SEA-BCMA MONO- AND COMBINATION THERAPY IN PATIENTS WITH RELAPSED/REFRACTORY MULTIPLE MYELOMA: UPDATED RESULTS OF A PHASE 1 STUDY (SGNBCMA-001)

James E Hoffman, MD[1]; Brea Lipe, MD[2]; Jason Melear, MD[3]; Michaela Liedtke, MD[4]; Mark A Schroeder, MD[5]; Ruben Niesvizky, MD[6]; Christopher Strouse, MD[7]; Christopher A Yasenchak, MD[8]; Damian J Green, MD[9]; Jeremy Sauer, PhD[10]; Yinghui Wang, MS[10]; Phoenix Ho, MD[10]; Al-Ola Abdallah, MD[11]

[1]Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL; [2]University of Rochester Medical Center, Wilmot Cancer Institute, Rochester, NY; [3]Texas Oncology, Austin, TX; [4]Stanford University Medical Center, Stanford, CA; [5]Siteman Cancer Center, Washington University School of Medicine, Saint Louis, MO; [6]Weill Cornell Medicine, New York, NY; [7]Holden Comprehensive Cancer Center, University of Iowa, Iowa City, IA; [8]Willamette Valley Cancer Institute and Research Center, Eugene, OR; [9]Fred Hutchinson Cancer Center, Seattle, WA; [10]Seagen Inc., Bothell, WA; [11]University of Kansas Medical Center, Kansas City, KS

Background

- Despite recent progress in the treatment of triple-class R/R (R/R to a proteasome inhibitor, immunomodulatory drug and anti-CD38 antibody) MM, patients remain in need of novel therapies with manageable toxicity and non-cross-resistant mechanisms of action.
- B-cell maturation antigen (BCMA) is expressed in most MM plasma cells.[1]
- SEA-BCMA is an investigational, humanized, nonfucosylated IgG1 monoclonal antibody directed to BCMA.
- Preclinical data for SEA-BCMA demonstrate promising antitumor activity and versatile combinability.[2]
- SGNBCMA-001 Part A was comprised of both monotherapy dose escalation (100–1600 mg flat dosing by intravenous infusion) and dose expansion at the highest tolerated dose. SEA-BCMA 1600 mg Q2W was generally well-tolerated and showed initial antitumor activity with an ORR of 14% (95% CI: 2.9, 34.9; n=22).[3]
- Here, we evaluated intensive SEA-BCMA dosing (Part B) and the addition of dexamethasone (DEX; Part C) to further assess the safety, tolerability, and clinical activity. Preliminary data from Part D, testing the combination of SEA-BCMA, pomalidomide (pom), and DEX in an earlier line of therapy, are also presented.

SEA-BCMA Proposed Mechanism of Action

- A sugar-engineered antibody, the nonfucosylated Fc region of SEA-BCMA has increased affinity for activating FcγRIIIa and minimal affinity for inhibitory FcγRIIb.

