Quality of Life of Metastatic Urothelial Cancer Patients Treated With Enfortumab Vedotin Following Platinum-containing Chemotherapy and a Checkpoint Inhibitor: Data From EV-201 Cohort 1

Bradley A. McGregor[1], Peter H. O'Donnell[2], Arjun Balar[3], Daniel Petrylak[4], Jonathan E. Rosenberg[5], Evan Y. Yu[6], David I. Quinn[7], Sonali Shah[8], Juan Pinelli[9], Zsolt Hepp[9], Matthew D. Galsky[10]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]University of Chicago, Chicago, IL, USA; [3]Perlmutter Cancer Center at NYU Langone Health, New York, NY, USA; [4]Yale Cancer Center, New Haven, CT, USA; [5]Memorial Sloan Kettering Cancer Center and Weill Cornell Medical College, New York, NY, USA; [6]Fred Hutchinson Cancer Research Center and University of Washington, Seattle, WA, USA; [7]University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [8]Astellas Pharma US, Inc., Northbrook, IL, USA; [9]Seattle Genetics, Bothell, WA, USA; [10]Tisch Cancer Institute, Icahn School of Medicine at Mount Sinai, New York, NY, USA

Background

- Patients with locally advanced or metastatic urothelial carcinoma (la/mUC) experience significant declines in health-related quality of life (HRQoL) due to a number of factors, including disease symptoms such as pain, decreased vigor (possibly resulting from radiotherapy), sleep disturbance, and social or psychological problems such as feelings of isolation and a sense of being a burden to friends and family.[1-3]
- EV-201 (NCT03219333) is a single-arm, 2-cohort study of enfortumab vedotin (EV) in patients with la/mUC who were treated with prior platinum-containing chemotherapy and anti-PD-1/L1 therapy (Cohort 1), or anti-PD-1/L1 therapy and no prior chemotherapy and are cisplatin-ineligible (Cohort 2).[4]
 - In Cohort 1, EV was associated with an objective response rate of 44% (95% confidence interval [CI], 35.1–53.2%),[4] compared with a historical response rate of 10%.[5]
 - 12% of patients in Cohort 1 had a complete response.[4]
 - Median time to response was 1.84 months (range, 1.2–9.2 months), and median duration of response was 7.6 months (range, 0.95–11.30+ months).[4]
 - Target lesions were reduced in 84% of evaluable patients, and median progression-free survival and overall survival were 5.8 months (95% CI, 4.9–7.5 months) and 11.7 months (95% CI, 9.1 months to not reached), respectively.[4]
 - The observed safety profile of EV was manageable and tolerable.[4]
- Based on results from the EV-201 trial, EV-ejfv received FDA approval in December 2019 for adults with la/mUC previously treated with anti-PD-1/L1 therapy and a platinum-containing chemotherapy in the neoadjuvant/adjuvant, la/mUC setting. This indication was approved under accelerated approval based on tumor response rate. Continued approval may be contingent upon verification and description of clinical benefit in confirmatory trials.[6]
- EV is now recommended in NCCN Clinical Practice Guidelines In Oncology (NCCN Guidelines®) as a preferred regimen (category 2A) for subsequent-line systemic therapy in patients with la/mUC who have already received platinum and anti-PD-1/L1 therapy.[7]

Objective

- To explore the impact of EV on HRQoL among patients with la/mUC who were treated with platinum-containing chemotherapy and anti-PD-1/L1 therapy through evaluation of exploratory patient-reported outcome (PRO) endpoints among Cohort 1 of the EV-201 trial.

Methods

EV-201 study design and population

- The full study methods and primary results for EV-201 have been previously published.[4]
- Cohort 1 comprised patients with la/mUC previously treated with both platinum-based chemotherapy and anti-PD-1/L1 therapy who experienced progression during or after their most recent therapy.
- Patients received EV 1.25 mg/kg (up to a maximum of 125 mg for patients ≥100 kg) intravenously on days 1, 8, and 15 of each 28-day cycle, and treatment continued until disease progression, unacceptable toxicity, consent withdrawal, or investigator decision.

HRQoL assessments

- PRO measures were included in EV-201 as exploratory endpoints for the assessment of HRQoL.
- 2 validated instruments were utilized (European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30 [EORTC QLQ-C30] v3 and EuroQol-5 Dimension-3 Level [EQ-5D-3L]),[8,9] with assessments completed at baseline and at the start of each cycle.
 - These PRO instruments have been developed and validated in several oncology disease states and have been used widely in both clinical trials and the clinic setting.
 - Both tools have been used to assess the effect of treatment on HRQoL in mUC trials, including the KEYNOTE-045 and CheckMate 275 trials.[10,11]

EORTC QLQ-C30[8]

- 30-item questionnaire consisting of 5 functional domains, 3 symptom scales, 5 single-item symptom questions, 1 financial impact of disease question, and 2 global quality of life (QoL) questions.
 - Each domain or question is scored 0 to 100.
 - For the global health status/QoL and functional domain scores, higher scores represent better QoL and functioning, respectively. For symptom scales, higher scores represent greater symptomatology.

EQ-5D-3L[9]

- 5-item self-reported measure of functioning and well-being, which assesses 5 dimensions of health, including mobility, self-care, usual activities, pain/discomfort, and anxiety/depression.
- Responses to the 5 items are converted to a weighted health state index (utility score) based on values derived from general population samples.
 - The health utility score is between 0 and 1 where 0 is death and 1 is perfect health.
- In addition to the utility score, this questionnaire also records the respondent's self-rated health status on a vertical graduated (0–100) visual analog scale (VAS).

Results

Study participants

- A total of 128 patients with la/mUC who were previously treated with platinum-containing chemotherapy and anti-PD-1/L1 therapy were enrolled in Cohort 1 (October 8, 2017–July 2, 2018).
 - 125 patients were treated with EV (Table 1); median follow-up as of March 1, 2019 was 10.2 months (range, 0.5–16.5 months), and median duration of treatment was 4.6 months.
 - 20 patients had treatment ongoing at the time of data cutoff.
 - Among all treated patients, 120 (96%) completed the EORTC-QLQ-C30 and 119 (95%) the EQ-5D questionnaire, at baseline.
- Across all cycles, while completion rates decreased due to treatment discontinuation (n=120/119 at baseline to n=12 at cycle 10), ≥86% of subjects with available data at each cycle completed both instruments.

Table 1. Demographics and disease characteristics of Cohort 1 (prior platinum-containing chemotherapy and anti-PD-1/L1 therapy) at baseline

Characteristic	Cohort 1 (N=125)
Male, n (%)	88 (70)
Median age, years (min, max)	69 (40, 84)
ECOG performance status, n (%)	
0	40 (32)
1	85 (68)
Primary tumor location, n (%)	
Bladder/other	81 (65)
Upper tract	44 (35)
Histology type, n (%)	
Urothelial carcinoma only	84 (67)
Urothelial carcinoma with squamous differentiation	15 (12)
Urothelial carcinoma with other histological variants	26 (21)
Metastasis, n (%)	125 (100)
Metastasis sites, n (%)	
Lymph nodes only	13 (10)
Visceral disease	112 (90)
Bone	51 (41)
Liver	50 (40)
Lung	53 (42)
Number of prior systemic la/mUC therapies	
Median (min, max)	3 (1, 6)
≥3, n (%)	63 (50)

ECOG, Eastern Cooperative Oncology Group; la/mUC, locally advanced or metastatic urothelial carcinoma.

EORTC QLQ-C30

- EORTC QLQ-C30 domain scores, inclusive of general QoL (Figure 1), functioning (Figure 2), and symptom scores (Figure 3), remained stable over time.
- Some domains demonstrated trends towards improvement across the study period, including physical functioning (Figure 2B), emotional functioning (Figure 2C), pain (Figure 3B), and fatigue (Figure 3C).

