SEA-CD40 is a Non-Fucosylated Anti-CD40 Antibody with Potent Pharmacodynamic Activity in Preclinical Models and Patients with Advanced Solid Tumors

Haley Neff-LaFord[1], Juneko E. Grilley-Olson[2], David C. Smith[3], Brendan Curti[4], Sanjay Goel[5], Timothy M. Kuzel[6], Svetomir N. Markovic[7], Oliver Rixe[8], David L. Bajor[9], Thomas F. Gajewski[10], Martin Gutierrez[11], Elisabeth I. Heath[12], John A. Thompson[13], Sahar Ansari[1], Shyra J. Gardai[1], Celine Jacquemont[1], Michael Schmitt[1], Andrew L. Coveler[13]

[1]Seattle Genetics, Inc., Bothell, WA; [2]University of North Carolina Lineberger Comprehensive Cancer Center/University of North Carolina Chapel Hill, Chapel Hill, NC; [3]University of Michigan, Ann Arbor, MI; [4]Providence Cancer Institute, Portland, OR; [5]Montefiore Medical Center, Bronx, NY; [6]Rush University Medical Center, Chicago, IL; [7]Mayo Clinic Minnesota, Rochester, MN; [8]University of New Mexico Comprehensive Cancer Center, Albuquerque, NM; [9]Case Western Reserve University/University Hospitals Cleveland Medical Center, Cleveland, OH; [10]University of Chicago Medical Center, Chicago, IL; [11]Hackensack University Medical Center, Hackensack, NJ; [12]Karmanos Cancer Institute, Detroit, MI; [13]Seattle Cancer Care Alliance/University of Washington, Seattle, WA

SEA-CD40 description and proposed mechanisms of action

- CD40 is a member of the tumor necrosis factor receptor superfamily expressed on antigen-presenting cells (APCs) that functions as a co-stimulatory receptor. [1]
- Antibodies targeting CD40 induce innate immune activation that can support generation of antigen-specific antitumor T cell responses in addition to inducing antibody-mediated target cell killing of CD40+ cancer cells. These multiple mechanisms may have therapeutic benefit.
- SEA-CD40 is an investigational agonistic non-fucosylated CD40-directed humanized monoclonal IgG1 antibody that exhibits enhanced binding to FcγRIIIa.
- Preclinically, the enhanced effector function of SEA-CD40 confers greater immune stimulation and antitumor activity relative to other CD40-directed therapies.

SEA-CD40 Proposed Mechanisms of Action

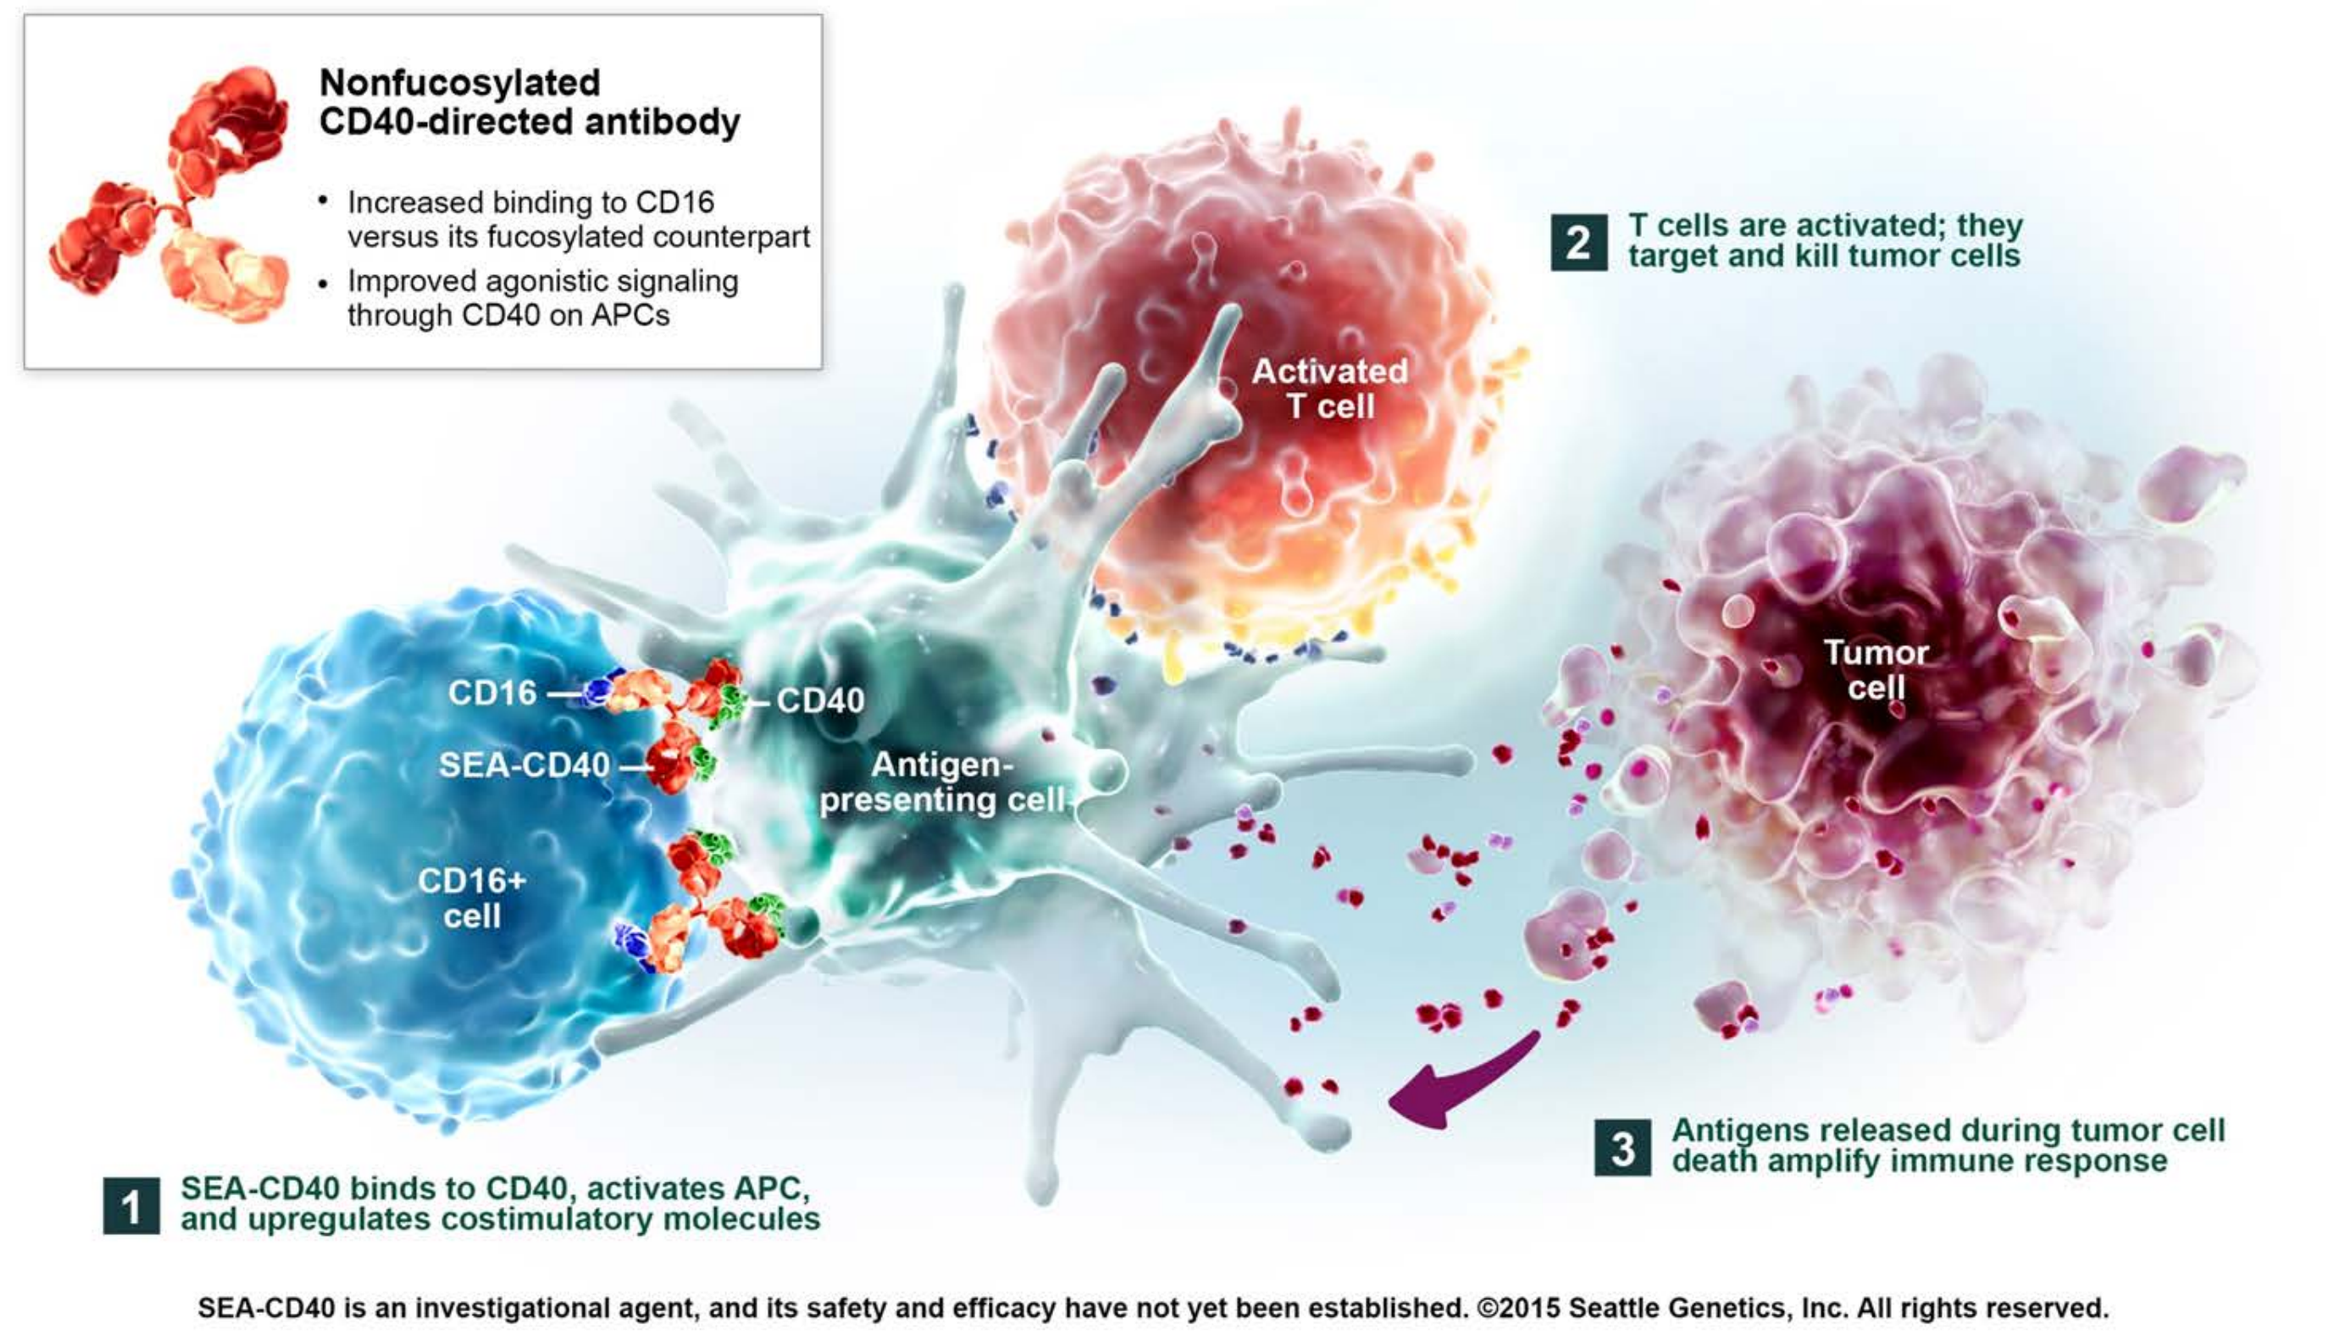
Nonfucosylated CD40-directed antibody
• Increased binding to CD16 versus its fucosylated counterpart
• Improved agonistic signaling through CD40 on APCs
2 T cells are activated; they target and kill tumor cells
Activated T cell
Tumor cell
CD16
SEA-CD40
CD40
Antigen-presenting cell
CD16+ cell
3 Antigens released during tumor cell death amplify immune response
1 SEA-CD40 binds to CD40, activates APC, and upregulates costimulatory molecules

SEA-CD40 is an investigational agent, and its safety and efficacy have not yet been established. ©2015 Seattle Genetics, Inc. All rights reserved.

