Phase 2 Trial of Brentuximab Vedotin (BV) with Pembrolizumab (Pembro) in Patients with Metastatic Non-Small Cell Lung Cancer or Metastatic Cutaneous Melanoma After Progression on Anti-PD-1 Therapy

Lee, Sylvia[1]; Jotte, Robert[2,3]; Gillespie-Twardy, Amanda[3,4]; Mehmi, Inderjit[5]; Chandra, Sunandana[6]; Zakharia, Yousef[7]; Hamid, Omid[5]; Watson, Graham[3,8]; Ward, Patrick[3,9]; Chaney, Marya[10]; Lu, Hailing[11]; Berndt, Jason[11]; Rathi, Kapil[11]; Knowles, Scott[11]; O'Connor, Brian P.[11]; Cowey, C. Lance[3,12]

[1]Fred Hutchinson Cancer Center, Seattle, WA; [2]Rocky Mountain Cancer Centers, Lone Tree, CO; [3]US Oncology Network, The Woodlands, TX; [4]Blue Ridge Cancer Care, Roanoke, VA; [5]The Angeles Clinic and Research Institute, A Cedars-Sinai Affiliate, Los Angeles, CA; [6]Northwestern University, Chicago, IL; [7]University of Iowa, Iowa City, IA; [8]Virginia Oncology Associates, Norfolk, VA; [9]Oncology Hematology Care, Cincinnati, OH; [10]Merck & Co., Inc., Rahway, NJ; [11]Seagen Inc., Bothell, WA; [12]Texas Oncology – Baylor University, Dallas, TX

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SITC or the author of this poster, **Sylvia Lee, leesm@uw.edu**

Objectives

To present the efficacy, safety, and exploratory biomarker data from an ongoing phase 2 study evaluating the efficacy and safety of BV+pembro in anti-PD-1 refractory solid tumors (SGN35-033).

Conclusions

- Treatment with BV+pembro was associated with selective depletion of Tregs and modulation of tumor microenvironment with increased CD8 T cell infiltration in responding patients.
- The observed pharmacodynamic activity demonstrates the potential for BV to re-sensitize anti-PD-1 resistant/refractory tumors to PD-1 inhibitors through CD30-directed depletion of Treg cells. Preliminary data in an exploratory analysis suggests that higher response may be observed in patients with tumors with higher PD-L1 expression (data not shown); future analyses are required to confirm this association.
- Anti-tumor responses were observed in patients with confirmed PD and prior CPI exposure within 90 days of receiving study treatment.
- BV+pembro was well tolerated in patients with R/R metastatic NSCLC and metastatic cutaneous melanoma. No new safety signals were observed.
- SGN35-033 is currently enrolling cohorts to investigate the activity of BV+pembro in frontline NSCLC and head and neck cancer (cohorts 5 and 6).

Background

- Anti-PD-1 therapy is the mainstay of frontline treatment for metastatic NSCLC and metastatic cutaneous melanoma.
 - Putative mechanisms of resistance to PD-1 inhibitors include activated Tregs and other immunosuppressive cells within the TME.
- BV is hypothesized to selectively deplete a subset of activated effector intratumoral Tregs that express CD30, potentially re-sensitizing tumors to anti-PD-1 therapy.
- SGN35-033 (NCT04609566) is an ongoing, multiple cohort, multicenter, open-label trial evaluating the efficacy and safety of BV+pembro in anti-PD-1 refractory solid tumors.

Study Design and Endpoints

Study Design

Patient population

Metastatic squamous or nonsquamous NSCLC (without known targetable EGFR, ALK, ROS1, or BRAF mutations)

Metastatic cutaneous melanoma

Treatment arms

- **Cohort 1** Primary refractory metastatic NSCLC
- **Cohort 2** Secondary refractory metastatic NSCLC
- **Cohort 3** Primary refractory metastatic cutaneous melanoma
- **Cohort 4** Secondary refractory metastatic cutaneous melanoma

Dose

BV 1.8 mg/kg Q3W + pembrolizumab 200 mg Q3W

Endpoints

Primary: Confirmed ORR per RECIST v1.1 by investigator

Secondary: DOR, PFS per RECIST v1.1 by investigator; iORR, iDOR and iPFS per iRECIST by investigator, safety evaluation per NCI CTCAE v5.0

Exploratory Endpoints

- Biomarker analysis at baseline and during treatment, including T cells in peripheral blood by flow cytometry, Treg and CD8 T cells by IHC in tumor biopsies, and gene expression profiles by RNAseq in tumor biopsies

Primary refractory (cohorts 1 and 3): Progressed without prior objective response during or after prior PD-1 inhibitor therapy within 3 months or SD for <6 months. Patients who progress while on adjuvant anti-PD1 therapy were eligible to enroll. Adjuvant courses of therapy without measurable disease do not apply to the SD for at least 6 months criterion.
Secondary refractory (cohorts 2 and 4): Progressed after having developed a prior objective response of CR/PR for at least 3 months or SD for at least 6 months
For NSCLC patients only: For patients on CPI treatment for <90 days, iRECIST confirmation of PD at least 4 weeks from the initial PD was required
For melanoma patients only: Treatment with a PD-1 CPI containing regimen within 90 days; iRECIST confirmation of PD at least 4 weeks from the initial PD was required
Unless otherwise specified, all analyses reflect the FAS and include all subjects who received any amount of study medication (BV or pembrolizumab).

Results

Patient Demographics and Disease Characteristics

	Metastatic NSCLC			Metastatic cutaneous melanoma		
	Primary refractory (n=12)	Secondary refractory (n=43)	Total (n=55)	Primary refractory (n=17)	Secondary refractory (n=41)	Total (n=58)
Age (years) median (range)	**67.5 (49, 80)**	**67.0 (55, 86)**	**67.0 (49, 86)**	**59.0 (23, 83)**	**65.0 (25, 86)**	**64.5 (23, 86)**
Sex, n (%)						
Male	11 (92)	24 (56)	35 (64)	11 (65)	25 (61)	36 (62)
Female	1 (8)	19 (44)	20 (36)	6 (35)	16 (39)	22 (38)
ECOG performance status, n (%)						
0	6 (50)	16 (37)	22 (40)	13 (76)	26 (63)	39 (67)
1	6 (50)	27 (63)	33 (60)	4 (24)	15 (37)	19 (33)
Time from initial diagnosis to first dose (months)						
n	12	40	52	14	34	48
Median	15.5	22.4	20.0	32.3	59.7	44.1
Min, max	5, 41	8, 138	5, 138	6, 132	8, 259	6, 259
Prior therapy, n (%)						
Prior ipilimumab/ anti-CTLA4 exposure	0	6 (14)	6 (11)	15 (88)	32 (78)	47 (81)

Data cutoff: 10AUG2023

Efficacy Analyses

- ORR was 8% to 22% across cohorts.
- DORs appear durable in some patients; however, data are immature and limited by small sample size.
- In an exploratory analysis, numerically higher response rates were observed in patients with tumors that had higher PD-L1 expression (data not shown).
- Most responders in cohorts 3 and 4 received study treatment within 90 days of receiving prior CPI therapy.

Antitumor Activity

	Metastatic NSCLC Primary refractory (n=12)	Metastatic NSCLC Secondary refractory (n=43)		Metastatic cutaneous melanoma Primary refractory (n=17)	Metastatic cutaneous melanoma Secondary refractory (n=41)
ORR (CR+PR) %, (95% CI)	8 (0.2, 38.5)	14 (5.3, 27.9)	**iORR (iCR+iPR)[a] %, (95% CI)**	18 (3.8, 43.4)	22 (10.6, 37.6)
DCR (CR+PR+SD) %, (95% CI)	67 (34.9, 90.1)	72 (56.3, 84.7)	**iDCR (iCR+iPR+iSD)[a] %, (95% CI)**	71 (44.0, 89.7)	80 (65.1, 91.2)

[a]One patient with melanoma experienced pseudoprogression but consolidated to an iCR; hence iRECIST is reported for melanoma cohorts.
Data cutoff: 10AUG2023

