A FIRST-IN-HUMAN TRIAL OF AN INTEGRIN BETA-6 TARGETED ANTIBODY-DRUG CONJUGATE (ADC), SGN-B6A, IN PATIENTS WITH ADVANCED SOLID TUMORS: INTERIM RESULTS OF A PHASE 1 STUDY (SGNB6A-001)

Antoine Hollebecque[1], Juanita Lopez[2], Sarina A Piha-Paul[3], Afshin Dowlati[4], Amita Patnaik[5], Vladimir Galvao[6], Bruno Bockorny[7], Kartik Sehgal[8], Ed Kingsley[9], Rachel E Sanborn[10], Solange Peters[11], Yan Sun[12], Gabriela Patilea-Vrana[12], Mingjin Yan[12], Natalya Nazarenko[12], Emiliano Calvo[13]

[1]Institut Gustave Roussy, Villejuif, France; [2]The Royal Marsden Hospital (Surrey), London, UK; [3]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [4]Seidman Cancer Center and Case Western Reserve University, Cleveland, OH, USA; [5]START San Antonio, San Antonio TX, USA; [6]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [7]Beth Israel Deaconess Medical Center, Boston, MA, USA; [8]Dana-Farber Cancer Institute, Boston, MA, USA; [9]Comprehensive Cancer Centers of Nevada, Las Vegas, Nevada, USA; [10]Earle A. Chiles Research Institute, Providence Cancer Institute, Portland, OR, USA; [11]Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland; [12]Seagen Inc., Bothell, WA, USA; [13]START Madrid-CIOCC, Centro Integral Oncológico Clara Campal, Madrid, Spain

BACKGROUND

- Integrin beta-6, a membrane-associated protein, plays an important role in tumor pathogenesis and invasiveness, and has been correlated with poor outcomes[1,2]
- Integrin beta-6 is highly expressed in non-small cell lung cancer, head and neck cancer, and esophageal cancer[1,2] and expressed at low levels in normal tissue
- SGN-B6A is an investigational antibody-drug conjugate designed to deliver MMAE to integrin beta-6-expressing cells via selective binding to the target, to elicit MMAE-directed cytotoxicity, bystander effect, and immunogenic cell death[3]
- Preclinical studies of SGN-B6A have shown antitumor activity[4,5]
- SGNB6A-001 (NCT04389632) is a first-in-human phase 1 trial evaluating SGN-B6A in advanced solid tumors, consisting of dose escalation (Part A) and dose expansion (Part B) cohorts. Initial safety and efficacy data is presented here

STUDY DESIGN

Dose Escalation (Part A)
Up to N=85

- NSCLC
- Esophageal cancer
- HNSCC
- Breast cancer
- Cutaneous squamous cancer
- Ovarian cancer
- Exocrine pancreatic adenocarcinoma
- Bladder cancer
- Cervical cancer

→

SGN-B6A Dosing Schedules

Q1W
21-day cycle (Days 1, 8, and 15)
0.8, 1.0, and 1.2 mg/kg

2Q3W
21-day cycle (Days 1 and 8)
1.2 and 1.25 mg/kg

Q3W
21-day cycle (Day 1)
1.5 and 1.8 mg/kg

2Q4W
28-day cycle (Days 1 and 15)
1.5, 1.8, and 2.0 mg/kg

→

Dose expansion (Part B)
Up to 6 disease-specific cohorts
N=40 each

- NSCLC
- ESCC
- HNSCC
- EAC/GEJ
- Biology Cohort
- Opportunity for additional expansion cohorts

Key Eligibility Criteria

- Must have histologically or cytologically confirmed metastatic or unresectable solid malignancy, relapsed or refractory disease, or intolerance to SOC therapies
- No previous receipt of an MMAE-containing agent or agent targeting integrin beta-6

Biomarker Evaluation

- All-comer population to be enrolled, no requirement for presence of integrin beta-6 expression
- Biomarker tissue expression to be evaluated retrospectively

Study Objectives

- Primary
 - Determine safety and tolerability of SGN-B6A
 - Identify the MTD, recommended dose, and schedule for SGN-B6A
- Secondary
 - Assess the antitumor activity of SGN-B6A
 - Assess the PK and immunogenicity of SGN-B6A

DEMOGRAPHICS AND CHARACTERISTICS AT BASELINE (DOSE ESCALATION, ALL TUMOR TYPES)

	Q1W 0.8, 1.0, 1.2 mg/kg (N=30)	2Q3W 1.2, 1.25 mg/kg (N=18)	Q3W 1.5, 1.8 mg/kg (N=8)	2Q4W 1.5, 1.8, 2.0 mg/kg (N=23)	All Schedules All Dose Levels (N = 79)
Age, median years (range)	60.0 (36–84)	61.0 (48–78)	56.0 (48–73)	69.0 (46–78)	62.0 (36–84)
Sex, n (%)					
Male	11 (36.7)	9 (50)	2 (25.0)	13 (56.5)	35 (44.3)
Female	19 (63.3)	9 (50)	6 (75.0)	10 (43.5)	44 (55.7)
ECOG Performance Status, n (%)					
0	11 (36.7)	9 (50)	2 (25.0)	5 (21.7)	27 (34.2)
1	19 (63.3)	9 (50)	6 (75.0)	18 (78.3)	52 (65.8)
Number of prior systemic metastatic therapies, median (range)	2.0 (1–19)	3.0 (1–7)	5.0 (2–8)	4.0 (1–7)	3.0 (1–19)
Disease Diagnosis, n (%)					
HNSCC	3 (10)	7 (39)	1 (13)	4 (17)	15 (19)
Breast Cancer	10 (33)	4 (22)	1 (13)	0	15 (19)
NSCLC	3 (10)	4 (22)	3 (38)	17 (74)	27 (34)
Esophageal Cancer	4 (13)	3 (17)	3 (38)	2 (9)	12 (15)
Cutaneous Squamous Cell Cancer	3 (10)	0	0	0	3 (4)
Ovarian Cancer	3 (10)	0	0	0	3 (4)
Exocrine Pancreatic Adenocarcinoma	2 (7)	0	0	0	2 (3)
Bladder Cancer	1 (3)	0	0	0	1 (1)
Cervical Cancer	1 (3)	0	0	0	1 (1)

Overall TEAEs and TR-TEAEs (Dose Escalation, All Tumor Types)

