ENFORTUMAB VEDOTIN, A NECTIN-4 DIRECTED ANTIBODY-DRUG CONJUGATE, DEMONSTRATES COMPELLING TOLERABILITY AND ANTITUMOR ACTIVITY WITH INTRAVESICAL INSTILLATION IN PRECLINICAL MODELS OF NON-MUSCLE INVASIVE BLADDER CANCER

Abstract No. 1140

Christopher Carosino,[1] Devra Olson,[1] Katie Snead,[1] Anthony Lee,[1] Lauren Farr,[1] Amit Garg,[1] Christine O'Day,[1] Esther Trueblood,[1] Jennifer Wright,[1] Mark Bieda,[1] Charles Caldwell,[1] Kelly Hensley,[1] Sean Allred,[1] Bernard Liu,[1] Masashi Shimazaki,[2] Sharsti Sandall[1]

[1]Seagen Inc., Bothell, WA, USA; [2]Astellas Research Institute of America, LLC, Northbrook, IL, USA

Background

- EV is the first and only treatment to demonstrate a survival benefit with a tolerable safety profile versus chemotherapy in patients with locally advanced/metastatic bladder cancer previously treated with platinum-based chemotherapy and immunotherapy.[1,2]
 - EV is approved in the US, Canada, Japan, Switzerland and Israel.
- Most bladder cancer diagnoses present as NMIBC.[4–7]
- NMIBC is typically treated with TURBT, with the addition of intravesical BCG or chemotherapy for patients with a high risk of recurrence.[4,8]
- BCG is considered the standard of care for high risk NMIBC; however, almost half of patients eventually experience relapse.[9]
 - Shortage of BCG has also impacted treatment options for NMIBC.[10]
- Radical cystectomy is the standard of care for BCG unresponsive disease.[8,11,12] Treatment options are limited to intravesical chemotherapy or PD-1/PD-L1 inhibitors for patients who may be reluctant to undergo surgery.[8,12]
- Local administration of EV via intravesical instillation could enable direct exposure to NMIBC cells with reduced systemic exposure and an improved safety profile compared with systemic administration.

Proposed Mechanism of Action of EV

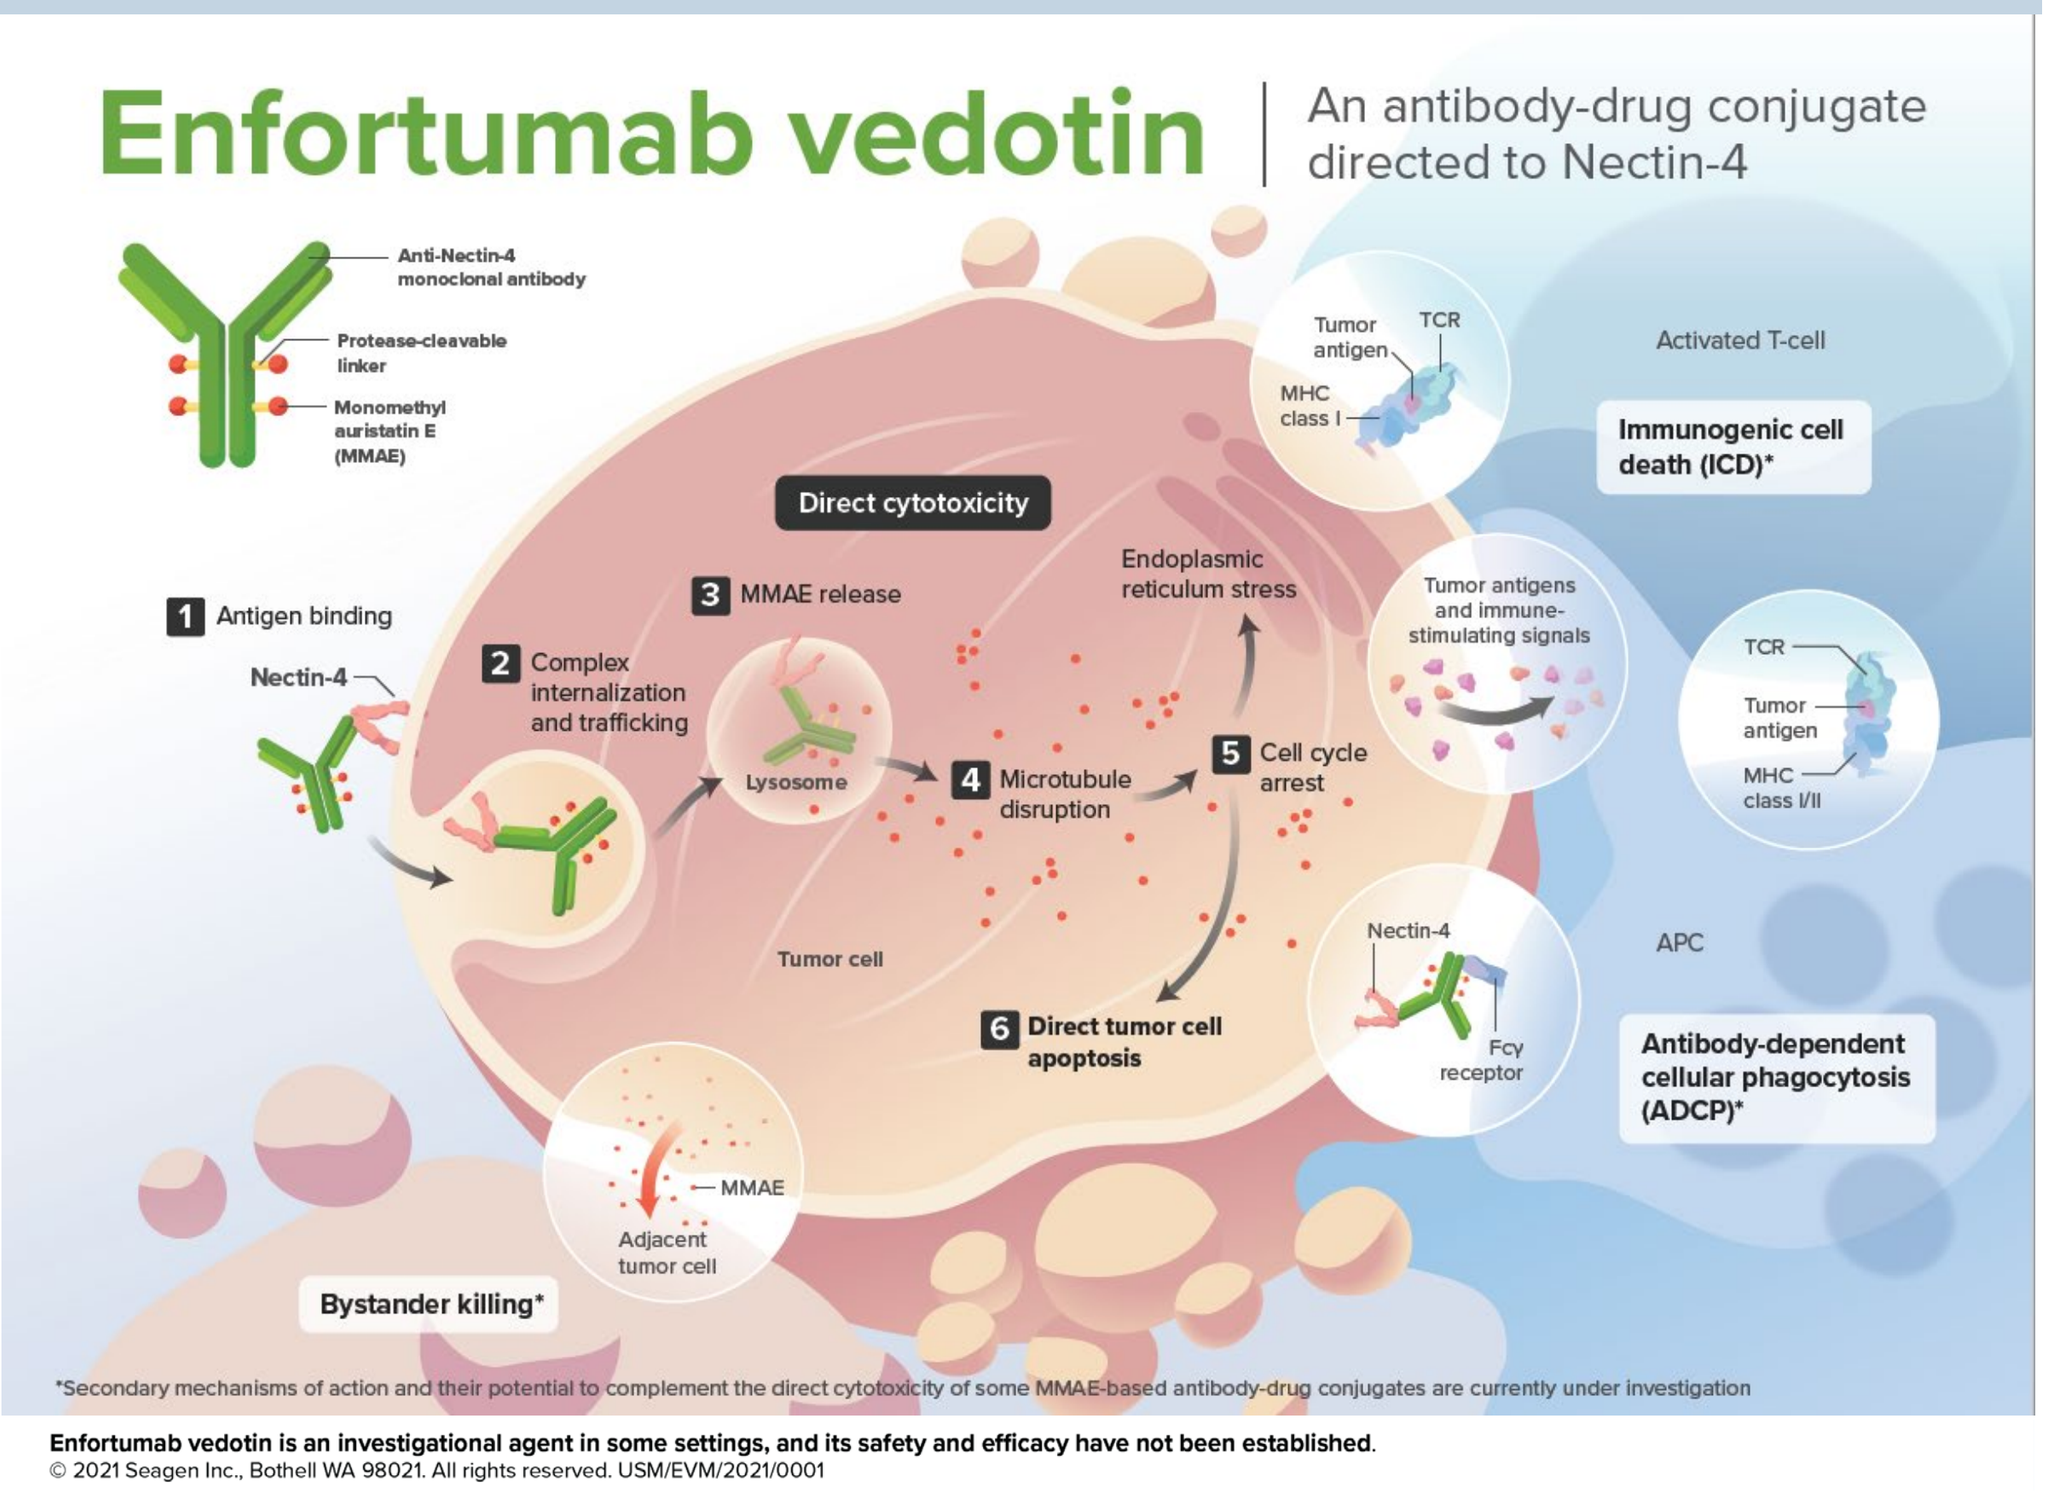
Enfortumab vedotin
An antibody-drug conjugate directed to Nectin-4
Anti-Nectin-4 monoclonal antibody
Protease-cleavable linker
Monomethyl auristatin E (MMAE)
1 Antigen binding
Nectin-4
2 Complex internalization and trafficking
Lysosome
3 MMAE release
Direct cytotoxicity
4 Microtubule disruption
Endoplasmic reticulum stress
5 Cell cycle arrest
6 Direct tumor cell apoptosis
Tumor cell
Bystander killing*
Adjacent tumor cell
MMAE
Immunogenic cell death (ICD)*
Activated T-cell
Tumor antigens and immune-stimulating signals
APC
Antibody-dependent cellular phagocytosis (ADCP)*
Fcγ receptor
*Secondary mechanisms of action and their potential to complement the direct cytotoxicity of some MMAE-based antibody-drug conjugates are currently under investigation
Enfortumab vedotin is an investigational agent in some settings, and its safety and efficacy have not been established.
© 2021 Seagen Inc., Bothell WA 98021. All rights reserved. USM/EVM/2021/0001