Duration of Response: Metastatic NSCLC and Metastatic Cutaneous Melanoma

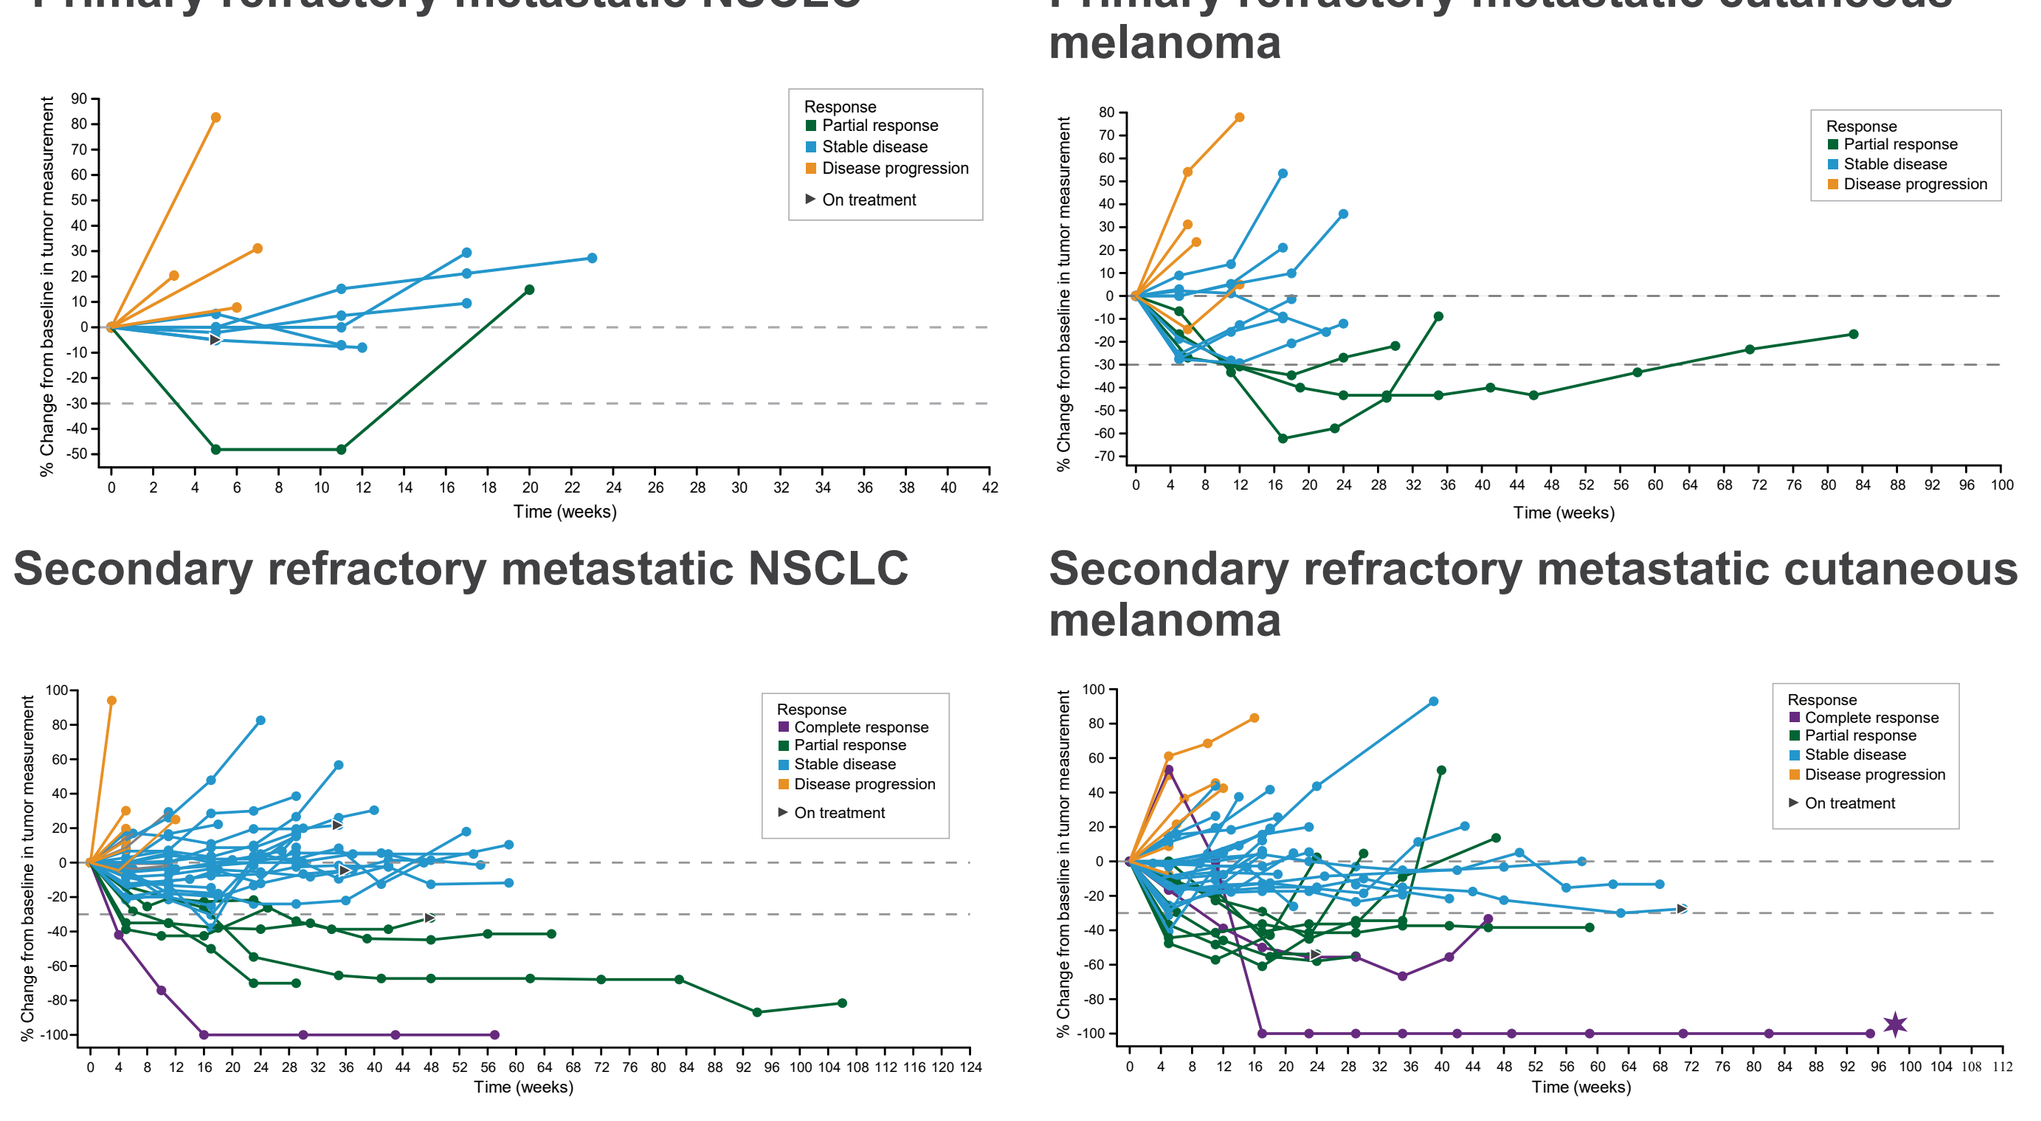
Primary refractory metastatic NSCLC
Primary refractory metastatic cutaneous melanoma
Secondary refractory metastatic NSCLC
Secondary refractory metastatic cutaneous melanoma
% Change from baseline in tumor measurement
Time (weeks)
Response
Complete response
Partial response
Stable disease
Disease progression
On treatment

Primary and Secondary refractory metastatic NSCLC (cohorts 1 and 2): Response Criterion: CR, PR, SD and PD per RECIST v1.1.
Primary and Secondary refractory metastatic cutaneous melanoma (cohorts 3 and 4): Response Criterion: iCR, iPR, iSD, iUPD, and iCPD per iRECIST.
✶ Indicates the patient with biopsy data shown in biopsy figure.
Data cutoff: 10AUG2023

Safety Summary

All TEAEs Occurring at ≥20% Frequency and Grade ≥3 TEAEs Occurring at ≥5% Frequency

TEAEs, n (%)	Metastatic NSCLC (N=55) Any grade (≥20%)	Metastatic NSCLC (N=55) Grade ≥3 (≥5%)	Metastatic cutaneous melanoma (N=58) Any grade (≥20%)	Metastatic cutaneous melanoma (N=58) Grade ≥3 (≥5%)
Any event	51 (93)	31 (56)	56 (97)	32 (55)
Fatigue	28 (51)	3 (5)	24 (41)	3 (5)
Nausea	28 (51)	3 (5)	20 (34)	–
Peripheral sensory neuropathy	20 (36)	–	21 (36)	–
Diarrhea	16 (29)	–	18 (31)	–
Constipation	15 (27)	–	17 (29)	–
Decreased appetite	13 (24)	3 (5)	–	–
Dyspnea	16 (29)	–	–	–
Vomiting	11 (20)	–	–	–
Pruritus	11 (20)	–	–	–
Acute kidney injury	–	3 (5)	–	–
Aspartate aminotransferase increased	–	3 (5)	–	–
Neutropenia	–	3 (5)	–	
Hypokalemia	–	–	–	4 (7)
Hepatitis	–	–	–	3 (5)

Note: "–" indicates TEAEs below 20% (all grade) or 5% (grade ≥3) threshold
AEs were defined as treatment-emergent if they were newly occurring or worsened following treatment with either brentuximab vedotin or pembrolizumab.
Data cutoff: 10AUG2023

Safety Summary, continued

- No new safety signals were observed.
- TESAEs were reported in 18 (33%) patients with metastatic NSCLC and in 29 (50%) patients with metastatic cutaneous melanoma. The most common TESAEs across all cohorts were vomiting (in 4% of patients) and cerebrovascular accident (in 3% of patients).
- TEAEs leading to treatment discontinuation were reported in 11 (20%) patients with metastatic NSCLC and in 8 (14%) patients with metastatic cutaneous melanoma.
- Treatment-emergent peripheral neuropathy events occurred in 27 (49%) patients with metastatic NSCLC and in 26 (45%) patients with metastatic cutaneous melanoma.
- 29 (26%) patients experienced immune-mediated TEAEs; 13 (12%) patients experienced events that were considered treatment-related. The most common (≥2 patients) treatment-related IMAEs were aspartate aminotransferase increased, hypothyroidism, pneumonitis, and rash maculopapular, all of which occurred in 2 patients each.
- 5 (4%) patients experienced TEAEs that led to death.
- The safety data are generally consistent with the approved safety profiles in the labels of BV and pembrolizumab.

