Enfortumab Vedotin (EV) With or Without Pembrolizumab (P) in Cisplatin-Ineligible Patients (Pts) With Previously Untreated Locally Advanced or Metastatic Urothelial Cancer (la/mUC); Additional 3-Month Follow-up on Cohort K Data

Terence W. Friedlander[1], Matthew I. Milowsky[2], Peter H. O'Donnell[3], Daniel P. Petrylak[4], Christopher J. Hoimes[5], Thomas W. Flaig[6], Nataliya Mar[7], Helen H. Moon[8], Rana R. McKay[9], Mehmet A. Bilen[10], Delphine Borchiellini[11], Marco Iafolla[12], Anne-Sophie Carret[13], Yao Yu[13], Maria Guseva[14], Ritesh Kataria[15], Jonathan E. Rosenberg[16]

[1]University of California at San Francisco, San Francisco, CA; [2]University of North Carolina Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [3]University of Chicago, Chicago, IL; [4]Yale Cancer Center, New Haven, CT; [5]Duke Cancer Institute, Duke University, Durham, NC; [6]University of Colorado Comprehensive Cancer Center, Aurora, CO; [7]University of California at Irvine, Irvine, CA; [8]Kaiser Permanente Southern California, Riverside, CA; [9]University of California at San Diego, San Diego, CA; [10]Emory University Winship Cancer Institute, Atlanta, GA; [11]Centre Antoine Lacassagne, Université Côte d'Azur, Nice, France; [12]University of Toronto, Toronto, Canada; [13]Seagen Inc., Bothell, WA; [14]Astellas Pharma, Northbrook, IL; [15]Merck & Co., Inc., Rahway, NJ; [16]Memorial Sloan Kettering Cancer Center, New York, NY

Objectives

To provide updated data from Cohort K of the EV-103 study on the efficacy, safety, and tolerability of EV+P in the 1L treatment of cisplatin-ineligible patients at a median follow-up time of 18 months (3 additional months of follow-up than the previous analysis)

Conclusions

At a median follow-up of 18 months, EV+P continues to show a high cORR with rapid and durable responses as 1L treatment in cisplatin-ineligible patients with la/mUC

- Median DOR has not been reached, with approximately two-thirds of objective responses lasting ≥12 months

While PFS and OS continue to evolve, 12-month results for PFS and OS for EV+P are trending similarly to results from EV-103 Dose-escalation

- Median PFS and OS have not been reached
- 12-month PFS was 54.5% and OS was 81.5%

With additional follow-up, there was no meaningful change in the safety profile of EV+P

- AEs for the combination remained manageable, with no new safety signals observed

EV monotherapy results were generally consistent with prior results in 2L+ la/mUC

EV-302 is investigating the potential benefit of 1L EV+P compared to chemotherapy in cisplatin-eligible and cisplatin-ineligible patients with la/mUC

Abbreviations

1L: first-line; 2L: second-line; AEs: adverse events; AESIs: adverse events of special interest; BICR: blinded independent central review; CI: confidence interval; cORR: confirmed objective response rate; CPS: combined positive score; CR: complete response; DCR: disease control rate; DOR: duration or response; ECOG PS: Eastern Cooperative Oncology Group performance status; EV: enfortumab vedotin; gem-carbo: gemcitabine and carboplatin; IV: intravenous; la/mUC: locally advanced or metastatic urothelial cancer; mDOR: median duration of response; MDRD: modification of diet in renal disease; Mono: monotherapy; mOS: median overall survival; mos: months; mPFS: median progression-free survival; ORR: objective response rate; OS: overall survival; P: pembrolizumab; PD: progressive disease; PD-1: programmed cell death protein 1; PD-L1: programmed death-ligand 1; PFS: progression-free survival; PR: partial response; RECIST: Response Evaluation Criteria in Solid Tumors; TEAE: treatment-emergent adverse event; TRAEs: treatment-related adverse events; wk: week; yrs: years

Acknowledgements

This study is being conducted in collaboration with Astellas Pharma, Inc., Northbrook, IL, and with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ.

The authors thank Gary Dorrell, MS, ELS, CMPP, employee of Seagen Inc., for providing editorial support in accordance with Good Publication Practice guidelines.