Dose Selection for Preclinical Studies

- A mouse orthotopic model was developed in SCID mice utilizing the human urothelial bladder cancer cell line, UM-UC-3, that was engineered to express human Nectin-4 and luciferase (UM-UC-3-hNectin4⁺-Luc⁺).
- Rat was selected for safety evaluation as EV binds with comparable affinity to rat and human Nectin-4 orthologs.

	Total Dose (mg)	Dose Concentration (mg/mL)	Dose Level[a] (mg/kg)	Dose/Bladder Surface Area[b] (mg/cm^2)
Mouse	0.75	15	30	0.5
Rat	2	5	10	0.4
	6	15	30	1.2
	12	30	60	2.5
	20	50	100	4.1
Human (approved total IV dose)	125	–	–	0.4

[a]Assumes 78 kg patient (mean body weight of population pharmacokinetic EV model population), 200 g rat, 25 g mouse. [b]Bladder surface area derived from the volume of bladder where $V=(4/3)\pi r^3$ and $SA=4\pi r^2$.[13] Note: Dose scaling on body surface area is not appropriate due to limited systemic exposure and local administration dose route; instead, total dose has been normalized to bladder tissue surface area to ensure relevant preclinical doses.[14]

Nectin-4 is Highly Expressed in NMIBC and Advanced Bladder Tumors

A

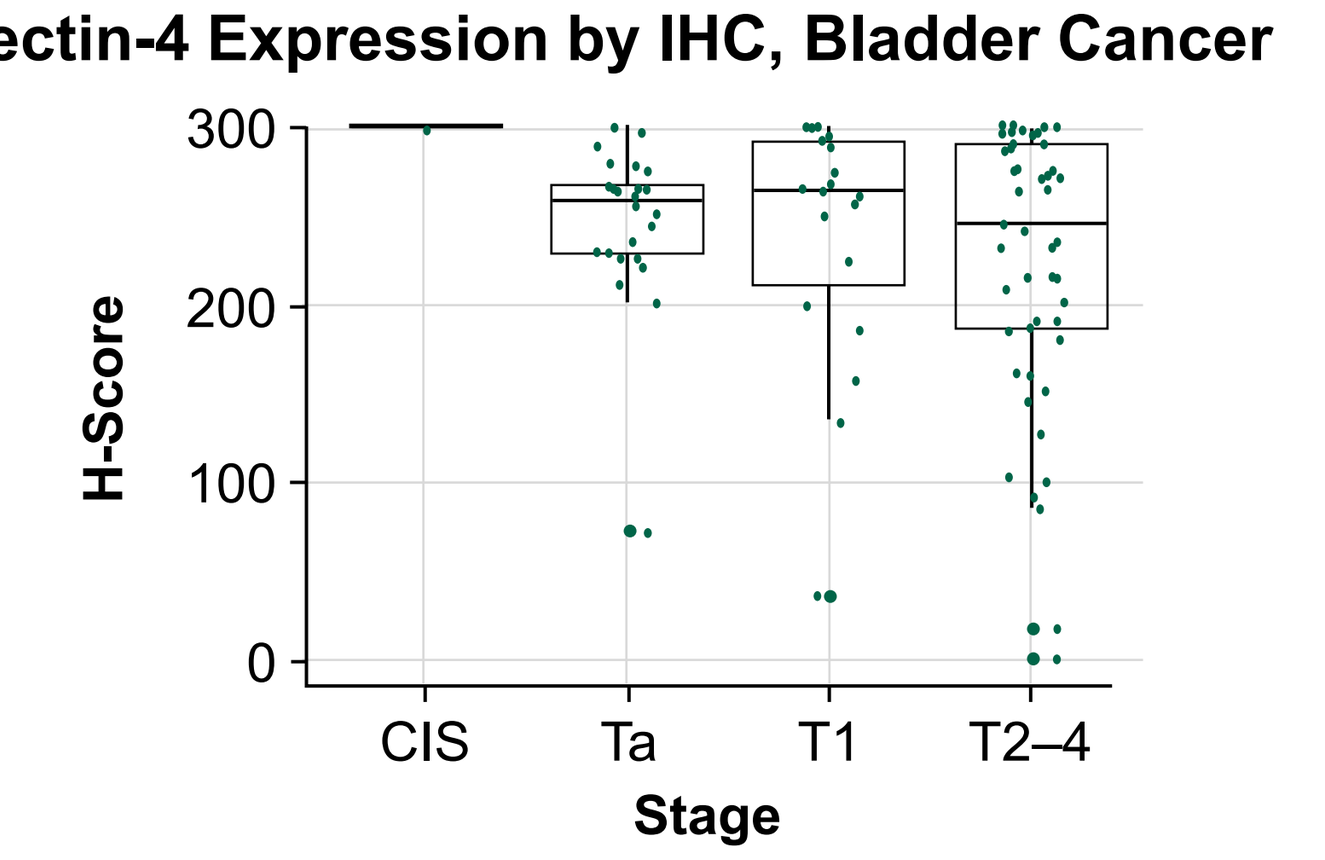
Nectin-4 Expression by IHC, Bladder Cancer
H-Score
300
200
100
0
CIS
Ta
T1
T2–4
Stage

B

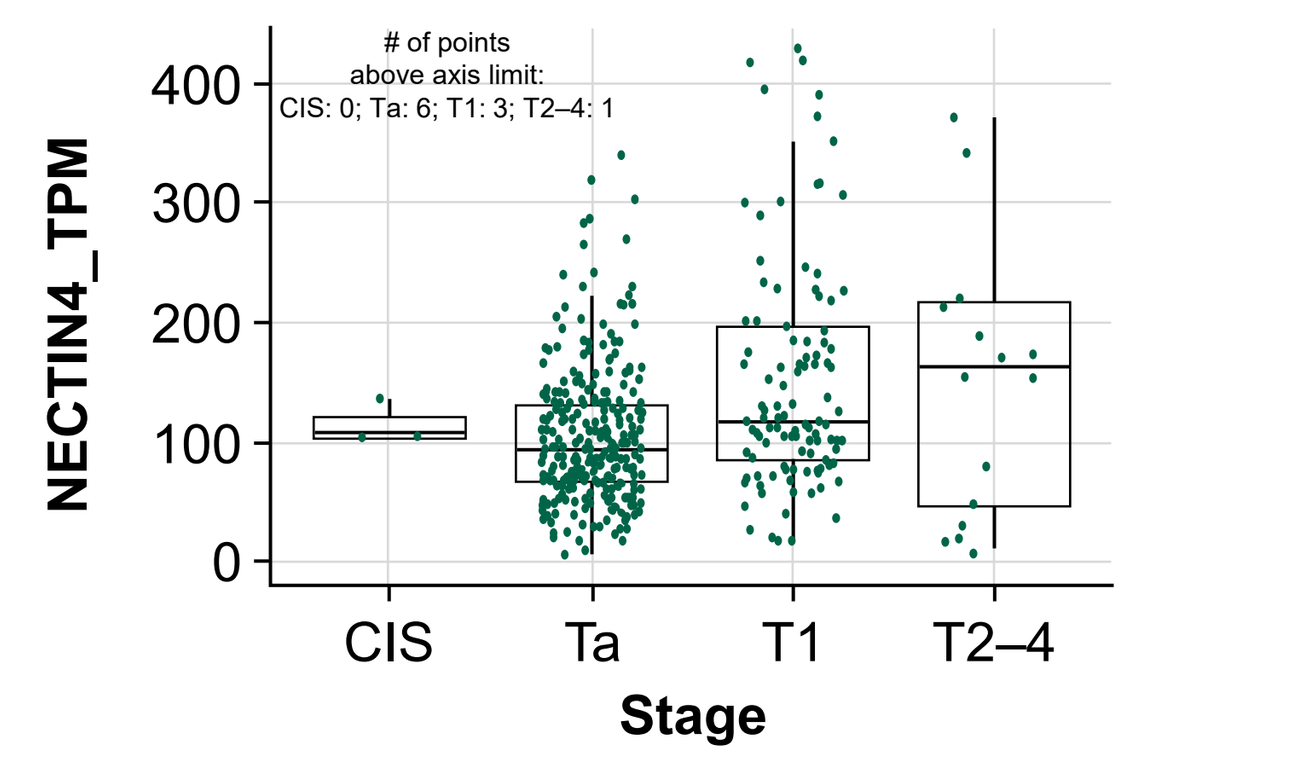
Nectin-4 RNA Expression, Bladder Cancer
From Hedegaard UROMOL study
of points above axis limit:
CIS: 0; Ta: 6; T1: 3; T2–4: 1
NECTIN4_TPM
400
300
200
100
0
CIS
Ta
T1
T2–4
Stage