Exploratory Biomarker Analyses

Transient Decrease in Tregs in Peripheral Blood in Patients Receiving BV+Pembro

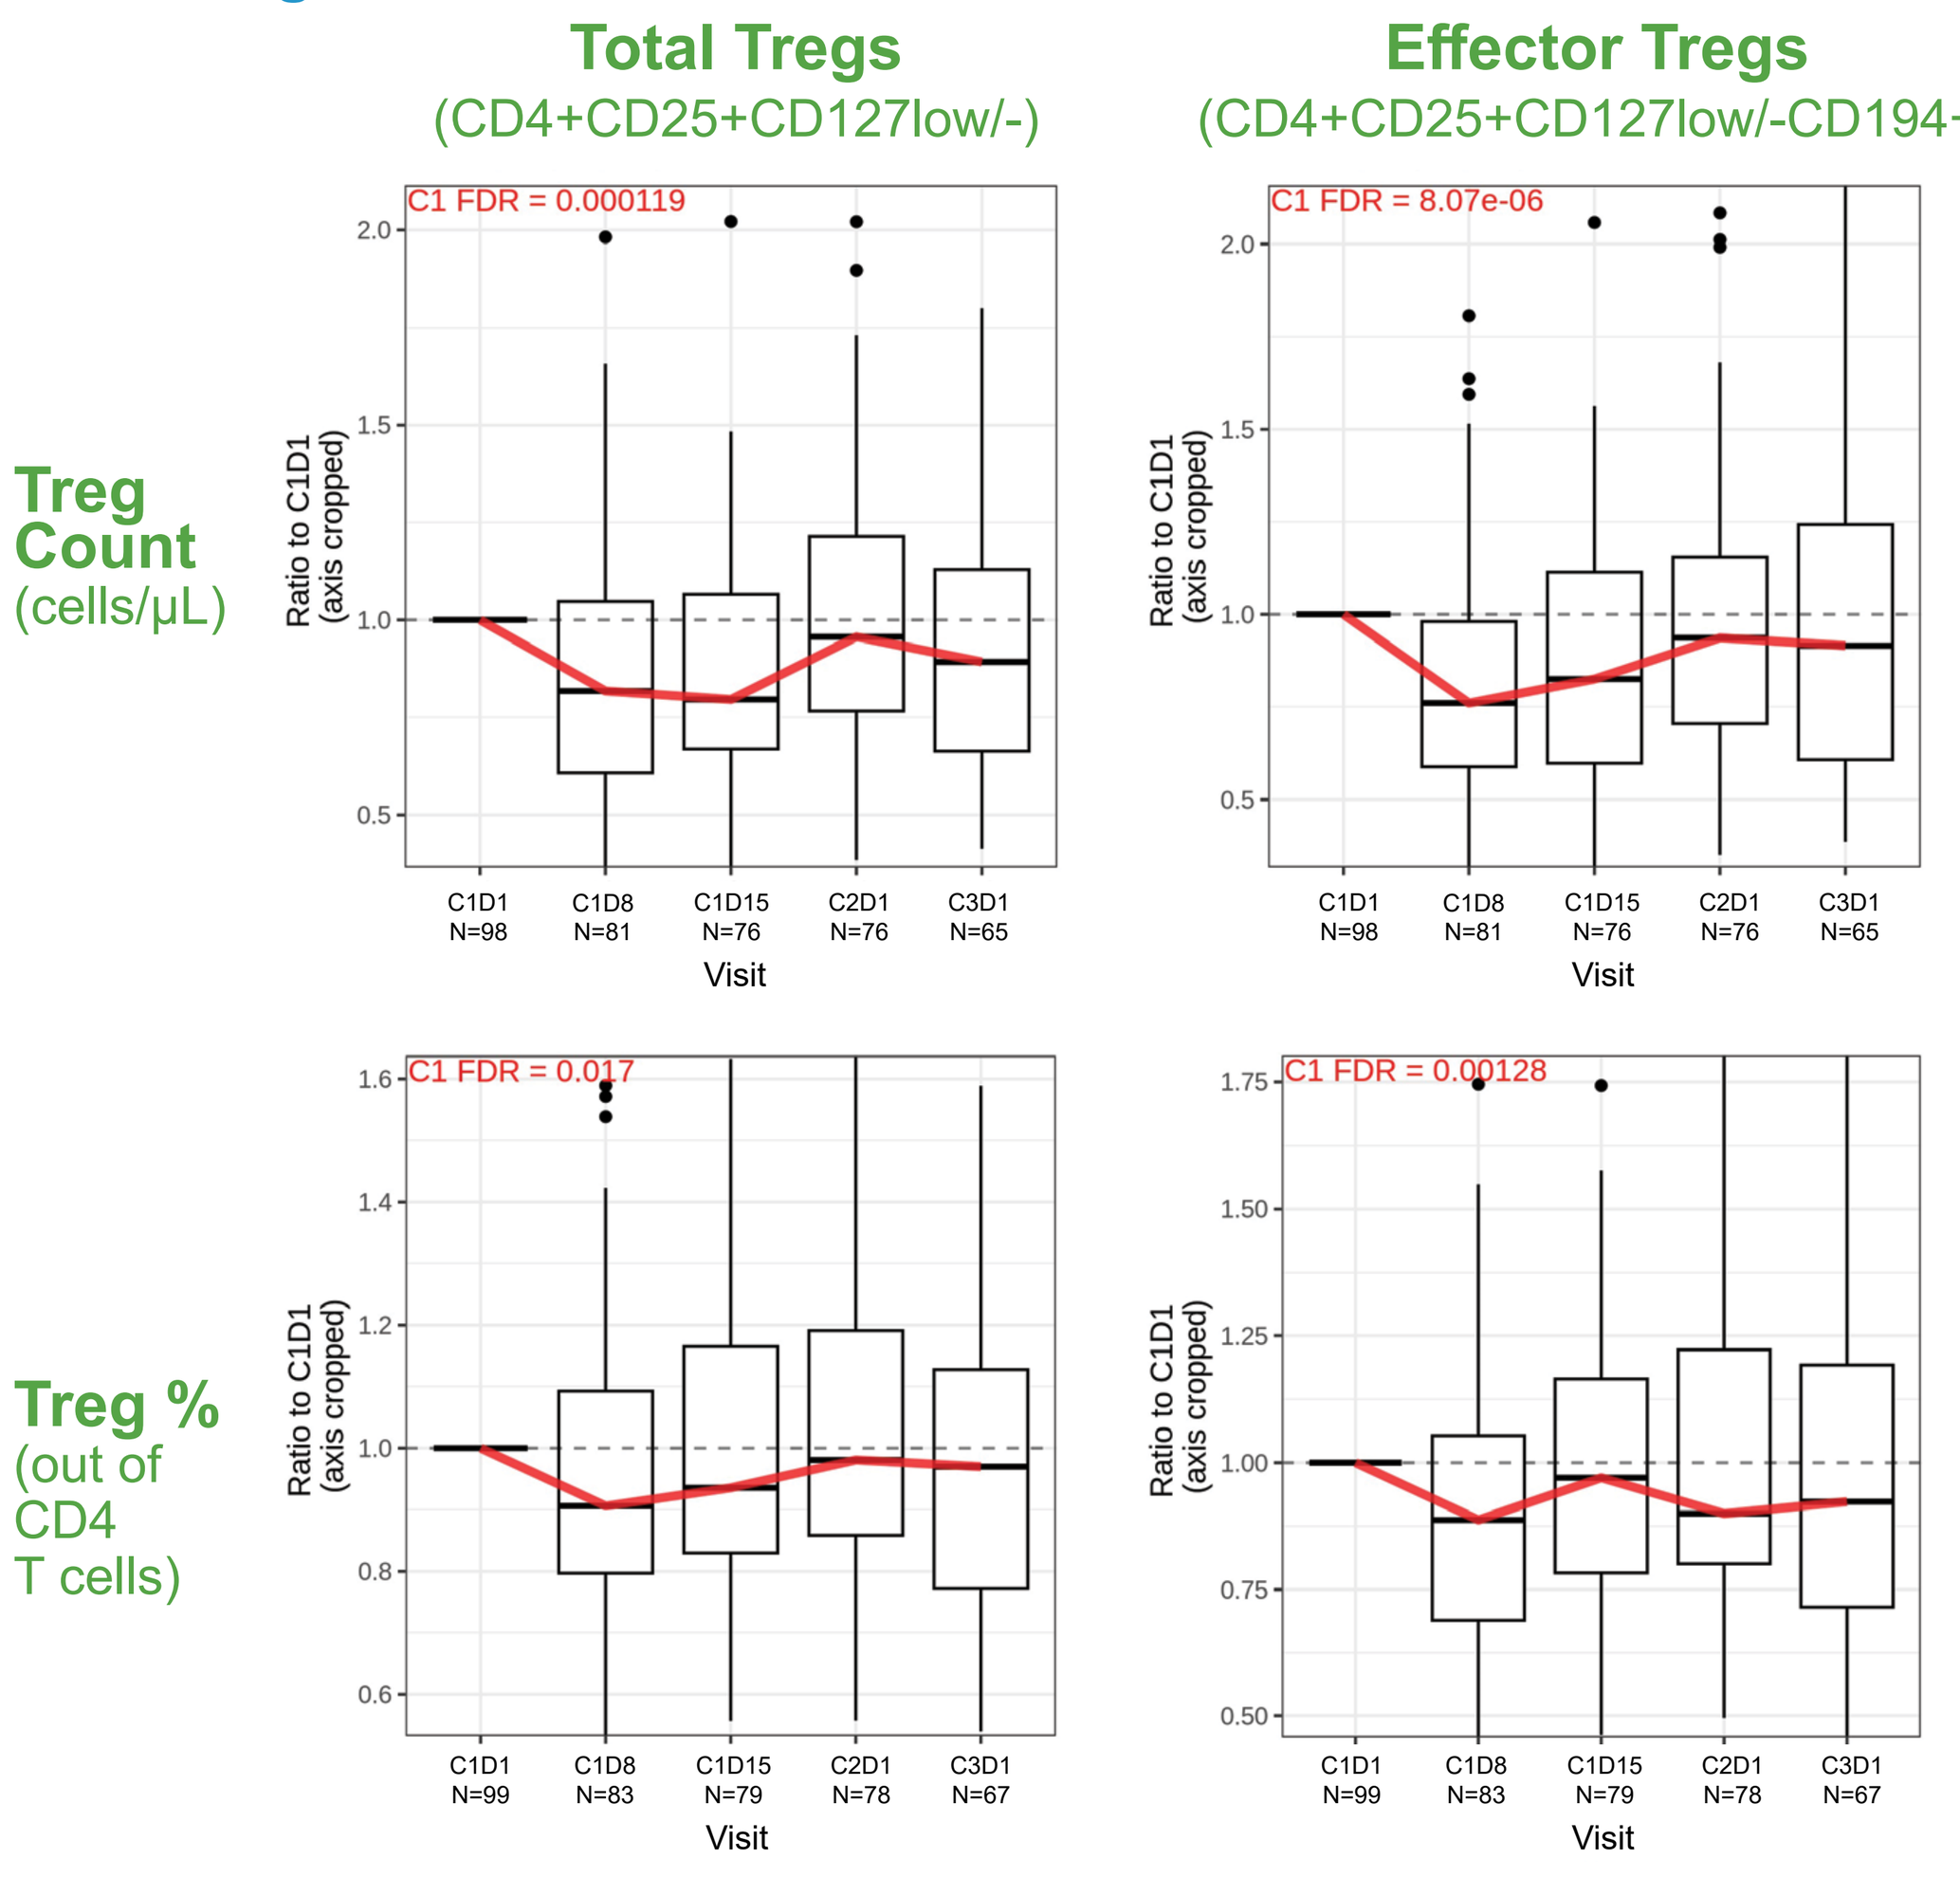
Total Tregs
(CD4+CD25+CD127low/-)
Effector Tregs
(CD4+CD25+CD127low/-CD194+)
Treg Count
(cells/µL)
Treg %
(out of CD4 T cells)
C1 FDR = 0.000119
C1 FDR = 8.07e-06
C1 FDR = 0.017
C1 FDR = 0.00128
Ratio to C1D1 (axis cropped)
C1D1 N=98
C1D8 N=81
C1D15 N=76
C2D1 N=76
C3D1 N=65
C1D1 N=99
C1D8 N=83
C1D15 N=79
C2D1 N=78
C3D1 N=67
Visit

The red numbers in each graph (C1 FDR) indicate False Discovery Rate comparing maximum change at C1D8 or C1D15 to C1D1; The numbers underneath each graph indicate the number of patient samples at each time point.