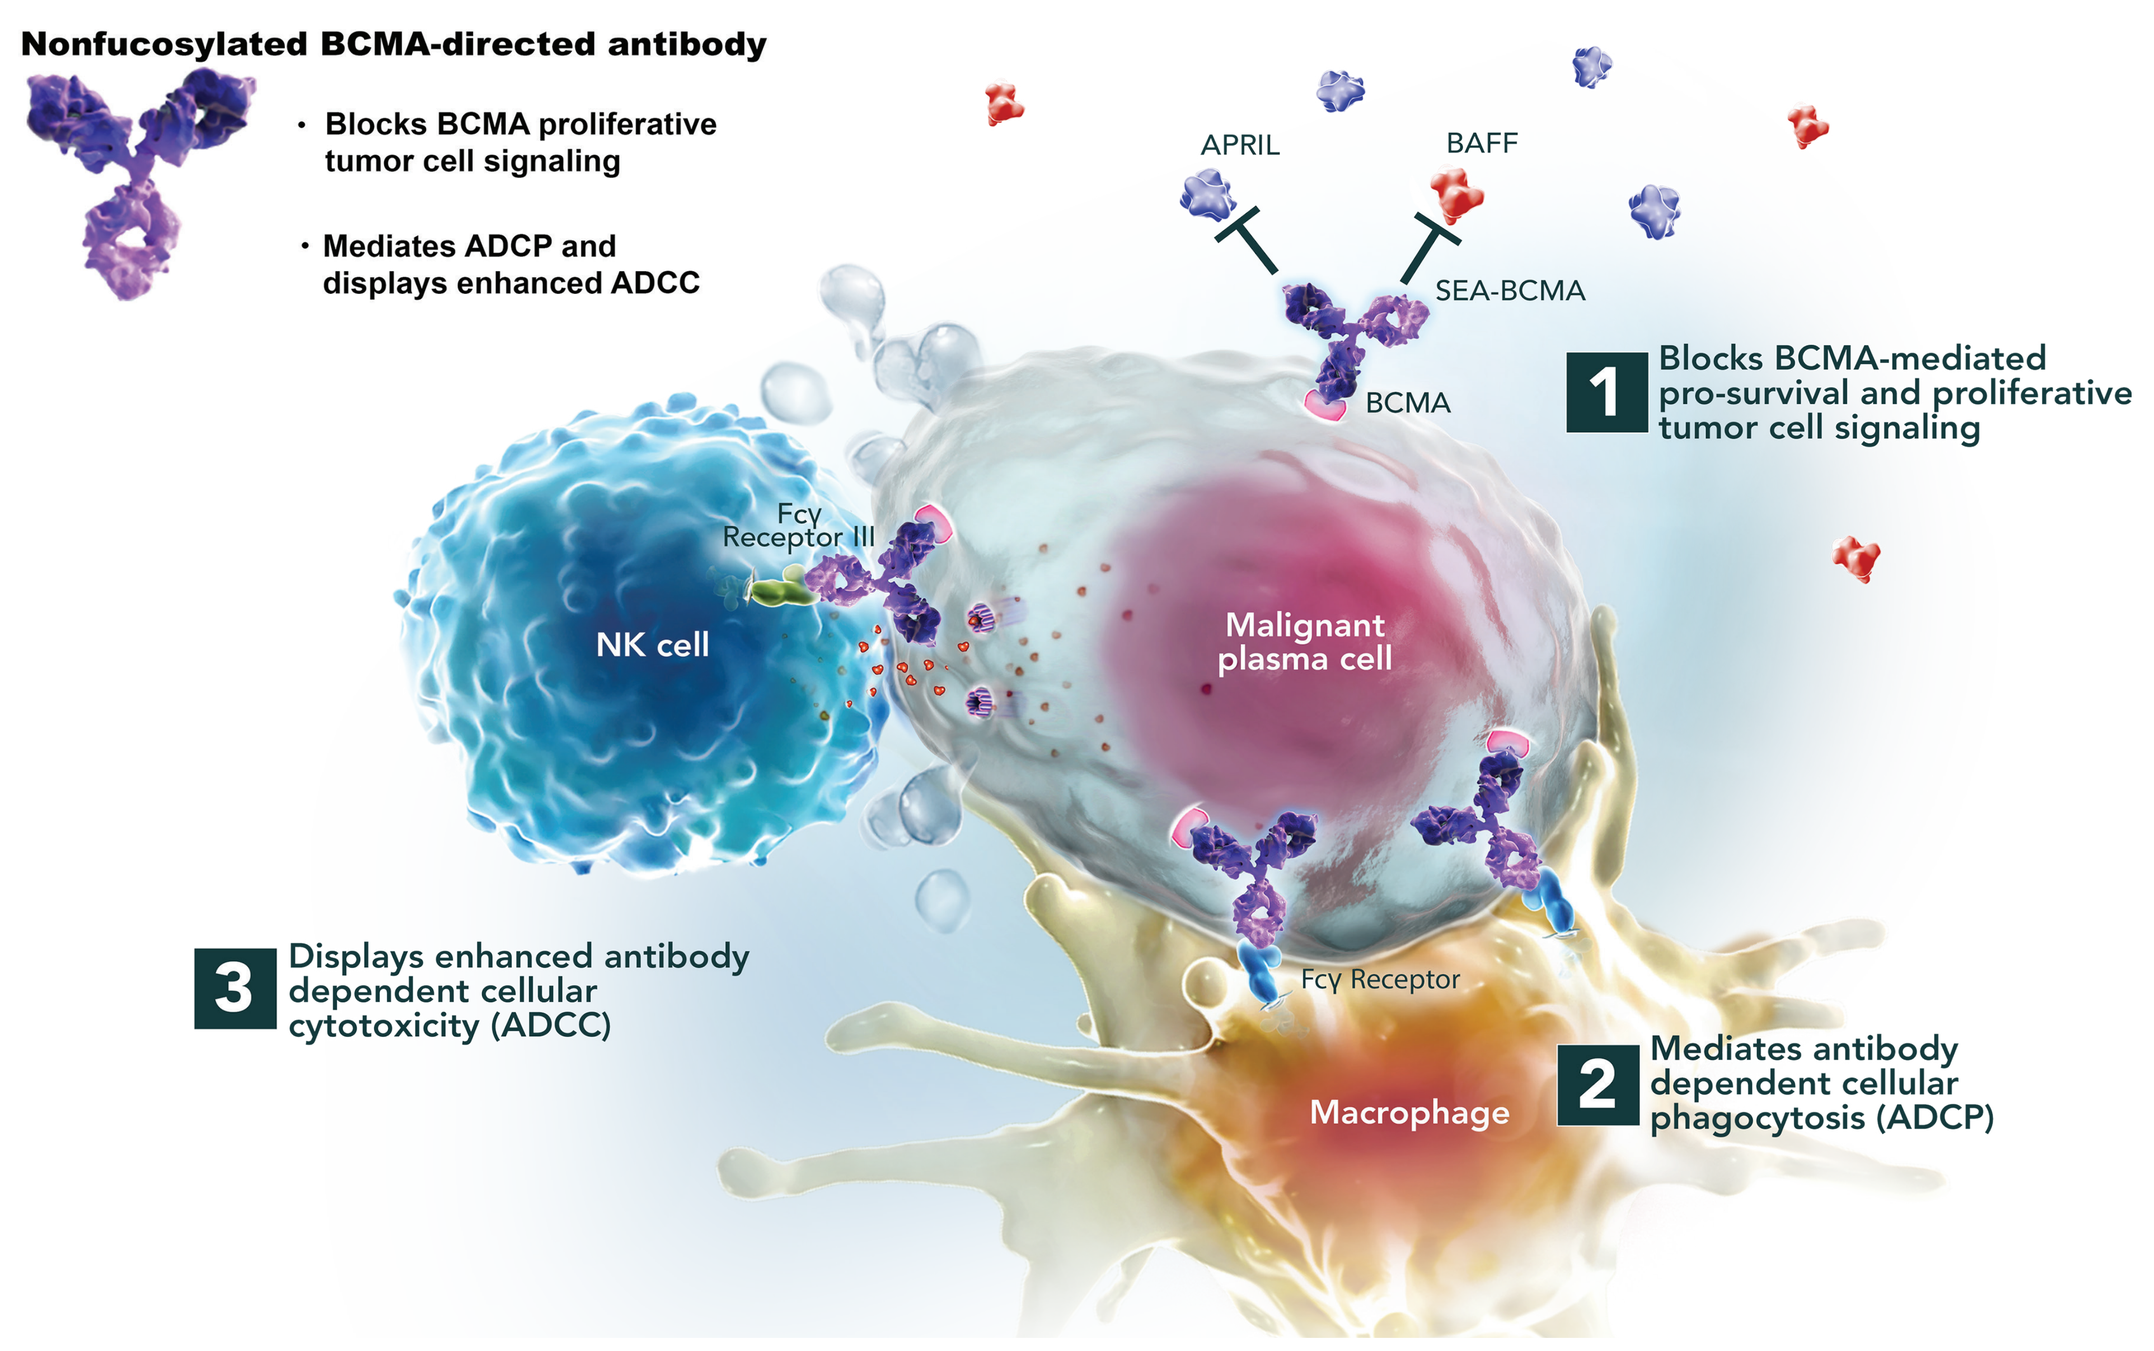
Nonfucosylated BCMA-directed antibody
• Blocks BCMA proliferative tumor cell signaling
• Mediates ADCP and displays enhanced ADCC
APRIL
BAFF
SEA-BCMA
BCMA
1 Blocks BCMA-mediated pro-survival and proliferative tumor cell signaling
Fcγ Receptor III
NK cell
Malignant plasma cell
3 Displays enhanced antibody dependent cellular cytotoxicity (ADCC)
Fcγ Receptor
Macrophage
2 Mediates antibody dependent cellular phagocytosis (ADCP)

SEA-BCMA is an investigational agent, and its safety and efficacy have not been established. ©2022 Seagen Inc. All rights reserved.

Methods

- SGNBCMA-001 (NCT03582033) is an ongoing phase 1, open-label, multicenter, dose escalation and expansion study to evaluate the safety, tolerability and antitumor activity of SEA-BCMA in adults with R/R MM not previously exposed to any other BCMA-directed therapy.
- Parts B and C enrolled patients who have received ≥3 prior lines of therapy for MM and are triple-class refractory. These expansion cohorts are testing
 - Intensive dosing (Part B; weekly induction dosing of SEA-BCMA for 8 weeks is followed by Q2W maintenance dosing) and
 - DEX (40 mg weekly) combination therapy with either standard SEA-BCMA dosing (Part C1) or intensive SEA-BCMA dosing (Part C2; 800 mg [dose level -1] or 1600 mg [dose level 1]).
- Part C2 has been amended to allow patients with prior exposure to BCMA-directed therapy and is continuing to enroll.
- Part D (testing the combination of SEA-BCMA with pom and DEX in patients who have received ≥2 prior lines of therapy) is enrolling.
- Responses are assessed per the 2016 International Myeloma Working Group criteria.

