ASCO Genitourinary Cancer Symposium
San Francisco, CA
February 13–15, 2020
Abstract No. TPS595

Study EV-103: New Cohorts Testing Enfortumab Vedotin Alone or in Combination with Pembrolizumab in Muscle Invasive Bladder Cancer

Christopher J. Hoimes[1], Jonathan E. Rosenberg[2], Daniel P. Petrylak[3], Anne-Sophie Carret[4], Carolyn Sasse[5], Marya Chaney[6], Thomas W. Flaig[7]

[1]Case Western Reserve University/University Hospitals Seidman Center, Cleveland, OH; [2]Memorial Sloan Kettering Cancer Center, New York, NY; [3]Yale Cancer Center, New Haven, CT; [4]Seattle Genetics, Inc., Bothell, WA; [5]Astellas Pharma, Inc., Northbrook, IL; [6]Merck & Co., Inc., Kenilworth, NJ, USA; [7]University of Colorado Comprehensive Cancer Center, Aurora, CO

Current Treatments for Muscle Invasive Bladder Cancer

- Up to 25% of patients diagnosed with urothelial cancers present with muscle invasive bladder cancer (MIBC)[1]
- Cisplatin-based chemotherapy is the standard of care for the neoadjuvant cisplatin-eligible MIBC population
- Many patients (>40%)[2] are cisplatin-ineligible for a variety of reasons, such as impaired renal function
- Radical cystectomy (RC) with bilateral lymph node dissection (PLND) is the standard of care for the neoadjuvant *cisplatin-ineligible* MIBC population
- Programmed cell death protein 1/programmed death-ligand 1 (PD-1/PD-L1) inhibitors are not approved for MIBC in the neoadjuvant setting
- There are no other treatments approved for *cisplatin-ineligible* patients

Enfortumab Vedotin is an Investigational Antibody-Drug Conjugate Targeting Nectin-4

- Enfortumab vedotin consists of a fully human monoclonal antibody targeting Nectin-4 and the microtubule-disrupting agent monomethyl auristatin E (MMAE), conjugated by a protease-cleavable linker
- Nectin-4 is a transmembrane cell adhesion molecule[3] that is expressed in multiple tumor types, including urothelial cancer[4]
- Nectin-4 was expressed in all 120 samples tested from patients with locally advanced or metastatic urothelial cancer (la/mUC) in the ongoing phase 2 study (EV-201; NCT3219333)[5] of enfortumab vedotin monotherapy; expression was uniformly high[5]