1. Vonderheide RH. Clin Cancer Res 2007

High FcγRIIIa binding differentiates SEA-CD40 from other CD40 agonists

	SEA-CD40[1]	Dacetuzumab	APX005M	ADC-1013	Selicrelumab	2141-V11[2]
Developer	Seattle Genetics	Seattle Genetics	Apexigen	Janssen/ Alligator	Roche	Rockefeller Univ.
Antibody class	Humanized IgG1	Humanized IgG1	Humanized IgG1	Fully human IgG1	Fully human IgG2	Fully human IgG2
Fc backbone[3] modification impact	↑FcRγIIIa binding	Native	↑FcγRIIa&b ↓FcγRIIIa binding	Native	Native	↑FcRγIIb binding
Antibody used for activity studies	SEA-CD40	Dacetuzumab	APX005M S267E[4]	G12[5]	21.4.1[6]	SGN-40/11B[7]

[1]Parent antibody is dacetuzumab; [2]Parent antibody is selicrelumab; [3]Vonderheide RH. Annu Rev Med 2020, 71:47-58; [4]Based on sequence in US patent US9676861B2; [5]Based on sequence in US patent application US20170226217A1; [6]Based on clone described in Cancer Immunology Research March 2015 3; 236; [7]Dacetuzumab sequence altered to increase FcγRIIb binding as a surrogate for 2141-V11

Binding to FcγRIIa

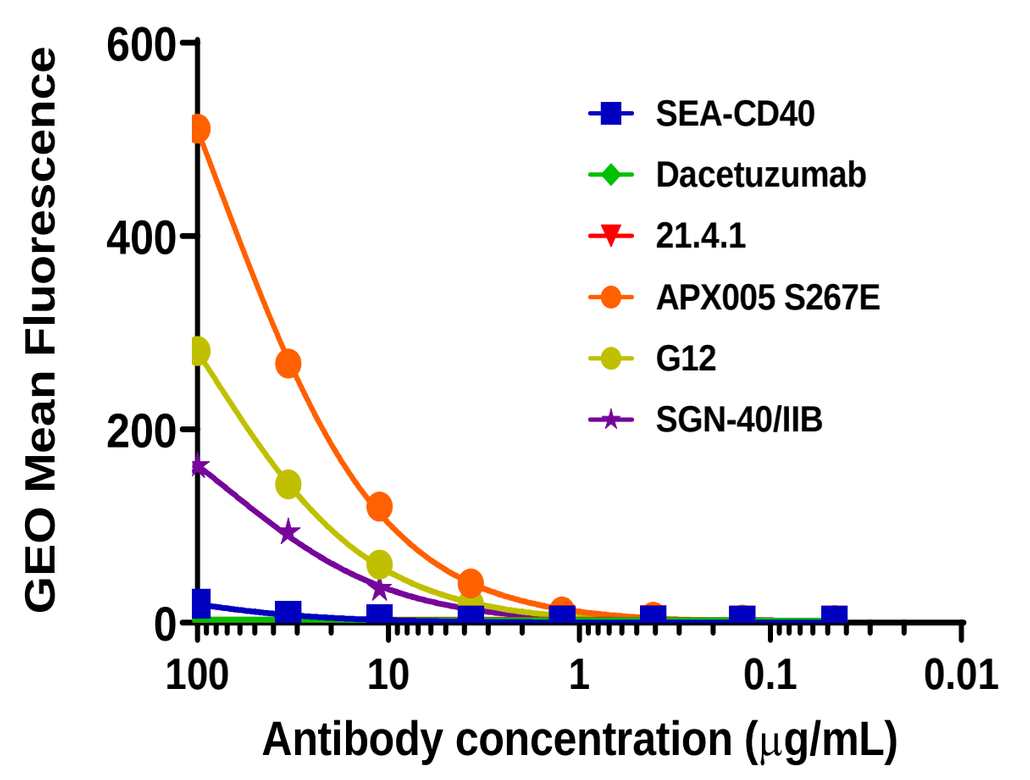
GEO Mean Fluorescence
SEA-CD40
Dacetuzumab
21.4.1
APX005 S267E
G12
SGN-40/IIB
Antibody concentration (μg/mL)

Binding to FcγRIIb

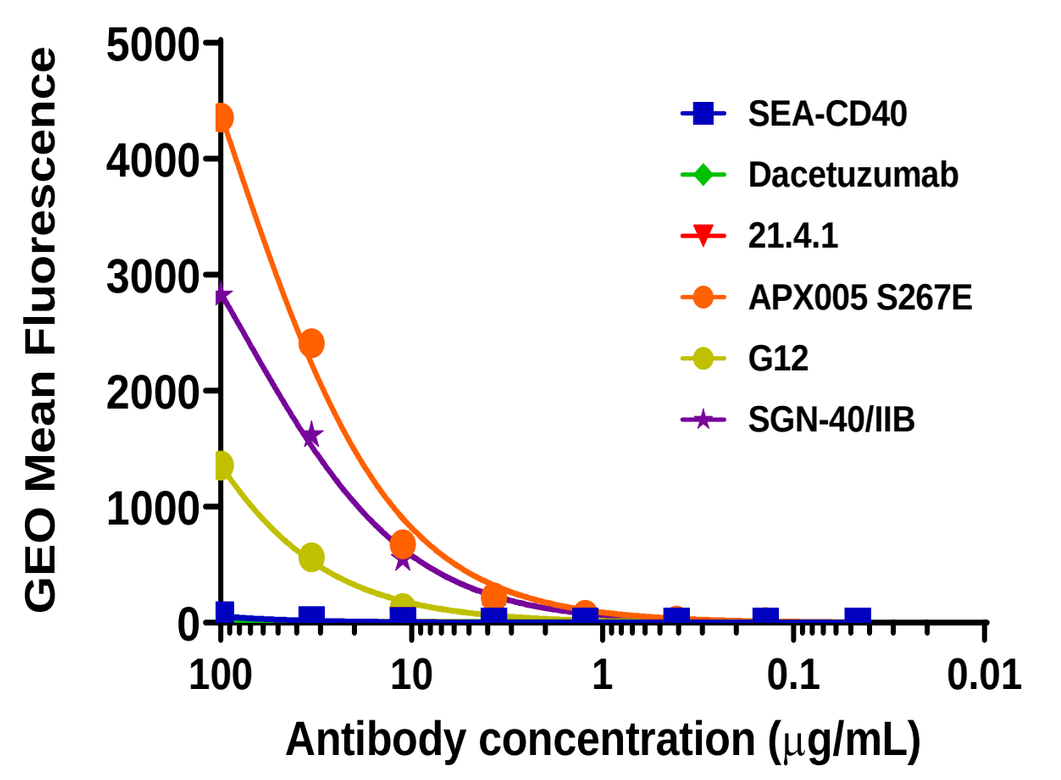
GEO Mean Fluorescence
SEA-CD40
Dacetuzumab
21.4.1
APX005 S267E
G12
SGN-40/IIB
Antibody concentration (μg/mL)

Binding to FcγRIIIa

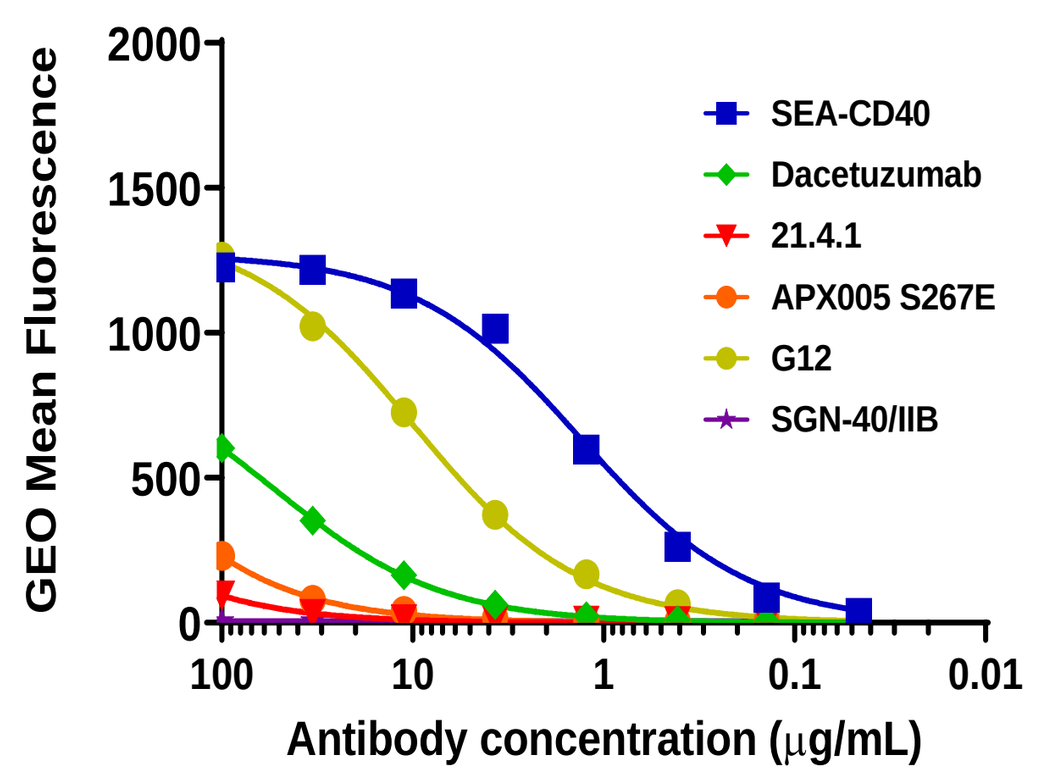
GEO Mean Fluorescence
SEA-CD40
Dacetuzumab
21.4.1
APX005 S267E
G12
SGN-40/IIB
Antibody concentration (μg/mL)

Antibody binding to Fcγ receptors was assessed using flow cytometry of CHO cells transfected with human FcγRIIa, IIb or IIIa. As expected, APX005 S267E bound to FcγRIIa and FcγIIb with the highest affinity, whereas SEA-CD40 had the highest affinity for FcγRIIIa. These data highlight the differential binding of the antibodies to FcγRs and allows for in vitro and in vivo exploration of the consequences of that binding.