SEA-BCMA Study Schema

Currently Conducting 3 Expansion Cohorts in Parallel

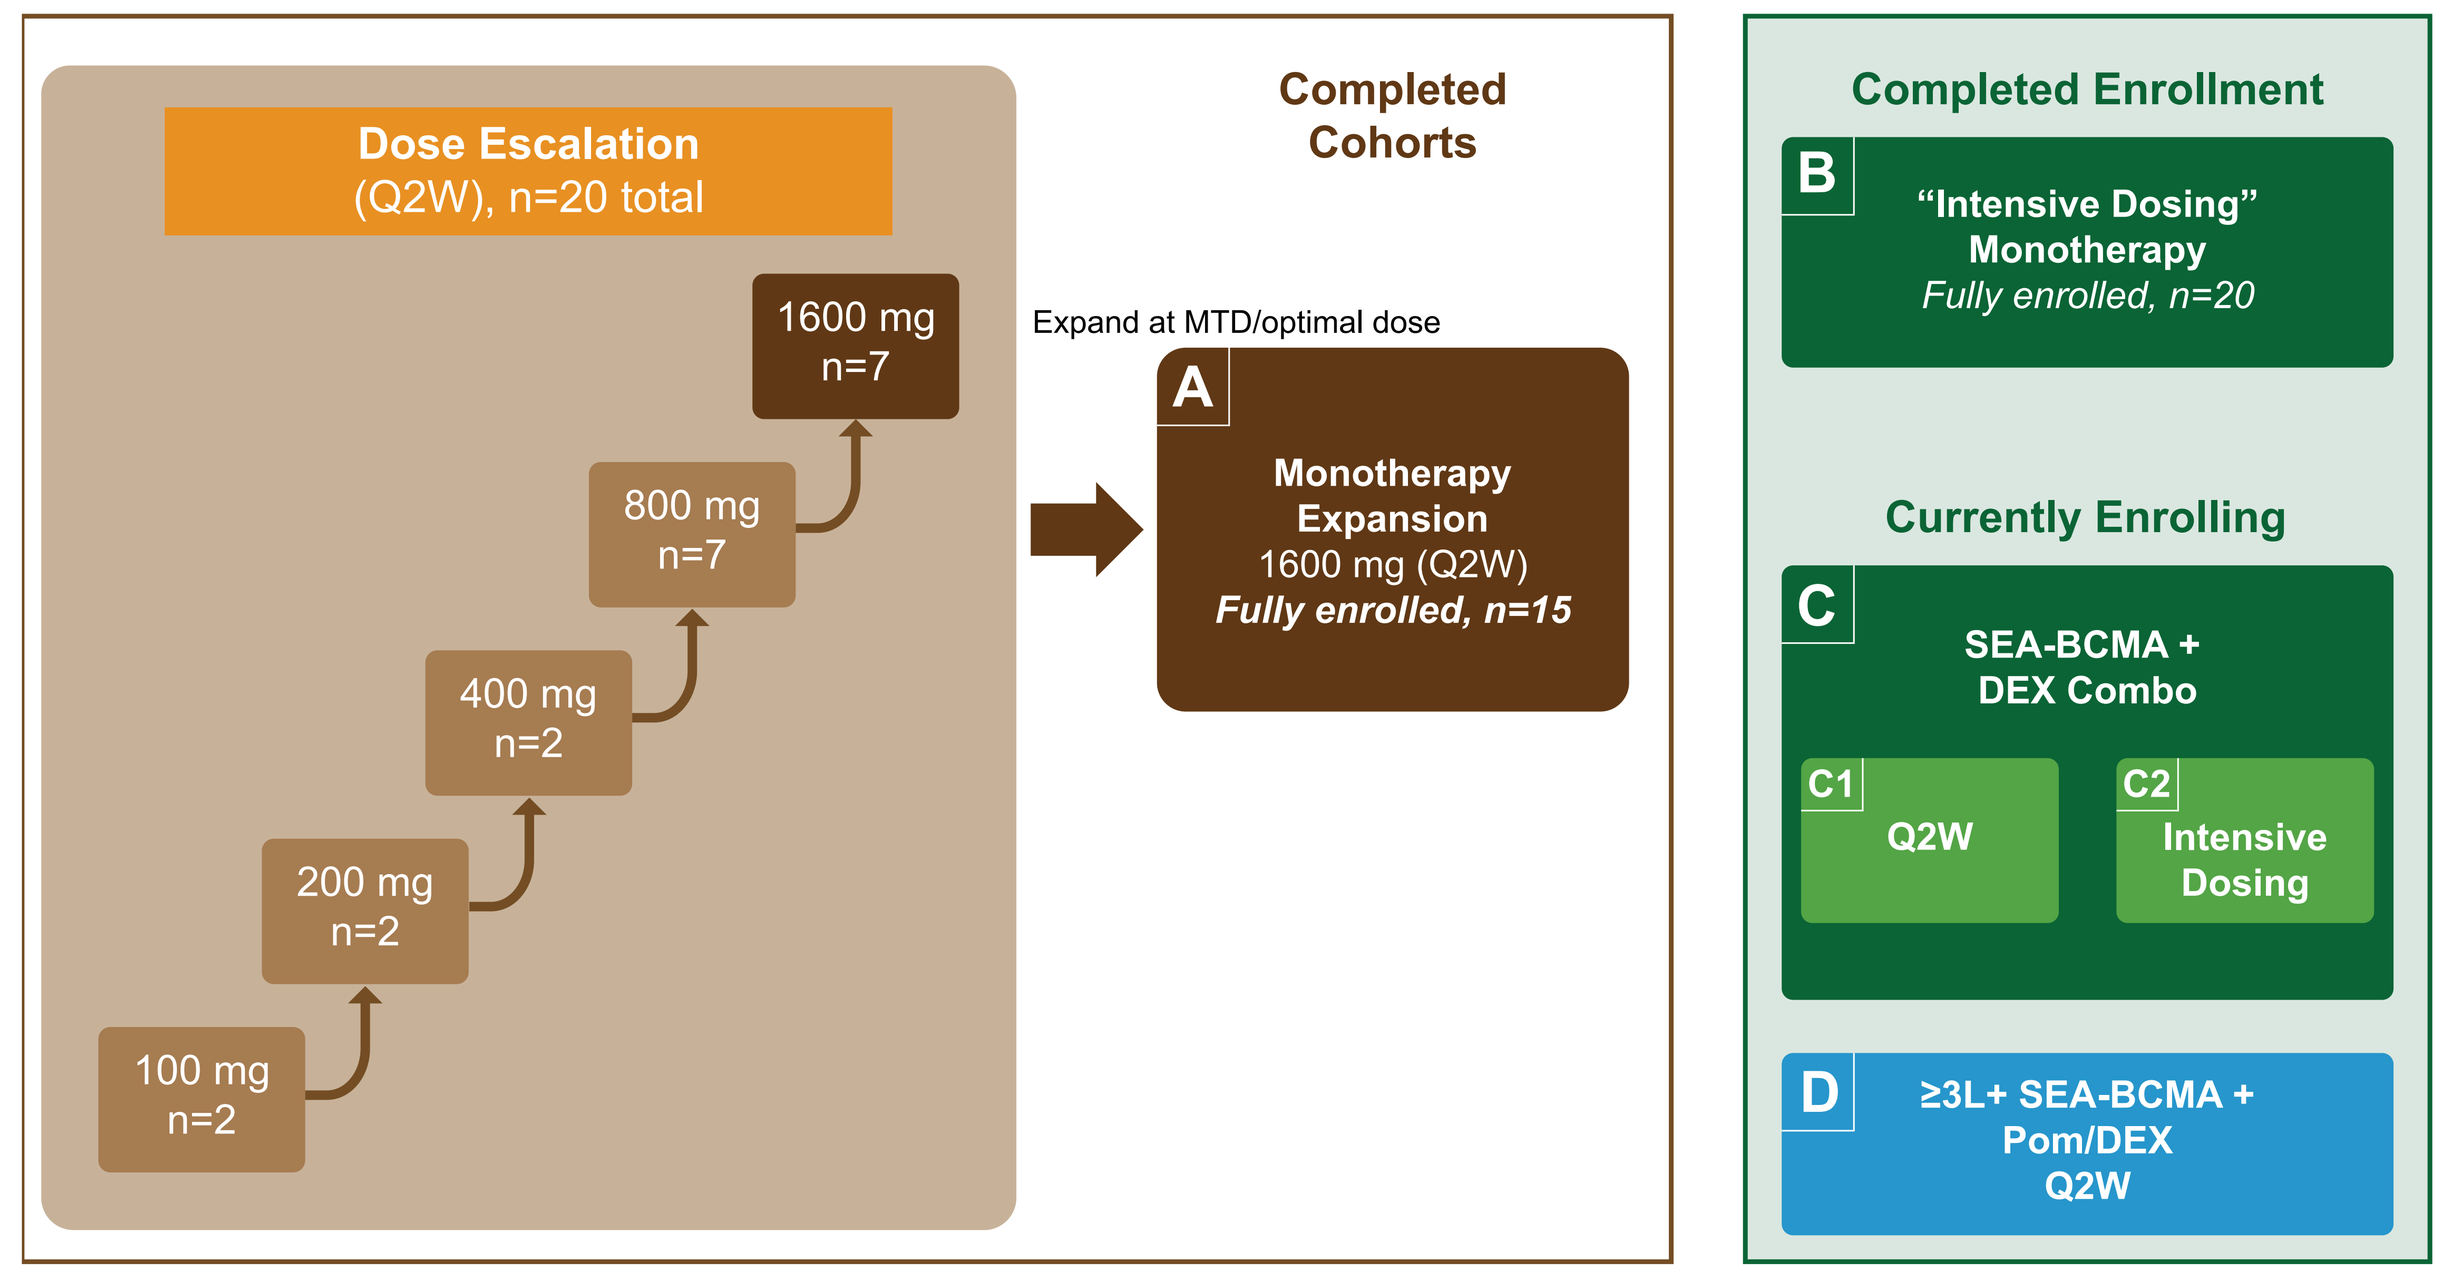
Dose Escalation
(Q2W), n=20 total
100 mg n=2
200 mg n=2
400 mg n=2
800 mg n=7
1600 mg n=7
Completed Cohorts
Expand at MTD/optimal dose
A
Monotherapy Expansion
1600 mg (Q2W)
Fully enrolled, n=15
Completed Enrollment
B
"Intensive Dosing" Monotherapy
Fully enrolled, n=20
Currently Enrolling
C
SEA-BCMA + DEX Combo
C1 Q2W
C2 Intensive Dosing
D
≥3L+ SEA-BCMA + Pom/DEX Q2W