Figure 1. EORTC QLQ-C30 global QoL score through cycle 10

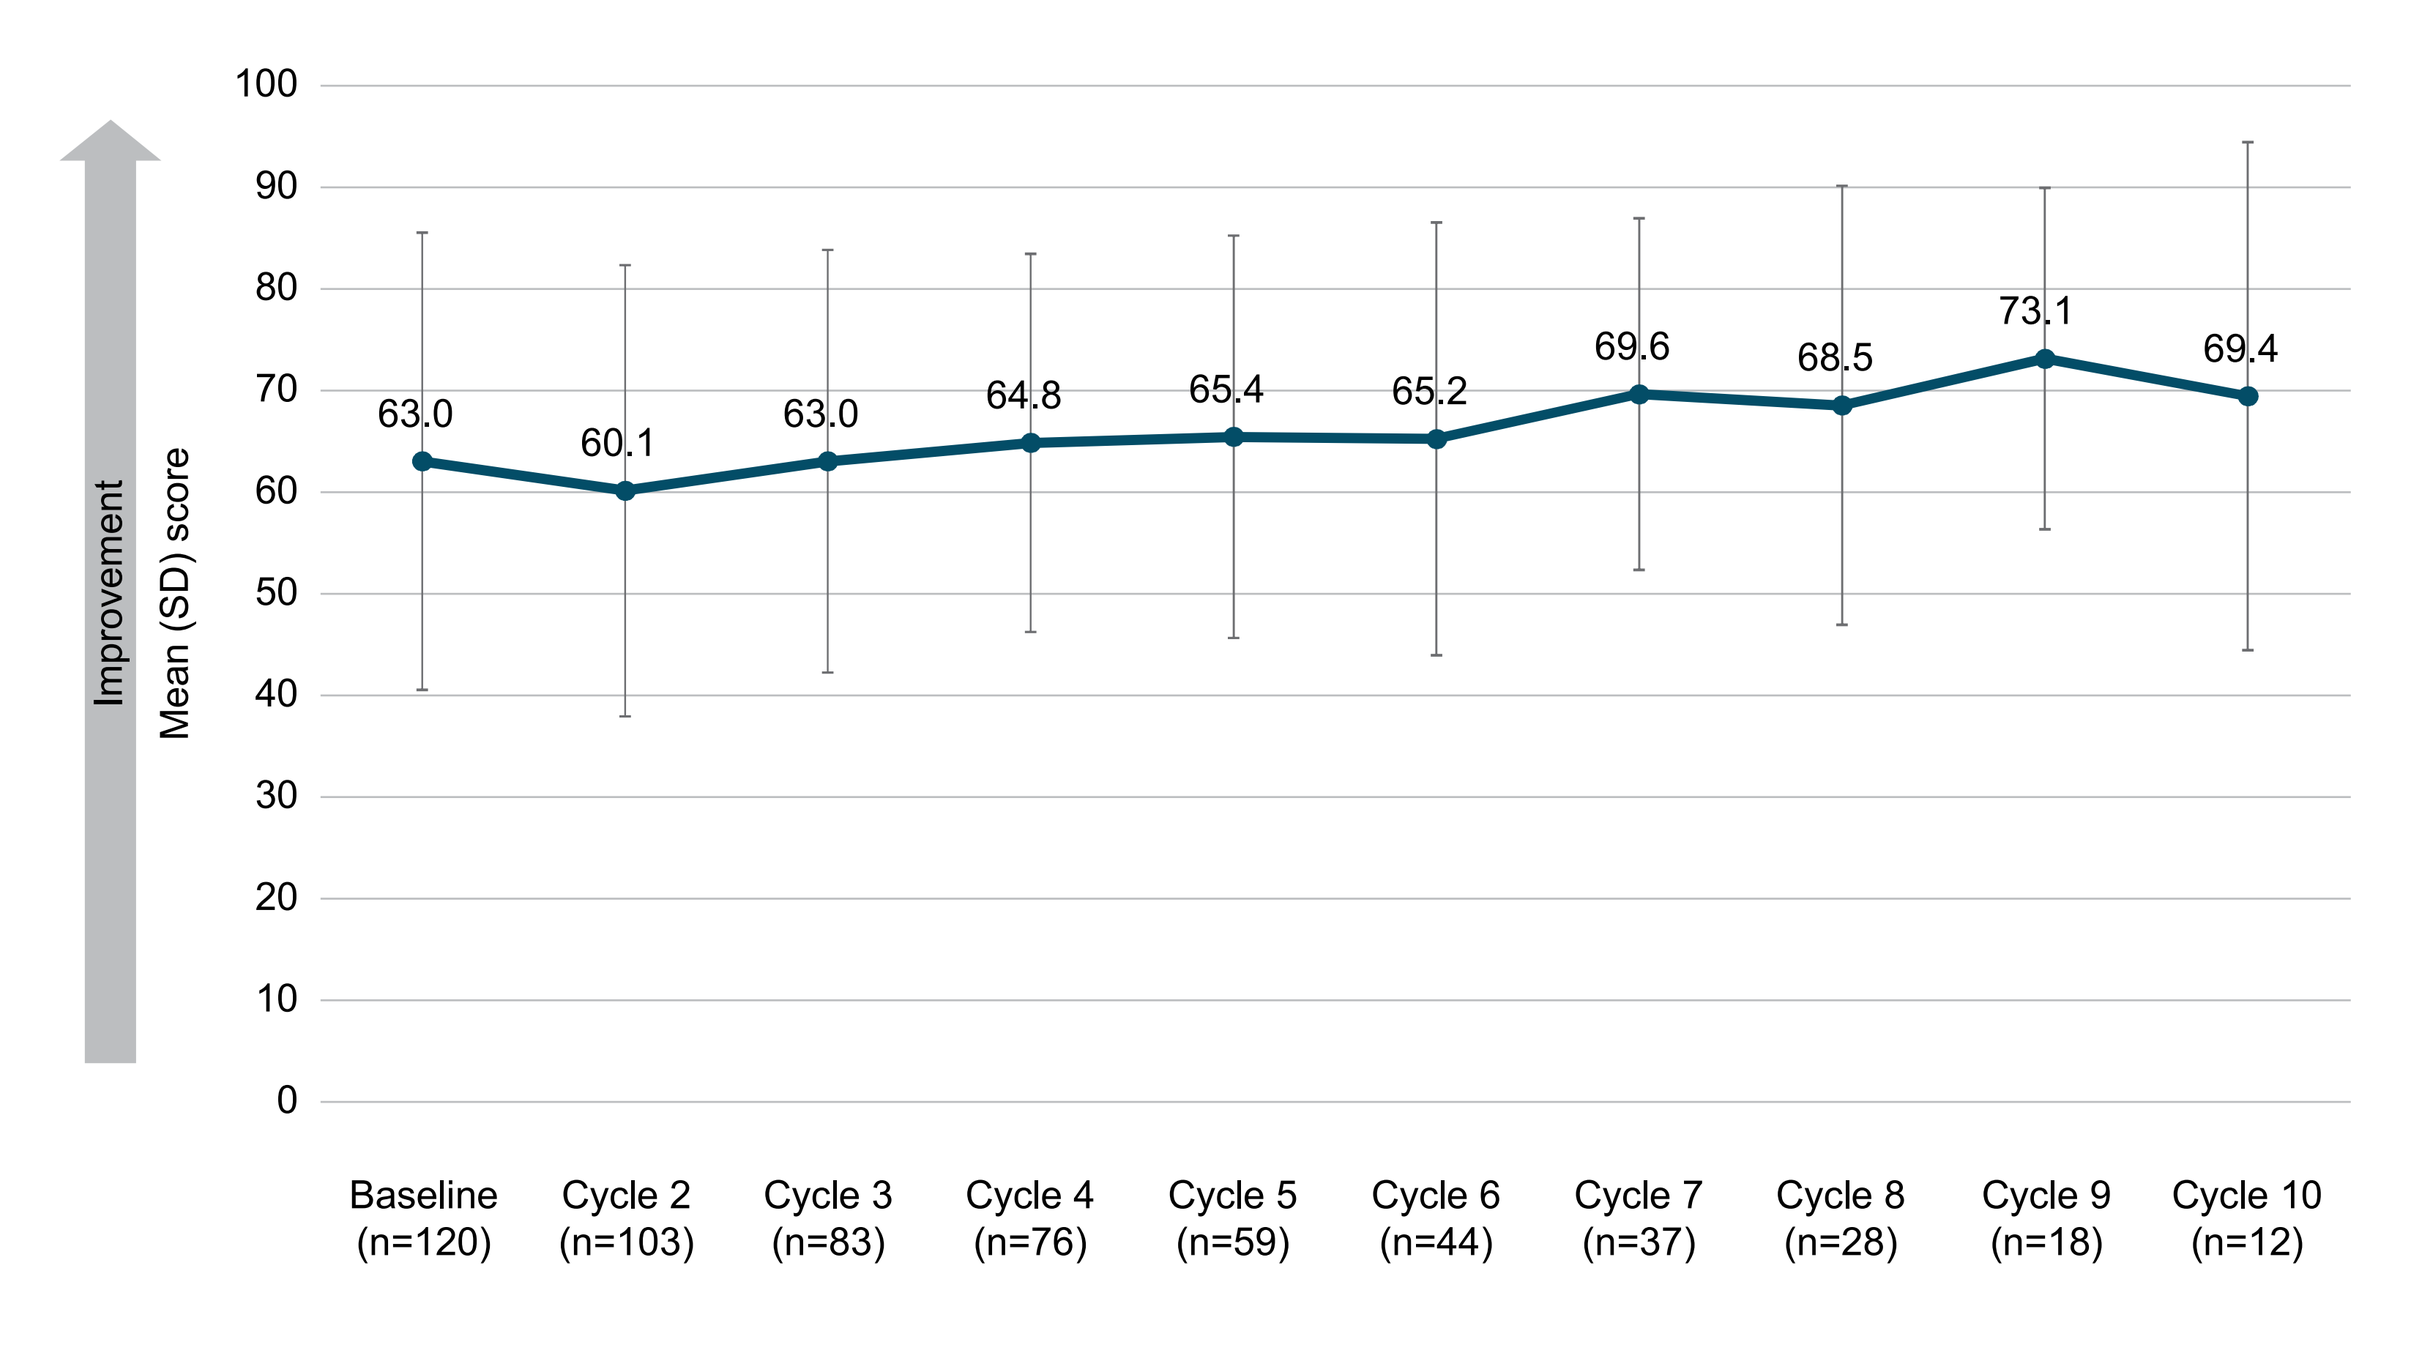
Improvement
Mean (SD) score
63.0
60.1
63.0
64.8
65.4
65.2
69.6
68.5
73.1
69.4
Baseline (n=120)
Cycle 2 (n=103)
Cycle 3 (n=83)
Cycle 4 (n=76)
Cycle 5 (n=59)
Cycle 6 (n=44)
Cycle 7 (n=37)
Cycle 8 (n=28)
Cycle 9 (n=18)
Cycle 10 (n=12)