	Q1W 0.8, 1.0, 1.2 mg/kg (N=30)	2Q3W 1.2, 1.25 mg/kg (N=18)	Q3W 1.5, 1.8 mg/kg (N=8)	2Q4W 1.5, 1.8, 2.0 mg/kg (N=23)	All Schedules All Dose Levels (N=79)
All TEAEs, n (%)	29 (96.7)	16 (88.9)	8 (100.0)	23 (100.0)	76 (96.2)
Treatment-related[a] TEAEs	22 (73.3)	10 (55.6)	7 (87.5)	18 (78.3)	57 (72.2)
Grade ≥3 TEAE, n (%)	21 (70.0)	11 (61.1)	2 (25.0)	15 (65.2)	49 (62.0)
Treatment-related[a] Grade ≥3 TEAE	9 (30.0)	5 (27.8)	2 (25.0)	7 (30.4)	23 (29.1)
Serious TEAEs, n (%)	15 (50.0)	10 (55.6)	2 (25.0)	10 (43.5)	37 (46.8)
Treatment-related[a] serious TEAEs	3 (10.0)	4 (22.2)	0	3 (13.0)	10 (12.7)
TEAEs leading to dose modification[b], n (%)	19 (63.3)	9 (50.0)	3 (37.5)	13 (56.5)	44 (55.7)
TEAEs leading to treatment discontinuation	3 (10.0)	3 (16.7)	0	1 (4.3)	7 (8.9)
TEAEs leading to dose reduction	7 (23.3)	3 (16.7)	1 (12.5)	5 (21.7)	16 (20.3)
TEAEs leading to dose delay/elimination	19 (63.3)	5 (27.8)	3 (37.5)	13 (56.5)	40 (50.6)
TEAEs leading to death, n (%)	0	1 (5.6)[c]	0	0	1 (1.3)

a Related to treatment with SGN-B6A as assessed by the investigator
b Patients may have more than one type of dose modification event
c Fatal TEAE of pneumonia not related to SGN-B6A per investigator assessment
Cutoff date: 24 AUG 2022

All TEAEs occurring in ≥20% of patients, in one or more groups*

	Q1W 0.8, 1.0, 1.2 mg/kg (N=30)		2Q3W 1.2, 1.25 mg/kg (N=18)		Q3W 1.5, 1.8 mg/kg (N=8)		2Q4W 1.5, 1.8, 2.0 mg/kg (N=23)		All Schedules All Dose Levels (N=79)	
	All Grade	Grade ≥3	All Grade	Grade ≥3	All Grade	Grade ≥3	All Grade	Grade ≥3	All Grade	Grade ≥3
Fatigue	11 (36.7)	3 (10.0)	7 (38.9)	0	3 (37.5)	0	8 (34.8)	2 (8.7)	29 (36.7)	5 (6.3)
Nausea	5 (16.7)	0	9 (50.0)	0	1 (12.5)	0	12 (52.2)	0	27 (34.2)	0
Diarrhea	10 (33.3)	0	4 (22.2)	3 (16.7)	2 (25.0)	0	7 (30.4)	0	23 (29.1)	3 (3.8)
Decreased appetite	10 (33.3)	0	1 (5.6)	0	2 (25.0)	0	6 (26.1)	1 (4.3)	19 (24.1)	1 (1.3)
Peripheral sensory neuropathy	7 (23.3)	1 (3.3)	4 (22.2)	0	1 (12.5)	0	5 (21.7)	1 (4.3)	17 (21.5)	2 (2.5)
Alopecia	7 (23.3)	0	3 (16.7)	0	1 (12.5)	0	5 (21.7)	0	16 (20.3)	0
Weight decreased	5 (16.7)	0	3 (16.7)	0	2 (25.0)	0	5 (21.7)	0	15 (19.0)	0
Abdominal pain	5 (16.7)	0	3 (16.7)	1 (5.6)	0	0	5 (21.7)	0	13 (16.5)	1 (1.3)
Anemia	6 (20.0)	2 (6.7)	4 (22.2)	0	2 (25.0)	1 (12.5)	1 (4.3)	0	13 (16.5)	3 (3.8)
Neutropenia	4 (13.3)	3 (10.0)	3 (16.7)	2 (11.1)	2 (25.0)	1 (12.5)	4 (17.4)	3 (13.0)	13 (16.5)	9 (11.4)
Dyspnea	3 (10.0)	2 (6.7)	4 (22.2)	1 (5.6)	1 (12.5)	0	4 (17.4)	1 (4.3)	12 (15.2)	4 (5.1)
Hypokalemia	5 (16.7)	0	4 (22.2)	2 (11.1)	1 (12.5)	0	2 (8.7)	0	12 (15.2)	2 (2.5)
Asthenia	5 (16.7)	1 (3.3)	4 (22.2)	2 (11.1)	0	0	1 (4.3)	1 (4.3)	10 (12.7)	4 (5.1)
Stomatitis	4 (13.3)	2 (6.7)	4 (22.2)	0	1 (12.5)	0	1 (4.3)	0	10 (12.7)	2 (2.5)
Alanine aminotransferase increased	1 (3.3)	0	1 (5.6)	0	2 (25.0)	0	5 (21.7)	0	9 (11.4)	0
Aspartate aminotransferase increased	2 (6.7)	0	1 (5.6)	0	2 (25.0)	0	4 (17.4)	0	9 (11.4)	0
Hyperglycemia	6 (20.0)	2 (6.7)	2 (11.1)	2 (11.1)	0	0	1 (4.3)	0	9 (11.4)	4 (5.1)
Dehydration	6 (20.0)	0	0	0	1 (12.5)	0	0	0	7 (8.9)	0
Hypertransaminasemia	1 (3.3)	0	2 (11.1)	1 (5.6)	2 (25.0)	0	0	0	5 (6.3)	1 (1.3)

* Preferred term ordered by frequency in all schedules, all dose levels
Treatment-emergent adverse events are newly occurring adverse events (not present at baseline) or adverse events that worsen after first dose of SGN-B6A. TEAEs that occurred on or after the first dose through 30 days after the last study treatment (SGN-B6A) were included in the analysis.
Cutoff date: 24 AUG 2022

- SGN-B6A demonstrated a manageable and tolerable safety profile at the explored dose levels and schedules.
- DLTs were reported in 1 patient in Q1W, 2 in 2Q3W, 0 in Q3W, and 2 patients in 2Q4W
 - Q1W: stomatitis
 - 2Q3W: diarrhea, neutropenia, and rash maculo-papular
 - 2Q4W: neutropenia and vomiting

Abbreviations

2Q3W: day 1 and day 8, 21-day cycle; 2Q4W: day 1 and day 15, on a 21-day cycle; acMMAE: antibody-conjugated monomethyl auristatin E; ADC: antibody-drug conjugate; cCR: confirmed complete response; CI: confidence interval; cORR: confirmed objective response rate; cPR: confirmed partial response; CR: complete response; cSCC: cutaneous squamous cell carcinoma; D: day; DLT: dose limiting toxicity; EAC: esophageal adenocarcinoma; ECOG: Eastern Cooperative Oncology Group; EC: esophageal cancer; ESCC: esophageal squamous cell carcinoma; GEJ: gastroesophageal junction adenocarcinoma; GPP3: Good Publication Practice; HNSCC: head and neck squamous cell carcinoma; ICMJE: International Committee of Medical Journal Editors; MMAE: monomethyl auristatin E; MTD: maximum tolerated dose; NA: no assessment; NE: not evaluable; NSCLC: non-small cell lung carcinoma; PD: progressive disease; PD-L1: programmed death-ligand 1; PK: pharmacokinetics; PR: partial response; Q1W: once every week, 21-day cycle; Q3W: once every 3 weeks, 21-day cycle; RECIST: Response Evaluation Criteria in Solid Tumors; SD: stable disease; SOC: standard of care; SoD: sum of diameters; TEAEs: treatment-emergent adverse events; TKI: tyrosine kinase inhibitor; TR-TEAEs: treatment-related treatment emergent adverse events