Color Key: brown box=prior lines of therapy must include at least a PI, an IMiD, and an anti-CD38 antibody; green boxes=triple-class refractory cohorts, blue box=≥3L + at least 2 consecutive cycles of both lenalidomide and a PI + pom naïve

- Parts B and C aim to optimize clinical activity in patients with triple-class refractory MM
- Part D aims to assess potential IMiD synergy in earlier line of therapy
- **Updated data from Parts B and C, and preliminary data from Part D are presented therein**

Abbreviations

3L, ≥2 lines of prior therapy; BCMA, B-cell maturation antigen; CI, confidence interval; CR, complete response; DEX, dexamethasone; DLT, dose-limiting toxicity; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; IMiD, immunomodulatory drug; IRR, infusion related reaction; MM, multiple myeloma; MR, minimal response; MTD, maximum tolerated dose; n, number of patients with disease characteristic; OR, overall response; ORR, overall response rate; PD, progressive disease; PI, proteasome inhibitor; pom, pomalidomide; PR, partial response; Q2W, once every 2 weeks; R/R, relapsed/refractory; SAE, serious adverse event; SD, stable disease; sFLC, serum free light chain; TEAE, treatment emergent adverse event; triple-class R/R, R/R to a proteasome inhibitor, immunomodulatory drug and anti-CD38 antibody; VGPR, very good partial response

References

1. Seckinger A, Delgado JA, Moser S, et al. Cancer Cell. 2017;31(3):396-410
2. Van Epps H, Anderson M, Yu C, et al. Cancer Res July 1 2018 (78) (13 Supplement) 3833; DOI: 10.1158/1538-7445.AM2018-3833
3. Hoffman JE, Lipe B, Melear J, et al.; Blood 2021; 138 (Supplement 1): 2740. doi: https://doi.org/10.1182/blood-2021-146047

Disclosures

This study is sponsored by Seagen Inc., Bothell, WA, USA. The institutions of JEH, BL, JM, ML, MAS, RN, CS, CAY, DJG, and AA received research funding from Seagen Inc. JS, YW and PH are employees of and hold ownership interest in Seagen Inc. Other disclosures: JEH BMS, Celgene, Seagen Inc.; BL: Janssen, BMS, Sanofi, GSK, Seagen Inc., Amgen, Harpoon; JM AstraZeneca, Janssen, TG Therapeutics; ML Adaptive, Allogene, Alnylam, BMS, Caelum, Gilead, GSK, Janssen, Karyopharm, Kite, Kura Oncology, Natera, Oncopeptides, Sanofi, Seagen Inc., Takeda; MAS: Incyte, Genentech Inc, Cellect Inc, Fortis, Seagen Inc., Amgen, Celgene; RN BMS, GSK, Janssen, Karyopharm, Takeda; CS nothing to disclose; CAY Beigene, Seagen Inc., Takeda. DJG Seagen Inc.; AA nothing to disclose.

