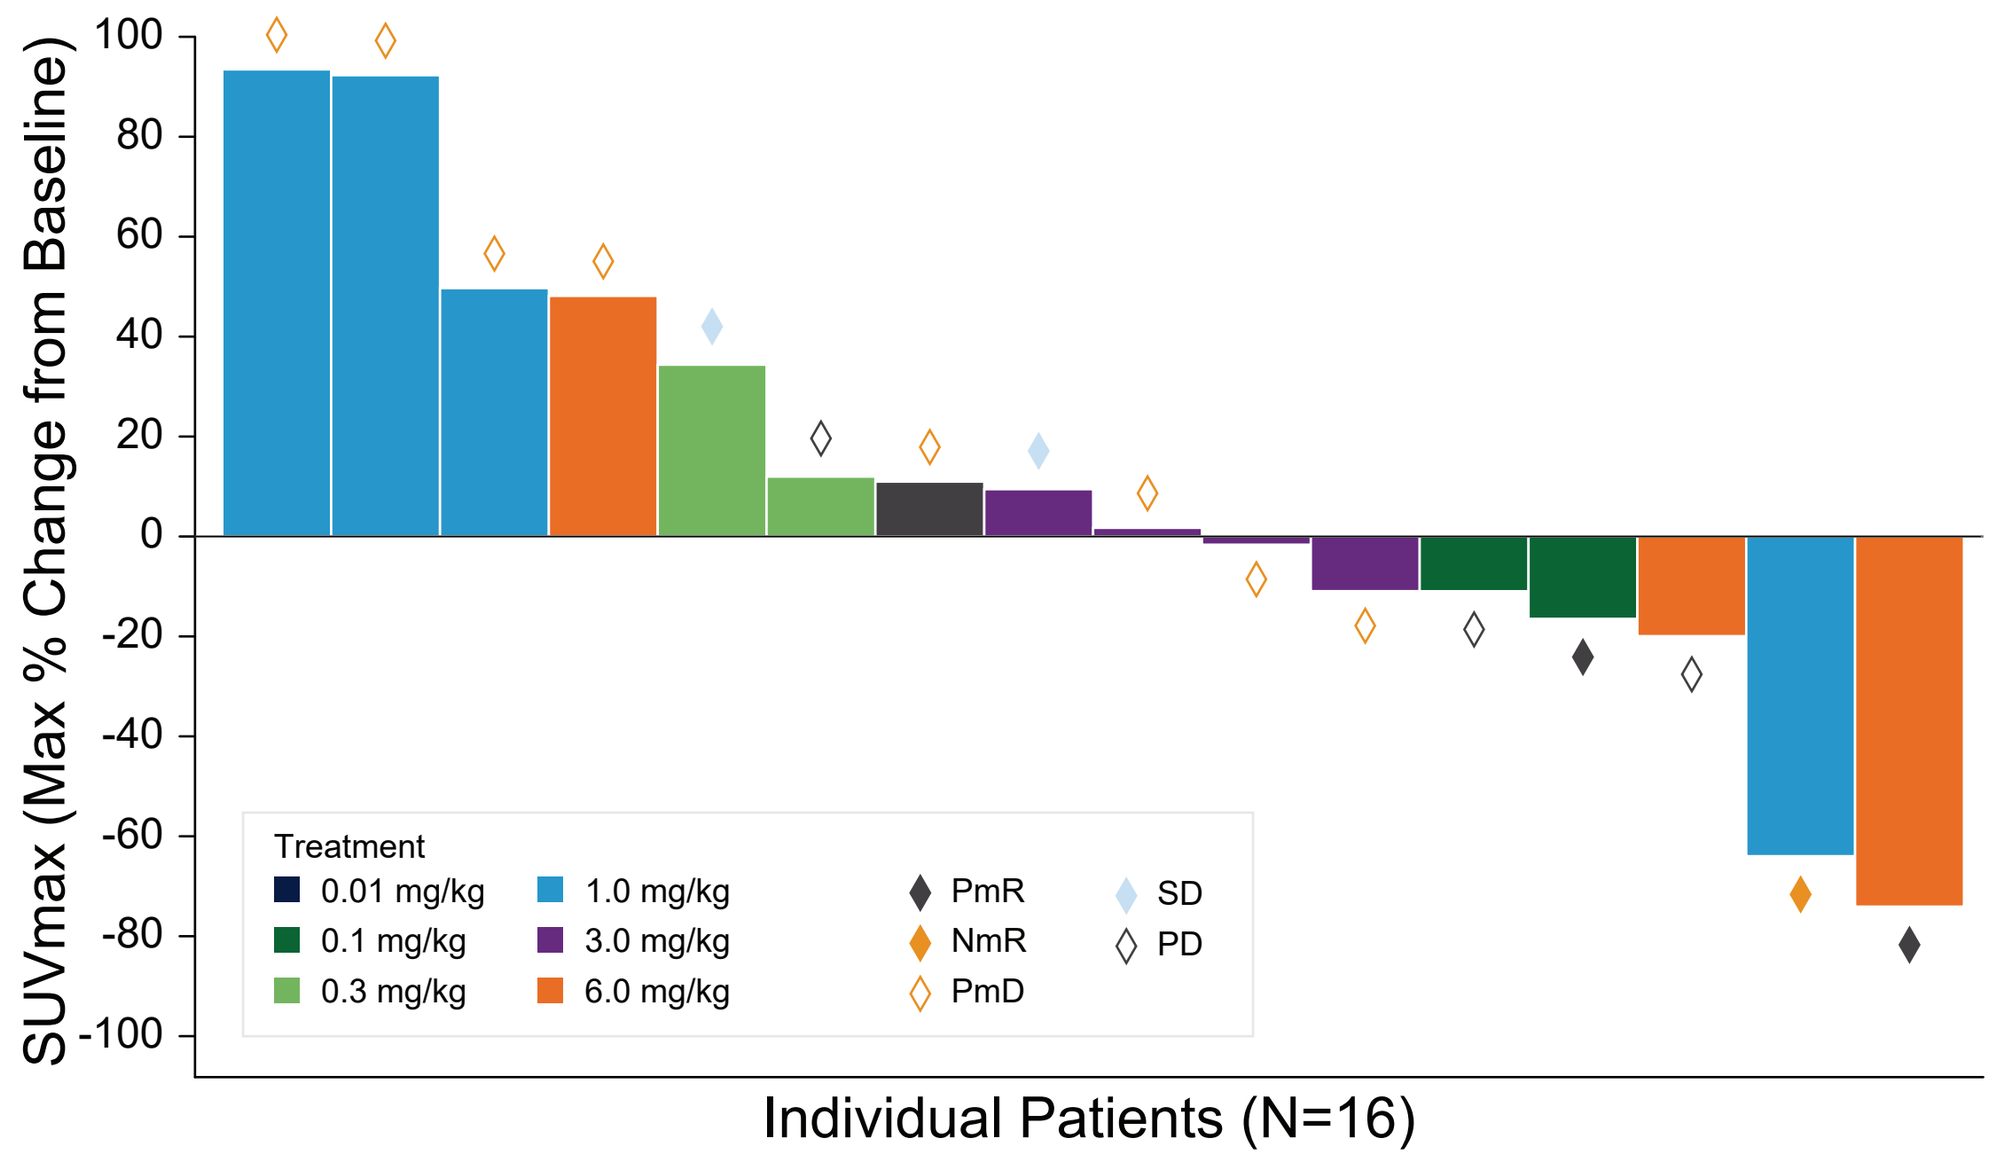

SUVmax (Max % Change from Baseline)
Treatment
0.01 mg/kg
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
6.0 mg/kg
PmR
NmR
PmD
SD
PD
Individual Patients (N=16)

	Total (N=16) n (%)
Best overall response[a,b]	
CmR	0
95% CI[c] for CmR rate	(0, 20.59)
PmR	2 (13)
95% CI[c] for PmR rate	(1.55, 38.35)
NmR/SD	3 (19)
PmD/PD	11 (69)
NE	0
Objective response rate (CmR+PmR)	2 (13)
95% CI[c] for ORR	(1.55, 38.35)

a CmR, PmR, NmR, and PmD per response criteria: Lugano with LYRIC.
b CmR, PmR, NmR, PmD, and NE are mutually exclusive.
c Two-sided 95% exact confidence interval, computed by using the Clopper-Pearson method (Collett 1991).

