Vedotin ADCs Induce ER Stress and Elicit Hallmarks of ICD Across Multiple Cancer Indications

Kerry Klussman[1], Elena-Marie Tenn[1], Shaylin Higgins[1], Rebecca Mazahreh[1], Katie Snead[1], Joseph Z. Hamilton[1], Bryan Grogan[1], Johann Sigurjonsson[1], Anthony Cao[1], Shyra Gardai[1], and Bernard A. Liu[1]

[1]Seagen Inc., Bothell, WA USA

Background

Effective cancer treatment requires durable elimination of malignant cells. Cytotoxic chemotherapeutic agents used to treat cancer often show initial anti-tumor efficacy, but fail to produce long-term durable responses in patients. The elicitation of durable responses and improved survival in response to cytotoxic agents may be associated with the induction of innate and adaptive immune response to the cancer. For example, tumor cells undergoing apoptosis following exposure to some cytotoxic agents emit immunostimulatory damage-associated molecular patterns (DAMPs), this form of cell death is termed immunogenic cell death (ICD). ICD can promote the recruitment and activation of both the innate and adaptive immune system, providing an additional mechanism to drive an anti-tumor response.

Immunogenic Cell Death (ICD)

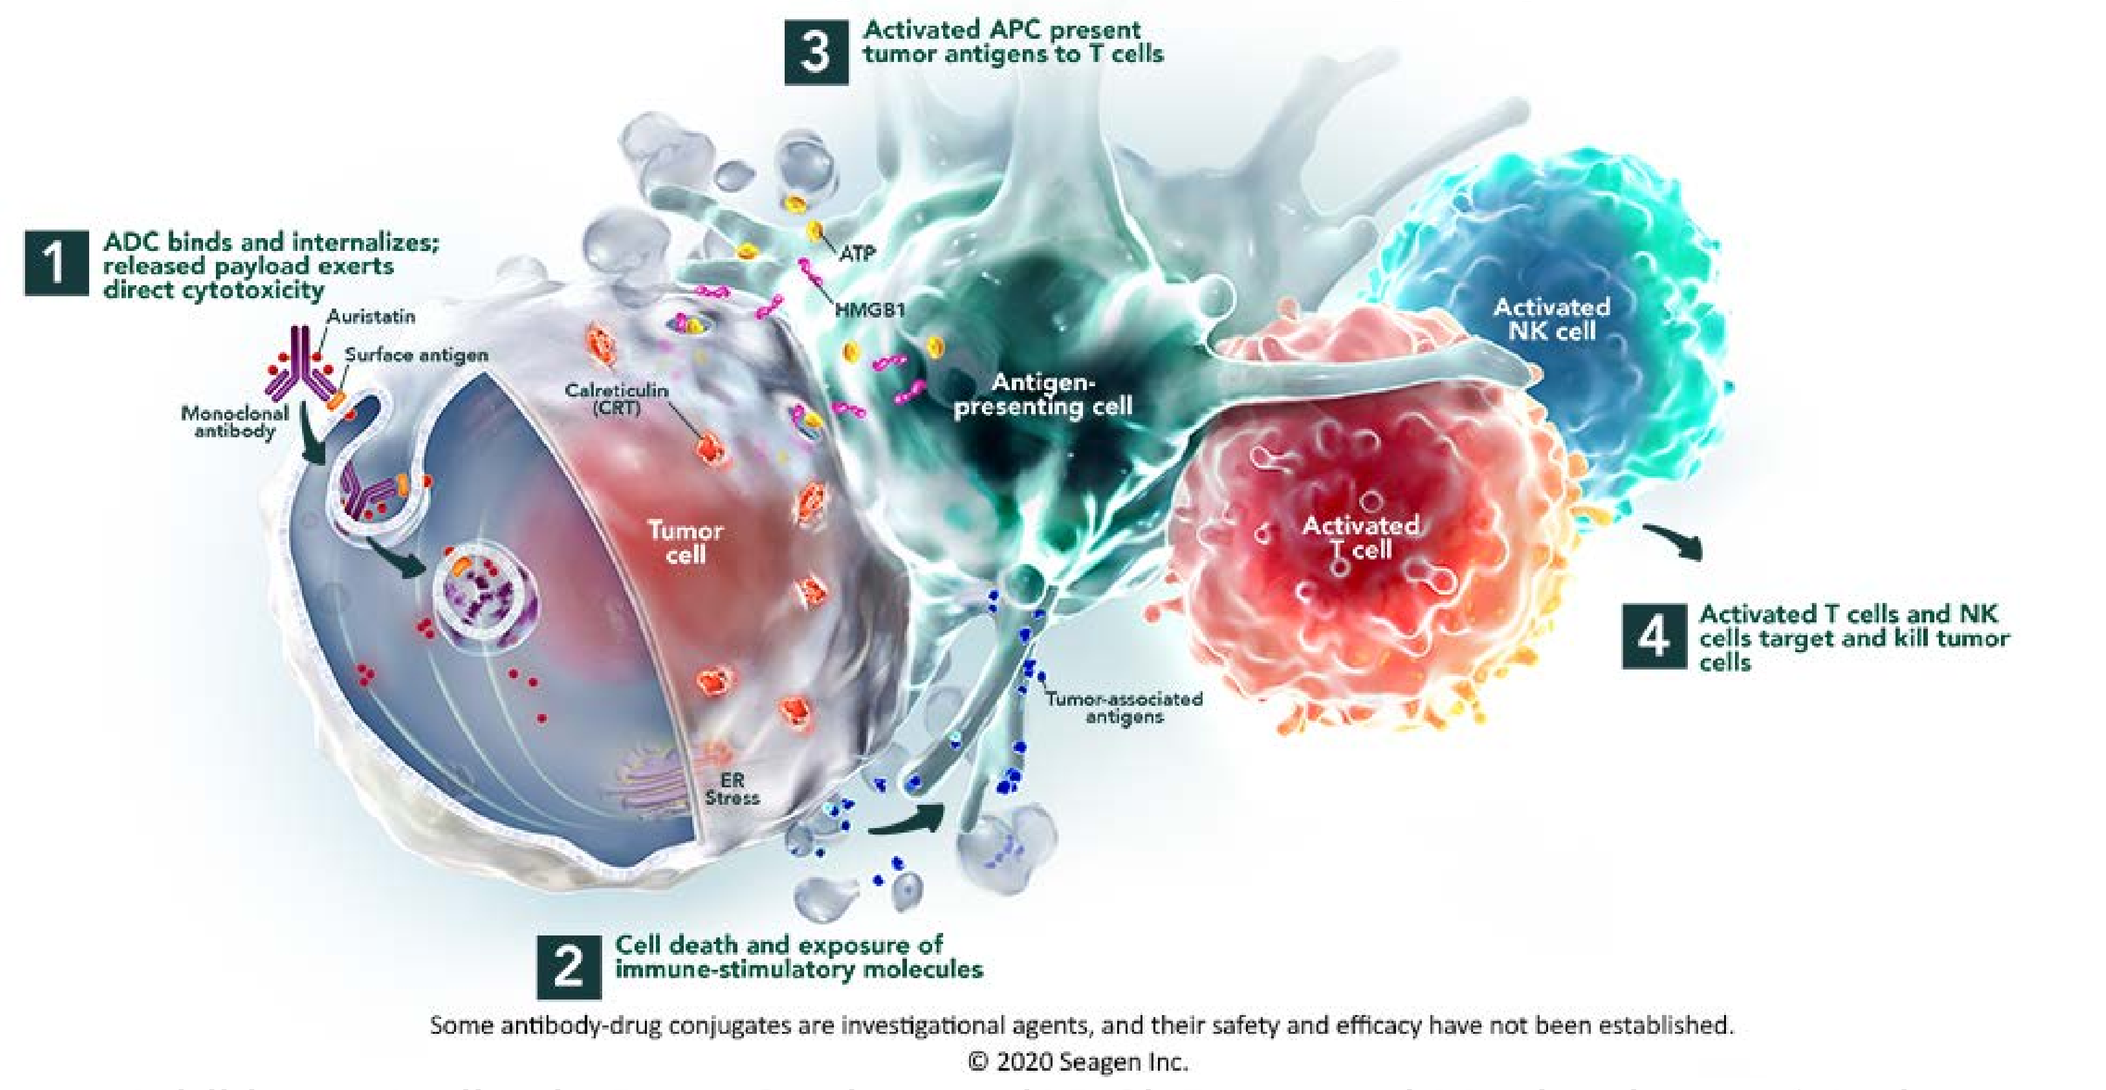
3 Activated APC present tumor antigens to T cells
1 ADC binds and internalizes; released payload exerts direct cytotoxicity
Auristatin
Surface antigen
Monoclonal antibody
Calreticulin (CRT)
ATP
HMGB1
Antigen-presenting cell
Activated NK cell
Tumor cell
Activated T cell
4 Activated T cells and NK cells target and kill tumor cells
Tumor-associated antigens
ER Stress
2 Cell death and exposure of immune-stimulatory molecules
Some antibody-drug conjugates are investigational agents, and their safety and efficacy have not been established.
© 2020 Seagen Inc.

Additional Antibody-Drug Conjugate (ADC) Proposed Mechanisms of Action

Payloads of ADC Programs in the Clinic

Vedotin and other clinically validated payloads

ADC Name	Abbreviations	Antibody	Payload Class	Disease Indication	Clinical Stage
Brentuximab vedotin (SGN-35)	BV	Brentuximab	MMAE (Auristatin)	Hodgkins Lymphoma	FDA Approved
Enfortumab vedotin (ASG22ME)	EV	Enfortumab	MMAE (Auristatin)	Urothelial	FDA Approved
Tisotumab vedotin (TF-011-MMAE)	TV	Tisotumab	MMAE (Auristatin)	Endometrial	Phase II
Ladiratuzumab vedotin (SGN-LIV1A)	LV	Ladiratuzumab	MMAE (Auristatin)	Breast	Phase II
Belantamab mafodotin (GSK2857916)	BM	Belantamab	MMAF (Auristatin)	Multiple Myeloma	FDA Approved
Trastuzumab emtansine (T-DM1)	T-DM1	Trastuzumab	DM1 (Maytansine)	Breast	FDA Approved
Trastuzumab deruxtecan (DS-8201)	T-Ex	Trastuzumab	Ex (Camptothecin)	Breast	FDA Approved