Figure 1: Enfortumab Vedotin Proposed Mechanism of Action

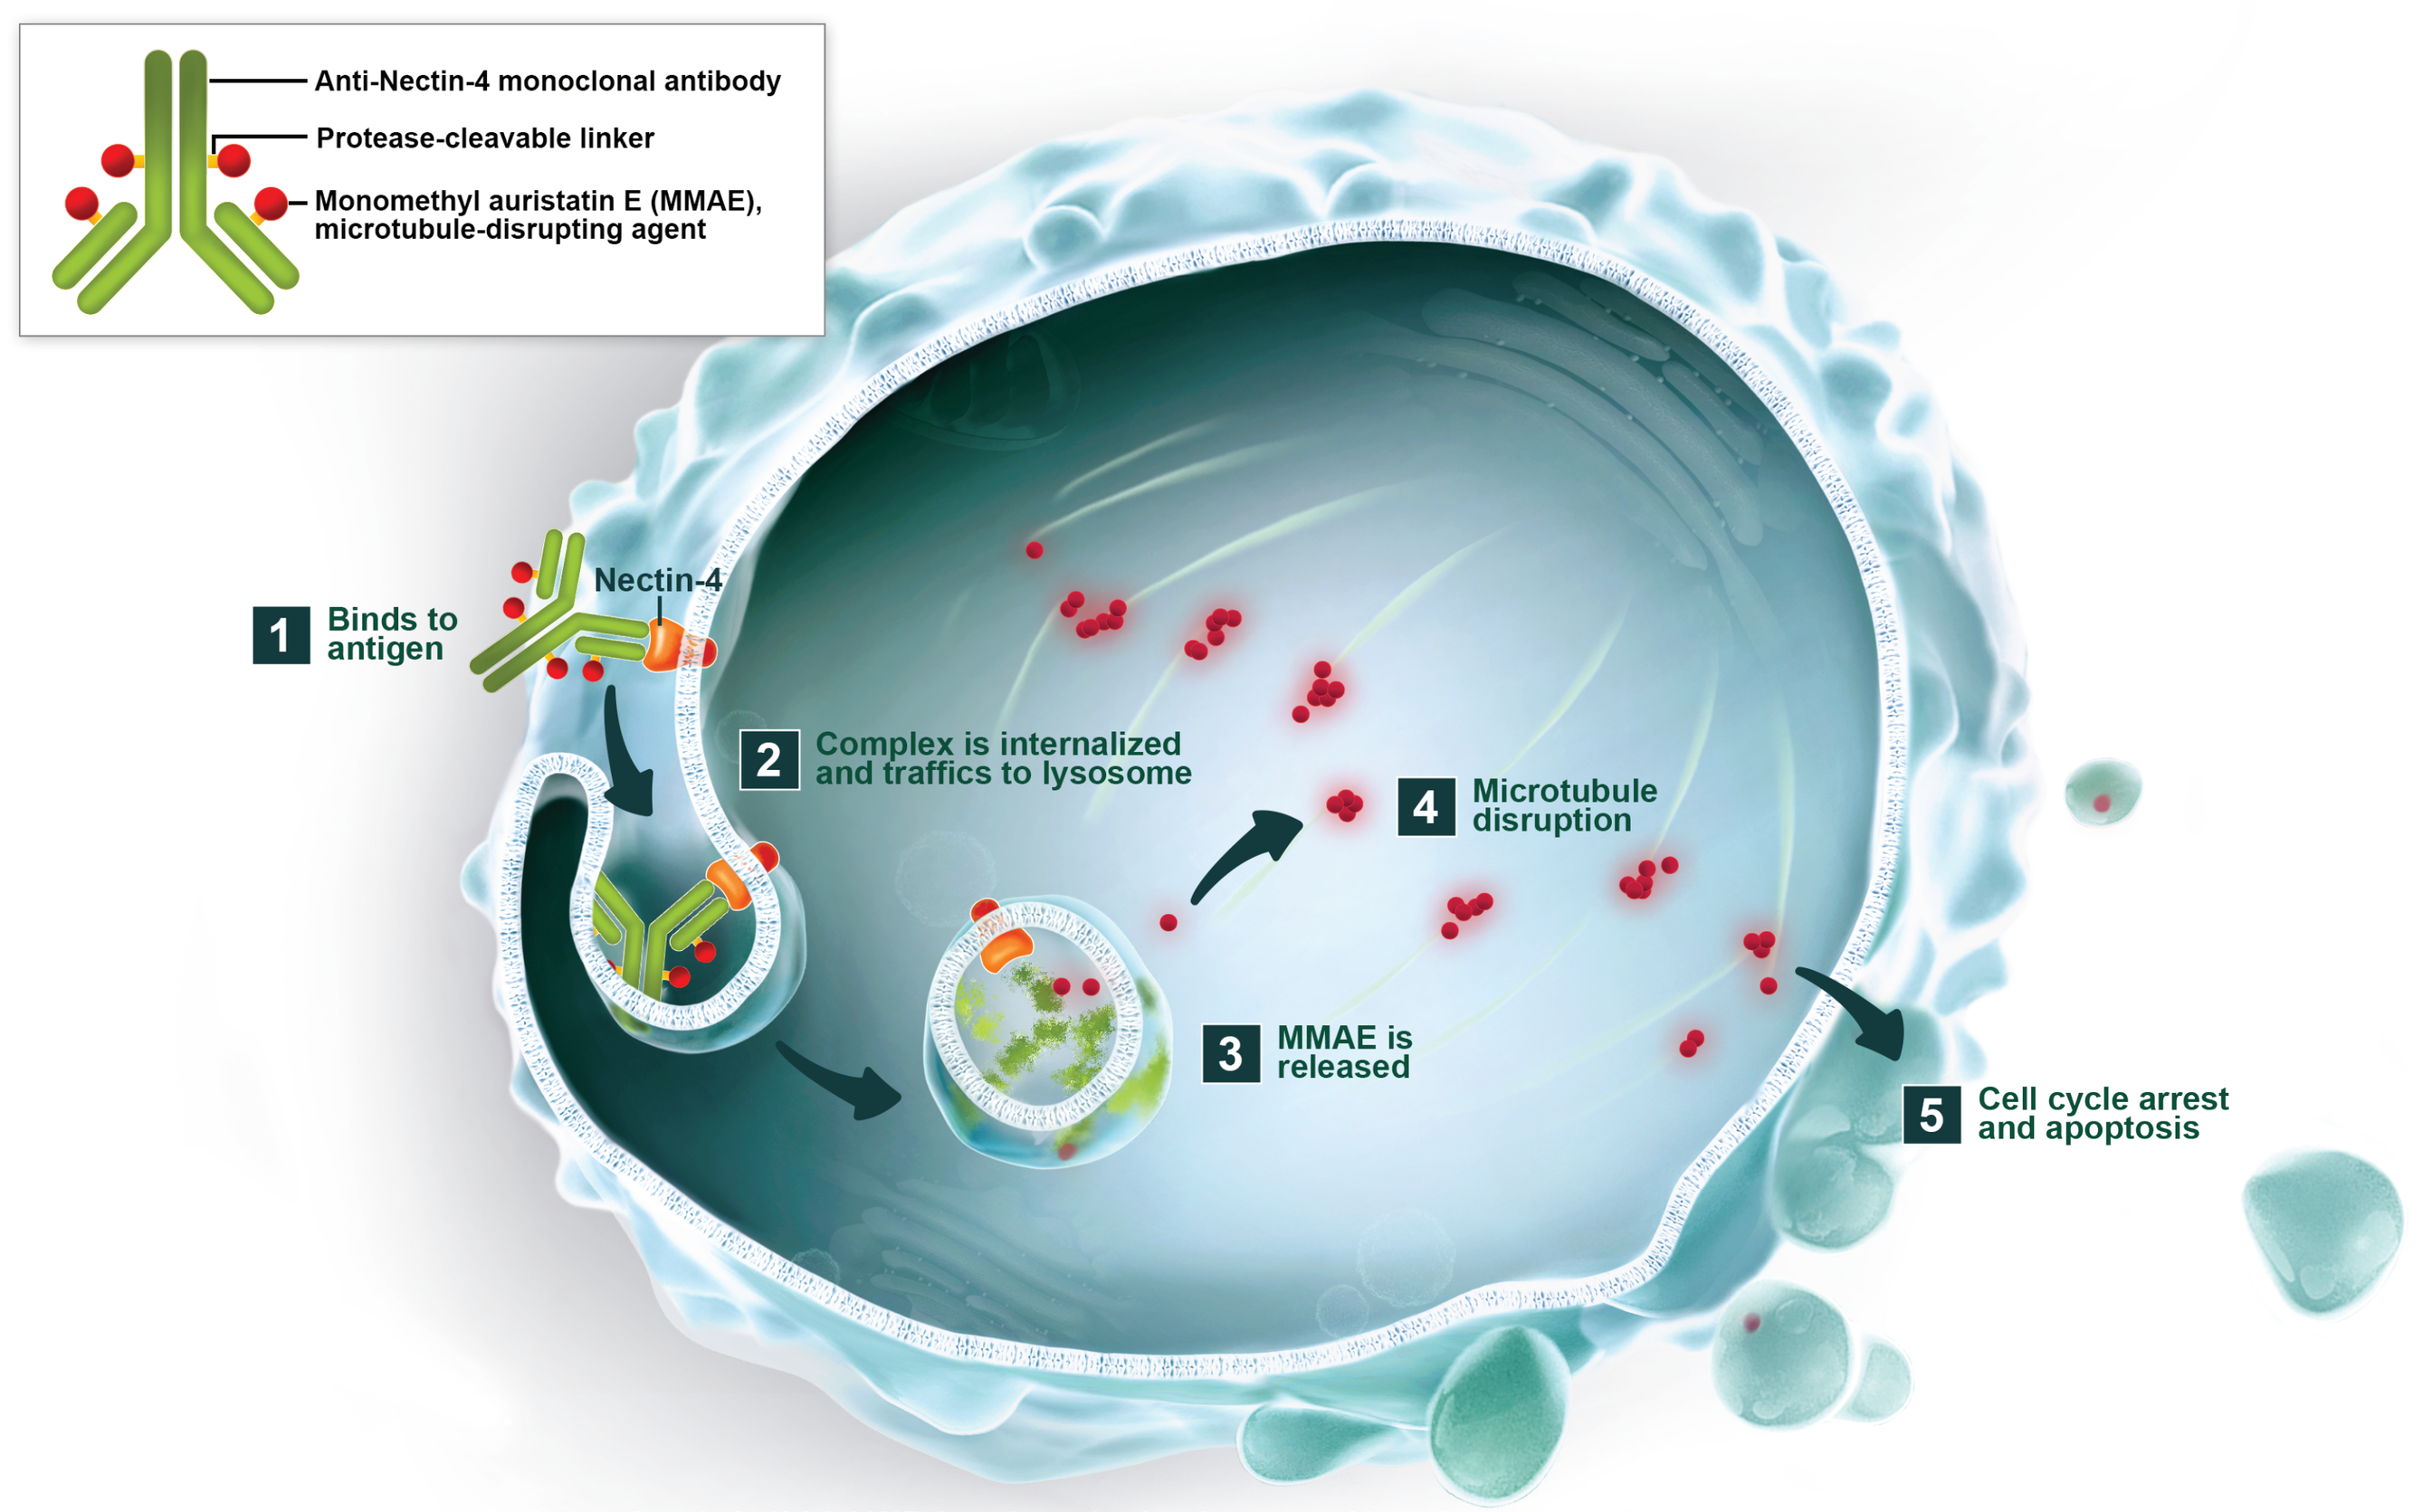
Anti-Nectin-4 monoclonal antibody
Protease-cleavable linker
Monomethyl auristatin E (MMAE), microtubule-disrupting agent
Nectin-4
1 Binds to antigen
2 Complex is internalized and traffics to lysosome
3 MMAE is released
4 Microtubule disruption
5 Cell cycle arrest and apoptosis

Enfortumab vedotin (ASG-22ME) is an investigational agent, and its safety and efficacy have not been established.
Enfortumab vedotin is being developed in collaboration with Astellas Pharma Inc. ©2020 Seattle Genetics, Inc. All rights reserved.