Results

Demographics and Disease Characteristics

Patient Characteristic	Part A (N=35)	Part B (N=20)	Part C1 (N=12)	Part C2 (N=8)	Part D (N=3)
Age (years)					
Median (Min–Max)	71.0 (51–84)	74.0 (57–91)	68.5 (48–76)	72.5 (68–83)	75.0 (68–76)
Age, n (%)					
≥65	28 (80)	18 (90)	9 (75)	8 (100)	3 (100)
Gender, n (%)					
Female	16 (46)	7 (35)	7 (58)	2 (25)	1 (33)
Male	19 (54)	13 (65)	5 (42)	6 (75)	2 (67)
ECOG Performance Status, n (%; Eligibility: 0–1)					
1	32 (91)	17 (85)	9 (75)	7 (88)	2 (67)
Number of prior systemic therapies for myeloma					
Median (Min–Max)	6.0 (3–15)	4.5 (3–10)	5.0 (3–10)	5.0 (3–6)	4.0 (2–5)
Refractory, n (%)					
To PI	25 (71)	19 (95)	12 (100)	8 (100)	3 (100)
To IMiD	26 (74)	19 (95)	12 (100)	8 (100)	2 (67)
To anti-CD38 antibody	26 (74)	20 (100)	12 (100)	8 (100)	2 (67)
Triple class refractory	23 (66)	19 (95)	12 (100)	8 (100)	1 (33)
Intolerant of any above classes	9 (26)	4 (20)	4 (33)	2 (25)	1 (33)
Extramedullary disease, n (%)					
Presence of bone lesions indicated by skeletal survey	23 (66)	9 (45)	7 (58)	7 (88)	–
Soft tissue plasmacytomas identified by radiography	5 (14)	–	–	1 (13)	–

Note: Parts A and D enrolled triple-class exposed, while Parts B and C required triple-class refractory.

Updated Safety Data Show SEA-BCMA was Generally Well-Tolerated

Parts B (Intensive Dosing), C1 (Combination Therapy with DEX), and C2 (Intensive Dosing Combination Therapy with DEX) Show Similar Tolerability with no DLTs Observed