References

1. Li F, Shang Y, Shi F, Zhang L, et al. Cancer Management and Research 2020;12 9599-9608
2. Elayadi A, Samli K-N, Prudkin L, et al. Cancer Res 2007;67: (12)
3. Klussman K, Tenn E-M, Higgins S, et al. J Immunother Cancer 2020;8(Suppl 3): A1-A559
4. Lyon R P, Gosink J J, Hale C J, et al. Cancer Res 2021
5. Lyon R P, Schmitt M W, Jonas M, et al. Cancer Res 2020

CONFIRMED OBJECTIVE RESPONSE RATE (DOSE ESCALATION, ALL TUMOR TYPES)

	Q1W 0.8, 1.0, 1.2 mg/kg (N=30)	2Q3W 1.2, 1.25 mg/kg (N=18)	Q3W 1.5, 1.8 mg/kg (N=8)	2Q4W 1.5, 1.8, 2.0 mg/kg (N=23)
cORR[a], n (%)	4 (13.3)	4 (22.2)	2 (25.0)	7 (30.4)
95% CI[b]	(3.8, 30.7)	(6.4, 47.6)	(3.2, 65.1)	(13.2, 52.9)
Best Overall Response[c] (Overall, n[%])				
cCR[d]	0	0	0	1 (4.3)
cPR	4 (13.3)	4 (22.2)	2 (25.0)	6 (26.1)
SD	17 (56.7)	7 (38.9)	3 (37.5)	7 (30.4)
PD	7 (23.3)	4 (22.2)	2 (25.0)	8 (34.8)
NE[e]	0	0	0	1 (4.3)
NA	2 (6.7)	3 (16.7)	1 (12.5)	0

a Responses were observed in HNSCC, NSCLC, EC, and cSCC
b Two-sided 95% exact confidence interval, computed using the Clopper-Pearson method (1934)
c Best overall response according to RECIST v1.1 CR or PR were confirmed with repeat scans >=28 days after the initial response
d cCR in NSCLC in 2Q4W
e Patients had post-baseline assessment and the best overall response was determined to be NE per RECIST v1.1
The efficacy-evaluable set includes all treated patients who had both a baseline and at least 1 evaluable post baseline disease assessment per RECIST v1.1 (assessed by investigator) or discontinued the treatment
Cutoff Date: 24 AUG 2022

SGN-B6A PHARMACOKINETICS

SGN-B6A mean concentration-time curves in Q1W, 2Q3W, Q3W, and 2Q4W dose escalation cohorts