Enfortumab Vedotin Monotherapy in Locally Advanced and Metastatic Urothelial Carcinoma

- Study EV-201 (NCT03288545) evaluated enfortumab vedotin monotherapy (1.25 mg/kg) in adults with la/mUC who previously received a PD-1 or PD-L1 inhibitor and a platinum-containing chemotherpay in the neoadjuvant/adjuvant, la/mUC setting
- 44% ORR in these heavily pre-treated patients (n=125)[5]

Enfortumab Vedotin Combined with Pembrolizumab in Locally Advanced and Metastatic Urothelial Carcinoma

- In the ongoing phase 1b/2 study EV-103 (enfortumab vedotin as monotherapy or in combination with other anticancer therapies for the treatment of bladder cancer):
 - Interim results from cisplatin-ineligible patients with la/mUC treated with enfortumab vedotin 1.25 mg/kg + pembrolizumab 200 mgin the 1L setting show an ORR of 73.3%, a manageable safety profile and no new safety signals have been identified
- Enfortumab vedotin + pembrolizumab may also be suitable for research in MIBC

Updated results from 1L patients with locally advanced or metastatic urothelial cancer treated with enfortumab vedotin + pembrolizumab presented on Friday, February 14, Poster Board A4/Abstract 441

Figure 2: Rationale for Combining Enfortumab Vedotin + Pembrolizumab

- Enfortumab vedotin and pembrolizumab each have single agent activity in la/mUC
- Preclinical studies show that antibody-drug conjugates (brentuximab vedotin, ladiratuzumab vedotin, and tisotumab vedotin)[6-8] linked to MMAE induce immunogenic cell death and may enhance anti-tumor immunity
- Clinical data suggests the combination of enfortumab vedotin + pembrolizumab may have the potential to induce greater antitumor activity in la/mUC compared to either agent alone

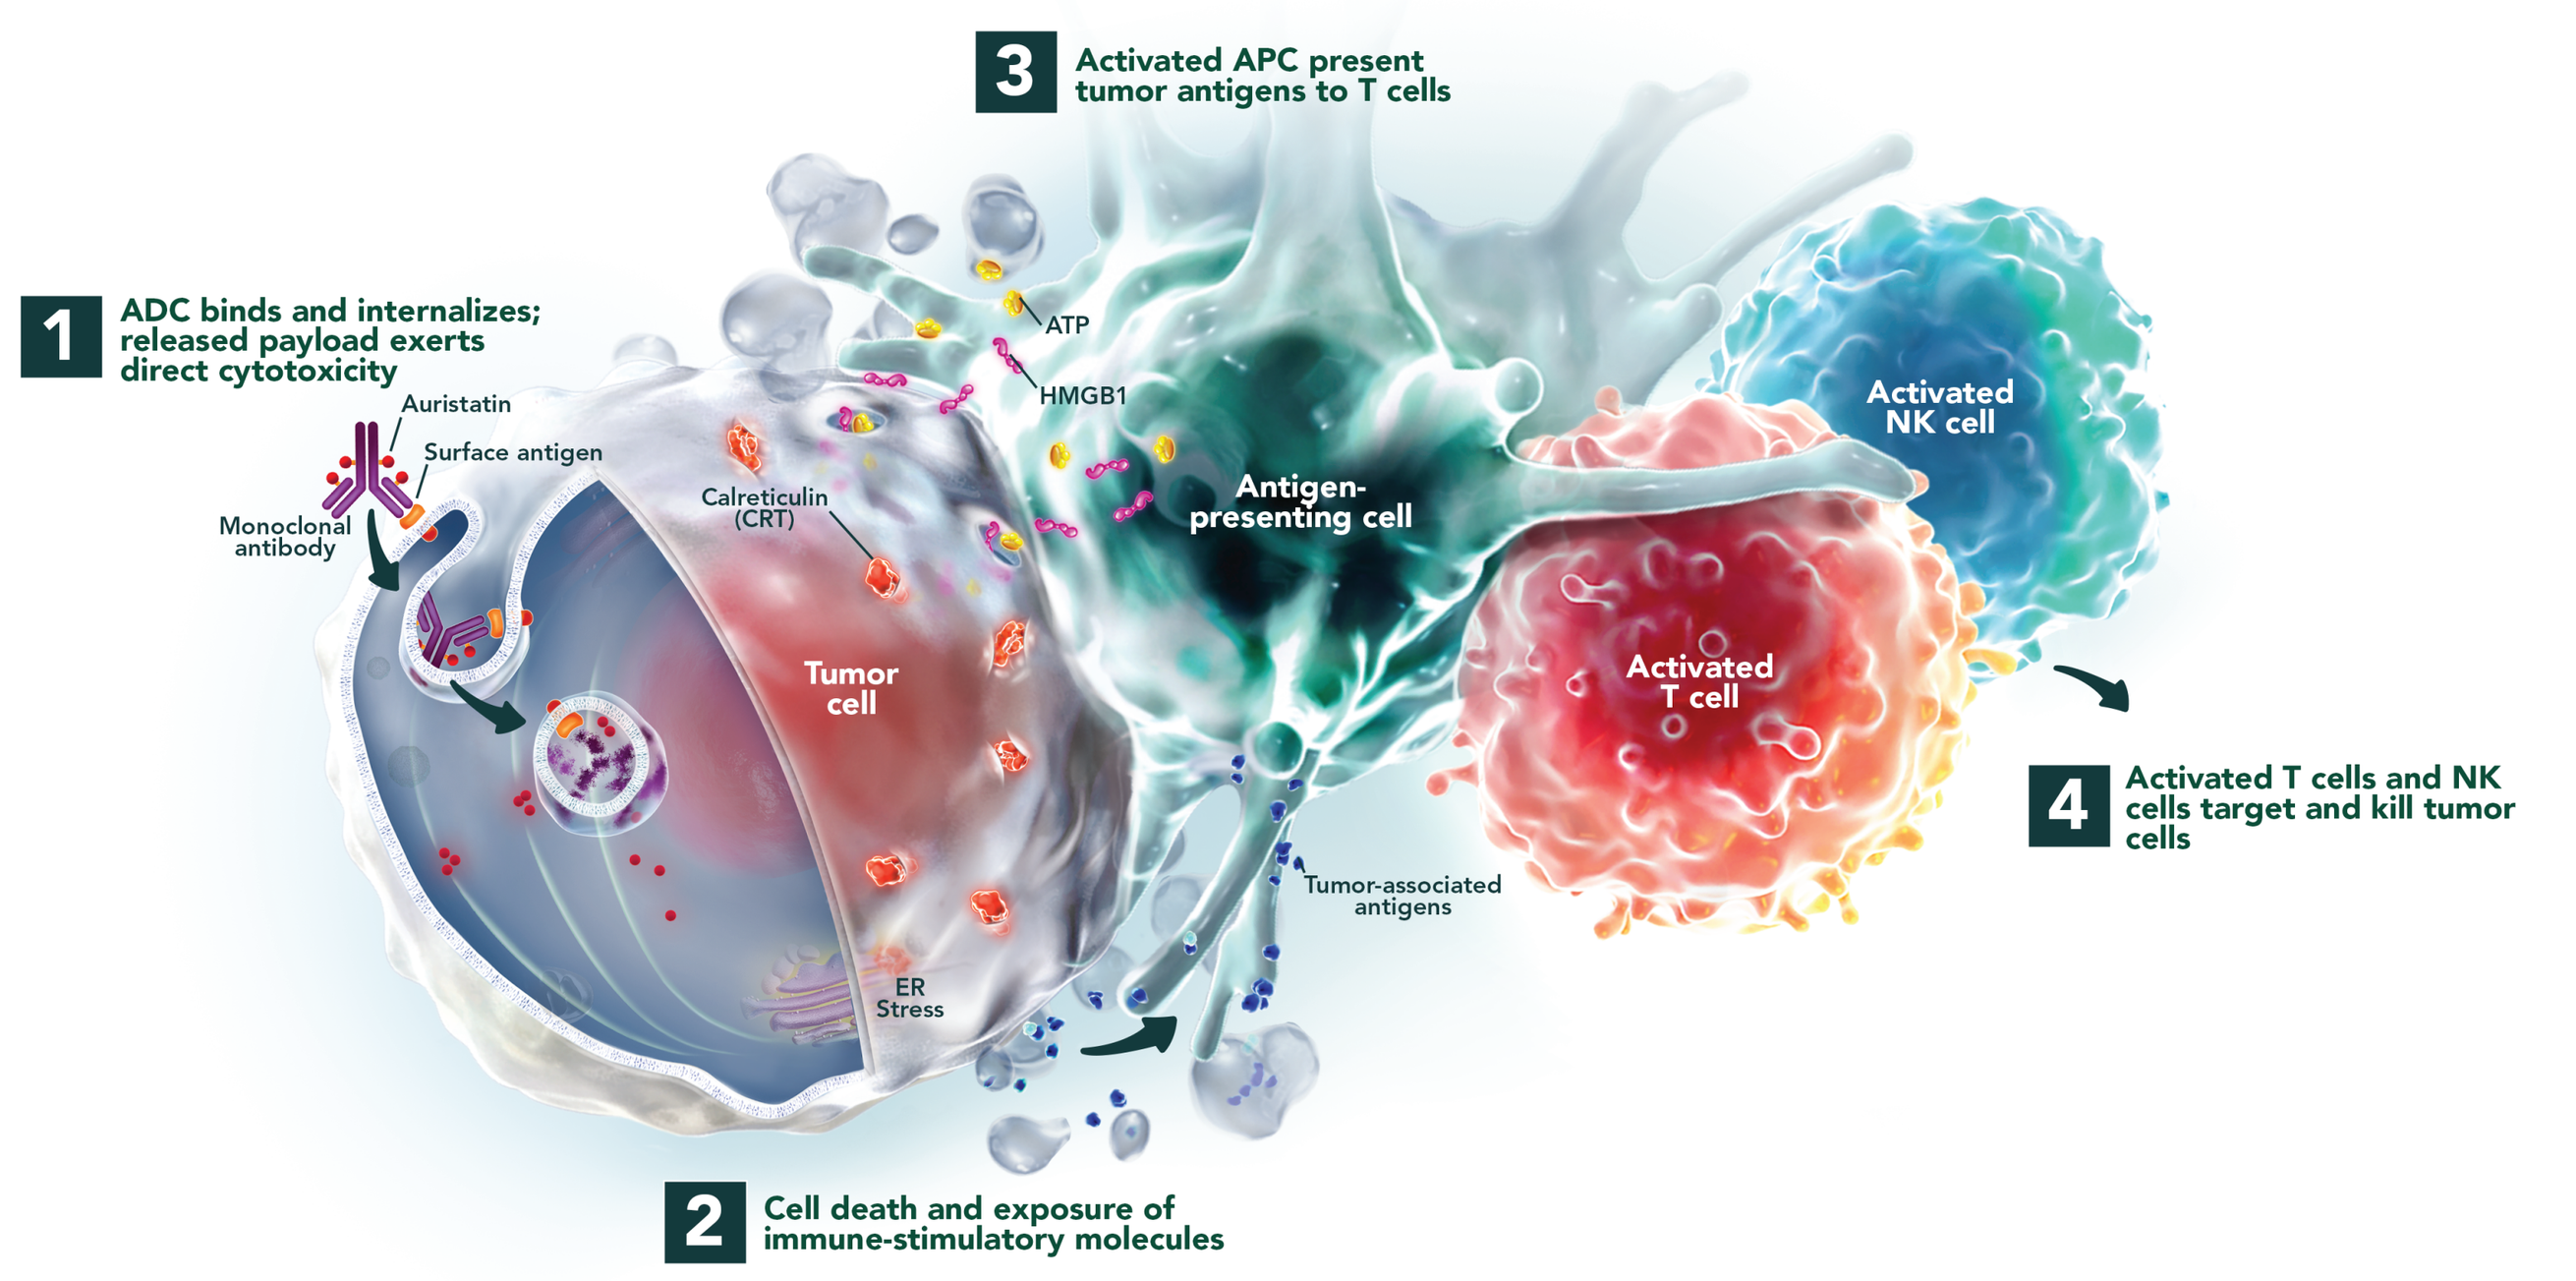
1 ADC binds and internalizes; released payload exerts direct cytotoxicity
Auristatin
Surface antigen
Monoclonal antibody
Calreticulin (CRT)
Tumor cell
ER Stress
ATP
HMGB1
2 Cell death and exposure of immune-stimulatory molecules
3 Activated APC present tumor antigens to T cells
Antigen-presenting cell
Tumor-associated antigens
Activated T cell
Activated NK cell
4 Activated T cells and NK cells target and kill tumor cells