References

1. Rosenberg JE. ESMO 2022: Oral presentation (Abstract 2895/LBA).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster,
Terence Friedlander, Terence.Friedlander@ucsf.edu

American Society of Clinical Oncology; June 2-6, 2023; Abstract No. 4568

Background

- Despite available therapeutic options, a significant unmet need remains for cisplatin-ineligible patients with la/mUC in the first-line setting
 - Gem-carbo followed by avelumab maintenance is only available to patients who do not progress after platinum-based chemotherapy
 - PD-1/L1 inhibitor monotherapy is only available to select patients
- EV+P received US accelerated approval in April 2023 in cisplatin-ineligible patients based on data from EV-103 (Dose Escalation/Cohort A and Cohort K), which demonstrated that this combination has rapid, durable responses (68% confirmed ORR [95% CI: 58.7 to 76.0]) and a manageable safety profile in patients with previously untreated la/mUC
- After a median follow-up of 18 months, an update on the results of EV-103 Cohort K[1] is presented

EV-103 Cohort K

Part of an open-label, multiple cohort, phase 1b/2 study in patients with urothelial carcinoma

Patient Population	Dose Escalation	Expansion Cohort A	Cohort K
Locally Advanced or Metastatic Urothelial Carcinoma (la/mUC)	EV+P Cisplatin-ineligible 1L (n=5)	EV+P Cisplatin-ineligible 1L (n=40)	1:1 Randomization EV+P or EV Cisplatin-ineligible 1L (N=151)

Stratification factors: Liver metastases (present/absent) and ECOG PS (0 or 1/2);
Exploratory endpoints: pharmacokinetics, antitherapeutic antibody, biomarkers of activity including baseline PD-L1 status and Nectin-4 expression, progression-free survival on subsequent therapy by investigator, patient reported outcomes
Data cutoff was 16SEP2022 except for time to objective response analysis and subgroup analysis of objective response, both of which had a data cutoff of 10JUN2022.

- **Dosing:** EV 1.25 mg/kg IV on days 1 and 8, and P 200 mg IV on day 1 of every 3-week cycle
- **Primary endpoint:** confirmed ORR by RECIST v1.1 per BICR
- **Key secondary endpoints:** confirmed ORR per RECIST v1.1 by investigator, DOR, DCR, PFS by BICR and by investigator, OS, safety/tolerability, and laboratory abnormalities

Statistical considerations

- The sample size was based on precision of the estimate for ORR characterized by 95% CIs
- No formal statistical comparisons between the 2 treatment arms

Results

Key Demographic and Baseline Disease Characteristics

Representative of the 1L cisplatin-ineligible la/mUC population

	EV+P (N=76)	EV Mono (N=73)
Male sex, n (%)	54 (71.1)	56 (76.7)
Age (yrs), median (range)	71 (51, 91)	74 (56, 89)
White race, n (%)	61 (80.3)	55 (75.3)
ECOG PS, n (%)		
0	33 (43.4)	28 (38.4)
1	33 (43.4)	35 (47.9)
2	10 (13.2)	10 (13.7)
Primary tumor location, n (%)[a]		
Lower tract	46 (60.5)	51 (69.9)
Upper tract	30 (39.5)	21 (28.8)
Metastasis disease site(s), n (%)[b]		
Bone	19 (25.0)	21 (28.8)
Liver	13 (17.1)	13 (17.8)
Lung	37 (48.7)	30 (41.1)
Metastasis category, n (%)		
Lymph node only	10 (13.2)	12 (16.4)
Visceral disease	64 (84.2)	60 (82.2)
Not applicable[c]	2 (2.6)	1 (1.4)

[a]1 patient in the EV Mono arm had primary disease at both the bladder and ureter
[b]Only key sites are identified. Patients may have had metastatic disease in more than 1 location
[c]Patients had locally advanced disease without metastasis to lymph nodes or distant organs