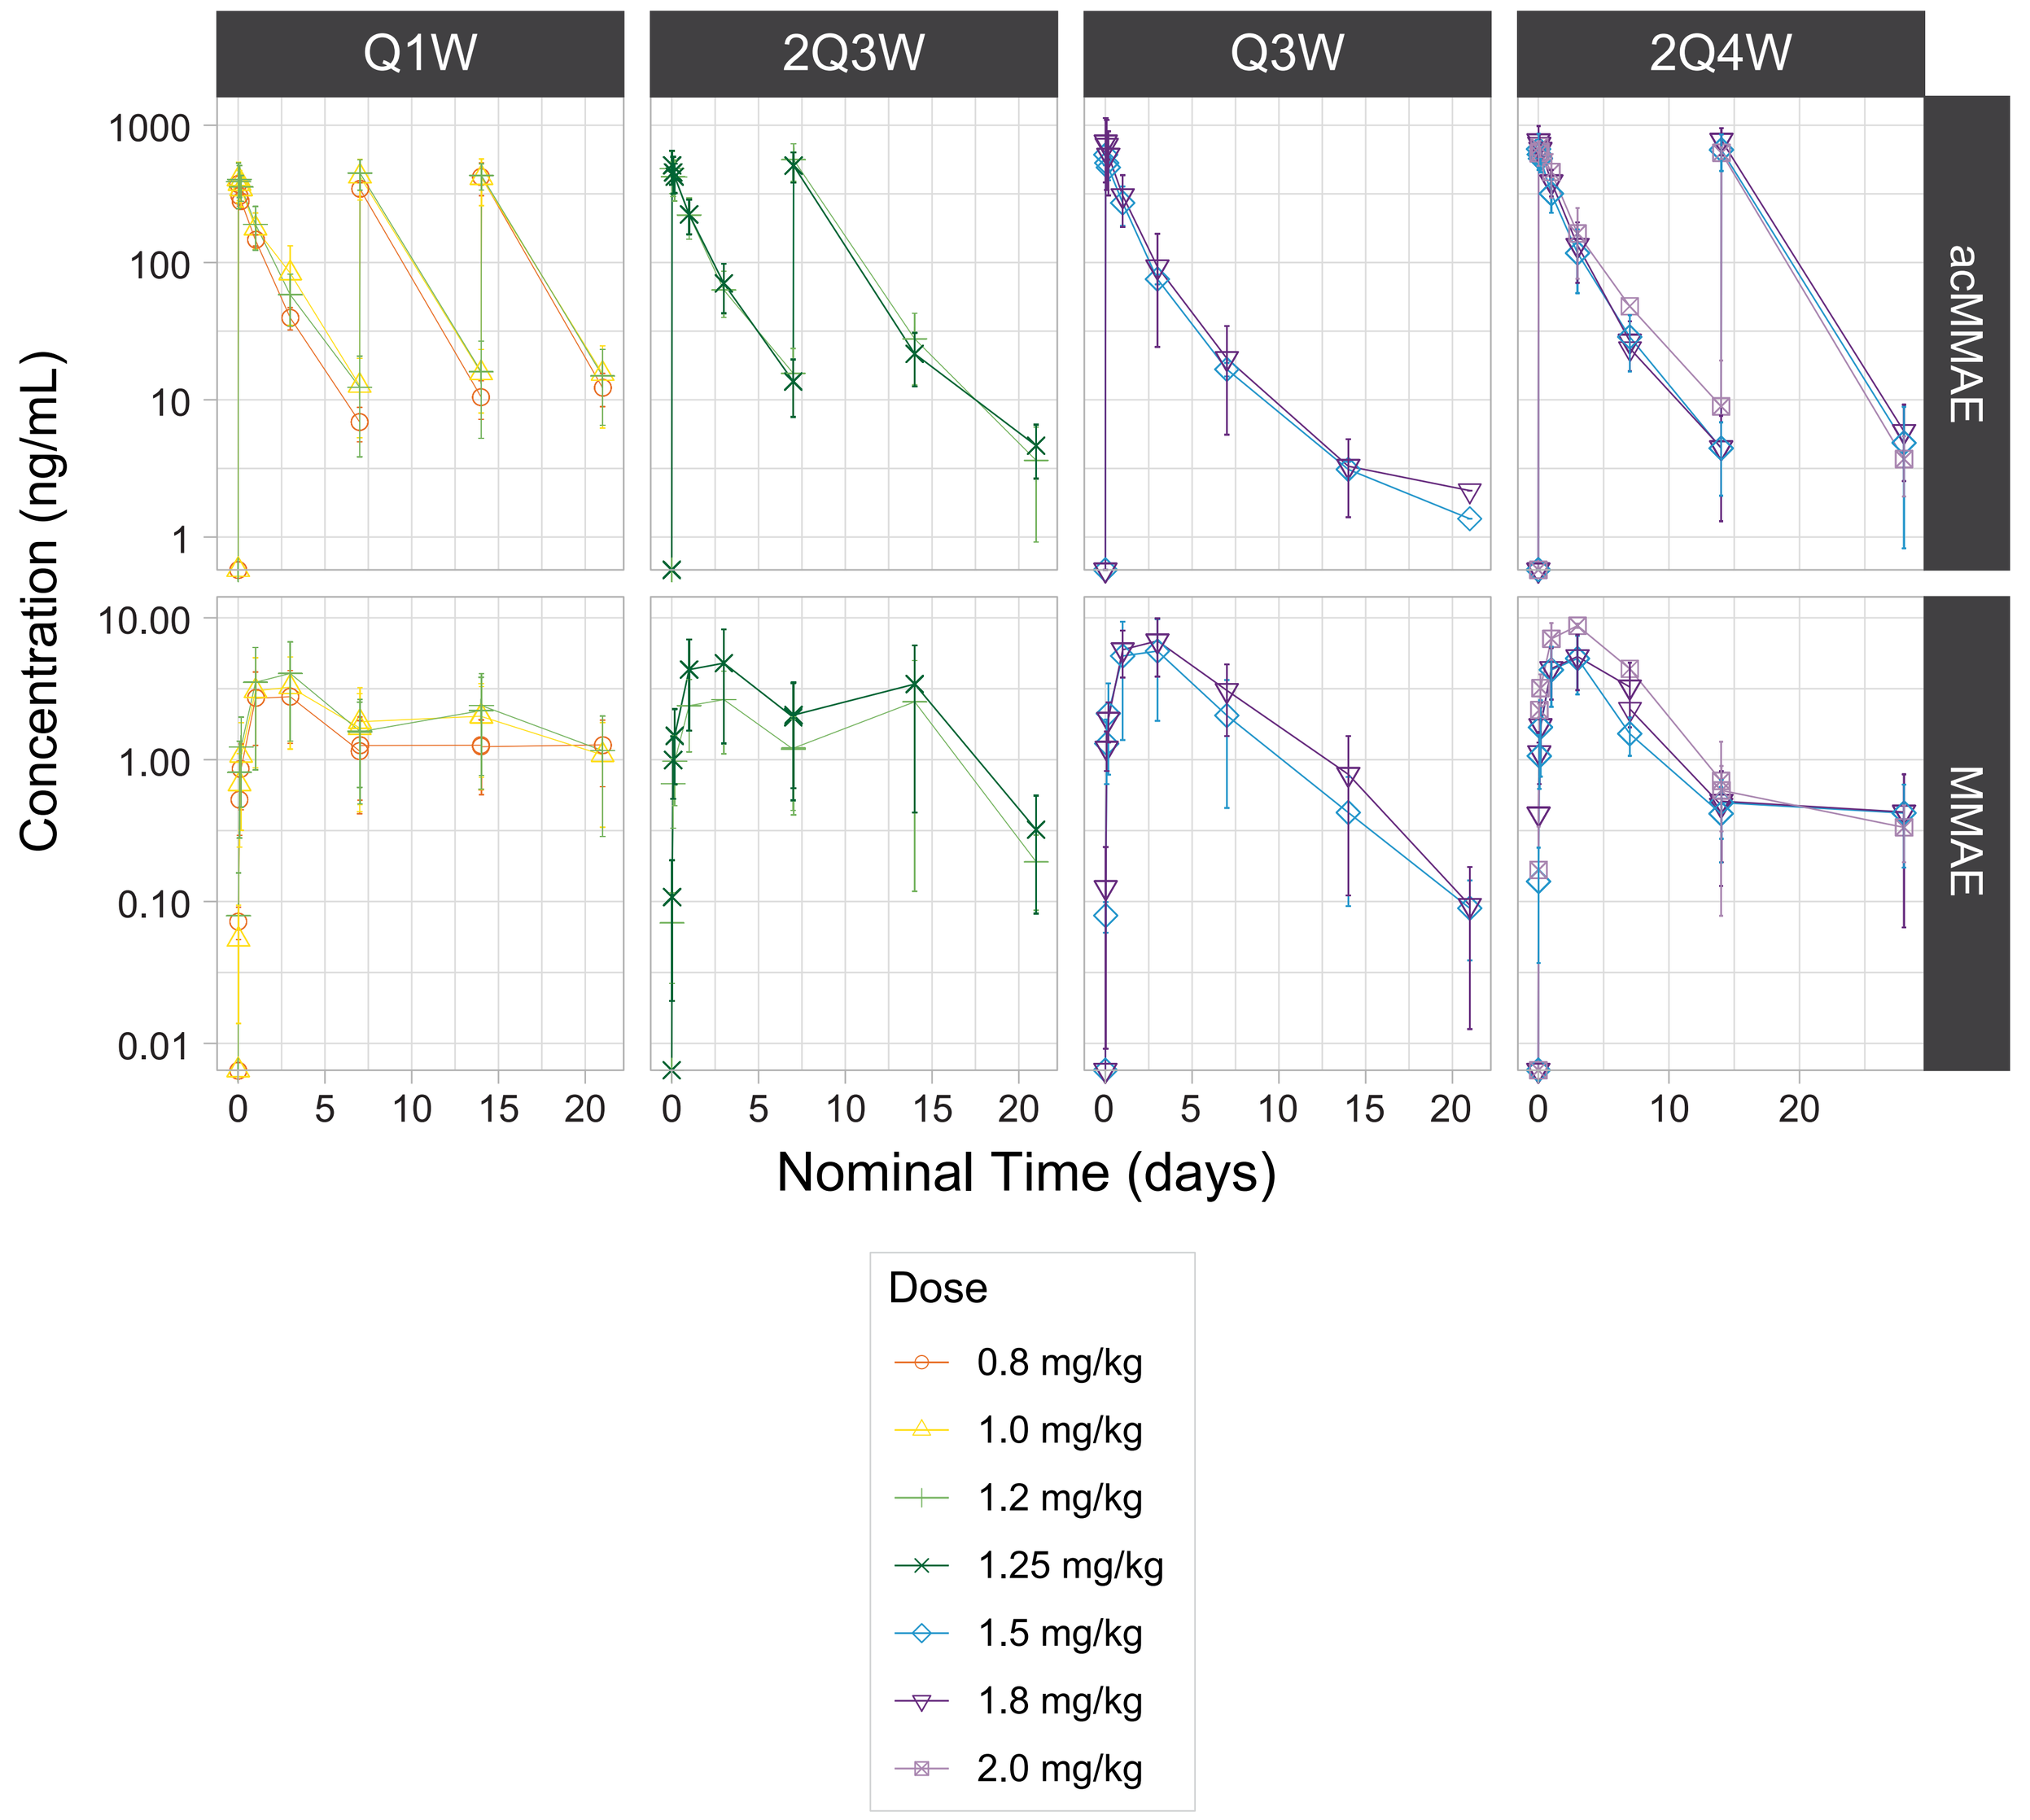
Q1W
2Q3W
Q3W
2Q4W
acMMAE
MMAE
Concentration (ng/mL)
Nominal Time (days)
Dose
0.8 mg/kg
1.0 mg/kg
1.2 mg/kg
1.25 mg/kg
1.5 mg/kg
1.8 mg/kg
2.0 mg/kg