© 2020 Seattle Genetics, Inc.

Figure 3: EV-103 Study Design (NCT03288545)

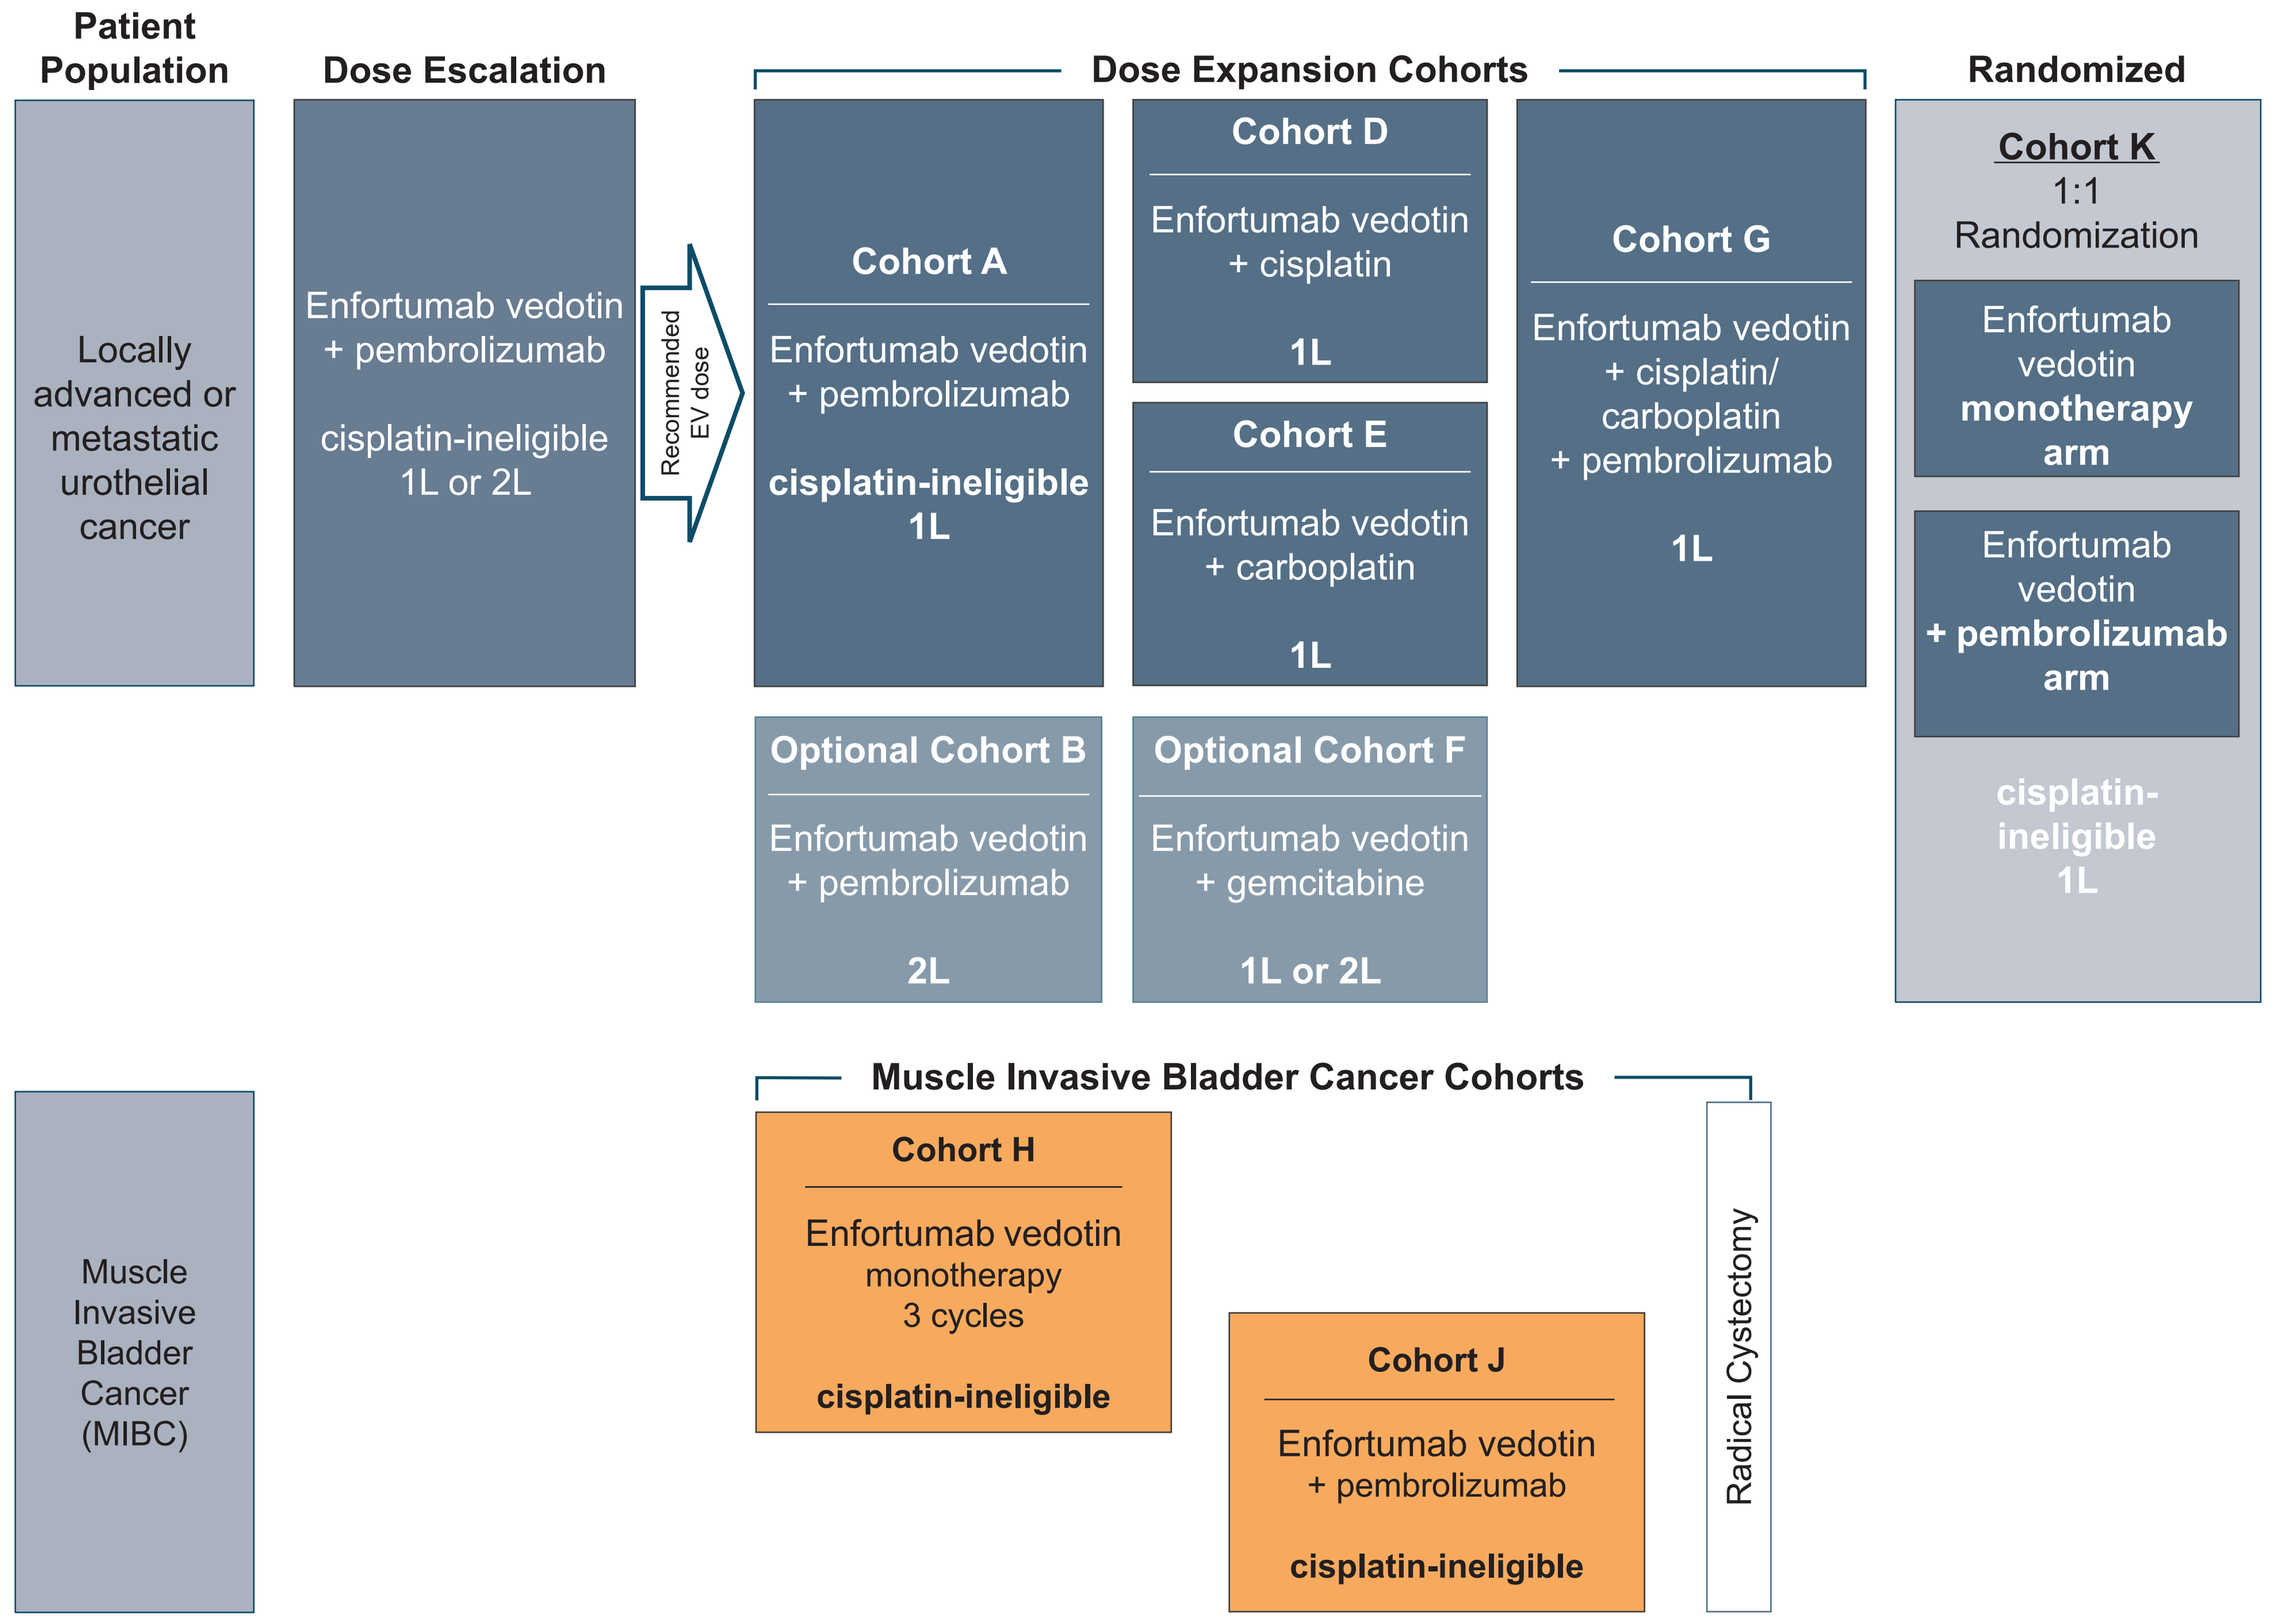
Patient Population
Dose Escalation
Dose Expansion Cohorts
Randomized
Locally advanced or metastatic urothelial cancer
Enfortumab vedotin + pembrolizumab
cisplatin-ineligible 1L or 2L
Recommended EV dose
Cohort A
Enfortumab vedotin + pembrolizumab
cisplatin-ineligible 1L
Cohort D
Enfortumab vedotin + cisplatin
1L
Cohort E
Enfortumab vedotin + carboplatin
1L
Cohort G
Enfortumab vedotin + cisplatin/ carboplatin + pembrolizumab
1L
Cohort K
1:1 Randomization
Enfortumab vedotin monotherapy arm
Enfortumab vedotin + pembrolizumab arm
cisplatin-ineligible 1L
Optional Cohort B
Enfortumab vedotin + pembrolizumab
2L
Optional Cohort F
Enfortumab vedotin + gemcitabine
1L or 2L
Muscle Invasive Bladder Cancer (MIBC)
Muscle Invasive Bladder Cancer Cohorts
Cohort H
Enfortumab vedotin monotherapy 3 cycles
cisplatin-ineligible
Cohort J
Enfortumab vedotin + pembrolizumab
cisplatin-ineligible
Radical Cystectomy

