Abstract CT164
American Association for Cancer Research
Orlando, FL
April 14-19, 2023

Tisotumab Vedotin in Squamous Cell Carcinoma of Head and Neck: Interim Analysis From innovaTV 207

Beatriz Cirauqui,[1] Sebastien Salas,[2] William N. William Jr,[3] Ariel E. Birnbaum,[4] Kristi Schmidt,[5] Xuesong Guan,[5] Ibrahima Soumaoro,[6] Leonardo Nicacio,[5] Fortunato Ciardiello[7]

[1]Catalan Institute of Oncology, Badalona, Barcelona, Spain; [2]Aix-Marseille Université, Marseille, France; [3]A Beneficência Portugesa de São Paulo, São Paulo, Brazil; [4]Lifespan Cancer Institute, Providence, Rhode Island, USA; [5]Seagen Inc., Bothell, WA, USA; [6]Genmab US, Inc., Plainsboro, NJ, USA; [7]Università degli studi della Campania Luigi Vanvitelli, Caserta, Italy

BACKGROUND

- Globally, an estimated 932,000 patients were diagnosed with head and neck cancer in 2020, and more than 467,000 deaths resulted from the disease[1]
 - SCCHN is the most common histology of head and neck cancers, representing 90% of diagnoses[2]
 - Patients living with SCCHN experience poor quality of life, with impairment in basic human functions such as chewing, swallowing, tasting, and communicating[3]
 - Currently, there is a significant unmet need for effective treatment options in patients with r/m SCCHN especially following treatment with platinum and immunotherapy[4,5]
- TV is a TF-directed ADC composed of 1) a fully human monoclonal antibody specific for TF, 2) the microtubule-disrupting agent MMAE, and 3) a protease-cleavable linker that covalently links MMAE to the antibody
 - In tumor cells, TF has been shown to promote tumor growth, angiogenesis, and metastasis[6,7]
 - TF is highly expressed in numerous solid tumors, including SCCHN, where expression has been reported in 63%-100% of patients[8,9]