SEA-CD40 drives superior immune activation in vitro

EBV Transformed B Cell Killing

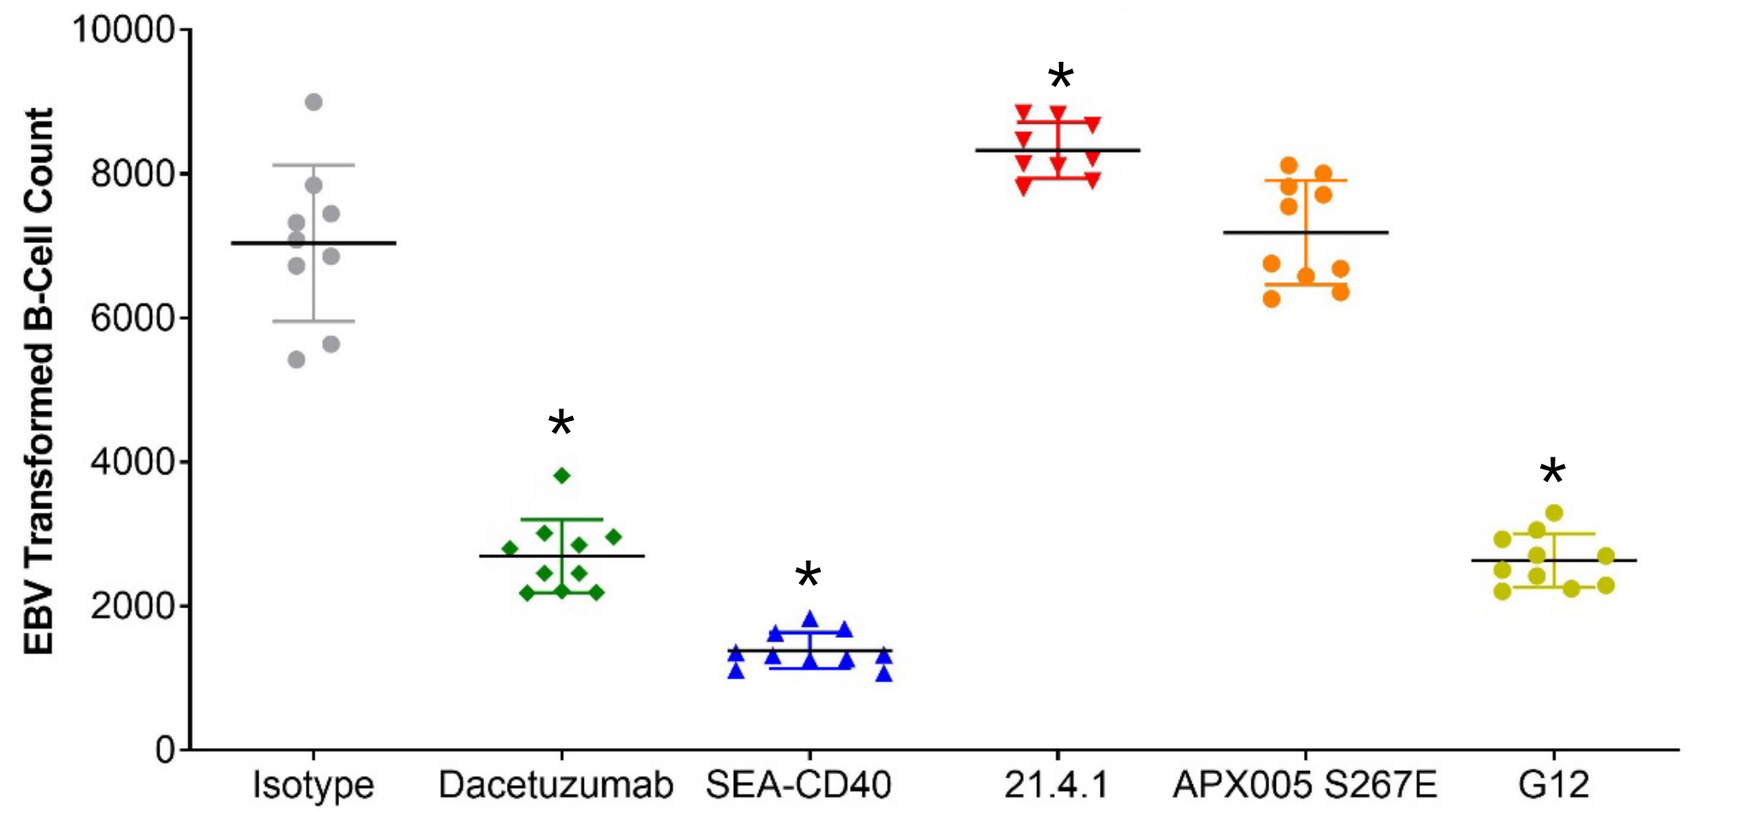
EBV Transformed B-Cell Count
Isotype
Dacetuzumab
SEA-CD40
21.4.1
APX005 S267E
G12

* = P≤0.0006 vs. isotype control, SEA-CD40 significantly different from all groups

CD8 T Cell Numbers

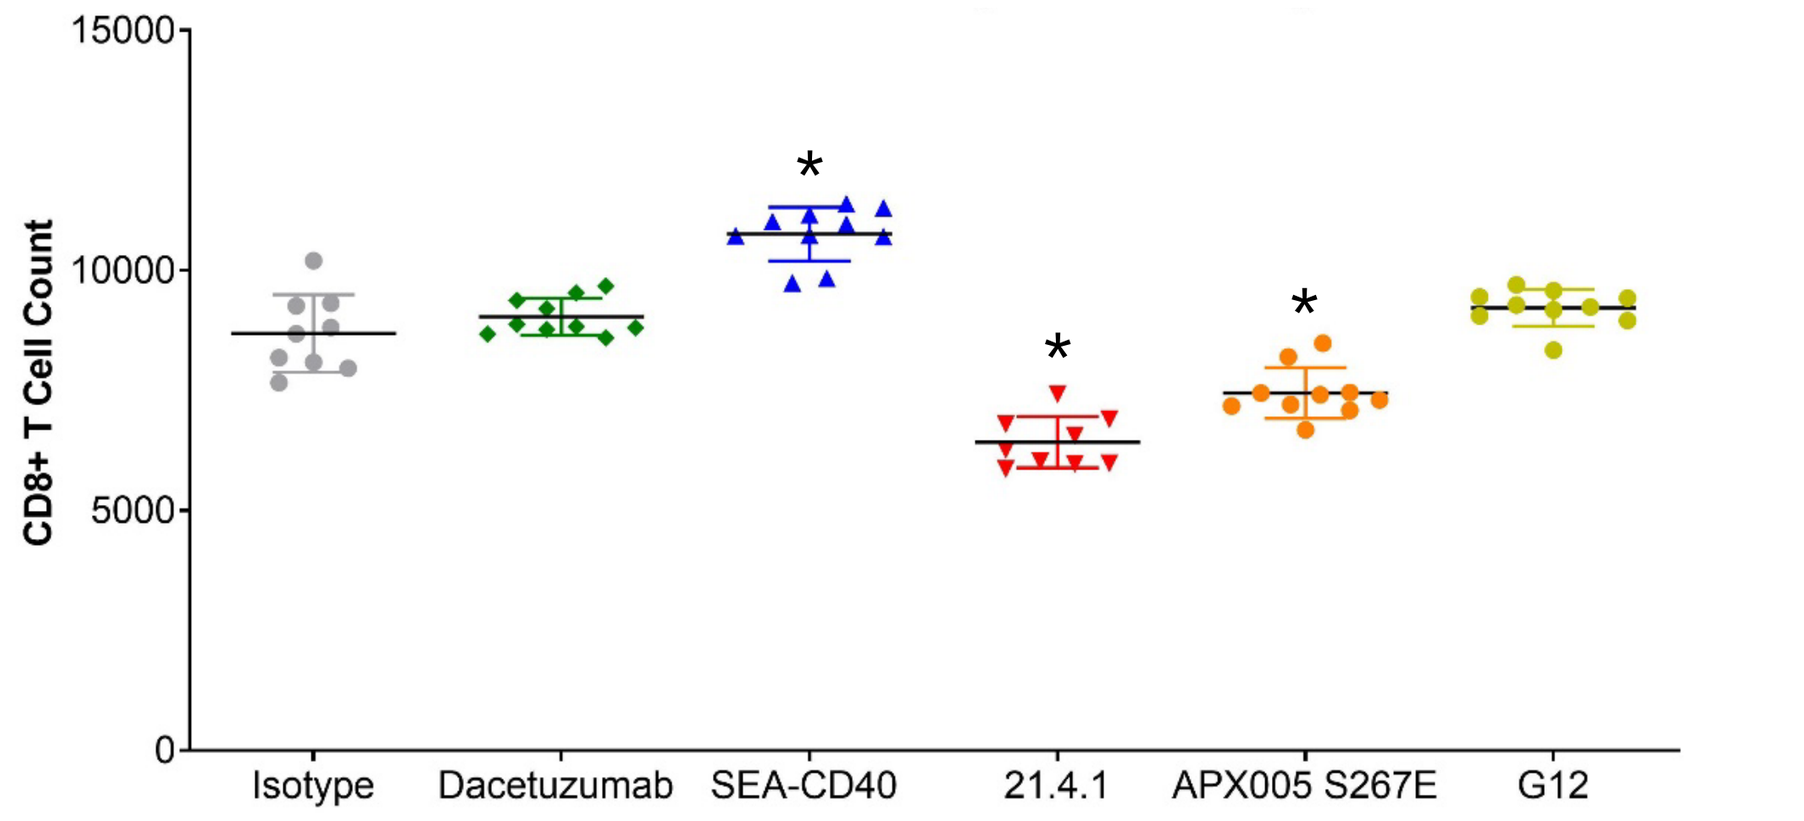
CD8+ T Cell Count
Isotype
Dacetuzumab
SEA-CD40
21.4.1
APX005 S267E
G12

* = P≤0.0001 vs. isotype control, SEA-CD40 significantly different from all groups

Increased SEA-CD40 binding to FcγRIIIa drives superior killing of CD40+ EBV-transformed B cells isolated from healthy human donors. Killing of CD40+ cells by human PBMCs occurred concomitant with promoting increased CD8+ T cell numbers (antibodies used at 1 μg/mL; P values determined by 1-way ANOVA).

MCP-1

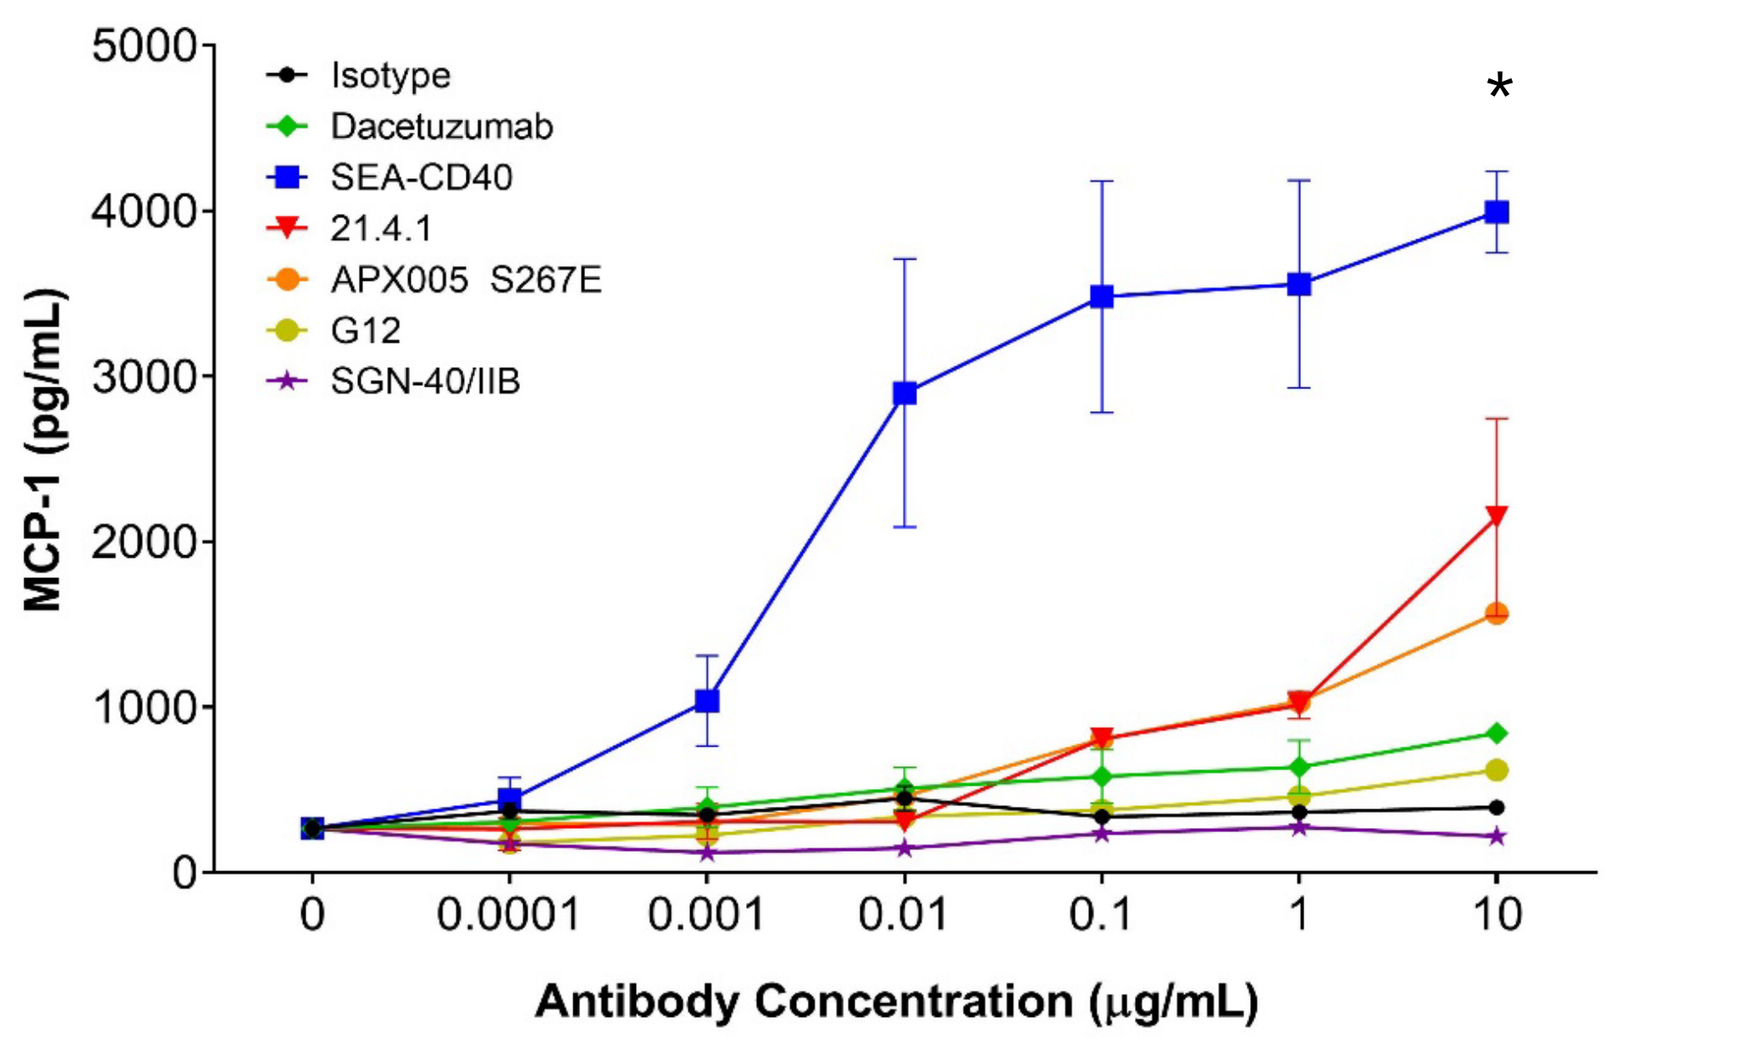
MCP-1 (pg/mL)
Isotype
Dacetuzumab
SEA-CD40
21.4.1
APX005 S267E
G12
SGN-40/IIB
Antibody Concentration (μg/mL)