Vedotin ADCs Induce ICD Markers Across Multiple Cancer Types

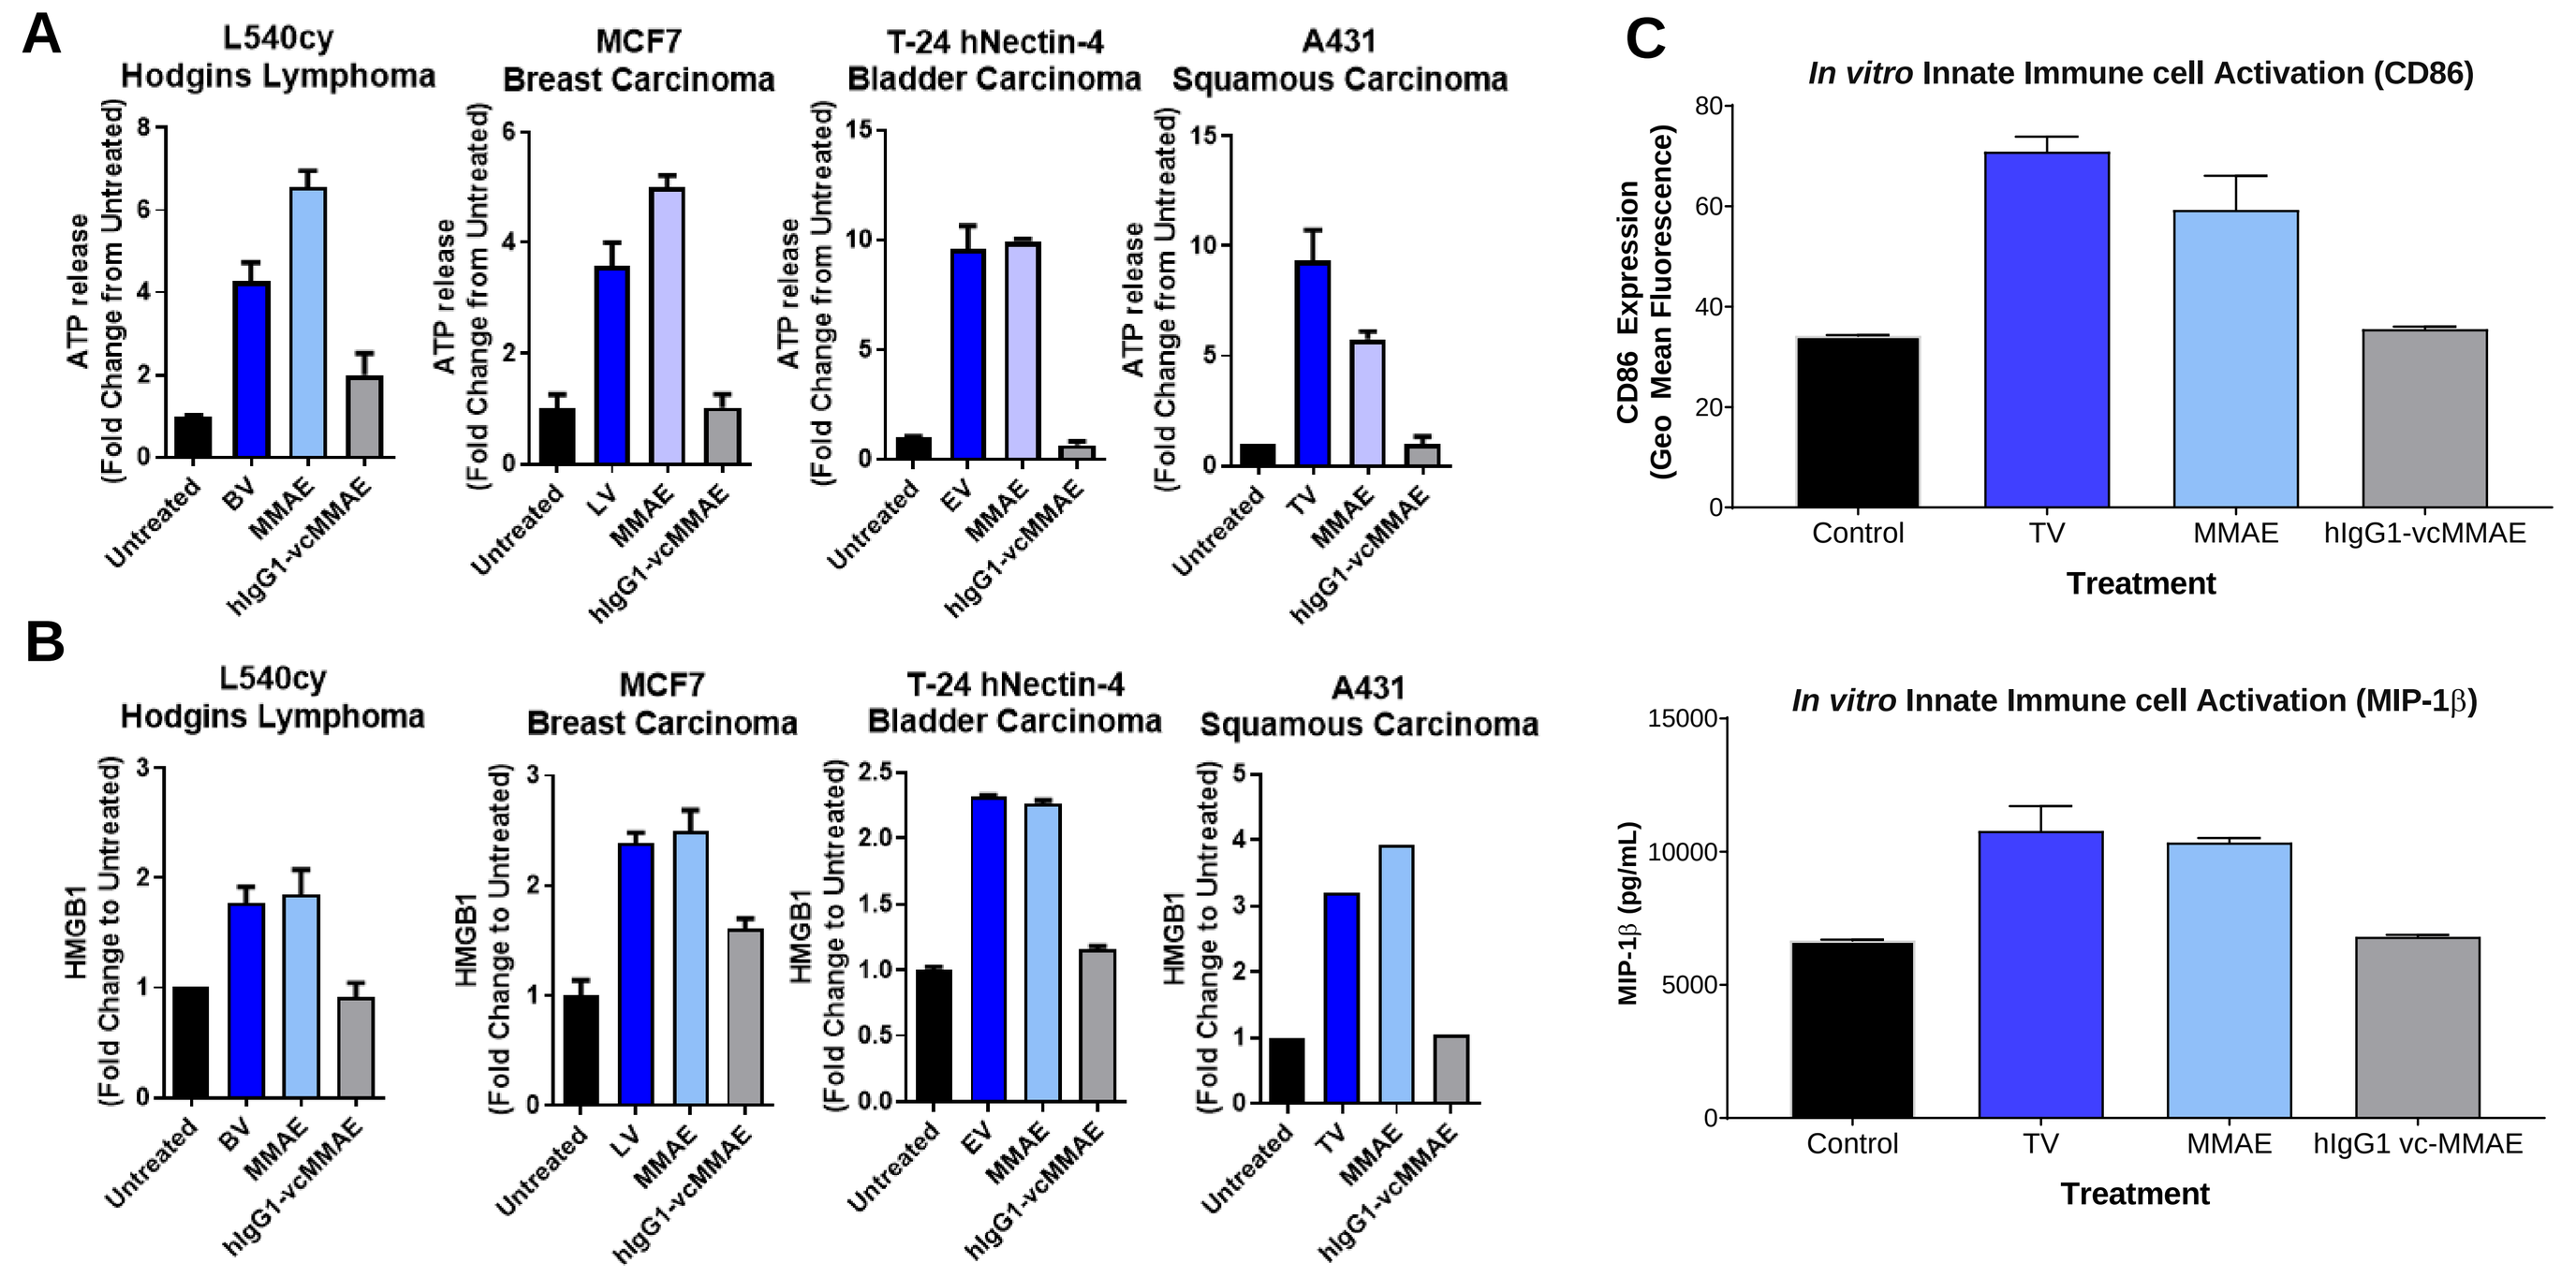
A
L540cy Hodgins Lymphoma
MCF7 Breast Carcinoma
T-24 hNectin-4 Bladder Carcinoma
A431 Squamous Carcinoma
ATP release (Fold Change from Untreated)
Untreated
BV
LV
EV
TV
MMAE
hIgG1-vcMMAE
B
HMGB1 (Fold Change to Untreated)
C
In vitro Innate Immune cell Activation (CD86)
CD86 Expression (Geo Mean Fluorescence)
Control
TV
MMAE
hIgG1-vcMMAE
Treatment
In vitro Innate Immune cell Activation (MIP-1β)
MIP-1β (pg/mL)
hIgG1 vc-MMAE

Figure 1. (A) Tumor cell lines cultured with the vedotin ADCs, free MMAE or non-binding ADC control (hIgG1-vcMMAE) demonstrate release of cytosolic ATP or (B) HMGB1 release in the media upon drug treatment, measured at 48hr. (C) Exemplary data of immune cell activation associated with ICD induced cell death is shown. Tumor cells treated with an ADC (TV) or non-binding ADC control were fed to PBMCs. Luminex cytokine analysis and flow cytometry were used to assess immune activation after 24hr.