Proposed Mechanism of Action

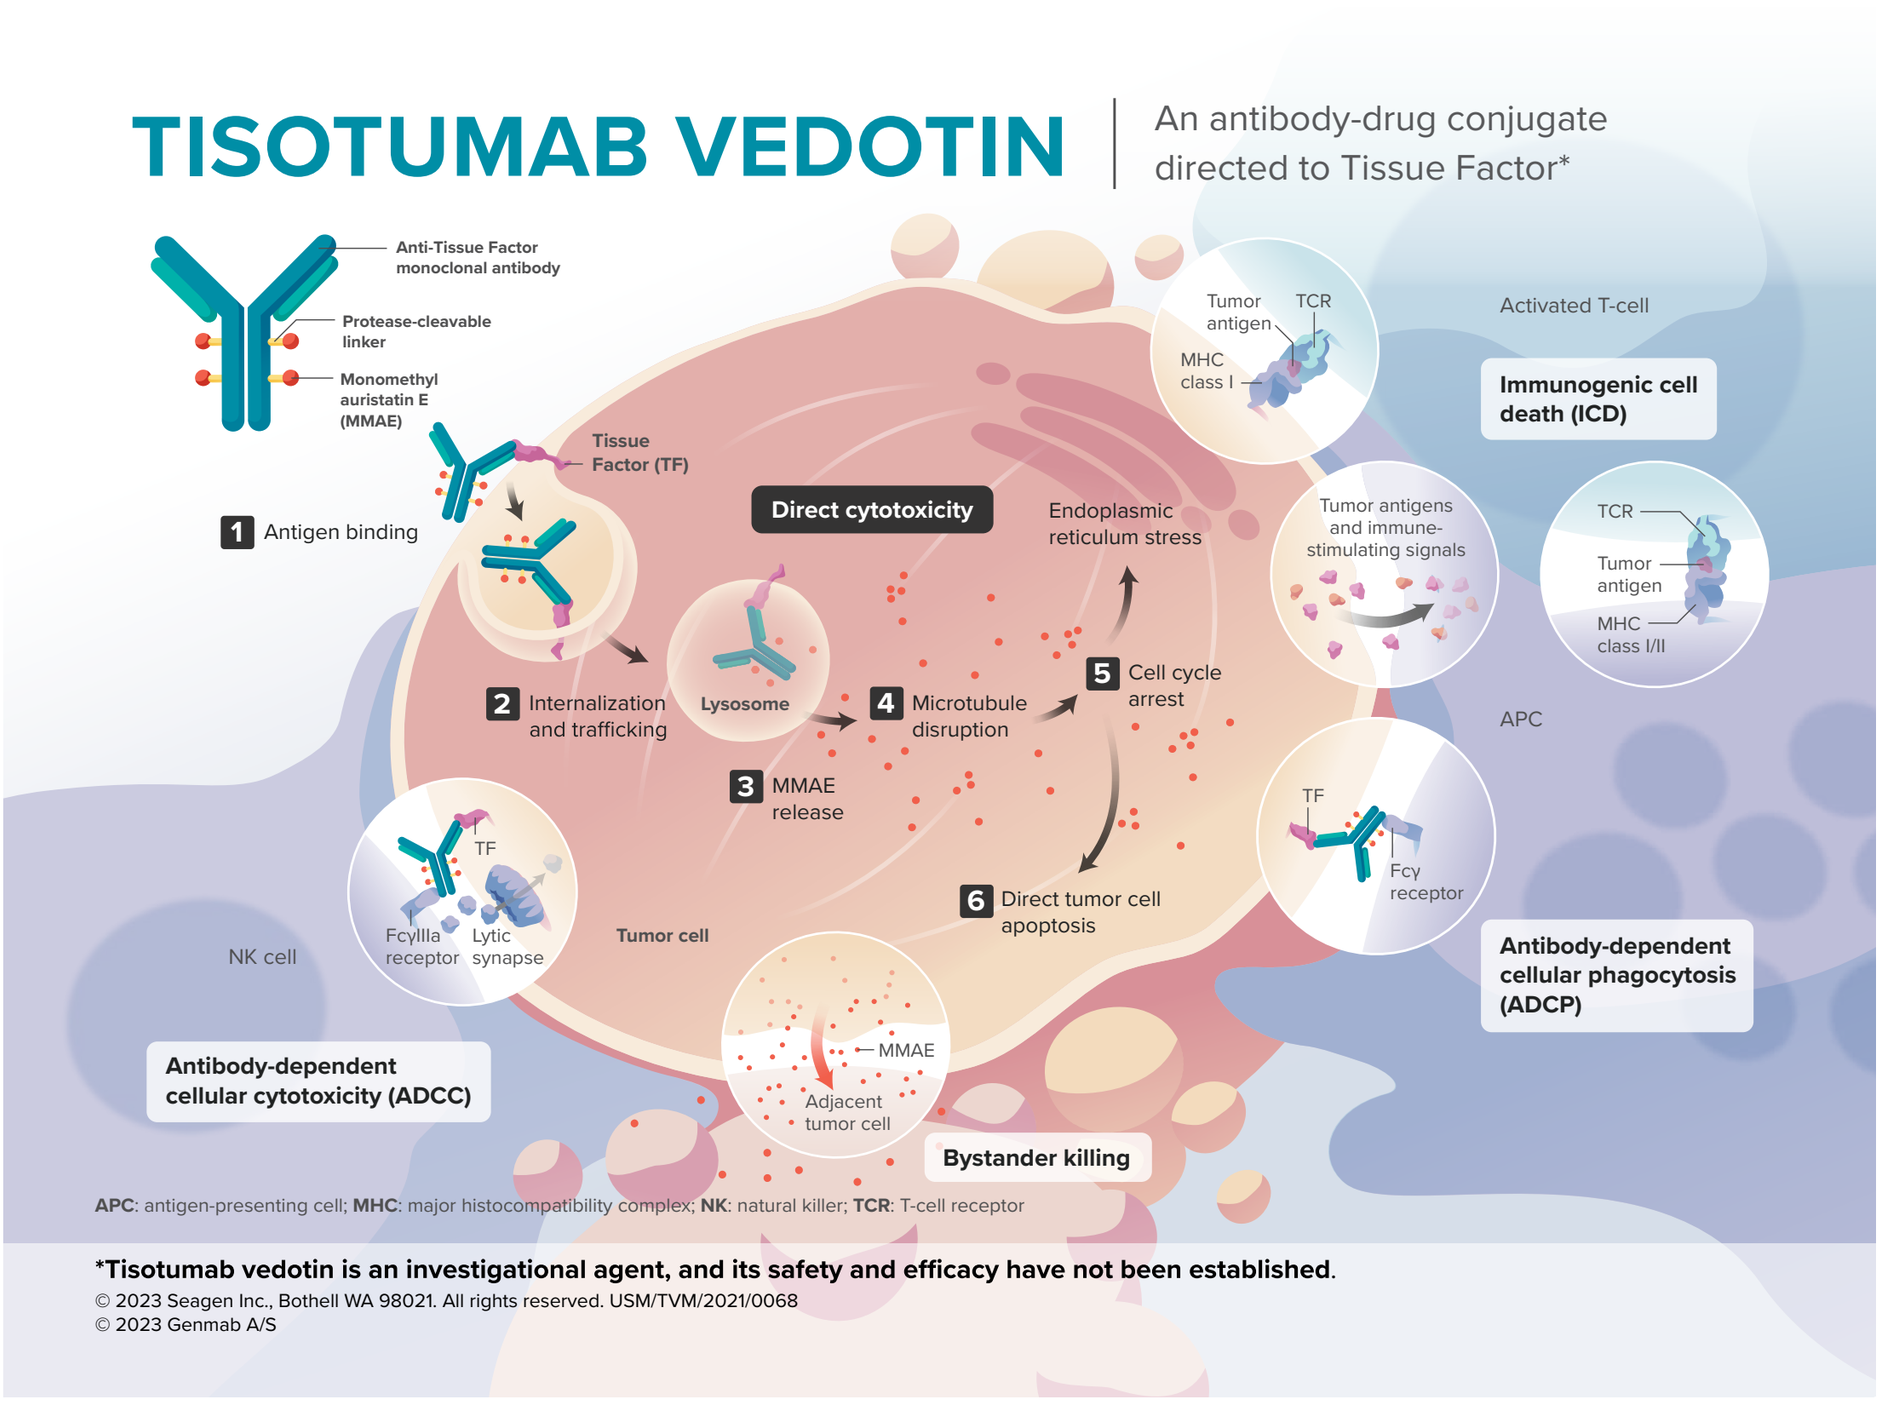
TISOTUMAB VEDOTIN
An antibody-drug conjugate directed to Tissue Factor*
Anti-Tissue Factor monoclonal antibody
Protease-cleavable linker
Monomethyl auristatin E (MMAE)
Tissue Factor (TF)
1 Antigen binding
2 Internalization and trafficking
Lysosome
3 MMAE release
4 Microtubule disruption
5 Cell cycle arrest
6 Direct tumor cell apoptosis
Direct cytotoxicity
Endoplasmic reticulum stress
Tumor antigens and immune-stimulating signals
Immunogenic cell death (ICD)
Activated T-cell
APC
Antibody-dependent cellular phagocytosis (ADCP)
Antibody-dependent cellular cytotoxicity (ADCC)
NK cell
Tumor cell
Bystander killing
Adjacent tumor cell
APC: antigen-presenting cell; MHC: major histocompatibility complex; NK: natural killer; TCR: T-cell receptor
*Tisotumab vedotin is an investigational agent, and its safety and efficacy have not been established.
© 2023 Seagen Inc., Bothell WA 98021. All rights reserved. USM/TVM/2021/0068
© 2023 Genmab A/S

