BRENTUXIMAB VEDOTIN IN COMBINATION WITH NIVOLUMAB, DOXORUBICIN, AND DACARBAZINE IN NEWLY DIAGNOSED PATIENTS WITH EARLY-STAGE HODGKIN LYMPHOMA (SGN35-027, TRIAL IN PROGRESS)

Ian Flinn, MD[1], Jeremy S. Abramson, MD[2], Linda Ho, MD[3], Hun Ju Lee, MD[4]

[1]Sarah Cannon Research Institute and Tennessee Oncology, Nashville, TN, USA; [2]Massachusetts General Hospital, Boston, MA, USA; [3]Seagen Inc., Bothell, WA, USA; [4]MD Anderson Cancer Center, Houston, TX, USA

Background and Rationale

- Brentuximab vedotin (BV) was the first antibody–drug conjugate to be approved in multiple cancer types, including treatment-naïve Stage 3 or 4 classical Hodgkin lymphoma (cHL) in combination with doxorubicin, vinblastine, and dacarbazine (AVD).[1,2]
- The combination of a CD30-directed monoclonal antibody, a protease-cleavable linker, and the microtubule-disrupting agent monomethyl auristatin E drives the anticancer activity (**Figure 1**).[3]
 - MMAE-mediated microtubule disruption induces cell cycle arrest and apoptosis.[3]
 - Direct cytotoxicity is at the heart of the multifaceted anticancer activity of BV, including the induction of immunogenic cell death, which promotes activation and recruitment of immune cells to tumors.[3–9]
- BV combined with doxorubicin and dacarbazine (BV+AD), omitting vinblastine, demonstrated notable and durable activity with low toxicity in 34 patients with non-bulky Stage 1 or 2 cHL, suggesting that vinblastine may not be required for efficacy.[10]
 - BV+AD resulted in interim and end of treatment (EOT) complete response (CR) rates of 94% and 97%, respectively.[10]
 - The 4-year progression-free survival (PFS) and overall survival (OS) estimates were 91% and 100%, respectively.[10]
 - Peripheral sensory neuropathy (PSN) was low grade and only one patient had persistent PSN at the last follow-up.[10]
- Nivolumab is a fully humanized monoclonal antibody that targets programmed cell death protein 1 (PD-1) and is approved as monotherapy for relapsed/refractory (R/R) cHL.[11]
- Nivolumab combined with multi-agent chemotherapy (N+AVD) was well tolerated and had promising activity in newly diagnosed advanced-stage cHL.[11]
- BV combined with nivolumab was well-tolerated and produced an 82% overall response rate (ORR) rate in 11 evaluable patients with newly diagnosed cHL who were ineligible for, or declined, conventional chemotherapy.[12]
- BV combined with nivolumab produced a 67% CR rate and an estimated 3-year PFS rate of 77% in 91 adults with R/R cHL in the first-line salvage setting.[13]
- The combination of BV and nivolumab with doxorubicin and dacarbazine (AN+AD) should hypothetically result in high response rates and be well tolerated, with potentially less toxicity than vinblastine-containing regimens.

