Abstract LB246

American Association for Cancer Research Annual Meeting
April 14–19, 2023; Orlando, FL, USA

ENFORTUMAB VEDOTIN, A NECTIN-4-DIRECTED ANTIBODY-DRUG CONJUGATE, DEMONSTRATES COMPELLING PRECLINICAL ANTITUMOR ACTIVITY IN NON-MUSCLE-INVASIVE BLADDER CANCER MODELS, ACCURATELY PREDICTING MINIMAL SYSTEMIC EXPOSURE WHEN ADMINISTERED BY INTRAVESICAL INSTILLATION IN PATIENTS

Devra Olson[1], Yen Lin Chia[1], Enahoro Iboi[1], Lauren Farr[1], Maddy Burcher[1], Anthony Lee[1], Kelly Hensley[1], Sean Allred[1], Abbie Wong[1], Masashi Shimazaki[2], Masamichi Mori[3], Sharsti Sandall[1], Christopher Carosino

[1]Seagen Inc., Bothell, WA, USA; [2]Astellas Research Institute of America LLC, Northbrook, IL, USA; [3]Astellas Pharma, Inc., Japan

Background

- Enfortumab vedotin (EV) is a monomethyl auristatin E (MMAE)-containing antibody-drug conjugate directed to Nectin-4, which is highly expressed in bladder cancers.
- EV improves survival in adults with previously treated locally advanced or metastatic urothelial carcinoma (la/mUC) and is approved in the US, Europe, Japan, and others[1].
- Most newly diagnosed bladder cancer cases are non-muscle invasive (NMIBC)[2-5]. Standard treatment of high-risk NMIBC involves transurethral resection followed by intravesical Bacillus Calmette-Guerin (BCG) or chemotherapy.
- While response to BCG is high, recurrence is common[6], and treatment options for patients with BCG-unresponsive tumors are limited.
- Previously, we demonstrated compelling preclinical antitumor activity of EV in NMIBC models with a favorable safety profile and minimal systemic exposure[7].
- Patients with high-risk BCG-unresponsive NMIBC are currently enrolled in a Phase I study that evaluates the safety, tolerability, and antitumor activity of intravesical EV (EV-104, NCT05014139)[8].
- Initial dose level for EV-104 was selected to be active and with minimal systemic exposure. Here, we present confirmatory data that EV and unconjugated MMAE are undetectable in the bloodstream at the clinical intravesical starting dose (125 mg).