- In the United States, TV received accelerated approval for adult patients with r/mCC with disease progression on or after chemotherapy at a dose of 2.0 mg/kg Q3W, based on results from innovaTV 204[10,11]
 - Additionally, TV administered at a dose of 2.0 mg/kg Q3W demonstrated encouraging preliminary evidence of antitumor activity and a tolerable safety profile in patients with r/m SCCHN that progressed after platinum combination with or without immunotherapy, with a confirmed ORR of 16% and DCR of 58%[12]
- Based on exposure-response data, the 1.7-mg/kg Q2W dosing regimen may achieve a higher dose intensity through increased AUC and higher exposure with elevated C_{trough} compared with the 2.0-mg/kg Q3W dosing regimen, potentially allowing for enhanced clinical activity[13]
- Here, we report the interim analysis from the ongoing cohort of TV 1.7 mg/kg Q2W for r/m SCCHN that has progressed on or after treatment with platinum based regimen and checkpoint inhibitor, if eligible

METHODS

- innovaTV 207 (NCT03485209) is an open-label, phase 2 multicenter study evaluating TV monotherapy or in combination for advanced solid tumors, including in patients with r/m SCCHN

innovaTV 207 Part C Study Design

Key Eligibility Criteria
- Relapsed, locally advanced or metastatic SCCHN or sqNSCLC[a]
- Prior therapy with platinum based regimen and/or CPI (if eligible)
- No more than 3 prior systemic lines of therapy[b]
- ECOG PS: 0-1

→

Safety Run-In
Tisotumab Vedotin
1.7 mg/kg
Days 1 and 15
of a 28-day cycle (Q2W)
(N=6)
SCCHN or sqNSCLC

→

Dose Expansion
Tisotumab Vedotin
1.7 mg/kg
Days 1 and 15
of a 28-day cycle (Q2W)
(N=30)
SCCHN or sqNSCLC

→

Endpoints
Primary Endpoint: Confirmed ORR by investigator, RECIST v1.1[c]
Key Secondary Endpoints:
- Safety
- Tolerability
- DCR
- PFS
- ORR
- OS

a Data for sqNSCLC are not presented herein.
b For patients with sqNSCLC, patients eligible for a tyrosine kinase inhibitor should have received no more than 4 lines of systemic therapy.
c Tumor response assessed every 6 weeks.

Abbreviations

ADC, antibody-drug conjugate; AE, adverse event; AESI, adverse event of special interest; AUC, area under the plasma concentration-time curve; CPI, checkpoint inhibitor; CR, complete response; C_{trough}, trough concentration; DCR, disease control rate; ECOG PS, Eastern Cooperative Oncology Group performance status; IHC, immunohistochemistry; MMAE, monomethyl auristatin E; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; PSN, peripheral sensory neuropathy; Q2W, Days 1 and 15 of a 28-day cycle; Q3W, Day 1 of a 21-week cycle; r/m, recurrent or metastatic; r/mCC, recurrent or metastatic cervical cancer; RECIST, Response Evaluation Criteria in Solid Tumours; SAE, serious adverse event; SCCHN, squamous cell carcinoma of head and neck; SD, stable disease; SOD, sum of diameters; sqNSCLC, squamous non-small cell lung cancer; TF, tissue factor; TNM, tumor node metastasis; TRAE, treatment-related adverse event; TV, tisotumab vedotin