n numbers represent the total number of patients completing the questionnaire at each cycle.
EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30; QoL, quality of life; SD, standard deviation.

Figure 2. Functional domains[a] from baseline to cycle 10

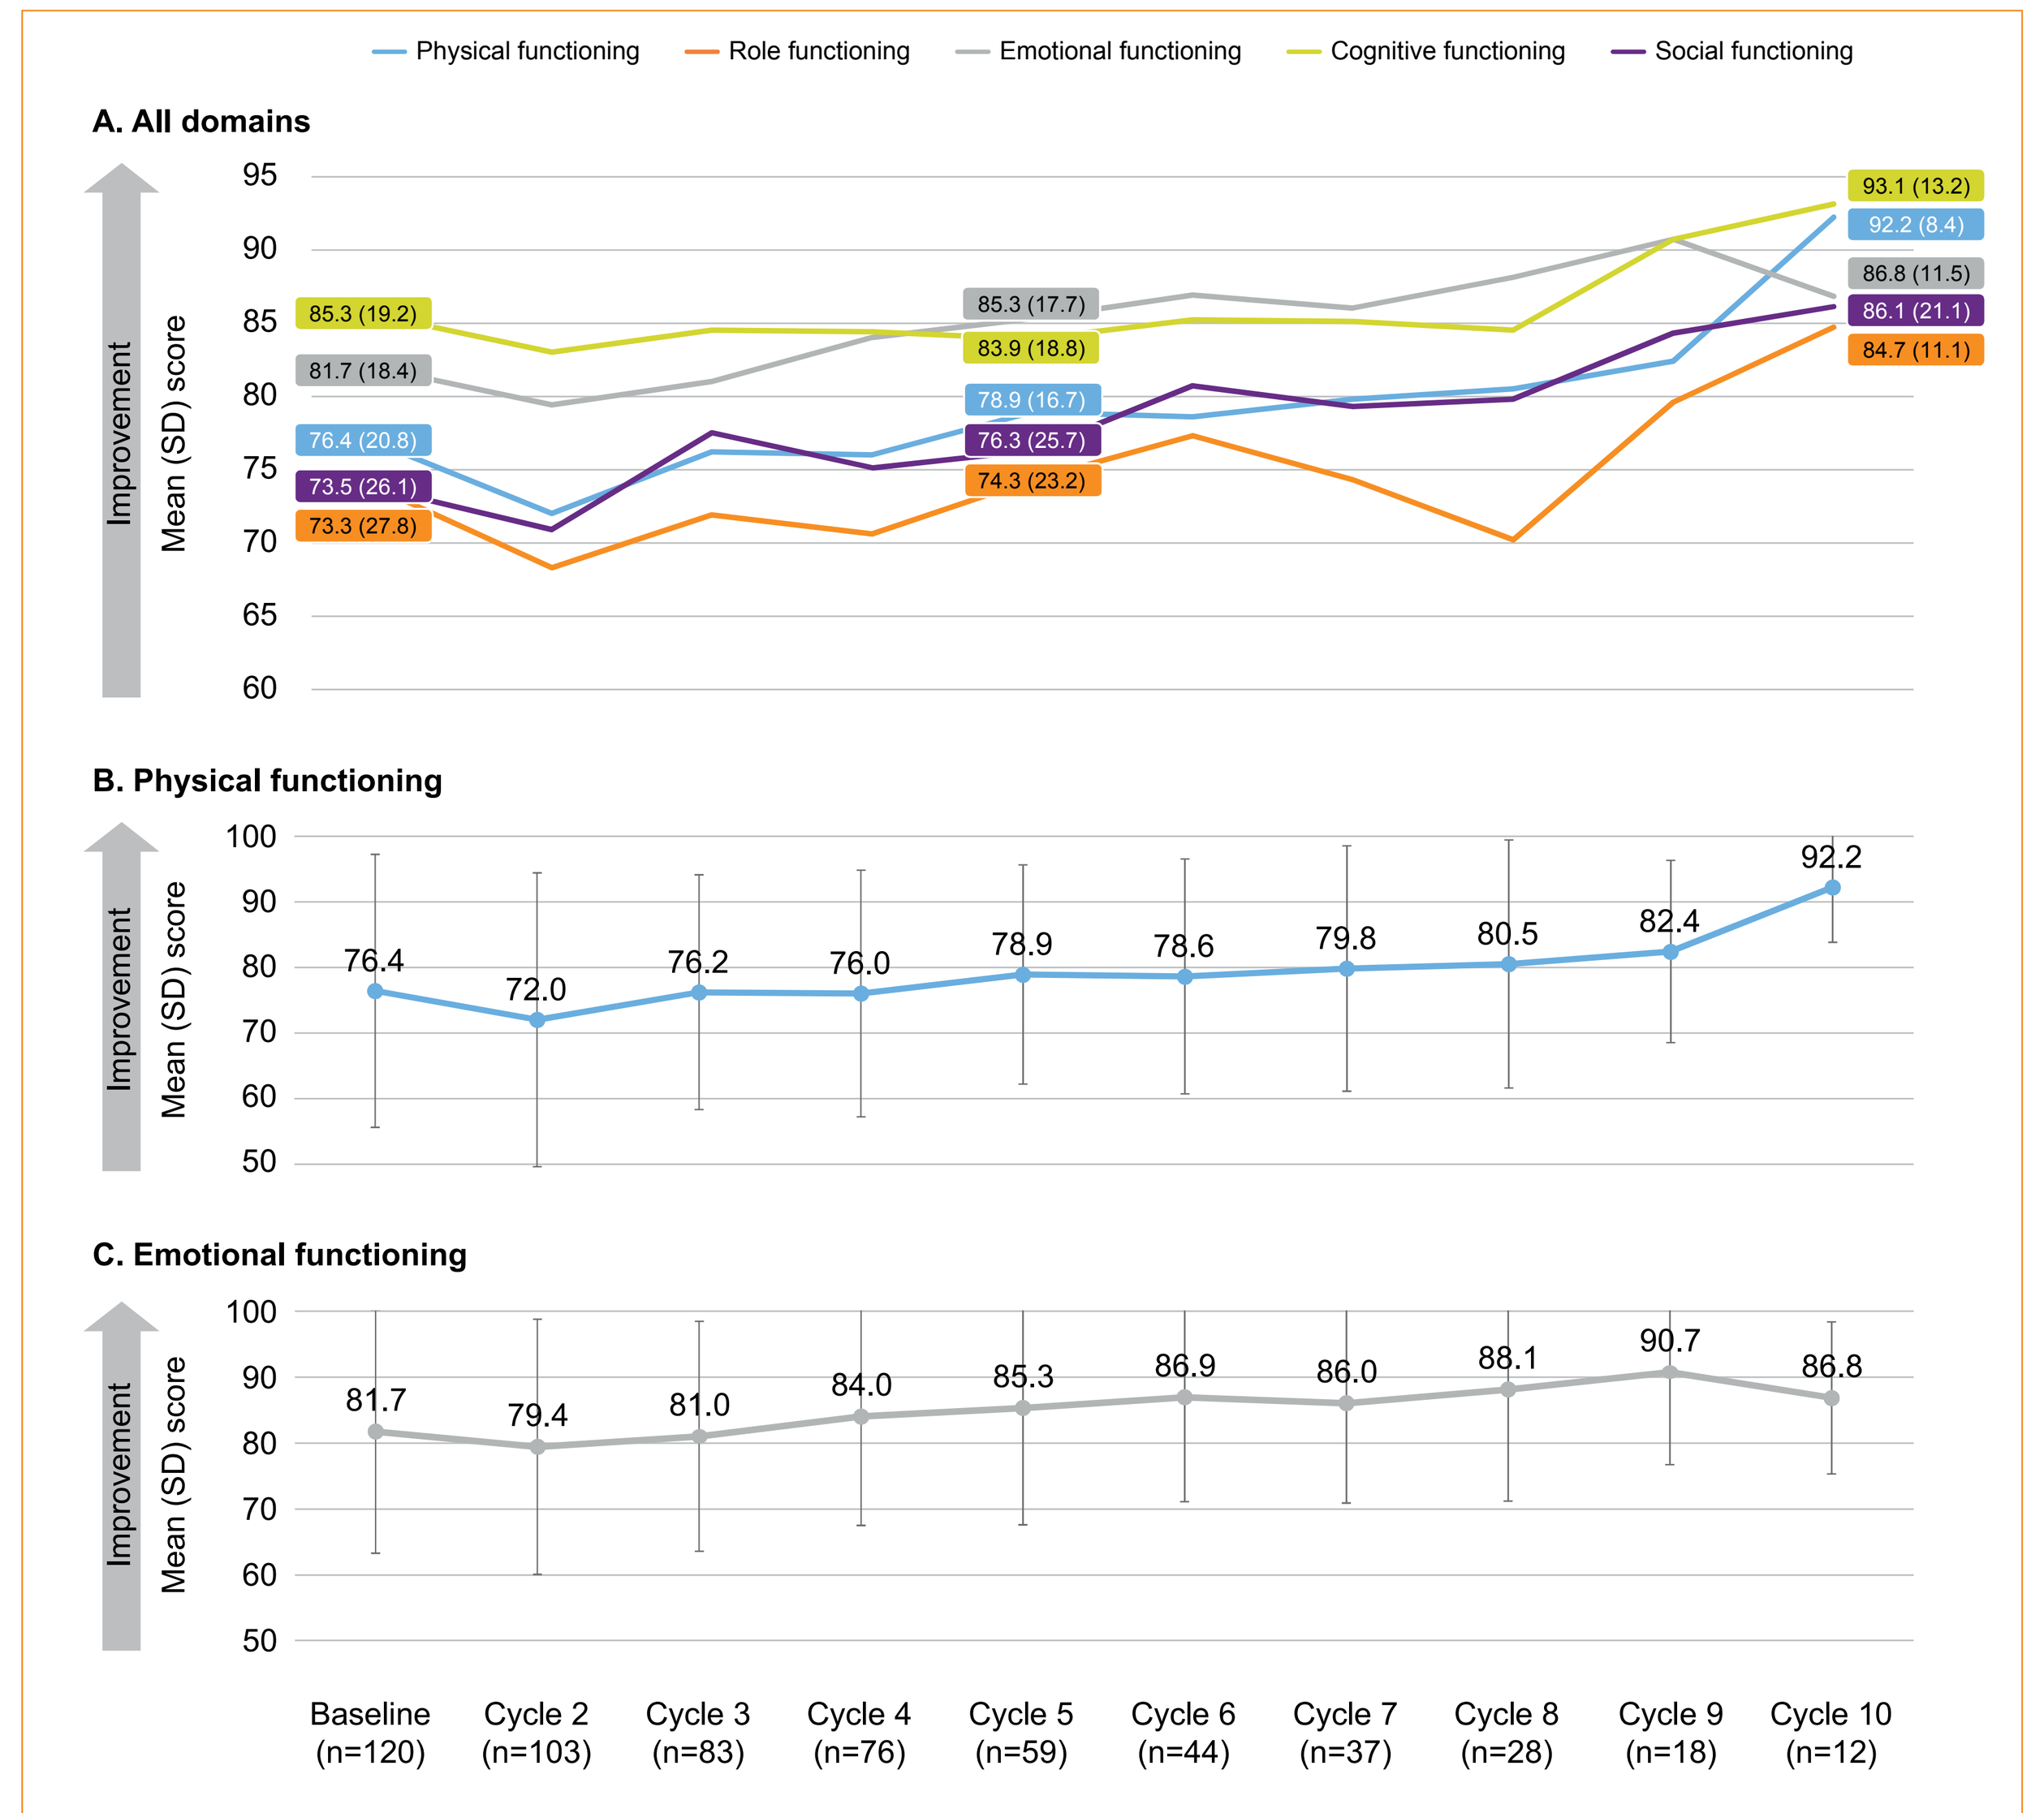
Physical functioning
Role functioning
Emotional functioning
Cognitive functioning
Social functioning
A. All domains
85.3 (19.2)
81.7 (18.4)
76.4 (20.8)
73.5 (26.1)
73.3 (27.8)
85.3 (17.7)
83.9 (18.8)
78.9 (16.7)
76.3 (25.7)
74.3 (23.2)
93.1 (13.2)
92.2 (8.4)
86.8 (11.5)
86.1 (21.1)
84.7 (11.1)
B. Physical functioning
76.4
72.0
76.2
76.0
78.9
78.6
79.8
80.5
82.4
92.2
C. Emotional functioning
81.7
79.4
81.0
84.0
85.3
86.9
86.0
88.1
90.7
86.8
Improvement
Mean (SD) score
Baseline (n=120)
Cycle 2 (n=103)
Cycle 3 (n=83)
Cycle 4 (n=76)
Cycle 5 (n=59)
Cycle 6 (n=44)
Cycle 7 (n=37)
Cycle 8 (n=28)
Cycle 9 (n=18)
Cycle 10 (n=12)

n numbers represent the total number of patients completing the questionnaire at each cycle.
[a] Score range, 0–100. Higher scores represent a positive effect on functioning.
SD, standard deviation.