- Both antibody-conjugated MMAE (acMMAE) and unconjugated MMAE exposures increased in an approximately dose-proportional manner across 0.8–2.0 mg/kg dose levels
- Limited accumulation was observed across all schedules

SGN-B6A: ANTITUMOR ACTIVITY

NSCLC Subset, Dose Escalation
Median 3.5 (range: 1–8) lines of prior therapy[a]

Best Percentage Change in Target Lesion SoD from Baseline per RECIST v1.1

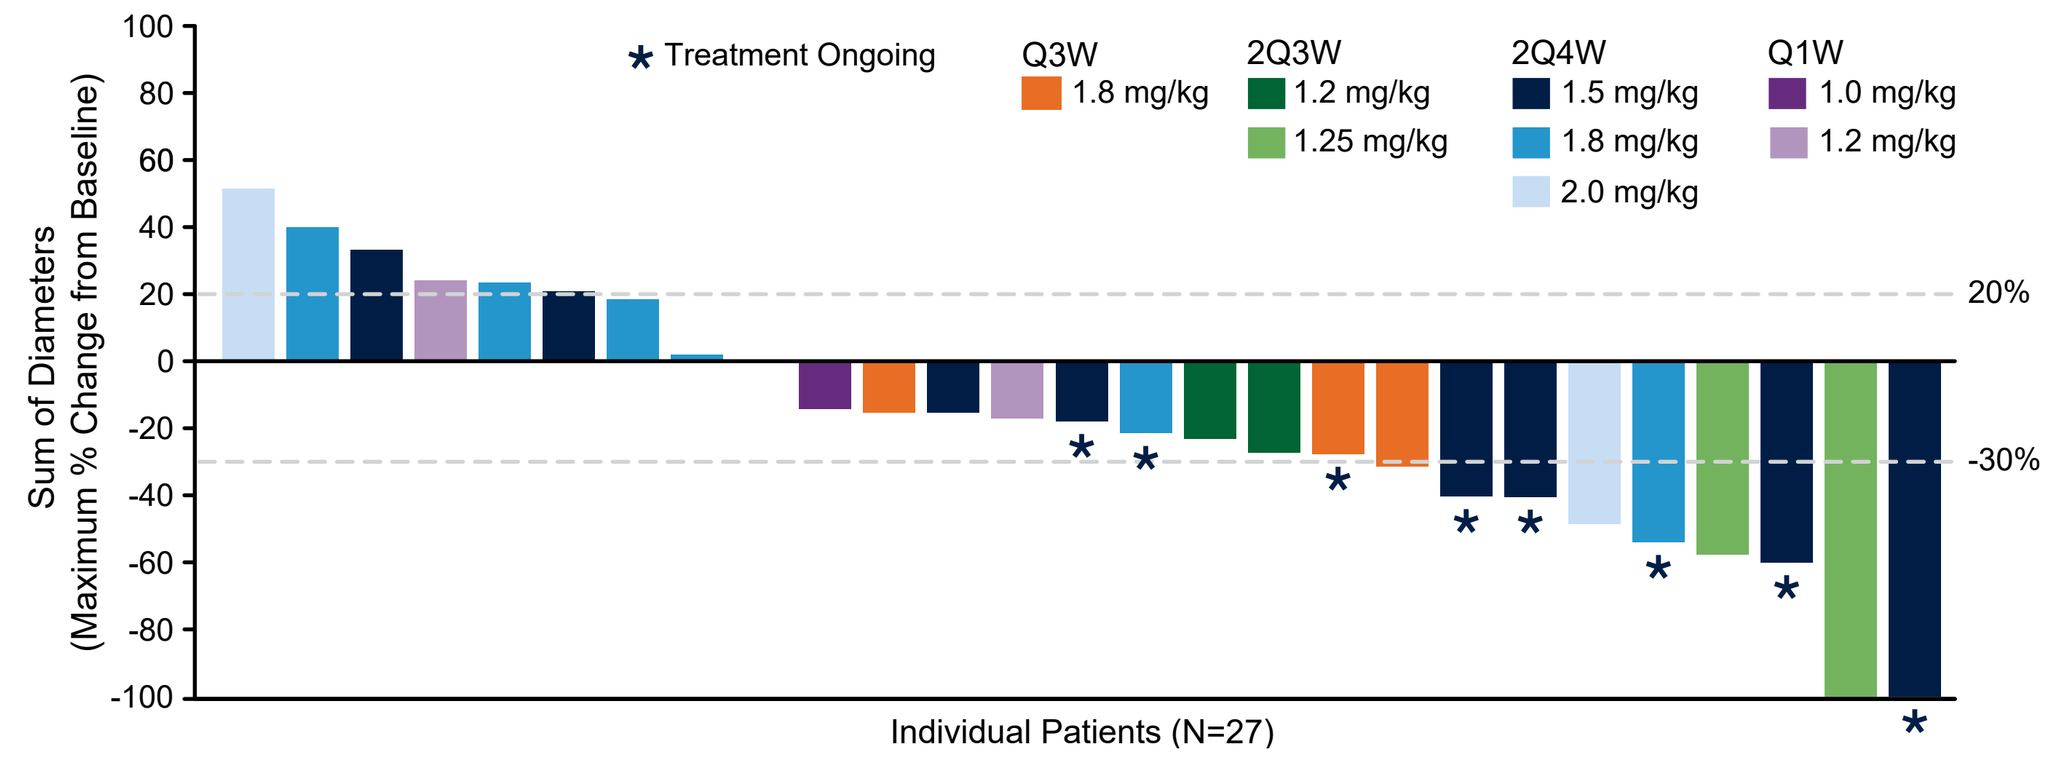
Sum of Diameters (Maximum % Change from Baseline)
Individual Patients (N=27)
Treatment Ongoing
Q3W
1.8 mg/kg
2Q3W
1.2 mg/kg
1.25 mg/kg
2Q4W
1.5 mg/kg
1.8 mg/kg
2.0 mg/kg
Q1W
1.0 mg/kg
1.2 mg/kg
20%
-30%