Summary of Disposition

The majority of patients remain on study

	EV+P (N=77)	EV Mono (N=74)
Patients treated, n	76	73
Patients on treatment, n (%)	19 (24.7)	6 (8.1)
Patients off treatment, n (%)	57 (74.0)	67 (90.5)
Reason for treatment discontinuation, n (%)		
Progressive disease	35 (45.5)	40 (54.1)
Adverse event	13 (16.9)	20 (27.0)
Patient decision	4 (5.2)	3 (4.1)
Physician decision	3 (3.9)	3 (4.1)
Other	2 (2.6)	1 (1.4)
Patients off study, n (%)	26 (33.8)	33 (44.6)
Reason for study discontinuation, n (%)		
Death	23 (29.9)	30 (40.5)
Patient withdrawal of consent	2 (2.6)	2 (2.7)
Other	1 (1.3)	1 (1.4)

Overall Response Rate by BICR

EV+P: 64.5% confirmed ORR with rapid response

	EV+P (N=76)	EV Mono (N=73)
Confirmed ORR, n (%) (95% CI)	**49 (64.5) (52.7, 75.1)**	**33 (45.2) (33.5, 57.3)**
Best overall response, n (%)		
Complete response	8 (10.5)	4 (5.5)
Partial response	41 (53.9)	29 (39.7)
Stable disease	17 (22.4)	25 (34.2)
Progressive disease	6 (7.9)	7 (9.6)
Not evaluable	3 (3.9)	5 (6.8)
No Assessment	1 (1.3)	3 (4.1)
Median time to objective response, mos (range)	2.07 (1.1, 6.6)	2.07 (1.9, 15.4)
Median number of treatment cycles (range)	12.0 (1, 34)	8.0 (1, 33)

EV+P

- 42/49 (85.7%) of responses observed at first assessment (week 9 ±1 wk)
- cORRs were consistent across all pre-specified subgroups
- 7/13 (53.8%) cORR observed in patients with liver metastases

EV monotherapy

- Activity is consistent with prior results in 2L+ la/mUC

Duration of Response per BICR

Median DOR for EV+P was not reached; 65.6% of responders were still responding at 12 months

	EV+P (N=76)	EV Mono (N=73)
Responders, n	49	33
Progression events, n	14[a]	14
mDOR (95% CI), mos	– (10.25, –)	13.2 (6.14, –)
DOR ≥12 mos, %	65.6	59.7