Figure 3. Symptom scales[a] from baseline to cycle 10

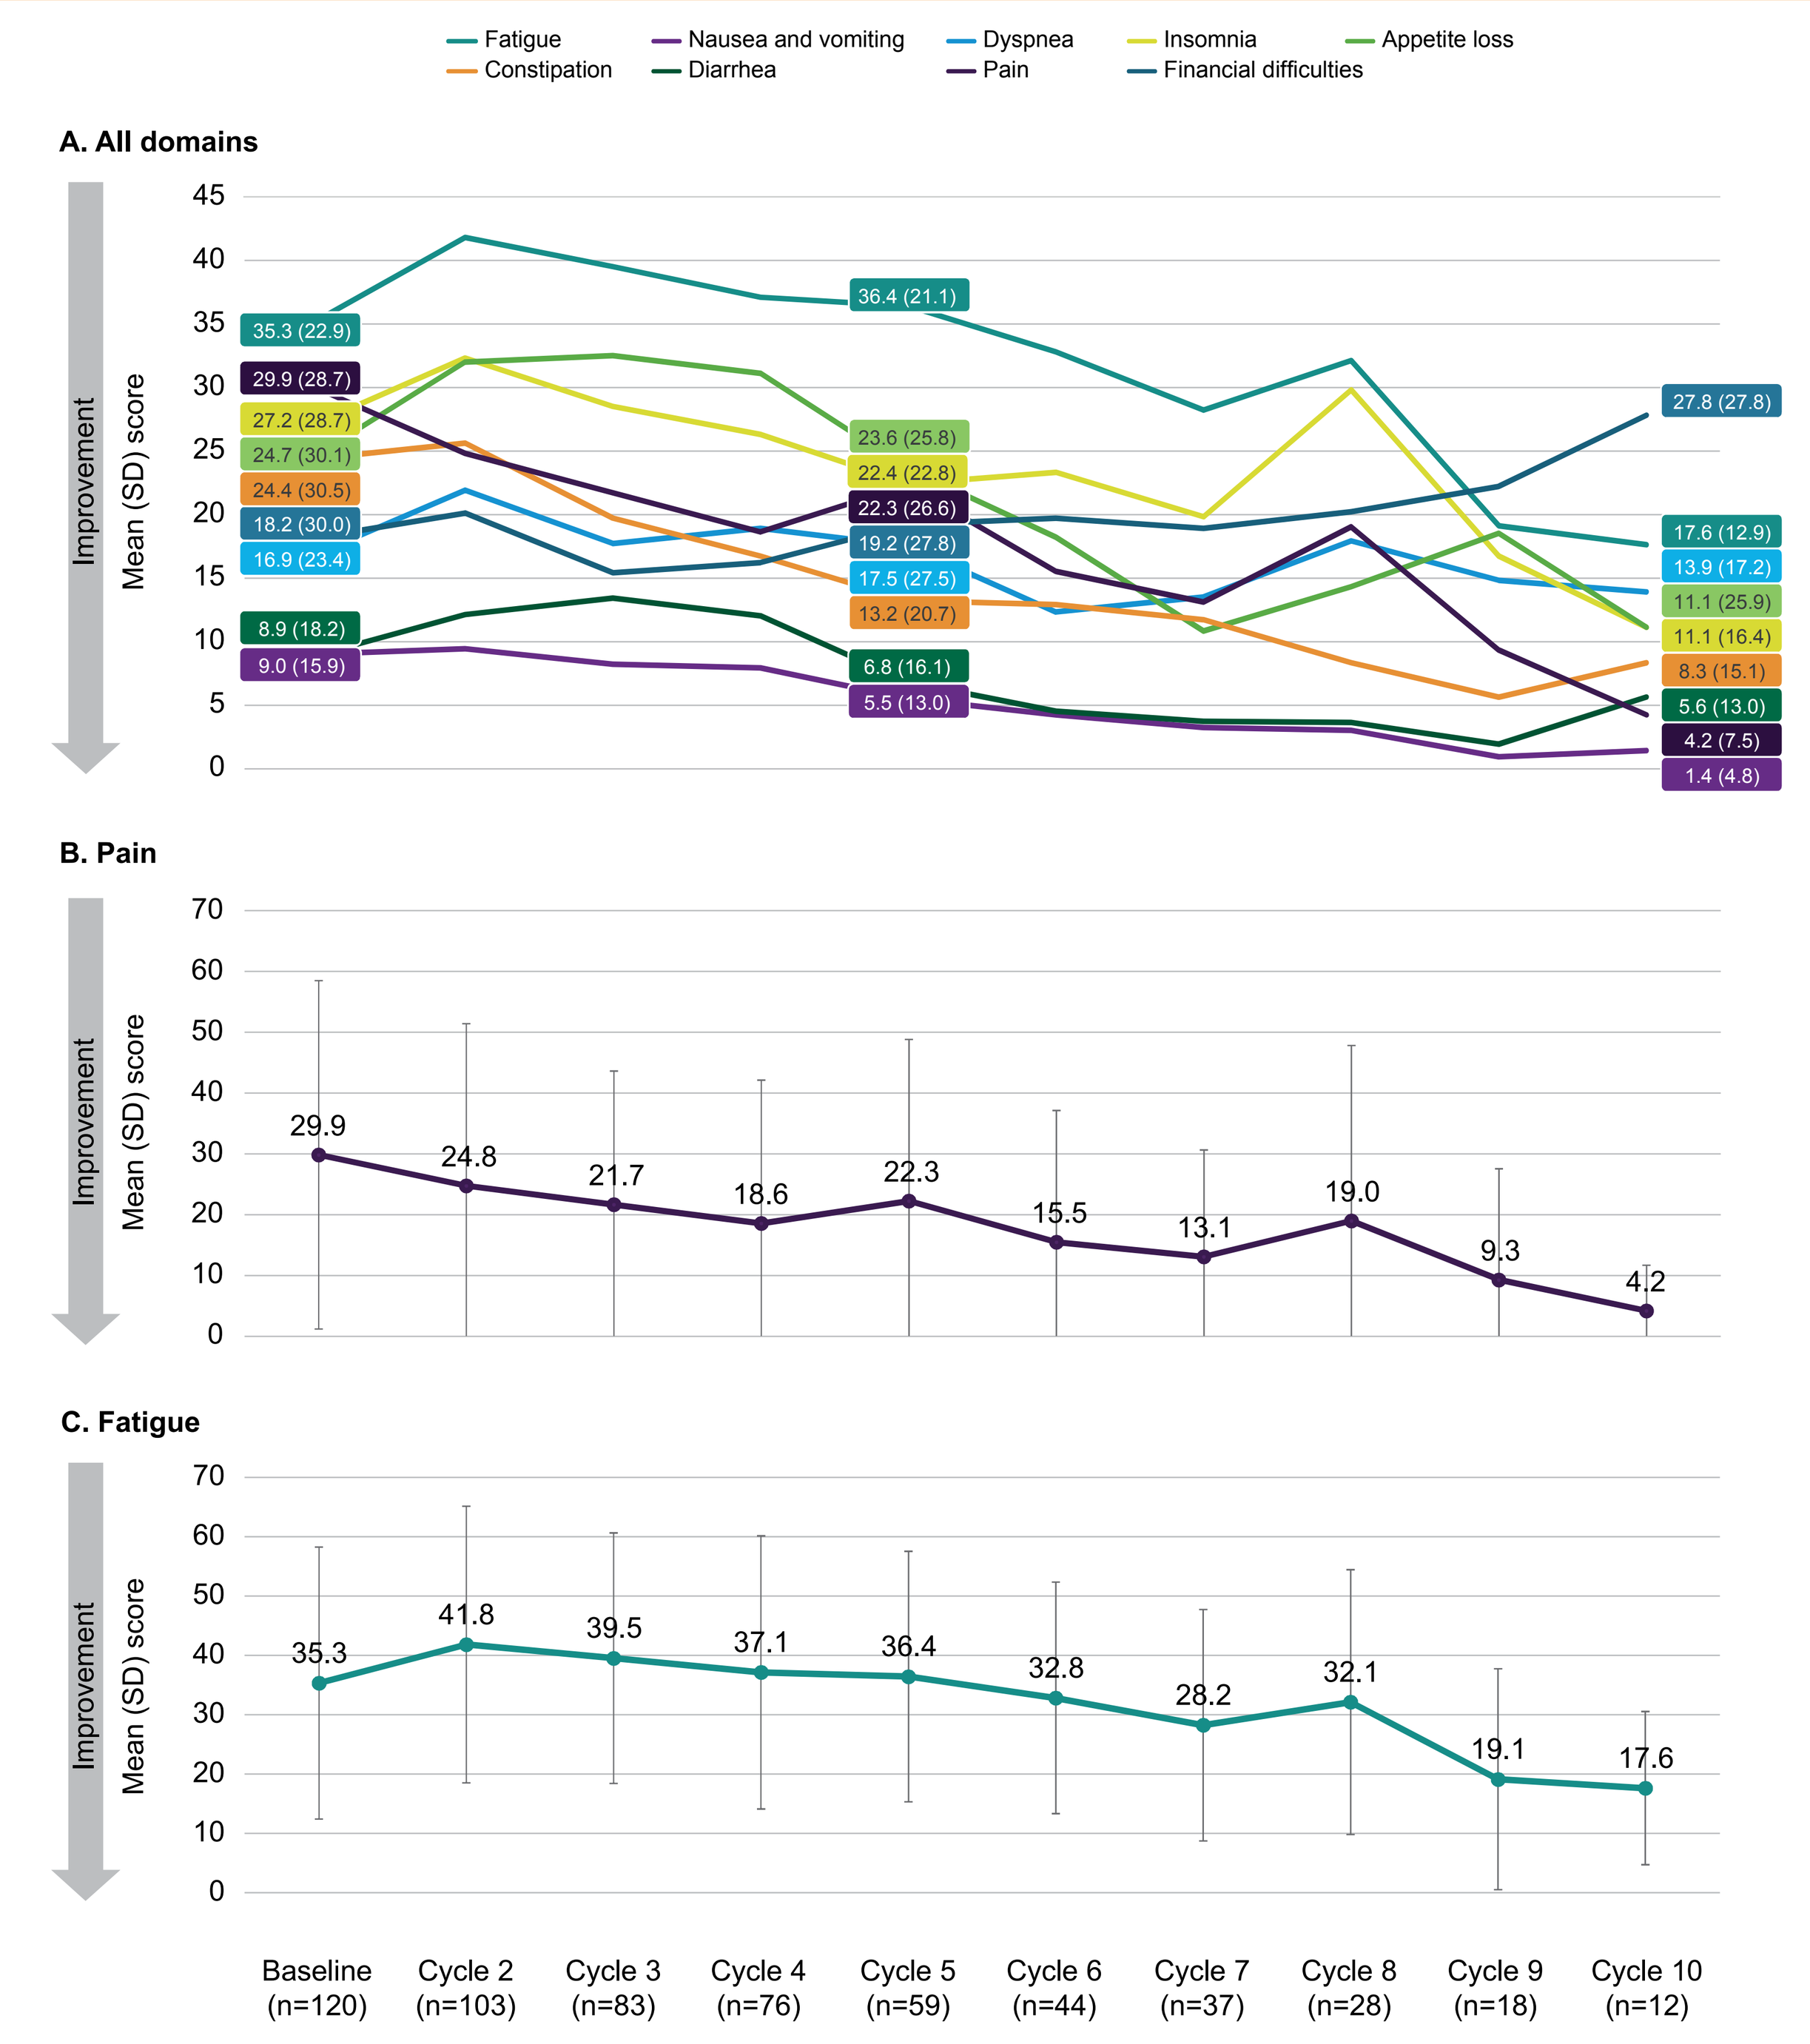
Fatigue
Constipation
Nausea and vomiting
Diarrhea
Dyspnea
Pain
Insomnia
Financial difficulties
Appetite loss
A. All domains
35.3 (22.9)
29.9 (28.7)
27.2 (28.7)
24.7 (30.1)
24.4 (30.5)
18.2 (30.0)
16.9 (23.4)
8.9 (18.2)
9.0 (15.9)
36.4 (21.1)
23.6 (25.8)
22.4 (22.8)
22.3 (26.6)
19.2 (27.8)
17.5 (27.5)
13.2 (20.7)
6.8 (16.1)
5.5 (13.0)
27.8 (27.8)
17.6 (12.9)
13.9 (17.2)
11.1 (25.9)
11.1 (16.4)
8.3 (15.1)
5.6 (13.0)
4.2 (7.5)
1.4 (4.8)
B. Pain
29.9
24.8
21.7
18.6
22.3
15.5
13.1
19.0
9.3
4.2
C. Fatigue
35.3
41.8
39.5
37.1
36.4
32.8
28.2
32.1
19.1
17.6
Improvement
Mean (SD) score
Baseline (n=120)
Cycle 2 (n=103)
Cycle 3 (n=83)
Cycle 4 (n=76)
Cycle 5 (n=59)
Cycle 6 (n=44)
Cycle 7 (n=37)
Cycle 8 (n=28)
Cycle 9 (n=18)
Cycle 10 (n=12)