Maximum reduction or minimum increase if no reduction.
2 patients (0.1 mg/kg and 6.0 mg/kg) had partial metabolic reduction
- 1 patient with DLBCL
- 1 patient with cHL

1 patient (1.0 mg/kg) with cHL had a 60% reduction of target lesions but had progressive metabolic disease

Antitumor Activity: Solid Tumor per RECIST v1.1 Criteria

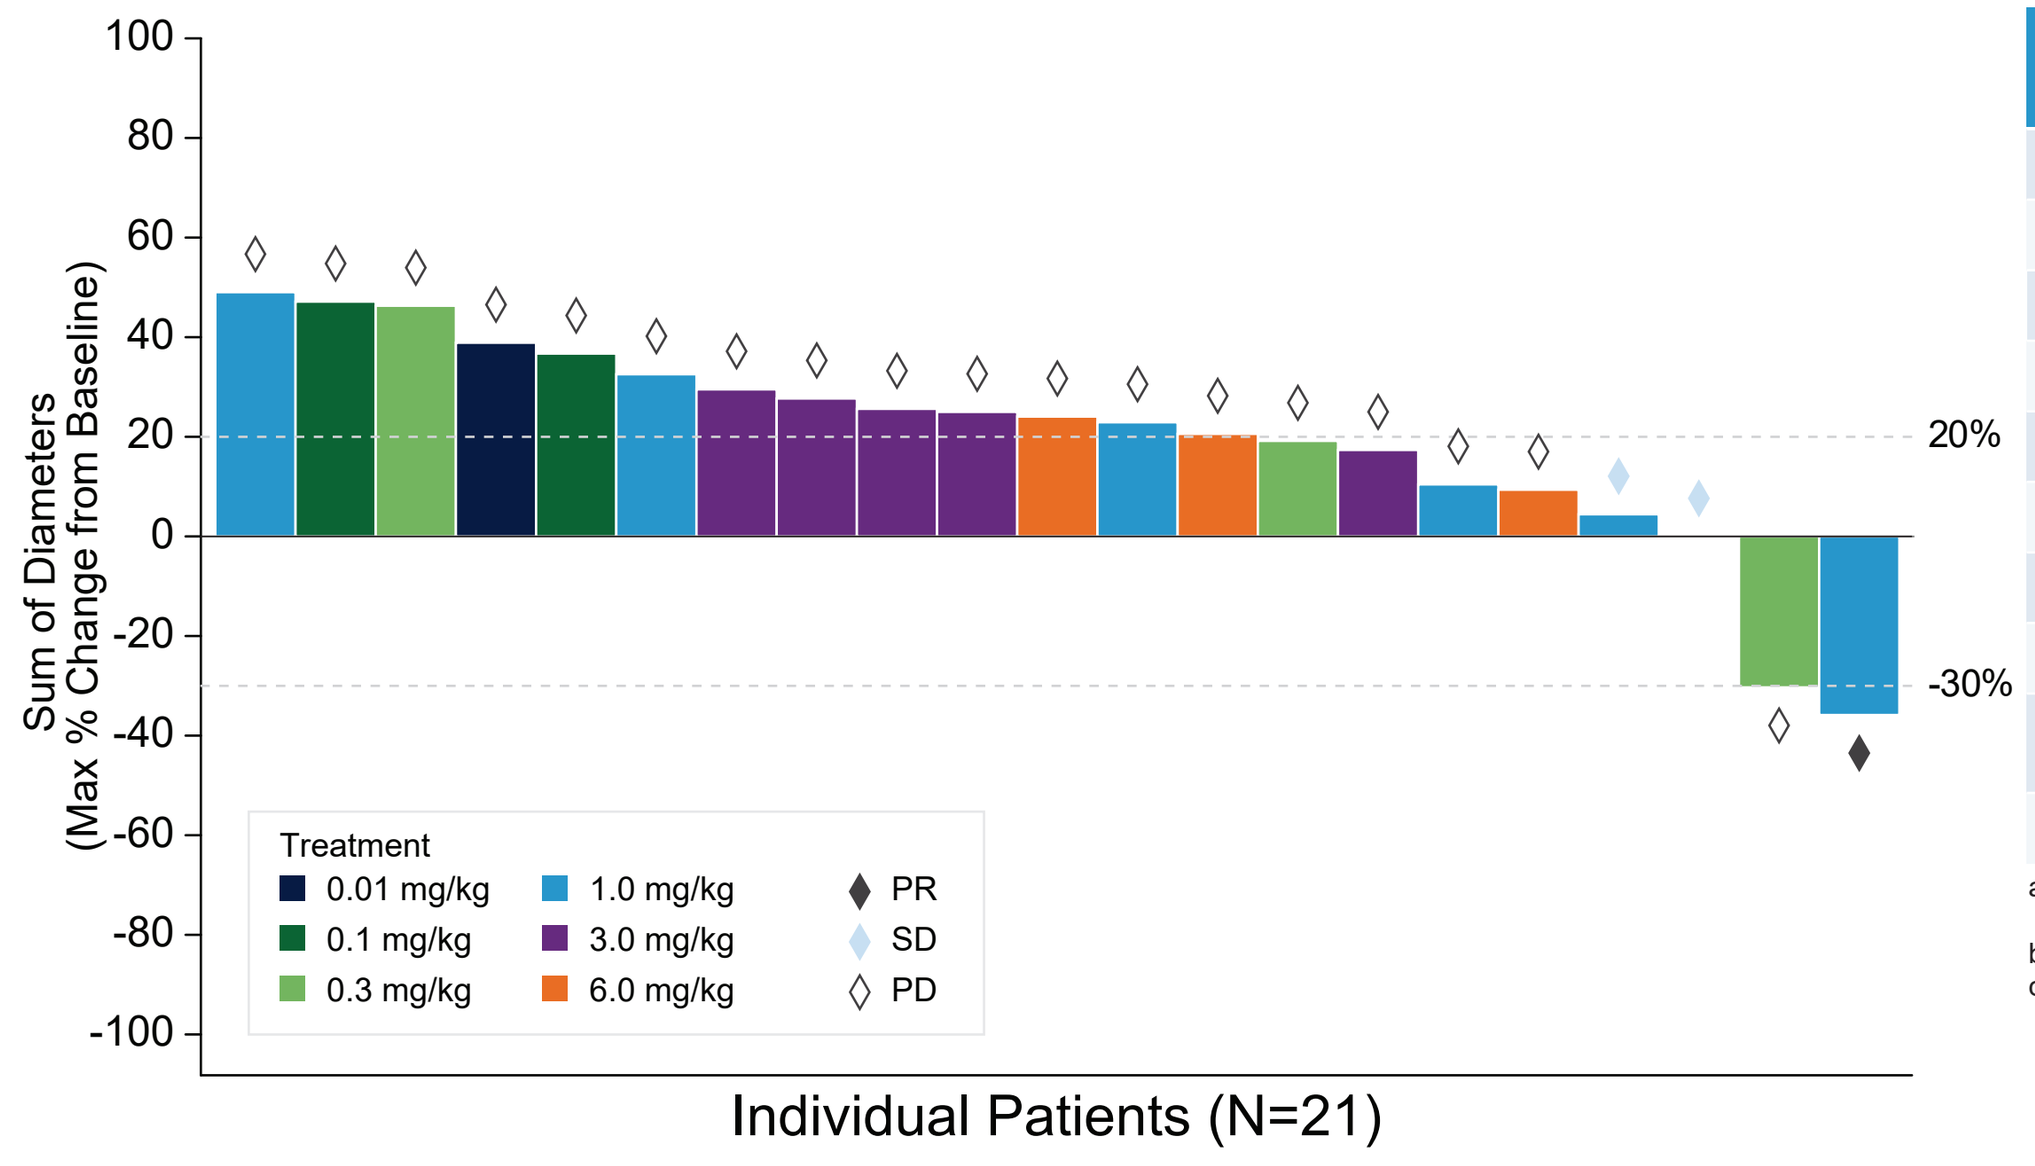

Sum of Diameters (Max % Change from Baseline)
20%
-30%
Treatment
0.01 mg/kg
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
6.0 mg/kg
PR
SD
PD
Individual Patients (N=21)

	Total (N=23) n (%)
Best overall response[a,b]	
CR	0
95% CI[c] for CR rate	(0, 14.82))
PR	1 (4)
95% CI[c] for PR rate	(0.11, 21.95)
SD	2 (9)
PD	18 (78)
NE	2 (9)
Objective response rate (CR+PR)	1 (4)
95% CI[c] for ORR	(0.11, 21.95)

a CR, PR, SD, and PD per response criteria: RECIST v1.1 (Eisenhauer 2009).
b CR, PR, SD, PD, and NE are mutually exclusive.
c Two-sided 95% exact confidence interval, computed using the Clopper-Pearson method (Collett 1991).