Paired Tumor Biopsies Collected from Responding Patients Suggest Increase in CD8 T Cell Infiltration After Treatment with BV+Pembro

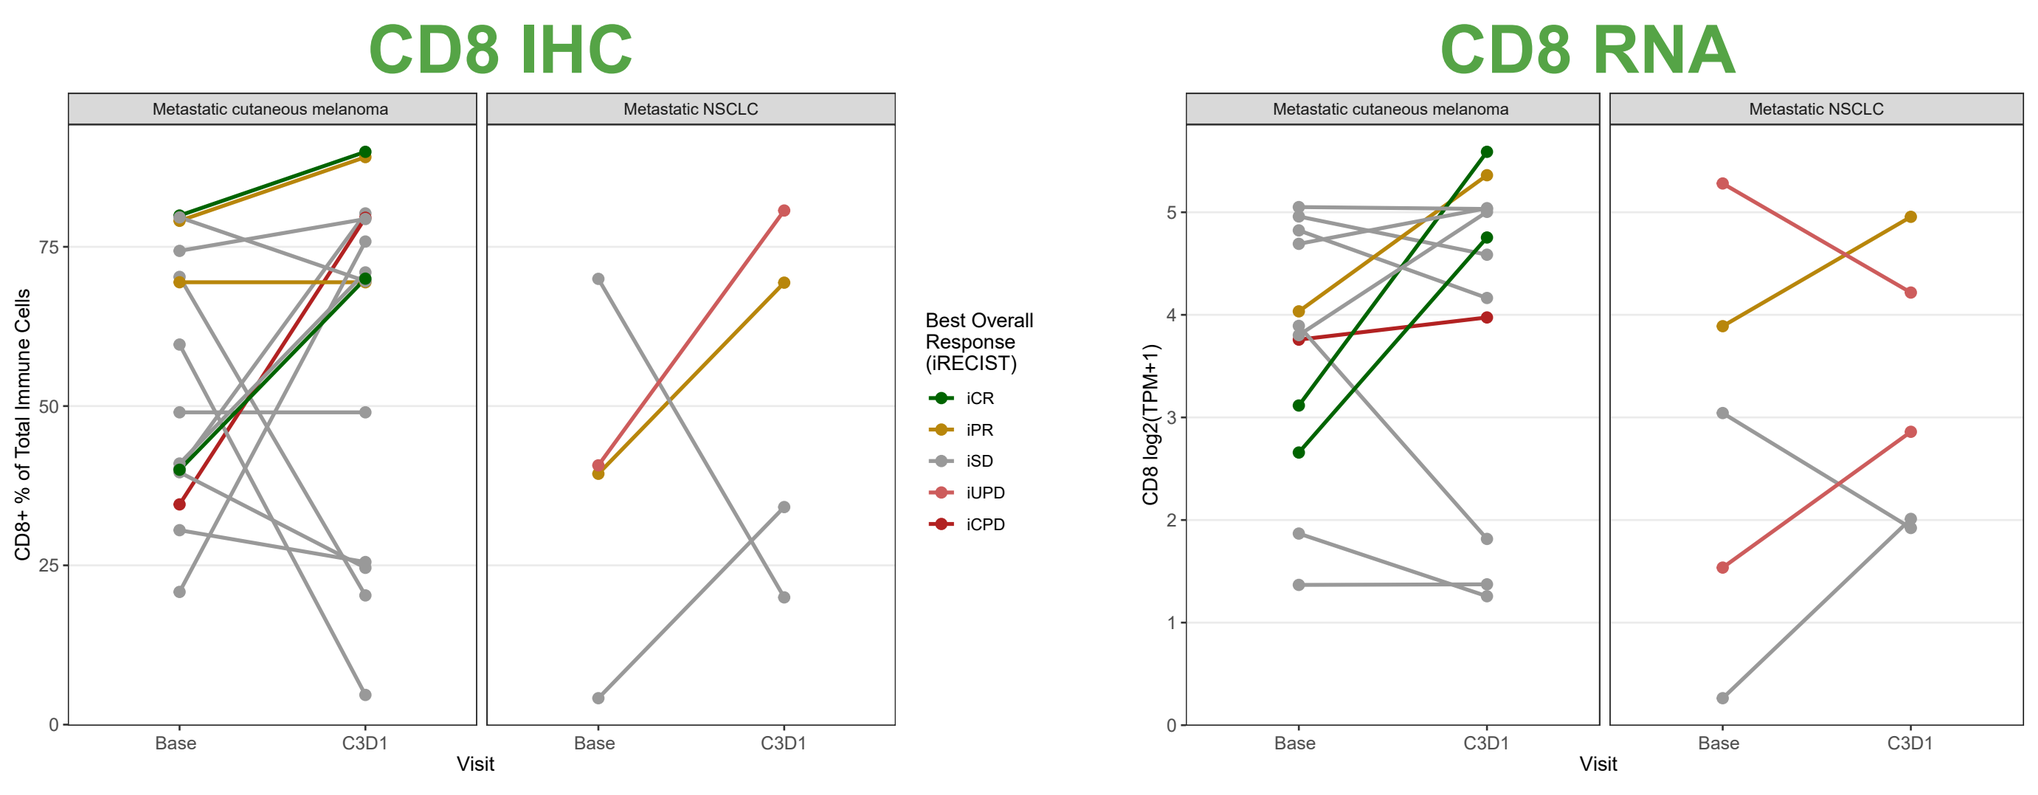
CD8 IHC
CD8 RNA
Metastatic cutaneous melanoma
Metastatic NSCLC
CD8+ % of Total Immune Cells
CD8 log2(TPM+1)
Best Overall Response (iRECIST)
iCR
iPR
iSD
iUPD
iCPD
Base
C3D1
Visit

CD8 T cells increased in 4/5 (80%) responding patients (iCR or iPR) and 7/14 (50%) non-responders (iSD or iPD). Note that some lines overlap.

CD8 expression increased in 4/4 (100%) responding patients (iCR or iPR) and 6/13 (46%) non-responders (iSD or iPD).

Paired Biopsies from Patient with Metastatic Cutaneous Melanoma Demonstrate Increase in CD8 and Decrease in Foxp3 After Treatment with BV+Pembro