Figure 1: Proposed Mechanism of Action

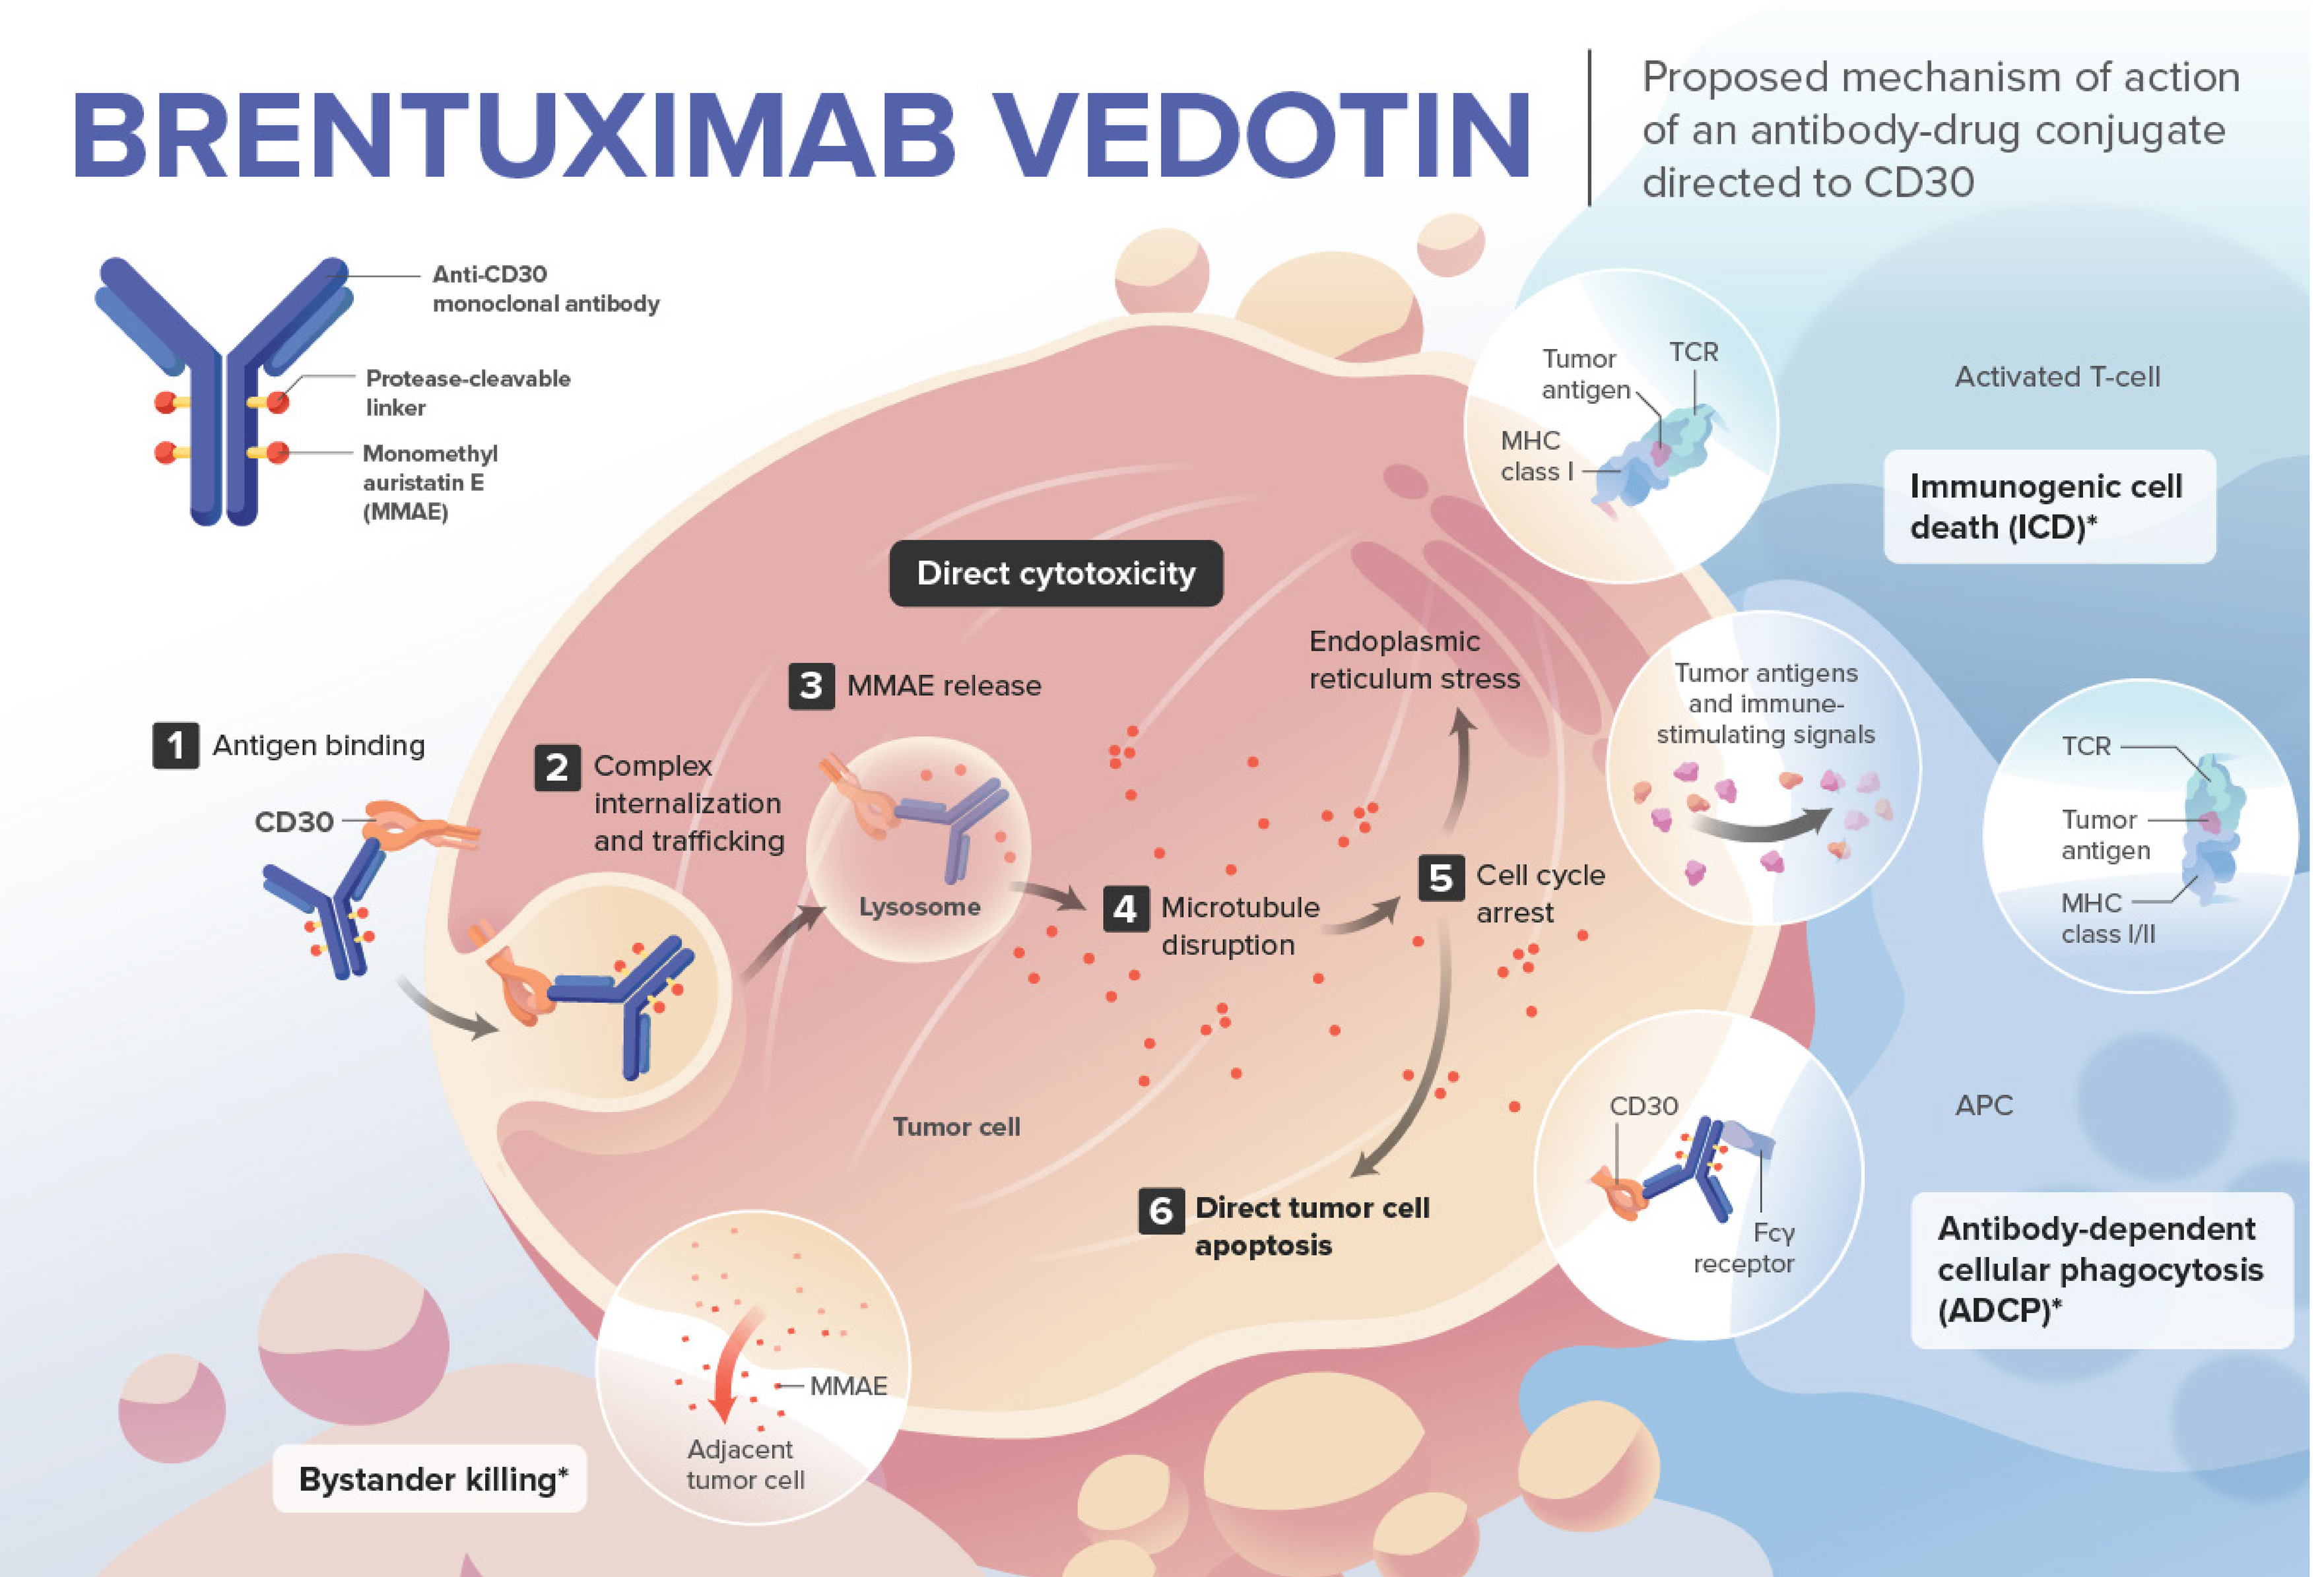
BRENTUXIMAB VEDOTIN
Proposed mechanism of action of an antibody-drug conjugate directed to CD30
Anti-CD30 monoclonal antibody
Protease-cleavable linker
Monomethyl auristatin E (MMAE)
Direct cytotoxicity
1 Antigen binding
CD30
2 Complex internalization and trafficking
3 MMAE release
Lysosome
4 Microtubule disruption
Endoplasmic reticulum stress
5 Cell cycle arrest
6 Direct tumor cell apoptosis
Tumor cell
Tumor antigen
TCR
MHC class I
Activated T-cell
Immunogenic cell death (ICD)*
Tumor antigens and immune-stimulating signals
TCR
Tumor antigen
MHC class I/II
CD30
Fcγ receptor
APC
Antibody-dependent cellular phagocytosis (ADCP)*
MMAE
Adjacent tumor cell
Bystander killing*