C

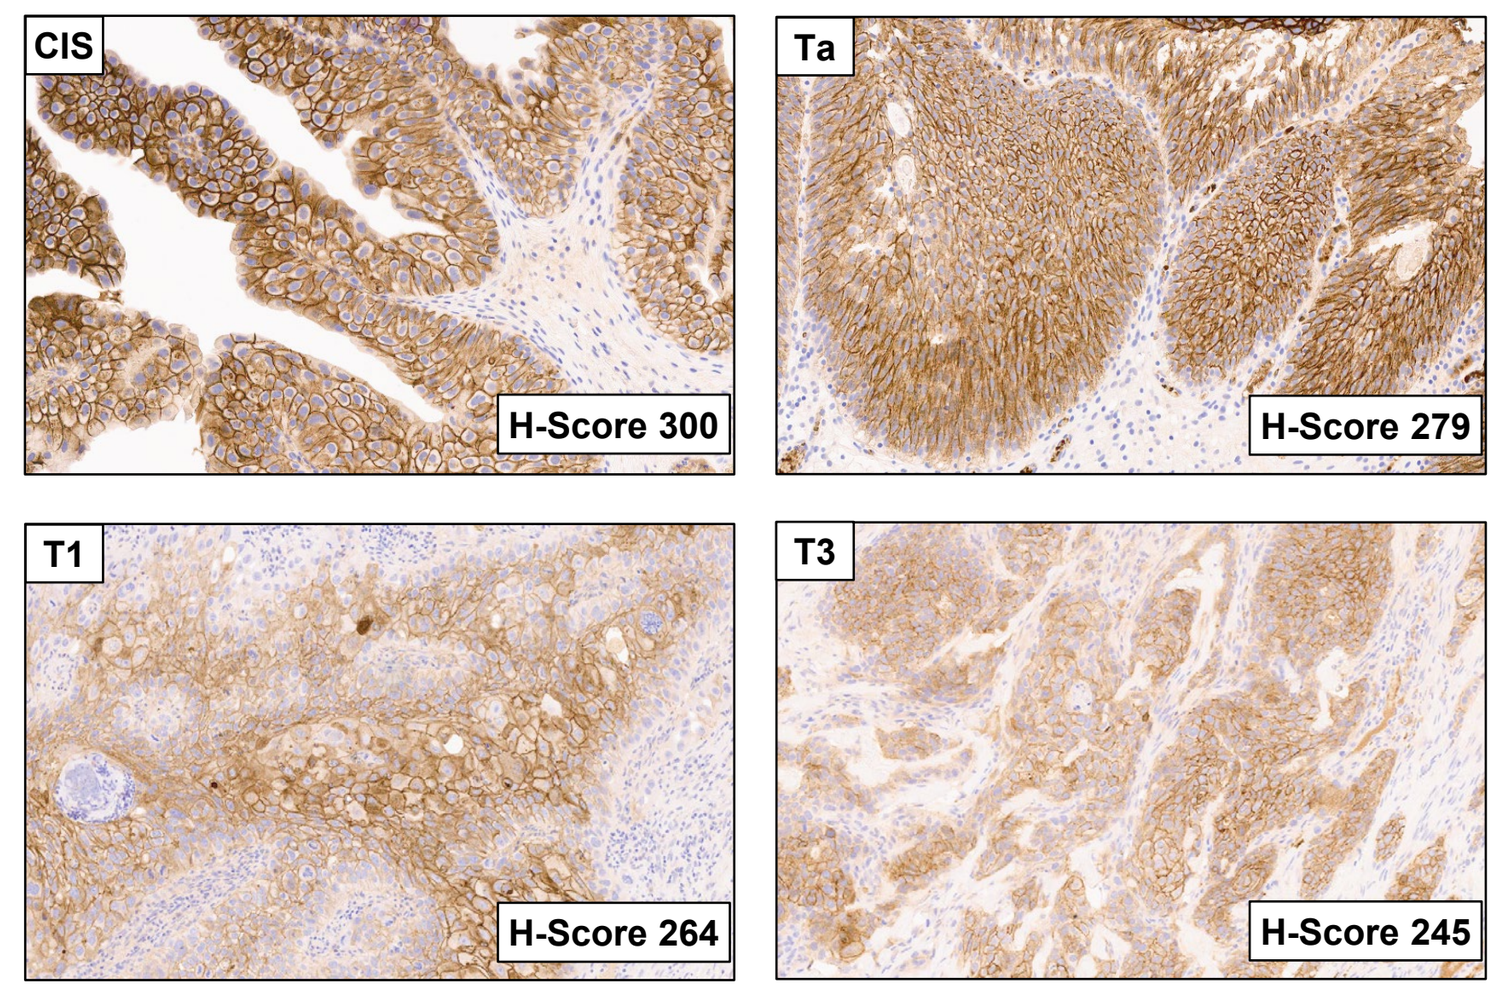
CIS
H-Score 300
Ta
H-Score 279
T1
H-Score 264
T3
H-Score 245

(A) Primary bladder tumor resections were stained using a proprietary IHC assay for Nectin-4 by Q² Solutions® (Durham, NC, USA). Nectin-4 was highly expressed across all stages of bladder cancer, including NMIBC (CIS [n=3], Ta [n=24], T1 [n=21]) and MIBC (T2–T4 [n=53]). **(B)** Nectin-4 mRNA levels are highly overlapping across all stages of bladder cancer (Number of points above axis limit [CIS: 0, Ta: 6, T1: 3, T2–4: 1]). RNA data were obtained from the UROMOL study.[15] **(C)** Representative fields of view at 10x magnification.

EV Maintains Cytotoxic Activity *In Vitro* Using Conditions that Mimic Intravesical Dosing

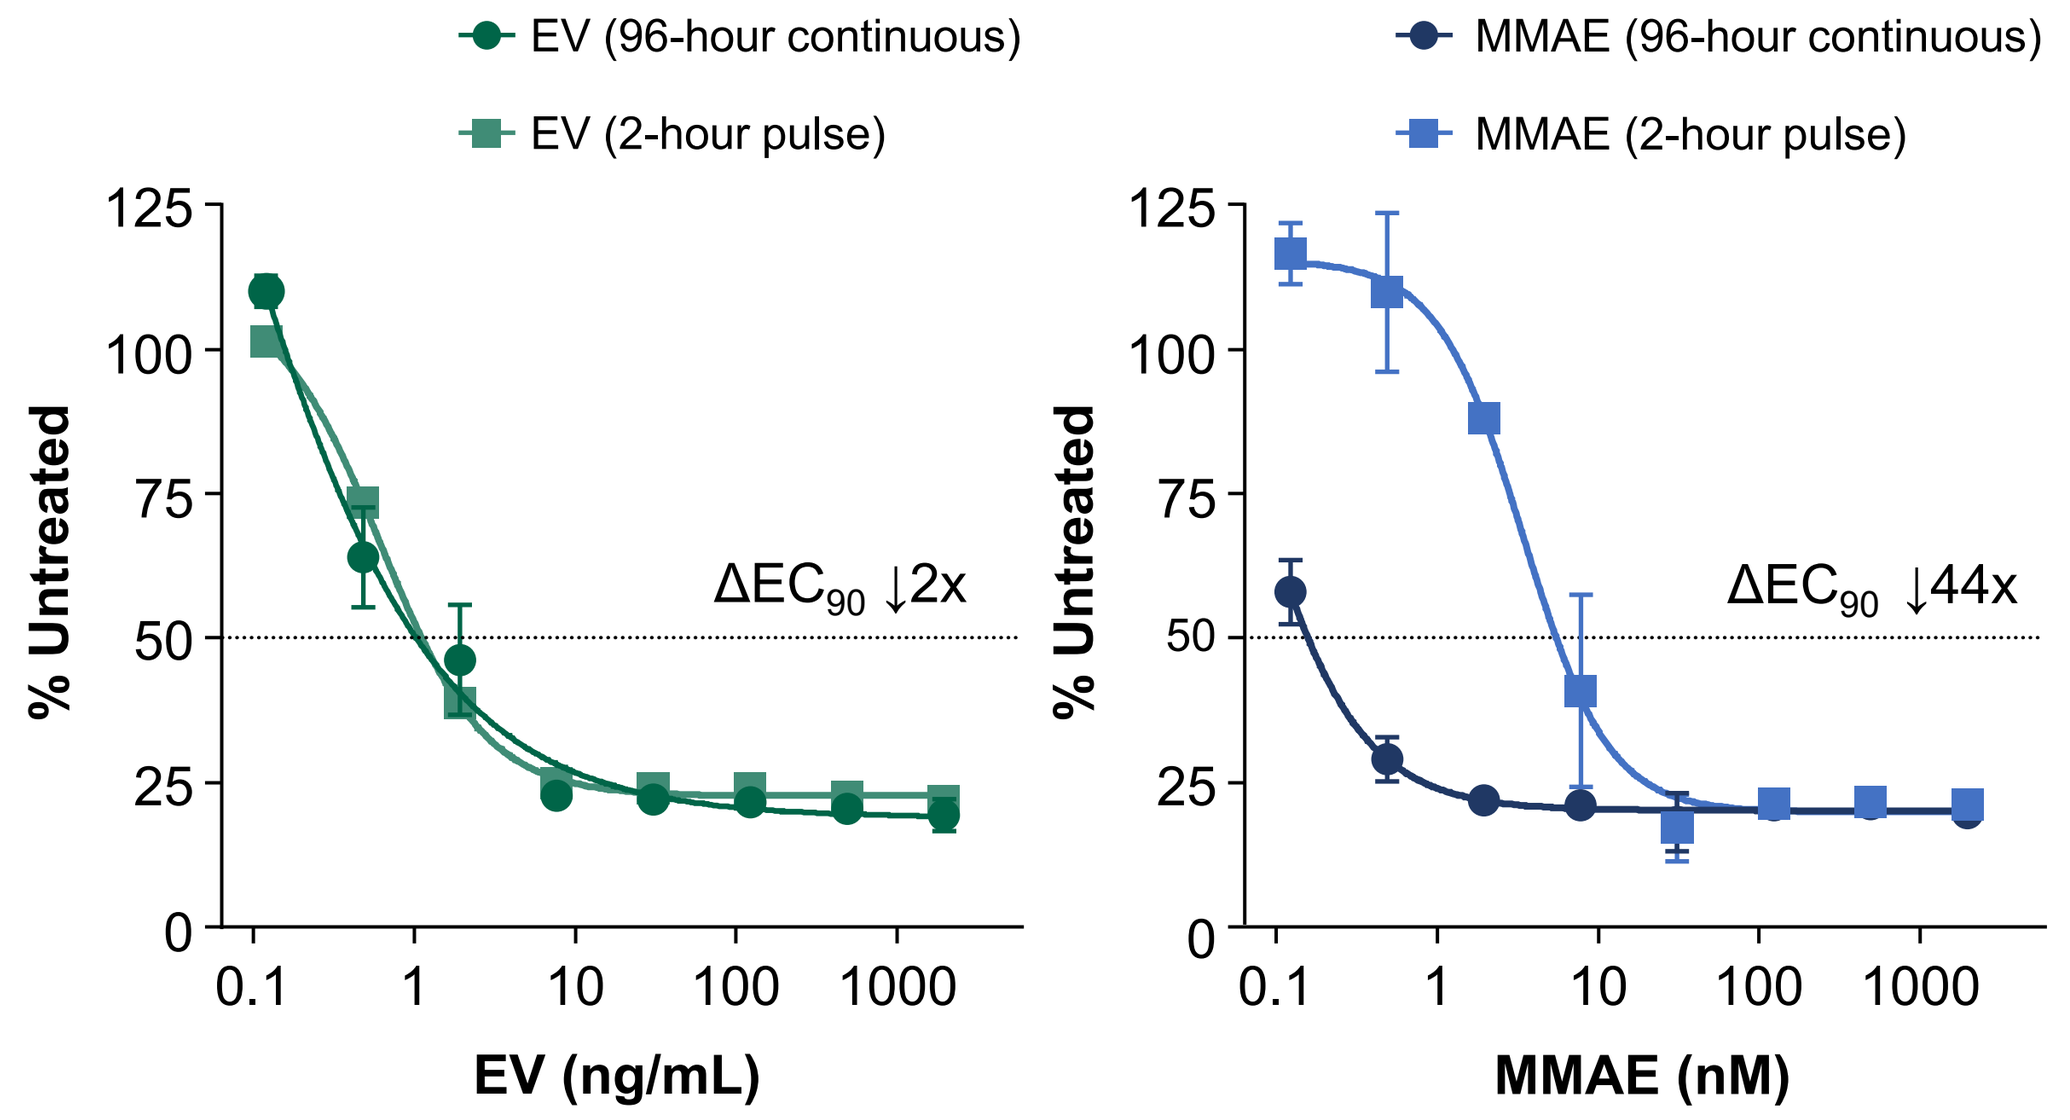
EV (96-hour continuous)
EV (2-hour pulse)
MMAE (96-hour continuous)
MMAE (2-hour pulse)
% Untreated
125
100
75
50
25
0
0.1
1
10
100
1000
ΔEC_{90} ↓2x
EV (ng/mL)
ΔEC_{90} ↓44x
MMAE (nM)

UM-UC-3-hNectin-4⁺ cells were exposed to EV, unconjugated anti–Nectin-4 antibody, unconjugated MMAE, or non-targeted vedotin ADC control. Intravesical dosing was modeled by exposing cells for 96 hours. Intravesical dosing was modeled with exposures of 2 and 24 hours followed by test article wash out. Cell death was measured using Cell TiterGlo® (Promega Corporation, Madison, WI, USA). Decreasing the exposure from 96 to 2 hours decreased potency (EC_{90}) by 44-fold for unconjugated MMAE but was almost unchanged for EV (2-fold). Control ADC and unconjugated anti–Nectin-4 (data not shown) did not result in sufficient activity to determine an EC_{90}.