Mouse orthotopic tumors effectively model NMIBC

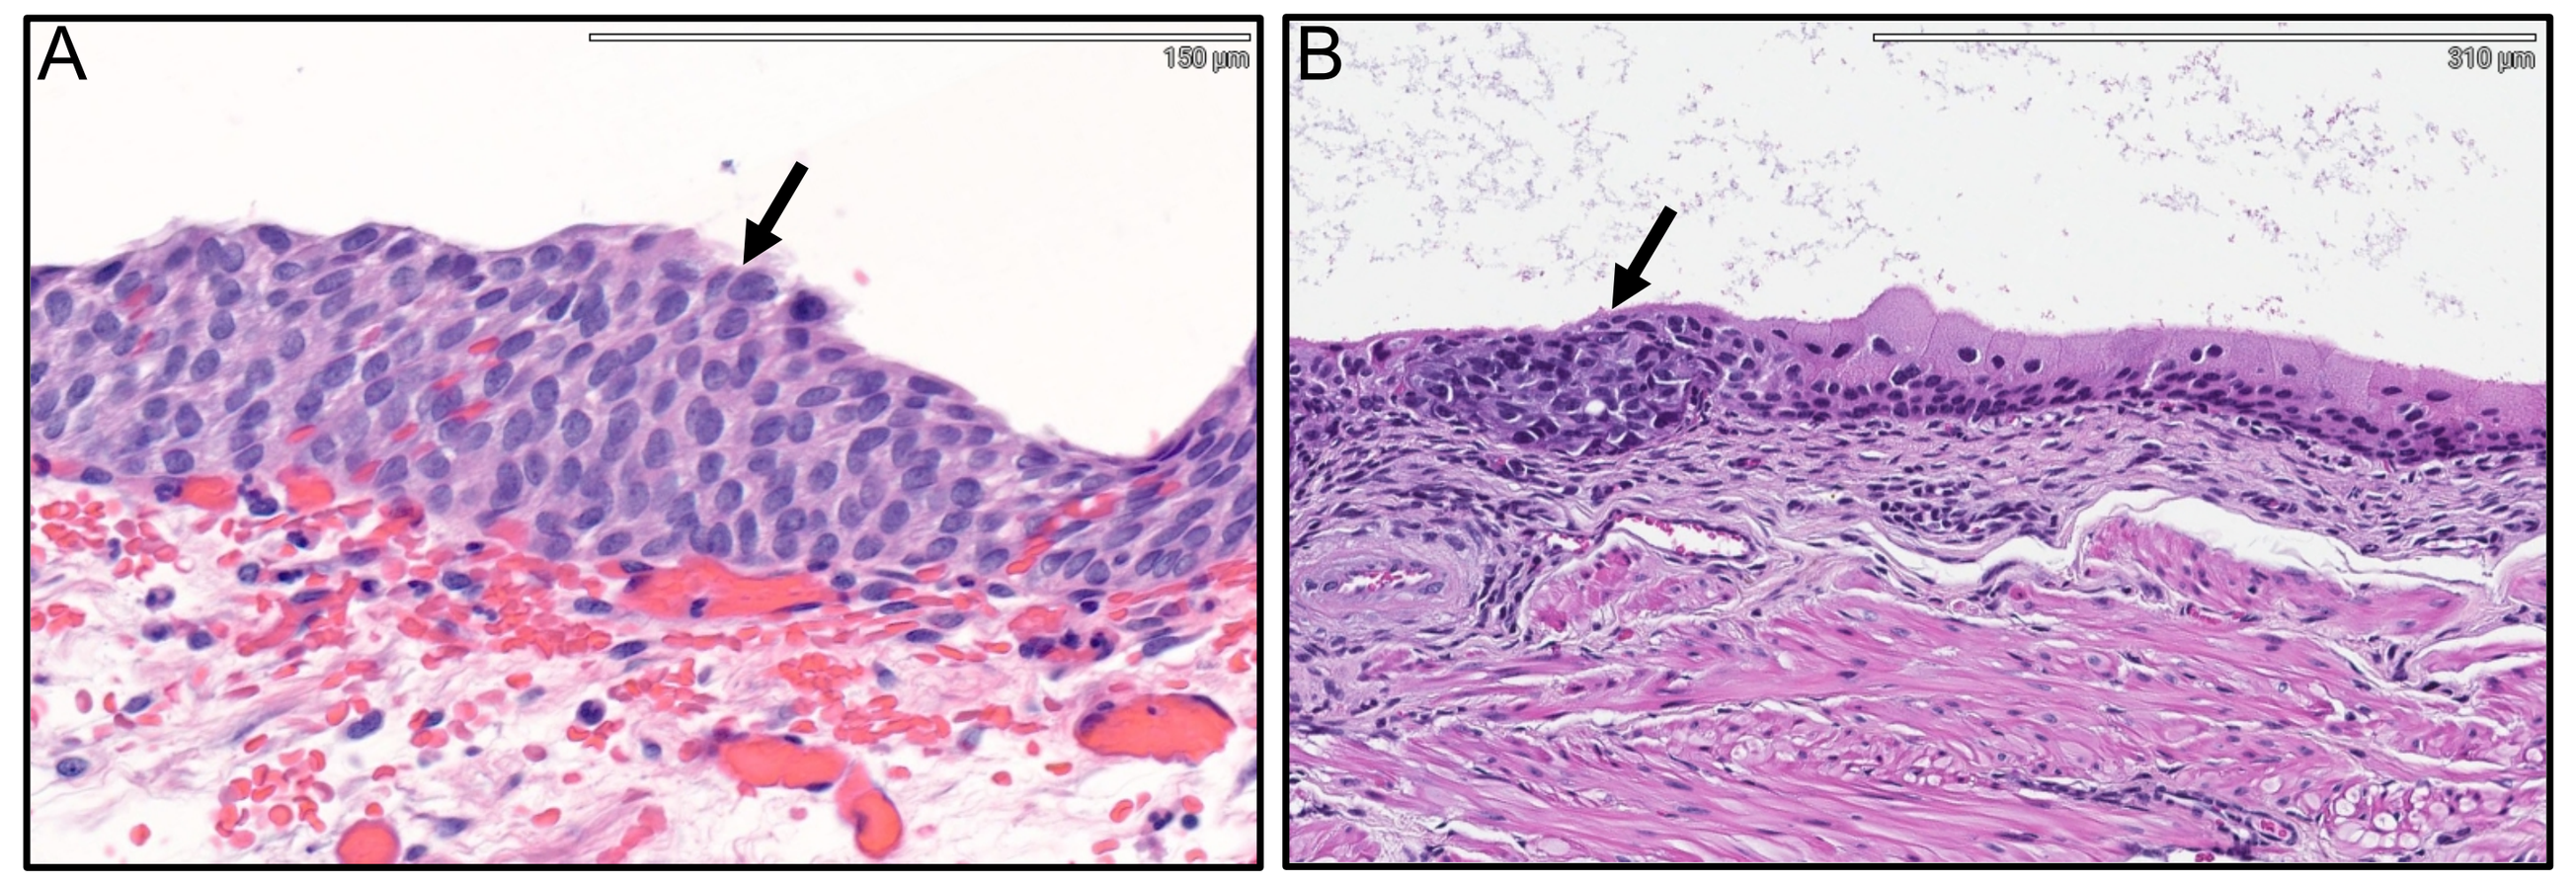
A
150 µm
B
310 µm

Figure 1. H&E staining of urothelial carcinoma in situ from a 68-year-old male (A) and mouse orthotopic xenograft bladder cancer (B) tissue demonstrating that the model effectively mimics NMIBC, with the induction of primarily epithelial tumors and, to a lesser degree, invasion into the submucosa (black arrows, 20x magnification). To generate a mouse model of NMIBC, SCID mice were orthotopically implanted with human UM-UC-3 bladder cancer cells transduced with hNectin-4 and luciferase genes (UM-UC-3-hNectin4+-Luc+) following chemical abrasion to the urothelium.

EV colocalizes with human Nectin-4-positive tumor cells after intravesical dosing

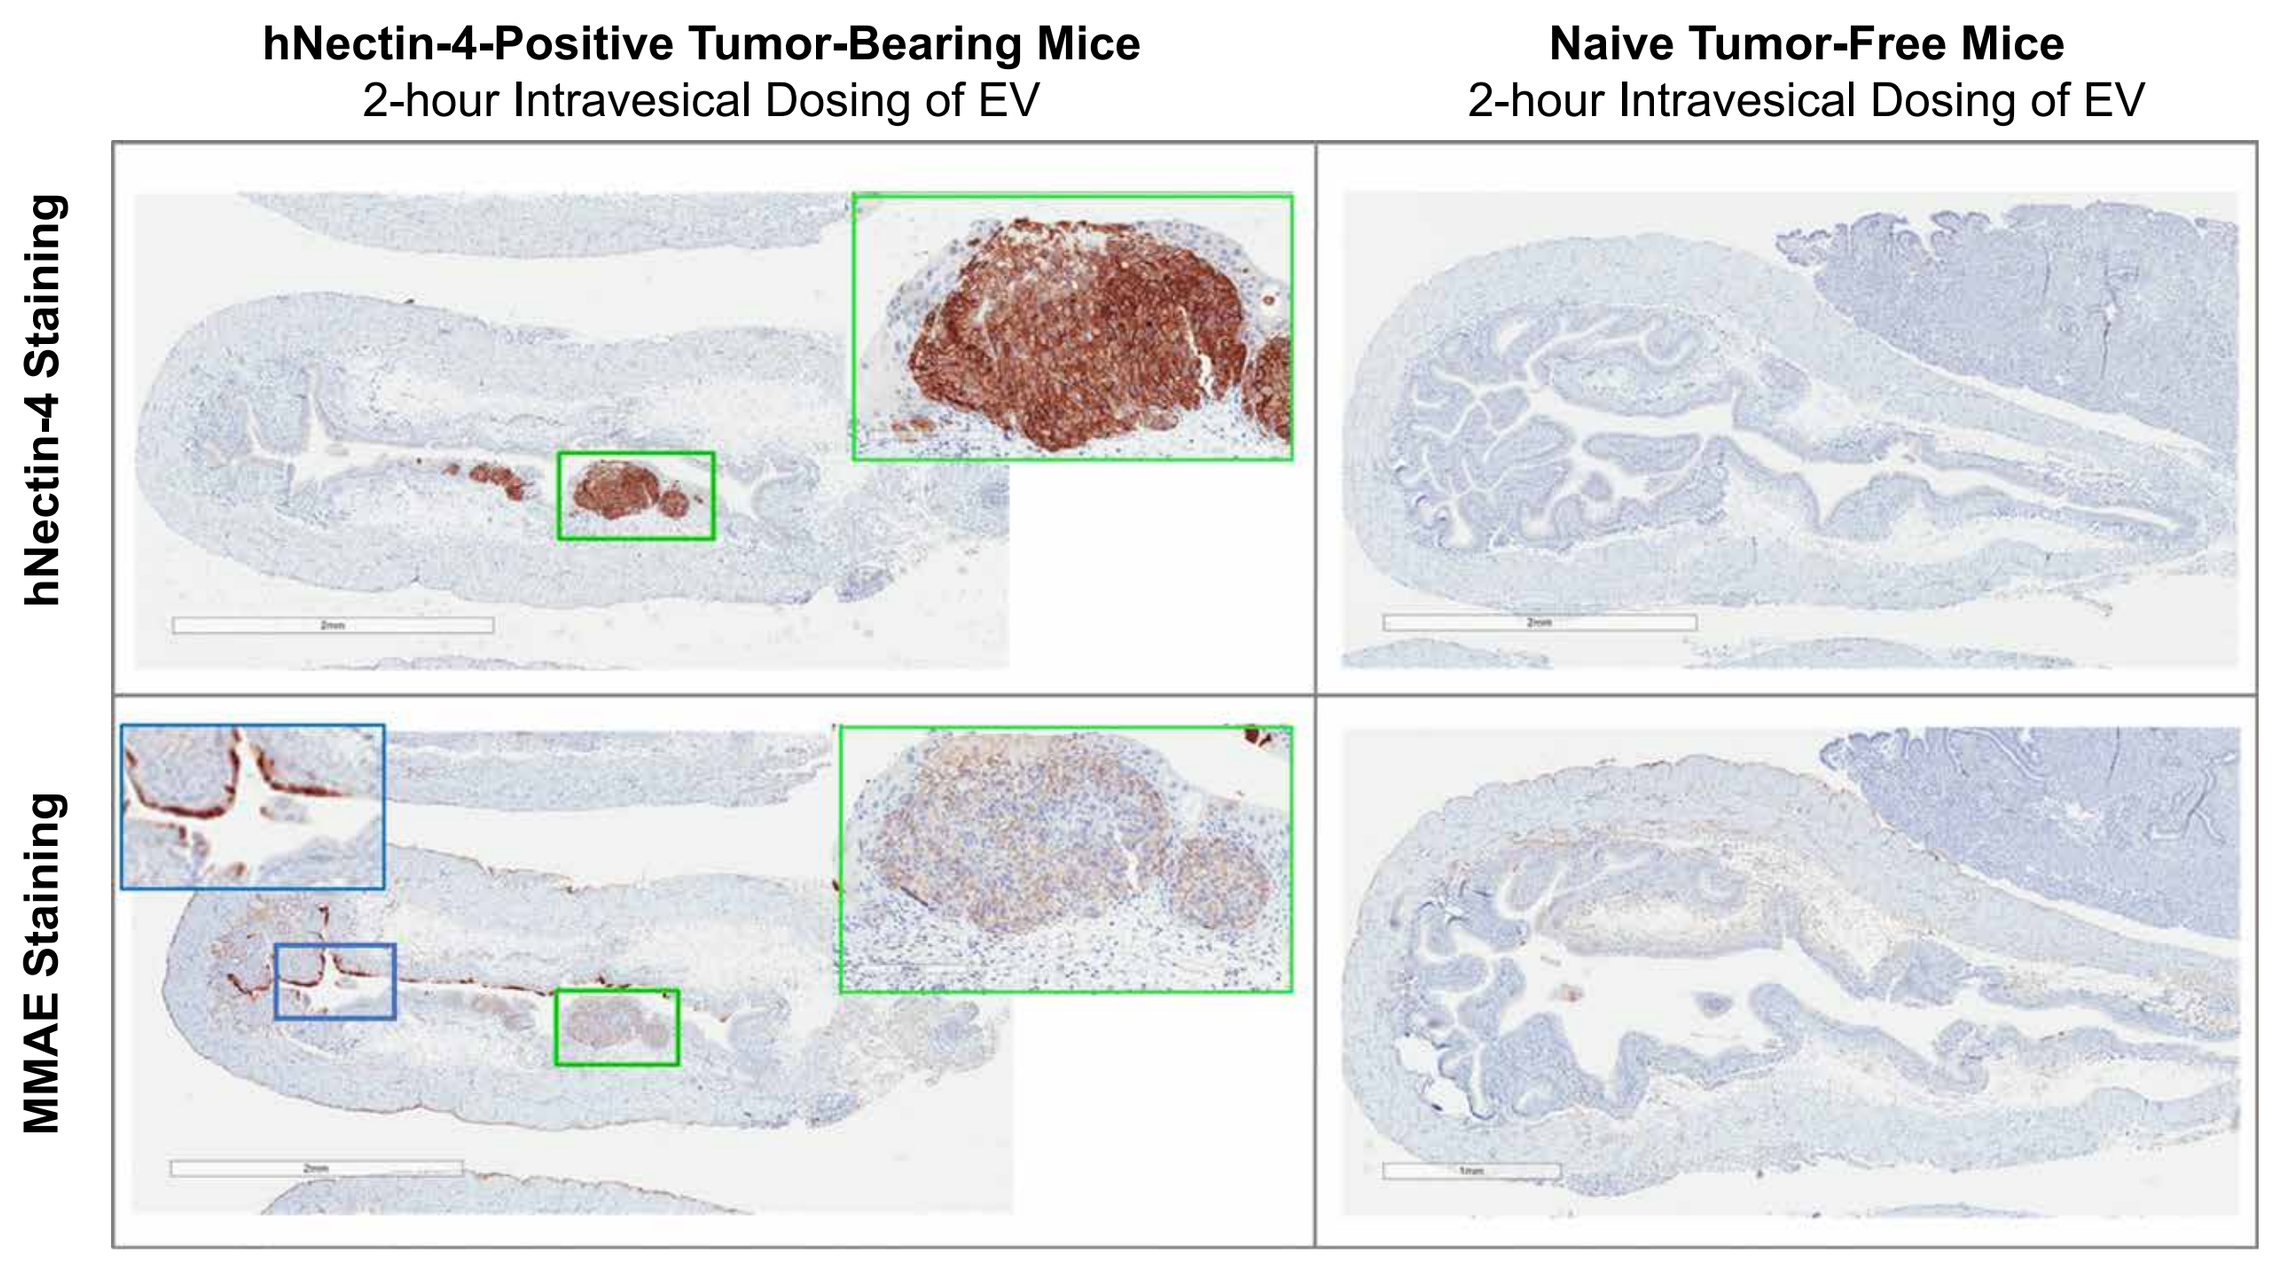
hNectin-4-Positive Tumor-Bearing Mice
2-hour Intravesical Dosing of EV
Naive Tumor-Free Mice
2-hour Intravesical Dosing of EV
hNectin-4 Staining
MMAE Staining