APC: antigen-presenting cell; **CD30:** cluster of differentiation 30; **MHC:** major histocompatibility complex; **TCR:** T-cell receptor

*Additional mechanisms of action and their potential to complement the direct cytotoxicity of some MMAE-based antibody-drug conjugates are currently under investigation

Brentuximab vedotin is an investigational agent in some settings, and its safety and efficacy have not been established.

© 2021 Seagen Inc., Bothell WA 98021. All rights reserved.

Study Design

- SGN35-027 (NCT03646123; EudraCT 2020-004027-17) is an open-label, multiple part, multicenter, phase 2 trial of BV in patients with cHL.
 - Part A will evaluate A+AVD when administered with growth factor prophylaxis in Stage 3 or 4 cHL.
 - Part B will evaluate AN+AD in patients with Stage 1 or 2 cHL with bulky mediastinal disease or Stage 3 or 4 cHL.
 - Part C will evaluate AN+AD in patients with Stage 1 or 2 cHL without bulky disease.
- Approximately 240 patients will be enrolled in this study: 40 in Part A, 50 in Part B, and 150 in Part C.
 - Parts A and B have completed enrollment.
- In Part C, patients will be administered AN+AD consisting of BV 1.2 mg/kg (A), nivolumab 240 mg (N), doxorubicin 25 mg/m² (+A), and dacarbazine 375 mg/m² (D). Each will be administered separately by intravenous infusion on Days 1 and 15 of each 28-day cycle for up to 4 cycles (**Figure 2**).