Copies of this poster obtained through QR, AR, and/or text key codes are for personal use only and may not be reproduced without written permission of the author,
Beatriz Cirauqui, bcirauqui@iconcologia.net

Disclosures

BC has received funding from Bristol Myers Squibb and Merck, Sharp & Dohme, and other support from Bristol Myers Squibb, Merck, Sharp & Dohme, Merck & Co, GEICAM, SEOM, SOLTI, and TTCC Group. WWJ has received other support from Bristol Myers Squibb, Eli Lilly and Company, Merck & Co, Roche/Genentech, Boehringer Ingelheim, AstraZeneca, Pfizer, Bayer, Sanofi, Takeda Pharmaceutical Company, Novartis, Libbs, and Janssen Pharmaceuticals. SS and AB have nothing to disclose. KS, XG, and LN are employees of and have equity ownership in Seagen Inc. IS is an employee of and has equity ownership in Genmab. FC has received other support from Roche, Merck KGaA, Bayer, Servier Laboratories, Merck, Sharp & Dohme, Amgen Inc, and Laboratories Pierre Fabre.

RESULTS

- At data cutoff on November 28, 2022, 15 patients with SCCHN are included in the analyses
 - The median number of prior lines for r/m disease is 2 (range 1-3)
 - All patients have received prior platinum therapy and most patients have received prior CPI, cetuximab, or taxanes in the r/m setting

Patient and Disease Characteristics

Characteristic	Part C SCCHN (N=15)
Age, years, median (range)	61 (40-69)
Sex, male, n (%)	12 (80.0)
Baseline ECOG PS, n (%)	
0	7 (46.7)
1	8 (53.3)
TNM stage at initial diagnosis, n (%)	
II	1 (6.7)
III	4 (26.7)
IV	9 (60.0)
IVA	4 (26.7)
IVB	5 (33.3)
Unknown	1 (6.7)
Diagnosis subtype	
Larynx	6 (40.0)
Oral cavity	4 (26.7)
Oropharynx	2 (13.3)
Hypopharynx	1 (6.7)
Sinus	1 (6.7)
Nasopharynx	1 (6.7)
Lines of prior systemic therapies in r/m setting, median (range)	2 (1-3)
Types of prior therapies, n (%)	
Systemic	15 (100)
Radiation	14 (93.3)
Surgery	10 (66.7)
Prior systemic therapy in all setting, n (%)	15 (100)
Carboplatin/cisplatin	15 (100)
Prior systemic therapy in r/m setting, n (%)	15 (100)
CPI[a]	14 (93.3)
Cetuximab	10 (66.7)
Taxane (nab-paclitaxel/docetaxel/paclitaxel)	8 (53.3)
Number of treated cycles, median (range)[b]	3 (2-7)
TF membrane expression, n/N (%)[c]	8/10 (80)

a CPI included pembrolizumab, ipilimumab, nivolumab, atezolizumab, durvalumab, and avelumab.
b Each cycle was 28 days with Q2W dosing regimen (Days 1 and 15 of a 28-day cycle).
c Of 15 patients, 10 patients had evaluable IHC for detection of TF membrane expression, which was defined as ≥1% expression.

Acknowledgements

- To the patients and their families for their participation in innovaTV 207
- This study was funded by Genmab (Copenhagen, Denmark), Seagen Inc. (Bothell, WA, USA), the Gynecologic Oncology Group (GOG), and the European Network of Gynaecological Oncological Trial Groups (ENGOT). Tisotumab vedotin is being co-developed by Genmab and Seagen Inc.
- Jennifer Yang, PhD, of Seagen Inc. provided medical writing and editorial support with funding from Seagen Inc., in accordance with Good Publication Practice guidelines.

References

1. Sung, et al. CA Cancer J Clin. 2021;71(3):209-49.
2. Klein and Grandis. Cancer Biol Ther. 2010;9(1):1-7.
3. McDowell, et al. Front Oncol. 2022;12:834068.
4. Borel, et al. Cancers (Basel). 2020;12(9):2691.
5. La, et al. Eur J Cancer Care (Engl). 2018;27(5):e12862.
6. Van den Berg, et al. Blood. 2012;119(4):924-32.
7. Förster, et al. Clin Chim Acta. 2006;364(1-2):12-21.
8. Kaido, et al. Hepatogastroenterology. 2005;52(65):1383-7.
9. Poon, et al. Clin Cancer Res. 2003;9(14):5339-45.
10. TIVDAK [Prescribing Information]. Bothell, WA: Seagen Inc. January 2022.
11. Coleman, et al. Lancet Oncol. 2021;22(5):609-19.
12. Hong, et al. Int J Radiat Oncol Biol Phys. 2022;112(5):e10-e11.
13. Voellinger, et al. ESMO TAT 2023. Poster 5P.