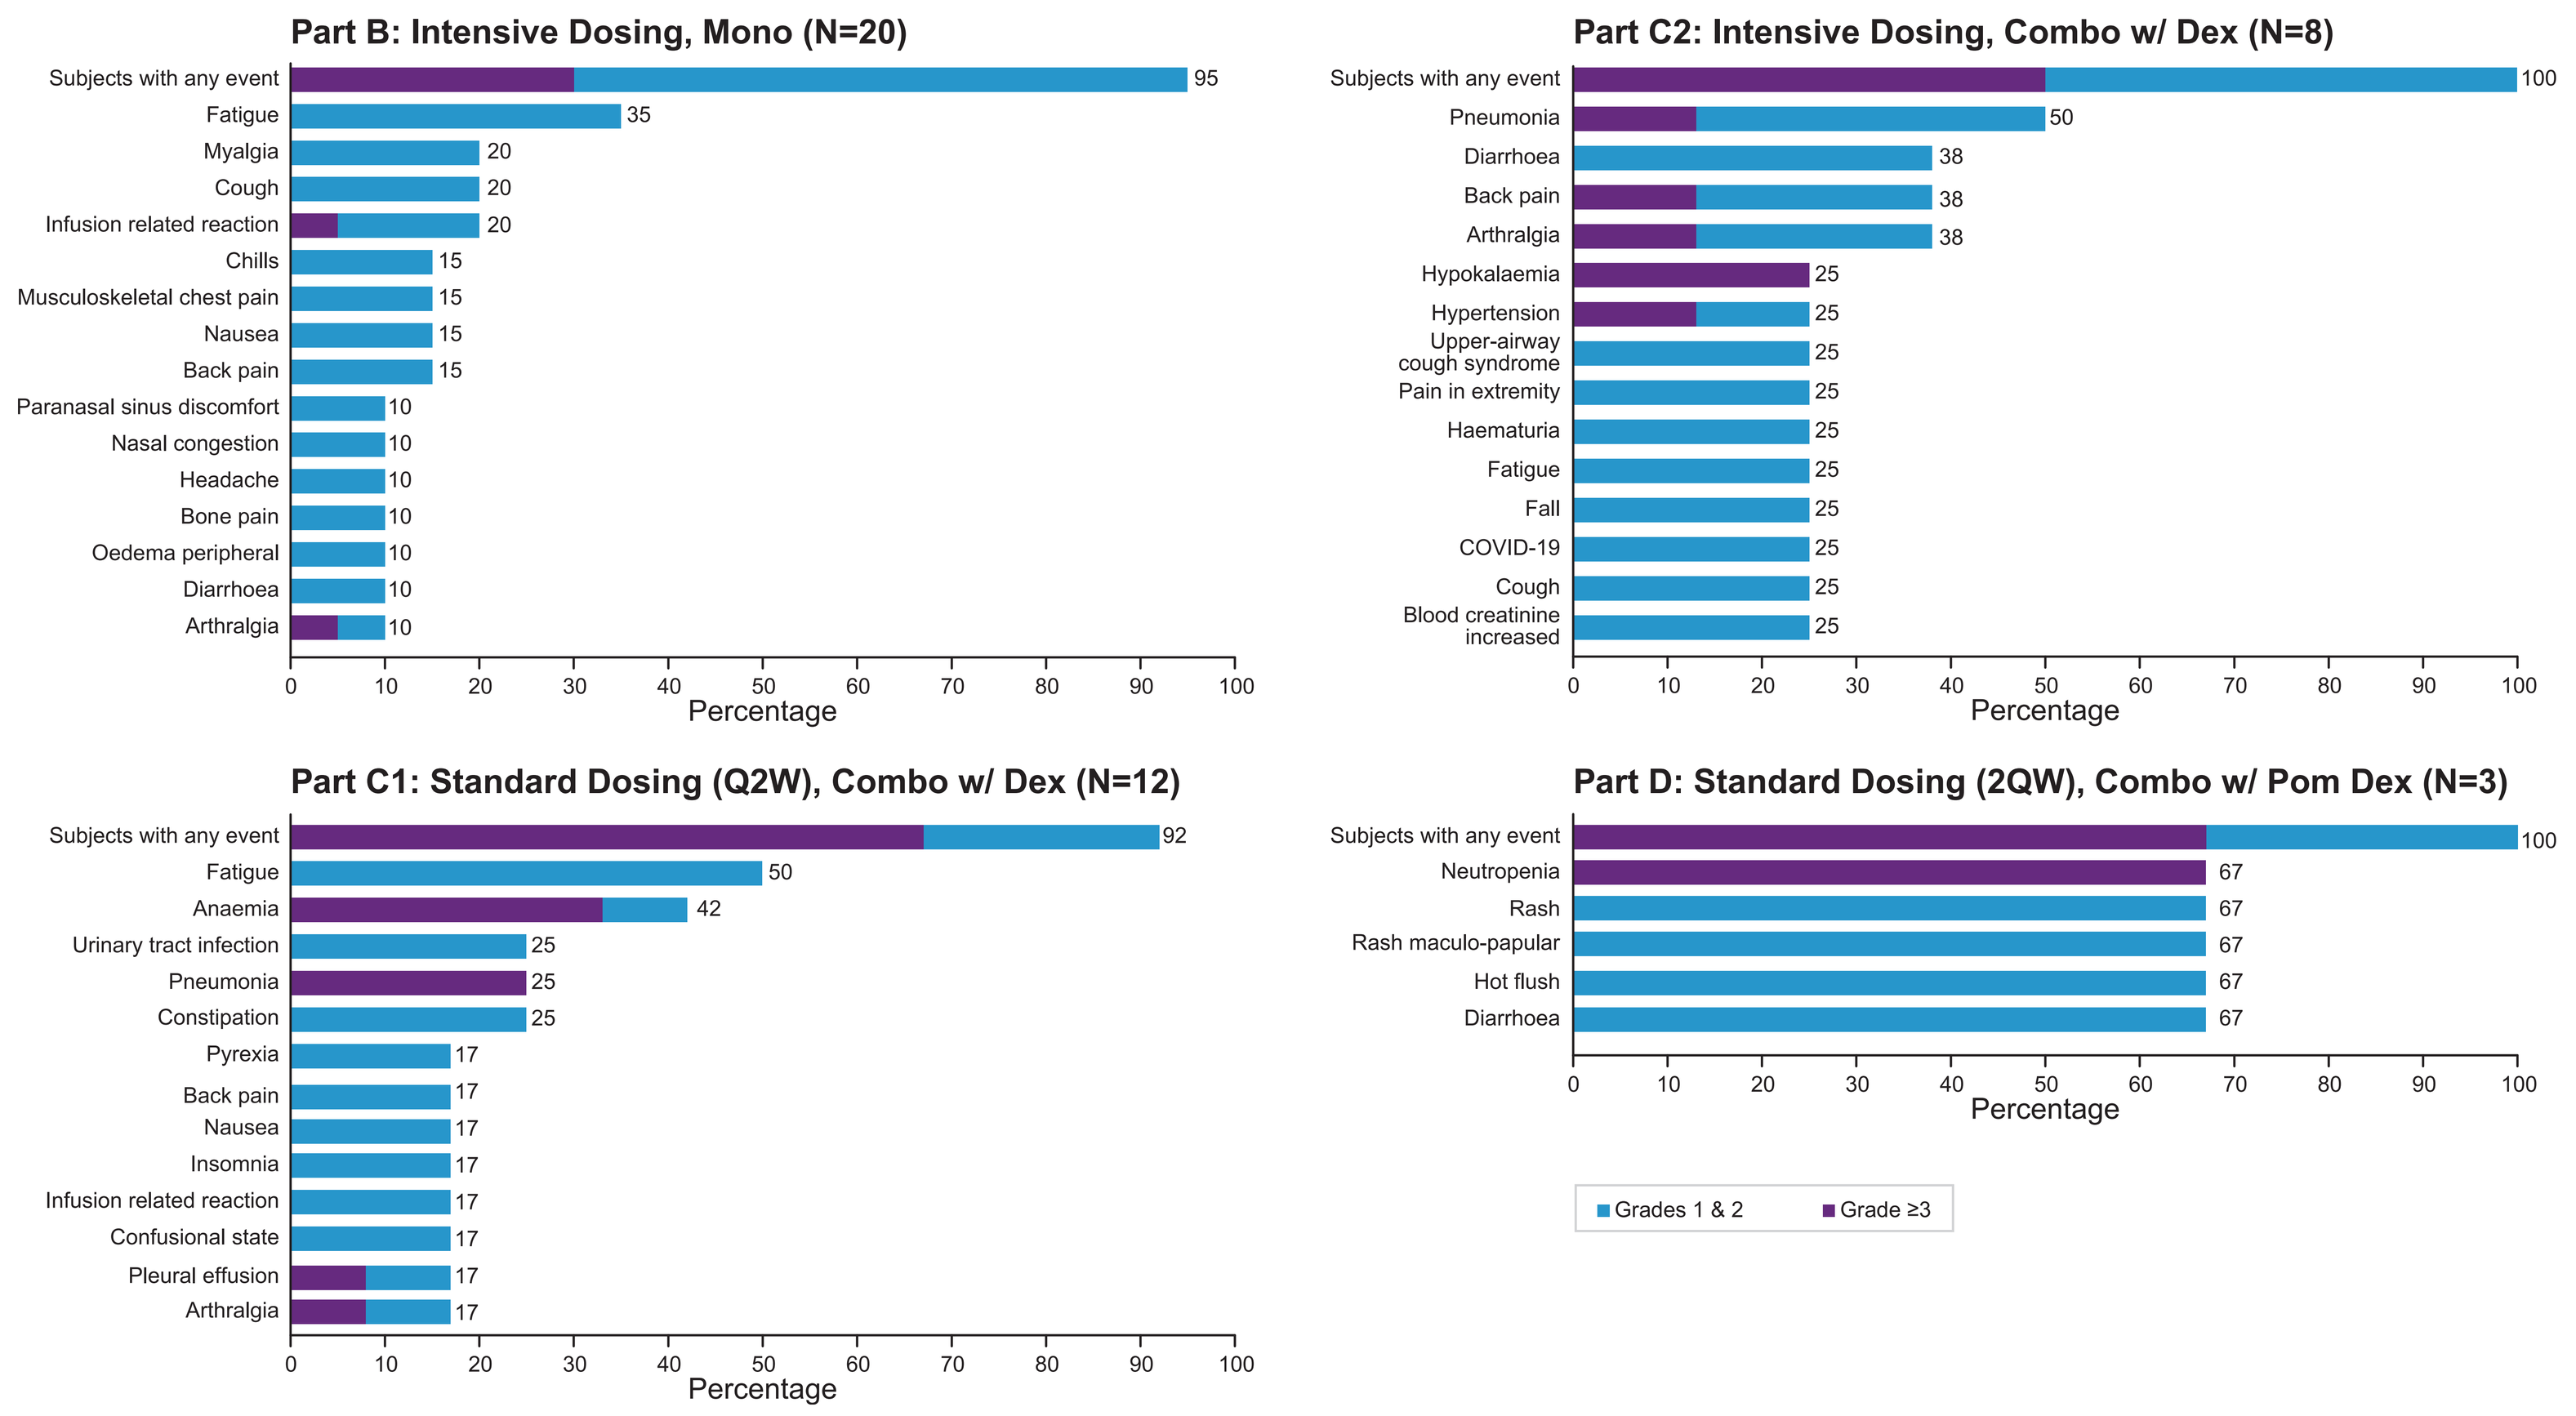
Part B: Intensive Dosing, Mono (N=20)
Subjects with any event 95
Fatigue 35
Myalgia 20
Cough 20
Infusion related reaction 20
Chills 15
Musculoskeletal chest pain 15
Nausea 15
Back pain 15
Paranasal sinus discomfort 10
Nasal congestion 10
Headache 10
Bone pain 10
Oedema peripheral 10
Diarrhoea 10
Arthralgia 10
Part C2: Intensive Dosing, Combo w/ Dex (N=8)
Subjects with any event 100
Pneumonia 50
Diarrhoea 38
Back pain 38
Arthralgia 38
Hypokalaemia 25
Hypertension 25
Upper-airway cough syndrome 25
Pain in extremity 25
Haematuria 25
Fatigue 25
Fall 25
COVID-19 25
Cough 25
Blood creatinine increased 25
Part C1: Standard Dosing (Q2W), Combo w/ Dex (N=12)
Subjects with any event 92
Fatigue 50
Anaemia 42
Urinary tract infection 25
Pneumonia 25
Constipation 25
Pyrexia 17
Back pain 17
Nausea 17
Insomnia 17
Infusion related reaction 17
Confusional state 17
Pleural effusion 17
Arthralgia 17
Part D: Standard Dosing (2QW), Combo w/ Pom Dex (N=3)
Subjects with any event 100
Neutropenia 67
Rash 67
Rash maculo-papular 67
Hot flush 67
Diarrhoea 67
Percentage
Grades 1 & 2
Grade ≥3