Figure 2: SGN35-027 Part C Study Design

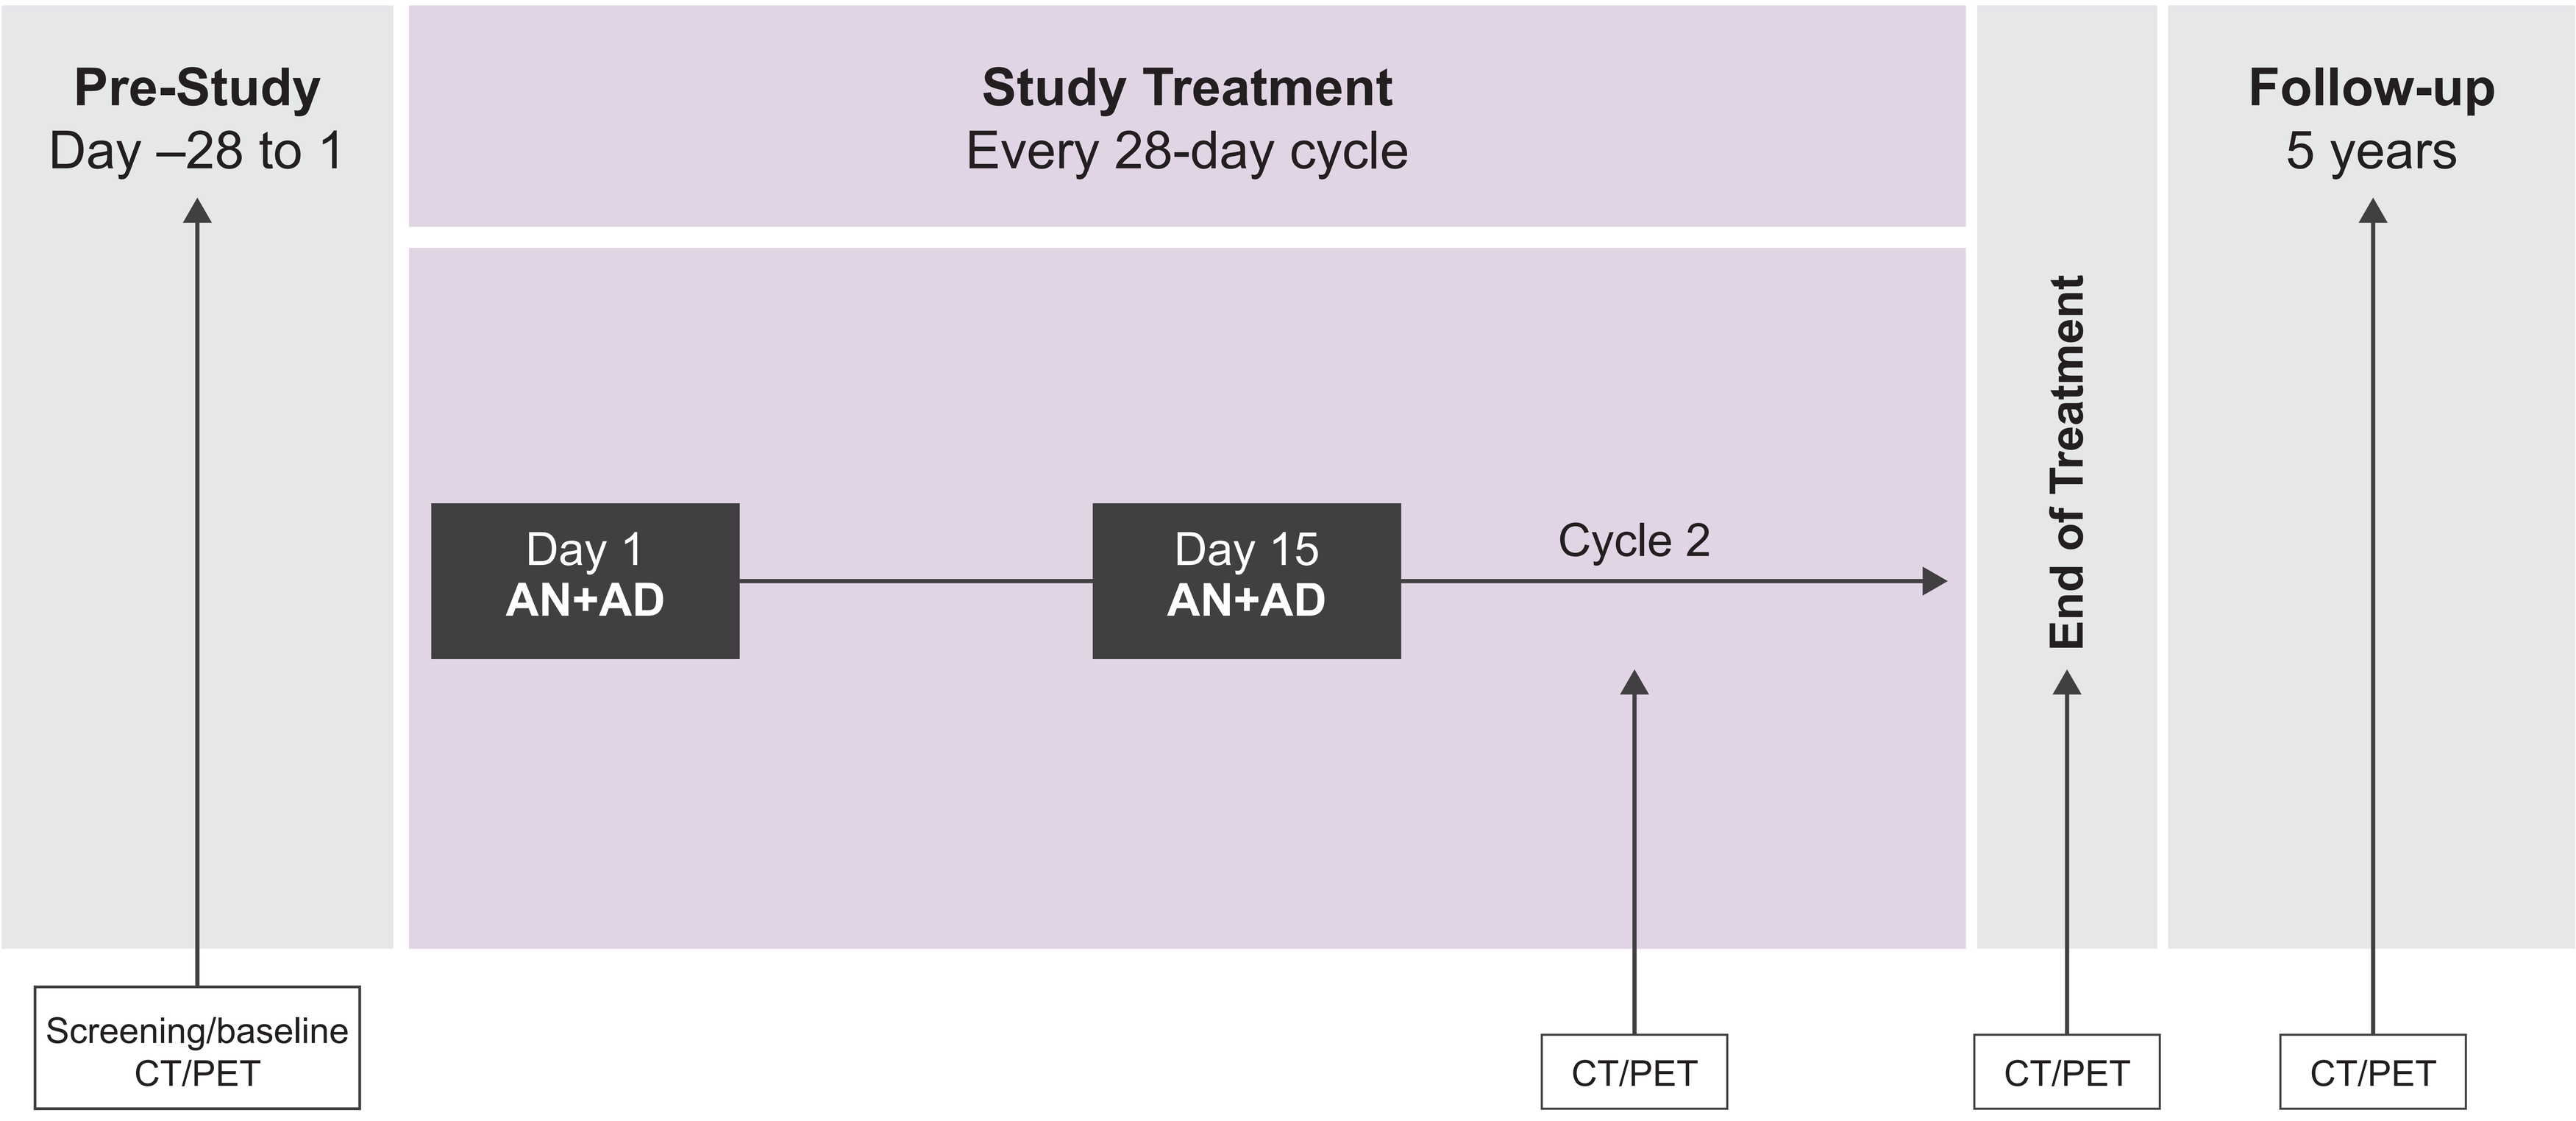
Pre-Study
Day –28 to 1
Study Treatment
Every 28-day cycle
Follow-up
5 years
Day 1
AN+AD
Day 15
AN+AD
Cycle 2
End of Treatment
Screening/baseline CT/PET
CT/PET
CT/PET
CT/PET