Figure 2. hNectin-4 and unconjugated MMAE colocalization was observed in hNectin-4 positive tumor-bearing mice treated with intravesical EV but not in naive mice, demonstrating that the specificity of EV for human Nectin-4 achieves localized delivery to tumor cells when administered via intravesical instillation.

Intravesical EV shows dose-responsive antitumor activity in a mouse orthotopic model of NMIBC

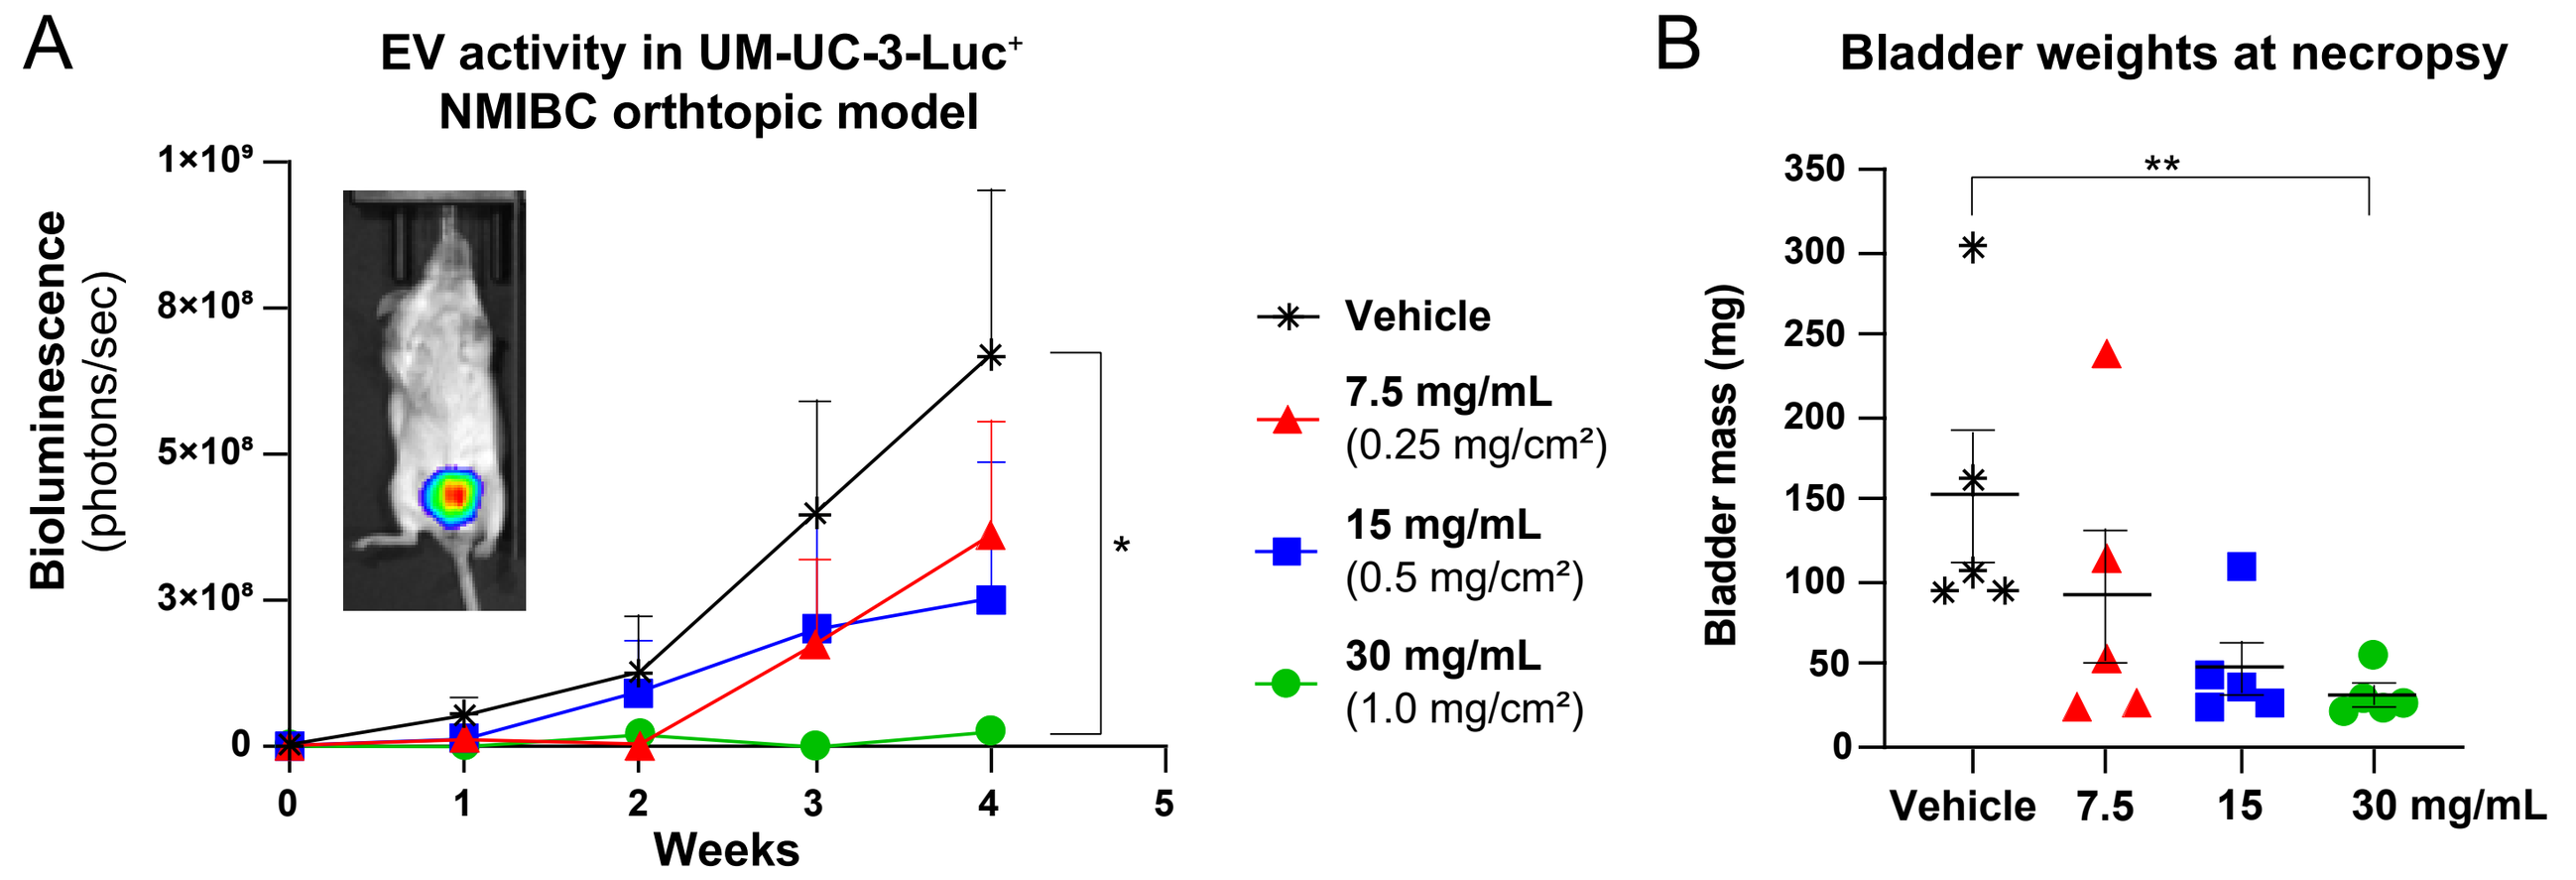
A
EV activity in UM-UC-3-Luc+ NMIBC orthtopic model
Bioluminescence (photons/sec)
Weeks
Vehicle
7.5 mg/mL (0.25 mg/cm²)
15 mg/mL (0.5 mg/cm²)
30 mg/mL (1.0 mg/cm²)
B
Bladder weights at necropsy
Bladder mass (mg)
Vehicle
7.5
15
30 mg/mL