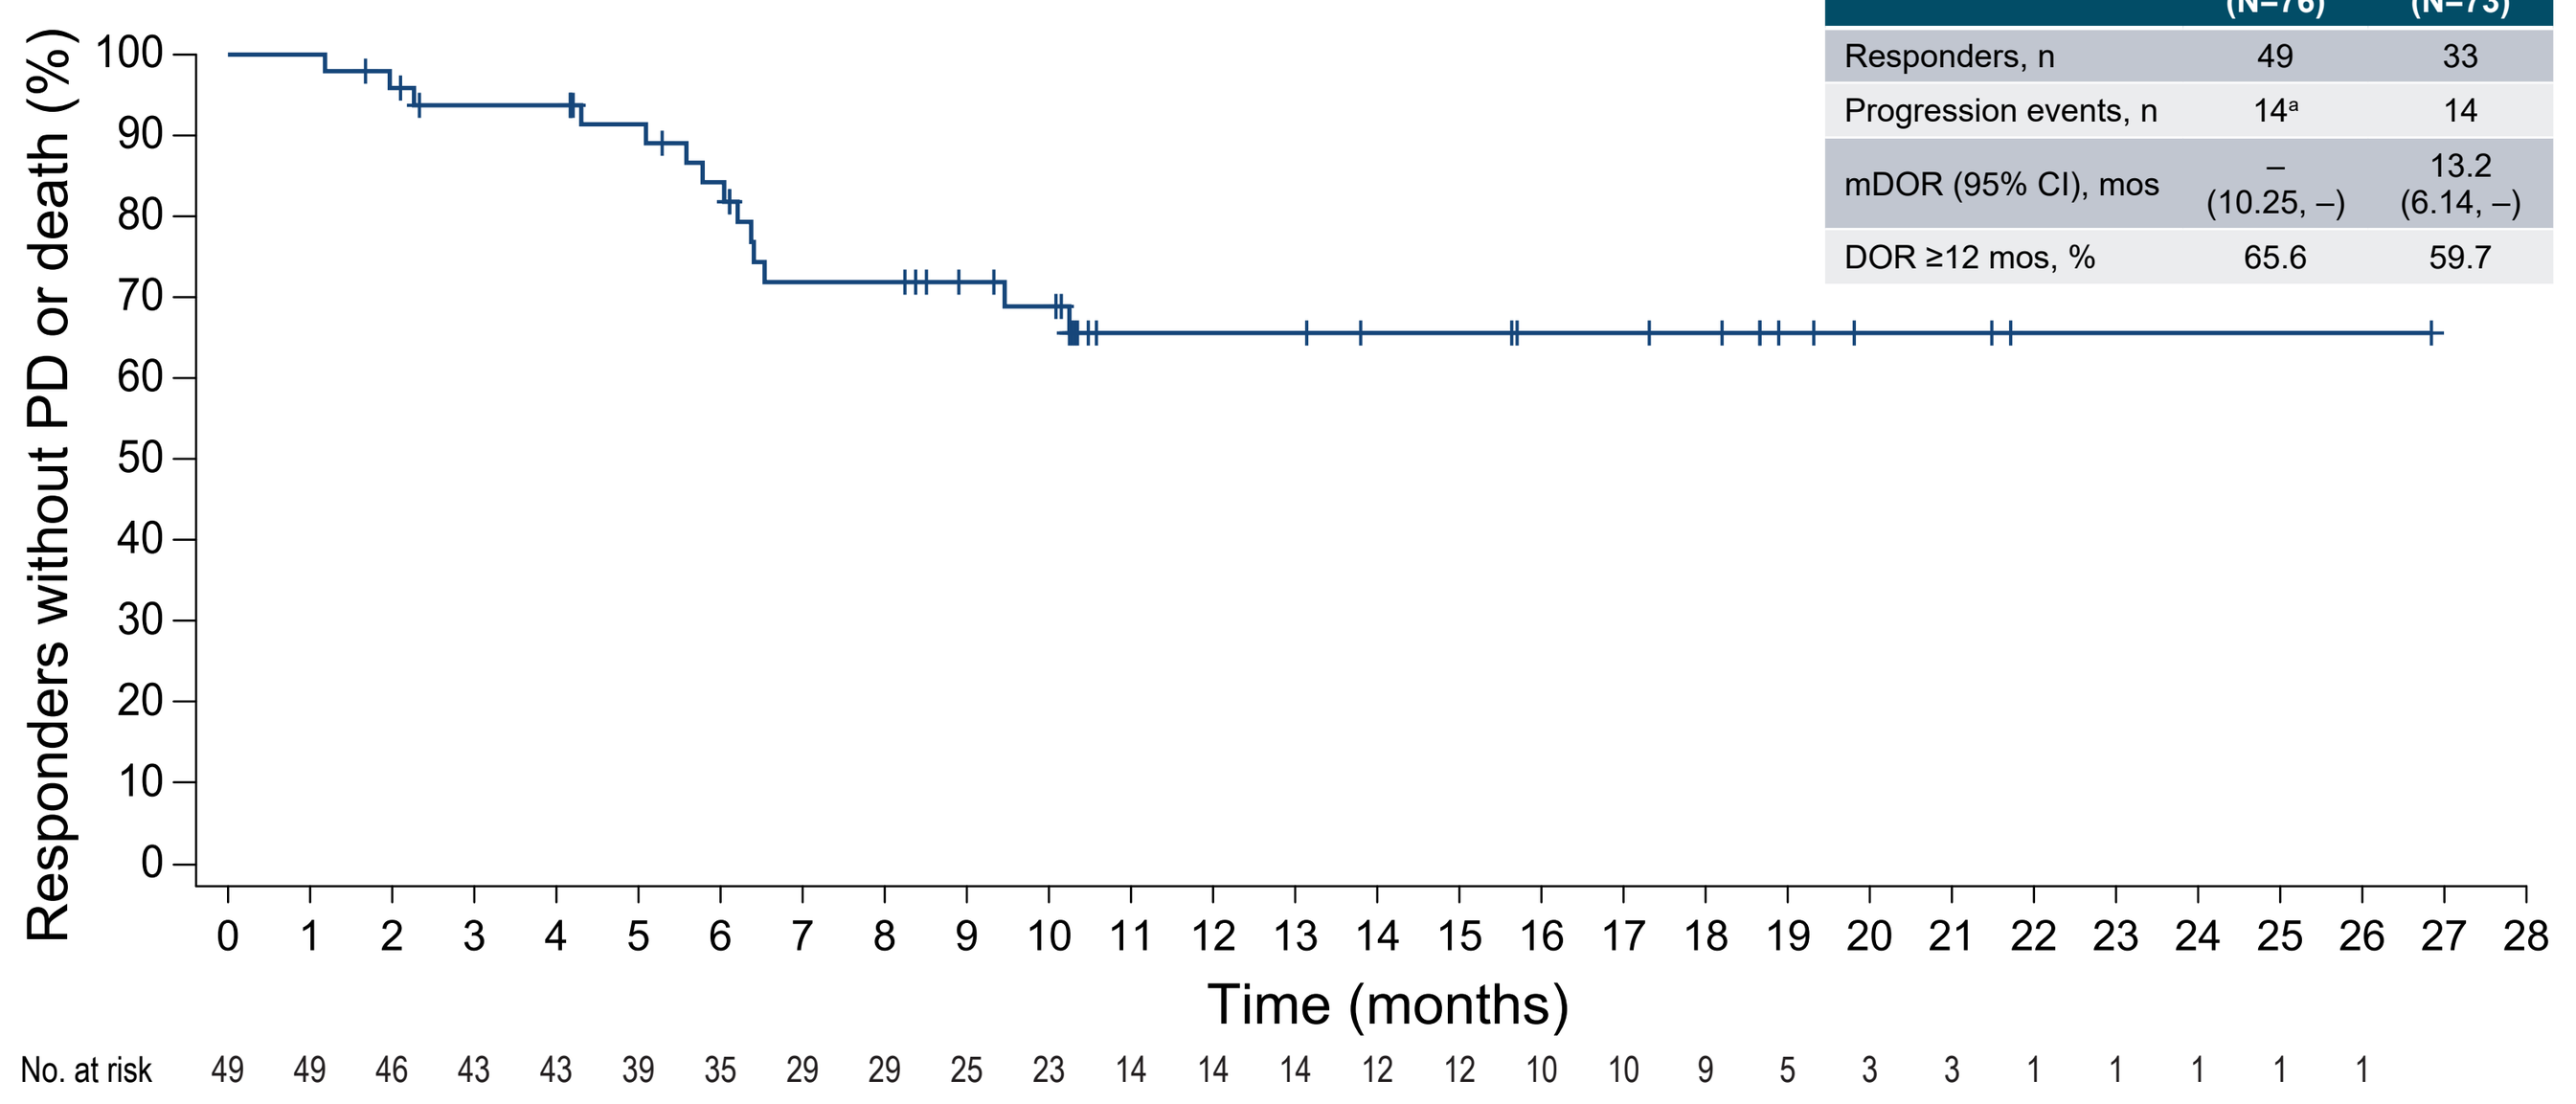
Responders without PD or death (%)
100
90
80
70
60
50
40
30
20
10
0
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28
Time (months)
No. at risk 49 49 46 43 43 39 35 29 29 25 23 14 14 14 12 12 10 10 9 5 3 3 1 1 1 1 1