Vedotin ADC Induces Immune Cell Recruitment and Activation *In Vivo*

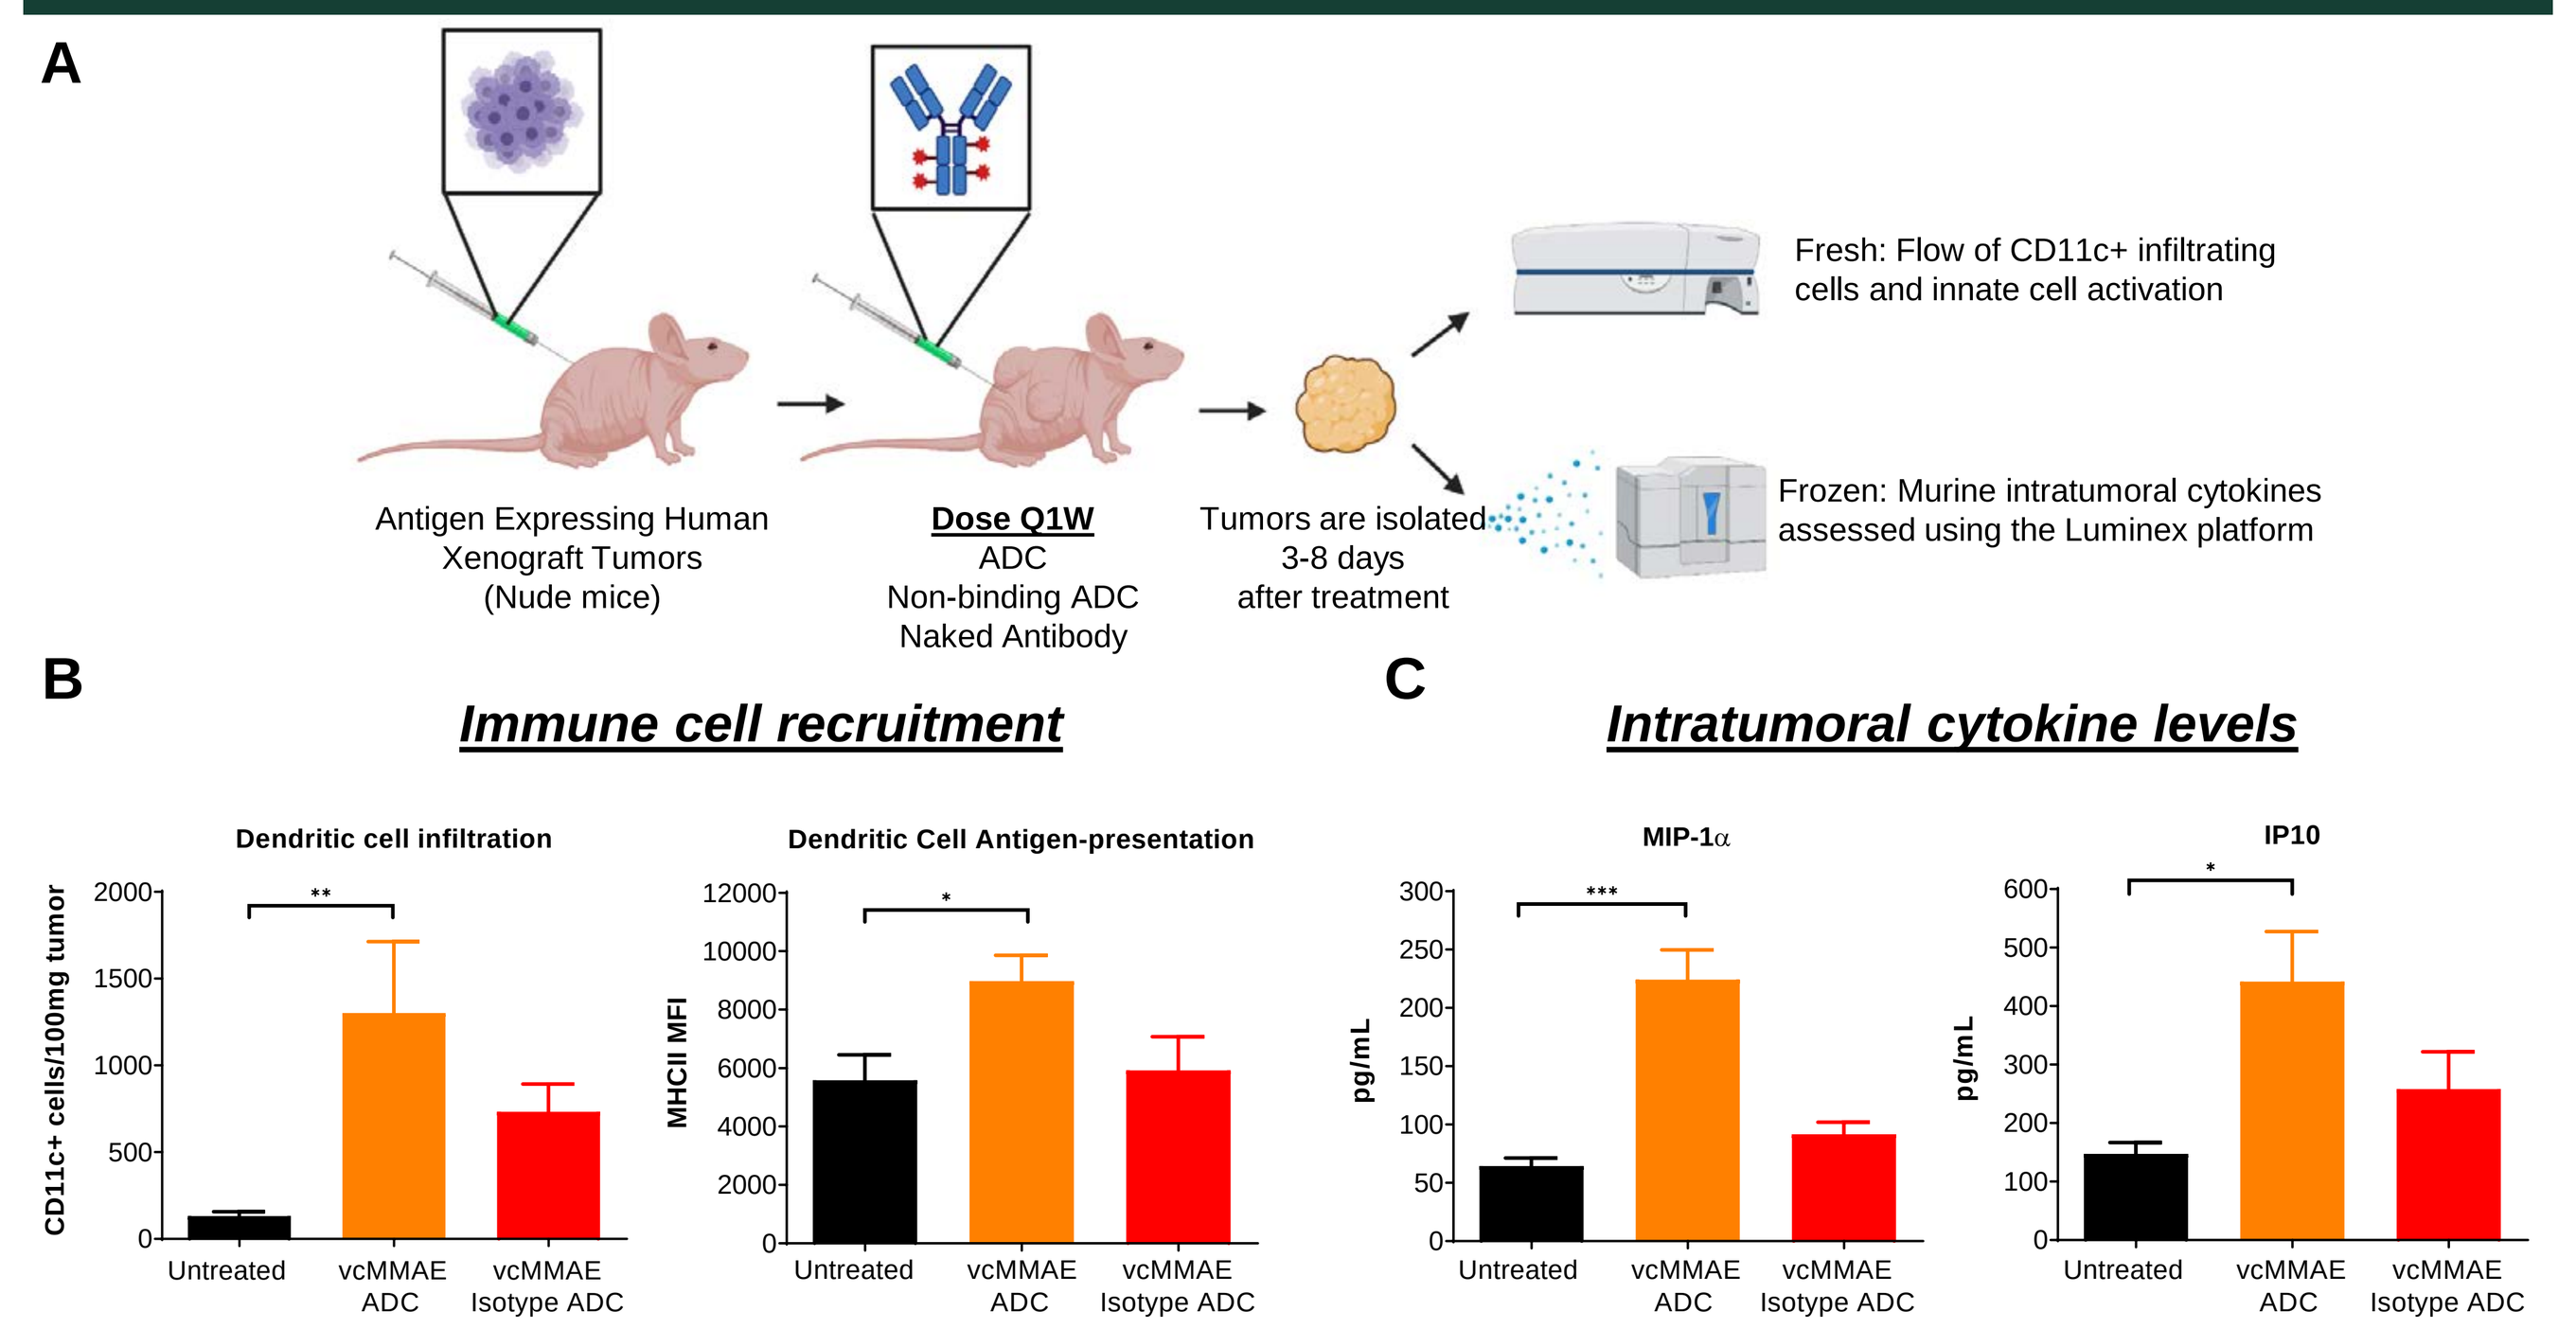
A
Antigen Expressing Human Xenograft Tumors (Nude mice)
Dose Q1W
ADC
Non-binding ADC
Naked Antibody
Tumors are isolated 3-8 days after treatment
Fresh: Flow of CD11c+ infiltrating cells and innate cell activation
Frozen: Murine intratumoral cytokines assessed using the Luminex platform
B
Immune cell recruitment
Dendritic cell infiltration
CD11c+ cells/100mg tumor
Dendritic Cell Antigen-presentation
MHCII MFI
C
Intratumoral cytokine levels
MIP-1α
IP10
pg/mL
Untreated
vcMMAE ADC
vcMMAE Isotype ADC

Figure 2. (A) Tumor xenografts isolated from animals treated with a vcMMAE ADC or non-binding vcMMAE Isotype ADC for 8 days, were dissociated, and subject to flow cytometry or cytokine profiling. (B) CD45 positive immune cells were stained for CD11c and activation observed by staining for the expression of MHC Class II on the cell surface. (C) Intratumoral cytokines were measured by Luminex.

Vedotin ADCs Upregulate MHC Class I in Tumors

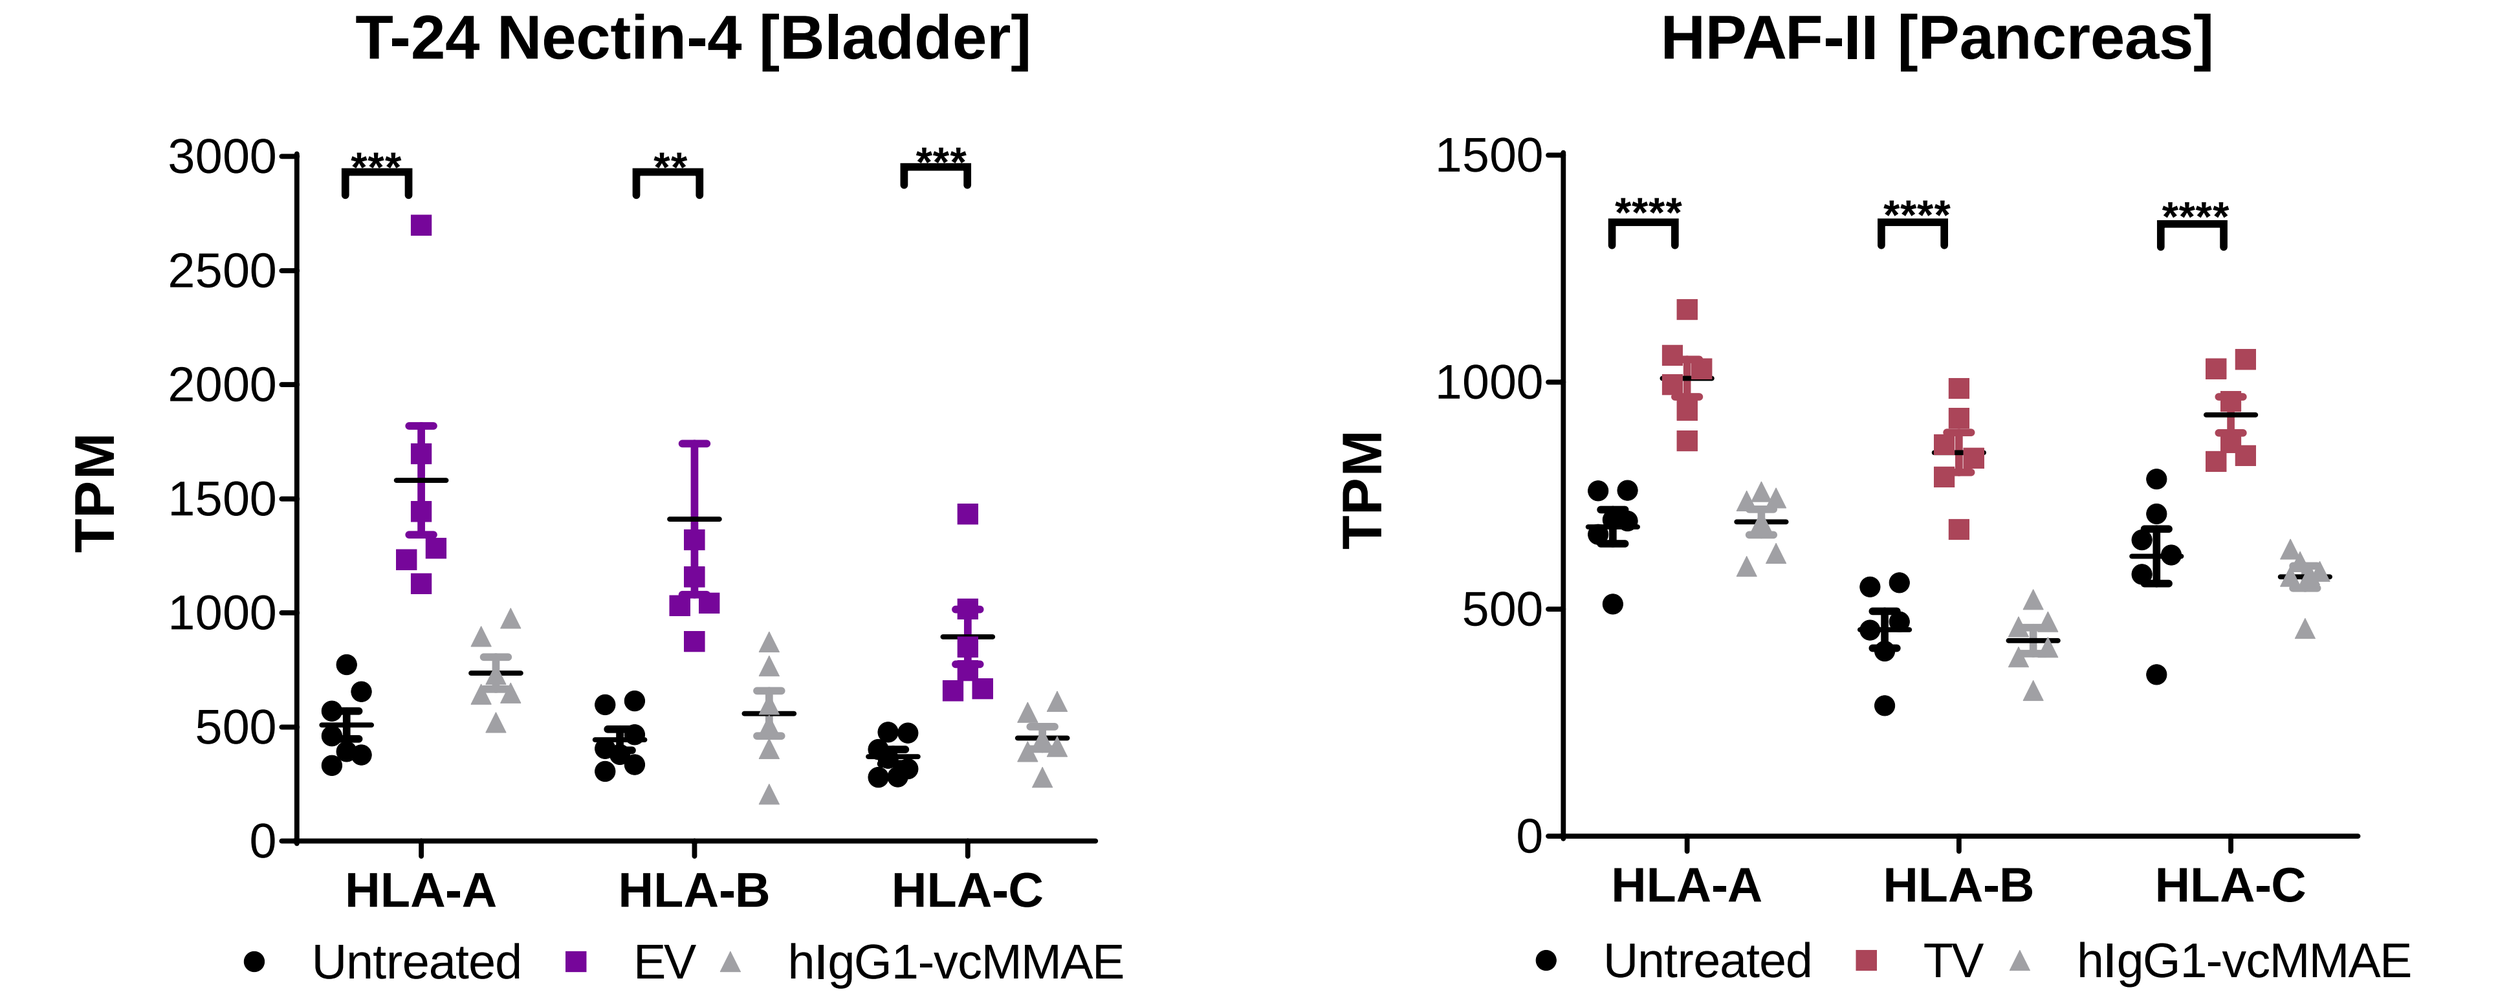
T-24 Nectin-4 [Bladder]
HPAF-II [Pancreas]
TPM
HLA-A
HLA-B
HLA-C
Untreated
EV
TV
hIgG1-vcMMAE

Figure 3. Tumor xenografts from animals treated with EV, TV or non-binding ADC control (hIgG1-vcMMAE) were isolated 5 days post treatment and transcripts for human MHC Class I (HLA-A,B,C) determined by RNA-seq. TPM, total reads per million.

