PHASE 3 STUDY OF TUCATINIB OR PLACEBO IN COMBINATION WITH TRASTUZUMAB AND PERTUZUMAB AS MAINTENANCE THERAPY FOR HER2+ METASTATIC BREAST CANCER (HER2CLIMB-05, TRIAL IN PROGRESS)

Erika Hamilton[1], Junji Tsurutani[2], Joohyuk Sohn[3], Giuseppe Curigliano[4], Ciara C. O'Sullivan[5], Miguel Martín[6], Konstantinos Tryfonidis[7], Libero Santarpia[8], Shan Yang[8], Véronique Diéras[9]

[1]Sarah Cannon Research Institute at Tennessee Oncology, Nashville, TN, USA; [2]Advanced Cancer Translational Research Institute at Showa University, Tokyo, Japan; [3]Yonsei Cancer Center, Seoul, Republic of Korea; [4]European Institute of Oncology, IRCCS, University of Milano, Milan, Italy; [5]Mayo Clinic, Rochester, MN, USA; [6]Hospital General Universitario Gregorio Marañón, Madrid, Spain; [7]Merck & Co., Inc., Rahway, NJ, USA; [8]Seagen Inc., Bothell, WA, USA; [9]Eugène Marquis Centre, Rennes, France

BACKGROUND

- The current 1L SOC for HER2+ MBC is trastuzumab plus pertuzumab and a taxane[1,2]
 - This regimen improved overall survival outcomes by 16 months compared with the prior SOC, trastuzumab and a taxane[3]
 - Despite advances in 1L SOC therapy, most patients progress on maintenance therapy with trastuzumab and pertuzumab[1]
- Tucatinib is an oral TKI approved in multiple countries in combination with trastuzumab and capecitabine for adult patients with HER2+ MBC, with or without BMs[4–7]
 - Tucatinib in combination with trastuzumab and capecitabine has been proven to demonstrate a statistically significant and clinically meaningful improvement in PFS and OS with a tolerable safety profile in patients with HER2+ MBC[4,7,8]
- As up to 50% of patients with HER2+ MBC will develop BMs; prevention and treatment of BMs is an urgent unmet clinical need[9]
 - Adding tucatinib to trastuzumab plus capecitabine also reduced the risk of disease progression or death in patients with active or stable BMs[4,7,8]
- The addition of tucatinib to 1L SOC maintenance therapy with trastuzumab and pertuzumab may extend PFS while maintaining QOL[10]
 - In patients with BMs, tucatinib has demonstrated the ability to improve PFS and OS and/or delay the emergence of BMs; therefore, it is thought patients in the 1L setting may also benefit from receiving tucatinib[8]