Figure 4: Enfortumab Vedotin + Pembrolizumab in Cisplatin-Ineligible MIBC Patients

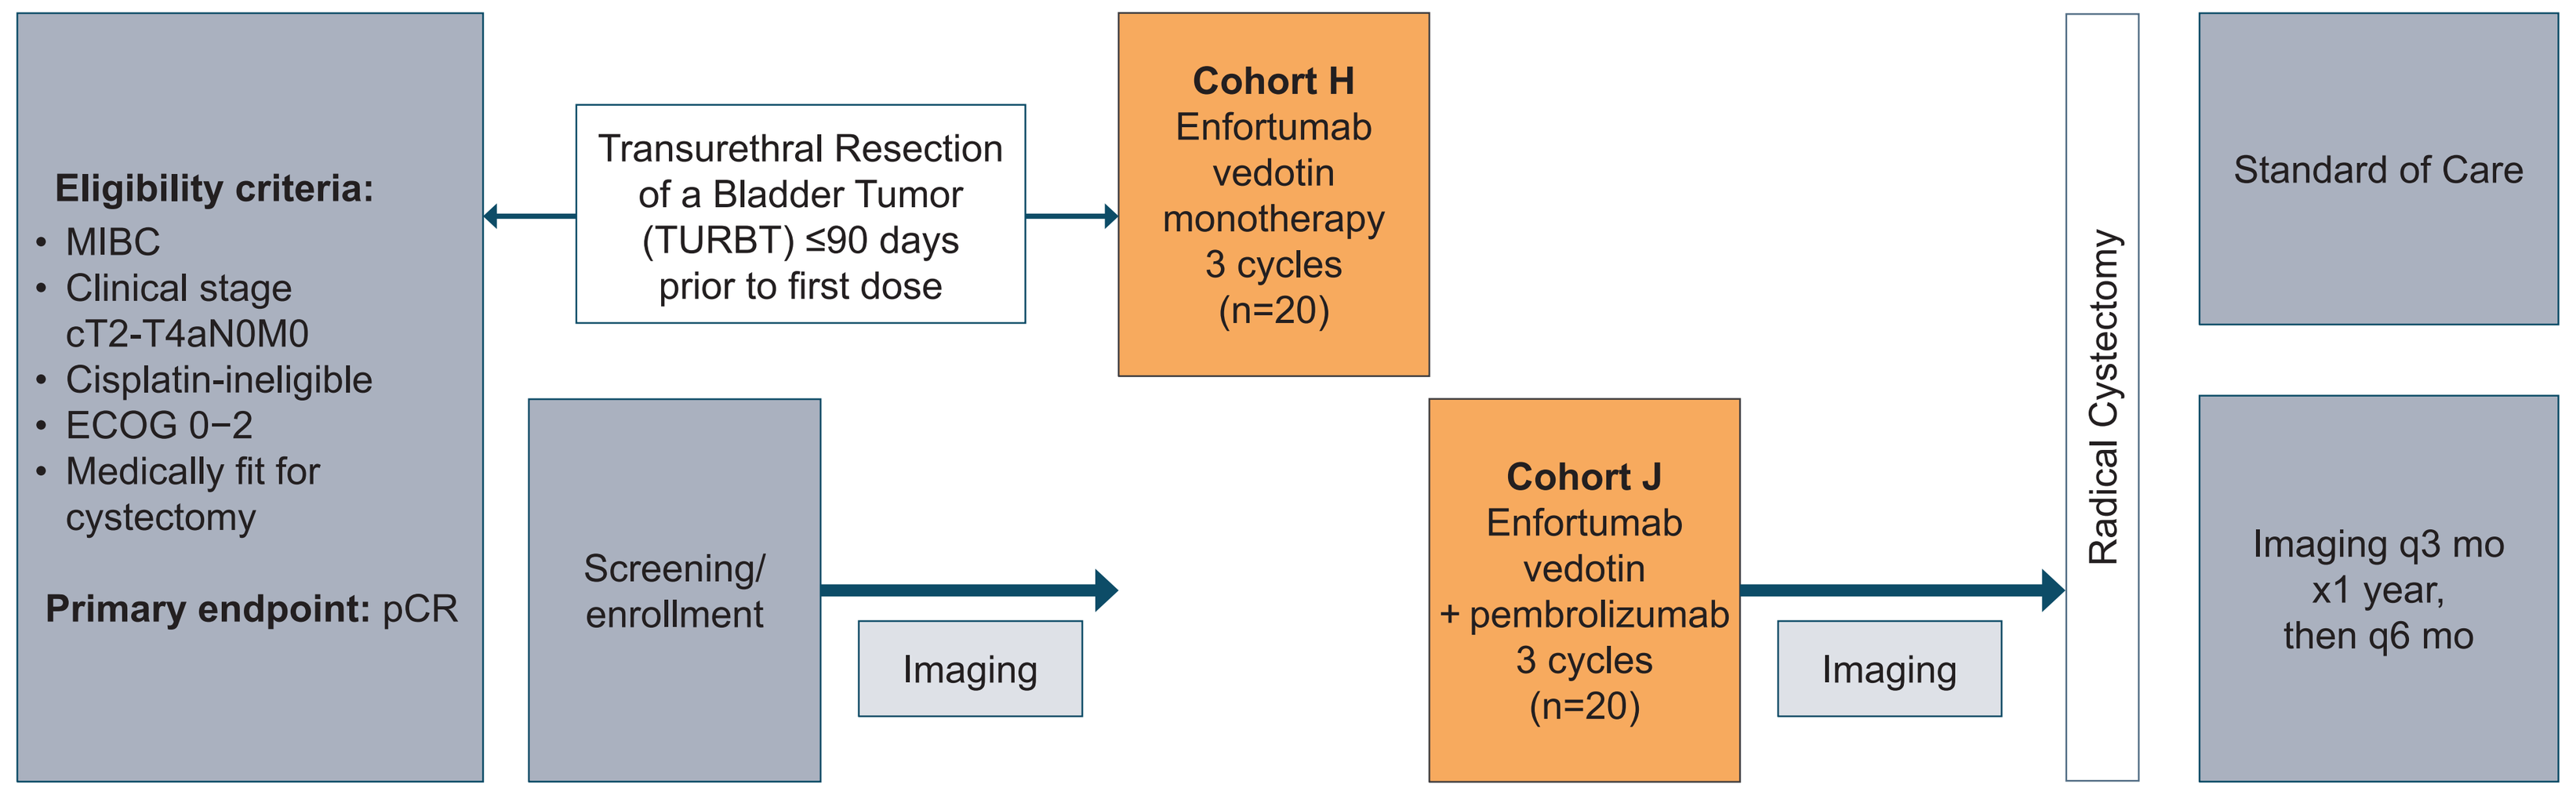
Eligibility criteria:
• MIBC
• Clinical stage cT2-T4aN0M0
• Cisplatin-ineligible
• ECOG 0−2
• Medically fit for cystectomy
Primary endpoint: pCR
Transurethral Resection of a Bladder Tumor (TURBT) ≤90 days prior to first dose
Screening/ enrollment
Imaging
Cohort H Enfortumab vedotin monotherapy 3 cycles (n=20)
Cohort J Enfortumab vedotin + pembrolizumab 3 cycles (n=20)
Imaging
Radical Cystectomy
Standard of Care
Imaging q3 mo x1 year, then q6 mo