Vedotin ADCs Promote Immune Memory for Anti-Tumor Response

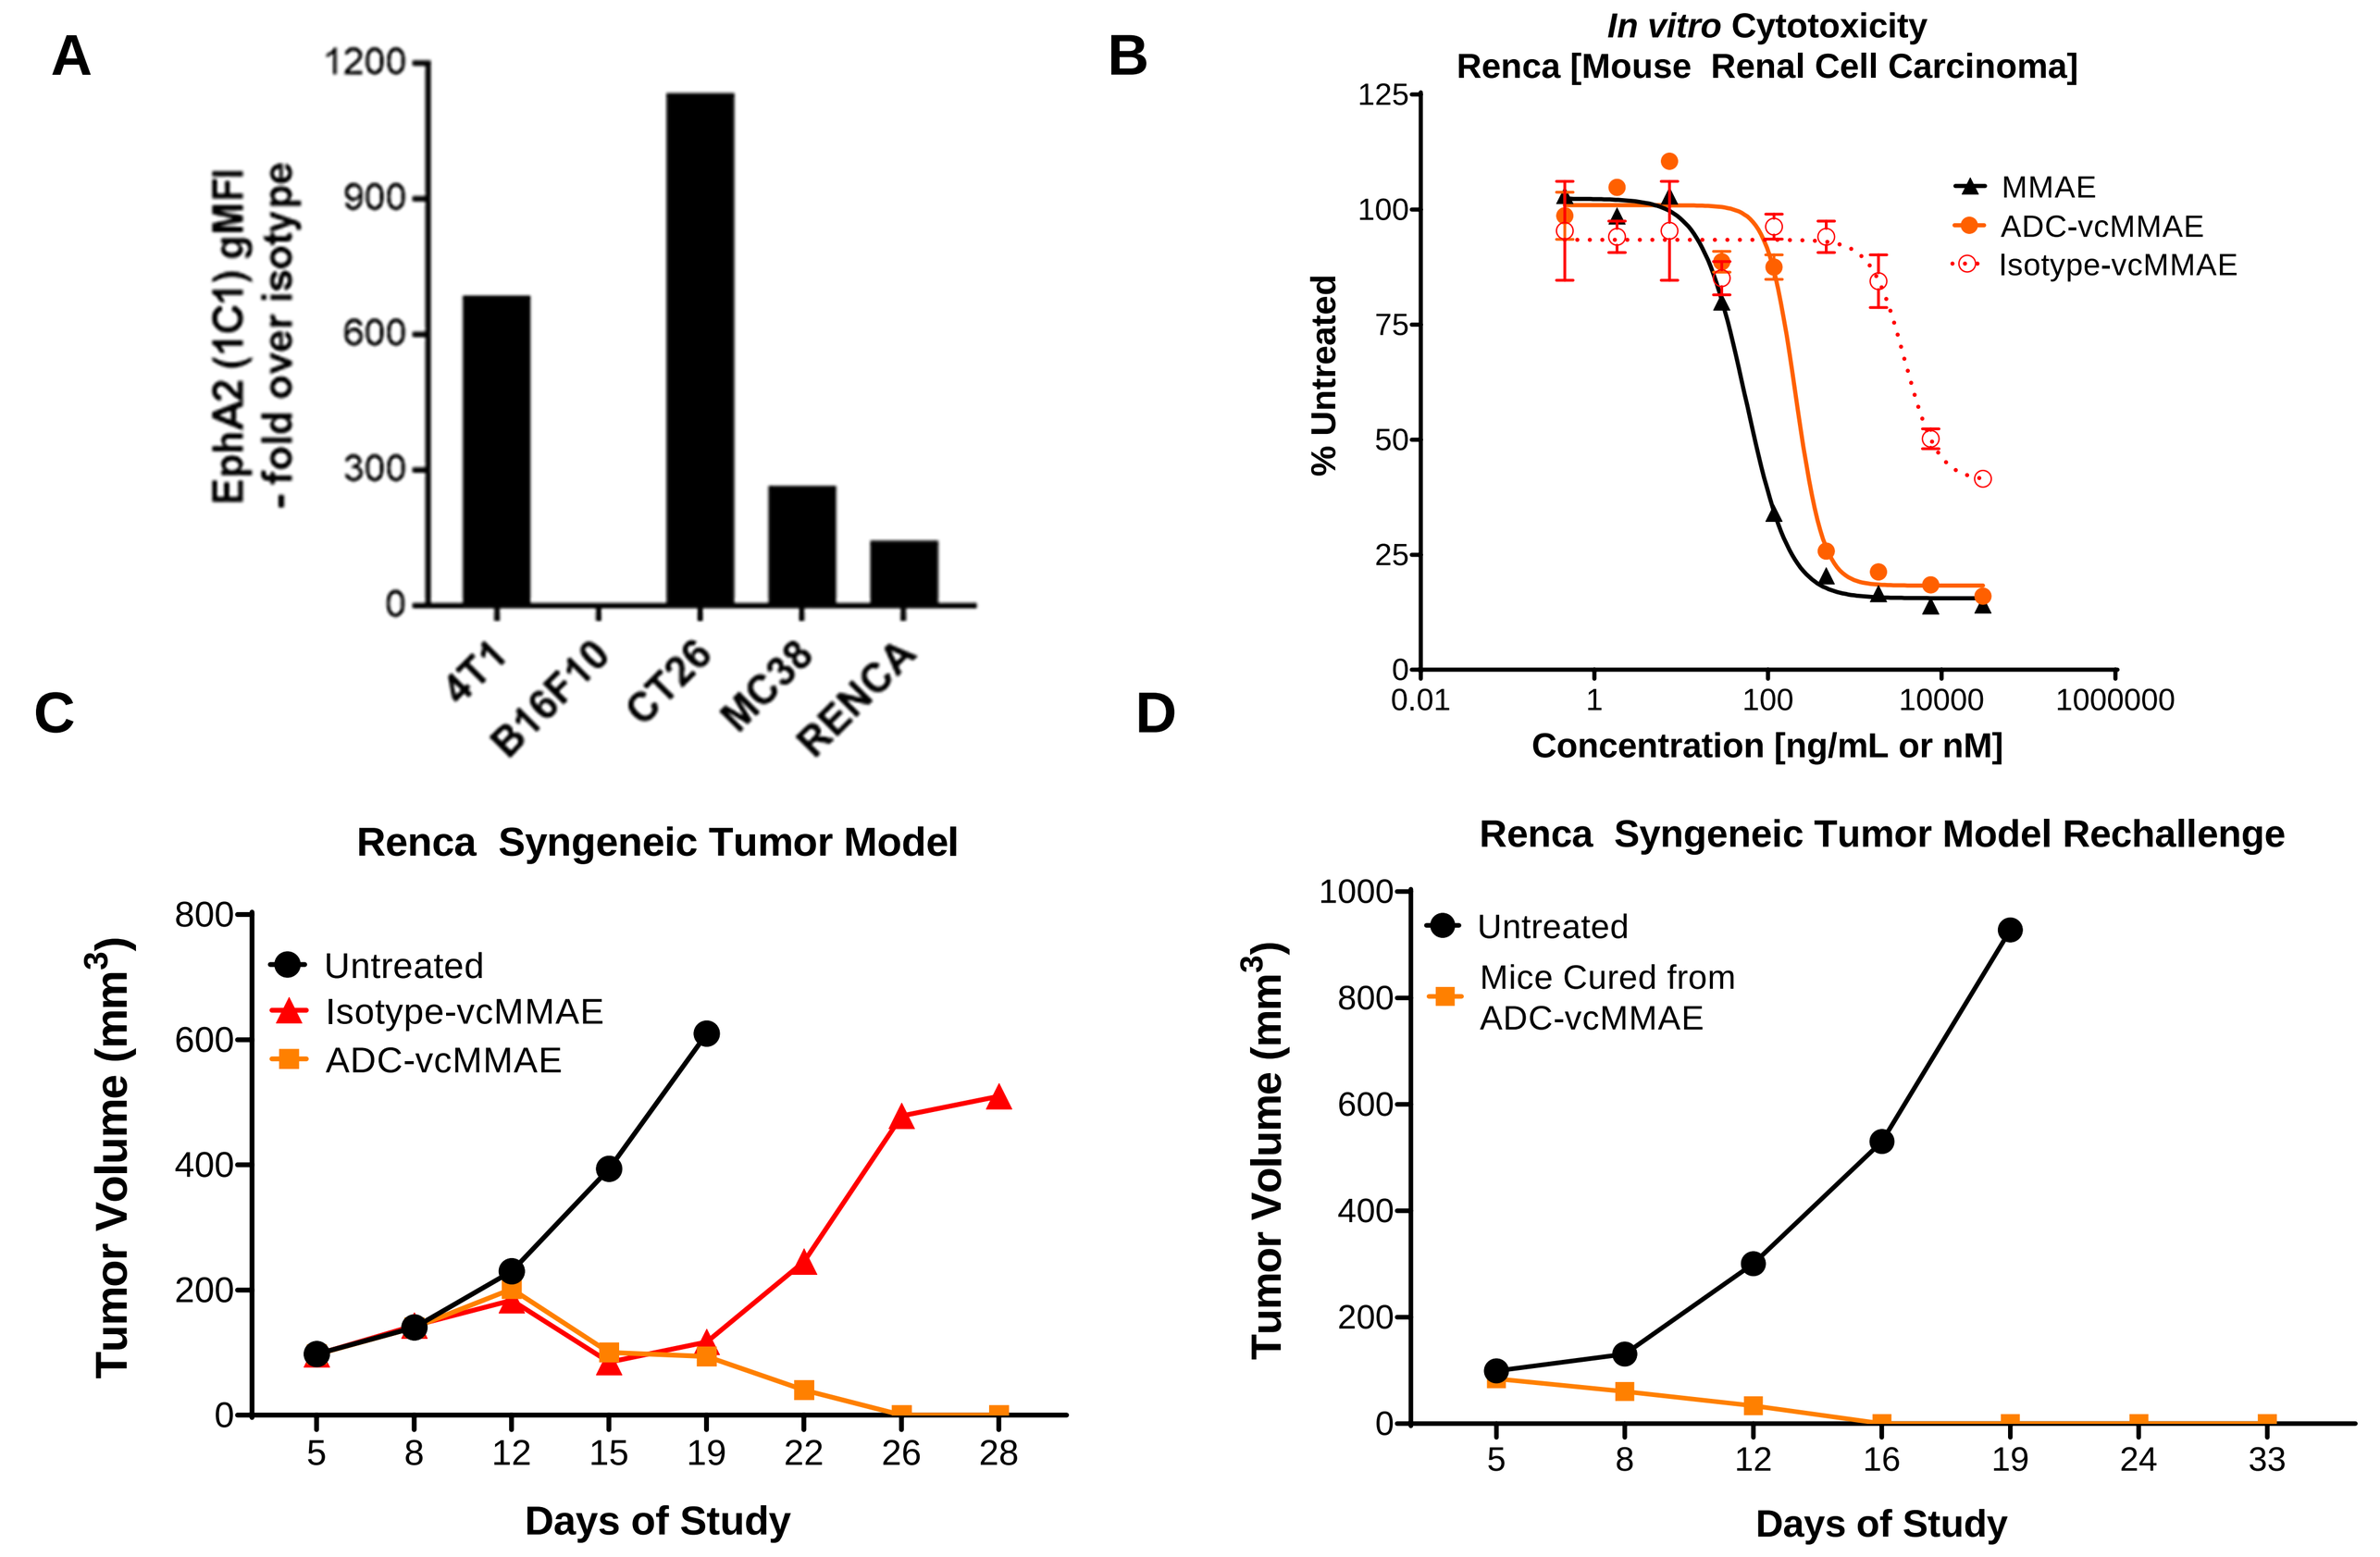
A
EphA2 (1C1) gMFI -fold over isotype
4T1
B16F10
CT26
MC38
RENCA
B
In vitro Cytotoxicity
Renca [Mouse Renal Cell Carcinoma]
% Untreated
MMAE
ADC-vcMMAE
Isotype-vcMMAE
Concentration [ng/mL or nM]
C
Renca Syngeneic Tumor Model
Tumor Volume (mm³)
Untreated
Isotype-vcMMAE
ADC-vcMMAE
Days of Study
D
Renca Syngeneic Tumor Model Rechallenge
Untreated
Mice Cured from ADC-vcMMAE

Figure 4. (A) Murine EphA2 expression on indicated syngeneic cell lines by flow cytometry. (B) Evaluation of EphA2-vcMMAE ADC in standard 96hr *in vitro* cytotoxicity assay (ng/mL). MMAE was also tested (nM). (C) Evaluation of the anti-tumor activity of EphA2-vcMMAE ADC *in vivo* in a Renca tumor model. (D) Upon re-challenge, tumor implants were rejected.