PROPOSED MECHANISM OF ACTION OF TUCATINIB

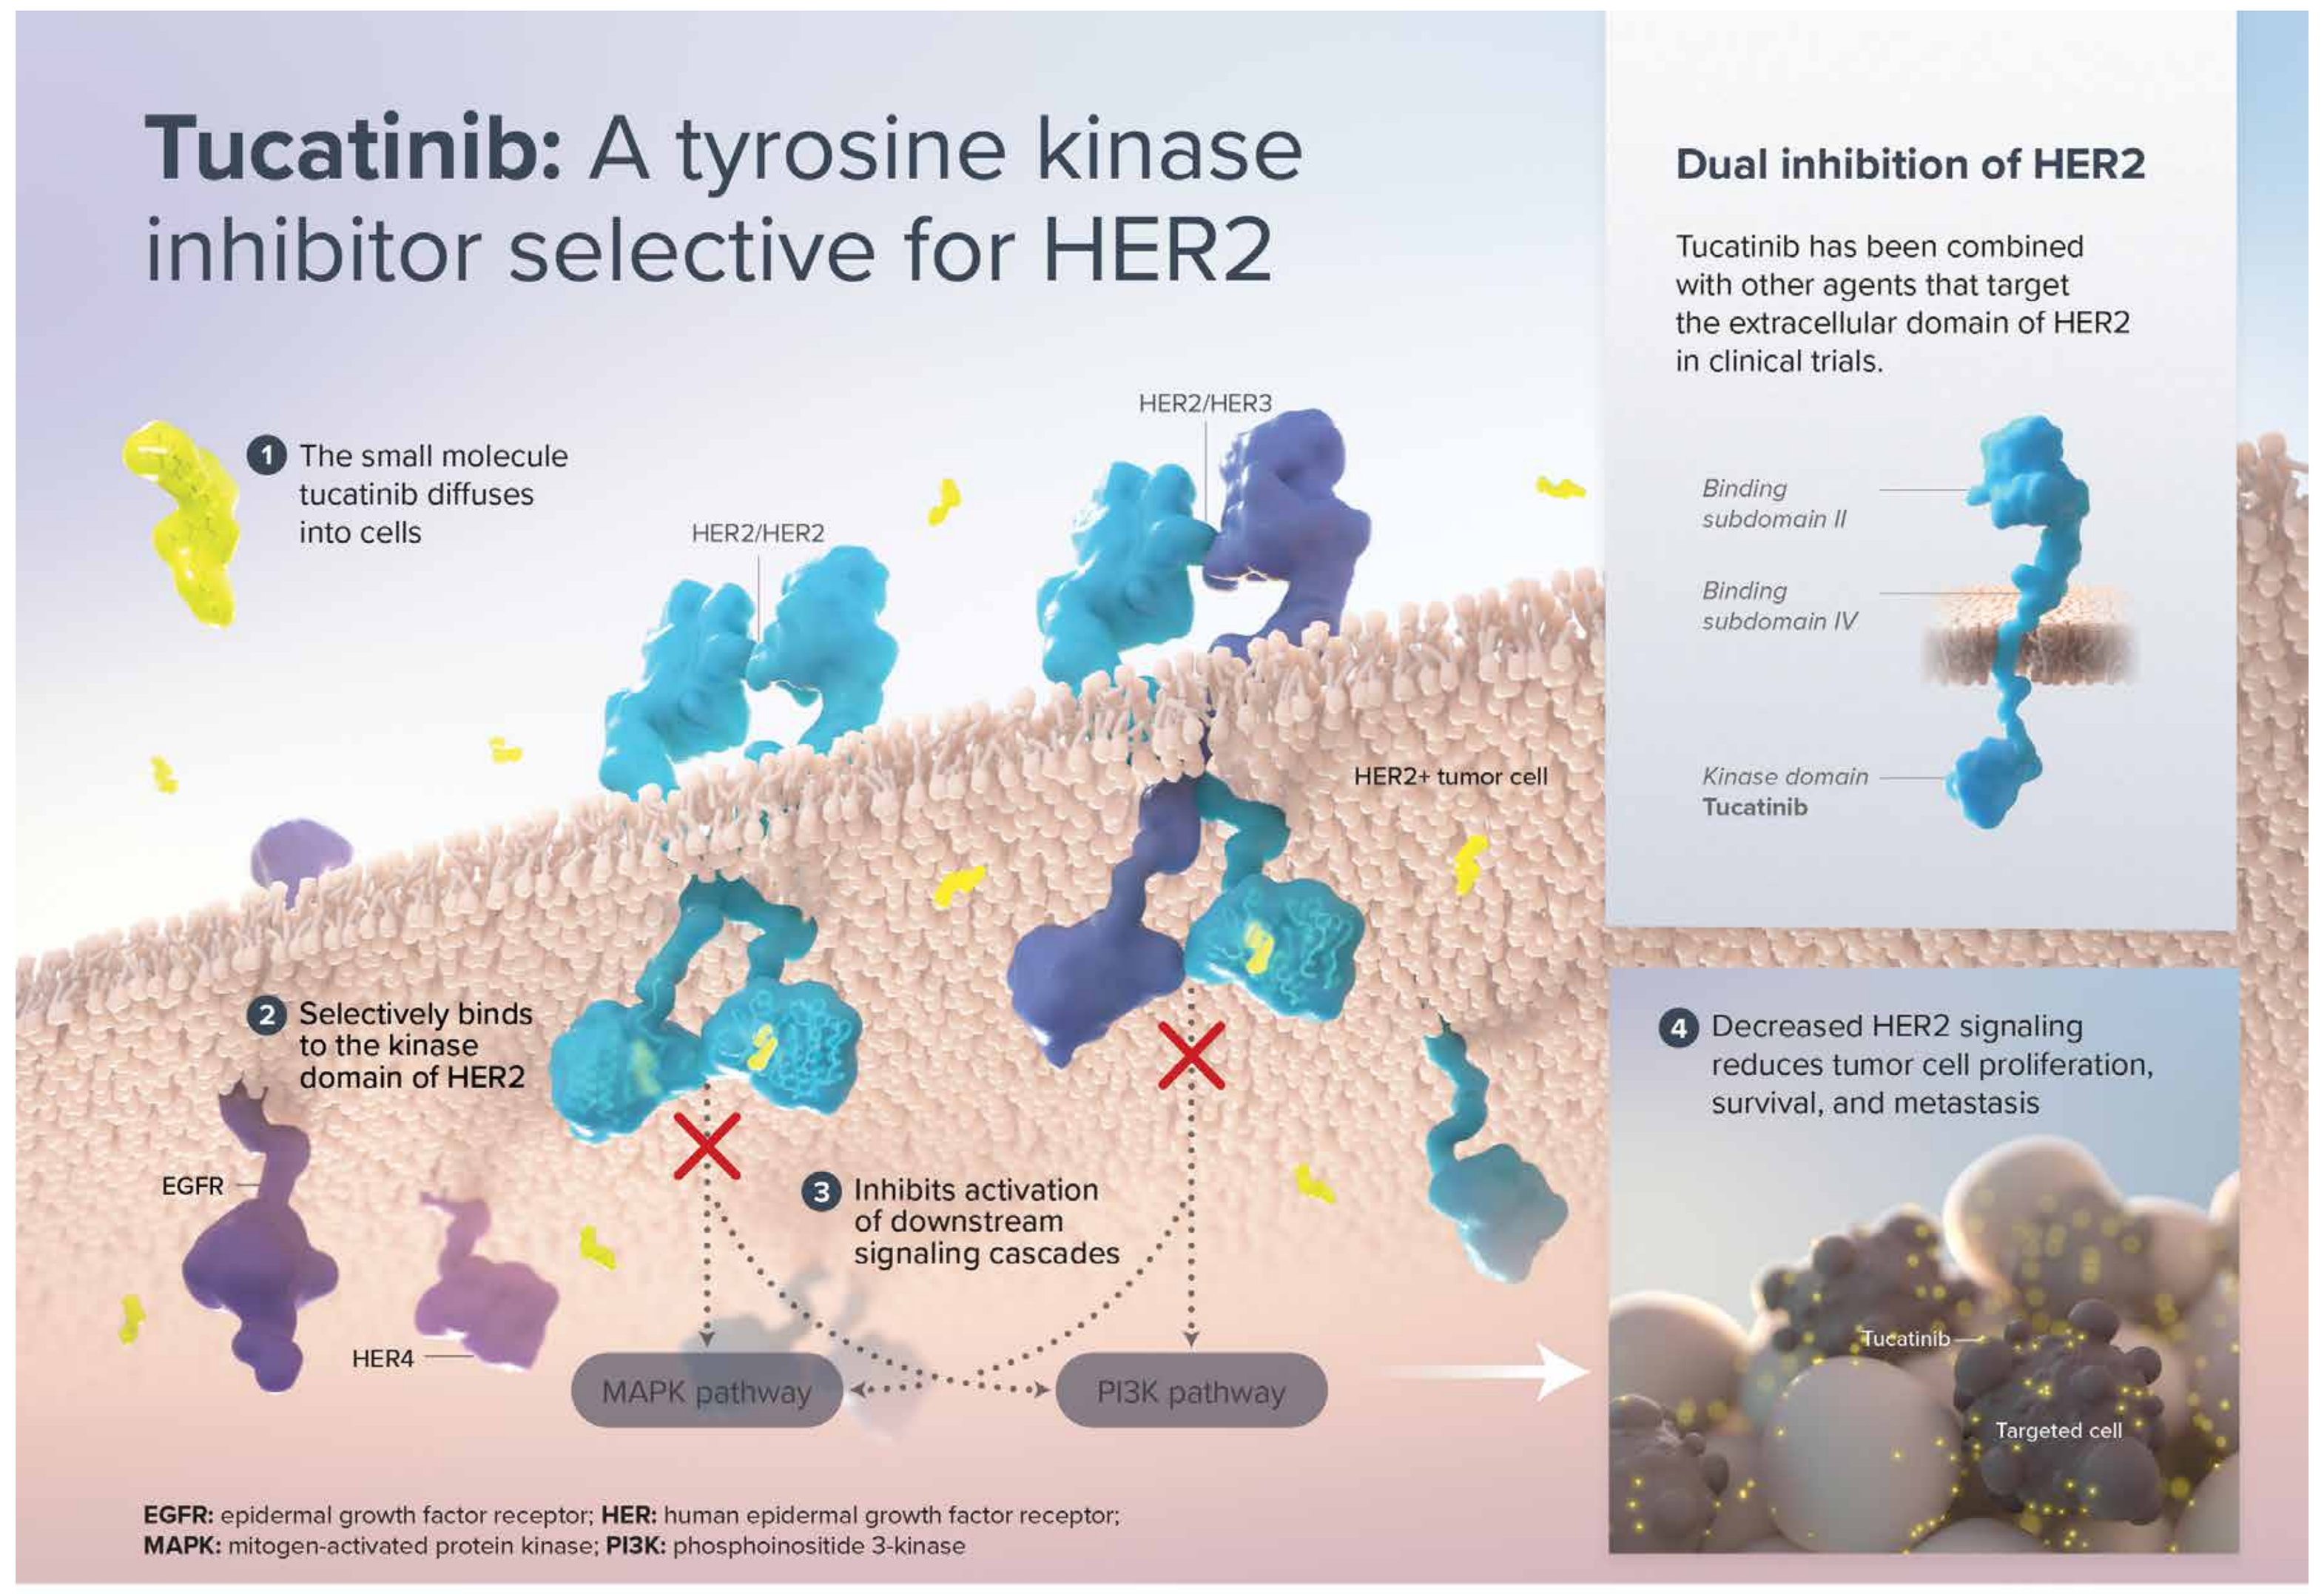
Tucatinib: A tyrosine kinase inhibitor selective for HER2
1 The small molecule tucatinib diffuses into cells
HER2/HER2
HER2/HER3
HER2+ tumor cell
2 Selectively binds to the kinase domain of HER2
EGFR
HER4
3 Inhibits activation of downstream signaling cascades
MAPK pathway
PI3K pathway
Dual inhibition of HER2
Tucatinib has been combined with other agents that target the extracellular domain of HER2 in clinical trials.
Binding subdomain II
Binding subdomain IV
Kinase domain Tucatinib
4 Decreased HER2 signaling reduces tumor cell proliferation, survival, and metastasis
Tucatinib
Targeted cell
EGFR: epidermal growth factor receptor; HER: human epidermal growth factor receptor;
MAPK: mitogen-activated protein kinase; PI3K: phosphoinositide 3-kinase