AN+AD, BV and nivolumab with doxorubicin and dacarbazine; CT, computed tomography; PET, positron emission tomography.

Eligibility Criteria

Table 1: Part C Key Inclusion Criteria

- Treatment-naïve, Ann Arbor Stage 1 or 2 cHL without bulky disease
- Histologically confirmed cHL according to the current World Health Organization Classification
- Bidimensional measurable disease as documented by PET/CT or CT imaging
 - Must have at least 1 lesion >1.5 cm in the longest diameter on cross-sectional imaging, measurable in 2 perpendicular dimensions on CT (or magnetic resonance imaging), and fluorodeoxyglucose avid by PET
- Eastern Cooperative Oncology Group performance status ≤2
- Age ≥12 years in the United States or ≥18 years in the rest of the world

cHL, classical Hodgkin lymphoma; CT, computed tomography; PET, positron emission tomography.

Table 2: Part C Key Exclusion Criteria

- Nodular lymphocyte predominant Hodgkin lymphoma
- History of another malignancy within 3 years of the first dose of study drug or any evidence of residual disease from a previously diagnosed malignancy
 - Exceptions are malignancies with a negligible risk of metastasis or death
- Active cerebral or meningeal disease related to the underlying malignancy
- Prior immunosuppressive chemotherapy, therapeutic radiation, or any immunotherapy within 4 weeks of the first study drug dose
- Prior treatment with an anti–PD-1, anti–PD-L1, anti–PD-L2, or anti–CTLA-4 antibody, or any other antibody or drug specifically targeting T-cell co-stimulation or checkpoint pathways
- Current therapy with other systemic anti-neoplastic or investigational agents

CTLA-4, cytotoxic T-lymphocyte-associated antigen 4; PD-1, programmed cell death protein 1; PD-L1/2, programmed death-ligand 1/2.

Efficacy Assessments

- Efficacy will be assessed by CT of the neck, chest, abdomen, and pelvis at baseline, Cycle 2 (Day 25–28) and EOT (within 30–37 days of last dose).
- PET will also be performed if CT is positive at prior timepoint in PET-negative disease.
- Disease response and progression will be assessed by investigators using the Lymphoma Response to Immunomodulatory Therapy Criteria (LYRIC) modification of the Lugano Classification Revised Staging System for nodal non-Hodgkin and Hodgkin lymphomas.[14]

Safety Assessments

- Safety assessments will consist of the surveillance and recording of adverse events (AEs) and concomitant medications, physical examination findings, and laboratory tests.
- Safety will be assessed by a safety monitoring committee (SMC) after at least 10 patients have completed treatment, with an additional SMC meeting occurring after approximately 50 have completed their EOT assessment. The SMC will also monitor the safety of patients through regular and/or ad hoc meetings that include review of AEs and laboratory abnormalities.
- The final safety visit will occur 100 days ±2 weeks after last dose of nivolumab or 30 days after last dose of BV, whichever is later.