ER Stress Induction is Important for ICD and Superior for Vedotin ADCs

A

Payload Class	Members	Average Drug Load	Mechanism of Action
Auristatins	MMAE (vedotin)	4	Microtubule Disruption
	MMAF (mafadotin)	4	
Maytansines	DM1 (emtansine)	4	
Camptothecins	Ex (exatecan)	8	Topoisomerase Inhibition

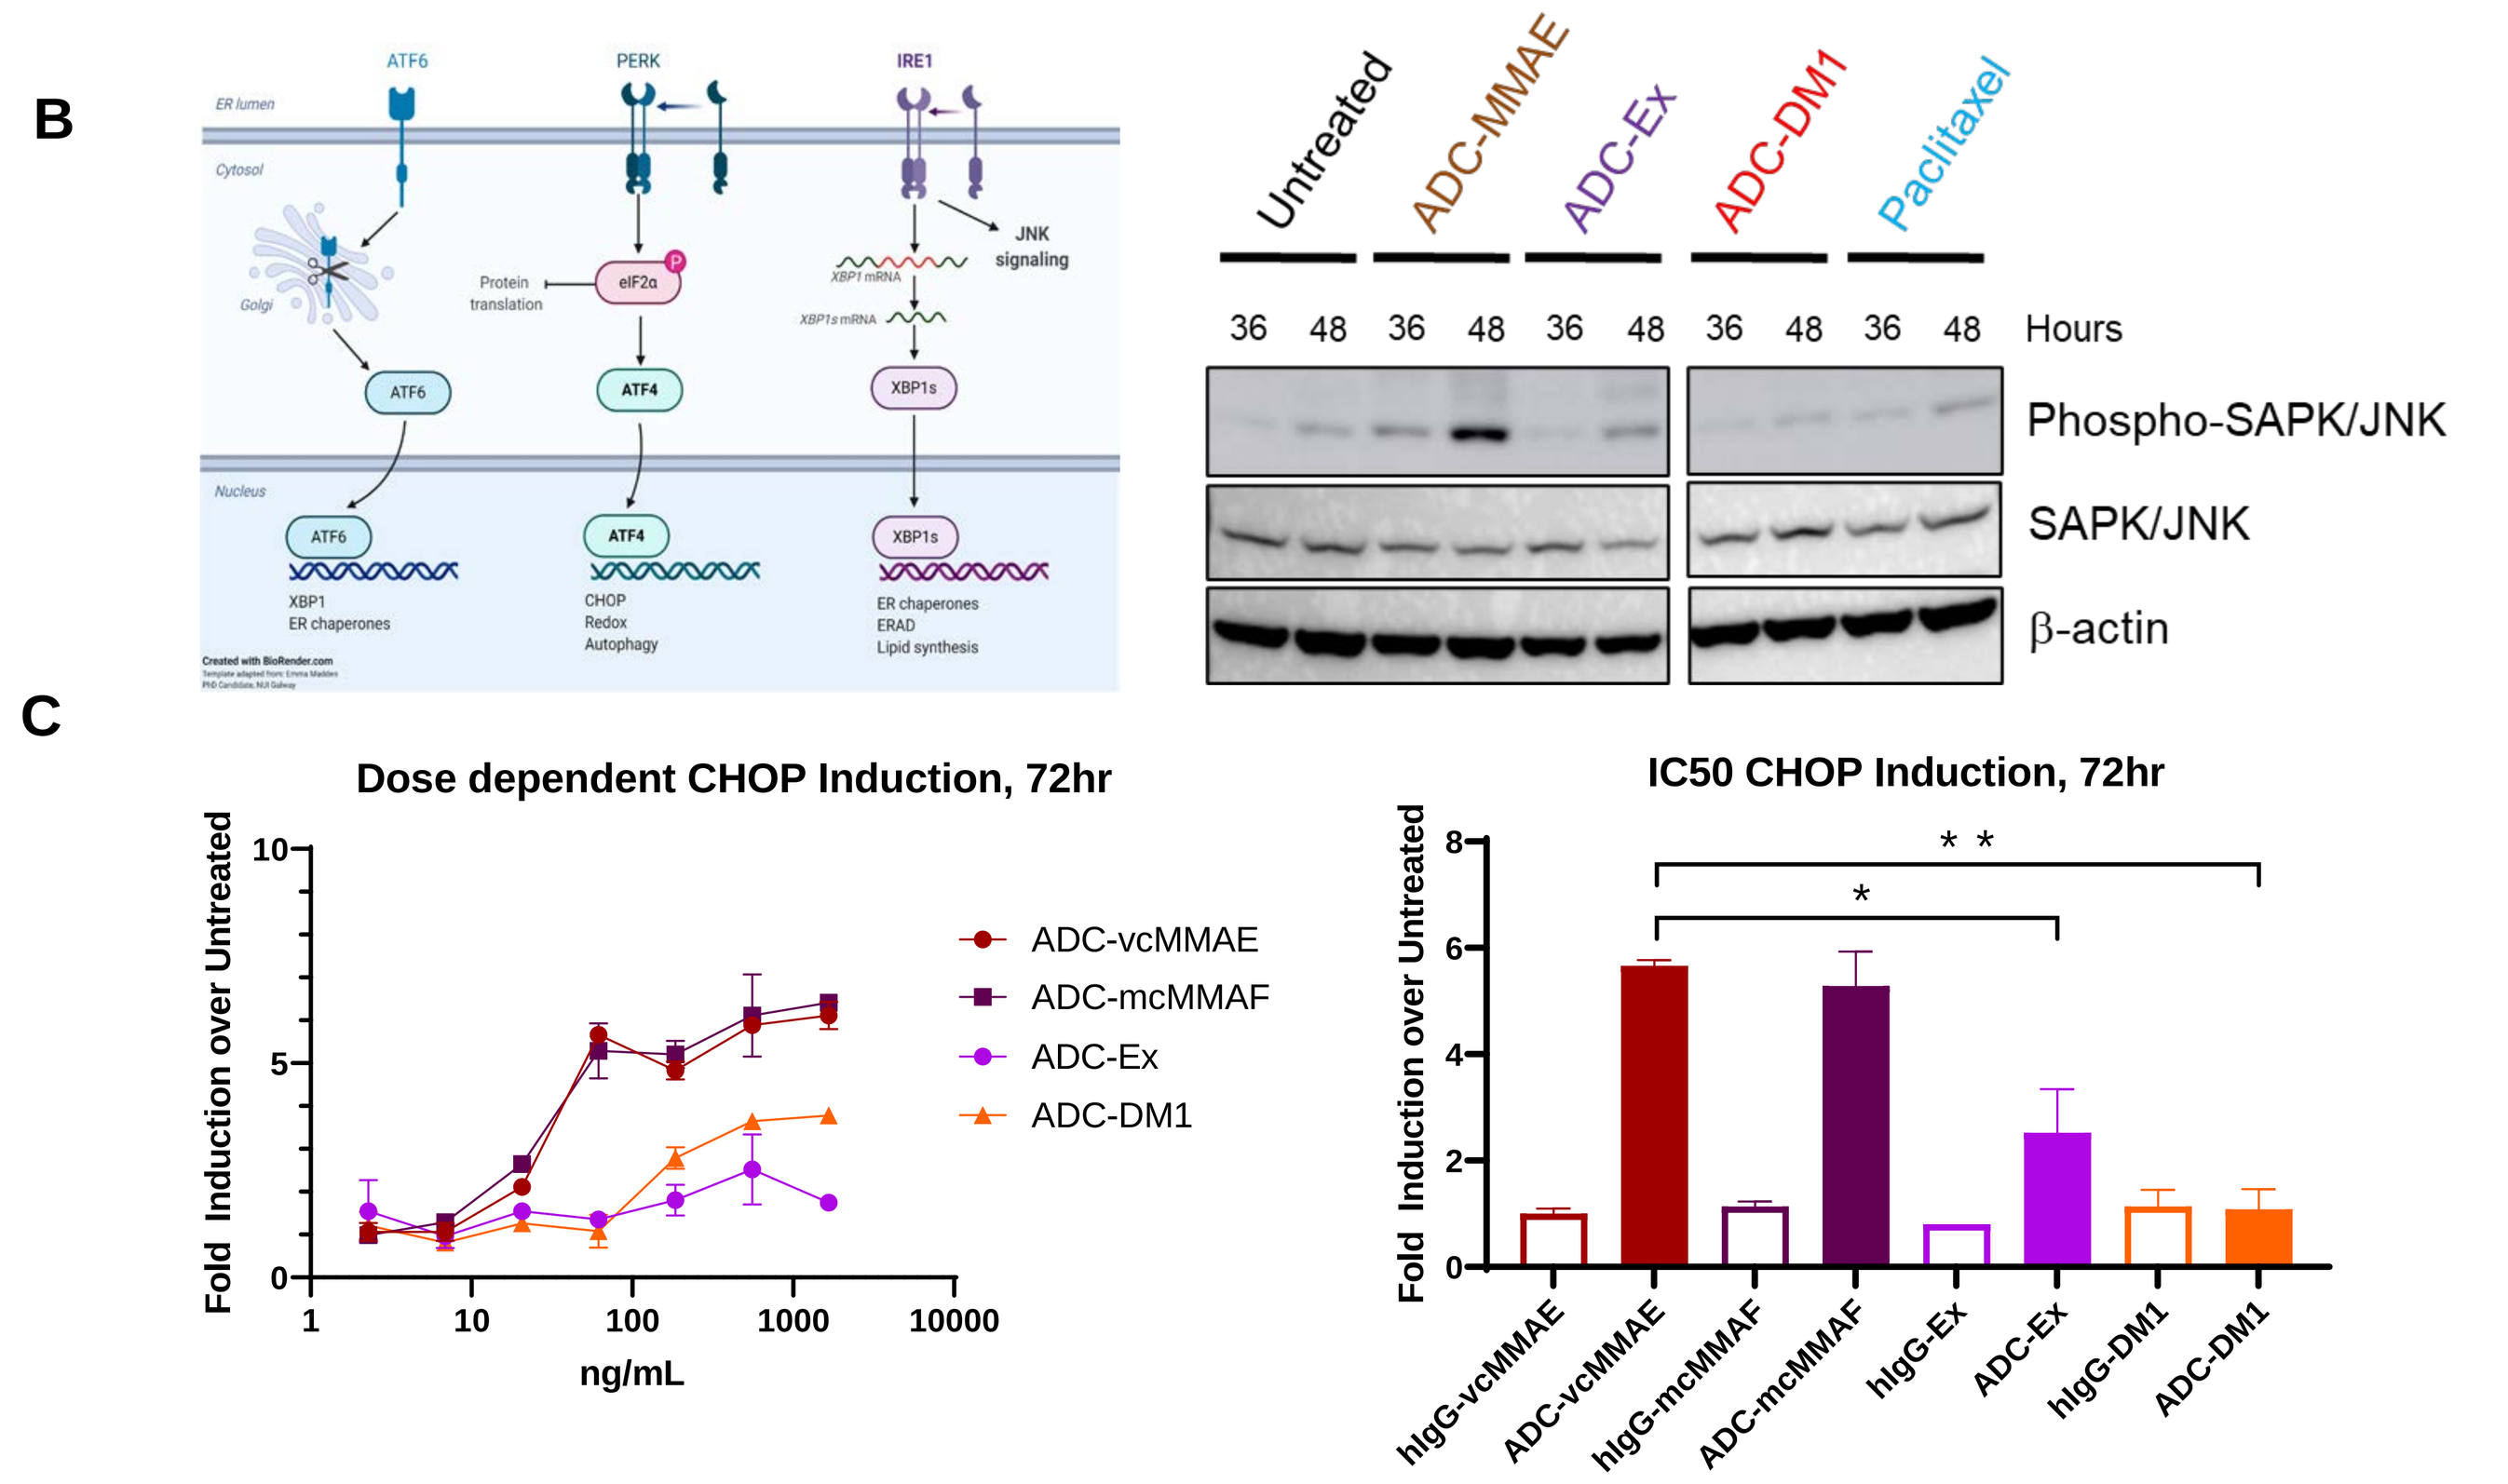
B
ATF6
PERK
IRE1
JNK signaling
Untreated
ADC-MMAE
ADC-Ex
ADC-DM1
Paclitaxel
36 48 Hours
Phospho-SAPK/JNK
SAPK/JNK
β-actin
C
Dose dependent CHOP Induction, 72hr
Fold Induction over Untreated
ng/mL
ADC-vcMMAE
ADC-mcMMAF
ADC-Ex
ADC-DM1
IC50 CHOP Induction, 72hr
hIgG-vcMMAE
ADC-vcMMAE
hIgG-mcMMAF
ADC-mcMMAF
hIgG-Ex
ADC-Ex
hIgG-DM1
ADC-DM1

Figure 5. (A) Table of various clinically approved ADC payloads. (B) Graphic of ER stress signaling response and Western blot analysis of MIA-PaCa-2 cells treated with ADC, paclitaxel at IC50 concentrations for 36hr or 48hr. (C) MIA-PaCa-2 cells expressing CHOP-driven luciferase reporter (Signosis, Inc.) were treated with ADCs over a dose range or at IC50 dose (cytotoxicity). CHOP induction is expressed by fold induction compared to untreated cells.