n numbers represent the total number of patients completing the questionnaire at each cycle.
[a] Score range, 0–100. Higher scores represent greater symptom burden.
SD, standard deviation.

EQ-5D-3L

- EQ-5D utility and VAS scores also remained stable throughout the treatment period (Figure 4).

Figure 4. EQ-5D-3L utility (A) and VAS (B) scores[a] at baseline, cycle 5, and cycle 10

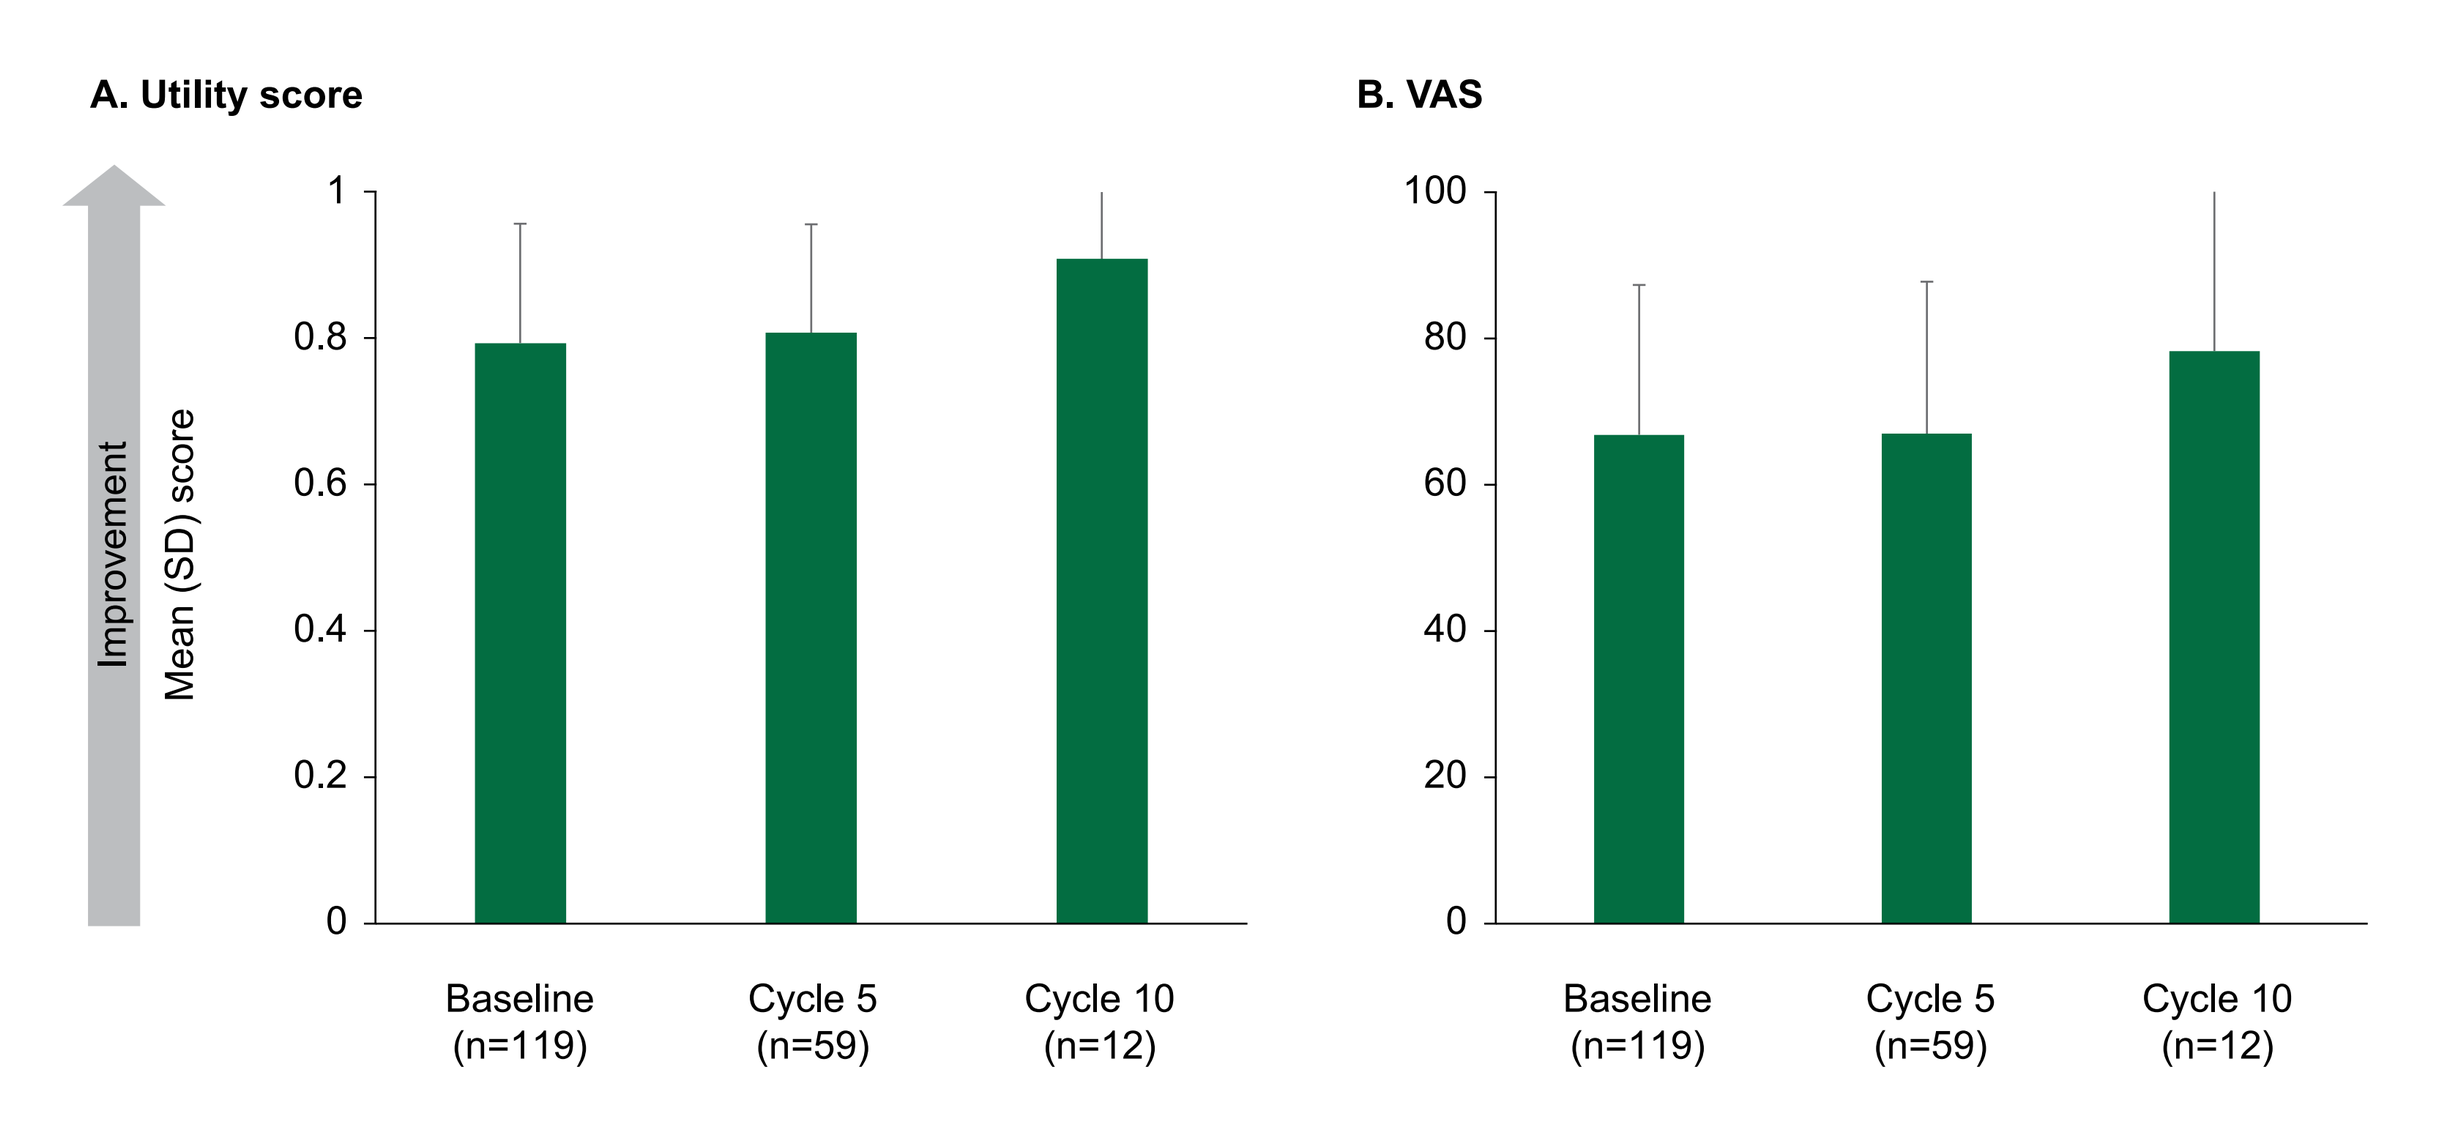
A. Utility score
B. VAS
Improvement
Mean (SD) score
Baseline (n=119)
Cycle 5 (n=59)
Cycle 10 (n=12)

n numbers represent the total number of patients completing the questionnaire at each cycle.
[a] Utility score range, 0–1; VAS, 0–100. Higher scores represent a positive effect on functioning/well-being.
EQ-5D-3L, EuroQol-5 Dimension-3 Level; SD, standard deviation; VAS, visual analog scale.