DISCLOSURE: CJH received honoraria from Seattle Genetics, Inc. DPP and TWF received research funding from Seattle Genetics, Inc./Astellas Pharma, Inc. JER held an consulting or advisory role with Seattle Genetics, Inc. ASC is an employee of and has an ownership interest in Seattle Genetics, Inc. CS is an employee of Astellas Pharma, Inc. MC is an employee of Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA.

Objectives

Primary Objective

- To assess antitumor activity of neoadjuvant enfortumab vedotin monotherapy or enfortumab vedotin + pembrolizumab as measured by the pathological complete response (pCR) rate by local pathology review in patients with T2-T4aN0M0 MIBC

Secondary Objectives

- To assess pCR rate by central pathology review
- To assess overall survival
- To assess pathological response rate per downstaging to ≤pT1pN0 by local and central pathology review
- To assess disease-free survival and progression-free survival per RECIST Version 1.1 by investigator assessment
- To assess safety and tolerability of enfortumab vedotin monotherapy or enfortumab vedotin in combination with pembrolizumab
- To assess the percentage of delay in planned surgeries due to treatment-related adverse events

Response Assessments

- pCR will be assessed after RC + PLND with curative intent by pathologic analysis (local and central review)
- Radiologic assessment by CT/urogram or MRI/urogram
 - Within 4 weeks of enrollment
 - 4 to 12 weeks after the last dose of study treatment and prior to RC + PLND
 - Every 3 months after the previous response assessment in the first year, and subsequently every 6 months until radiologically confirmed progressive disease

Eligibility

Key Criteria

- Histologically confirmed MIBC (mixed cell types eligible if UC predominant; neuroendocrine tumors are ineligible) at stage cT2-T4a per TURBT within 90 days prior to the first dose of treatment and N0M0 by CT/MRI
- Ineligible for cisplatin-based chemotherapy at time of enrollment due to at least 1 of the following: ECOG performance status of 2, CrCl (calculated or measured) ≥30 and <60 mL/min, hearing loss/dysfunction, age, and/or allergy to cisplatin
- No prior systemic treatment, chemoradiation, or radiation therapy for MIBC
- May have received prior intravesical Bacillus Calmette–Guérin or intravesical chemotherapy for non-muscle invasive bladder cancer
- ≥18 years of age
- Medically fit (i.e., eligible for surgery) and scheduled for RC + PLND
- Tumor samples with associated pathology reports available prior to enrollment and sufficient for pathology review and biomarker analysis
- Adequate hematologic and organ function tests
- ECOG performance status of 0, 1, or 2

Study Sites and Completion Date

- 27 active sites in the US
- Study start: Oct 2017
- MIBC Cohort H open: Jan 2020