Antitumor Activity in Patients with SCCHN

- Confirmed ORR is 40% (95% CI, 16.3-67.7), with 1 CR and 5 PRs
- DCR (proportion of patients with confirmed CR or PR, or SD) is 60% (95% CI, 32.3-83.7)
- Median PFS, as evaluated per investigator, is 4.4 months (95% CI, 1.4-6.8)

Maximum Change in Target Lesion Size

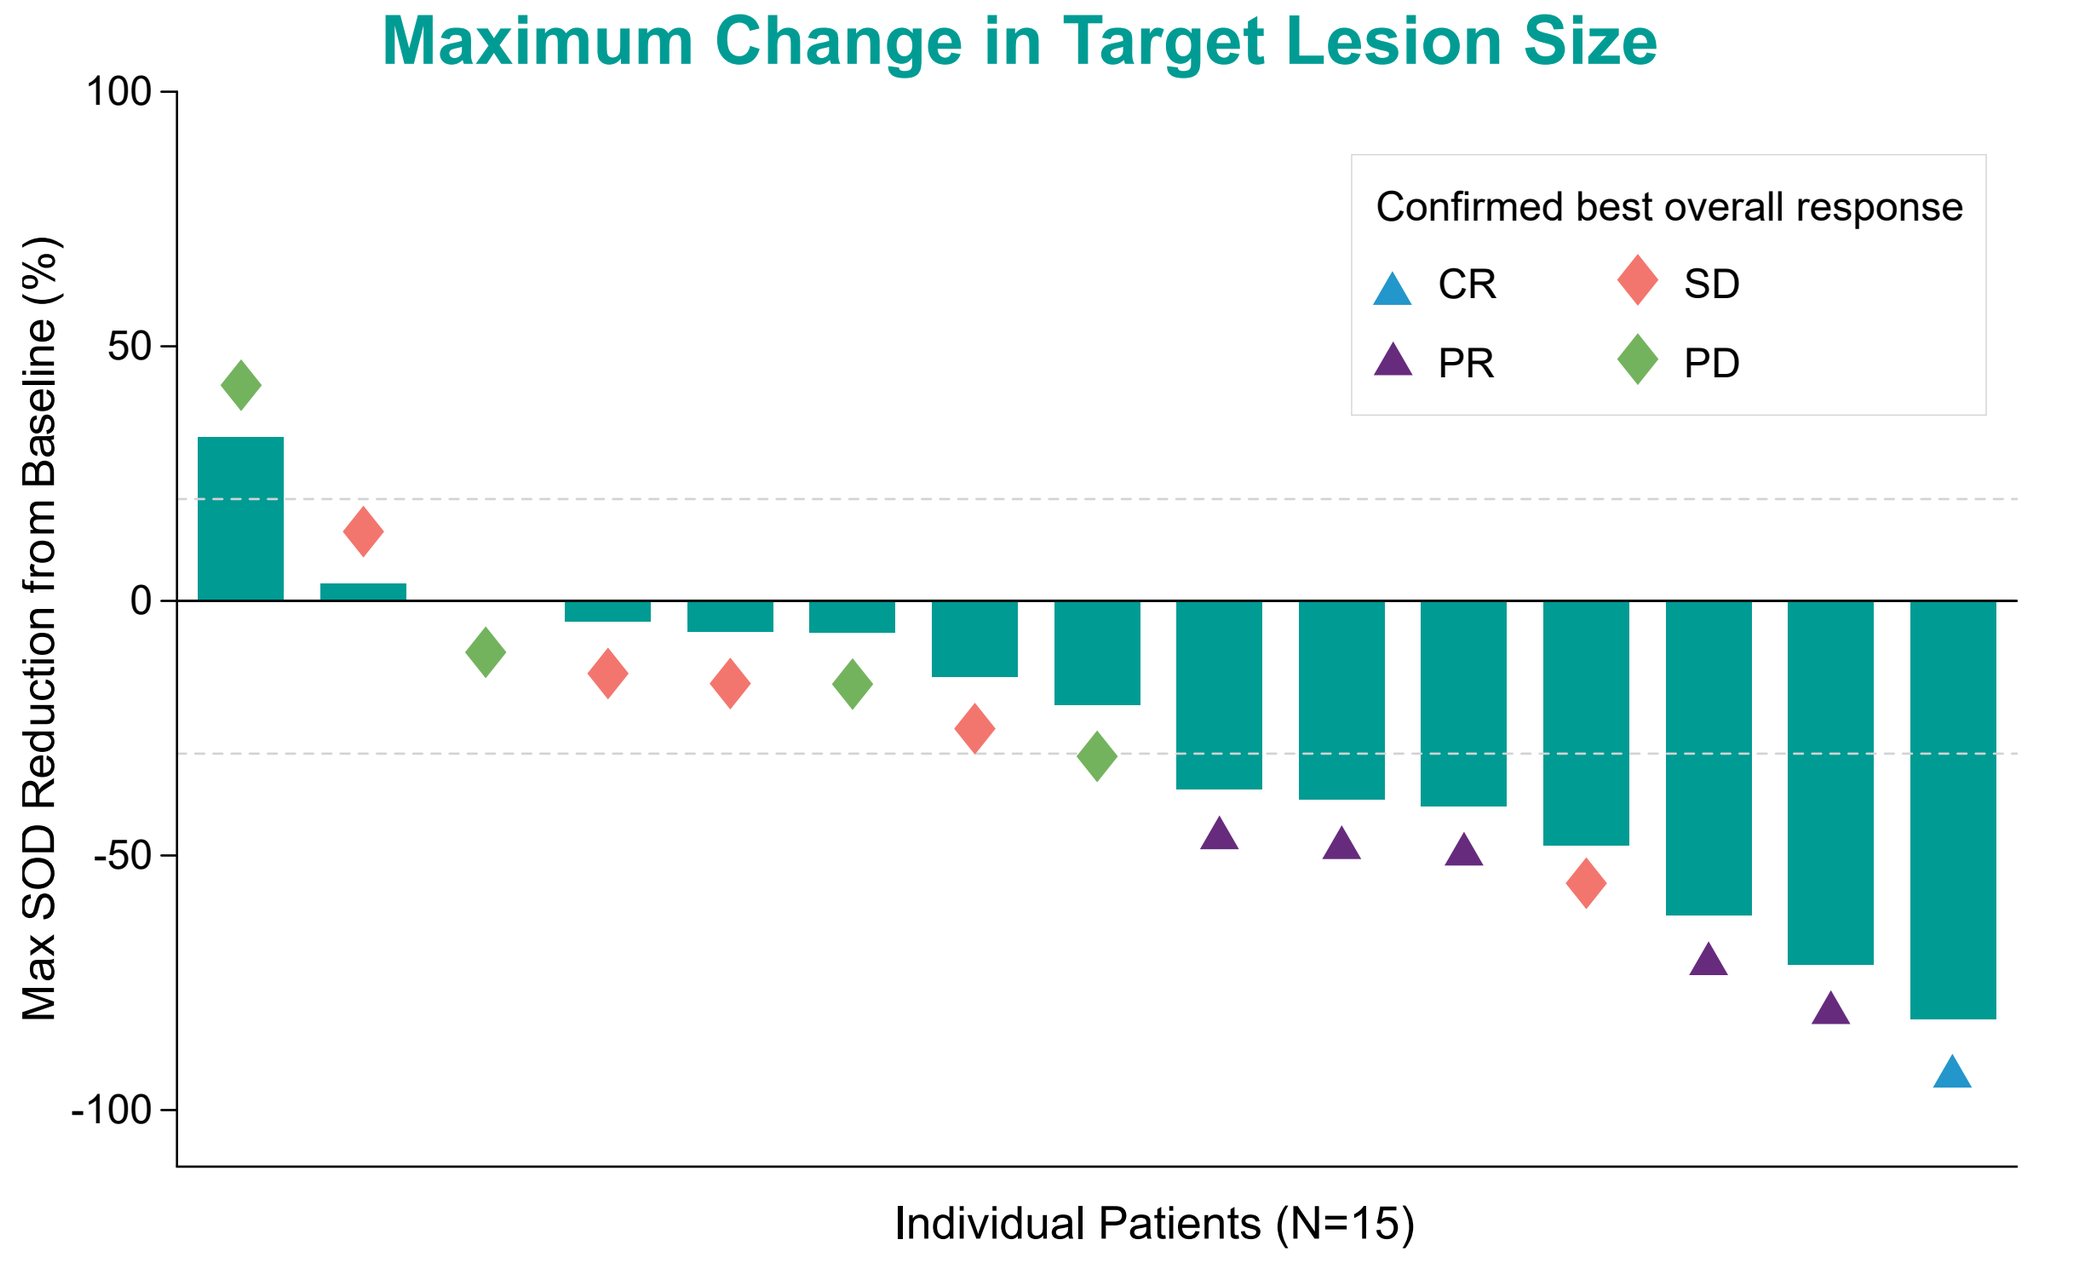
100
50
0
-50
-100
Max SOD Reduction from Baseline (%)
Confirmed best overall response
CR
SD
PR
PD
Individual Patients (N=15)