Tucatinib is an investigational agent, and its safety and efficacy have not been established. There is no guarantee that tucatinib will receive regulatory approval and become commercially available for uses being investigated.

© 2023 Seagen Inc. Bothell WA 98021. All rights reserved. USM/TUC/2019/0018

Abbreviations

1L, first-line; AE, adverse event; ASCO CAP, American Society of Clinical Oncology College of American Pathologists; BICR, blinded independent central review; BID, twice a day; BM, brain metastasis; CNS, central nervous system; CNS-PFS, time from randomization to investigator-assessed disease progression in brain; ECOG, Eastern Cooperative Oncology Group; EGFR, epidermal growth factor receptor; EORTC QLQ-C30, European Organization for the Research and Treatment of Cancer Core Quality of Life Questionnaire; EOT, end of treatment; EQ-5D-5L, 5-level EQ-5D; EU, European Union; HER2, human epidermal growth factor receptor 2; HER2+, HER2-positive; HR+, hormone receptor positive; HR–, hormone receptor negative; HR-QOL, health-related quality of life; IV, intravenous; LA, locally advanced; LMD, leptomeningeal disease; MBC, metastatic breast cancer; MRI, magnetic resonance imaging; OS, overall survival; PFS, progression-free survival; PO, by mouth; PK, pharmacokinetics; QOL, quality of life; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; SOC, standard of care; SC, subcutaneous; T-DM1, trastuzumab emtansine; TKI, tyrosine kinase inhibitor; UK, United Kingdom; US, United States.

References

1. Baselga J, et al. N Engl J Med. 2012;366(2):109-19.
2. Giordano SH, et al. J Clin Oncol. 2014;32(19):2078-99.
3. Swain SM, et al. Lancet Oncol. 2013;14(6):461-71.
4. Curigliano G, et al. J Clin Oncol. 2021;39(15):1043.
5. TUKYSA® Prescribing Information, Seagen Inc., 2020.
6. Tukysa (tucatinib) European summary of product characteristics. 2021.
7. Murthy RK, et al. N Engl J Med. 2020;382(7):597-609.
8. Lin NU, et al. J Clin Oncol. 2020;38(23):2610-19.
9. Leone JP, Lin NU. Curr Oncol Rep. 2019;21(6):49.
10. Mueller V, et al. Eur J Cancer. 2021;153:223-33.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster, Erika Hamilton, **ehamilton@tnonc.com**.