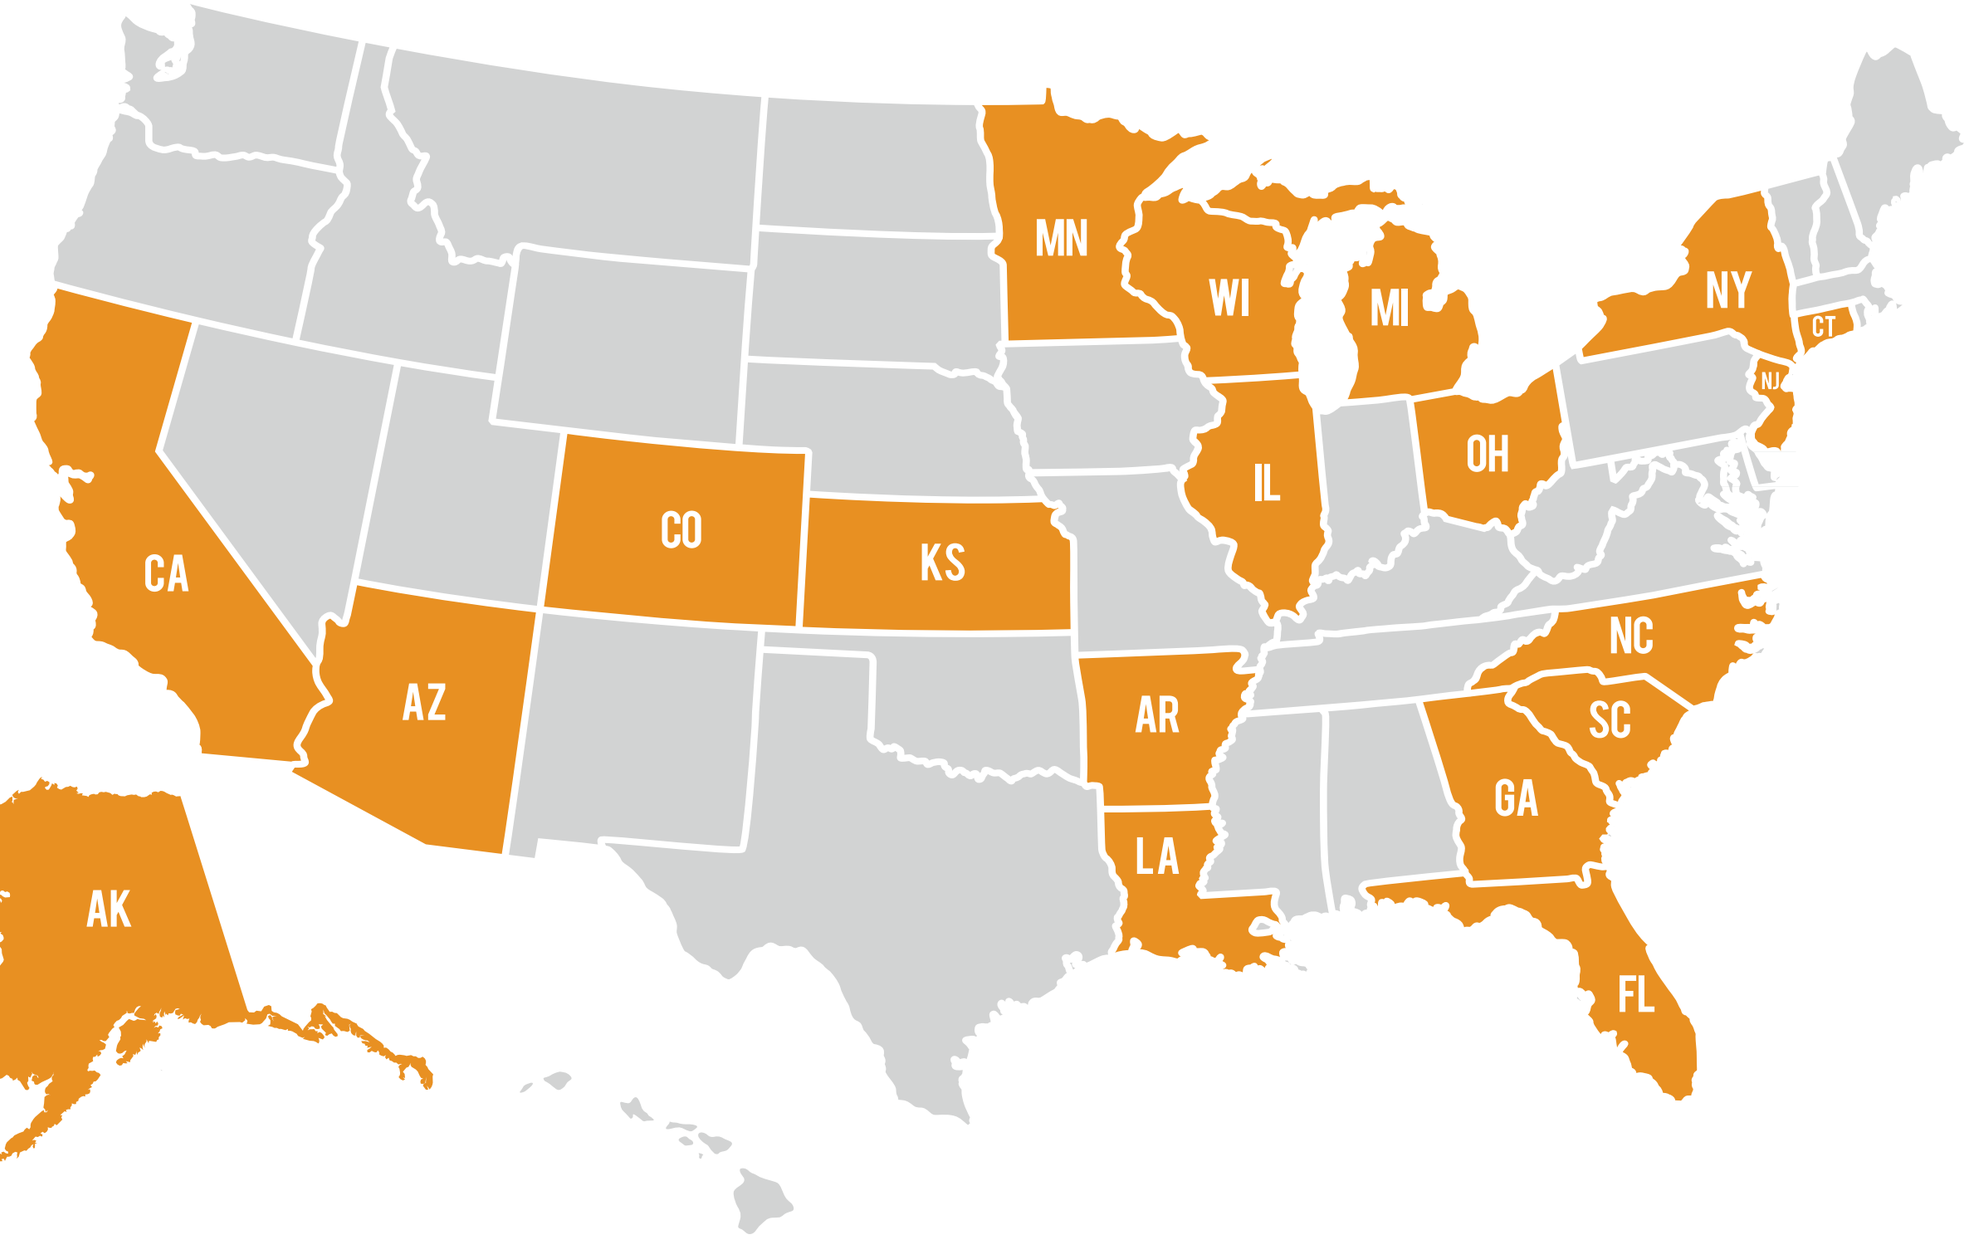
MN
WI
MI
NY
CT
NJ
OH
IL
CO
KS
CA
AZ
AR
NC
SC
GA
LA
AK
FL

References

1 Nielsen ME et al. Cancer. 2014;120:86–95
2 Dash A et al. Cancer. 2006;107:506–13
3 Challita-Eid PM et al. Cancer Res. 2016;76:3003–13
4 Petrylak DP et al. J Clin Oncol. 2017;35:106
5 Rosenberg JE et al. J Clin Oncol. 2019;37:(29):2592–600
6 Cao et al. AACR 2016. Cao et al. Cancer Res 2017;77(13 suppl): Abstract 5588.
7 Cao et al. Cancer Res 2018;78(13 Suppl): Abstract 2742.
8 Alley et al. Cancer Res 2019;79(13 Suppl): Abstract 221.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.