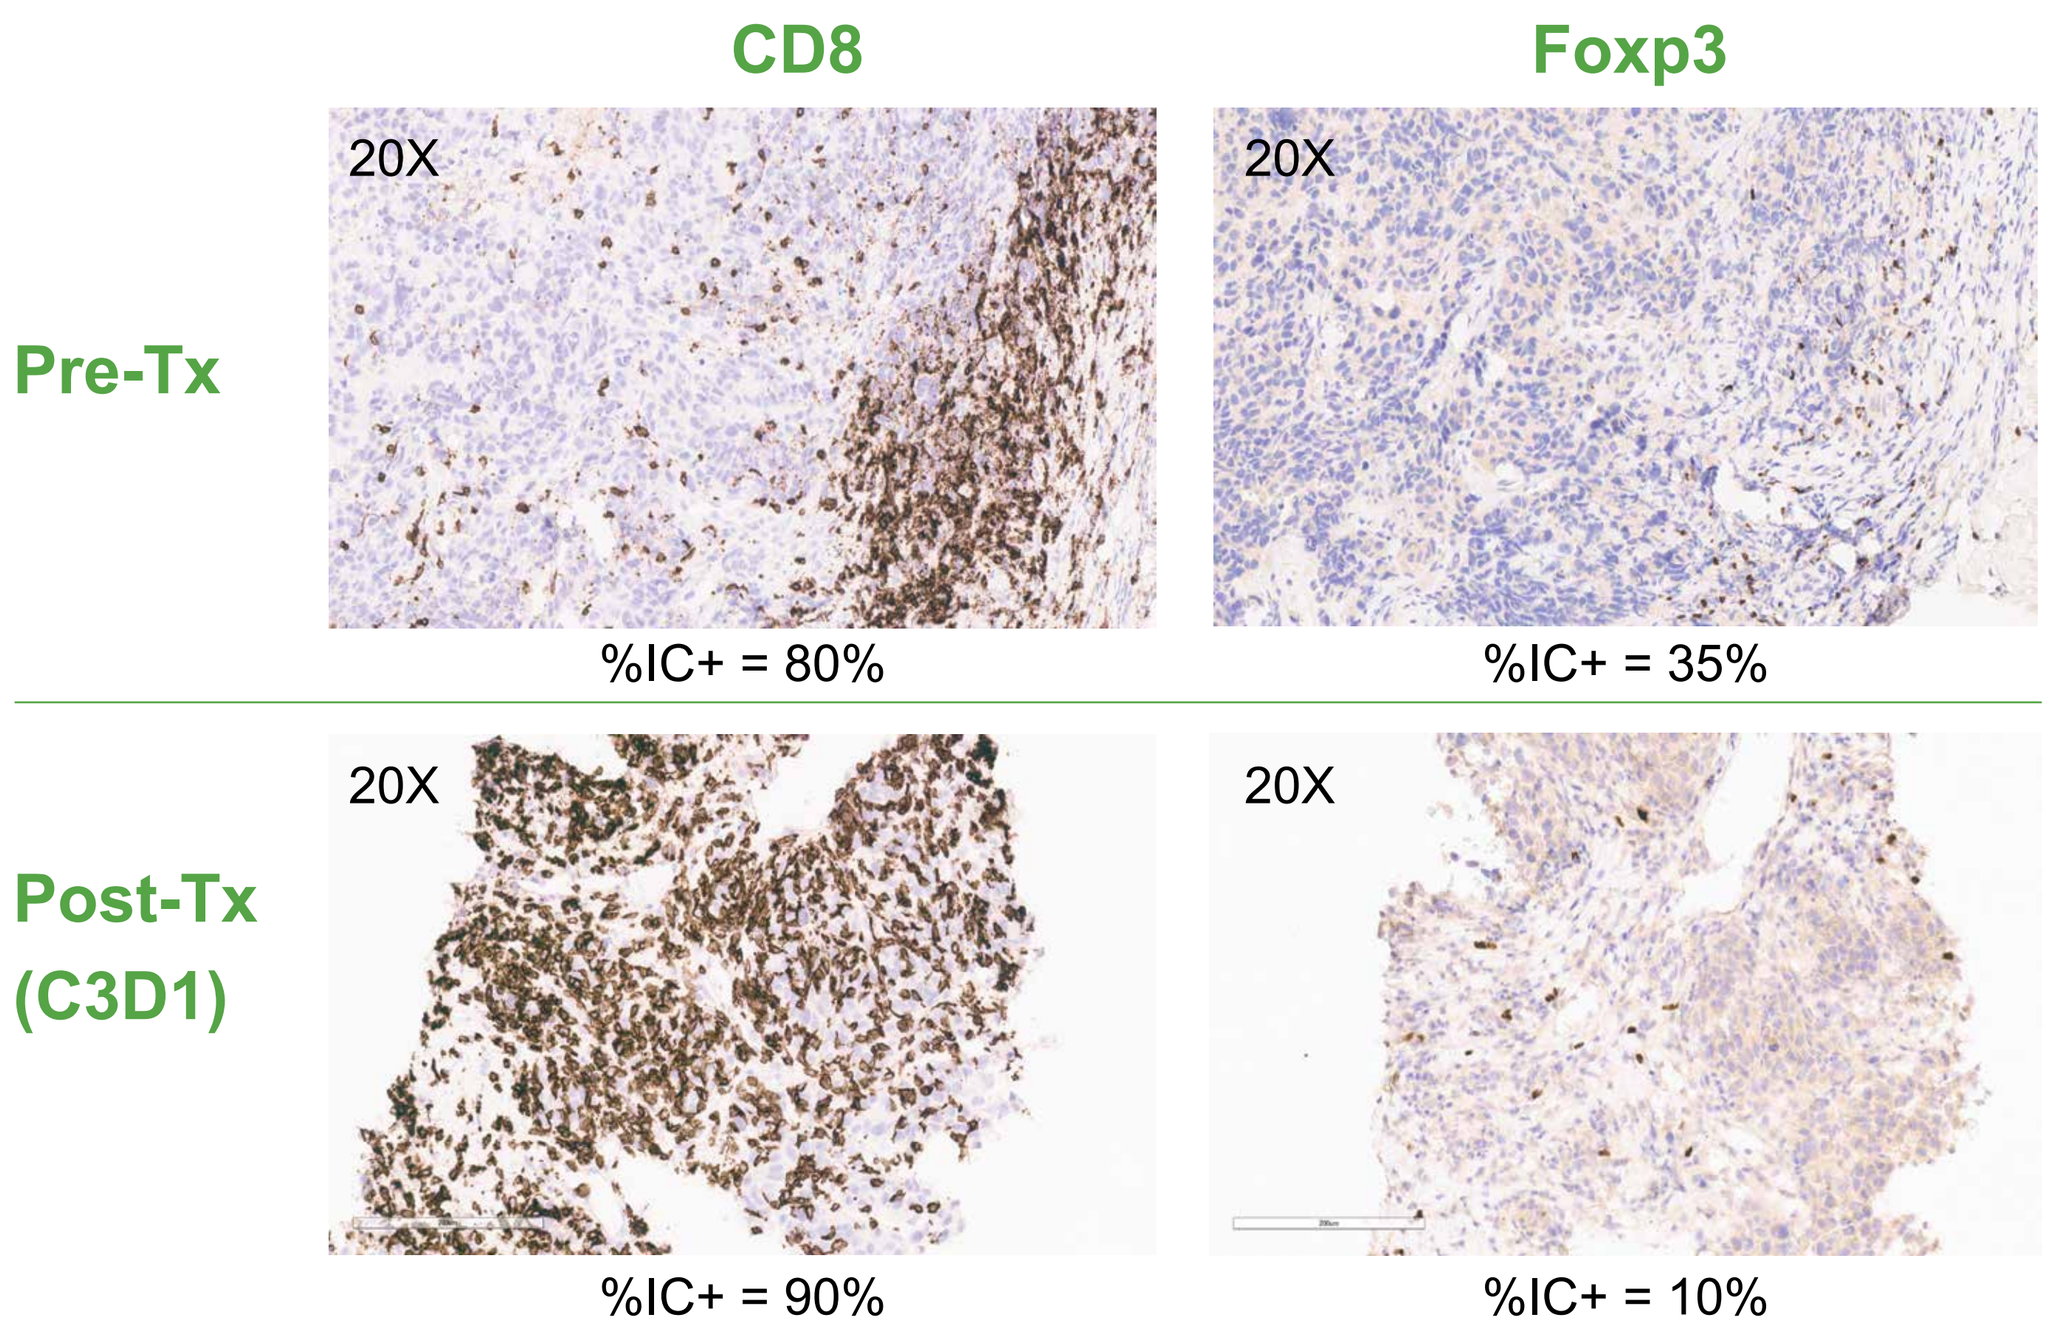
CD8
Foxp3
Pre-Tx
Post-Tx
(C3D1)
20X
20X
20X
20X
%IC+ = 80%
%IC+ = 35%
%IC+ = 90%
%IC+ = 10%

Baseline and C3D1 biopsy were collected from a melanoma patient who had complete response after pseudo-progression, as indicated by the star on the spider plot for secondary refractory cutaneous melanoma (cohort 4) patients. The brown color indicates cells stained positive for CD8 and Foxp3. The numbers below each graph show pathologist scoring. "% IC" means the percentage of cells out of total immune cells that stained positive.

Responding Patients Showed a Trend Toward Higher CD8 and Foxp3 Protein Expression in Baseline Tumor Biopsies

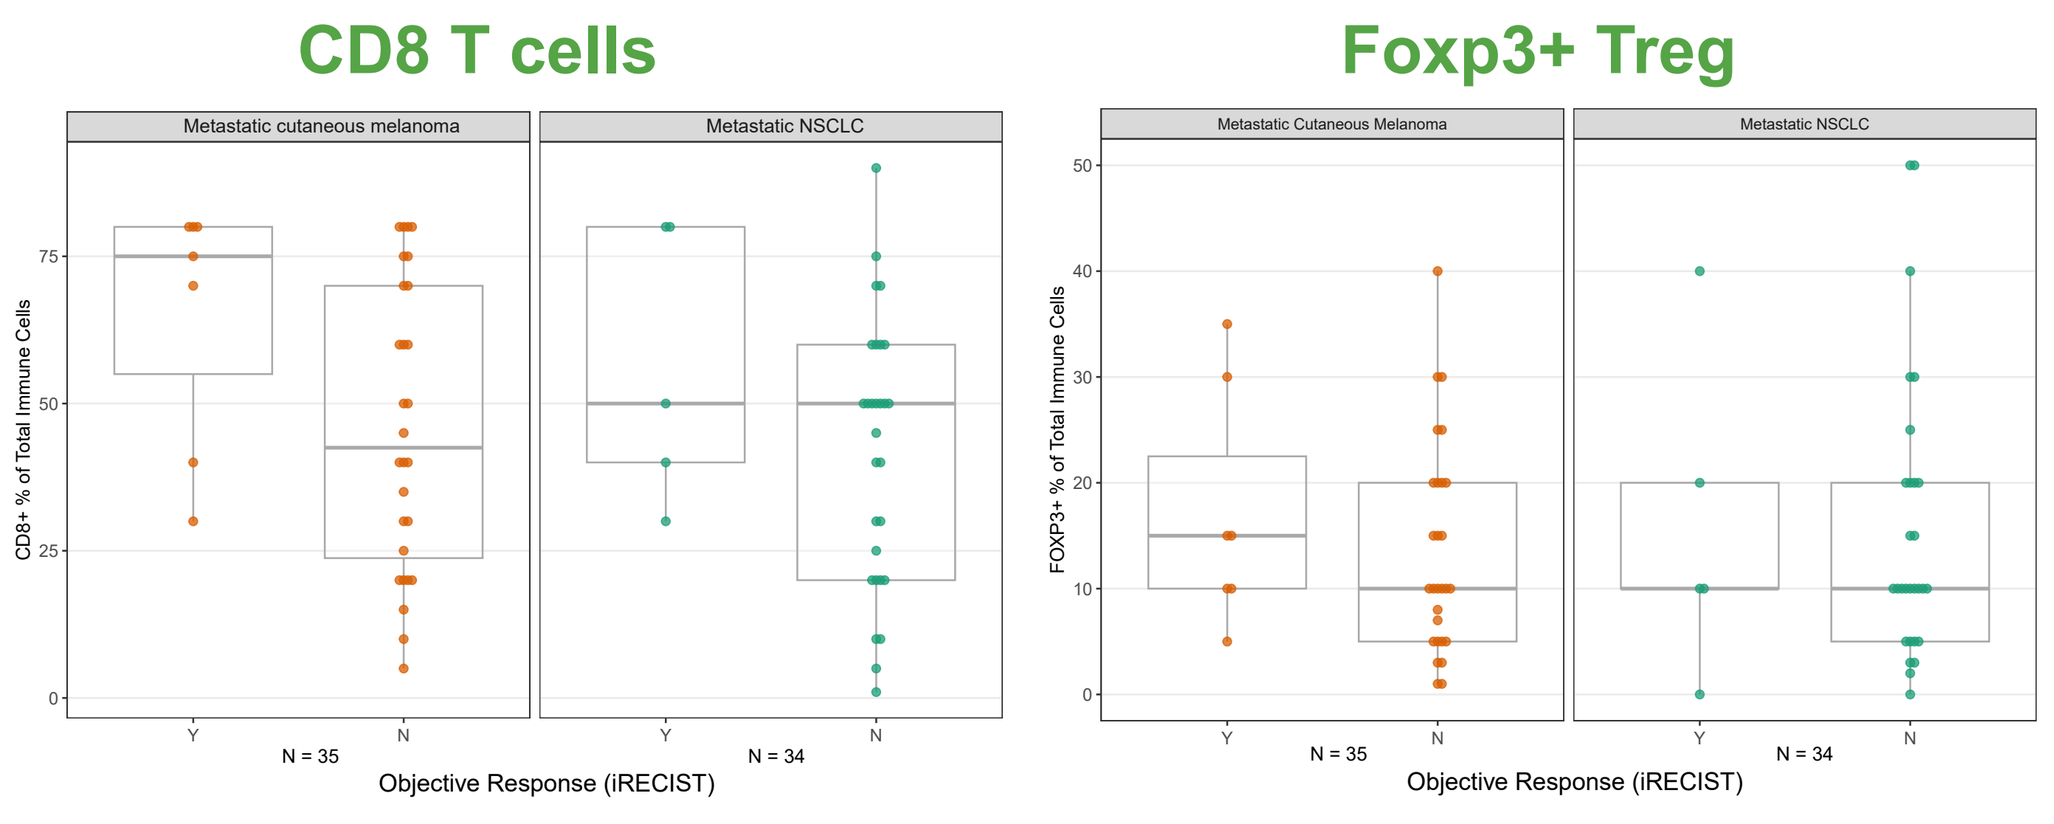
CD8 T cells
Foxp3+ Treg
Metastatic cutaneous melanoma
Metastatic NSCLC
CD8+ % of Total Immune Cells
FOXP3+ % of Total Immune Cells
N = 35
N = 34
Objective Response (iRECIST)