	NSCLC Total (N=27)
cORR, n (%)	**9 (33.3)**
95% CI	(16.5, 54.0)
Best Overall Response (Overall, n [%])	
cCR	1 (3.7)
cPR	8 (29.6)
SD	8 (29.6)
PD	9 (33.3)
NE	1 (3.7)
NA	0

a Prior therapy includes platinum and anti-PD/L-1 therapy. In patients with driver mutations prior therapy includes TKI and cytotoxic therapy

EC Subset, Dose Escalation
Median 3.0 (range: 1–5) lines of prior therapy

Best Percentage Change in Target Lesion SoD from Baseline per RECIST v1.1

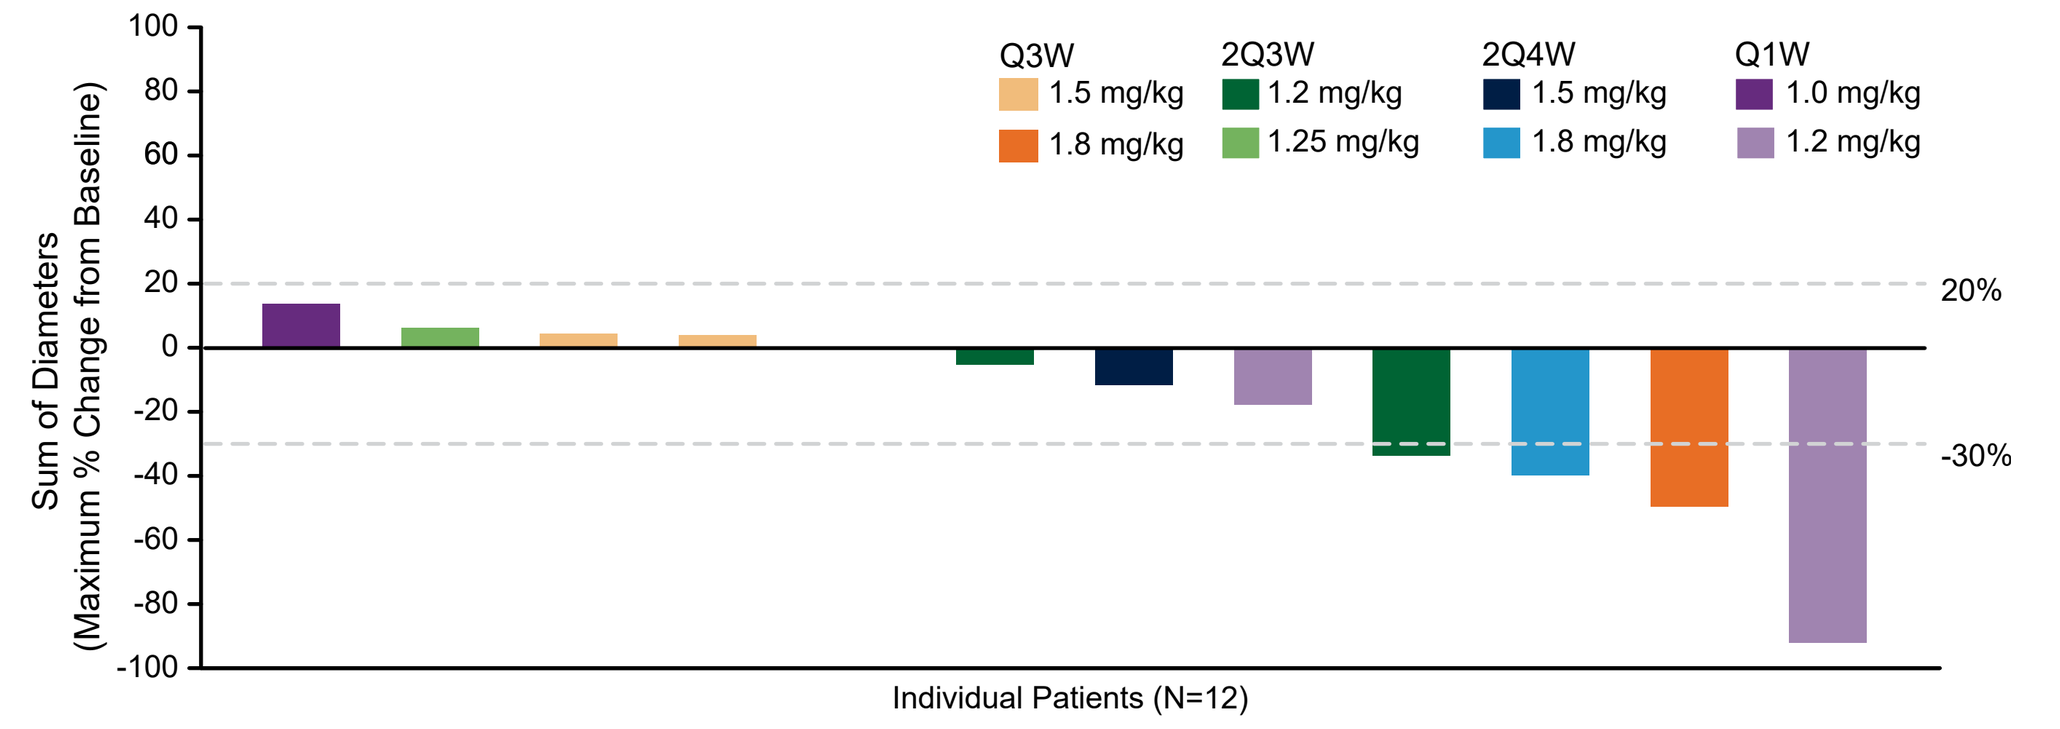
Sum of Diameters (Maximum % Change from Baseline)
Individual Patients (N=12)
Q3W
1.5 mg/kg
1.8 mg/kg
2Q3W
1.2 mg/kg
1.25 mg/kg
2Q4W
1.5 mg/kg
1.8 mg/kg
Q1W
1.0 mg/kg
1.2 mg/kg
20%
-30%