Percentage Change in Target Lesion Size

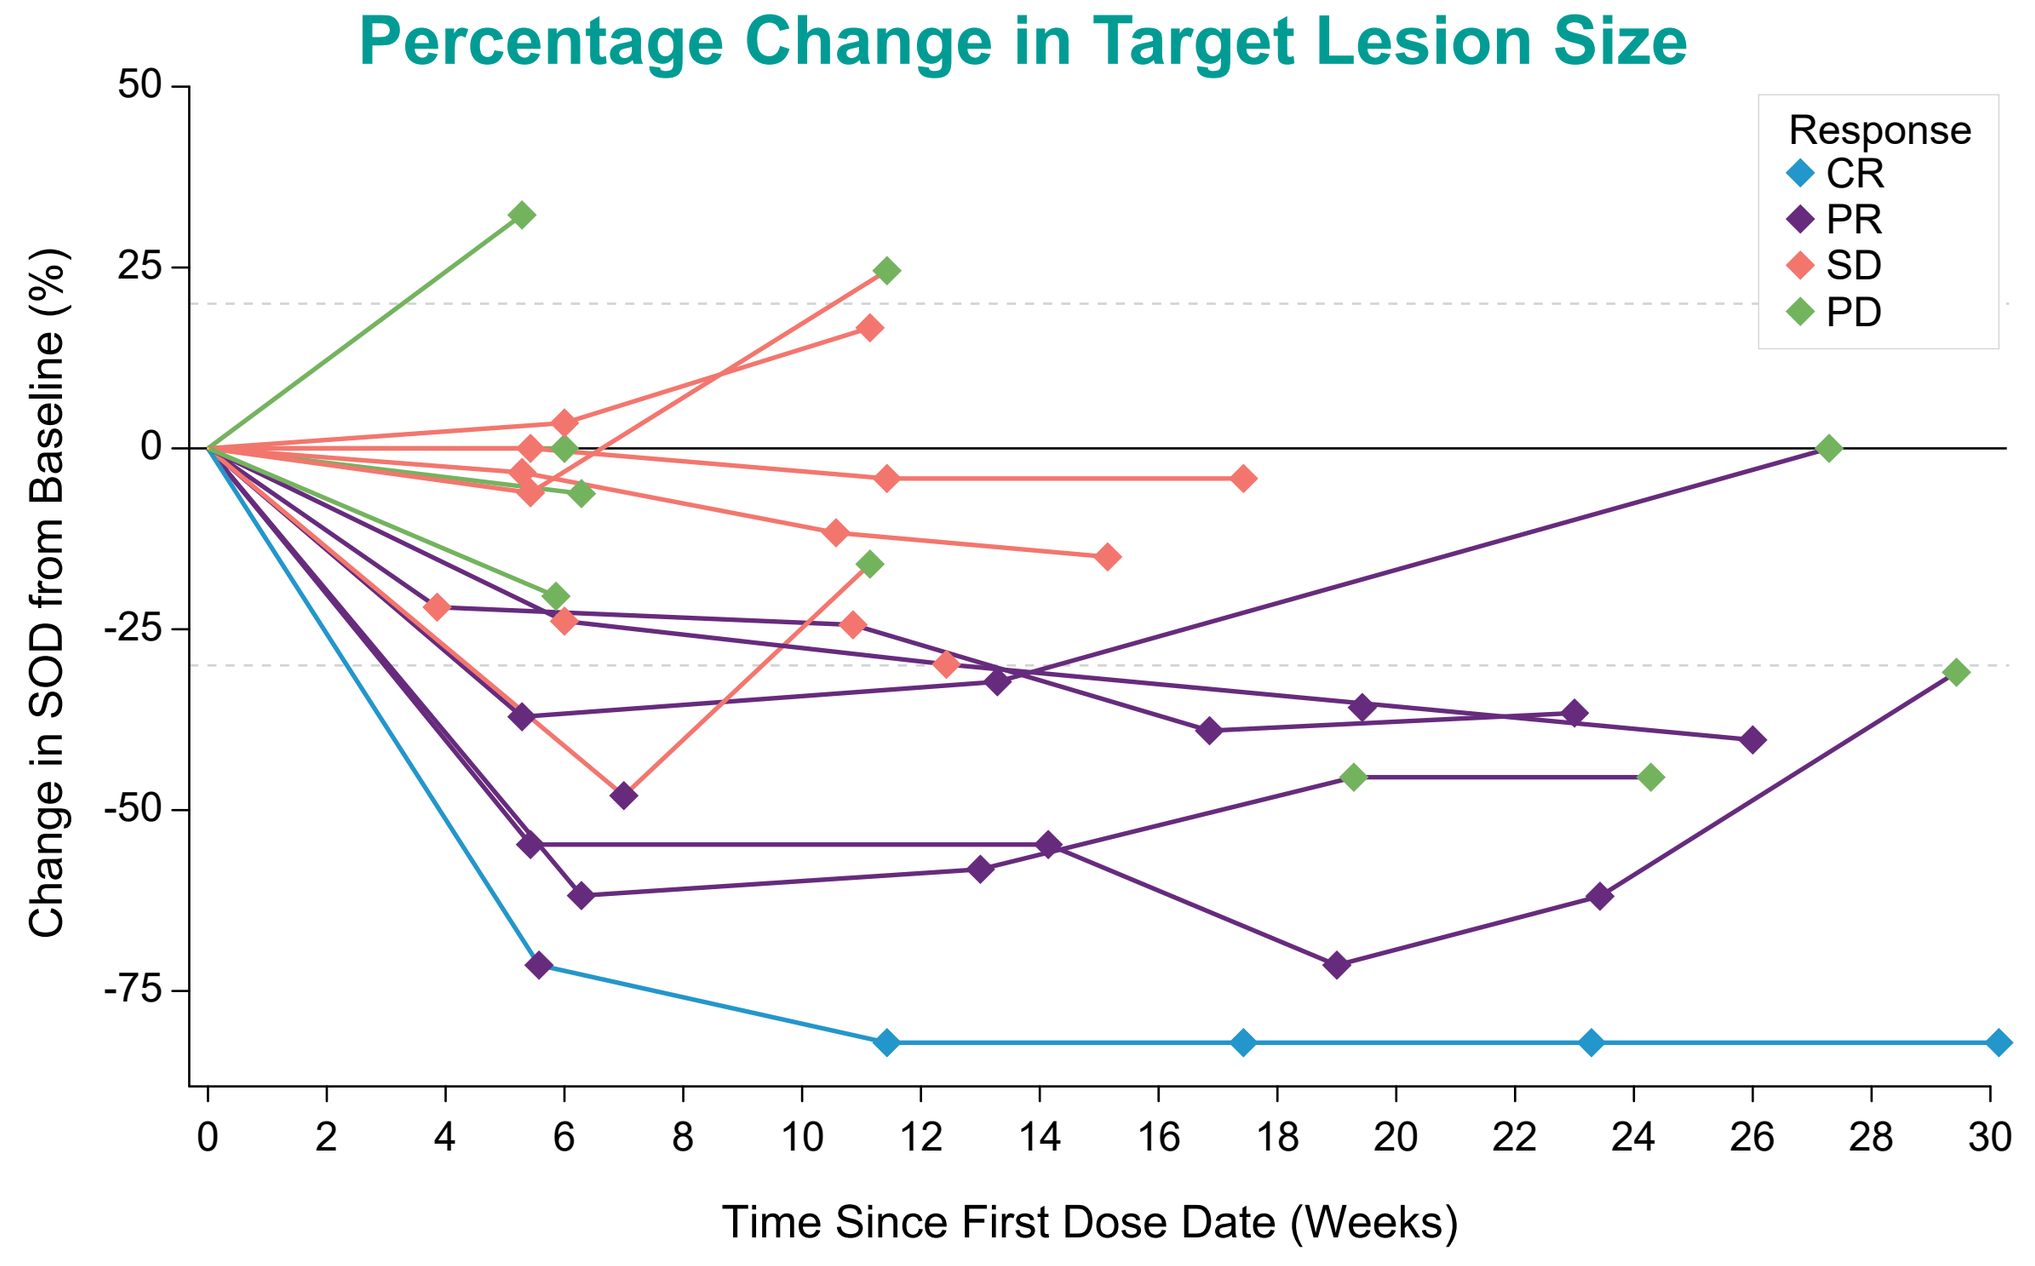
50
25
0
-25
-50
-75
Change in SOD from Baseline (%)
Response
CR
PR
SD
PD
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Time Since First Dose Date (Weeks)