Limitations

- Pre-treatment HRQoL data are not available and no on-treatment comparator is available as the trial was a single-arm study.
- There was a substantial decrease in available data over time, due to decreasing patient numbers across treatment cycles; decreasing numbers may have resulted from incomplete follow-up at data cutoff (patients who remained on treatment and had not yet reached 10 cycles) as well as treatment discontinuation.
- Variability and small sample size limit definitive conclusions.

Conclusions

- Cohort 1 of the EV-201 trial demonstrated a clinically meaningful response and a manageable safety profile among patients with la/mUC previously treated with platinum-containing chemotherapy and anti-PD-1/L1 therapy[4]; EV-ejfv is now an approved therapy in this patient population based on response rate results from the trial.[6]
- PRO data collected during EV-201 complement the efficacy findings, supporting maintenance of overall HRQoL and modest improvement in symptoms important to patients, including pain and fatigue, with ongoing treatment.

References

1. Perlis N, et al. Qual Life Res. 2014;23(8):2153-67. **2.** Smith AB, et al. BJU Int. 2017;121(4):549-57. **3.** Singer S, et al. Support Care Cancer. 2013;21(5):1383-93. **4.** Rosenberg JE, et al. J Clin Oncol. 2019;37(29):2592-600. **5.** Vaughn DJ, et al. J Clin Oncol. 2002;20(4):937-40. **6.** US Food and Drug Administration. FDA grants accelerated approval to enfortumab vedotin-ejfv for metastatic urothelial cancer; 2019 [URL: https://www.fda.gov/drugs/resources-information-approved-drugs/fda-grants-accelerated-approval-enfortumabvedotin-ejfv-metastatic-urothelial-cancer]. Accessed December 20, 2019. **7.** Referenced with permission from the NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Bladder Cancer V.3.2020. © National Comprehensive Cancer Network, Inc. 2020. All rights reserved. Accessed January 17, 2020. To view the most recent and complete version of the guideline, go online to NCCN.org. NCCN makes no warranties of any kind whatsoever regarding their content, use or application and disclaims any responsibility for their application or use in any way. **8.** Aaronson NK, et al. J Natl Cancer Inst. 1993;85(5):365-76. **9.** Rabin R, de Charro F. Ann Med. 2001;33(5):337-43. **10.** Vaughn DJ, et al. J Clin Oncol. 2018;36(16):1579-87. **11.** Necchi A, et al. J Clin Oncol. 2017;35(15_suppl):4526.

DISCLOSURES

This study was sponsored by Seattle Genetics and Astellas. BAM has served as an advisory board member and consultant for Astellas, AstraZeneca, Bayer, Exelixis, Genentech, Janssen, Nextar, Pfizer, and Seattle Genetics and has received research funding from BMS, Calithera Biosciences, Exelixis, and Seattle Genetics. PHO'D has acted as a consultant and advisory board member for Merck; has equity ownership in Allergan and PRescriptIQ; has received honoraria from Astellas, AstraZeneca, Genentech, Harrison Consulting Group, Inovio, Janssen, Kantar Health, Merck, OncLive, Parexel, Quintiles, and Seattle Genetics; has received research funding grants from Acerta, AstraZeneca, BMS, Boehringer, Genentech, Janssen, Merck, and Seattle Genetics; has other relationships with Advance Medical and Janssen; and is named as a co-inventor on a pending patent for a genomic prescribing system for medication prescribing. AB has served as an advisory board member and consultant for Astellas, AstraZeneca, Cerulean Pharma, Genentech, Incyte, Merck, Pfizer, and Seattle Genetics; has received honoraria from AstraZeneca, Genentech, and Merck; and has received research funding from AstraZeneca, Genentech, Merck, and Seattle Genetics. DP has served as an advisory board member and consultant for Astellas, AstraZeneca, Bayer, Bellicum Pharma, Dendreon, Exelixis, Ferring, Johnson & Johnson, Lilly, Millennium, Medivation, Pfizer, Roche, Sanofi, and Tyme; has equity ownership in Bellicum Pharma and Tyme; has received research funding from Agensys, Astellas, Medivation, AstraZeneca, Bayer, Clovis onc, Dendreon, Endocyte, Genentech, Innocrin Pharma, Johnson & Johnson, Lilly, MedImmune, Millennium, Merck, Novartis, Pfizer, Progenics, Roche, Sanofi, Seattle Genetics, and Sotio; and has provided expert testimony for Celgene and Sanofi. JER has served as an advisory board member and consultant for Adicet Bio, Agensys, AstraZeneca, Bayer, BioClin Therapeutics, BMS, EMD Serono, Fortress Biotech, Genentech, Inovio Pharma, Lilly, Merck, Pharmacyclics, QED Therapeutics, Sanofi, Seattle Genetics, Sensi Biotherapeutics, and Western Oncolytics; has equity ownership in Illumina and Merck; has received honoraria from AstraZeneca, BMS, Chugai Pharma, Medscape, Peerview, UpToDate, and Vindico; holds patents/royalties for a predictor of platinum sensitivity; has received research funding from Agensys, AstraZeneca, Bayer, Genentech, Incyte, Mirati Therapeutics, Oncogenex, Novartis, Seattle Genetics, and Viralytics; and has received travel expenses from BMS and Genentech. EYY has served as a consultant or in an advisory role for Janssen, Bayer, Merck, AstraZeneca, EMD Serono, Incyte, Amgen, Tolmar, QED, Dendreon, Seattle Genetics, and Pharmacyclics; has received research funding from Daiichi Sankyo; and has received research funding to his institution from Agensys, Astellas, Dendreon, Genentech/Roche, Bayer, Merck, and Seattle Genetics. DIQ is an employee of the University of Southern California; has received research funding to his institution from Seattle Genetics, MSD, and Novartis; has served as an advisor/board member for Astellas, AstraZeneca, Bayer, BMS, Dendreon, Exelixis, Roche, Janssen, MSD, Novartis, Pfizer, and Sanofi; and has received travel/accommodation/expenses from Pfizer, MSD, AstraZeneca, BMS, and Roche. SS is a contractor for Astellas. JP was an employee of Seattle Genetics at the time of study. ZH is an employee of Seattle Genetics. MDG has served as an advisory board member and consultant for Aileron Therapeutics, Astellas, AstraZeneca, BioMotiv, BMS, Dendreon, Dracen, EMD Serono, Genentech, GSK, Incyte, Inovio Pharma, Janssen, Lilly, Merck, Novartis, NuMab, Pfizer, and Seattle Genetics; has equity ownership in Rappta Therapeutics; holds patents/royalties for 'Methods and compositions for treating cancer and related Methods (20120322792)'; and has received research funding from AstraZeneca, BMS, Dendreon, Genentech, Janssen, Merck, and Novartis.

ACKNOWLEDGMENTS

Medical writing support was provided by Elizabeth Harvey of Curo, a division of Envision Pharma Group, and funded by Seattle Genetics. Lisa Bloudek of Curta Consulting provided editorial assistance during abstract development and Shang-Ying Liang of Seattle Genetics provided statistical support and QC of results.

Corresponding authors: Bradley A. McGregor (Bradley_McGregor@DFCI.HARVARD.EDU) or Zsolt Hepp (zhepp@seagen.com) or Sonali Shah (sonali.shah.contractor@astellas.com).

Please scan this Quick Response (QR) code with your smartphone app to view an electronic version of this poster.
If you don't have a smartphone, access the poster via the internet at: http://bit.ly/2v1AWIW

Poster presented at the Academy of Managed Care Pharmacy (AMCP) 2020 Virtual Learning Event, April 20–24, 2020
© Copyright 2020