	EC Total (N=12)
cORR, n (%)	**4 (33.3)**
95% CI	(9.9, 65.1)
Best Overall Response (Overall, n [%])	
cCR	0
cPR	4 (33.3)
SD	4 (33.3)
PD	4 (33.3)
NE	0
NA	0

HNSCC Subset, Dose Escalation
Median 3.0 (range: 1–4) lines of prior therapy

Best Percentage Change in Target Lesion SoD from Baseline per RECIST v1.1

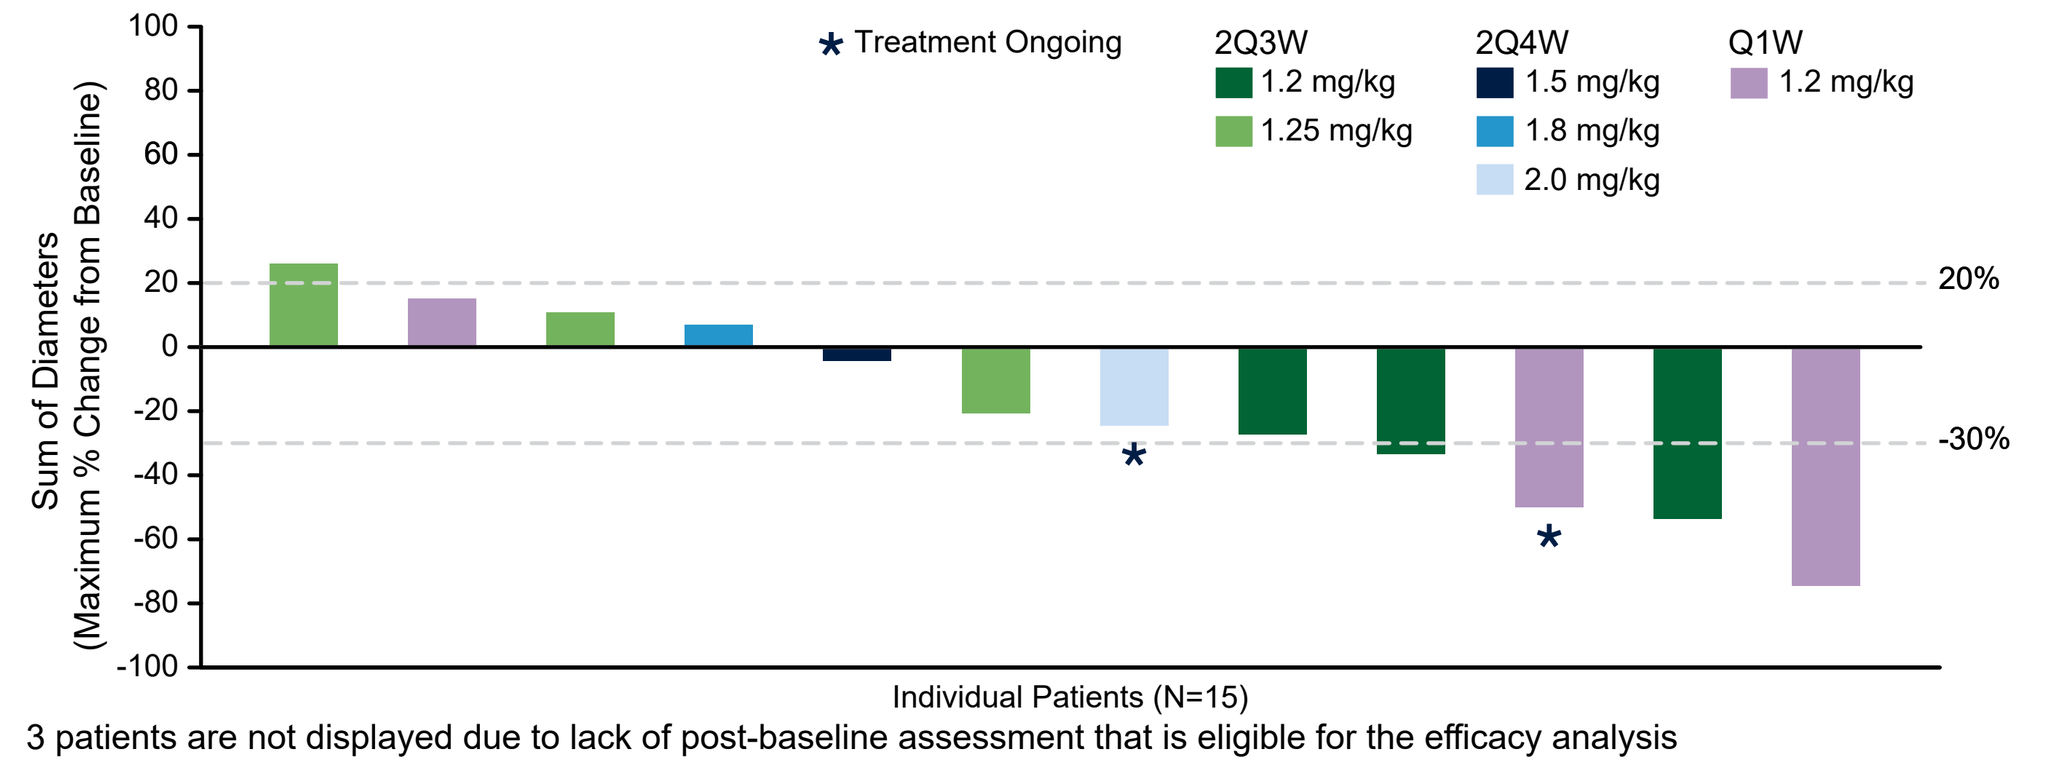
Sum of Diameters (Maximum % Change from Baseline)
Individual Patients (N=15)
Treatment Ongoing
2Q3W
1.2 mg/kg
1.25 mg/kg
2Q4W
1.5 mg/kg
1.8 mg/kg
2.0 mg/kg
Q1W
1.2 mg/kg
20%
-30%

	HNSCC Total (N=15)
cORR, n (%)	**3 (20.0)**
95% CI	(4.3, 48.1)
Best Overall Response (Overall, n [%])	
cCR	0
cPR	3 (20.0)
SD	7 (46.7)
PD	3 (20.0)
NE	0
NA	2 (13.3)

3 patients are not displayed due to lack of post-baseline assessment that is eligible for the efficacy analysis