* = P=0.0002 vs. isotype control, SEA-CD40 significantly different from all groups

IP-10

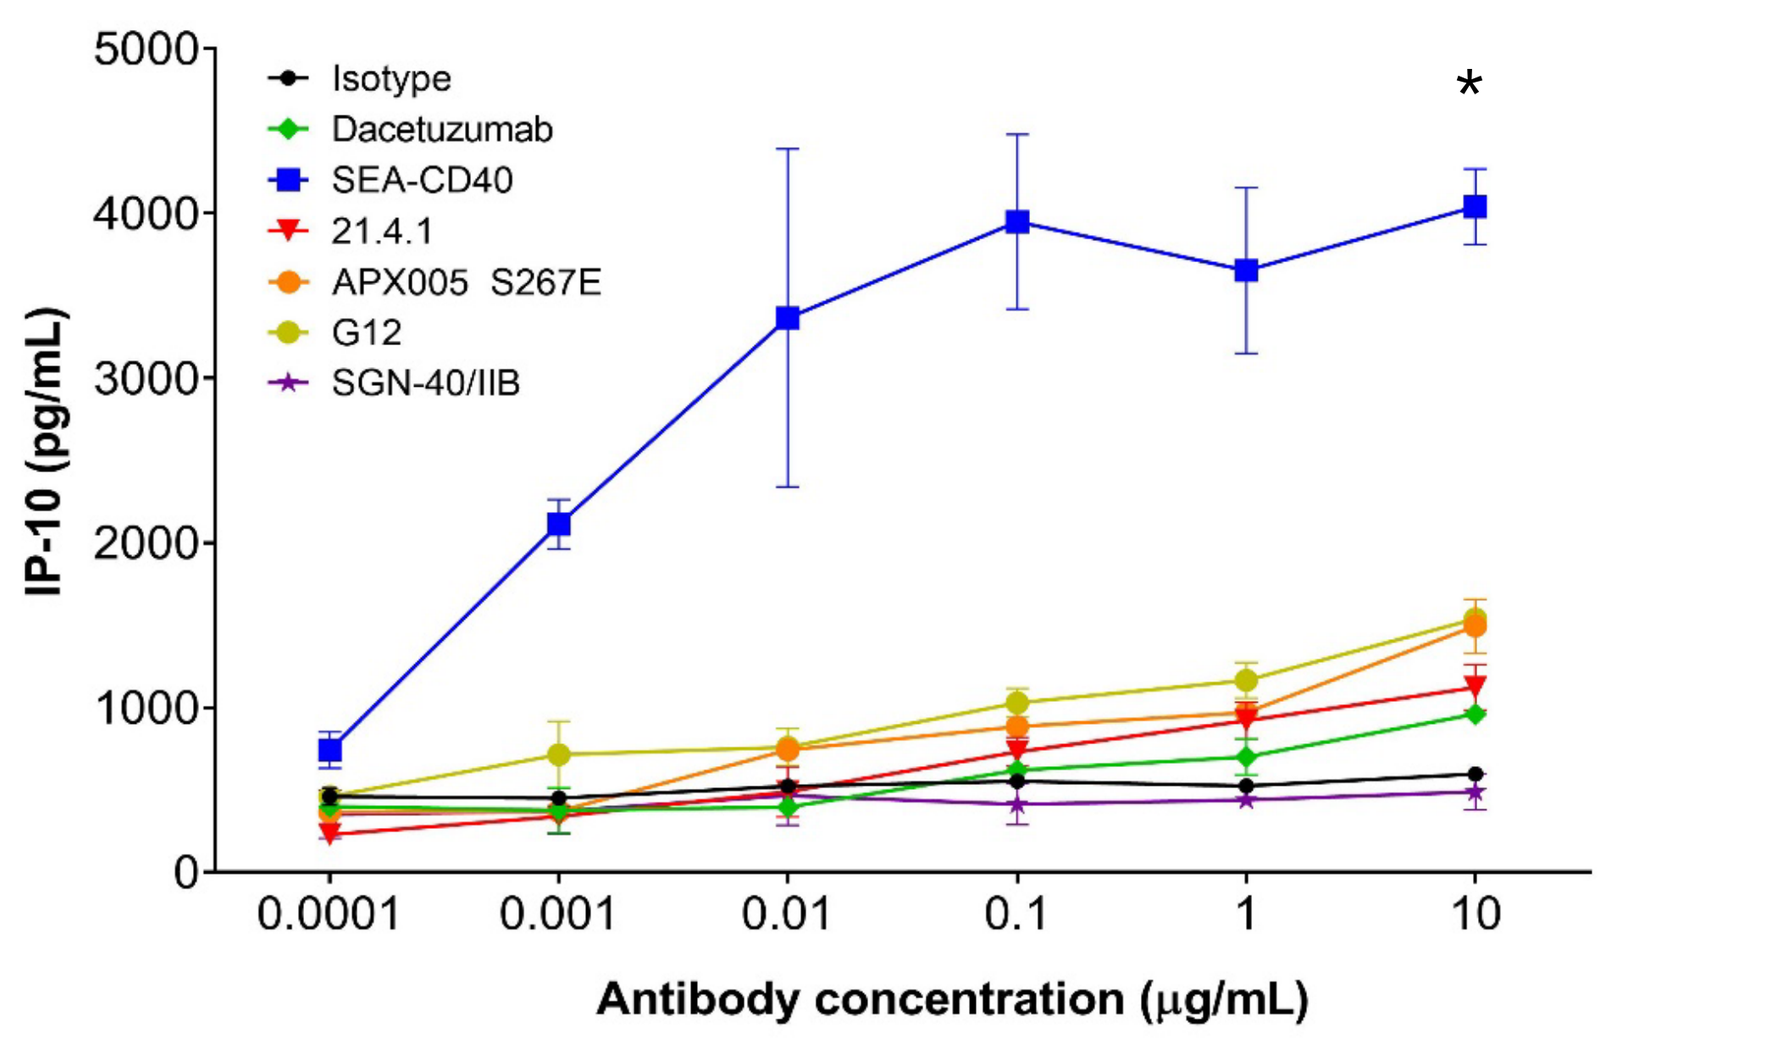
IP-10 (pg/mL)
Isotype
Dacetuzumab
SEA-CD40
21.4.1
APX005 S267E
G12
SGN-40/IIB
Antibody concentration (μg/mL)

* = P≤0.0001 compared to all groups

SEA-CD40 is more potent than other CD40 agonists at driving innate immune cell-mediated cytokine and chemokine production in vitro (P values determined by 1-way ANOVA).

SEA-CD40 drives robust anti-tumor activity alone and in combination with an anti-PD-1 antibody in mouse models

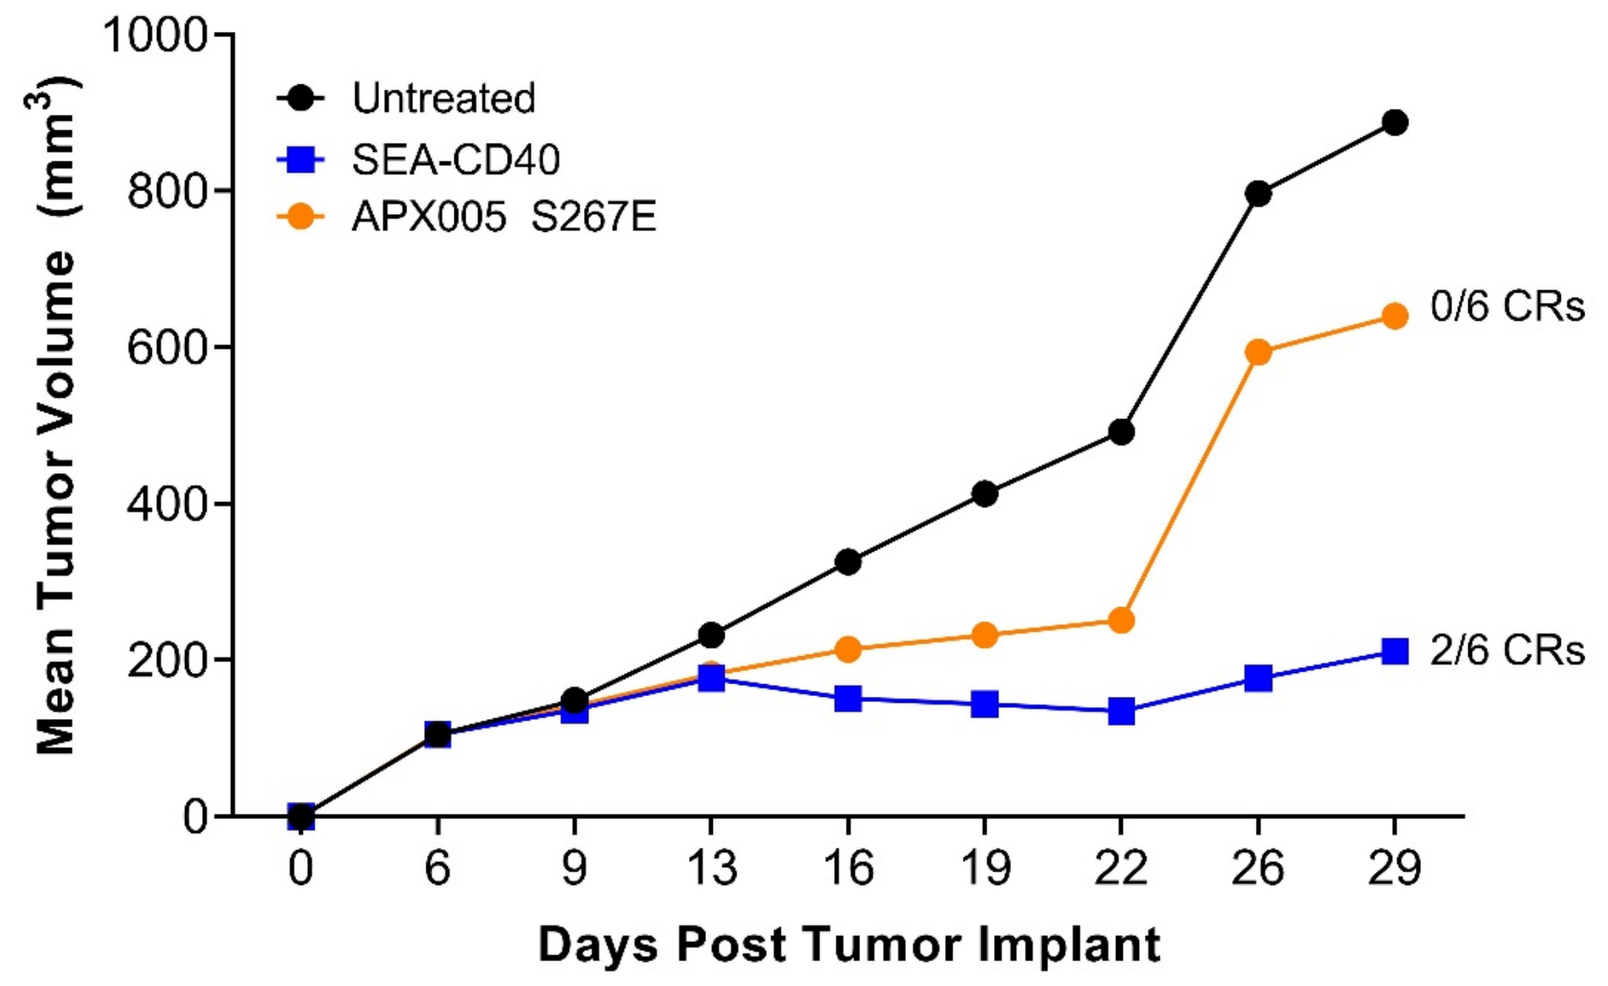
Mean Tumor Volume (mm³)
Untreated
SEA-CD40
APX005 S267E
0/6 CRs
2/6 CRs
Days Post Tumor Implant