[a]1 additional disease progression observed since the previous analysis[1]

Progression-Free Survival per BICR and Overall Survival

Median PFS and OS for EV+P were not reached

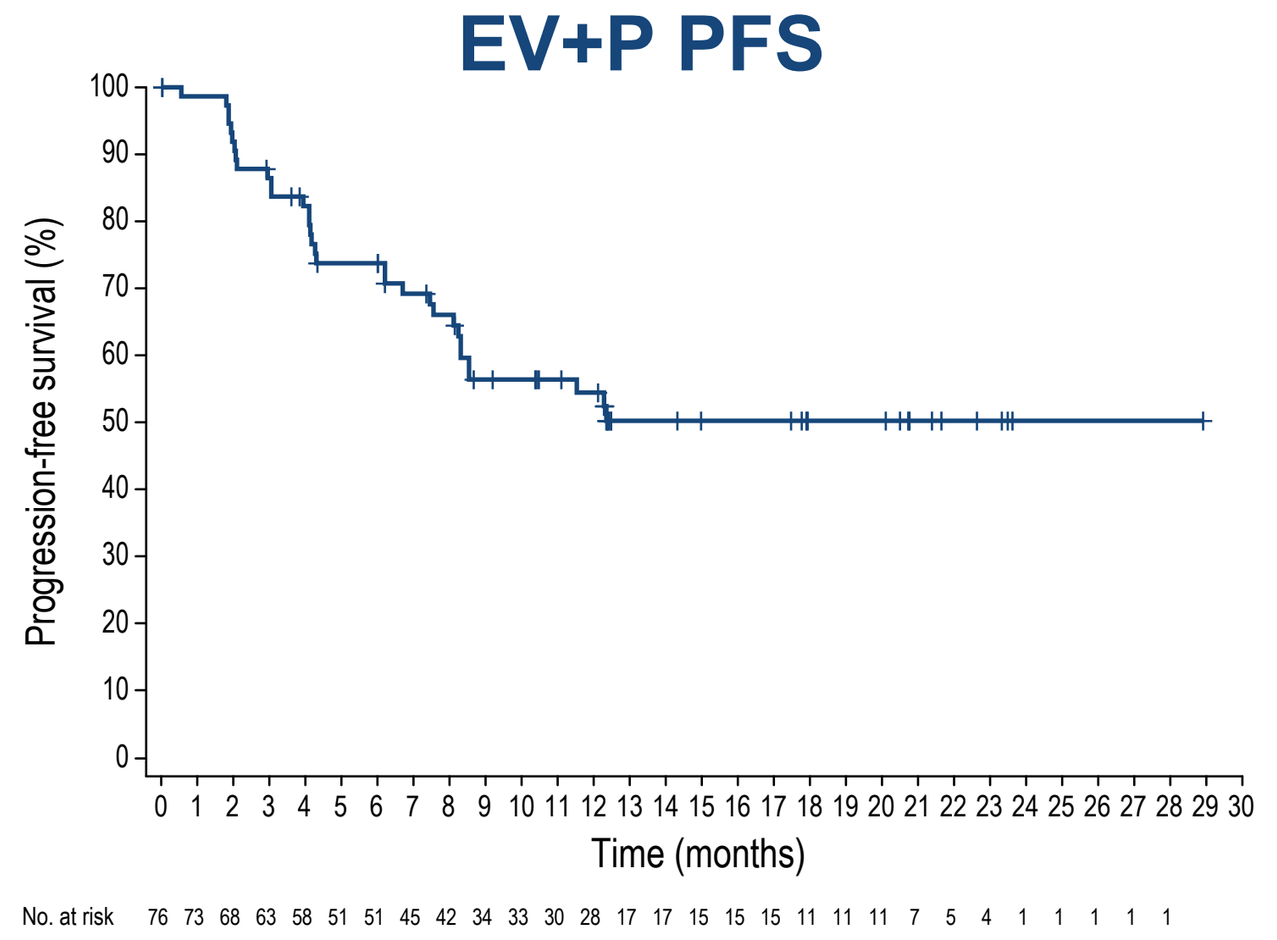
EV+P PFS
Progression-free survival (%)
Time (months)
No. at risk 76 73 68 63 58 51 51 45 42 34 33 30 28 17 17 15 15 15 11 11 11 7 5 4 1 1 1 1 1

	EV+P (N=76)	EV Mono (N=73)
PFS events, n	33	37
mPFS (95% CI), mos	– (8.31, –)	8.2 (6.05, 15.28)
PFS at 12 mos, %	54.5	40.3