Figure 3. In the NMIBC mouse model, engraftment of UM-UC-3-hNectin4+-Luc+ cells and disease progression was confirmed via bioluminescence imaging. Mice were administered intravesical EV weekly for 4 weeks (50 µL/dose). **A.** Mean tumor growth inhibition (TGI) measured via bioluminescence of 46%, 62%, and 96% was observed with 7.5, 15, and 30 mg/mL intravesical EV, respectively, compared to vehicle control at 4 weeks post-dose (*p = 0.0317 [Mann-Whitney t-test]. **B.** Mice treated with 30 mg/mL EV exhibited significantly less bladder weight at necropsy than vehicle-treated mice (**p = 0.0079 [Mann-Whitney t-test]).

Intravesical EV compared to IV administration has low systemic exposure in NMIBC orthotopic mouse model

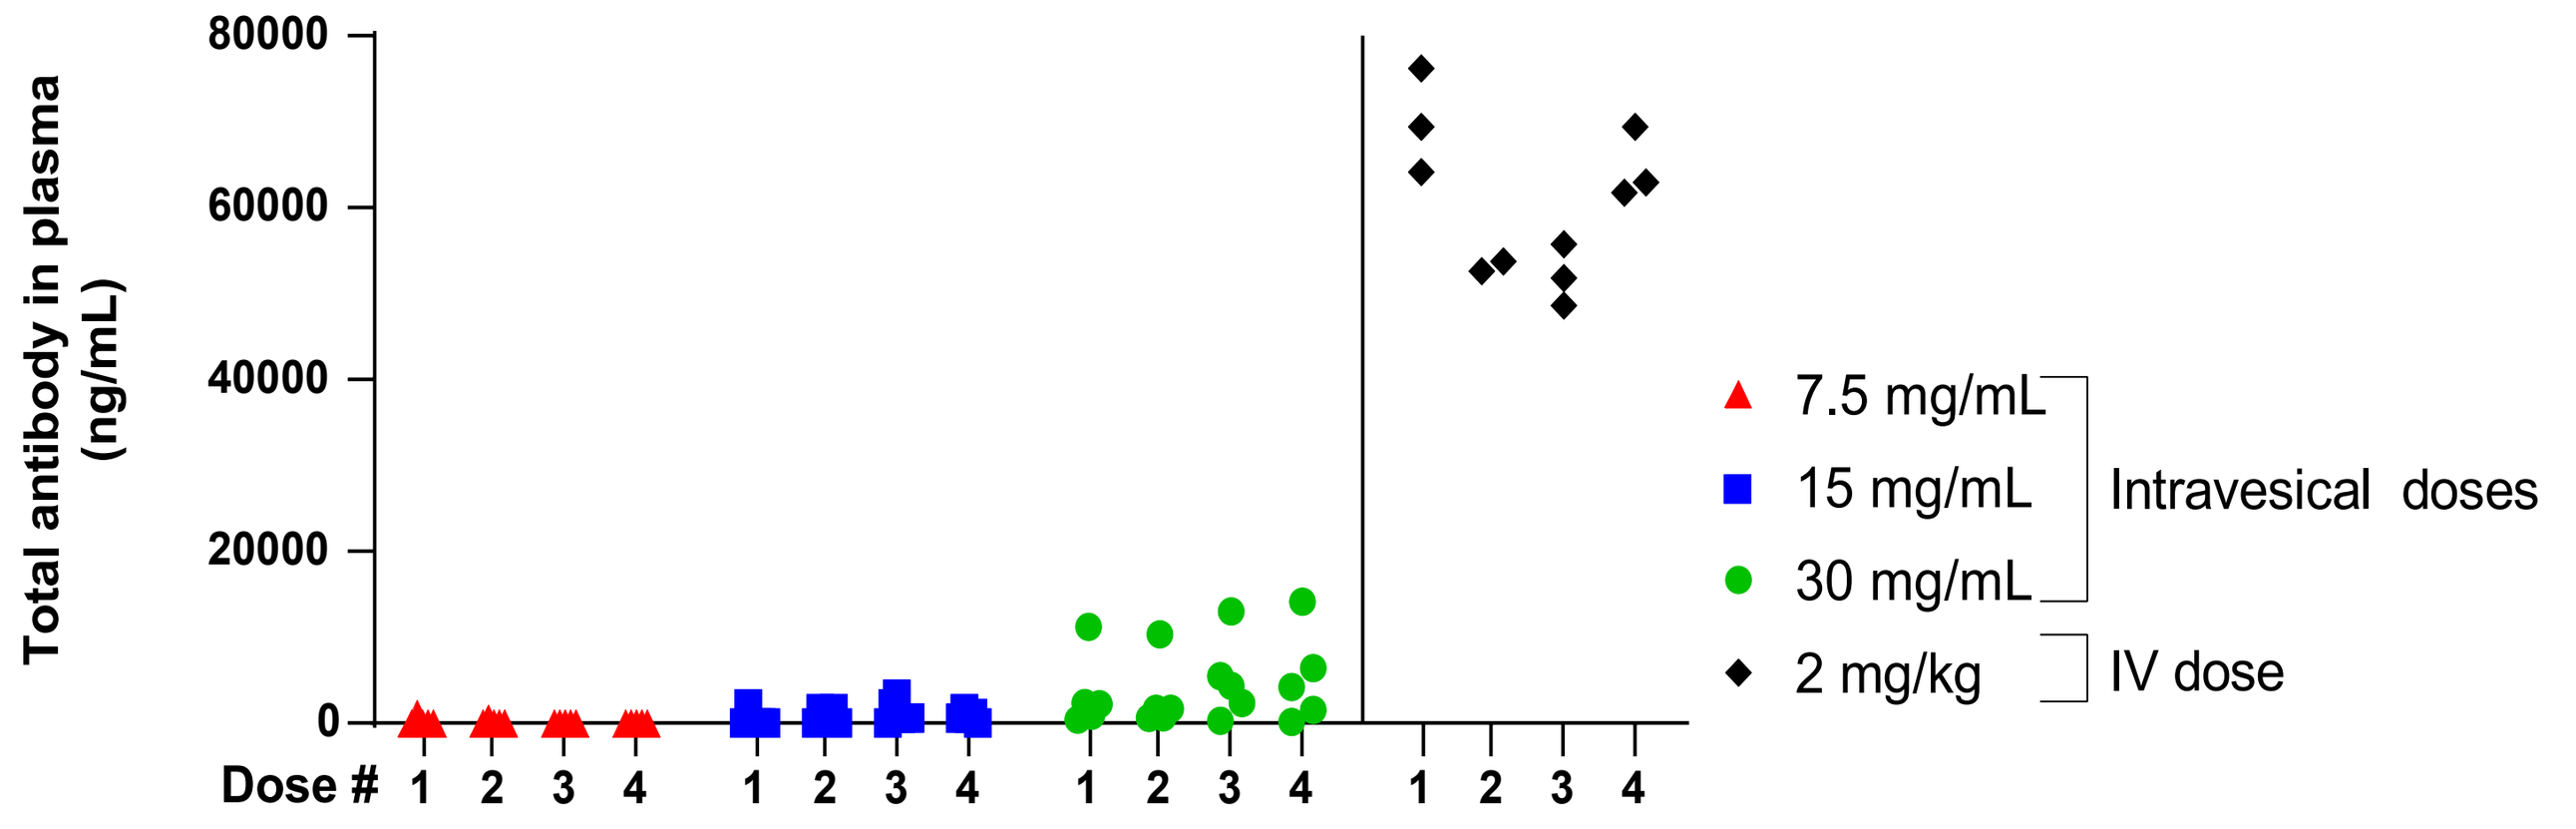
Total antibody in plasma (ng/mL)
Dose #
7.5 mg/mL
15 mg/mL
30 mg/mL
Intravesical doses
2 mg/kg
IV dose