ICD Potential of Clinical ADC Payloads

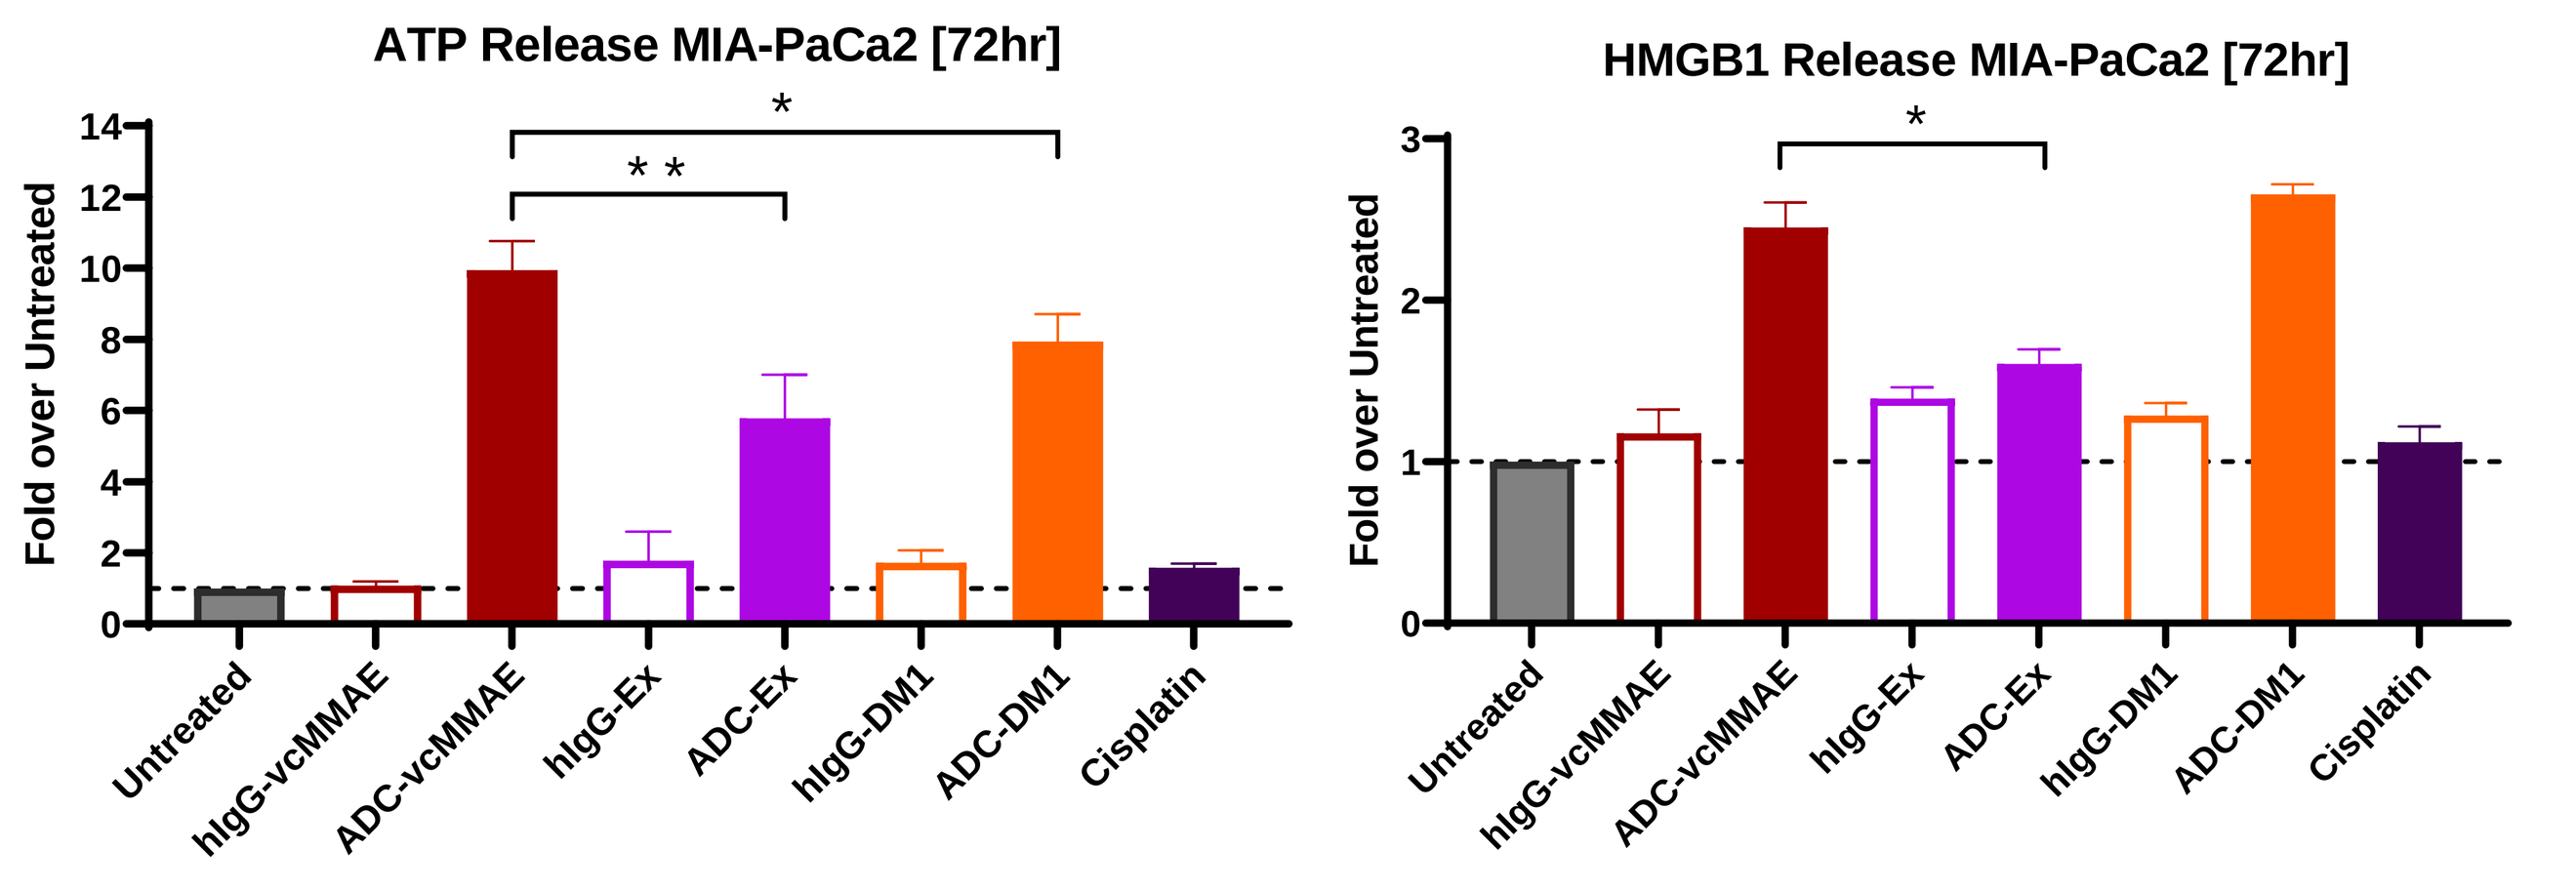
ATP Release MIA-PaCa2 [72hr]
HMGB1 Release MIA-PaCa2 [72hr]
Fold over Untreated
Untreated
hIgG-vcMMAE
ADC-vcMMAE
hIgG-Ex
ADC-Ex
hIgG-DM1
ADC-DM1
Cisplatin

Figure 6. Supernatants were collected from MIA-PaCa-2 pancreatic tumor cells treated with 1 μg/mL ADCs for 72hr and ATP release determined by Cell Titer Glo and HMGB1 secretion by ELISA was performed.

Immune Activation Assessment of ADC Payloads

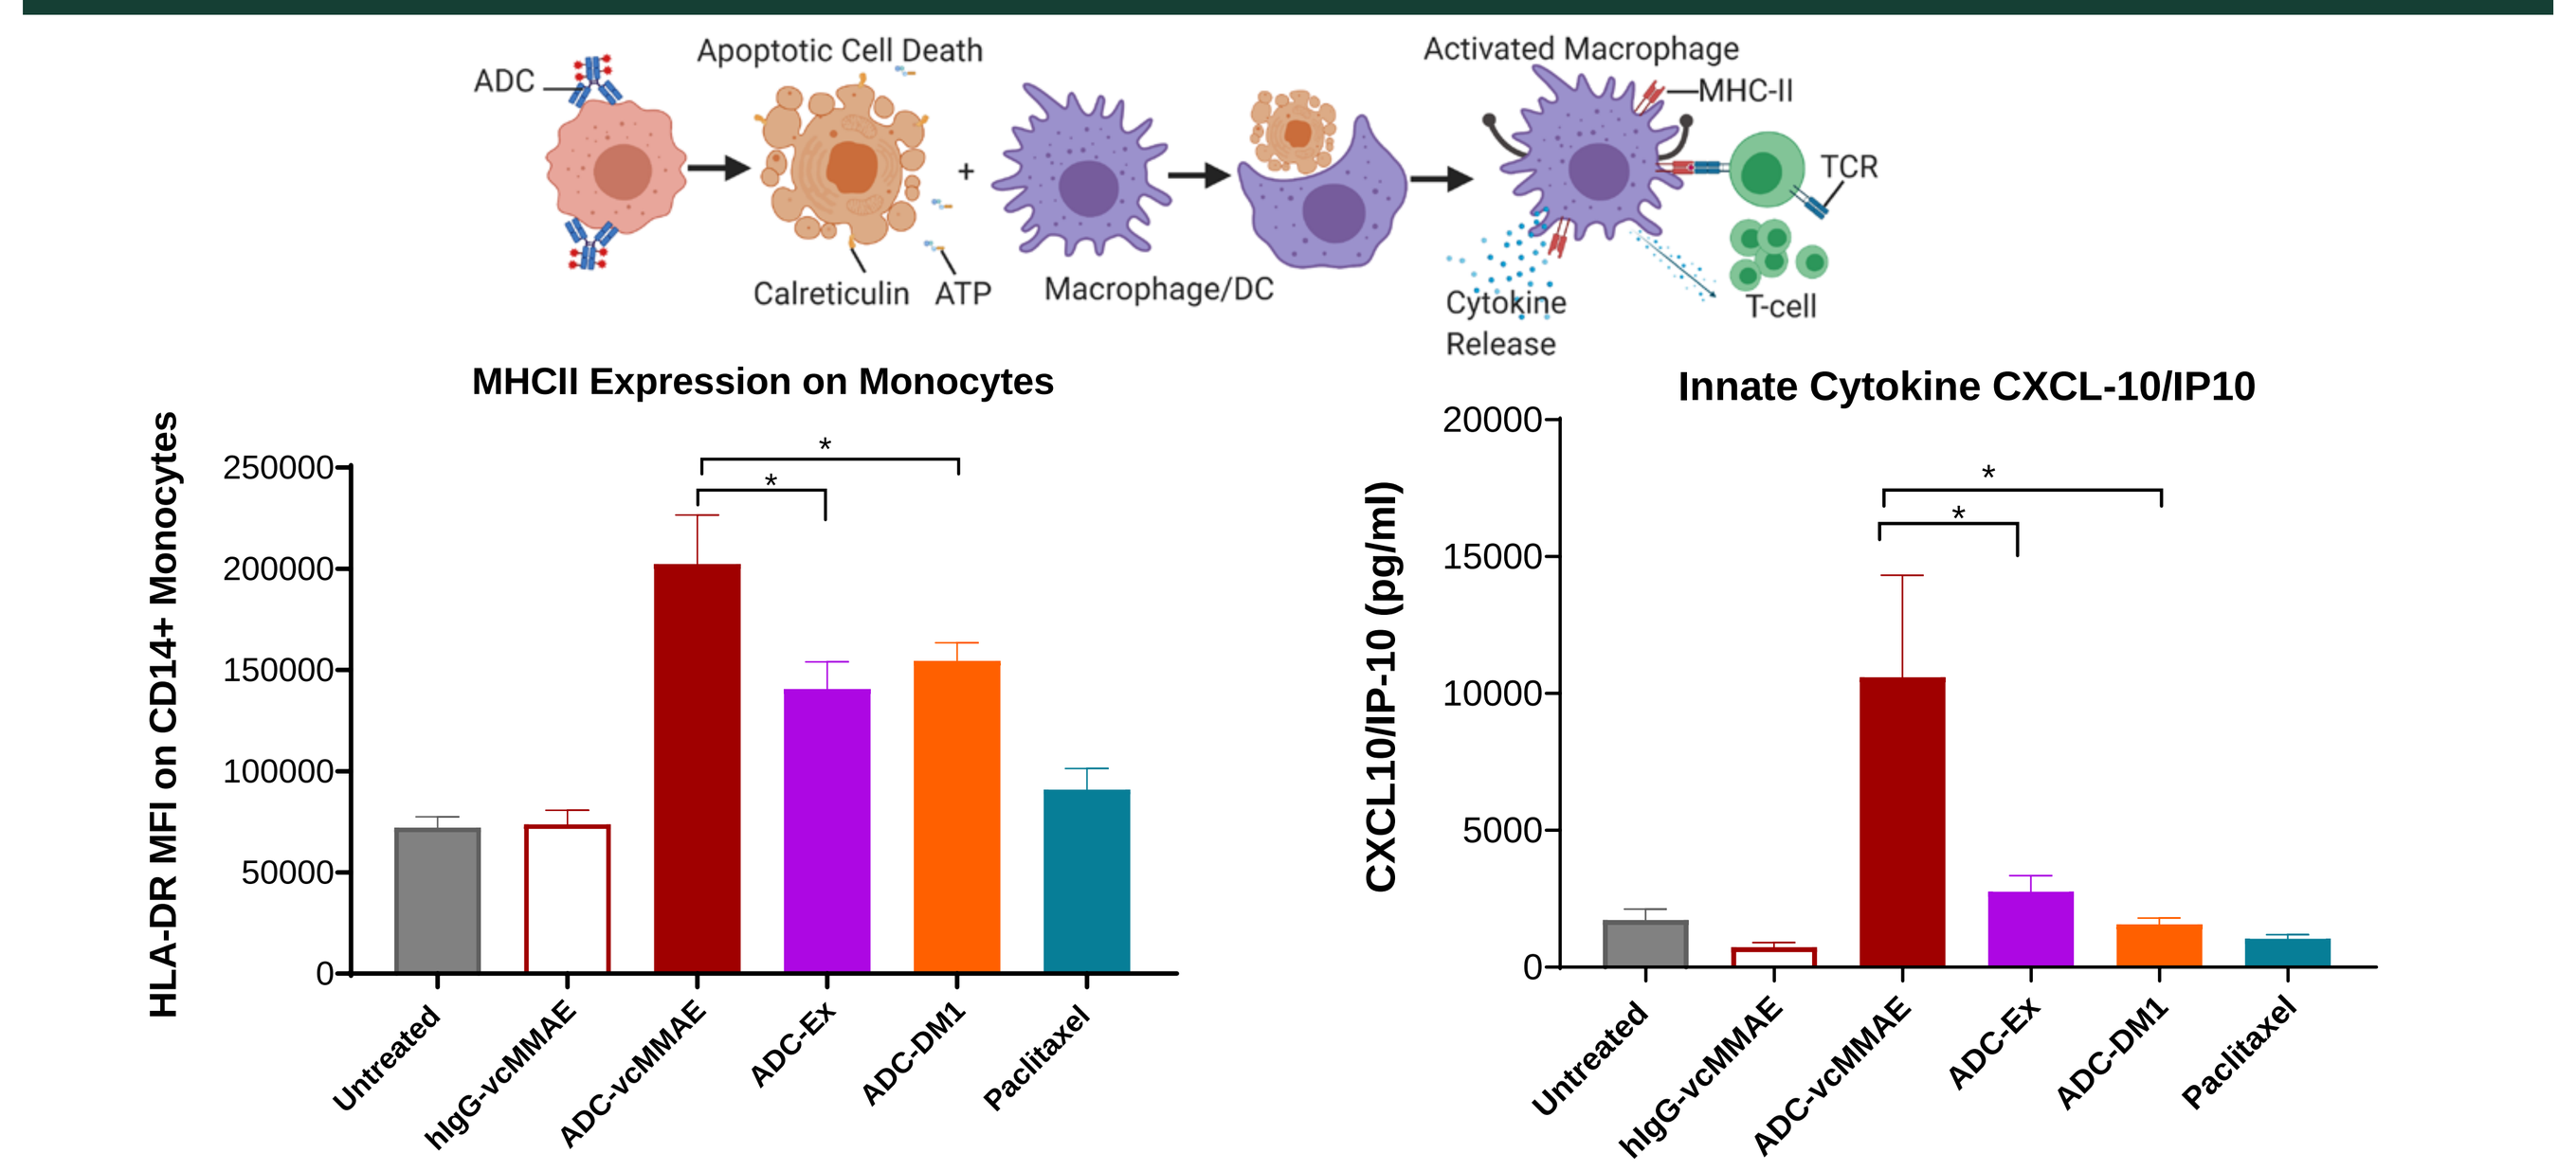
ADC
Apoptotic Cell Death
Calreticulin
ATP
Macrophage/DC
Activated Macrophage
MHC-II
TCR
Cytokine Release
T-cell
MHCII Expression on Monocytes
HLA-DR MFI on CD14+ Monocytes
Innate Cytokine CXCL-10/IP10
CXCL10/IP-10 (pg/ml)
Untreated
hIgG-vcMMAE
ADC-vcMMAE
ADC-Ex
ADC-DM1
Paclitaxel