SEA-CD40 leads to enhanced tumor reduction compared to APX005 S267E, demonstrating that engagement of specific FcγRs by the antibody backbone may be critical for driving anti-tumor activity. Mice expressing human CD40 were implanted with the syngeneic A20 tumor model, antibodies were dosed at 0.3 mg/kg.

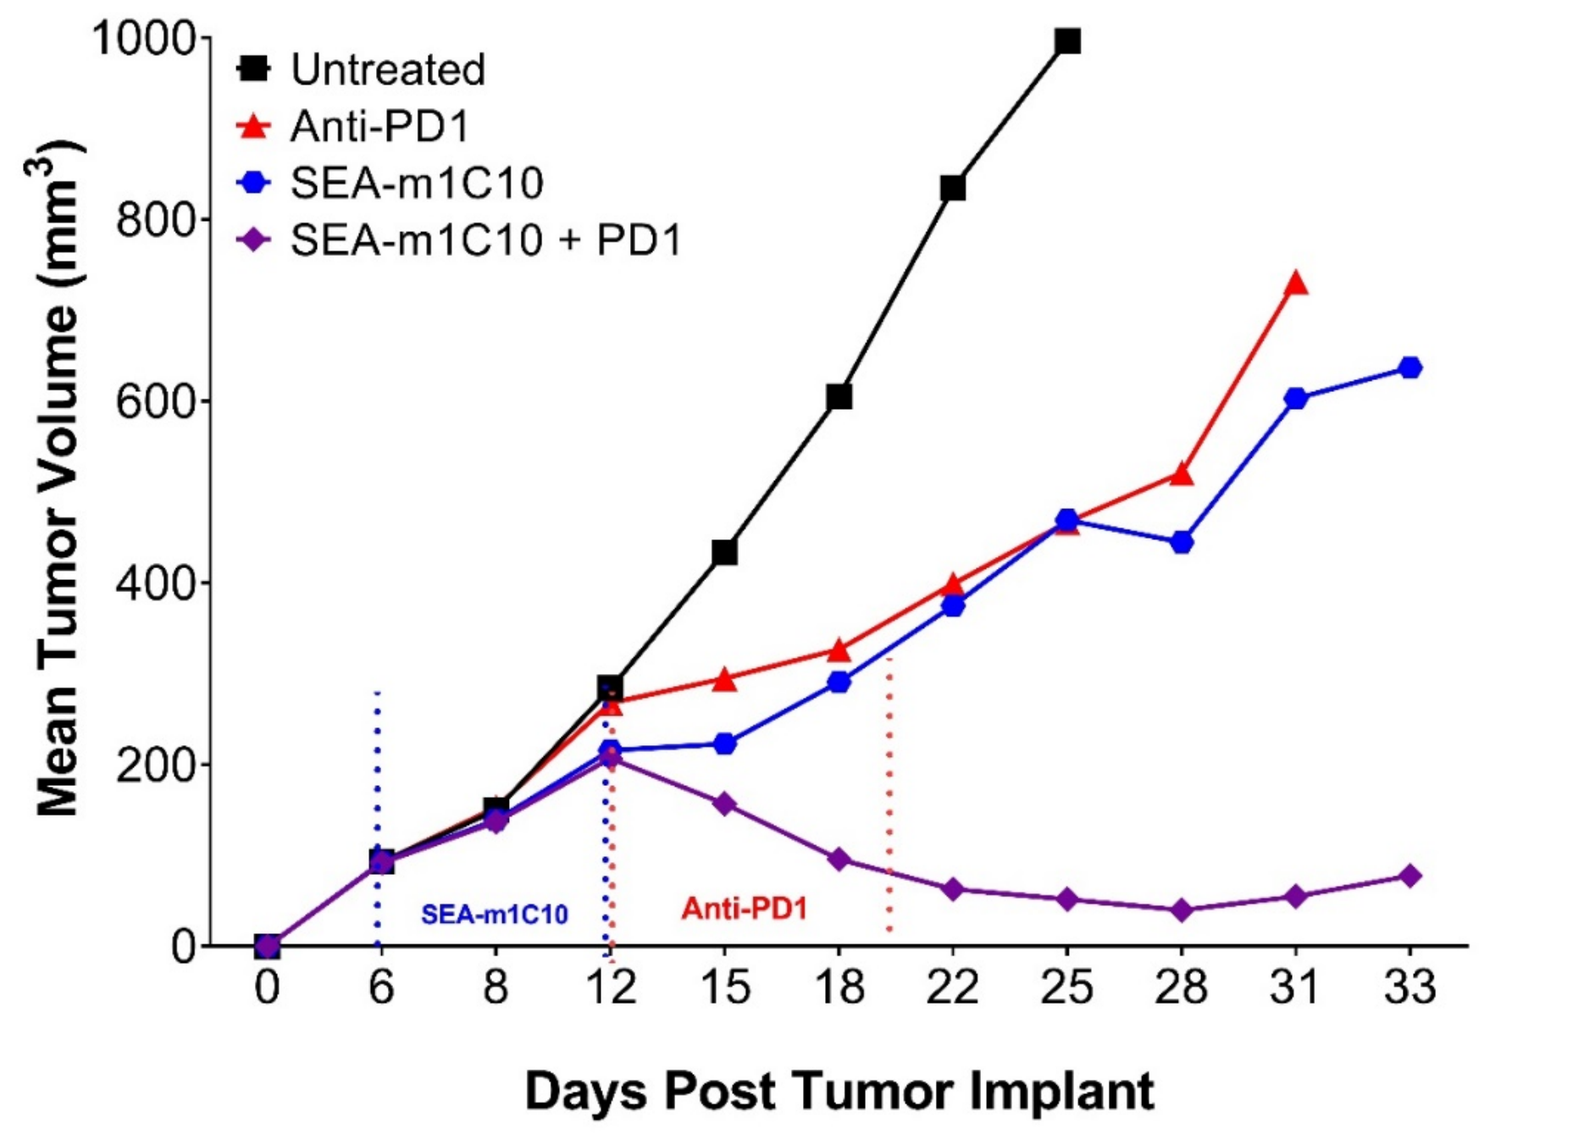
Mean Tumor Volume (mm³)
Untreated
Anti-PD1
SEA-m1C10
SEA-m1C10 + PD1
SEA-m1C10
Anti-PD1
Days Post Tumor Implant

A20 tumor-bearing mice treated with sub-therapeutic doses of SEA-m1C10 (murine analog of SEA-CD40) or the anti-PD1 surrogate antibody exhibit minimal tumor growth delay (antibodies dosed at 1 mg/kg). However, when given together, these two agents synergize to drive increased anti-tumor activity and complete regressions.