HNSCC, 2Q3W (Dose Expansion)[a]
Median 3 (range: 1–6) lines of prior therapy

Best Percentage Change in Target Lesion SoD from Baseline per RECIST v1.1

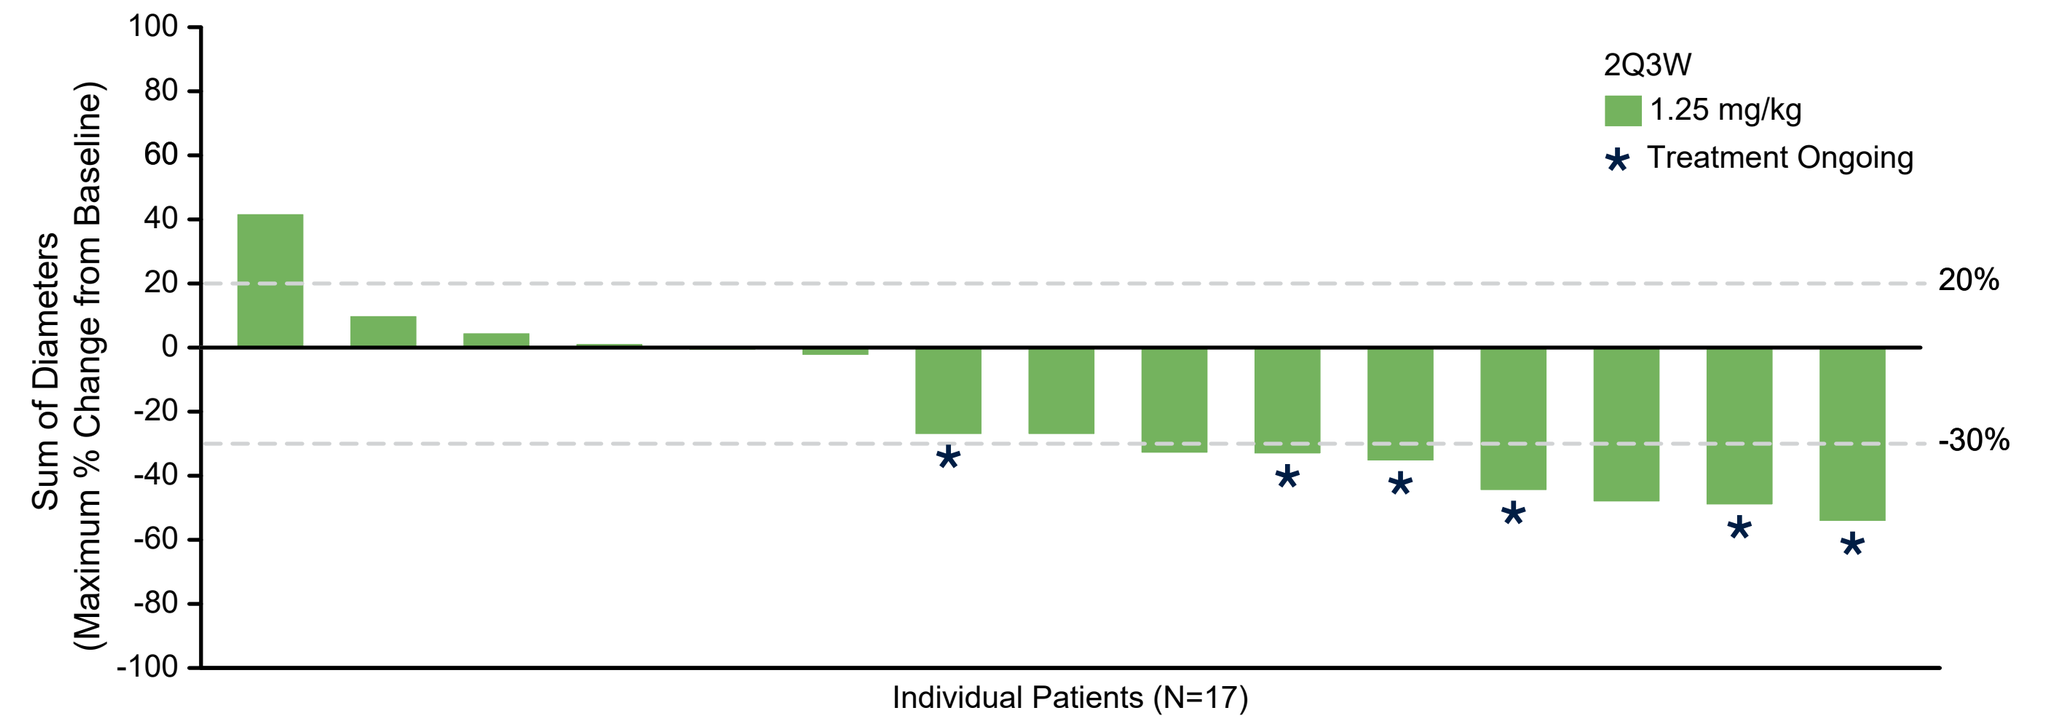
Sum of Diameters (Maximum % Change from Baseline)
Individual Patients (N=17)
2Q3W
1.25 mg/kg
Treatment Ongoing
20%
-30%

	2Q3W, HNSCC 1.25 mg/kg (N=17)
cORR, n (%)	**5 (29.4)**
95% CI	(10.3, 56.0)
Best Overall Response (Overall, n [%])	
cCR	0
cPR	5 (29.4)
SD	5 (29.4)
PD	5 (29.4)
NE	1 (5.9)
NA	1 (5.9)

- To date, the HNSCC safety profile is consistent with the 2Q3W escalation cohort
- 17 of 18 (94%) treated patients experienced TEAEs and 9 of 18 (50%) treated patients experienced Grade ≥3 TEAEs

a HNSCC dose expansion cohort has reached interim analysis by the time of data cutoff
2 patients are not displayed due to the lack of post baseline assessment that is eligible for the efficacy analysis

CONCLUSIONS

- SGN-B6A demonstrated a manageable and tolerable safety profile. Intermittent dosing schedules (2Q3W, 2Q4W) are being evaluated further
- SGN-B6A exposure is approximately dose-proportional and has no significant accumulation
- Initial antitumor activity is encouraging and has triggered expansion cohorts in NSCLC, HNSCC, and ESCC, which are currently ongoing

Acknowledgements
This study was sponsored by Seagen Inc., Bothell, WA, USA. The authors thank all our patients and families for their participation in the study and all research personnel for their support of this trial. Yan Sun, MS, is an employee of TechData Service Company LLC, King of Prussia, PA, USA, under funding by Seagen Inc. Hanna Thomsen, PhD (employee and stockholder of Seagen Inc.) provided medical writing and editorial support in accordance with GPP3 guidelines. All authors met the ICMJE criteria for authorship.

Ethical Statement
The trial is being conducted in compliance with the Declaration of Helsinki and International Conference on Harmonization Guidelines for Good Clinical Practice. All patients, or their legal representatives, provided informed consent. All participating sites have been approved by a corresponding institutional review board or independent ethical committee per the participating institution.

Abstract No. 731
Society for Immunotherapy of Cancer; November 8–12, 2022; Boston, MA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SITC or the author of this poster, **Antoine Hollebecque, antoine.hollebecque@gustaveroussy.fr**