Y: responders (CR or PR); N: Patients did not achieve an objective response

Immune Response Pathway-related Gene Sets are Enriched in Responders as Compared to Non-responders in Baseline Tumor Biopsies

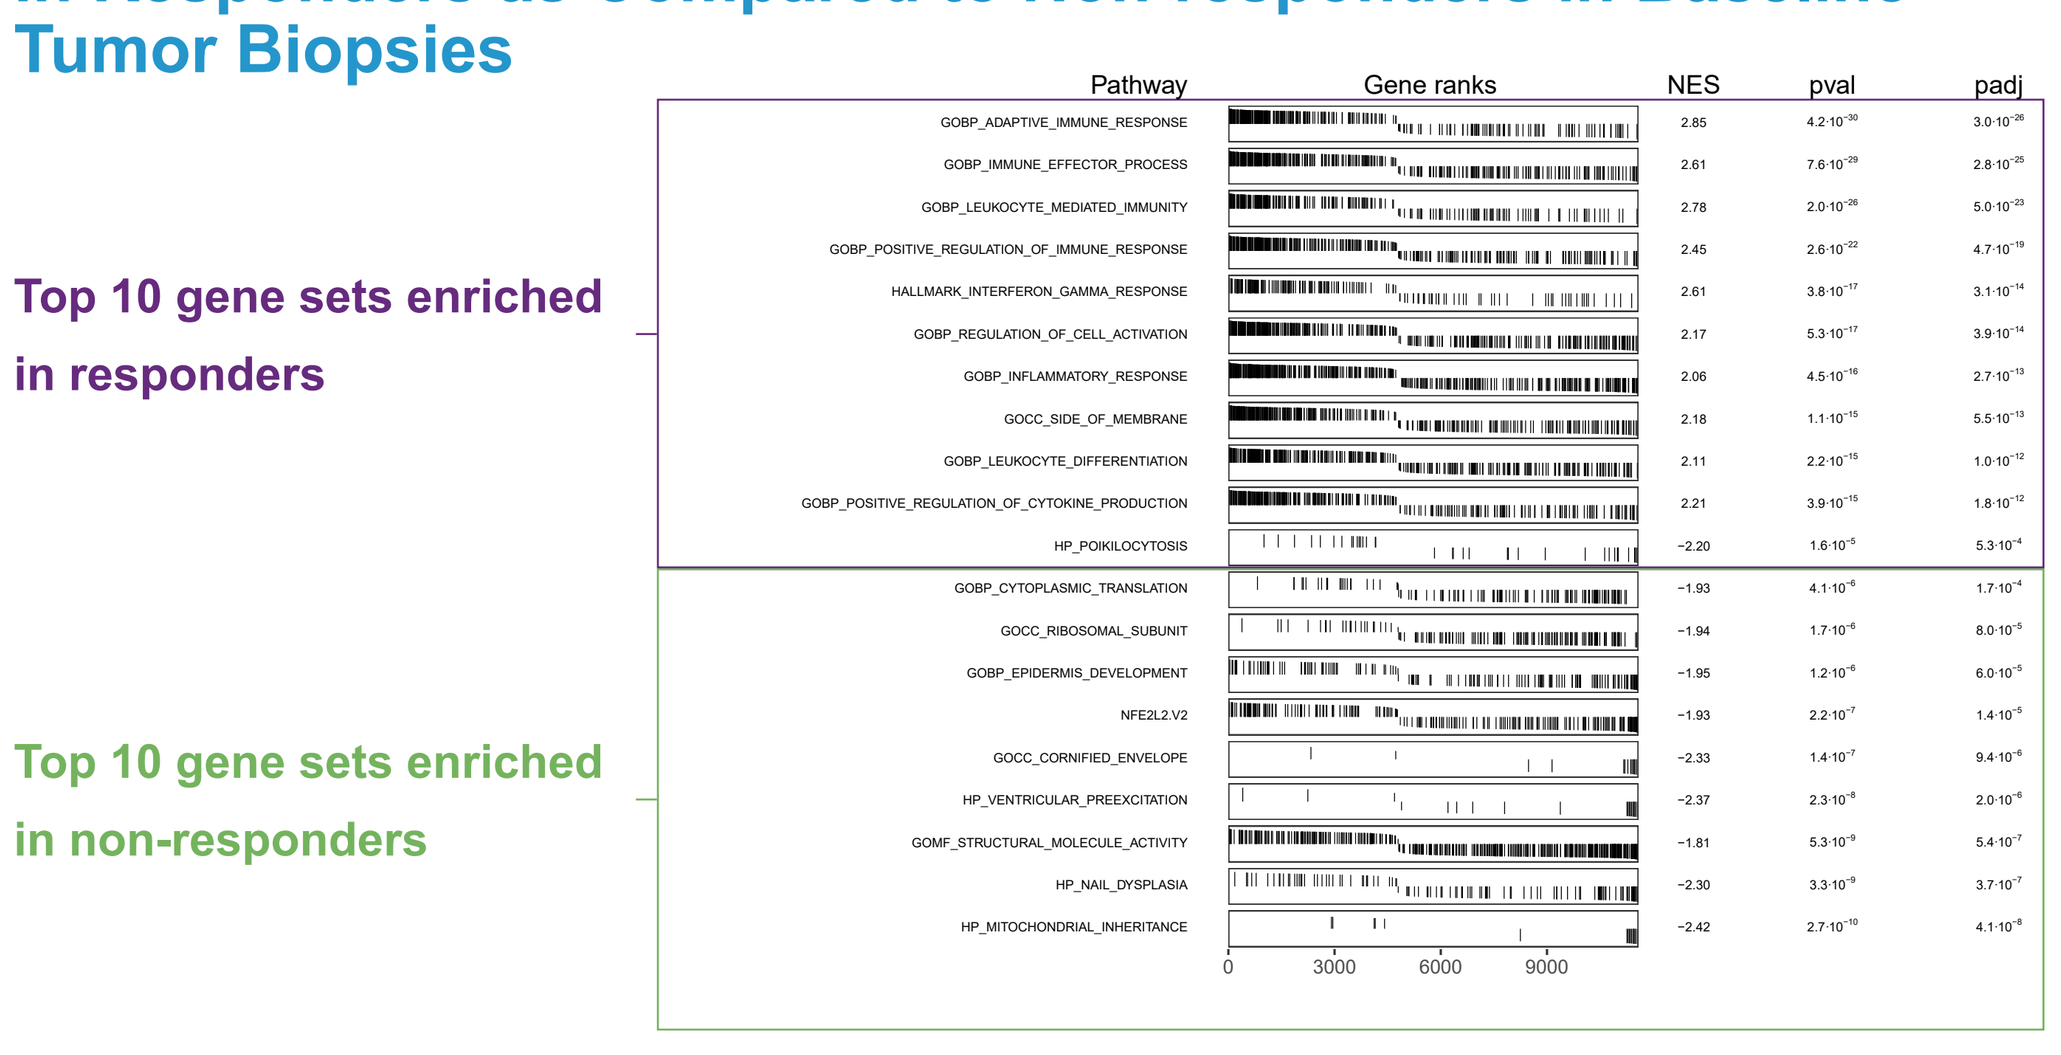
Top 10 gene sets enriched in responders
Top 10 gene sets enriched in non-responders
Pathway
Gene ranks
NES
pval
padj

Gene Set Enrichment Analysis (GSEA) used ranked log2 fold change between responders (iCR+iPR) and non-responders (iSD+iCPD+iUPD+NE/NA).
Evaluated 15,712 gene sets, including "hallmark", oncogenic gene signature, gene ontology (GO CC, BP, and MF) and human phenotype ontology sets.
"Collapsed" enriched gene sets with common genes, plots show representative set names.

Antigen Presentation, T Cell Signaling, and Treg Genes in Baseline Tumor Biopsies May be Associated with a Favorable Immune Environment for Response to BV+Pembro

54 intersecting leading-edge genes among 58 positively enriched gene sets when comparing iCR+iPR to iSD+iUPD+iCPD +NE/NA using GSEA