Intravesical EV Results in Antitumor Activity in an Orthotopic Model of NMIBC

A

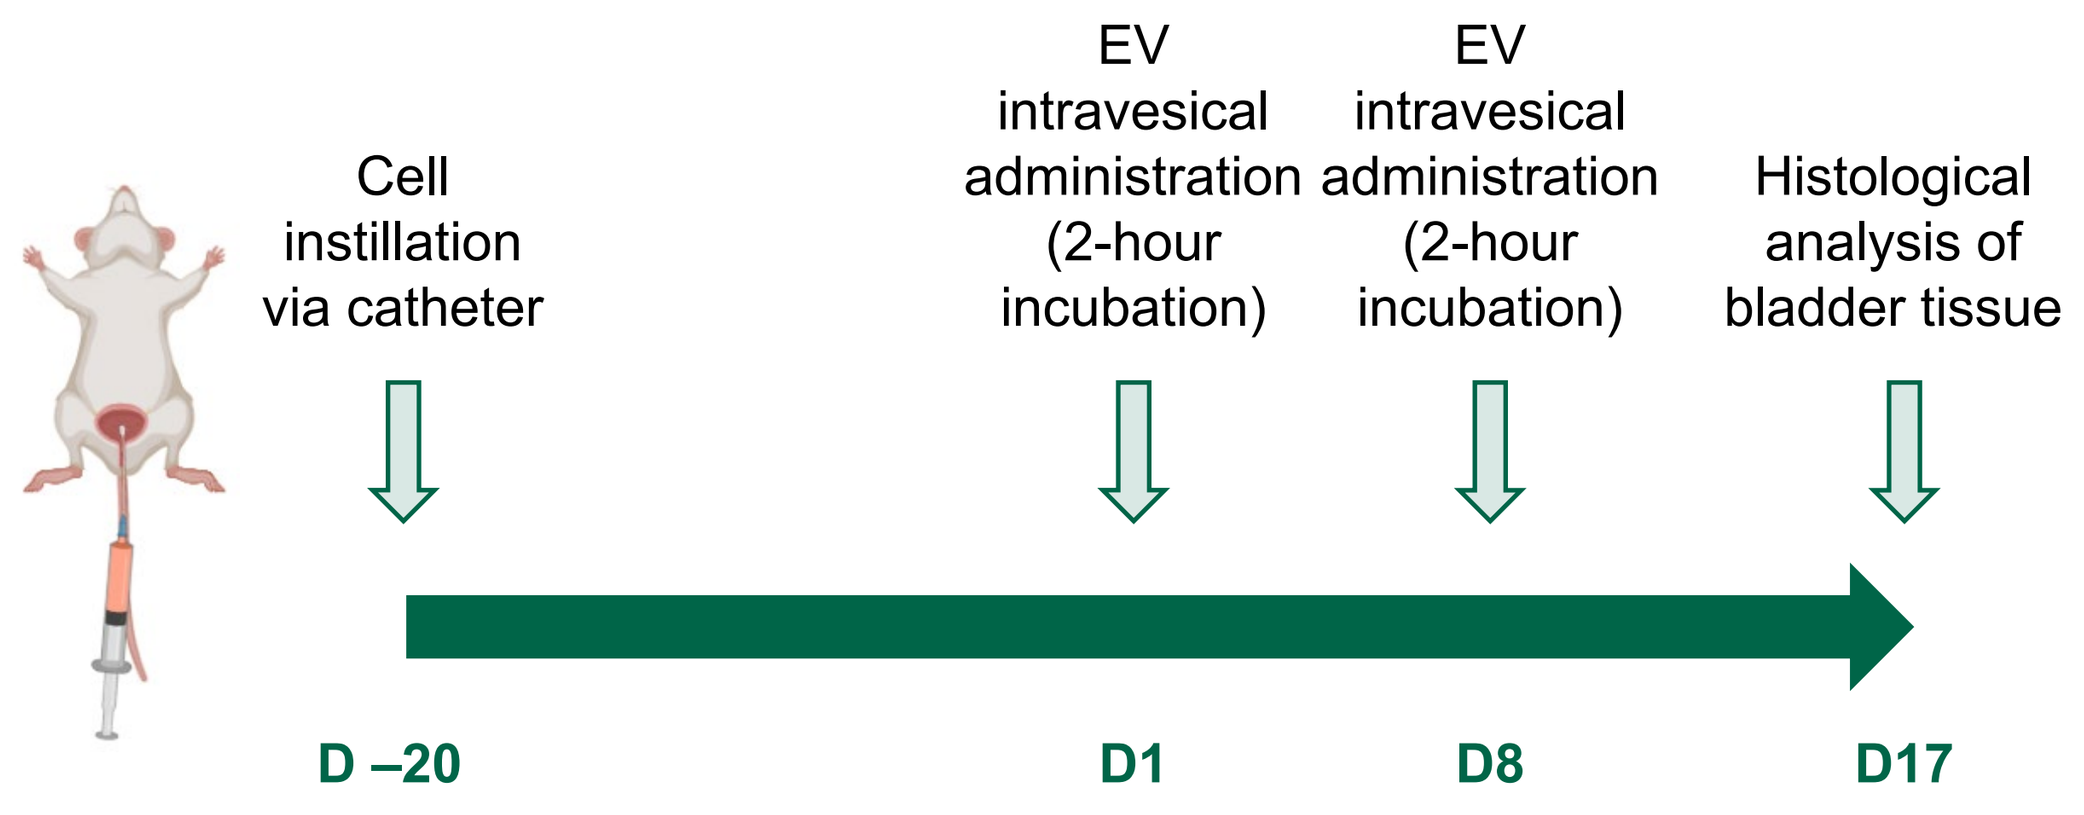
Cell instillation via catheter
EV intravesical administration (2-hour incubation)
EV intravesical administration (2-hour incubation)
Histological analysis of bladder tissue
D –20
D1
D8
D17