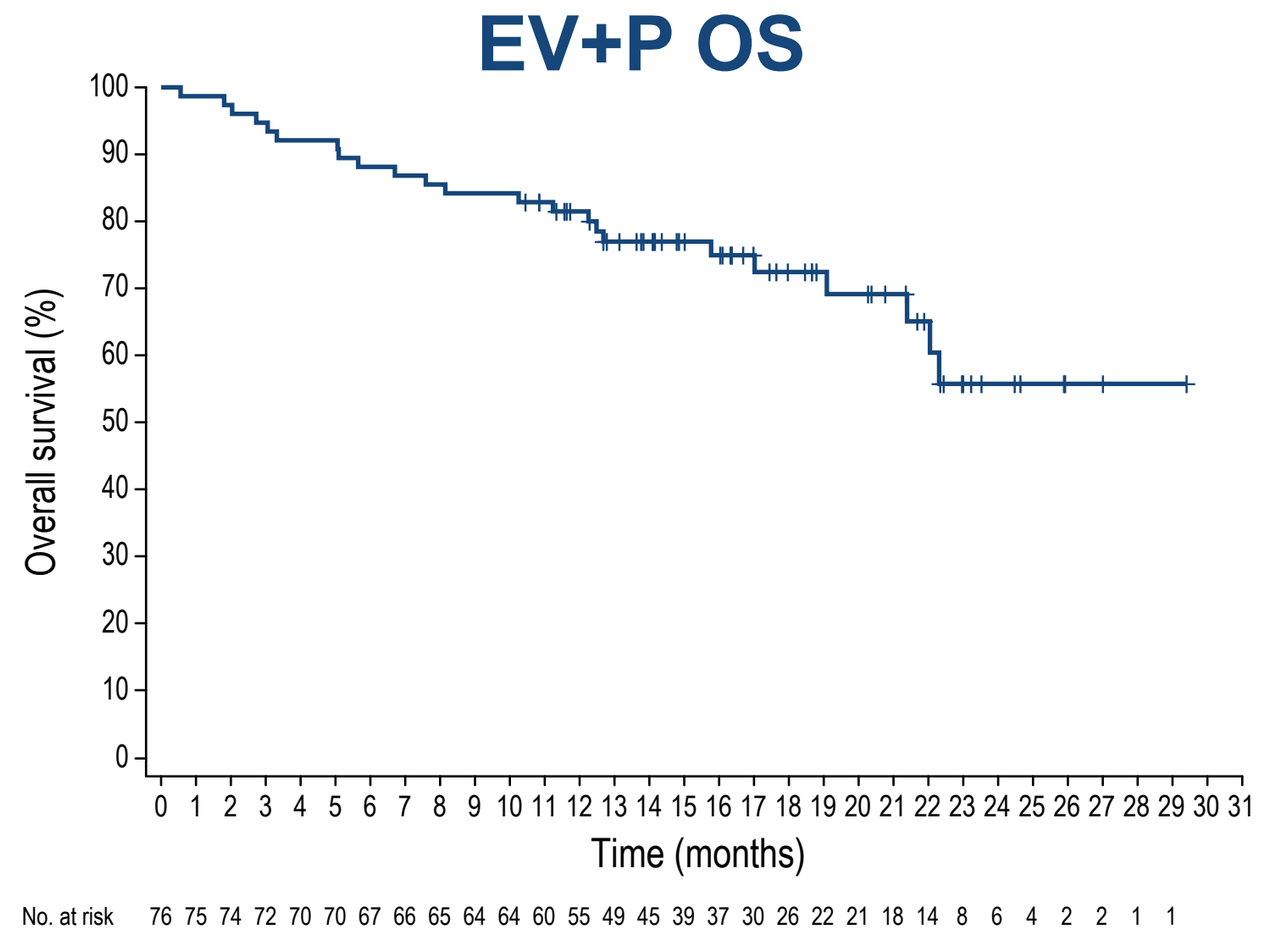
EV+P OS
Overall survival (%)
Time (months)
No. at risk 76 75 74 72 70 70 67 66 65 64 64 60 55 49 45 39 37 30 26 22 21 18 14 8 6 4 2 2 1 1

	EV+P (N=76)	EV Mono (N=73)
OS events, n	23	30
mOS (95% CI), mos	– (21.39, –)	21.7 (15.47, –)
OS at 12 mos, %	81.5	69.7
Median follow-up time, mos	17.6	18.2