- In Part A SEA-BCMA was generally well-tolerated with a single DLT (prolonged IRR) observed, and the MTD was not identified in the 5 dose levels tested.[3]
- Part B completed the safety run-in (n=6) and completed enrollment at 1600 mg SEA-BCMA intensive dosing.
- Part C1 completed the safety run-in (n=6) and continued enrollment for SEA-BCMA 1600 mg Q2W + DEX.
- Part C2 dose optimization is completing safety run-ins at 800 mg SEA-BCMA intensive dosing (dose level –1) and at 1600 mg SEA-BCMA intensive dosing (dose level 1).
- No DLTs were observed in Part B, the safety run-in portions of either Part C1 or C2 thus far, and among the first 3 patients in Part D; safety and tolerability were similar to that observed in Part A.
- No patients in Parts B, C2, and D discontinued treatment due to TEAEs. In Part C1, 1 patient discontinued treatment due to a TEAE unrelated to SEA-BCMA. The majority of treatment discontinuations were due to PD.
- No deaths related to SEA-BCMA occurred during the study to date; all deaths within 30 days of SEA-BCMA dosing were due to myeloma.

Preliminary Results for SEA-BCMA Antitumor Activity and DOR

SEA-BCMA Demonstrated Single Agent Activity and Response Rates Increased with Intensive Dosing and Combination Therapy

	Standard Q2W Dosing SEA-BCMA 1600 mg		Intensive Dosing (Weekly x 2 Cycles, Then Q2W) SEA-BCMA		
	Part A Monotherapy[a] (N=22)	Part C1 + DEX (N=12)	Part B Monotherapy (N=20)	Part C2 + DEX (N=8)	Part D Q2W + pom/DEX (N=3)
Best overall response (confirmed), n (%)					
Stringent complete response	0	1 (8)	1 (5)	0	2 (67)
Complete response	0	0	0	0	0
Very good partial response	1 (5)	0	0	1 (13)	0
Partial response	2 (9)	1 (8)	3 (15)	2 (25)	1 (33)
Minimal response	1 (5)	1 (8)	1 (5)	0	0
Stable disease	13 (59)	7 (58)	11 (55)	5 (63)	0
Progressive disease	5 (23)	2 (17)	4 (20)	0	0
Overall response rate (ORR), n (%)	3 (14)	2 (17)	4 (20)	3 (38)	3 (100)
95% CI[b] for ORR	(2.9, 34.9)	(2.1, 48.4)	(5.7, 43.7)	(8.5, 75.5)	(29.2, 100)
ORR by dosing schedule, n (%)	**5 (15)**		**7 (25)**		

a Escalation and Expansion Phase dosed at 1600 mg.
b Two-sided 95% exact confidence interval, computed using the Clopper-Pearson method (1934).

Duration of Response (Overall)