Figure 7. Upregulation of MHC Class II (HLA-DR) on myeloid cells within PBMC was assessed by flow cytometry following 48hr co-incubation of PBMC with L540cy cells dosed with ADCs (24hr IC50 concentration). 24hr supernatants were assessed by Luminex multiplex assay for cytokine levels.

Payload Evaluation on Trastuzumab Backbone

A

ADC	Antibody	Payload
T-DM1	Trastuzumab	DM1
T-MMAE	Trastuzumab	MMAE
T-Ex	Trastuzumab	Exatecan

Evaluation of ER Stress Pathway Signaling

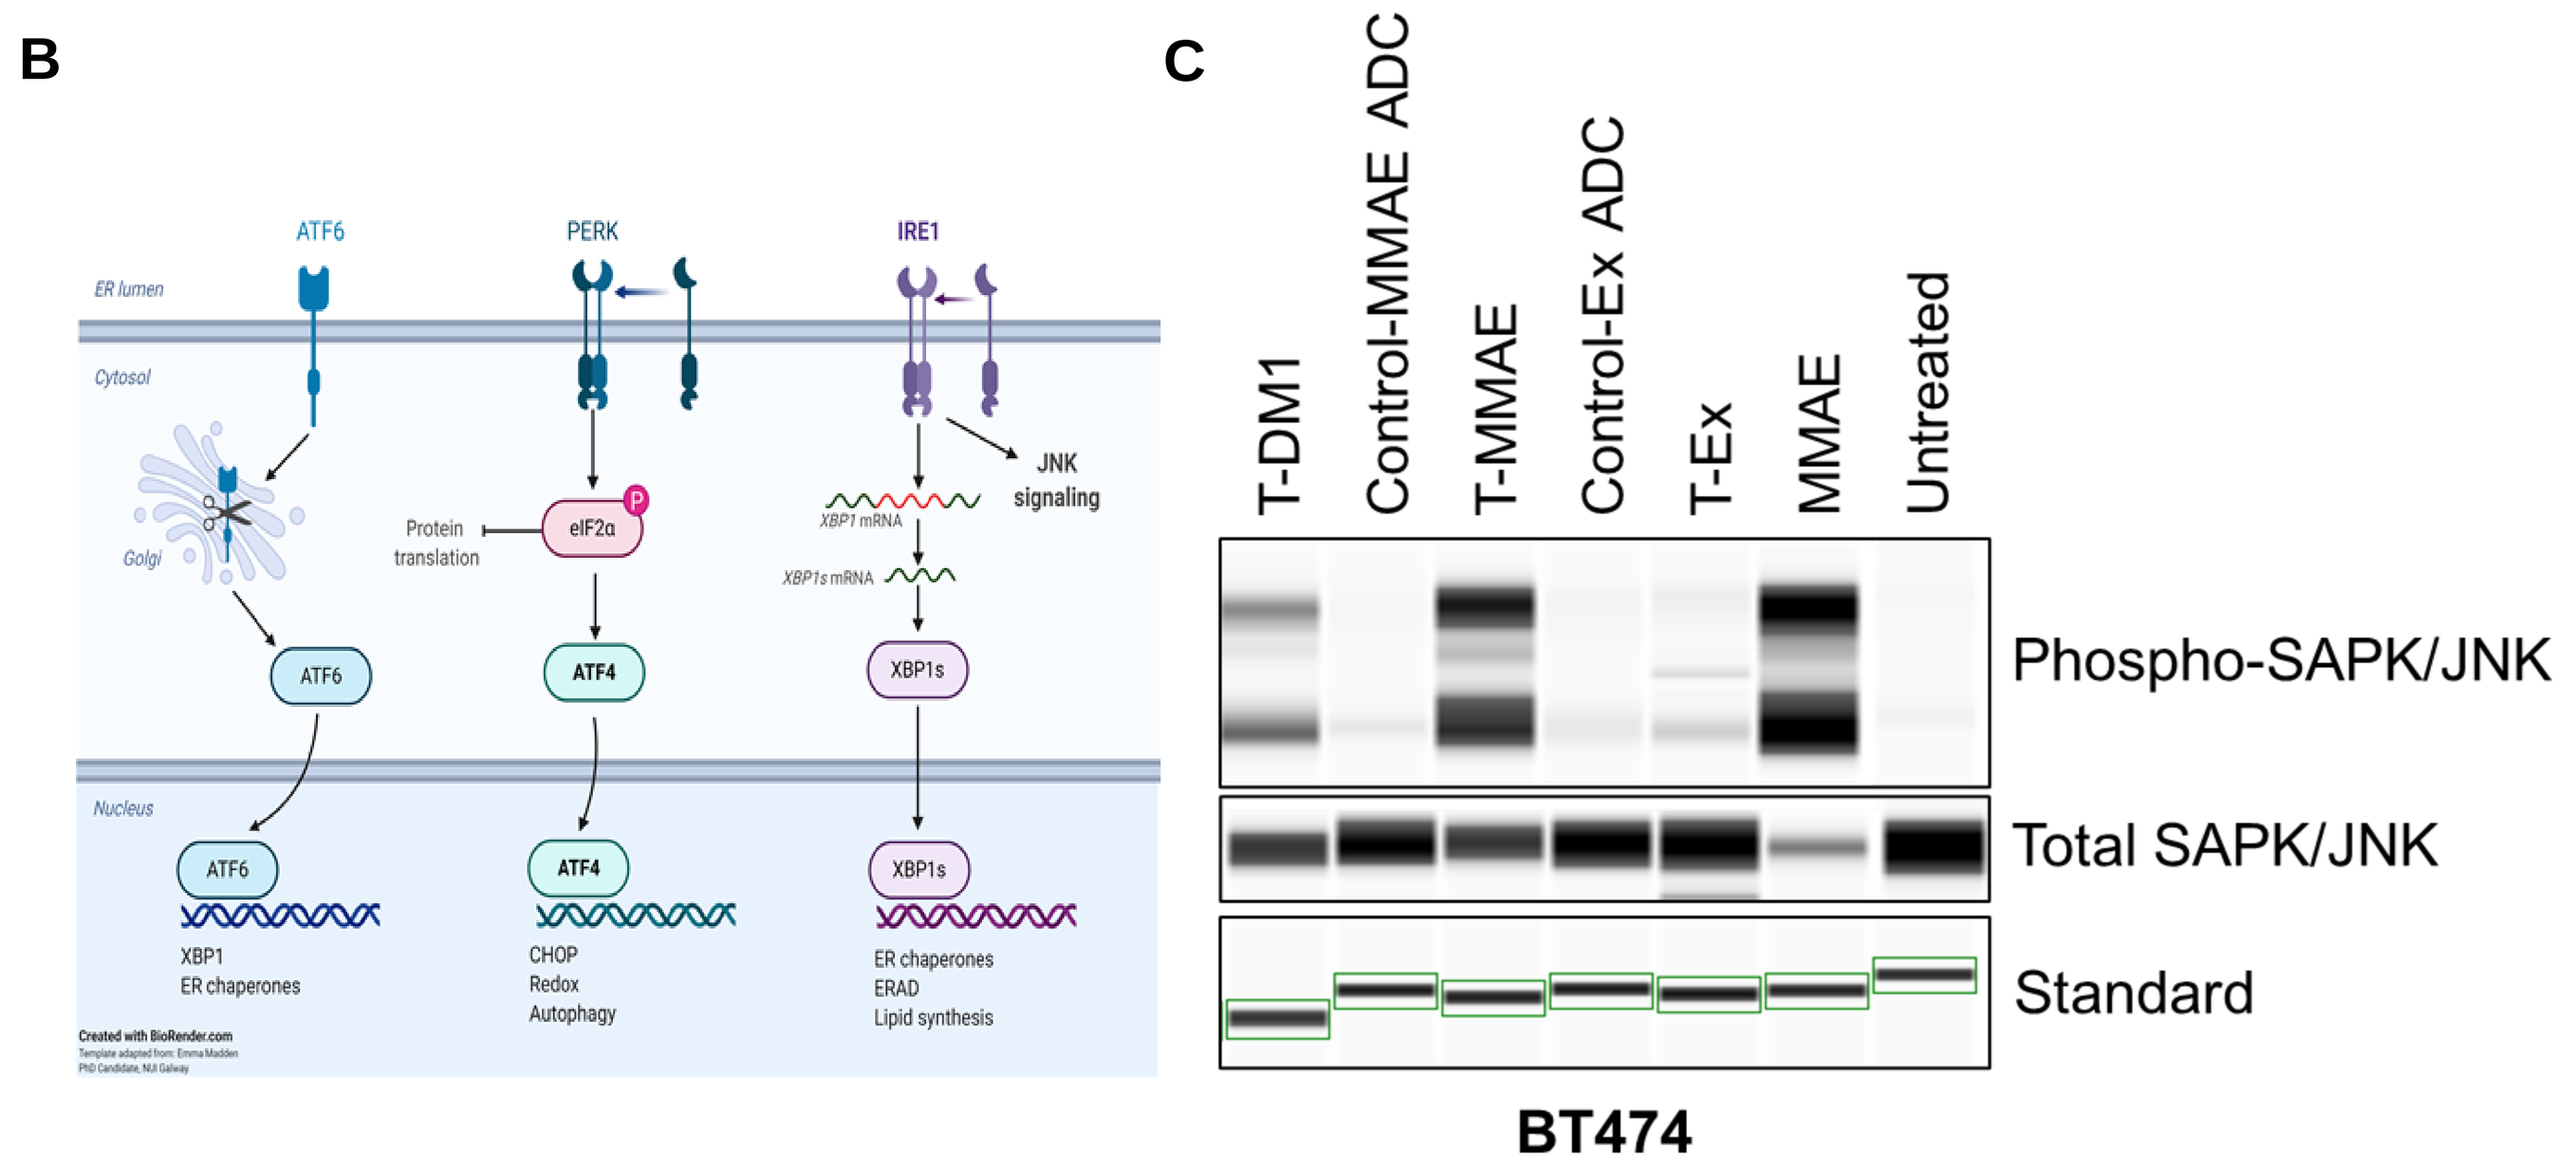
B
ATF6
PERK
IRE1
JNK signaling
C
T-DM1
Control-MMAE ADC
T-MMAE
Control-Ex ADC
T-Ex
MMAE
Untreated
Phospho-SAPK/JNK
Total SAPK/JNK
Standard
BT474