Maximum reduction or minimum increase if no reduction.
1 patient (1.0-mg/kg dose level) with PR – gastric cancer
1 patient (0.3-mg/kg dose level) with PD had 31% reduction of target lesions, but developed new lesions – gastric cancer

SEA-TGT Pharmacokinetics

- SEA-TGT pharmacokinetics were approximately dose proportional at doses ranging from 0.3 to 6.0 mg/kg
- For more information see 2023 AACR Abstract #5668; Poster 17, Section 44

Conclusion

- SEA-TGT demonstrated a manageable and tolerable safety profile
- 1 DLT, pruritic rash, was observed in 1 patient at 6.0 mg/kg and MTD was not reached
- Immune-mediated AEs were limited to infusion- related reactions and rashes
- **1.0 mg/kg Q3W was selected as the optimal biological dose** for the expansion cohorts based on PK and pharmacodynamic data (2023 AACR Abstract #5668; Poster 17, Section 44)
- Initial antitumor activity is encouraging and warrants further exploration of SEA-TGT as a single agent and in combination

Abbreviations

AACR: American Association for Cancer Research; AE: Adverse event; CD: cluster of differentiation; cHL: classic Hodgkin lymphoma; CI: confidence interval; CmR: complete metabolic response; CR: complete response; CTCAE: common terminology criteria for Adverse Events; CUI: Clinical Utility Index; DLBCL: diffuse large B cell lymphoma; DLT: dose-limiting toxicity; ECOG: Eastern Cooperative Oncology Group; Fc: fragment crystallizable; GE: gastroesophageal; GPP3: Good Publication Practice; HNSCC: head and neck squamous cell carcinoma; ICMJE: International Committee of Medical Journal Editors; LYRIC: Lymphoma Response to Immunomodulatory Therapy Criteria; MAD: maximum acceptable dose; Max: maximum; MedDRA: Medical Dictionary from Regulatory Activities; MOA: mechanism of action; MTD: maximum tolerated dose; NE: not evaluable; NK: natural killer; NmR: no metabolic response; NSCLC: non-small cell lung cancer; ORR: objective response rate; PD: progressive disease; PK: pharmacokinetics; PmD: progressive metabolic disease; PmR: partial metabolic response; PR: partial response; PTCL-NOS: Peripheral T-cell lymphoma not otherwise specified; Q3W: every 3 weeks; RECIST v1.1: Response Evaluation Criteria in Solid Tumors version 1.1; SD: stable disease; TEAE: treatment-emergent adverse event; TE SAE: treatment-emergent serious adverse event; TIGIT: T-cell immunoreceptor with immunoglobulin and immunoreceptor tyrosine-based inhibitory motif domain; TNBC: triple-negative breast cancer; Tregs: regulatory T-cells

Acknowledgements

This study was sponsored by Seagen Inc., Bothell, WA, USA. The authors thank all patients and families for their participation in the study and all research personnel for their support of this trial. Stacey Therrien, MPH (employee and stockholder of Seagen Inc) and Irene Park, PhD (employee and stockholder of Seagen) provided medical writing and editorial support in accordance with GPP3 guidelines. All authors met the ICMJE criteria for authorship.

Disclosures

EGC: Grant/Contract/Consultant/Advisory/Speaker Bureau/Other – Roche, Thermo Fisher, AZ, Novartis, Taiho, Biogene, Genetech, Elipses Pharma, LaRoche, Neomed Therapeutics, Lilly, Boehringer Ingelheim, Janssen Global Services, Seagen Inc, Alkeme, BMS, MabDiscovery, Anaveon, F-Star Therapeutics, Sanofi. AFT: Employment, Stock, Travel – Seagen Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster, **Elena Garralda Cabanas, egarralda@vhio.net**

Abstract No. CT265
American Association for Cancer Research; Orlando, FL; April 14–19, 2023