B

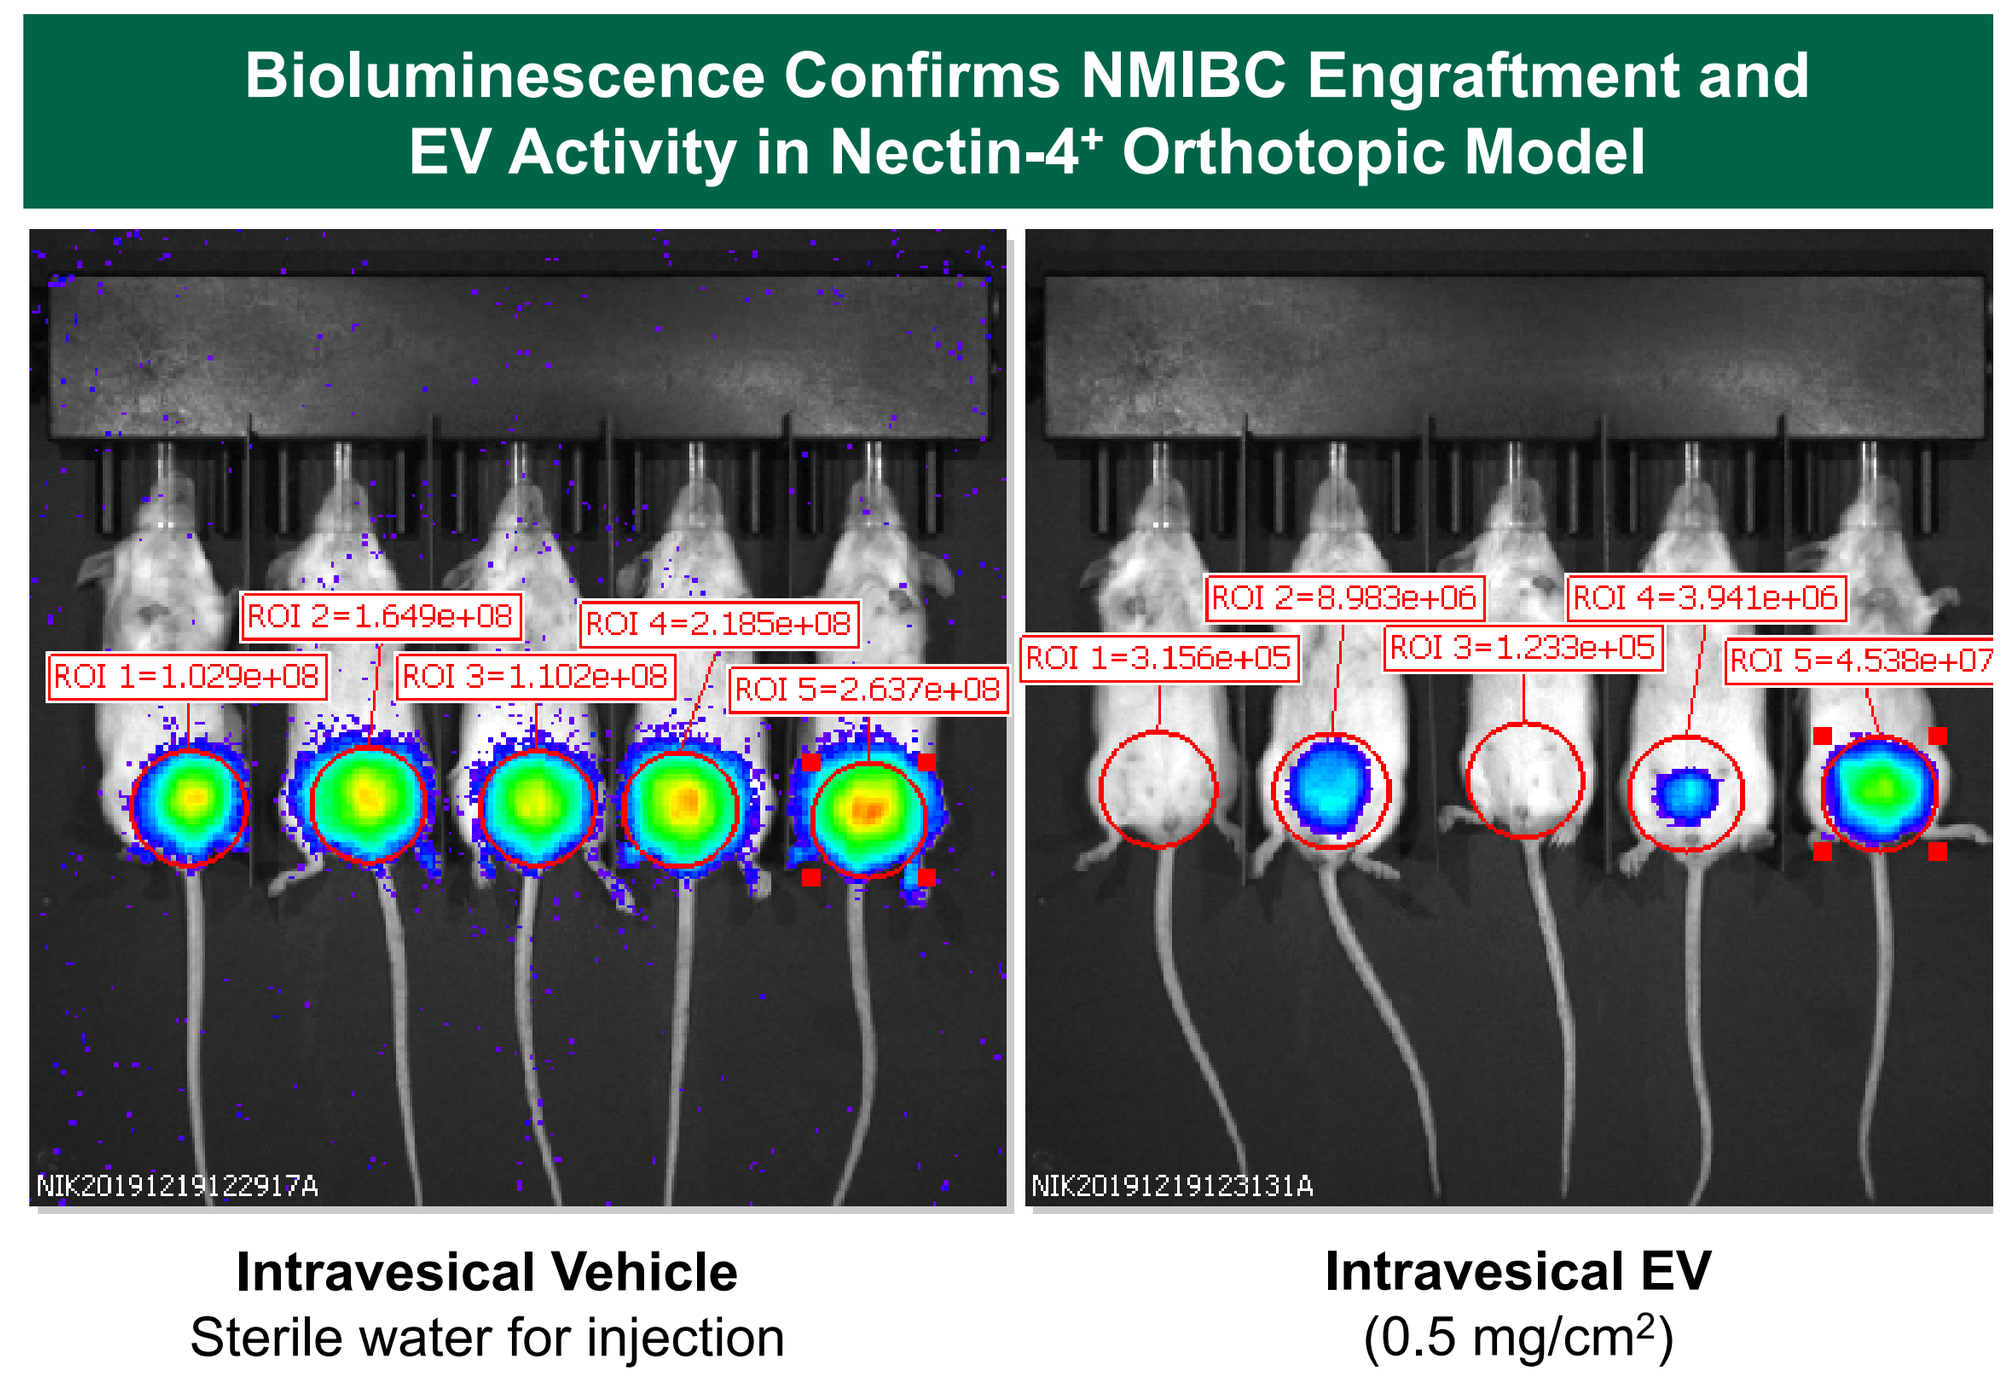
Bioluminescence Confirms NMIBC Engraftment and EV Activity in Nectin-4⁺ Orthotopic Model
ROI 1=1.029e+08
ROI 2=1.649e+08
ROI 3=1.102e+08
ROI 4=2.185e+08
ROI 5=2.637e+08
ROI 1=3.156e+05
ROI 2=8.983e+06
ROI 3=1.233e+05
ROI 4=3.941e+06
ROI 5=4.538e+07
NIK20191219122917A
NIK20191219123131A
Intravesical Vehicle
Sterile water for injection
Intravesical EV
(0.5 mg/cm²)

C

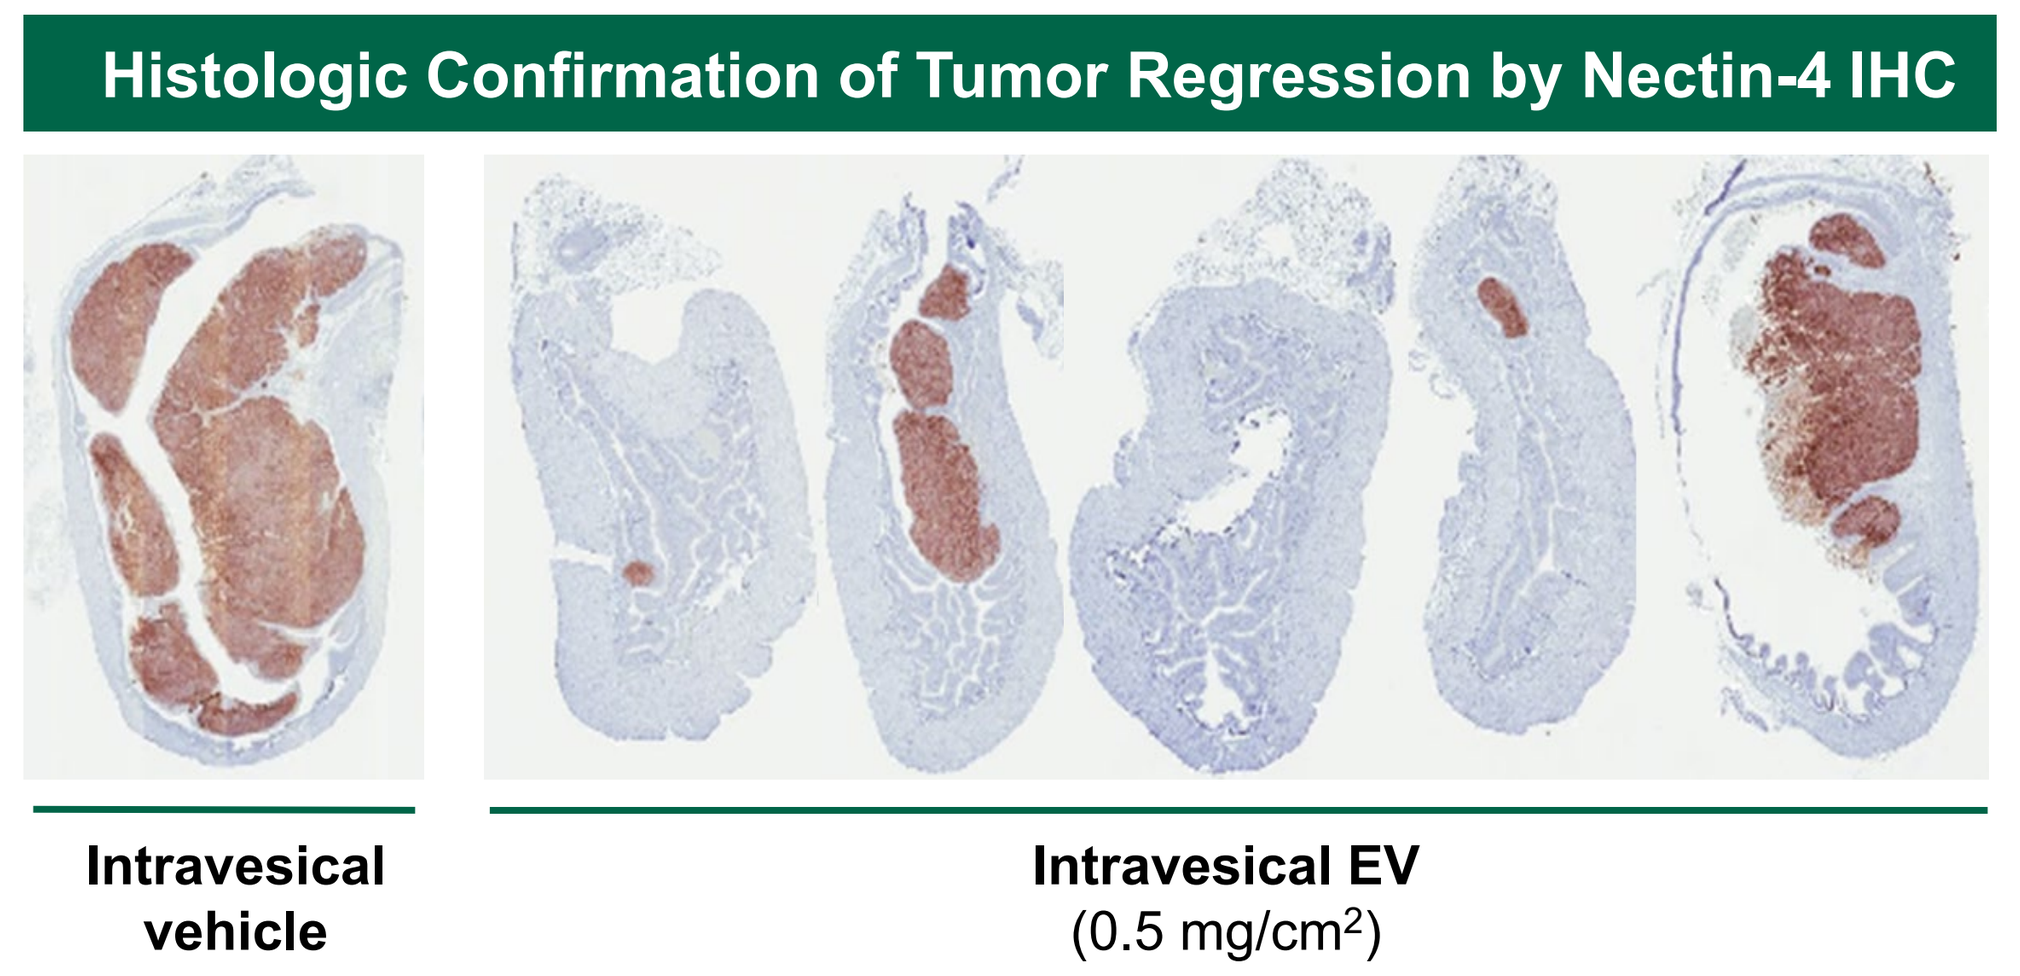
Histologic Confirmation of Tumor Regression by Nectin-4 IHC
Intravesical vehicle
Intravesical EV
(0.5 mg/cm²)