STUDY SCHEMA

- HER2CLIMB-05 (NCT05132582) is a phase 3, randomized, double-blind study evaluating tucatinib or placebo in combination with trastuzumab plus pertuzumab as maintenance therapy in the 1L setting for patients with unresectable LA or metastatic HER2+ breast cancer following SOC induction therapy

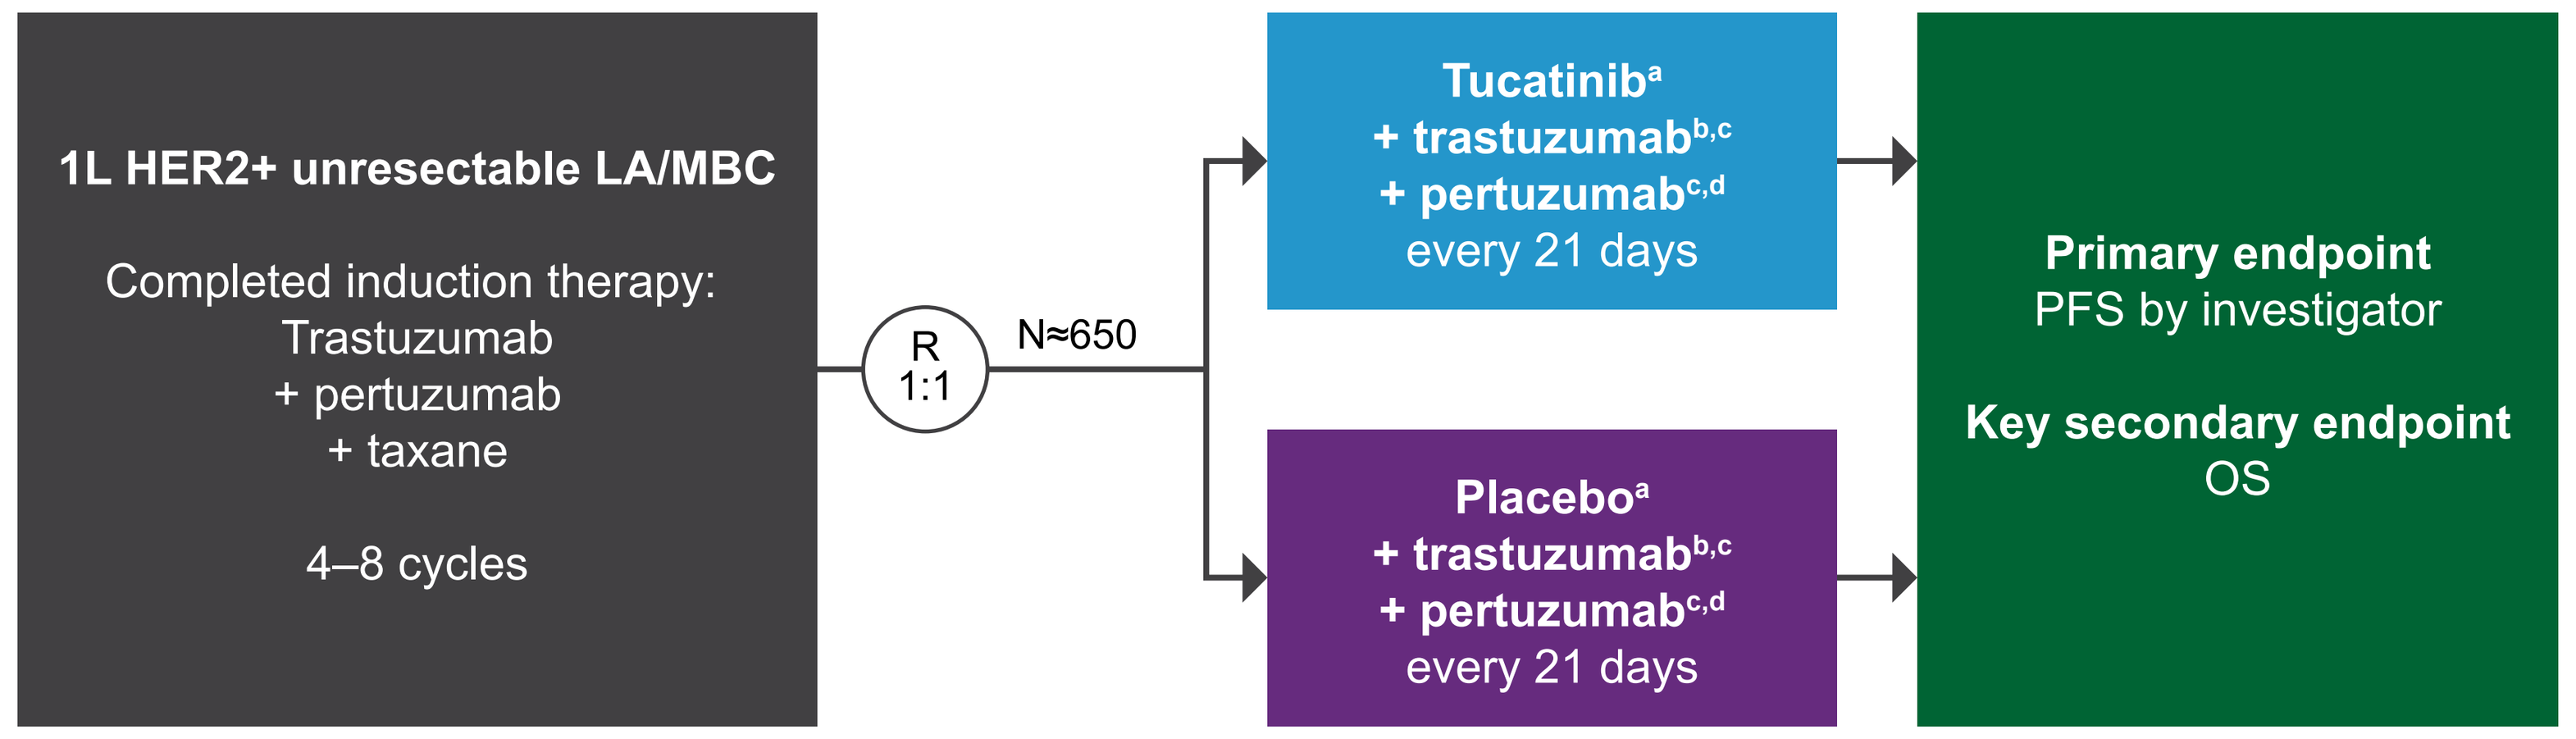
1L HER2+ unresectable LA/MBC
Completed induction therapy: Trastuzumab + pertuzumab + taxane
4–8 cycles
R 1:1
N≈650
Tucatinib[a] + trastuzumab[b,c] + pertuzumab[c,d] every 21 days
Placebo[a] + trastuzumab[b,c] + pertuzumab[c,d] every 21 days
Primary endpoint
PFS by investigator
Key secondary endpoint
OS