Figure 4. Concentration of total antibody in plasma collected from mice bearing NMIBC-like tumors described in **Figure 1** 24 hours following each intravesical dose or female rats following IV administration in a GLP toxicity study. After weekly x 4 week (Q1Wx4) instillations, exposures to EV remained substantially lower than those observed on the same schedule after 2 mg/kg IV dosing, which had no adverse effects.

Intravesical EV shows minimal systemic exposure with a promising safety profile in a GLP study

- Six weekly intravesical doses of EV or control were administered to female rats to characterize the potential toxicity and pharmacokinetics of intravesical EV.
- The no-observed-effect-level of intravesical EV was 0.4 mg/cm² (10 mg/kg), and the no-observed-adverse-effect-level (microscopic findings observed) of EV (1.2 mg/cm² [30 mg/kg]) was 24-fold higher than the approved human IV dose (1.25 mg/kg).
- Low and transient serum concentrations of intravesical EV at the high dose were 35-fold lower than clinically observed serum concentrations with IV administration of EV[7,8] (**Figure 5**).

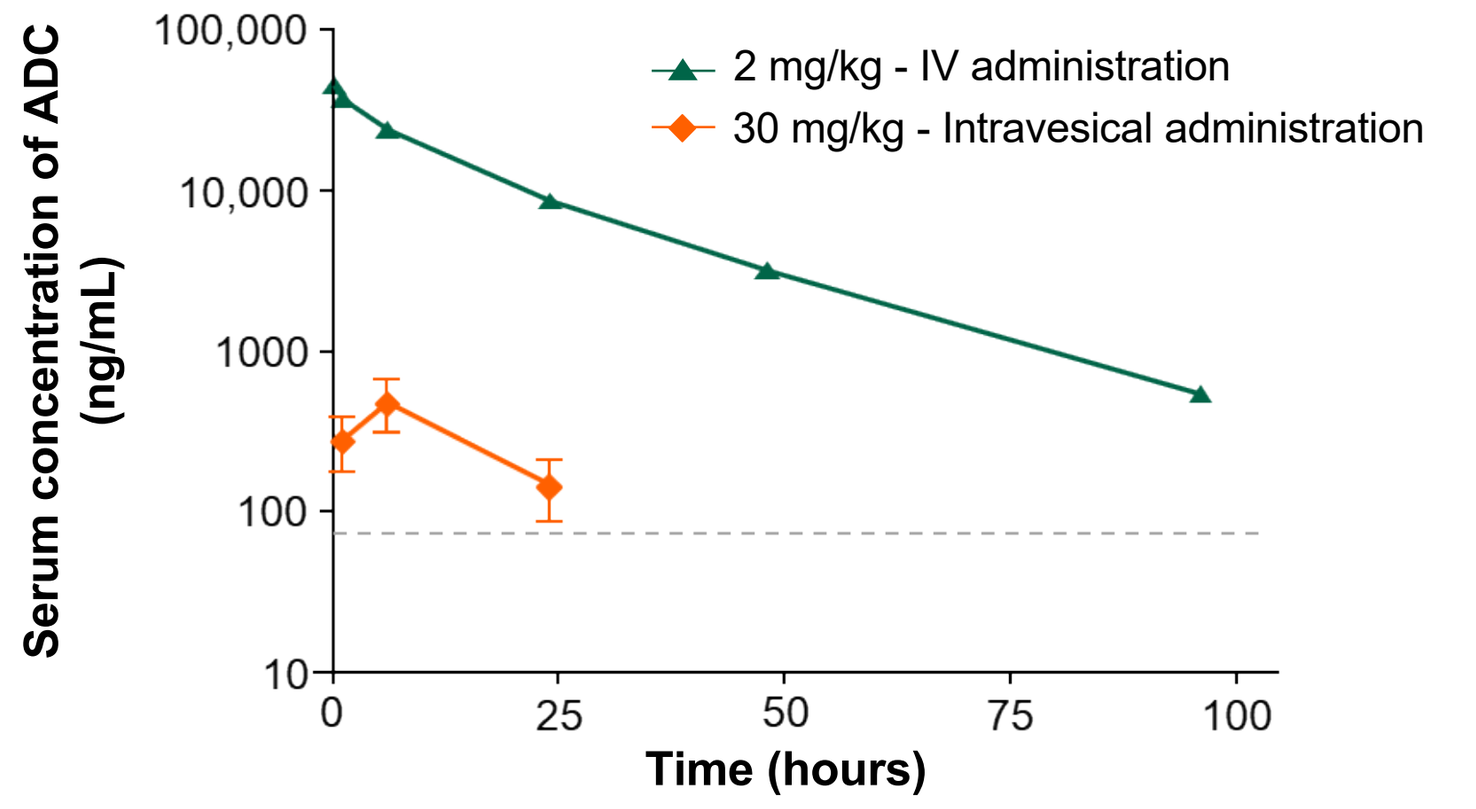
Serum concentration of ADC (ng/mL)
Time (hours)
2 mg/kg - IV administration
30 mg/kg - Intravesical administration

Figure 5. Serum collected from female rats treated with IV EV at 2 mg/kg exhibited higher concentrations of EV than serum from rats treated with intravesical EV at 30 mg/kg (1.2 mg/cm²). Datapoints represent the mean ± SEM. Dashed line shows the limit of detection. In the intravesical arm, EV was only detectable up to 24 hours post instillation, with no serum unconjugated MMAE detected.