Study Endpoints

Table 3: Study Endpoints

Primary Endpoint
- CR rate at EOT

Secondary Endpoints
- ORR
- Duration of response
- Duration of CR
- Event-free survival
- PFS
- OS
- Safety and tolerability

CR, complete response; EOT, end of treatment; ORR, overall response rate; OS, overall survival; PFS, progression-free survival.

Statistical Analyses

- Safety and efficacy endpoints will be summarized with descriptive statistics.
- The CR rate at EOT and its 2-sided 95% confidence intervals will be presented.
- Time-to-event endpoints will be analyzed using Kaplan–Meier methodology.

Summary

- AN+AD is a promising treatment combination for patients with early-stage HL.
- Part C of the SGN35-027 trial will evaluate whether AN+AD results in improved response rates and tolerability in treatment-naïve patients with early-stage cHL compared to vinblastine-containing regimens.
- Parts A and B have completed enrollment.
- Part C enrollment is open and ongoing at 54 study sites in Australia, Spain, and the US.

References

1. Gauzy-Lazo L, et al. (2020). SLAS Discov 25(8):843-68.
2. Connors JM, et al. (2018). N Engl J Med 378(4):331-44.
3. Sutherland MS, et al. (2006). J Biol Chem 281(15):10540-7.
4. Li F, et al. (2016). Cancer Res 76(9):2710-9.
5. Gardai SJ, et al. (2016). Haematologica 101(S5):53.
6. Müller P, et al. (2014). Cancer Immunol Res 2(8):741-55.
7. Oflazoglu E, et al. (2007). Blood 110(13):4370-2.
8. Heiser RA, et al. Presented at: AACR Annual Meeting; April 14-18, 2018; Chicago, IL, USA; Abstract 1789.
9. Herrera AF, et al. (2018). Blood 131(11):1183-94.
10. Abramson JS, et al. (2021). Hematol Oncol 39(S2):Abstract 198.
11. Ramchandren R, et al. (2019). J Clin Oncol 37(23):1997-2007.
12. Friedberg JW, et al. Presented at: ISHL - 11th International Symposium on Hodgkin Lymphoma Annual Meeting; October 27-29, 2018; Cologne, Germany; Abstract T027 (0153).
13. Advani RH, et al. (2021). Blood 138(6):427-38.
14. Cheson BD, et al. (2016). Blood 128(21):2489-96.

Disclosures: Study funded by Seagen Inc. Ian Flinn reports consultancy agreements with AbbVie, AstraZeneca, BeiGene, Genentech, Gilead, Great Point, Iksuda, Janssen, Juno, Kite, MorphoSys, Novartis, Nurix, Pharmacyclics, Roche, Seagen, Servier, Takeda, TG Therapeutics, Unum, Verastem, and Yingli; and research funding from AbbVie, Acerta, Agios, ArQule, AstraZeneca, BeiGene, Calithera, Celgene, Century Therapeutics, Constellation, Curis, Forma, Forty Seven, Genentech, Gilead, Great Point, Hutchisom MediPharma, IBM Bio, Iksuda, Incyte, Infinity, Janssen, Juno, Karyopharm, Kite, Loxo, Merck, MorphoSys, Novartis, Nurix, Pfizer, Pharmacyclics, Portola, Rhizen, Roche, Seagen, Servier, Takeda, Teva, TG Thera, Trillium, Triphase, Unum, Verastem, Vincerx, and Yingli. Jeremy Abramson reports consultancy agreements with Allogene, AstraZeneca, BeiGene, Bluebird Bio, Bristol-Myers Squibb, C4 Therapeutics, Celgene, Century Therapeutics, Epizyme, Genentech, Genmab, Incyte, Karyopharm, Kite Pharma, Kymera, MorphoSys, Mustang Bio, Ono Pharma, and Regeneron; and research funding from Bristol-Myers Squibb and Seagen Inc. Linda Ho is an employee of and reports equity ownership in Seagen Inc. Hun Ju Lee has no relationships to disclose.

Acknowledgments: The authors wish to thank the patients and their families and the co-investigators and study teams at the various sites for their participation in this study. Medical writing support was provided by Suparna Abraham, PharmD, and editorial support by Travis Taylor, BA, all of Scion, London, supported by Seagen Inc.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from the author of this poster, **Ian Flinn, iflinn@tnonc.com**

Abstract No. 1369
Presented at The 63rd Annual Meeting of the American Society of Hematology (ASH); December 11–14, 2021; Atlanta, Georgia, USA.