D

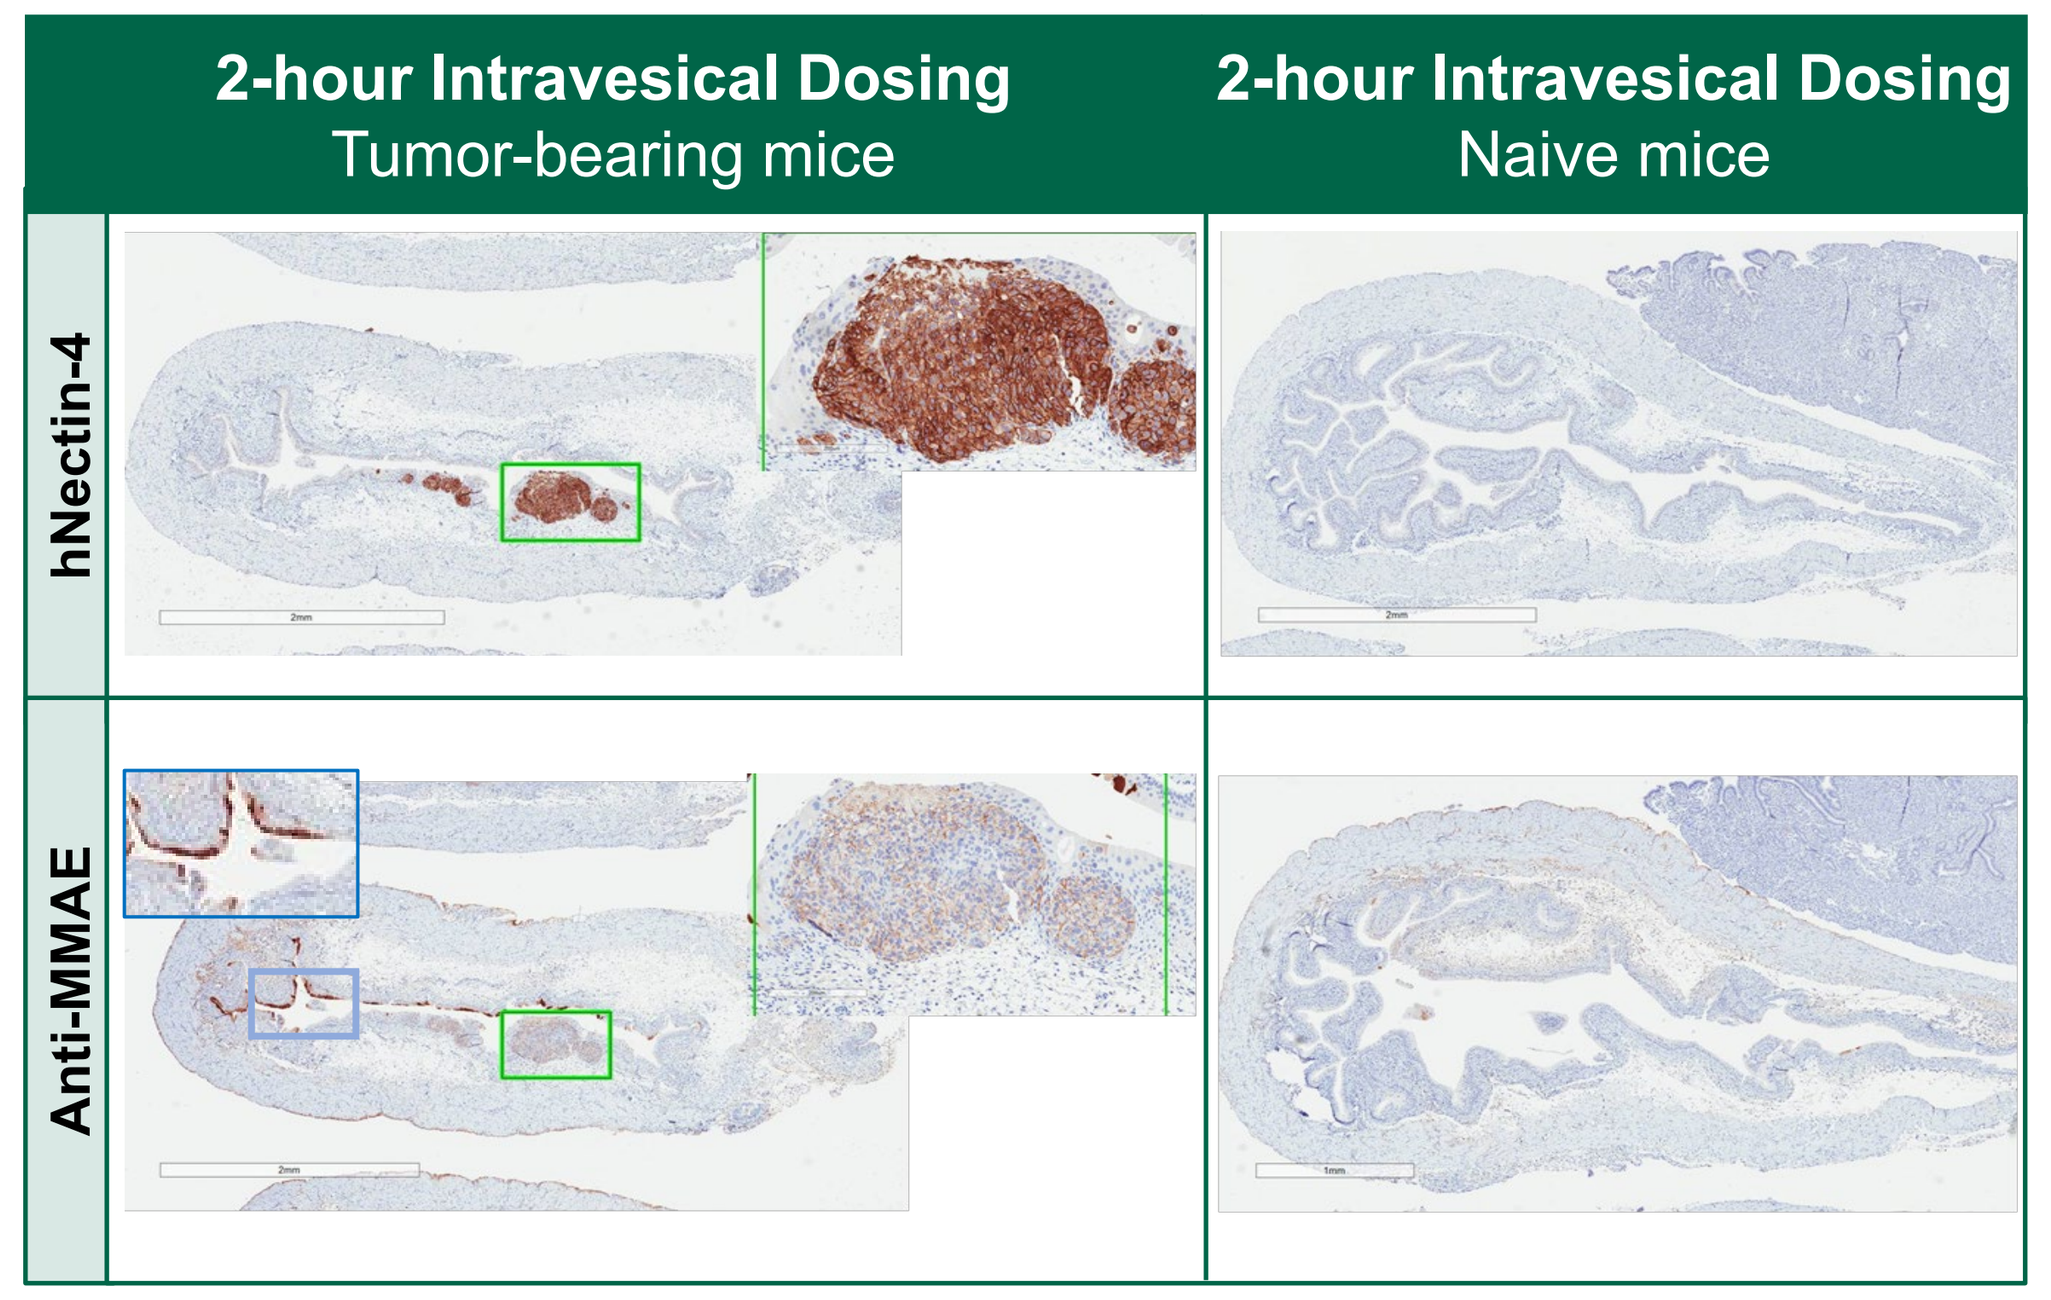
2-hour Intravesical Dosing
Tumor-bearing mice
2-hour Intravesical Dosing
Naive mice
hNectin-4
Anti-MMAE

SCID mice were sedated and orthotopically implanted following chemical abrasion with UM-UC-3-hNectin4⁺-Luc⁺. Engraftment and disease progression was confirmed via bioluminescence imaging. **(A)** Intravesical EV was administered once weekly for 2 weeks (0.5 mg/cm^2; dwell time 2 hours). **(B)** TGI of 97% was calculated on Day 17 by analyzing total flux units (photons/second) compared to control. **(C)** Tumor regression was confirmed by decreased immunohistochemical staining for Nectin-4 (M22-488, an anti–Nectin-4 antibody conjugated with Alexa 488). **(D)** Six hours following the first EV dose, tumors were collected and stained for Nectin-4 (top row) and anti-MMAE (bottom row). IHC analysis showed the presence of Nectin-4 in bladder tumor tissue with co-localization of drug detected by a biotin conjugated anti-MMAE primary antibody (15.22).