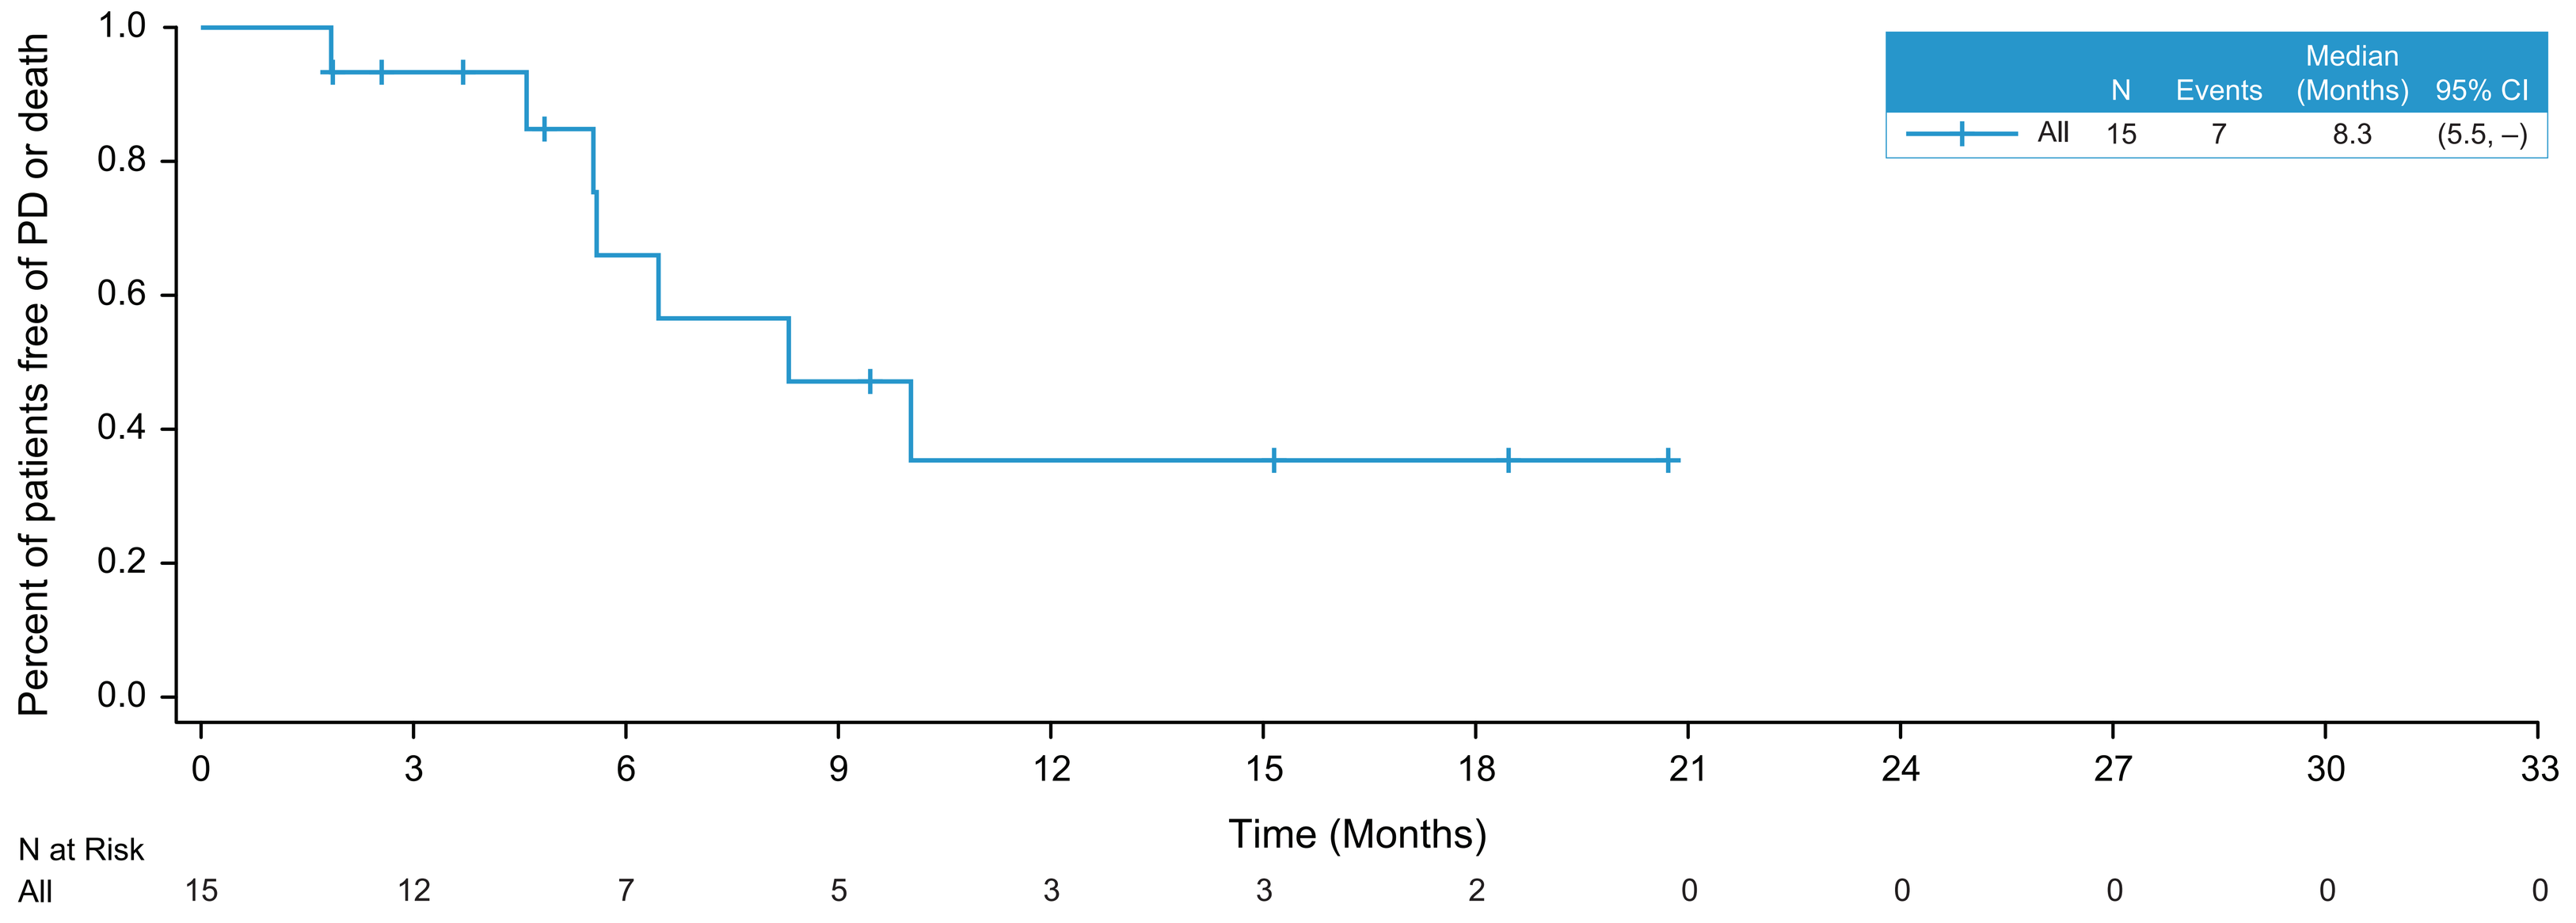
Percent of patients free of PD or death
1.0
0.8
0.6
0.4
0.2
0.0
0 3 6 9 12 15 18 21 24 27 30 33
Time (Months)
N at Risk
All 15 12 7 5 3 3 2 0 0 0 0 0
N Events Median (Months) 95% CI
All 15 7 8.3 (5.5, –)

- Both intensive SEA-BCMA dosing (Part B) and the addition of DEX (Part C1) showed increased ORR relative to Q2W monotherapy (Part A).
- For the combination of the intensive SEA-BCMA dosing schedule and the addition of DEX (Part C2), the ORR across both SEA-BCMA dose levels (800 mg [n=3] and 1600 mg [n=5]) was 38% (n=8; 95% CI: 8.5, 75.5).
- The combined ORR of both intensive dosing cohorts Parts B and C2 relative to standard dosing (Q2W) cohorts Parts A [1600 mg SEA-BCMA] and C1 were 25% versus 15%, respectively.
- The overall median DOR from 15 patients with a confirmed response across Parts A (1600 mg SEA-BCMA), B, C1, C2, and D was 8.3 months (95% CI: 5.5, –).
- In Part A, the median DOR was 10 months (95% CI: 5.6, –), 1 patient remains on treatment. In Part B, the median DOR was 6.5 months (95% CI: 1.8, –); 4 patients remain on treatment, including 2 responders. In Part D, the median DOR was 8.3 months (95% CI: –, –); 2 responding patients remain on treatment.
- In Parts C1 and C2, the median DOR was not yet reached (1 responding patient remains on treatment in Part C1, and 2 responding patients remain on treatment in Part C2).

Conclusions

- Updated results for SEA-BCMA continue to demonstrate a favorable safety profile and combination potential.
- Encouraging preliminary data suggest that both the intensive dosing schedule and combination with DEX may increase clinical activity over Q2W monotherapy.
- Part C2 and Part D (testing the combination of SEA-BCMA with pomalidomide and DEX), are enrolling.
- SEA-BCMA shows potential to provide R/R MM patients with the option of a novel therapy with manageable toxicity and combinability in regimens with other classes of myeloma therapies.

Abstract No. 4265
ASH Annual Meeting & Exposition; New Orleans, Louisiana, December 10–13, 2022

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the author, **James E Hoffman, j.hoffman4@med.miami.edu**