Randomization will be stratified by diagnosis (de novo vs recurrent MBC), hormone receptor status (positive vs negative), and presence or history of BMs (yes vs no).
a Tucatinib/placebo 300 mg will be administered PO from Cycle 1 Day 1 onward, BID on each day of study treatment.
b IV trastuzumab will be given at a dose of 6 mg/kg once every 21 days. Alternatively, trastuzumab may be administered as an SC dose, at a fixed dose of 600 mg once every 21 days. SC trastuzumab does not require a loading dose.
c A fixed dose of trastuzumab + pertuzumab (600 mg pertuzumab, 600 mg trastuzumab, and 20,000 units hyaluronidase) can be administered every 21 days by SC administration, in lieu of trastuzumab and pertuzumab administered IV individually.
d Pertuzumab 420 mg will be administered every 21 days intravenously over 30-60 minutes.

STUDY TREATMENT

- Patients will receive tucatinib 300 mg or placebo PO BID, trastuzumab 6 mg/kg IV or 600 mg SC, and pertuzumab 420 mg IV every 21-day cycle
- In some countries, patients will receive pertuzumab 600 mg, trastuzumab 600 mg, and 20,000 units hyaluronidase SC every 21-day cycle in place of trastuzumab and pertuzumab individually
- Patients with HR+ tumors may receive endocrine therapy per institutional SOC

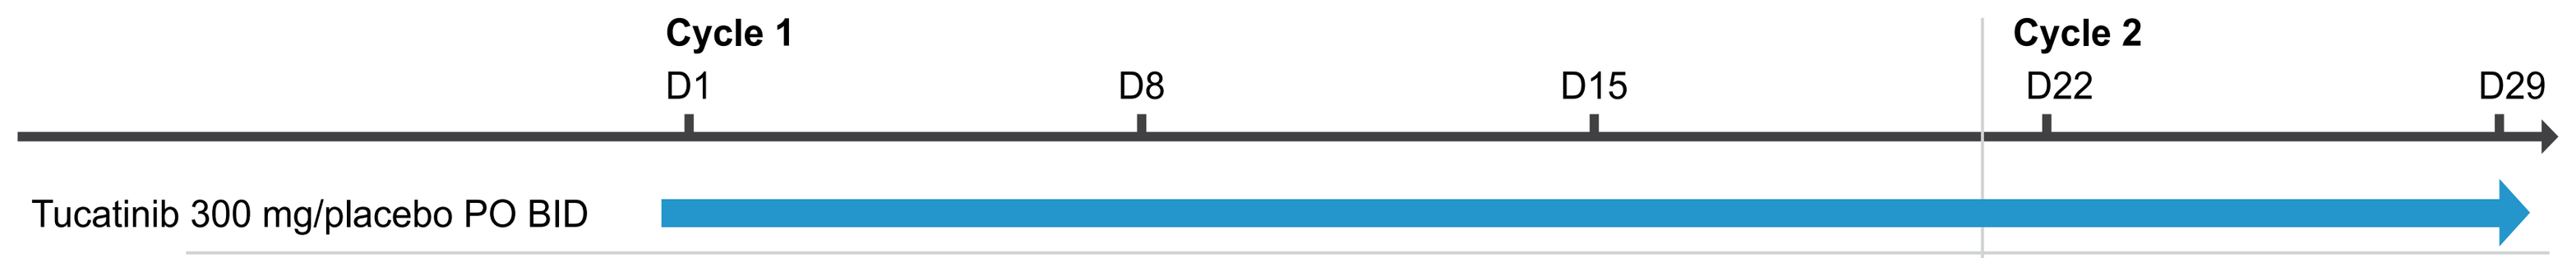
Cycle 1
D1
D8
D15
Cycle 2
D22
D29
Tucatinib 300 mg/placebo PO BID
Trastuzumab + pertuzumab