Total Dose and Concentration Drive Bladder Tissue Drug Concentrations

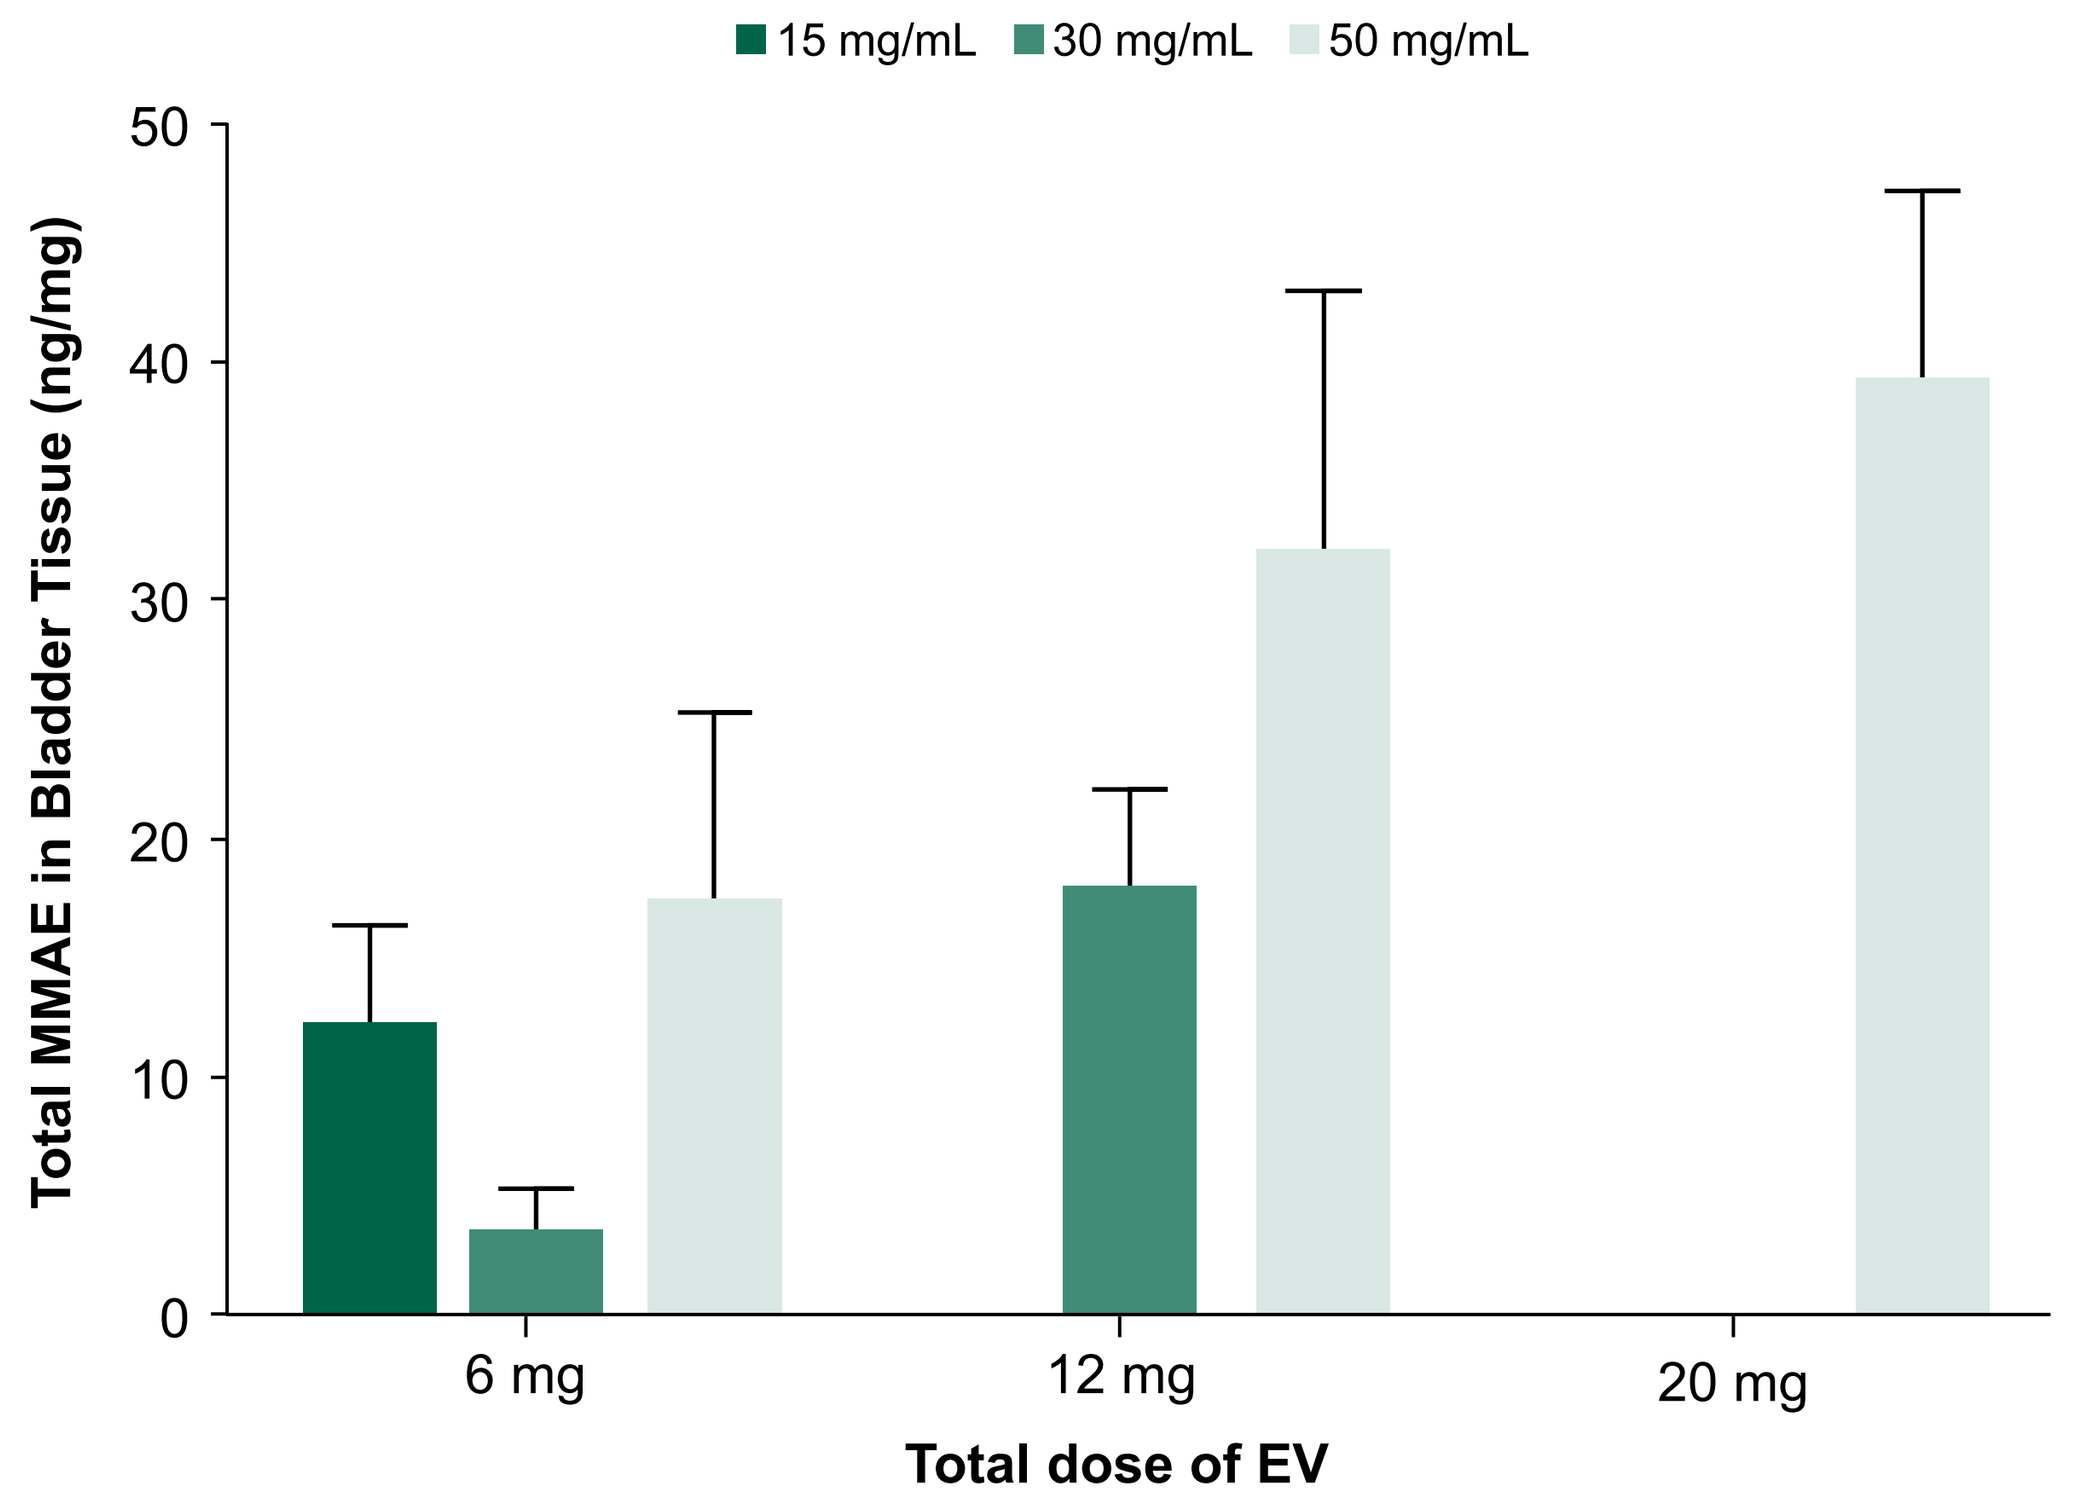
15 mg/mL
30 mg/mL
50 mg/mL
Total MMAE in Bladder Tissue (ng/mg)
50
40
30
20
10
0
6 mg
12 mg
20 mg
Total dose of EV

Single intravesical doses of EV were administered to female rats at doses of 0.3–4.1 mg/cm^2 at 15–50 mg/mL with a dwell time of 2 hours, representing doses of up to 100 mg/kg on a mass basis. Bladder levels of total MMAE at 24 hours were normalized to bladder tissue weight. MMAE was detected in bladder tissue for up to 7 days with peak concentrations within 24 hours of dose administration. Serum MMAE was generally undetectable (<1 ng/mL) and all dose levels well tolerated. There was no meaningful difference in total MMAE levels when concentration was held constant and dwell time varied from 30–120 minutes.

Intravesical EV Demonstrated a Promising Safety Profile in a Repeat Dose Toxicity Study

- To evaluate the potential for toxicity and characterize the pharmacokinetics of intravesical EV, 6 weekly intravesical doses of EV or control were administered to female rats.

Weekly Intravesical Dose Level	Clinical Signs, Clinical Pathology, Organ Weights, Mortality, Macroscopic Findings	Microscopic Findings	Conclusions
0.1 mg/cm^2 (3 mg/kg)	No EV-related findings	No EV-related findings	–
0.4 mg/cm^2 (10 mg/kg)	No EV-related findings	No EV-related findings	No-observed-effect-level (NOEL)
1.2 mg/cm^2 (30 mg/kg)	No EV-related findings	Minimal/slight mitotic/apoptotic figures in the transitional epithelium of the kidney and bladder	No-observed-adverse-effect-level (NOAEL)

- There were no microscopic findings in previously identified target tissues, including the skin and bone marrow.
- The no-effect level of EV is equivalent to >20-fold the approved human IV dose.