SEA-CD40 treatment elicits consistent immune changes across species

A. Cytokine production in patients

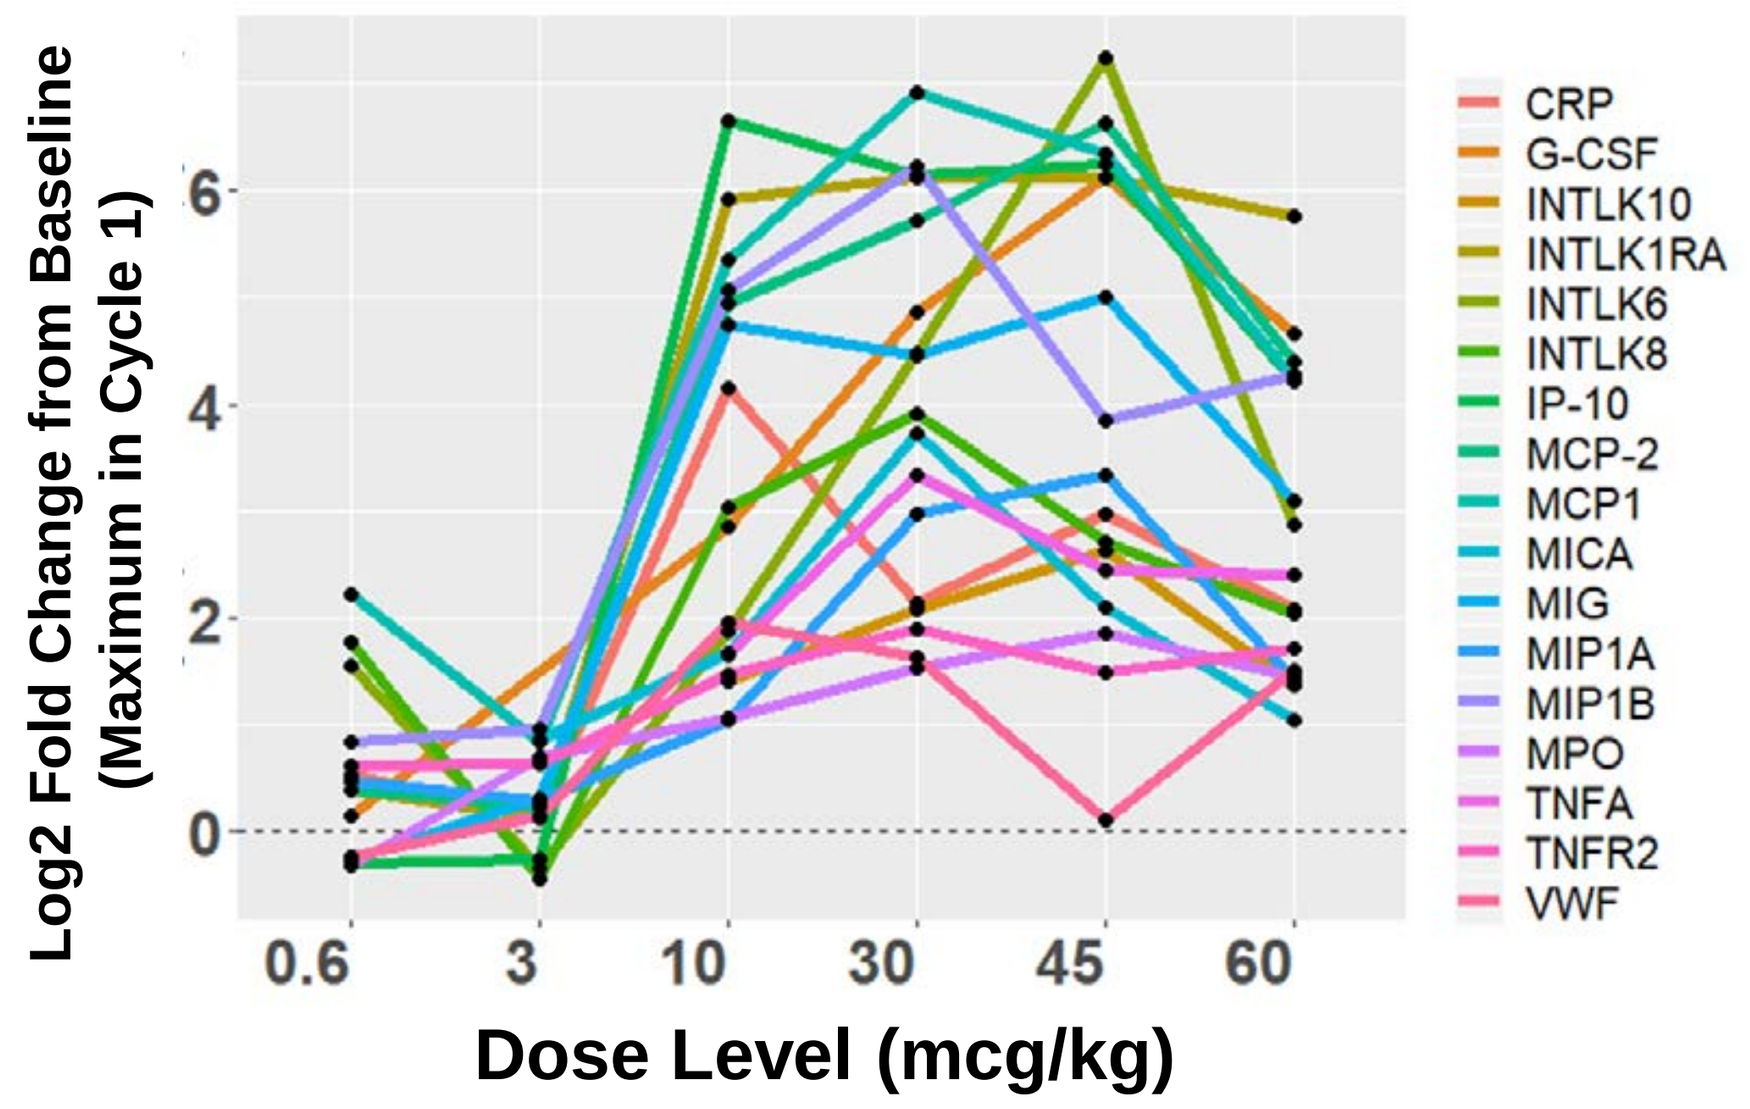
Log2 Fold Change from Baseline (Maximum in Cycle 1)
Dose Level (mcg/kg)
CRP
G-CSF
INTLK10
INTLK1RA
INTLK6
INTLK8
IP-10
MCP-2
MCP1
MICA
MIG
MIP1A
MIP1B
MPO
TNFA
TNFR2
VWF

B. MCP-1 levels across species

Patients

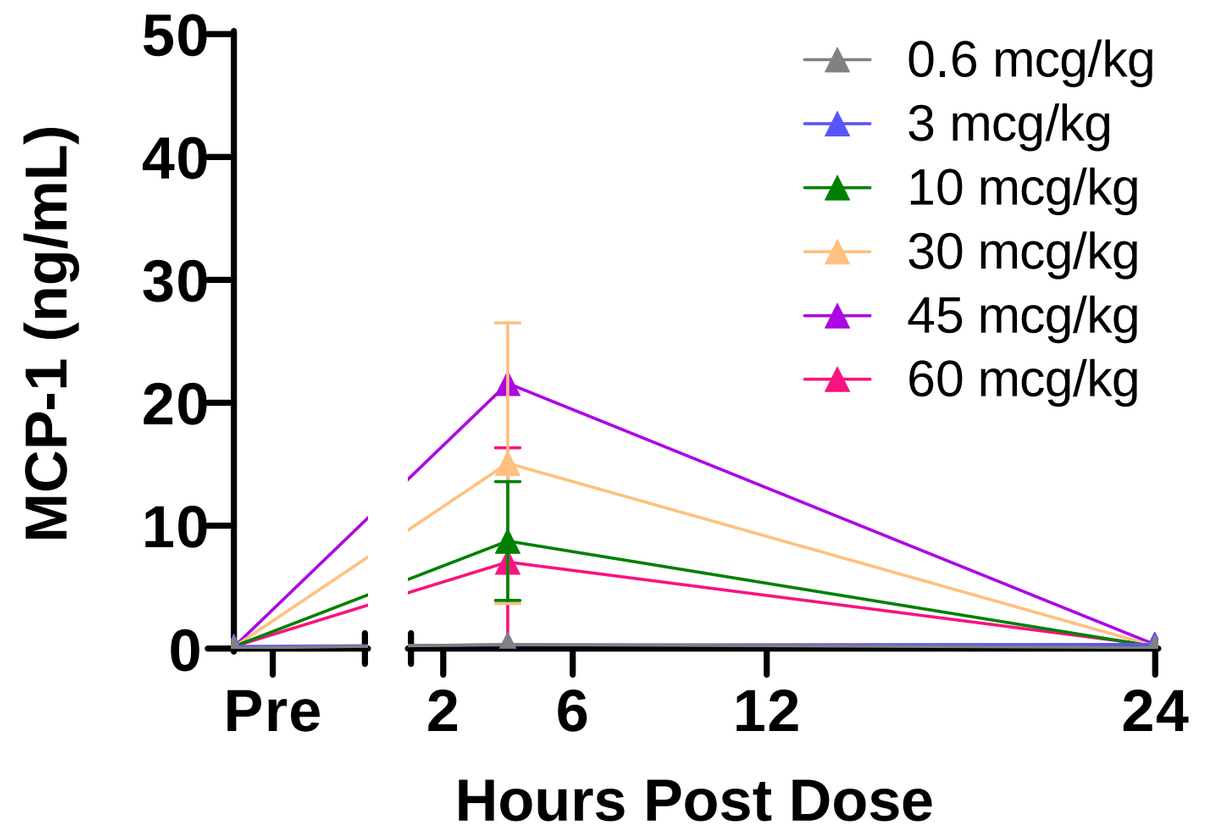
MCP-1 (ng/mL)
0.6 mcg/kg
3 mcg/kg
10 mcg/kg
30 mcg/kg
45 mcg/kg
60 mcg/kg
Hours Post Dose

Cynomolgus Monkeys

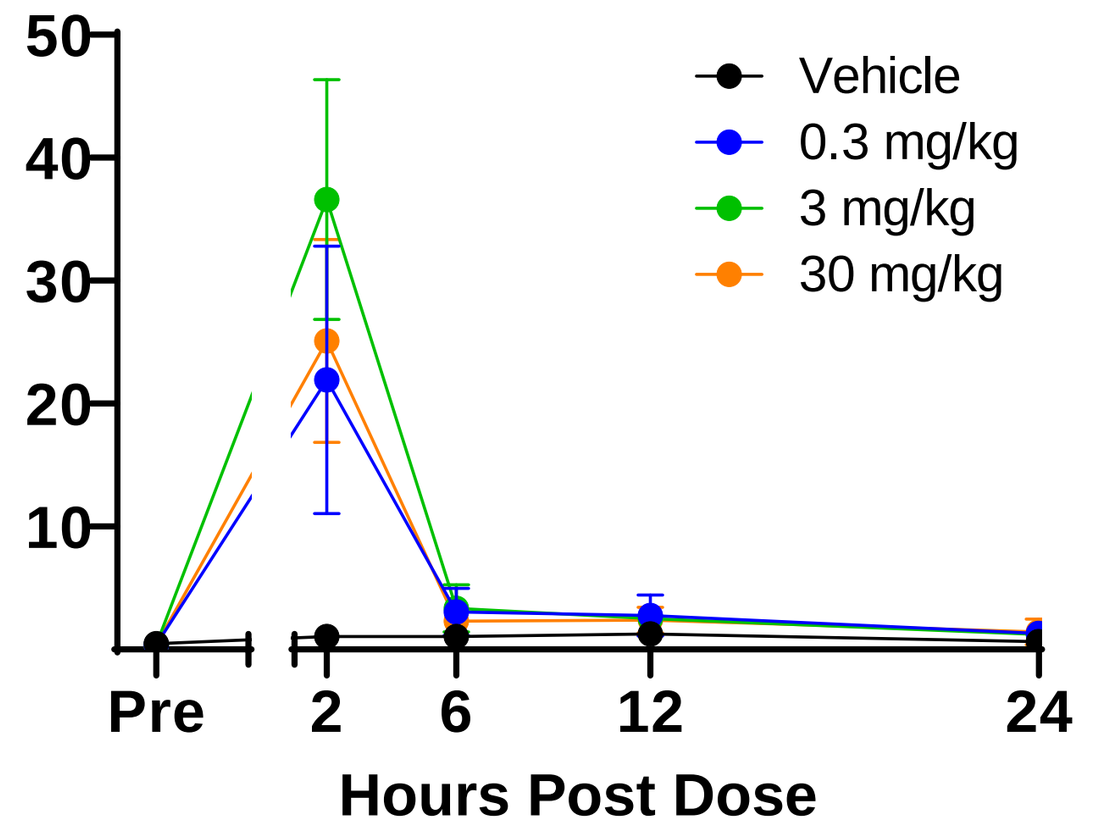
Vehicle
0.3 mg/kg
3 mg/kg
30 mg/kg
Hours Post Dose