Study treatment will continue until unacceptable toxicity, disease progression, withdrawal of consent, or study closure. If a patient is found to have radiographic progressive disease per RECIST v1.1 criteria based on isolated CNS progression without progression of extracranial disease, the patient may be eligible to continue study treatment after completion of local treatment of BMs.

ELIGIBILITY

Key Inclusion Criteria

- Centrally confirmed HER2+ breast carcinoma per 2018 ASCO CAP guidelines
- Unresectable locally advanced or metastatic disease
 - If recurrent (after [neo]adjuvant therapy), must be ≥6 months treatment free from any trastuzumab or pertuzumab received for advanced HER2+ disease
- Received 4–8 cycles (21-day cycles) of previous treatment with trastuzumab, pertuzumab, and taxane as 1L therapy for advanced HER2+ breast cancer with no evidence of disease progression
- Known hormone receptor status (per local guidelines; may be HR+ or HR−)
- ECOG performance status score of 0 or 1
- **CNS inclusion criteria:** Based on screening contrast brain MRI at baseline or at screening, patients may have any of the following:
 - No evidence of BMs
 - Untreated BMs which are asymptomatic, and if identified on prior brain imaging, without evidence of progression since starting 1L induction therapy with trastuzumab, pertuzumab, and taxane
 - Previously treated BMs which are asymptomatic or must not have progressed since treatment

Key Exclusion Criteria

- Prior treatment with any anti-HER2 and/or anti-EGFR TKI including pyrotinib, lapatinib, tucatinib, neratinib, and afatinib (except neratinib if given in the extended adjuvant setting and ≥12 months have elapsed since the last neratinib dose prior to the start of study drug)
- Unable to undergo contrast MRI of the brain
- **CNS exclusion criteria:** Based on screening brain MRI and clinical assessment, patients must not have any of the following:
 - Symptomatic BM
 - Progression of BMs since starting 1L trastuzumab, pertuzumab, and taxane
 - Ongoing use of systemic corticosteroids at a total daily dose of >2 mg of dexamethasone (or equivalent)
 - Any untreated brain lesion in an anatomic site which may pose a risk to patient
 - Known or suspected LMD
 - Poorly controlled (>1/week) seizures or other persistent neurologic symptoms

OBJECTIVES

Primary Objective	Endpoints
Evaluate antitumor activity of tucatinib in combination with trastuzumab and pertuzumab	**Primary endpoint:** PFS by investigator per RECIST v1.1 **Key secondary endpoint:** OS
Other Secondary Objectives	**Endpoints**
Evaluate overall PFS and PFS in the brain	PFS by BICR per RECIST v1.1, and CNS-PFS
Assess the change in HR-QOL	Time to deterioration of HR-QOL, defined as time to 10-point decrease in the global health status/QOL scale of the EORTC QLQ-C30 questionnaire
Evaluate the safety and tolerability of tucatinib in combination with trastuzumab and pertuzumab	• AEs • Clinical laboratory assessments • Incidence of dose holding, dose reductions, and discontinuations of tucatinib • Incidence of dose holding and discontinuations of trastuzumab and pertuzumab
Evaluate the PK of tucatinib	Plasma concentrations of tucatinib
Exploratory Objectives	**Endpoints**
Identify somatic alterations potentially associated with resistance to study treatment	Somatic alterations associated with resistance to tucatinib
Evaluate health utilities	HR-QOL utilities as assessed with the EQ-5D-5L instrument
Evaluate HR-QOL	Change from baseline in global health status/QOL, and physical and role functional scales of the EORTC QLQ-C30