Intravesical EV shows minimal systemic exposure with a promising safety profile in a GLP study

Table 1. Prediction of human clinical effective dose from preclinical data

		Dose[a] (mg/cm²)	Fold of Recommended Clinical Starting Dose
Fold of approved IV dose with known safety profile if 100% bioavailable (F%=100) *(1.25 mg/kg @ 100 kg max=125 mg total dose)*		0.4	1x
Approved IV dose plasma exposure predicted by observed bioavailability (ADC Cmax)		≤ 1%[b]	1x
Active intravesical dose for in vivo antitumor activity[c]		0.5	1.3x
GLP toxicity study results:	NOEL	0.4	1x
	1/6th the NOAEL[d]	0.2	0.5x
Recommended clinical starting dose (125 mg in 25 mL; 5 mg/mL)		0.4	

BLQ=below the limit of quantification; NOAEL=no-observed-adverse-effect level; NOEL=no-observed-effect level
[a]Dose as total dose/bladder surface area; dose scaling on body surface area not appropriate due to limited systemic exposure and local administration dose route, instead total dose has been normalized to bladder tissue surface area (FDA CDER. Estimating the maximum safe starting dose in initial clinical trials for therapeutics in adult healthy volunteers, Jul 2005).
[b]Fold of IV systemic exposure where IV dose is 100% bioavailable and intravesical dose is <1% bioavailable. Fold calculated by (125 mg dose/78 kg patient)*F1/(1.25 mg/kg*F100).
[c]Dose tested in an orthotopic mouse model of bladder cancer.
[d]Rat STD10 is > highest dose of 1.2 mg/cm² at 15 mg/mL tested in the GLP study, therefore as 1/10th the STD10 could not be determined, the NOEL and 1/6th the NOAEL was used to calculate the starting dose.

Table 2. Translation of doses across species

	Total Dose (mg)	Dose Concentration (mg/mL)	Dose Level[a] (mg/kg)	Dose/Bladder Surface Area[b] (mg/cm²)
Mouse (activity model)	0.38	7.5	15	0.25
	0.75	15	30	0.5
	1.5	30	60	1.0
Rat (toxicity model)	2	5	10	0.4
	6	15	30	1.2
	12	30	60	2.5
	20	50	100	4.1
Human (approved total IV dose and EV-104 dose)[c]	125	5	-	0.4

[a]Assumes 78 kg patient (mean body weight of population PK EV model population), 200 g rat, 25 g mouse.
[b]Bladder surface area derived from the volume of bladder where V=(4/3)πr3 and SA=4πr2.
[c]Assumes 1.25 mg/kg at max 100 kg for approved IV total dose.
Note: Dose scaling on body surface area is not appropriate due to limited systemic exposure and local administration dose route; instead, total dose has been normalized to bladder tissue surface area to ensure relevant preclinical doses.

Clinical trial design optimized to maximize concentration and potential for efficacy and safety

- The study was designed with minimal feasible volume of 25 mL to maximize concentration and potential for efficacy at minimal systemic exposure.
 - Minimal volume reduces the potential for urinary urgency during dwell time
- Fluid restriction recommended to minimize dilution by urine production (estimated ~50% reduction in dilution[10]).

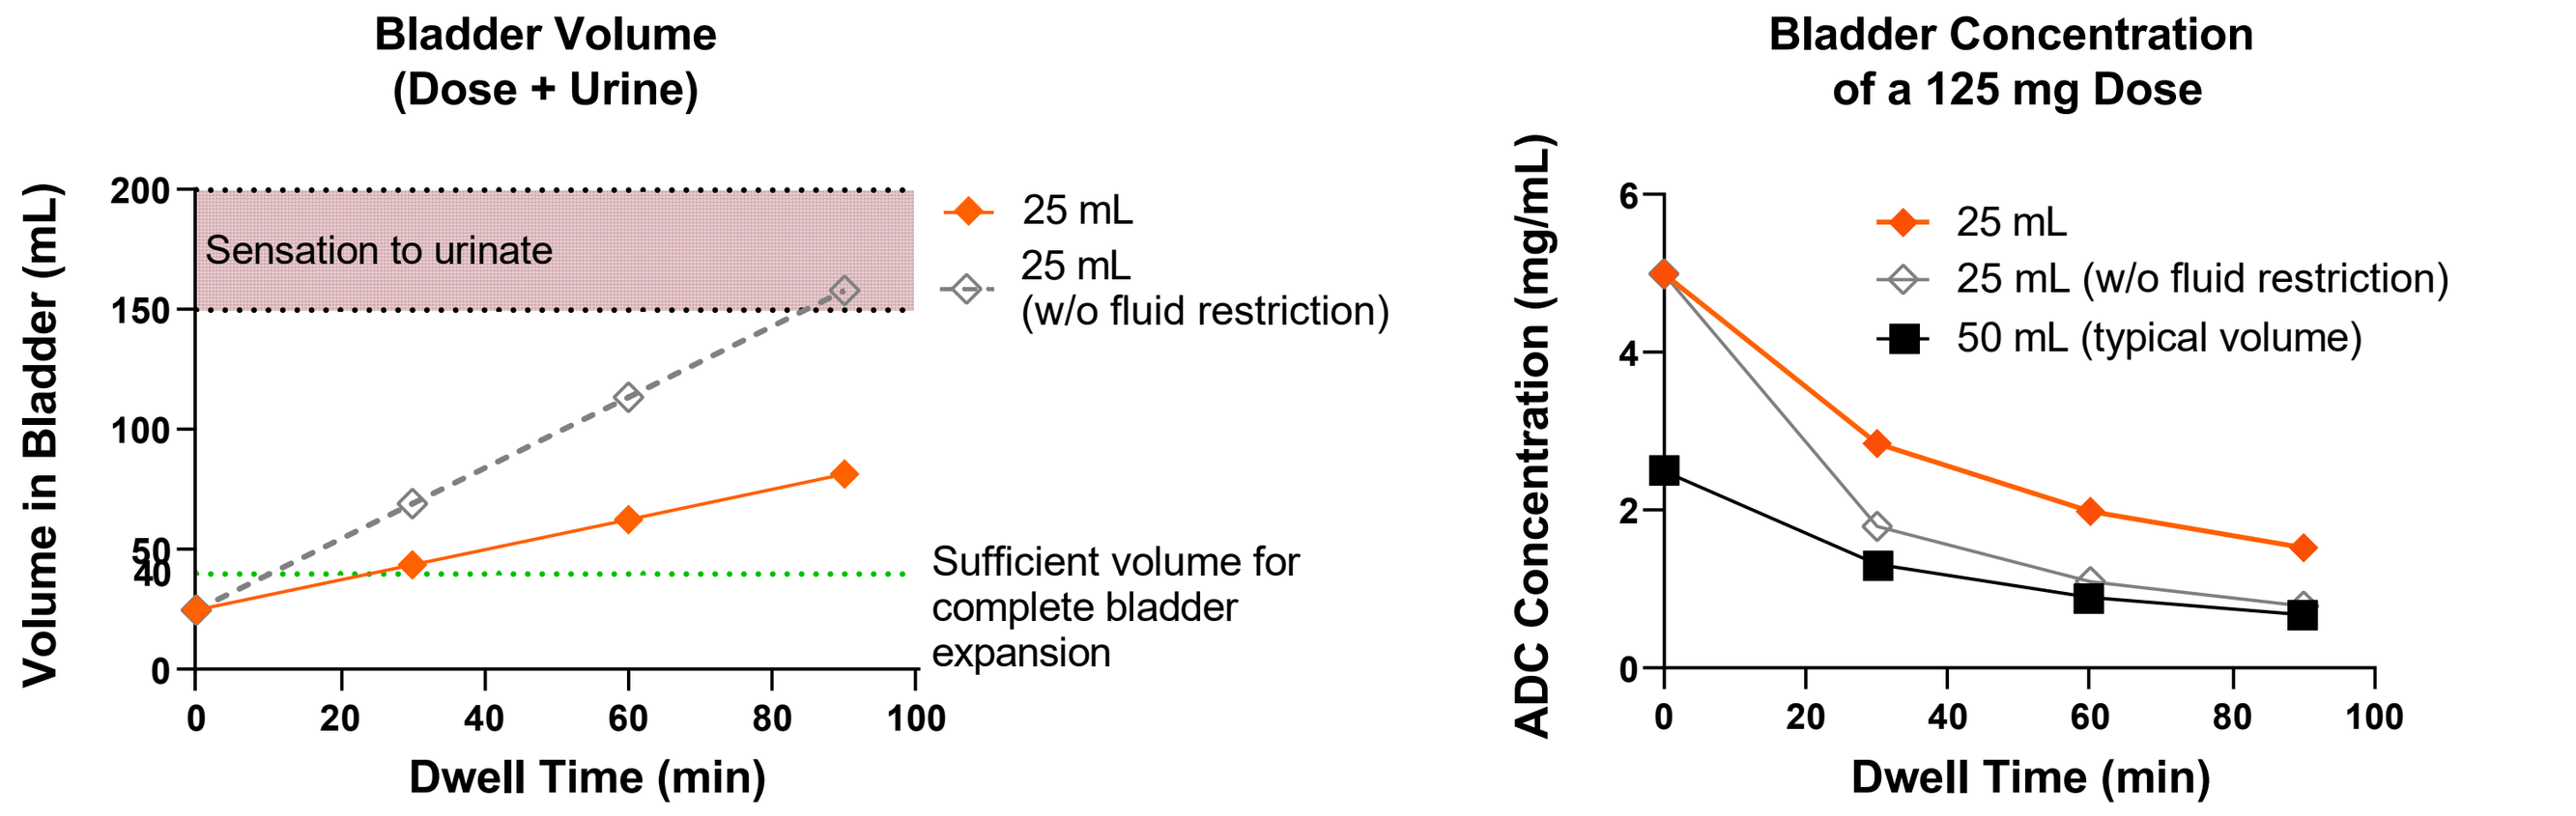
Bladder Volume (Dose + Urine)
Volume in Bladder (mL)
Dwell Time (min)
Sensation to urinate
25 mL
25 mL (w/o fluid restriction)
Sufficient volume for complete bladder expansion
Bladder Concentration of a 125 mg Dose
ADC Concentration (mg/mL)
Dwell Time (min)
25 mL
25 mL (w/o fluid restriction)
50 mL (typical volume)