C. B cell depletion across species

Patients

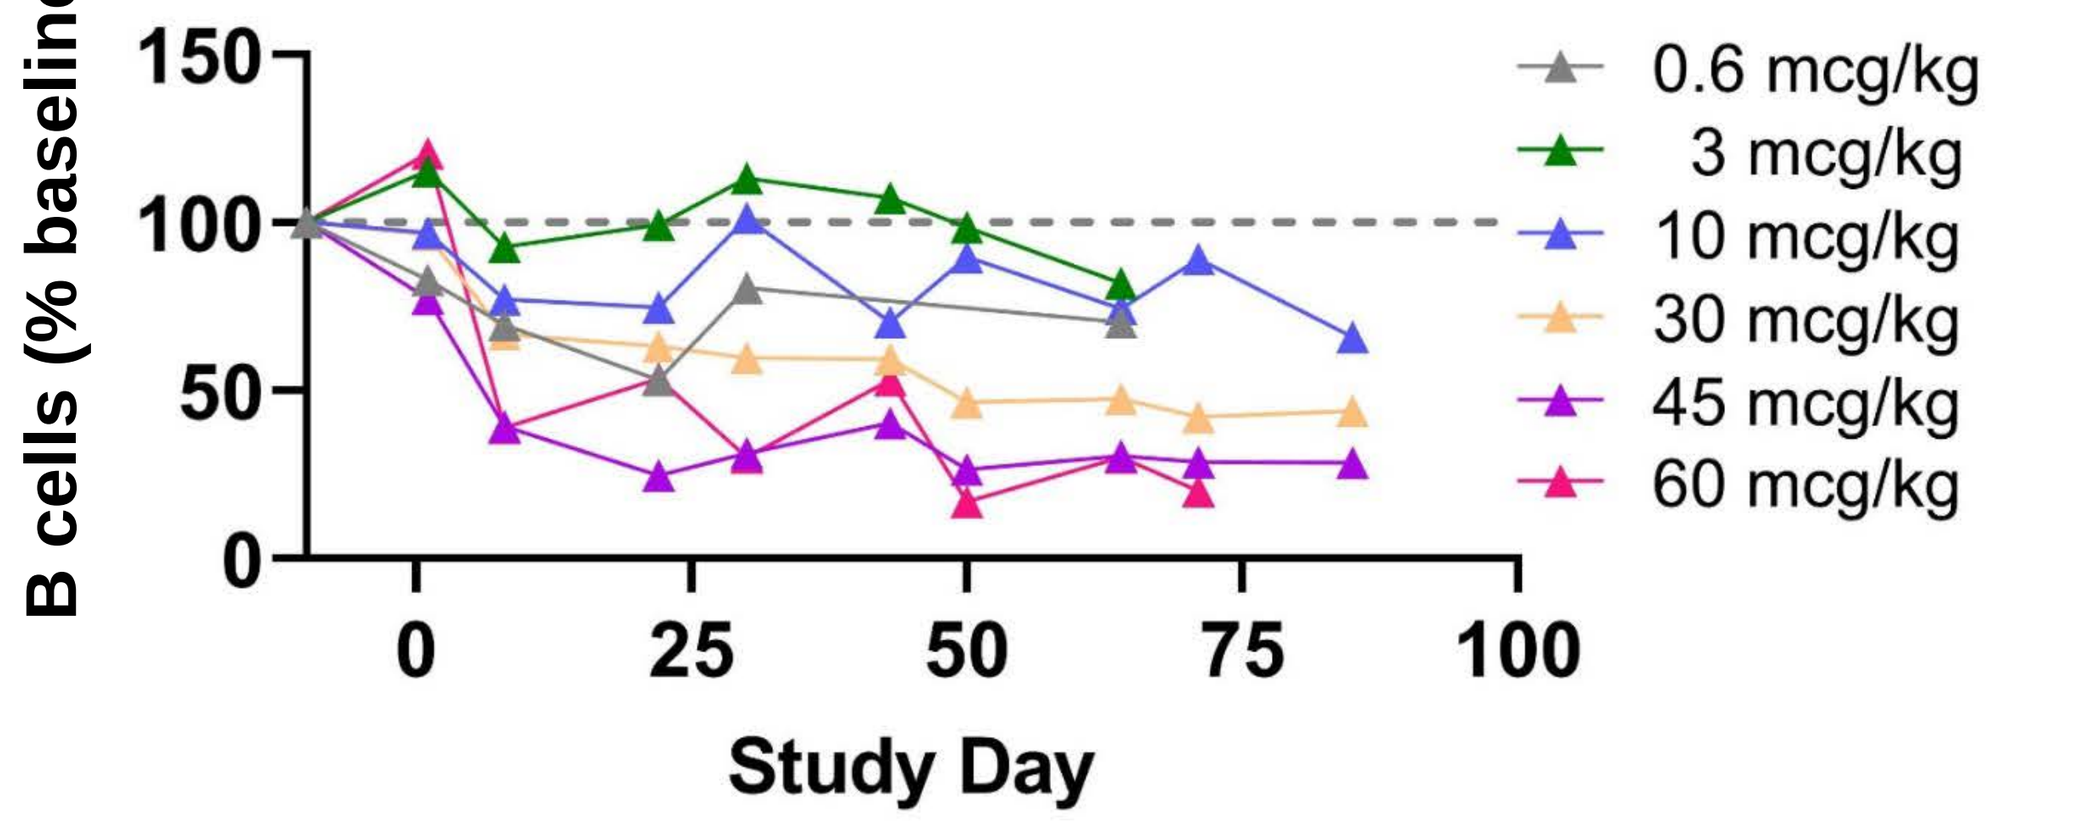
B cells (% baseline)
0.6 mcg/kg
3 mcg/kg
10 mcg/kg
30 mcg/kg
45 mcg/kg
60 mcg/kg
Study Day

Cynomolgus Monkeys

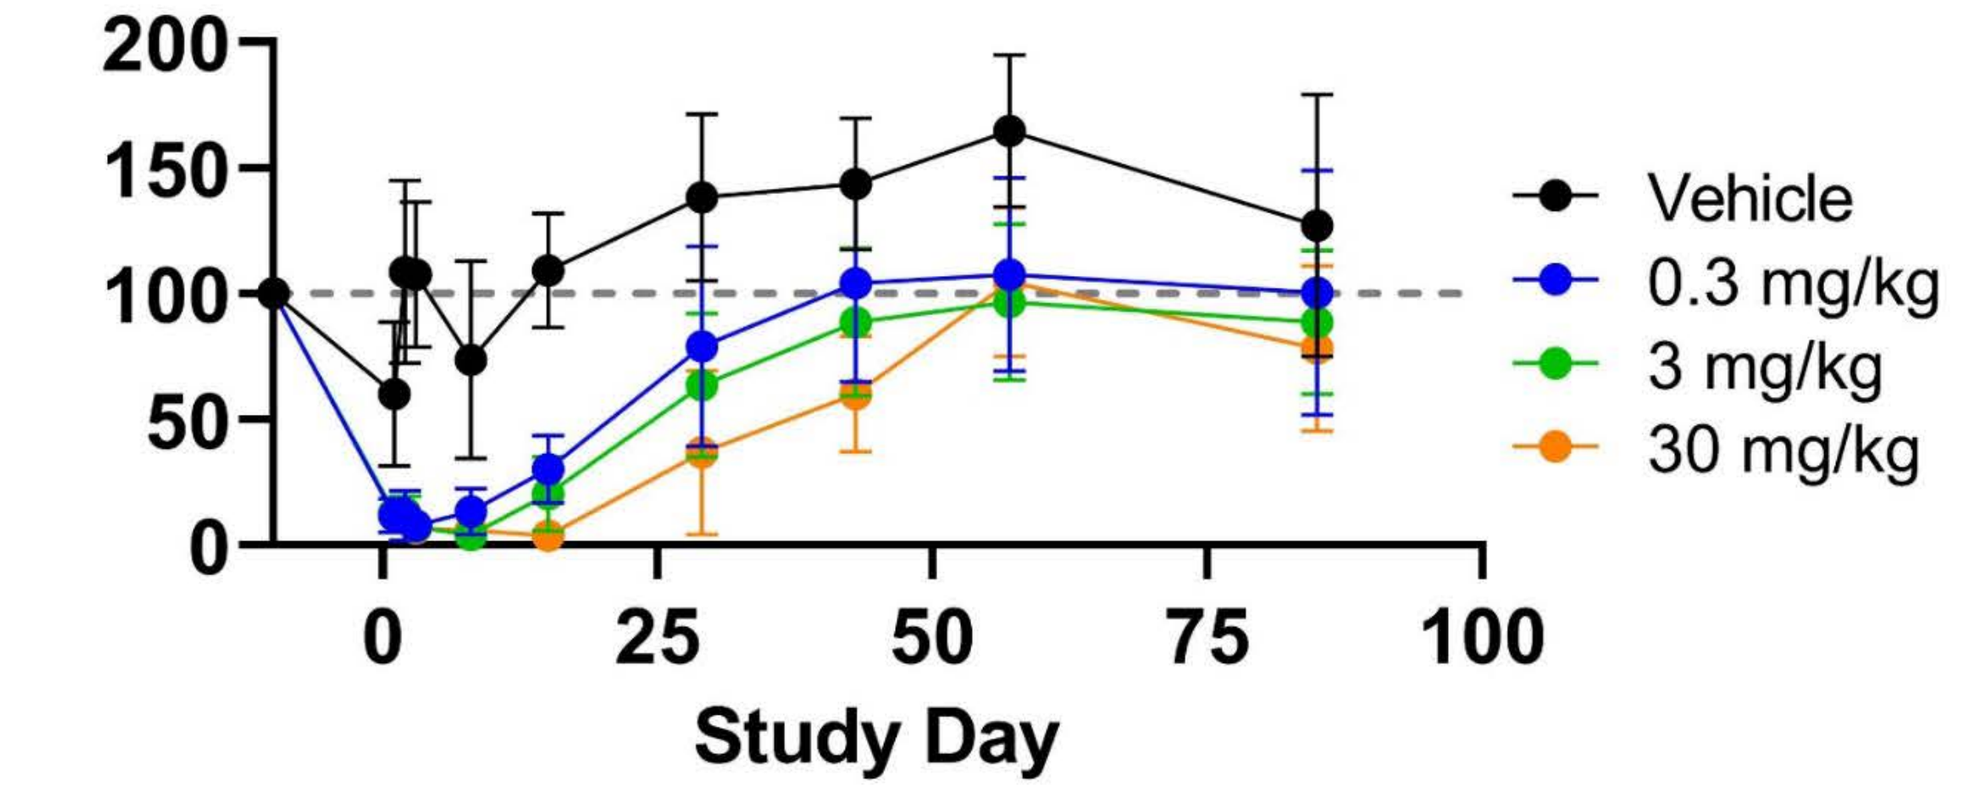
Vehicle
0.3 mg/kg
3 mg/kg
30 mg/kg
Study Day

Across species, SEA-CD40 causes dose-dependent increases in cytokines/chemokines associated with both innate and adaptive immune cell activation (panels **A and B**) as well as CD40+ B cell depletion (**C**). Depletion of CD40+ macrophages was not observed. Similar trends were observed in mice expressing human CD40 (data not shown). *Clinical data from Phase 1 trial (NCT02376699) in patients with advanced malignancies.*

SEA-CD40 drives T cell activation and APC, NK, and T cell trafficking in patients