Safety Overview in Patients with SCCHN

- 11 patients have received 3 or more cycles of treatment
- 11 patients have received a dose modification due to any reason
- 13 patients have experienced any grade TRAEs, and 4 patients have experienced grade 3 TRAEs
 - The most common TRAEs occurring in >20% of patients are asthenia (n=7), PSN (n=7), vomiting (n=5), and conjunctivitis (n=4)
 - 2 patients have discontinued treatment because of a TRAE (dry eye [n=1] and PSN [n=1])
- Treatment-related SAEs have occurred in 2 patients, both of which were peripheral sensorimotor neuropathy

Event, n (%)	Part C SCCHN (N=15)
TRAEs[a]	
Any grade	13 (86.7)
Grade 3[b]	4 (26.7)
Any grade TRAEs occurring in ≥20% of patients	
Asthenia	7 (46.7)
PSN	7 (46.7)
Vomiting	5 (33.3)
Conjunctivitis	4 (26.7)
Alopecia	3 (20.0)
Diarrhea	3 (20.0)
Myalgia	3 (20.0)
Pruritus	3 (20.0)

Event, n (%)	Part C SCCHN (N=15)
Any grade AESIs occurring in ≥10% of patients[a]	
Ocular event	8 (53.3)
Conjunctivitis	4 (26.7)
Lacrimation increased	3 (20.0)
Dry eye	2 (13.3)
Conjunctival hyperaemia	2 (13.3)
Vision blurred	2 (13.3)
Peripheral neuropathy event	9 (60.0)
Peripheral sensory neuropathy	7 (46.7)
Peripheral sensorimotor neuropathy	2 (13.3)
Bleeding event	5 (33.3)
Epistaxis	2 (13.3)

a Defined as newly occurring or worsening AE after the first dose and with AE start date on or before 30 days after the last dose of the study drug.
b 4 patients experienced 7 cases of grade 3 TRAEs, including: dry eye, keratitis, fatigue, neutropenia, PSN, and peripheral sensorimotor neuropathy.

CONCLUSIONS

- Interim results from Part C of the innovaTV 207 study suggest that TV has encouraging antitumor activity at a 1.7-mg/kg Q2W dosing regimen, with a confirmed ORR of 40% and median PFS of 4.4 months in patients with previously treated r/m SCCHN
 - Additionally, TV at a 1.7-mg/kg Q2W dosing regimen had an acceptable safety profile
- innovaTV 207 is an ongoing study; Part C continues to enroll patients in SCCHN and sqNSCLC to evaluate the clinical benefit of TV at a 1.7-mg/kg Q2W dosing regimen in eligible patients