ASSESSMENTS

- Disease assessments: PFS per RECIST v1.1 will be assessed by investigator and BICR; treatment decisions will be made based upon local radiologic assessment
 - Brain MRI: required for all patients at screening and at EOT, and is to be repeated for patients with a baseline or history of BM every 9 weeks. For patients without a baseline or history of BMs, MRI is to be repeated every 27 weeks
 - Survival and other follow-up assessments: until study closure or withdrawal of consent
- PK assessments: blood samples of trough tucatinib drug levels will be collected in all patients on Day 1 of Cycles 2 to 6, prior to administration of tucatinib. On Day 1 of Cycle 3, blood samples will also be collected postdose for PK assessments of peak levels of tucatinib 1 to 4 hours after administration of tucatinib
- Safety assessments: summaries of AEs and changes in laboratory test results, vital signs, and physical examination findings
- HR-QOL assessments: at protocol-specified time points using standardized assessment tools including the EQ-5D-5L instrument and the EORTC QLQ-C30

SUMMARY

- HER2CLIMB-05 will determine whether adding tucatinib to 1L SOC maintenance therapy with trastuzumab and pertuzumab will extend PFS while maintaining HR-QOL in patients with HER2+ MBC
- In addition, the effect of adding tucatinib to 1L SOC for treatment and potential prevention of BMs is of great interest
- Enrollment is ongoing in the US, Canada, the UK, Belgium, France, Spain, Australia, Japan, and South Korea, with additional sites planned

Acknowledgements

Kyrillos Soryal, PharmD, of Seagen Inc. provided medical writing and editorial support with funding from Seagen Inc., in accordance with Good Publication Practice guidelines. Reused with permission from the European Society for Medical Oncology (ESMO). This abstract was accepted and previously presented by Véronique Diéras at ESMO-BC 2022, FPN (Final Publication Number): 415, Annals of Oncology, Volume 33, 2022 Supplement 3. All rights reserved.

Disclosures

This study was sponsored by Seagen Inc., Bothell, WA, USA in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

Dr. Hamilton: Pfizer, Genentech/Roche, Lilly, Daiichi Sankyo, Mersana, AstraZeneca, Novartis, Seagen Inc., Arcus Biosciences, iTeos Therapeutics, Janssen, Loxo, Relay Therapeutics, Greenwich LifeSciences, Orum Therapeutics, Ellipses Pharma, Olema Pharmaceuticals, Stemline Therapeutics, Tubulis GmbH, and Verascity Science (to institution - consulting / advisory role); AstraZeneca, Hutchison MediPharma, OncoMed, MedImmune, Stem CentRx, Genentech/Roche, Curis, Verastem, Zymeworks, Syndax, Lycera, Rgenix, Novartis, Mersana, Millennium, TapImmune Inc., Lilly, Pfizer, Tesaro, Boehringer Ingelheim, H3 Biomedicine, Radius Health, Acerta Pharma, Macrogenics, AbbVie, Immunomedics, Fujifilm, eFFECTOR Therapeutics, Merus, Nucana, Regeneron, Leap Therapeutics, Taiho Pharmaceutical, EMD Serono, Daiichi Sankyo, ArQule, Syros Pharmaceuticals, Clovis Oncology, CytomX Therapeutics, InventisBio, Deciphera, Sermonix Pharmaceuticals, Sutro Biopharma, Zenith Epigenetics, Arvinas, Harpoon, Black Diamond Therapeutics, Orinove, Molecular Templates, Seagen Inc., Compugen, G1 Therapeutics, Karyopharm Therapeutics, Dana Farber Cancer Hospital, Onconova Therapeutics, Shattuck Labs, PharmaMar, Olema Pharmaceuticals, Immunogen, Plexxikon, Amgen, Akeso Biopharma, ADC Therapeutics, AtlasMedx, Aravive, Ellipses Pharma, Incyte, MabSpace Biosciences, ORIC Pharmaceuticals, Pieris Pharmaceuticals, Pionyr, Repertoire Immune Medicines, Treadwell Therapeutics, Jacobio, Accutar Biotech, Artios, Bliss Biopharmaceutical, Cascadian Therapeutics, Cullen-Florentine, Dantari, Duality Biologics, Elucida Oncology, Infinity Pharmaceuticals, Relay Therapeutics, Tolmar, Torque, BeiGene, Context Therapeutics, K-Group Beta, Kind Pharmaceuticals, Loxo, Oncothyreon, Orum Therapeutics, Prelude Therapeutics, and Profound Bio (to institution - research funding). Dr. Santarpia and Dr. Yang are employees of and have equity ownership in Seagen Inc.

Abstract Presentation Number: CT065
American Association for Cancer Research; April 14–19, 2023

Seagen®