Intravesical EV Limits Systemic Exposure

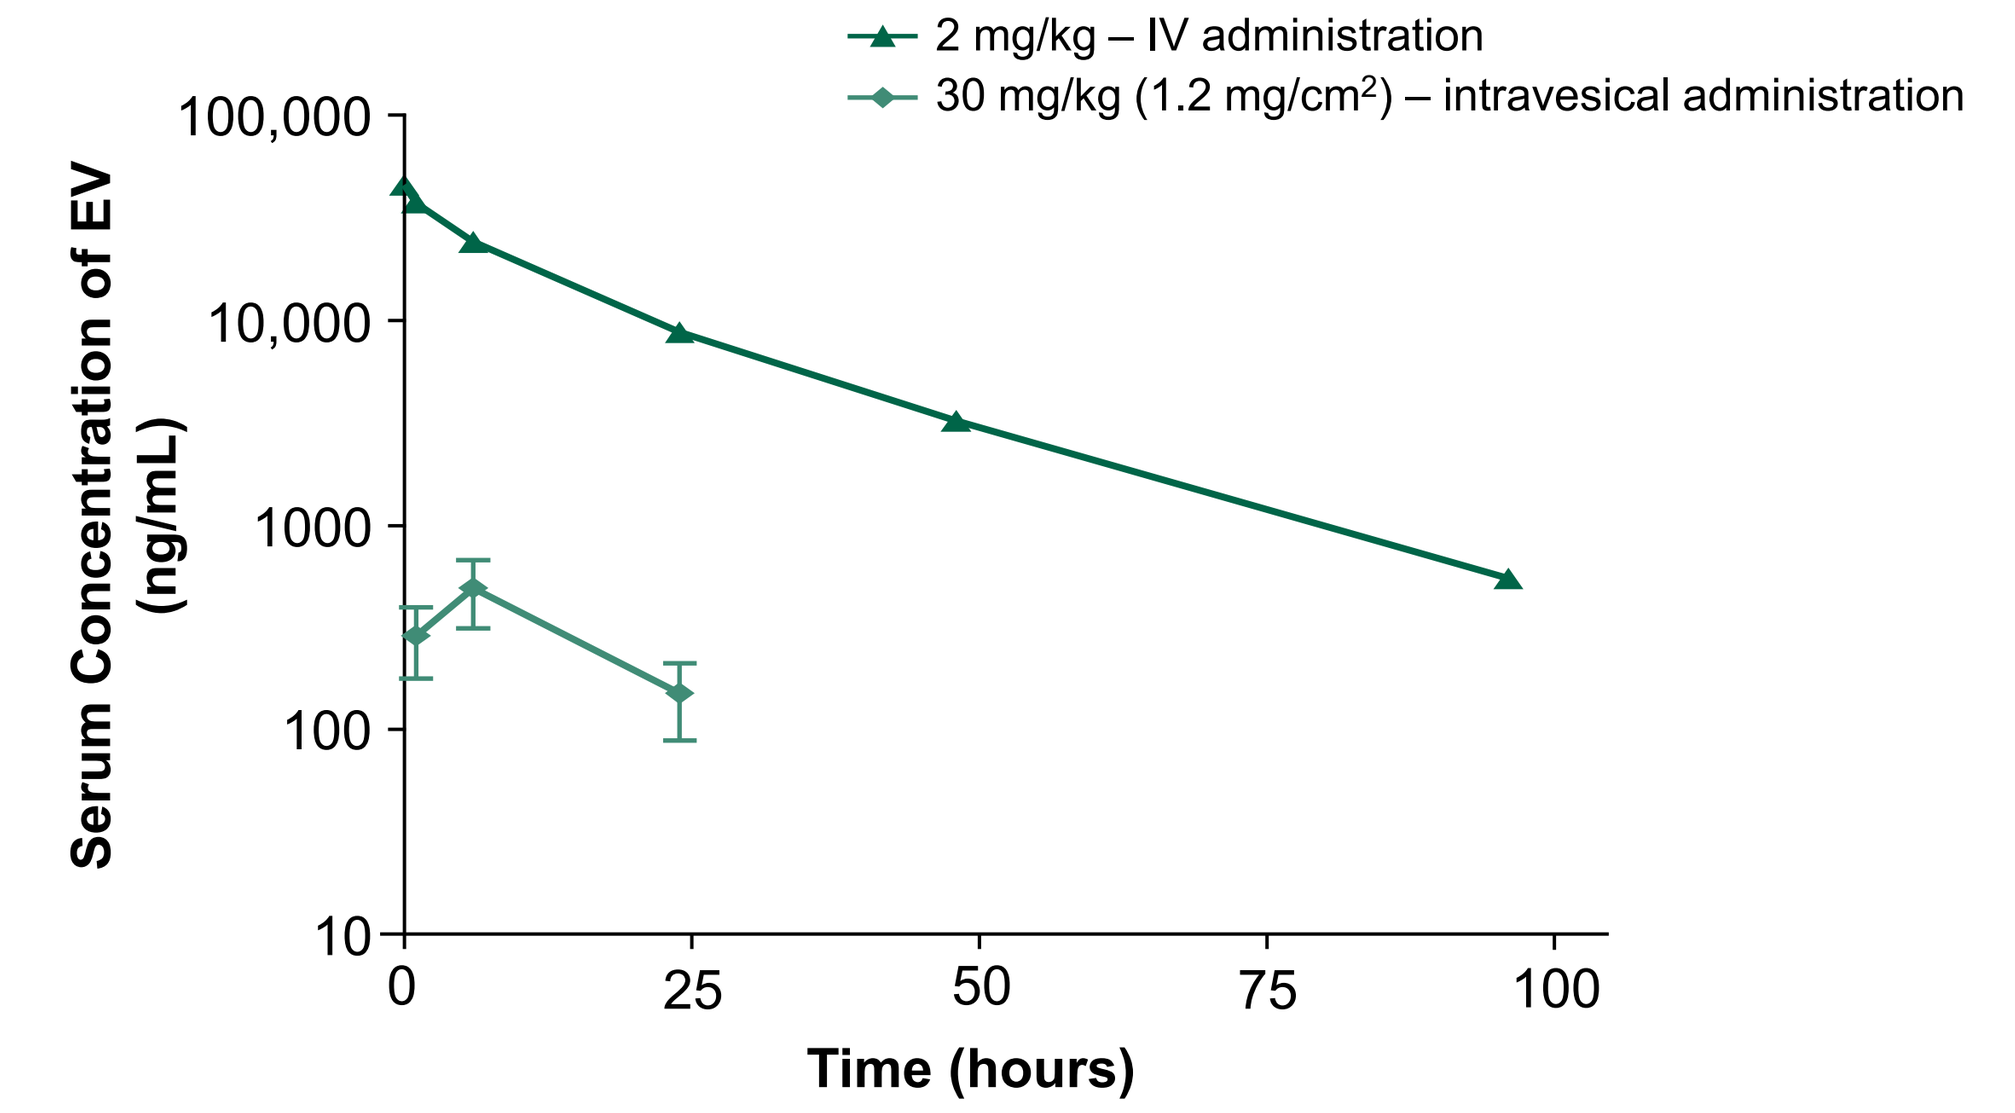
2 mg/kg – IV administration
30 mg/kg (1.2 mg/cm²) – intravesical administration
Serum Concentration of EV (ng/mL)
100,000
10,000
1000
100
10
0
25
50
75
100
Time (hours)

Low and transient serum concentrations of intravesical EV were determined following the first dose of EV by a validated ELISA-based assay (mean ± SEM). Mean EV C_{max} was ≤750 ng/mL (>35-fold lower than the IV C_{max} of the clinically approved dose) and there was no detectable serum MMAE by the validated mass spectrometry assay. Estimation of the serum area under the time–concentration curve was limited to only the highest EV dose (30 mg/kg) only detectable up to 24 hours post-instillation. Serum concentrations of unconjugated MMAE were below the lower limit of quantitation (<10 pg/mL).

Conclusions

- Nectin-4, the target of EV, was highly prevalent across early- and late-stage bladder tumors, including NMIBC.
- Preclinical models of translationally relevant doses of intravesical EV in vitro and in vivo demonstrated antitumor activity in human bladder cancer cells expressing Nectin-4.
- Delivering a higher total dose and concentration of EV may enhance activity and increase tissue drug levels more so than changes to volume instilled or dwell time.
- Intravesical administration of EV was well tolerated with no detectable local or systemic toxicities, and low and transient systemic absorption.
- Low systemic absorption of intravesical EV may lower the incidence of most common adverse events observed with systemically administered EV.
- These preclinical findings provide evidence to support further investigation of intravesical EV in patients with NMIBC (EV-104; NCT05014139).

References

1. PADCEV [package insert]. Northbrook, IL: Astellas Pharma US, Inc., and Bothell, WA: Seagen, Inc. Updated November 2021. 2. Petrylak D, et al. *J Clin Oncol*. 2022;Suppl 6:abstract 435. 3. Powles T, et al. *N Engl J Med*. 2021;384:1125-1135. 4. Chang SS, et al. *J Urol*. 2016;196:1021-1029. 5. Woldu SL, et al. *BJU Int*. 2017;119:371-380. 6. Li R, et al. *BMC Urol*. 2020; 20:97. 7. Kates M, et al. *Clin Cancer Res*. 2020;26:882-891. 8. Kikuchi E, et al. *Int J Urol*. 2020;27:108-116. 9. Hussain M, et al. *J Clin Oncol*. 2009;34:5680-5684. 10. American Urological Association. October 2020. Announcement about BCG Production, Supply. https://www.auanet.org/ about-us/bcg-shortage-info. Accessed March 1, 2022. 11. Lavallée LT, et al. *PLoS One*. 2014;9:e111281. 12. Babjuk M, et al. *Eur Urol*. 2019;76:639-657. 13. Andersson KE, et al. *Physiol Rev*. 2004;84:935-986. 14. U.S. Food and Drug Administration. Estimating the Maximum Safe Starting Dose in Initial Clinical Trials for Therapeutics in Adult Healthy Volunteers 2005. https://www.fda.gov/media/ 72309/download. Accessed March 1, 2022. 15. Hedegaard J, et al. *Cancer Cell*. 2016;30:27-42.

Abbreviations
ADC, antibody-drug conjugate; BCG, Bacillus Calmette-Guérin; CIS, carcinoma in situ; C_{max}, maximum concentration; ΔEC_{90}, change in EC_{90}; EC_{90}, concentration inducing 90% of maximal response; ELISA, enzyme-linked immunosorbent assay; EV, enfortumab vedotin; IHC, immunohistochemistry; IV, intravenous; MIBC, muscle invasive bladder cancer; MMAE, monomethyl auristatin E; NMIBC, non-muscle invasive bladder cancer; OS, overall survival; PD-1, programmed cell death protein-1; PD-L1, programmed death-ligand 1; RNA, ribonucleic acid; SCID, severe combined immune deficient; SEM, standard error of mean; TGI, tumor growth inhibition; TPM, transcripts per million; TURBT, transurethral resection of bladder tumor.

Disclosures
Study funded by Seagen Inc., and Astellas Research Institute of America, LLC. All authors are employees of Seagen Inc., except for MS who is an employee of Astellas Research Institute of America, LLC.

Ethical Conduct of Research Statement
All studies described were performed according to applicable national laws and guidelines with institutional IACUC approval.

Acknowledgements
Medical writing support was provided by Annie Rowe, PhD, and editorial support by George Chappell, MSc, all of Scion, London, supported by Seagen Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster,
Christopher Carosino (CCarosino@seagen.com).

Presented at the American Association for Cancer Research Annual Meeting 2022; April 8–13, 2022; New Orleans, LA, USA