Vedotin ADC Demonstrate Strong Activation of Multiple ICD Hallmarks

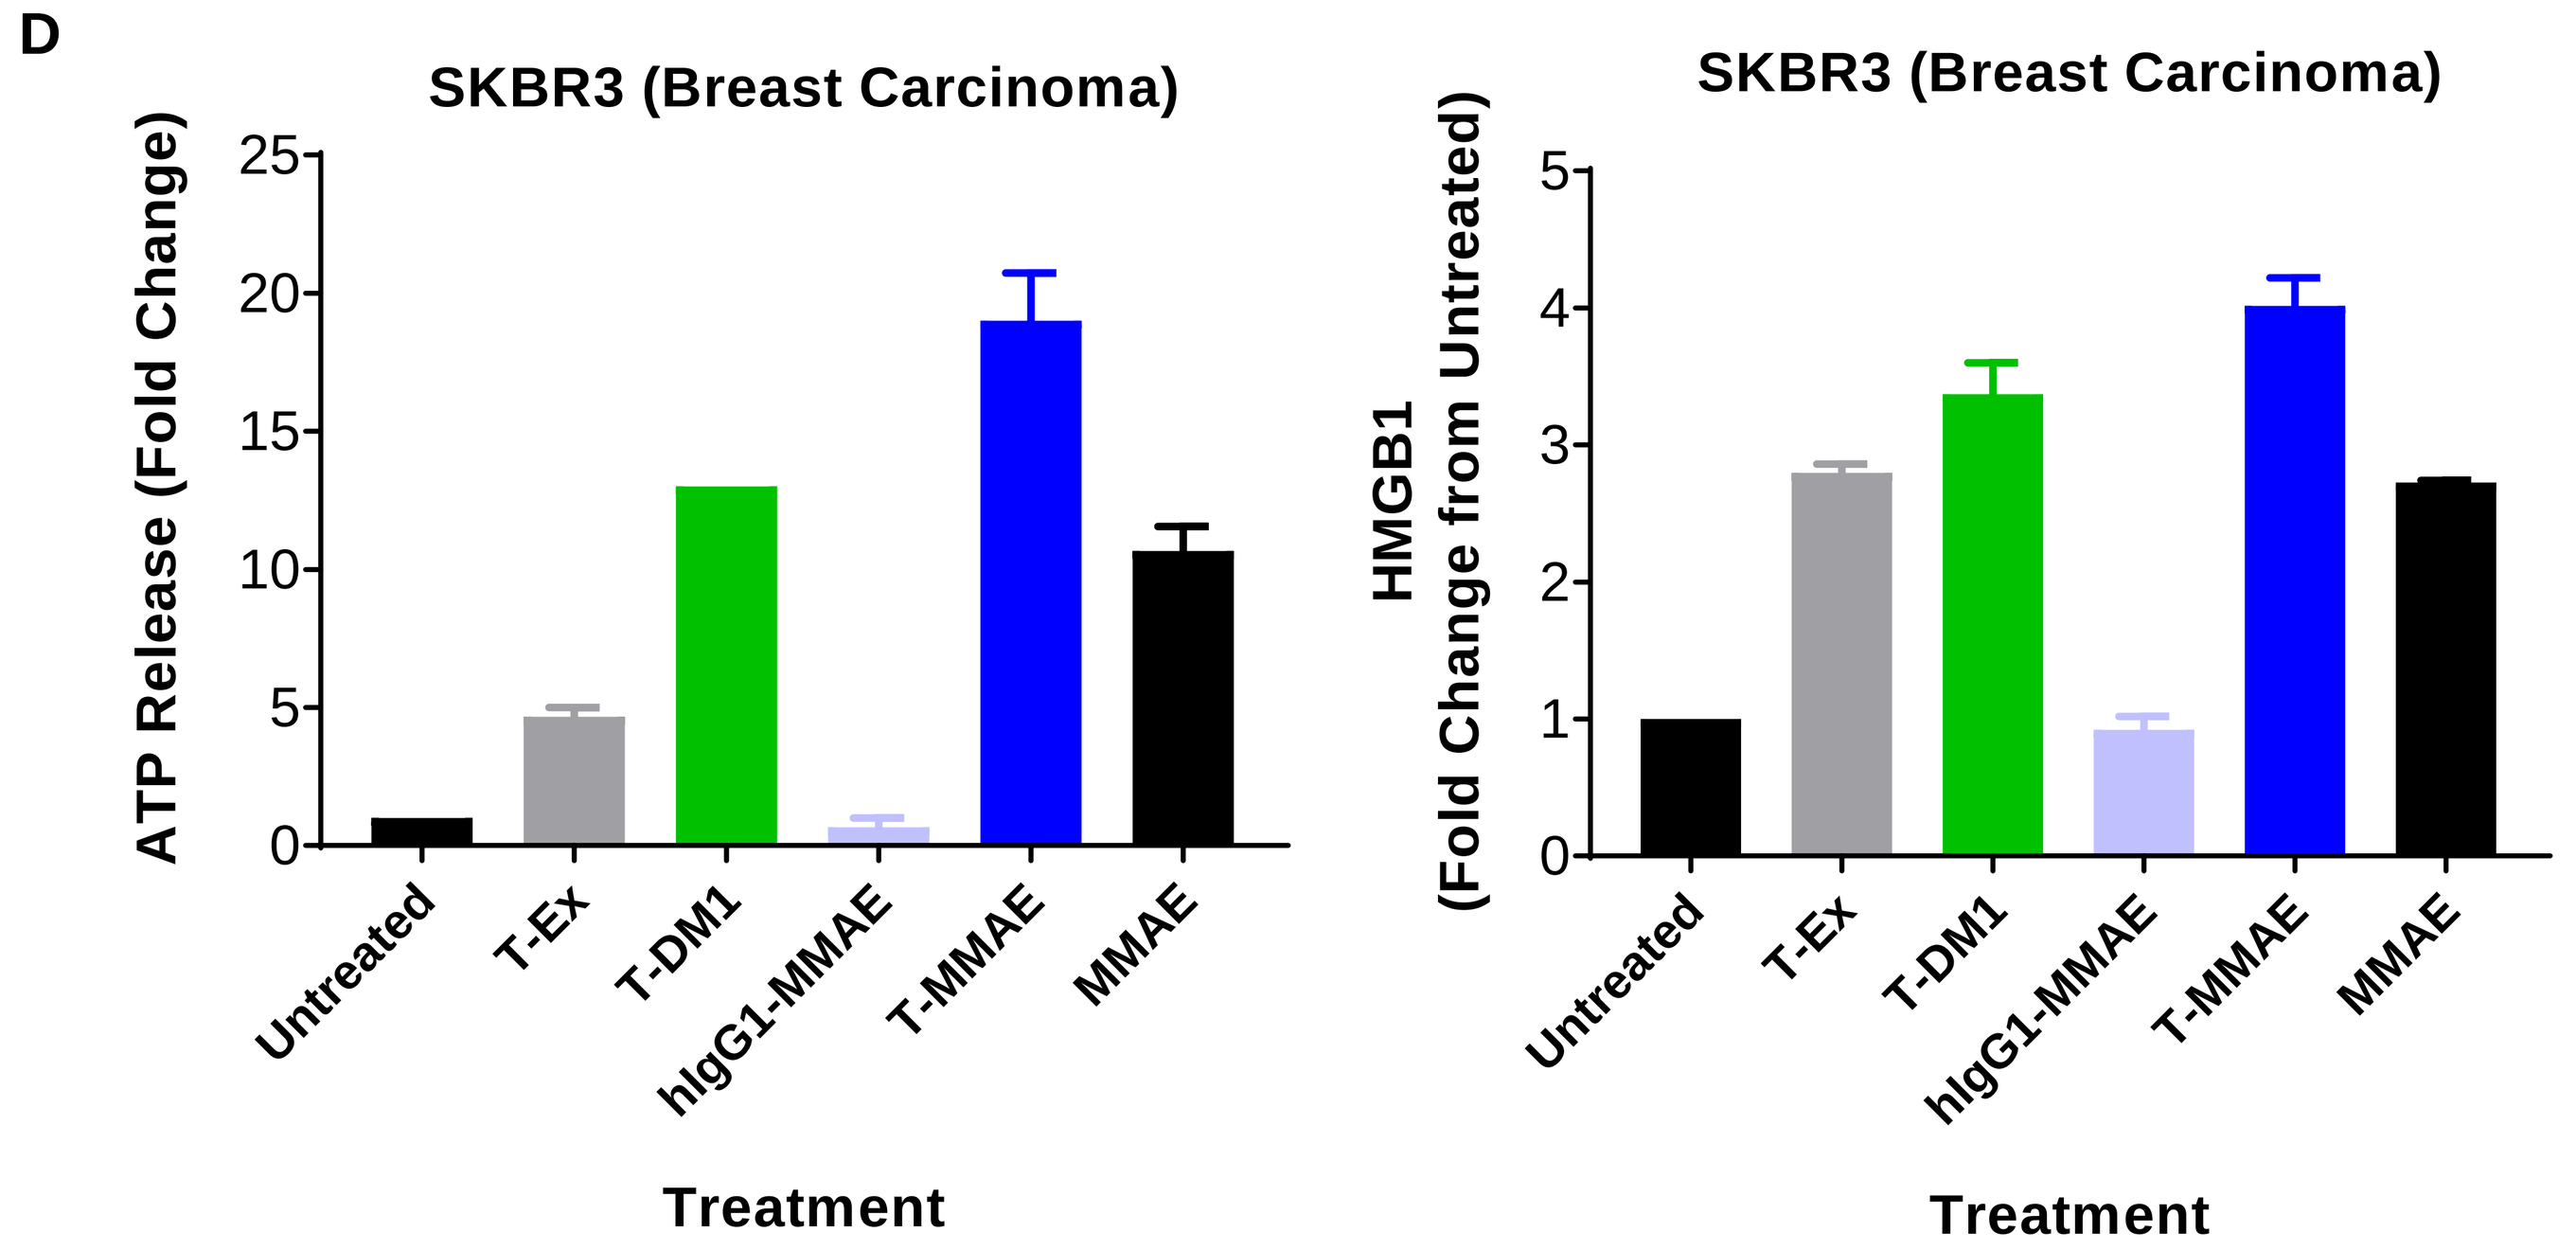
D
SKBR3 (Breast Carcinoma)
ATP Release (Fold Change)
HMGB1 (Fold Change from Untreated)
Untreated
T-Ex
T-DM1
hIgG1-MMAE
T-MMAE
MMAE
Treatment

Figure 8. (A) Table of trastuzumab ADCs evaluated. (B) Graphic of ER stress signaling response. (C) Simple Western (Wes™ Protein Simple) data of BT474 cells treated with ADCs or drug for 72hr. (D) Trastuzumab ADCs dosed at 1 μg/mL or free MMAE (100nM) demonstrate differential in ICD response in SKBR3 HER2-expressing breast cancer cells. Media collected after 48hr or 72hr treatment was used to measure ATP and HMGB1 levels.

Conclusions

- Vedotin ADCs induce ER stress and tumor cell death in a manner evoking ICD hallmarks across multiple cancer indications.
- Tumor-bearing mice treated with vedotin-based ADCs resulted in the promotion of immune cell recruitment and activation in tumors.
- Vedotin-based ADCs included production of innate cytokines and upregulation of MHC Class I/II expression, supporting a role in activating both the innate and adaptive immune responses.
- To further our understanding of the potent and broad ability of vedotin ADCs to induce ICD, we have also begun to examine the ICD potential of different classes of ADC payloads including other microtubule inhibitors (auristatins and maytansines), and DNA damaging agents (DNA alkylators or topoisomerase inhibitors).
- Initial data indicate differences in ICD induction by these agents and totality of the data helps build the rationale for vedotin-based ADCs as preferred partners for immune checkpoint blockade agents.

Acknowledgements

We would like to thank our colleagues Angela Epp, Weiping Zeng and Elizabeth Gray for the Renca studies, Kaleb Smith for HPAF II study, and John Gosink for RNA-seq data and analysis.

Poster No. 618
Society for Immunotherapy of Cancer, Virtual; November 9–14, 2020

Disclosures: Seagen Inc. provided research funding, KK, EMT, SH, RM, KS, JZH, BG, JS, SG, and BAL are employees of and have equity ownership in Seagen Inc., current affiliation for AC is Celgene, Inc., San Francisco, California.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster, kklussman@seagen.com