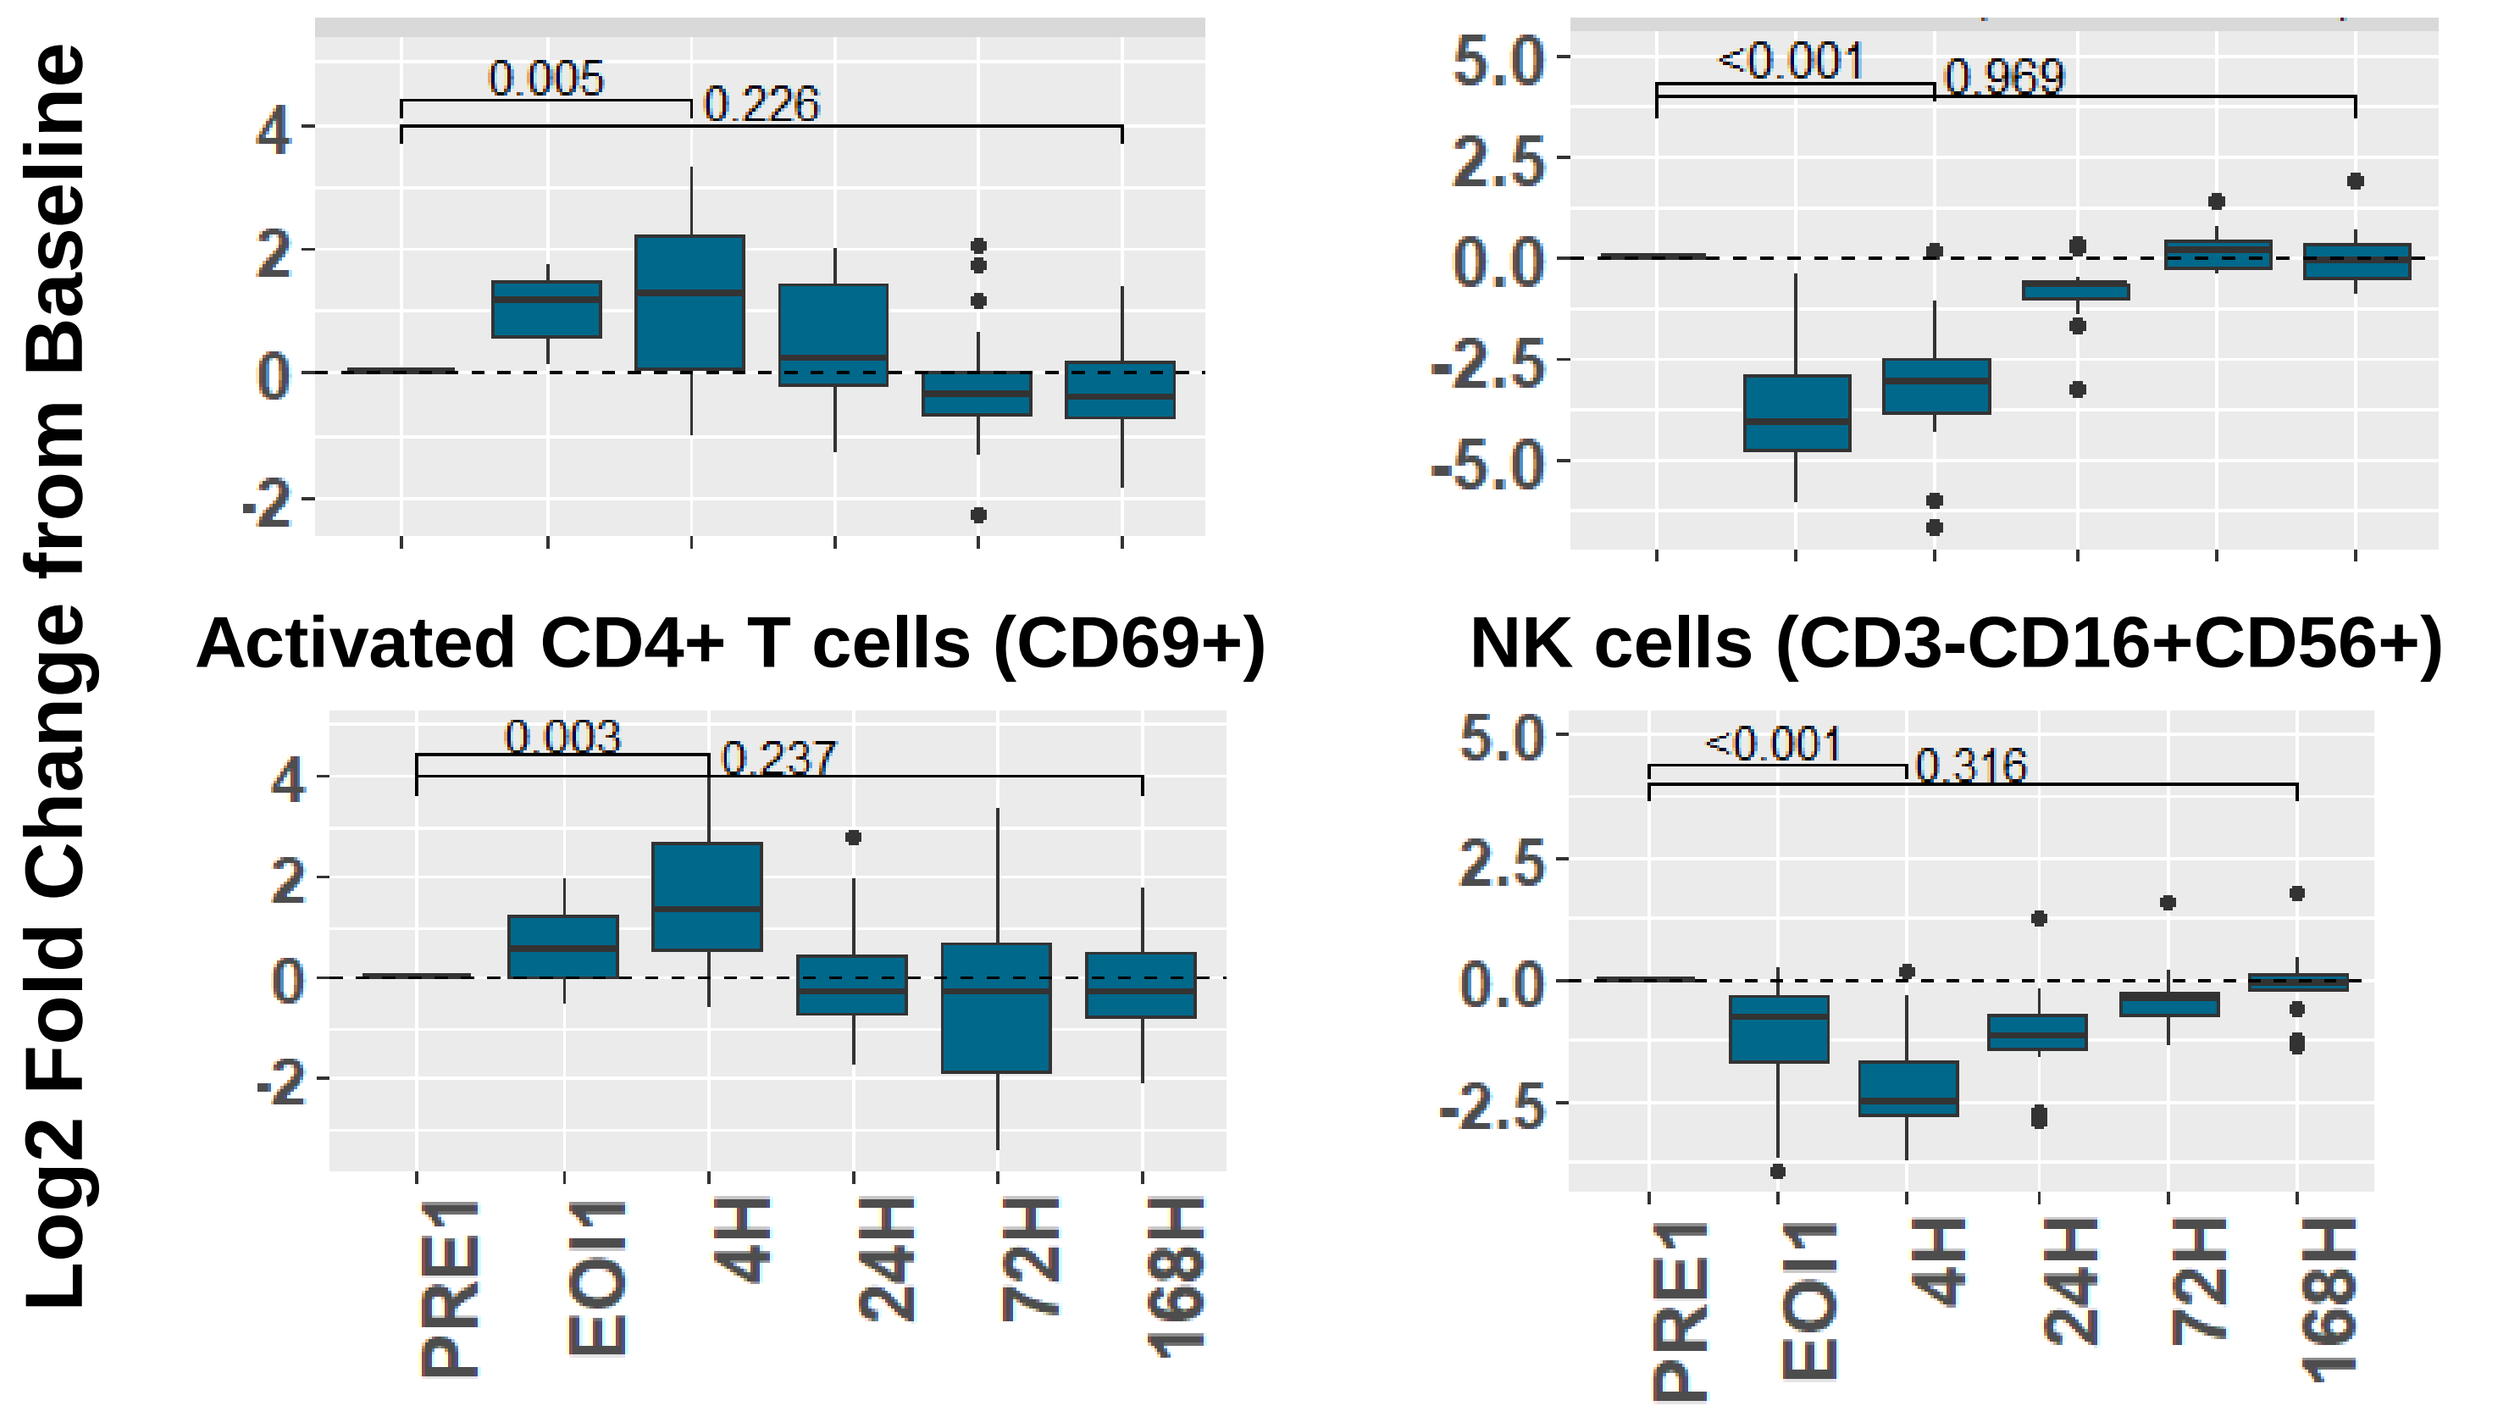
Activated CD8+ T cells (CD69+)
Monocytes (CD3-CD14+)
Activated CD4+ T cells (CD69+)
NK cells (CD3-CD16+CD56+)
Log2 Fold Change from Baseline
0.005
0.226
<0.001
0.969
0.003
0.237
<0.001
0.316
PRE1
EOI1
4H
24H
72H
168H

T cells

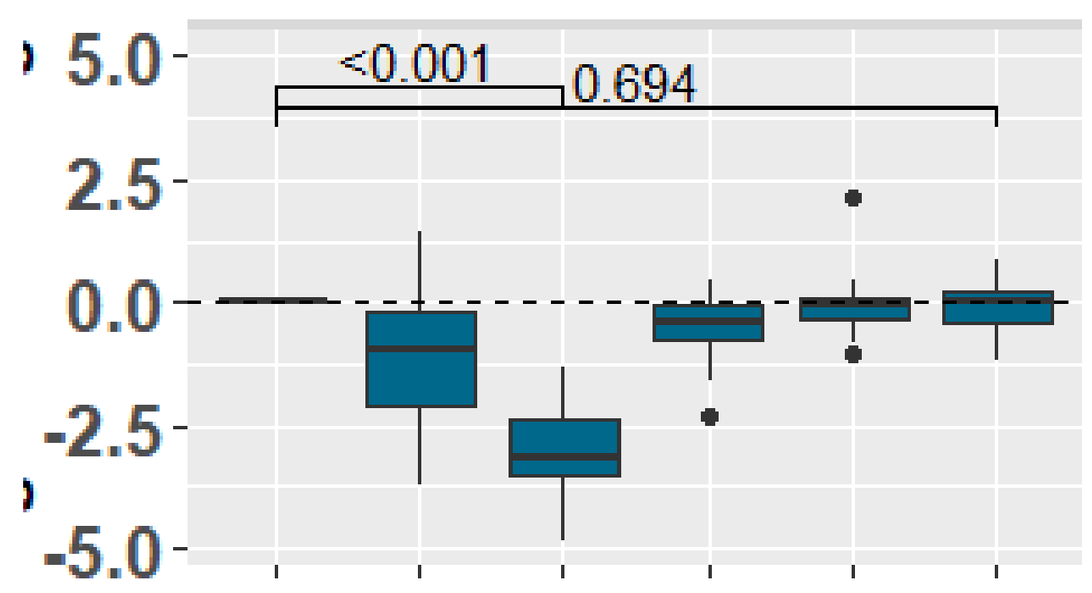
<0.001
0.694

Patients were treated with 30 mcg/kg SEA-CD40 once every 3 weeks. Circulating immune cells were assessed by flow cytometry. Cycle 1 data shown, P values determined by paired t-test. *Clinical data from Phase 1 trial (NCT02376699) in patients with advanced malignancies.*

Conclusions

- SEA-CD40 has increased in vitro and in vivo activity compared to other CD40-targeted mAbs suggesting that enhanced effector function and FcγRIIIa binding enables optimal immune cell agonism in contrast to other backbone modifications.
- SEA-CD40 induces an immune activation signature that translates from preclinical to clinical sample sets and is associated with single agent and anti-PD-1 combinatorial anti-tumor activity in preclinical models.
- An ongoing phase 1 clinical trial (NCT02376699) is actively enrolling in the United States and includes a new cohort assessing the combination of SEA-CD40, gemcitabine, nab-paclitaxel and pembrolizumab in the frontline treatment of patients with metastatic pancreatic ductal adenocarcinoma.

Abstract No. 7012, Poster #5535
AACR Virtual Annual Meeting II; June 22 – June 24, 2020

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.
Contact: Haley Neff-LaFord (hnlaford@seagen.com)

SeattleGenetics