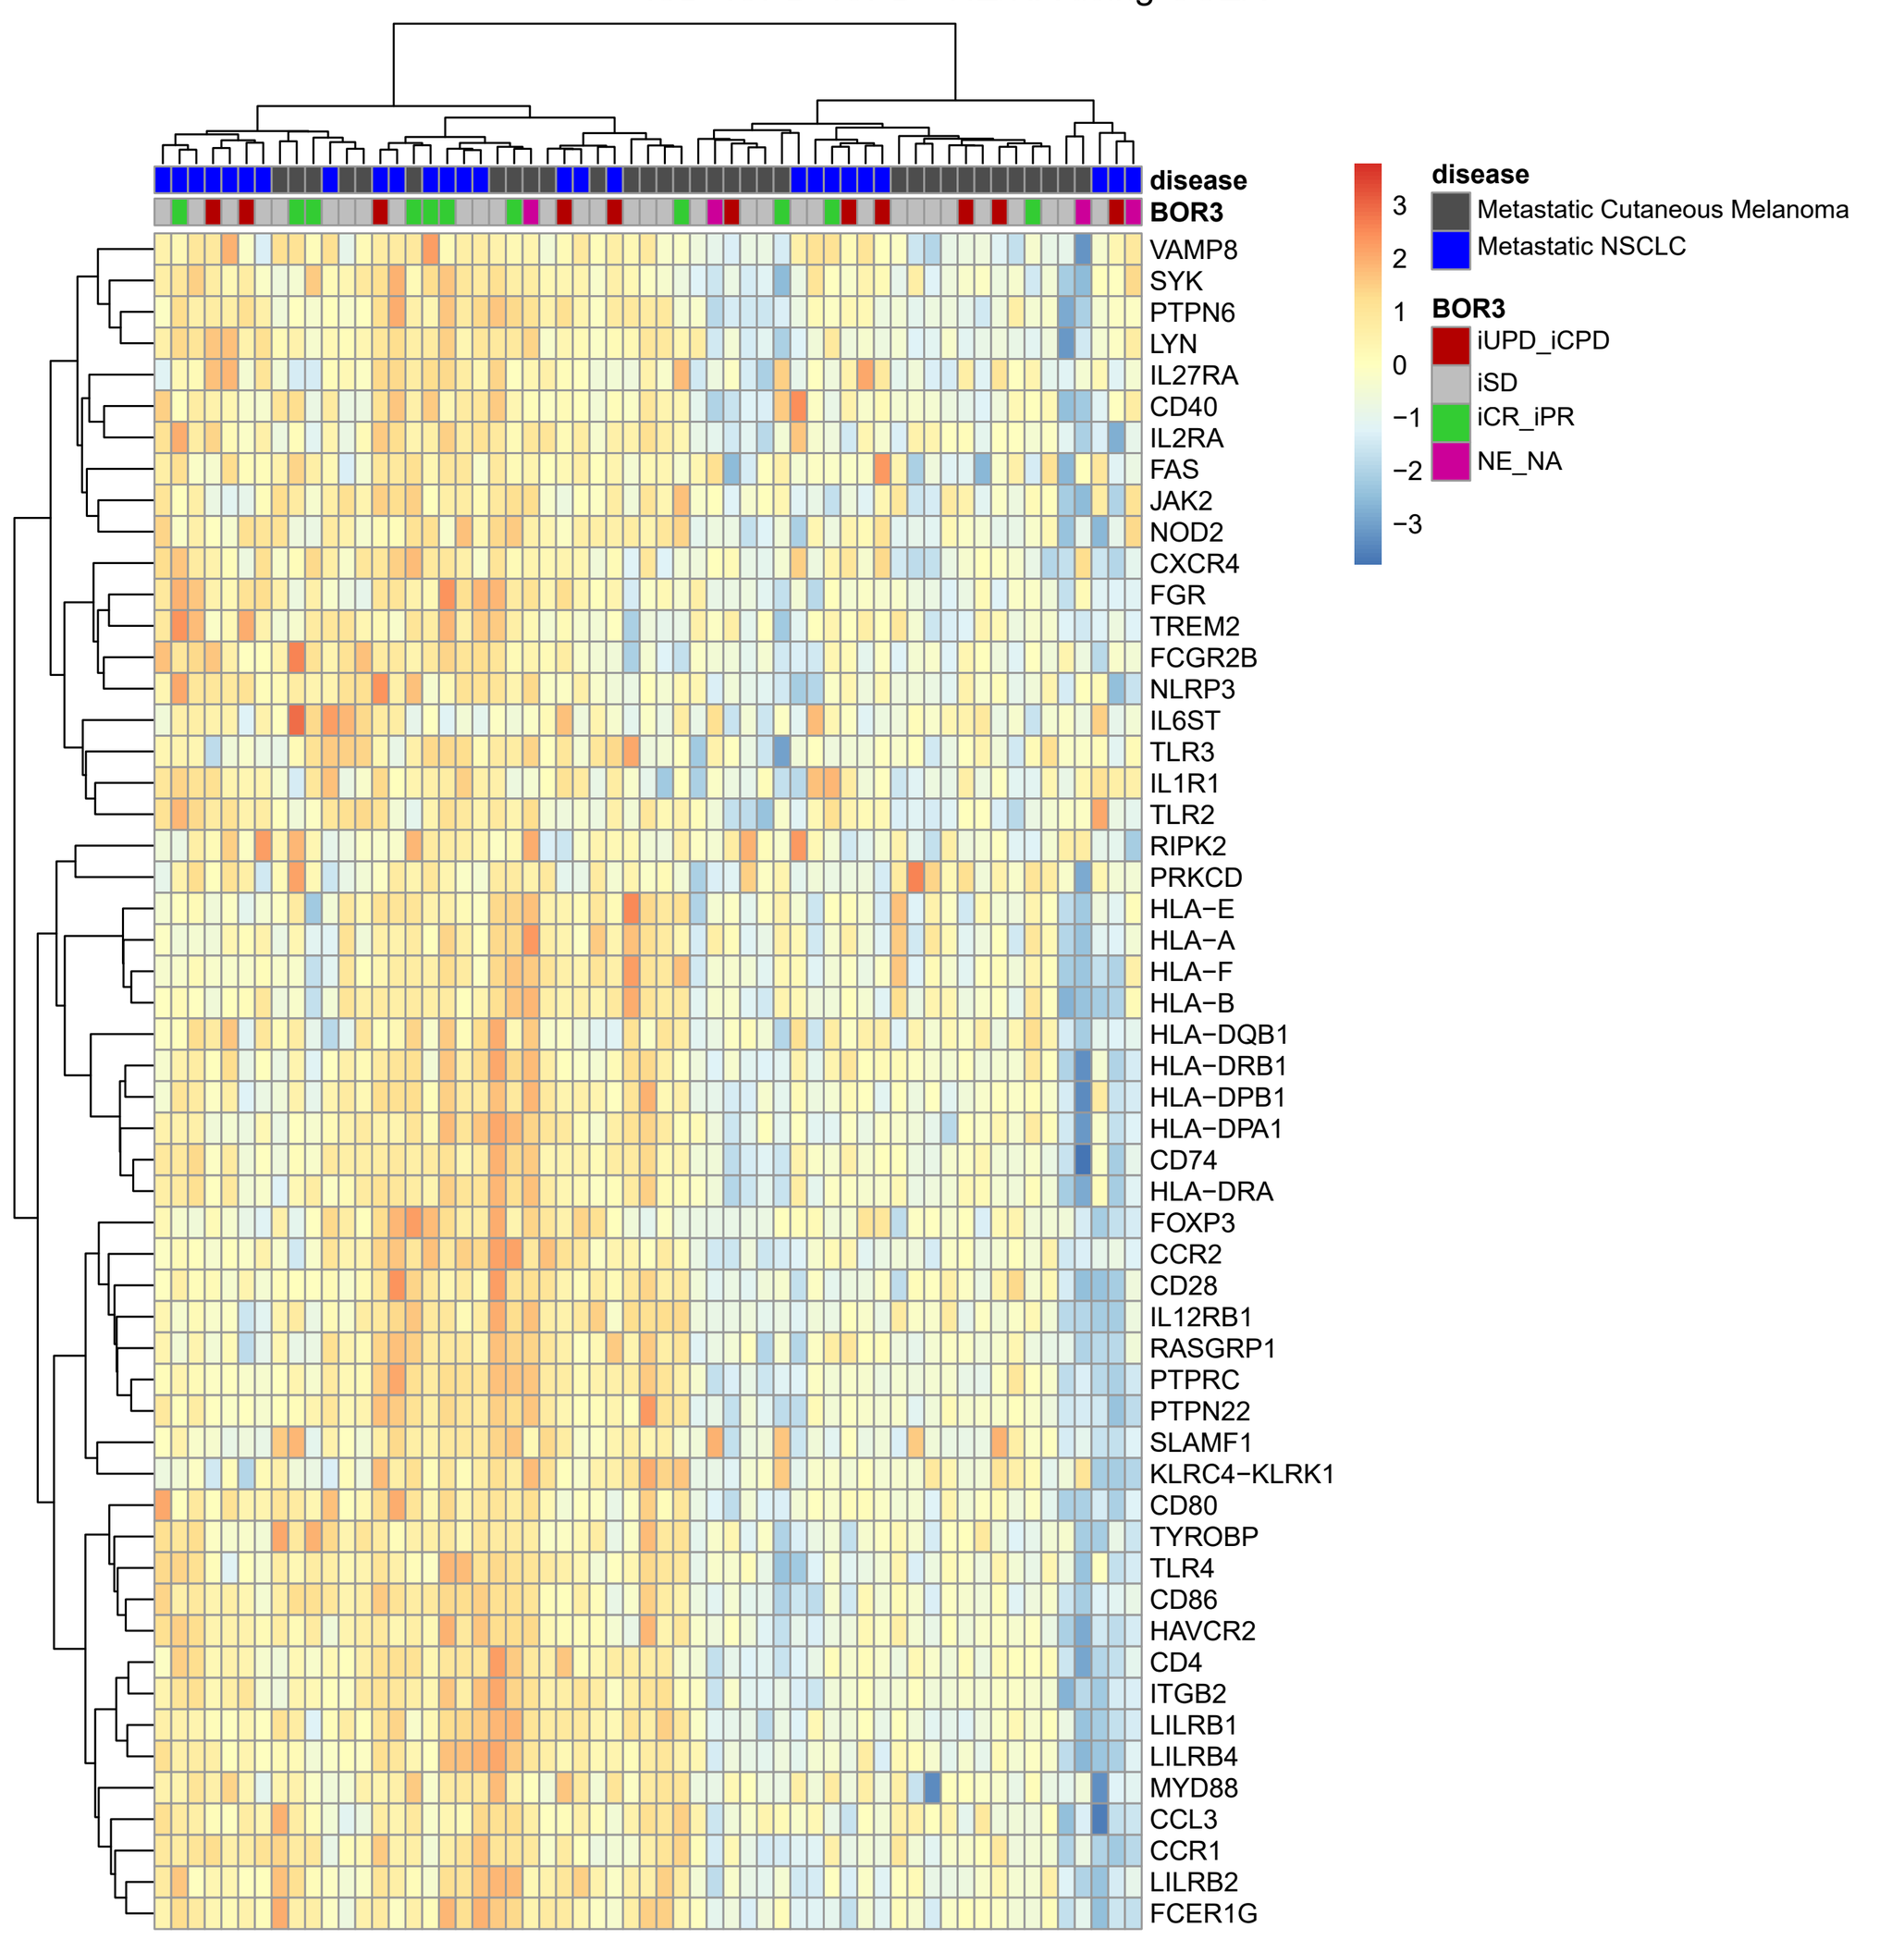
disease
BOR3
Metastatic Cutaneous Melanoma
Metastatic NSCLC
iUPD_iCPD
iSD
iCR_iPR
NE_NA
VAMP8
SYK
PTPN6
LYN
IL27RA
CD40
IL2RA
FAS
JAK2
NOD2
CXCR4
FGR
TREM2
FCGR2B
NLRP3
IL6ST
TLR3
IL1R1
TLR2
RIPK2
PRKCD
HLA−E
HLA−A
HLA−F
HLA−B
HLA−DQB1
HLA−DRB1
HLA−DPB1
HLA−DPA1
CD74
HLA−DRA
FOXP3
CCR2
CD28
IL12RB1
RASGRP1
PTPRC
PTPN22
SLAMF1
KLRC4−KLRK1
CD80
TYROBP
TLR4
CD86
HAVCR2
CD4
ITGB2
LILRB1
LILRB4
MYD88
CCL3
CCR1
LILRB2
FCER1G