Figure 6. In EV-104, volume minimization to 25 mL with fluid restriction maximizes concentration and potential for completed dwell time.

Disclosures and Acknowledgments

Astellas Pharma Inc. and Seagen Inc. provided research funding and sponsored the study. DO, YLC, EI, LF, MB, AL, KH, SA, AW, SS, and CC are employees of and have equity ownership in Seagen Inc. MS and MM are employees of Astellas affiliates and stockholders of Astellas Pharma Inc. Iliyana Mikell, an employee of Seagen Inc., provided writing and editorial assistance.

Clinical PK data confirm no detectable systemic exposure at 125 mg intravesical dosing

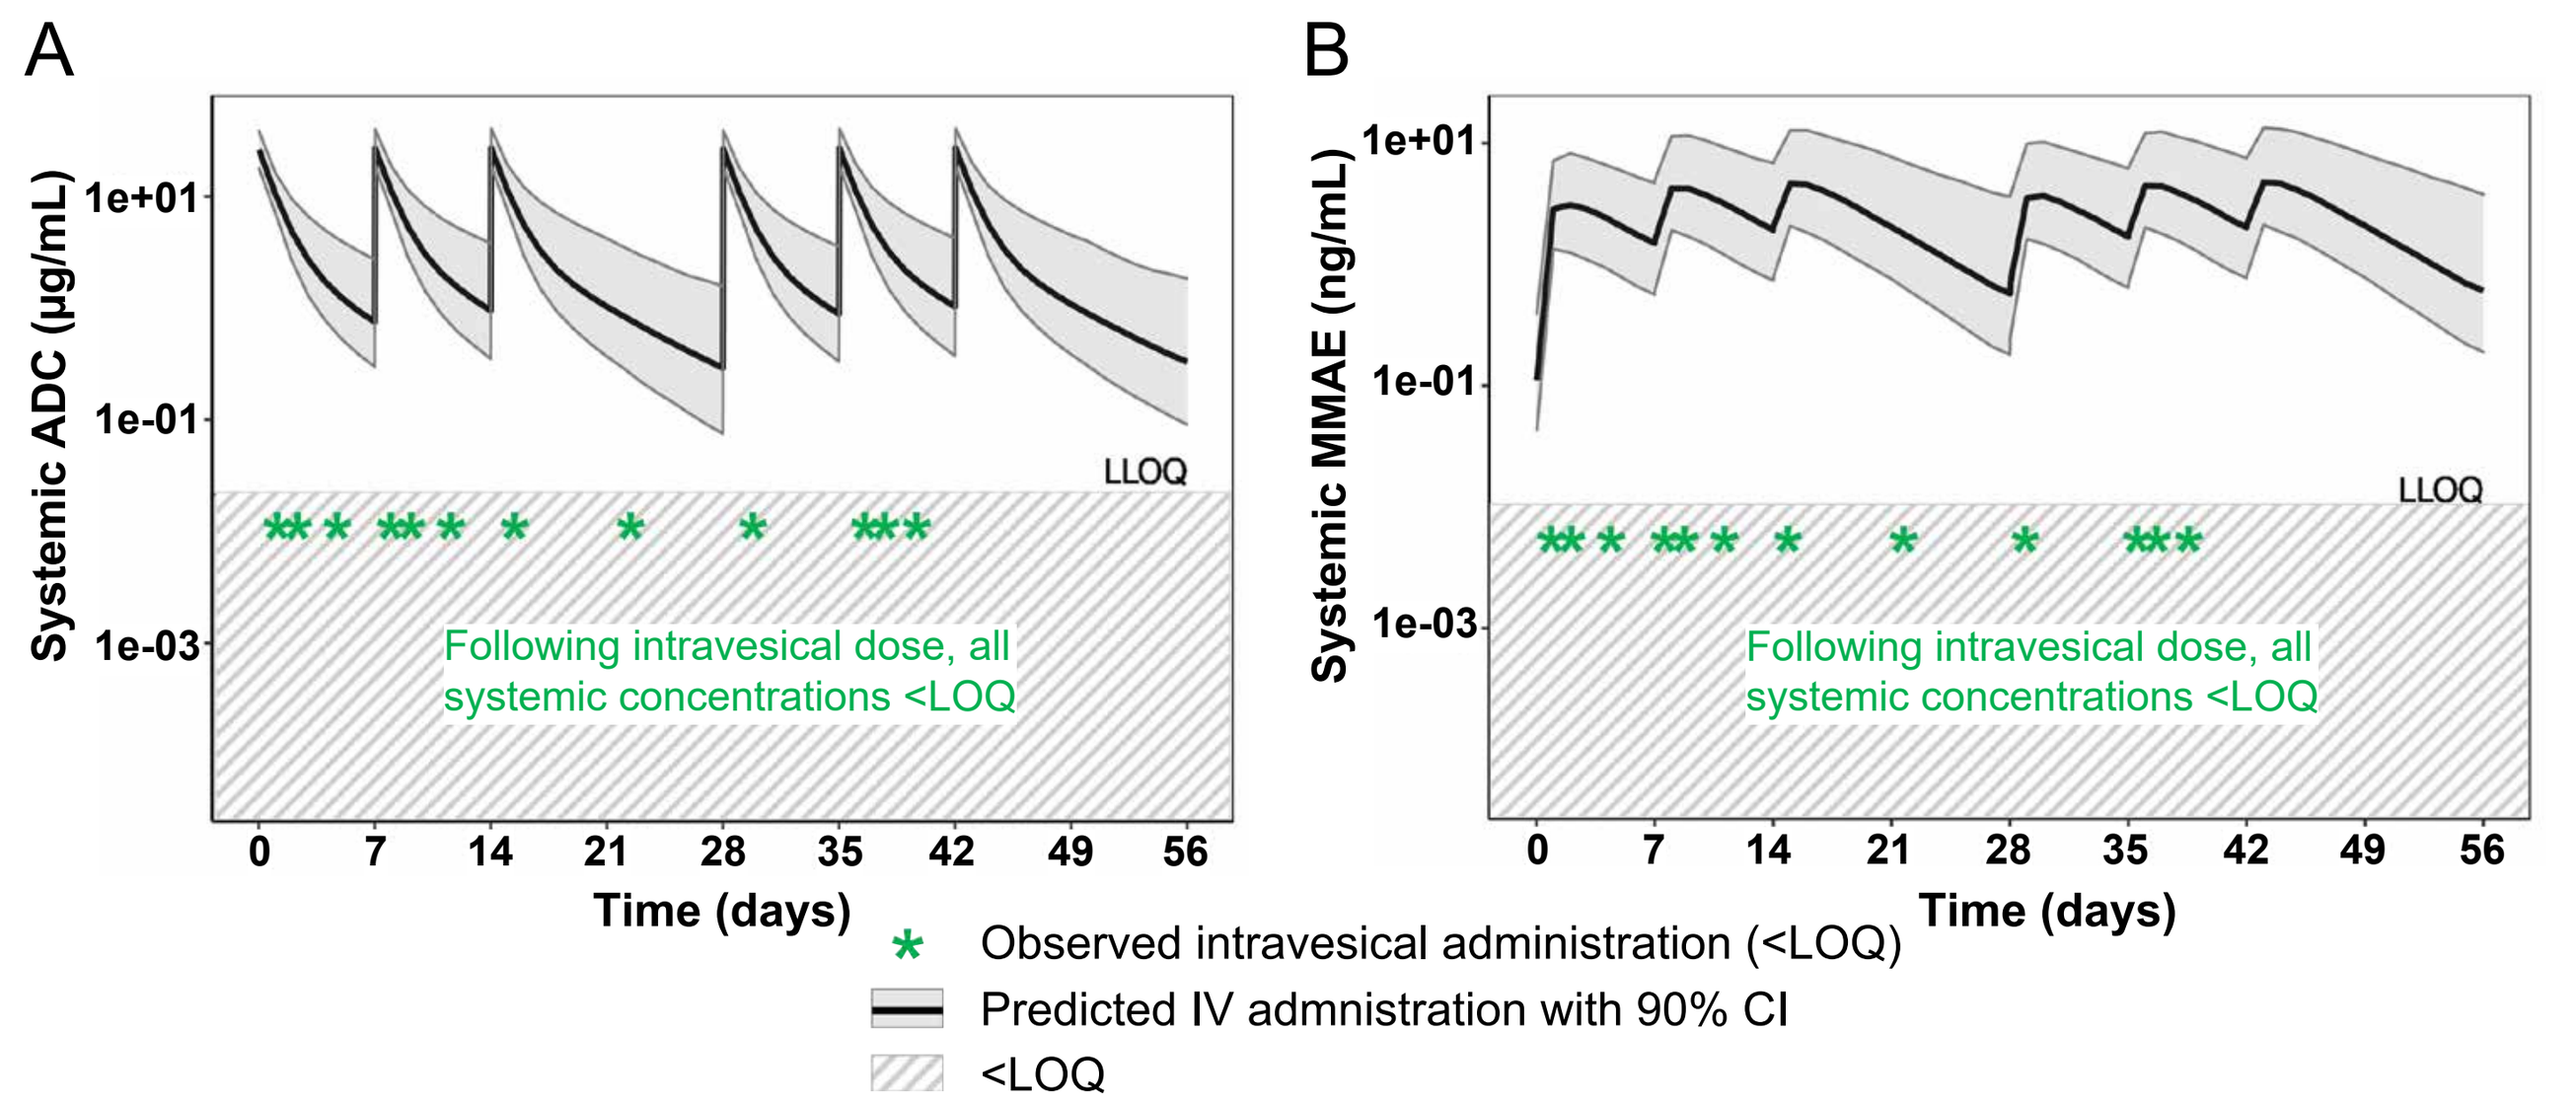
A
Systemic ADC (µg/mL)
LLOQ
Following intravesical dose, all systemic concentrations <LOQ
Time (days)
B
Systemic MMAE (ng/mL)
LLOQ
Following intravesical dose, all systemic concentrations <LOQ
Time (days)
Observed intravesical administration (<LOQ)
Predicted IV admnistration with 90% CI
<LOQ