Treatment-Related Adverse Events

Most common AEs with EV+P were fatigue, peripheral sensory neuropathy, maculopapular rash, and alopecia

TRAEs any grades by preferred term ≥20% of patients	EV+P (N=76) n (%)		EV Mono (N=73) n (%)	
	Any grade	Grade ≥3	Any grade	Grade ≥3
Overall	76 (100.0)	49 (64.5)	68 (93.2)	34 (46.6)
Fatigue	43 (56.6)	7 (9.2)	29 (39.7)	6 (8.2)
Peripheral sensory neuropathy	41 (53.9)	1 (1.3)	32 (43.8)	2 (2.7)
Rash maculopapular	36 (47.4)	13 (17.1)	21 (28.8)	1 (1.4)
Alopecia	35 (46.1)	0	27 (37.0)	0
Pruritus	30 (39.5)	3 (3.9)	19 (26.0)	1 (1.4)
Dysgeusia	23 (30.3)	0	25 (34.2)	0
Weight decreased	23 (30.3)	3 (3.9)	22 (30.1)	1 (1.4)
Diarrhea	22 (28.9)	5 (6.6)	20 (27.4)	4 (5.5)
Decreased appetite	20 (26.3)	0	28 (38.4)	0
Nausea	21 (27.6)	0	26 (35.6)	1 (1.4)
Dry eye	16 (21.1)	0	8 (11.0)	0

Serious TRAEs

- 19 (25.0%) EV+P; 11 (15.1%) EV Mono

TRAEs leading to death (per investigator)

- 3 (3.9%) EV+P (pneumonitis, respiratory failure, sepsis)
- 2 (2.7%) EV Mono (multiple organ dysfunction, respiratory failure)

Treatment-Related Adverse Events of Special Interest for EV

The majority of treatment-related AESIs were low grade

	EV+P (N=76) n (%)		EV Mono (N=73) n (%)	
	Any grade	Grade ≥3	Any grade	Grade ≥3
Skin reactions	51 (67.1)	16 (21.1)	33 (45.2)	5 (6.8)
Peripheral neuropathy	48 (63.2)	2 (2.6)	40 (54.8)	2 (2.7)
Ocular disorders	20 (26.3)	0	21 (28.8)	0
Dry eye	20 (26.3)	0	21 (28.8)	0
Blurred vision	2 (2.6)	0	5 (6.8)	0
Corneal disorders	0	0	4 (5.5)	0
Hyperglycemia	11 (14.5)	5 (6.6)	8 (11.0)	7 (9.6)
Infusion-related reactions	3 (3.9)	0	4 (5.5)	0

- Skin reactions were observed more frequently with EV+P
- Peripheral neuropathy remains the most common reason for study treatment discontinuation

Treatment-Emergent Adverse Events of Special Interest for Pembrolizumab

	EV+P (N=76) n (%)	
	Any grade	Grade ≥3
Severe skin reactions[a]	21 (27.6)	15 (19.7)
Hypothyroidism	10 (13.2)	0
Pneumonitis	7 (9.2)	4 (5.3)
Adrenal insufficiency	3 (3.9)	0
Colitis	3 (3.9)	1 (1.3)
Hyperthyroidism	3 (3.9)	0
Infusion reactions	3 (3.9)	0
Hepatitis	2 (2.6)	2 (2.6)
Myasthenic syndrome	2 (2.6)	2 (2.6)
Myositis	2 (2.6)	0
Pancreatitis	2 (2.6)	1 (1.3)
Hypophysitis	1 (1.3)	0
Myocarditis	1 (1.3)	0
Nephritis	1 (1.3)	1 (1.3)
Thyroiditis	1 (1.3)	0

- Pembrolizumab TEAEs were consistent with previously observed results with pembrolizumab monotherapy, except for severe skin reactions, which were reported with a higher incidence in this study.

[a]There are differences in the rates of skin reactions reported for EV treatment-related AESIs and pembrolizumab TEAEs of special interest because the analyses for reporting these events were conducted using different methods developed for EV and pembrolizumab monotherapies