Connectivity map of 54 leading edge genes showing protein-protein interactions

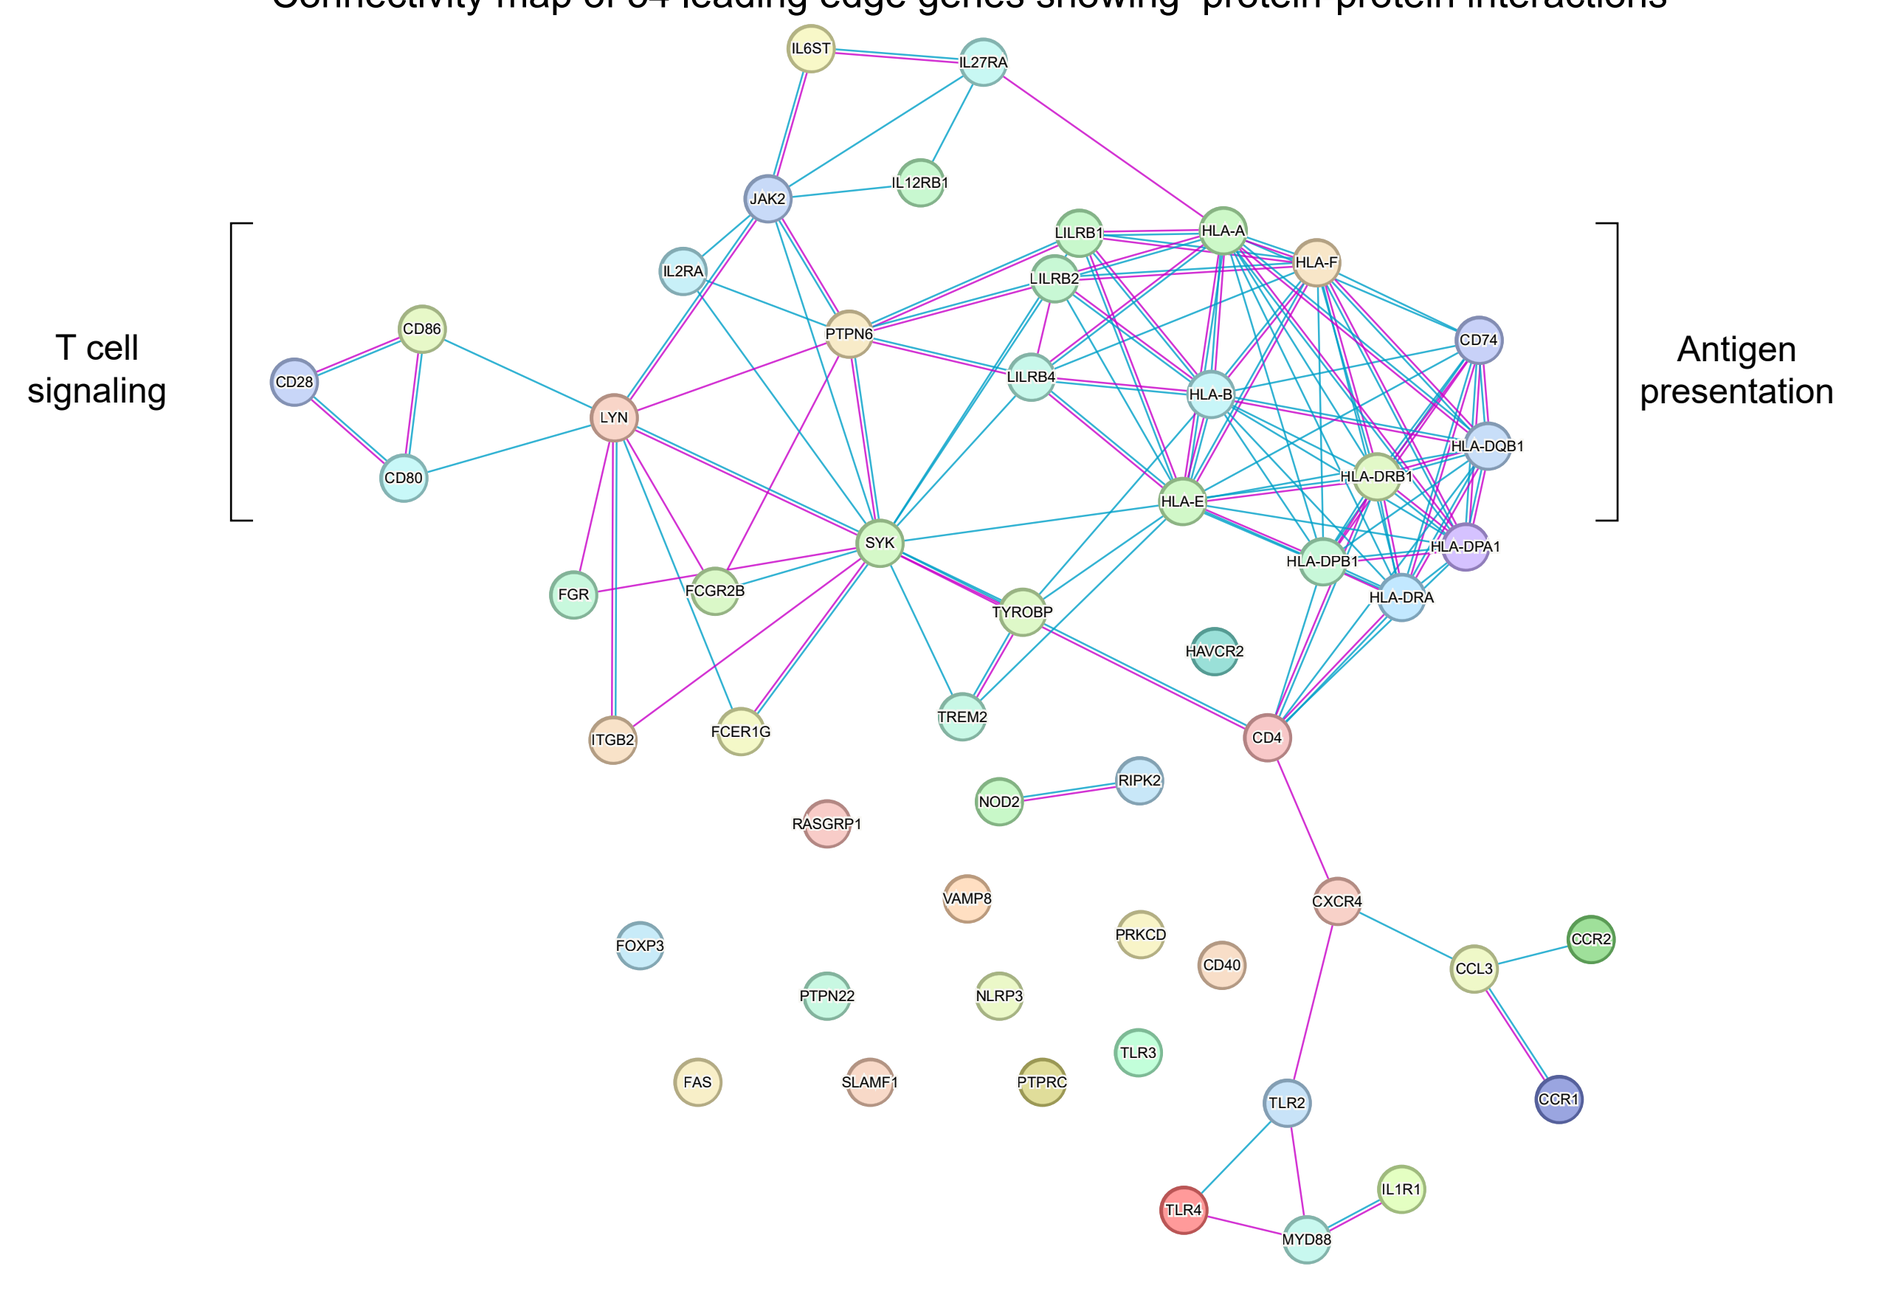
T cell signaling
Antigen presentation

Abbreviations

AEs: adverse events; ALK: anaplastic lymphoma kinase; BOR3: best overall response grouped into 3 categories; BP: biological process; BV: brentuximab vedotin; CC: cellular component; CD: cluster of differentiation; CPI: checkpoint inhibitor; CR: complete response; DCR: disease control rate; DOR: duration of response; ECOG: Eastern Cooperative Oncology Group; EGFR: epidermal growth factor receptor; FAS: full analysis set; FDR: false discovery rate; GO: gene ontology; GSEA: gene set enrichment analysis; HP: human phenotype; iCPD: immune confirmed progression; IHC: immunohistochemistry; IMAEs: immune-mediated AEs; iRECIST: immune-RECIST; iUPD: immune unconfirmed progressive disease; LE: leading edge (gene identified using GSEA); MF: molecular function; NCI CTCAE: National Cancer Institute Common Terminology Criteria for Adverse Events; NE: not evaluable; NA: not available subjects including those who discontinued treatment with no post-baseline response; NES: normalized enrichment score; NSCLC: non-small cell lung cancer; ORR: objective response rate; padv: adjusted p-value; PD-L1: programmed cell death ligand 1; pembro: pembrolizumab; PFS: progression-free survival; PR: partial response; pval: p-value; RECIST: Response Evaluation Criteria in Solid Tumors; RNAseq: ribonucleic acid sequencing; R/R: relapsed/refractory; SD: stable disease; TEAE: treatment-emergent adverse events; TESAEs: treatment-emergent serious adverse events; TME: tumor microenvironment; Tregs: regulatory T-cells

Acknowledgements

Thank you to the patients and their families for their participation in the study, and to all research personnel for their support of this important trial.
Direct funding for this research was issued by Seagen Inc. through the joint financial support of Seagen Inc. and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.
Clinical analysis support was provided by Eeman Shaikh (Seagen Inc.). Translational research support was provided by Ryan Heiser (Seagen Inc.). Medical writing support was provided by Lauren Angotti (BioBridges LLC) with funding from Seagen Inc.

Disclosures

SL: Travel expenses from Dava Oncology. MC: Employee of Merck, Inc. HL, JB, KR, SK, and BO are employees of Seagen, Inc.

Society for Immunotherapy of Cancer (SITC); San Diego, CA; November 1-5, 2023
Abstract No. 699