Figure 7. Following intravesical administration, systemic ADC (A) and unconjugated MMAE (B) are undetectable in the circulation and do not overlap with predicted IV exposure at the approved dose (1.25 mg/kg on days 1,8,15 of 28-day cycle). Graphs show predicted concentration time profile of PK of IV administration and observed PK of intravesical administration, which is below the Limit of Quantification (<LOQ). In EV-104, all patients received 125 mg and had undetectable systemic exposure of ADC (Lower Limit of Quantification [LLOQ] = 0.0214 µg/mL) and unconjugated MMAE (LLOQ = 0.01 ng/mL) at all timepoints for all cycles (see **Table 3** for sample collection schedules).

Table 3. Clinical PK collections

Study Phase	Study Day	PK Collections (Blood)			
		Time	Relative Time	PK	ATA
Screening	Days -28 to 1	N/A	N/A		
Induction weeks 1, 2, and 6	Day 1	Predose	START of instillation	X	X
		Post-void	END of dwell time	X	
		2 h		X	
	Day 2	24 h	END of dwell time	X	
	Day 4	72 h		X	
Induction weeks 3, 4, and 5 and maintenance phase months 4 to 12	Day 1	Predose	START of instillation	X	X
		Post-void	END of dwell time	X	
		2 h		X	
EOT				X	X
Follow-up					

ATA= antitherapeutic antibody; EOT=end of treatment; PK=pharmacokinetic

Conclusions

- Intravesical EV in a mouse orthotopic model of NMIBC resulted in dose-dependent antitumor activity and low systemic EV exposure, supporting that the antitumor activity is driven by local exposure within the bladder.
- In a repeat-dose GLP toxicology study in rats, no systemic toxicities were observed at intravesical doses up to 6-fold higher than the maximum tolerated IV dose in that toxicology model, consistent with the minimal systemic exposure of both EV and unconjugated MMAE.
- Key preclinical learnings were incorporated in the design of EV-104, in which no systemic EV or MMAE was detectable in patients treated with 125 mg EV by intravesical administration, confirming the nonclinical model prediction of minimal systemic absorption.
- These findings provide evidence that intravesical administration of EV in NMIBC is a promising approach that limits systemic exposure, supporting the potential for a favorable safety and activity profile, and warrant continued investigation of intravesical EV in patients with NMIBC.

References

[1] Powles T, Rosenberg JE, Sonpavde GP, et al. N Engl J Med. 2021;384:1125–1135; **[2]** Chang SS, Boorjian SA, Chou R, et al. J Urol. 2016;196:1021–1029; **[3]** Woldu SL, Bagrodia A, and Lotan Y. BJU Int. 2017;119:371–380; **[4]** Kates M, Matoso A, Choi W, et al. Clin Cancer Res. 2020;26:882–89; **[5]** Li R, Li Y, Song J, et al. BMC Urol. 2020;20:97; **[6]** Matulay JT, Li R, Hensley PJ, et al. J Urol. 2021;205(6):1612-1621; **[7]** Carosino C, Olson D, Snead K et al. Cancer Res 15 June 2022; 82 (12_Supplement): 1140; **[8]** "A Study of Intravesical Enfortumab Vedotin For Treatment of Patients With Non-muscle Invasive Bladder Cancer (NMIBC)", https://clinicaltrials.gov/ct2/show/NCT05014139; **[9]** PADCEV [prescribing information]. Northbrook, IL: Astellas Pharma US, Inc., and Bothell, WA: Seagen, Inc. October 2022. https://astellas.us/docs/PADCEV_label.pdf; **[10]** Wientjes MG, Badalament RA, Au JL. Cancer Chemother Pharmacol. 1993;32(4):255-262

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster, dolson@seagen.com