Poster #56

Real-World Treatment Patterns and Clinical Outcomes with First-Line Therapy in Cisplatin-Eligible and Ineligible Patients With Advanced Urothelial Carcinoma

Guru P Sonpavde[1], Matthew Galsky[2], Phoebe Wright[3], Zsolt Hepp[3], Nancy Chang[3], Candice L Willmon[3], Steve Sesterhenn[4], Yutong Liu[5], Alicia K Morgans[1]

[1]Dana-Farber Cancer Institute, Boston, MA; [2]Tisch Cancer Institute, Icahn School of Medicine at Mount Sinai, New York, NY; [3]Seagen Inc., Bothell, WA; [4]Astellas Pharma Inc., Northbrook, IL; [5]Genesis Research, Hoboken, NJ

Background

- Locally advanced or metastatic urothelial carcinoma (la/mUC) has a poor long-term prognosis.[1]
- For patients with la/mUC, cisplatin-based chemotherapy is the preferred treatment; however, up to 50% of patients are unfit for such therapy.[2,3]
- In patients who are cisplatin-ineligible, carboplatin-based therapy may be offered as an alternative first-line (1L) therapy.[2,3]
- Therapies targeted at programmed death 1 or programmed death-ligand 1 (PD-1/L1) have received approval in the US since 2016 and are recommended in a subset of patients with la/mUC (such as those ineligible for platinum-containing chemotherapy or with tumors with high expression of PD-1/L1 biomarker).[4]
- With these recent therapeutic advances, data describing real-world treatment patterns and clinical outcomes in patients with la/mUC receiving 1L therapy are limited.
- The objective of this study was to describe contemporary treatment patterns and real-world overall survival (OS) among patients with la/mUC receiving 1L therapy stratified by their eligibility for cisplatin therapy.

Methods

- This was a retrospective, observational study using data from the nationwide Flatiron Health longitudinal electronic health record-derived database, comprising de-identified, patient-level structured and unstructured data curated via technology-enabled abstraction.
- Eligible patients were adults diagnosed with la/mUC from May 1, 2016 to October 31, 2020, selected from the Flatiron Health database, and were followed until death or end of data availability in June 2021.
- Patients were stratified by cisplatin eligibility (during the baseline period and based on the Galsky criteria[5]) and considered at the initiation of each treatment line.
- Oncologist-defined, rule-based lines of therapy were evaluated as defined by Flatiron's methodology (oncologist-defined using pre-established validated criteria developed by Flatiron).
- Median OS among patients treated in the 1L setting was assessed from the start of 1L therapy: overall, by cisplatin eligibility, and by treatment.
- Adjusted hazard ratios (HRs) and median OS were estimated using multivariate Cox regression models adjusted for metastasis site, age at index, sex, Eastern Cooperative Oncology Group performance status (ECOG PS), smoking status, and PD-1/L1 status.

Results

- Of 4,300 patients who met the inclusion criteria, 3,311 (77.0%) received 1L therapy; 1,475 (44.5%) were cisplatin-eligible and 1,836 (55.5%) were cisplatin-ineligible.
- Compared with patients who were cisplatin-ineligible, patients who were cisplatin-eligible were younger (mean age, 69.0 vs 75.0 years), had a higher creatinine clearance (CrCl; median CrCl, 80.7 vs 45.3 mL/min), and were less likely to have a documented poor ECOG PS (ECOG PS ≥2: 0% vs 29.2%; **Table 1**).

Treatment patterns

- 23% of patients did not receive any 1L therapy.
- Fewer than half of the patients who received 1L therapy subsequently received second-line (2L) therapy (n=1,471/3,311; 44.4%).
- A higher proportion of patients in the cisplatin-eligible group received 2L therapy, compared with the cisplatin-ineligible group (52.0% vs 38.3%).
- Patients who were cisplatin-eligible were more likely than those who were cisplatin-ineligible to receive systemic therapy: 79.5% of patients vs 75.1% in 1L and 41.3% versus 28.7% in 2L.
- Among patients who received 1L therapy, 879 of the 1,475 (59.6%) patients who were cisplatin-eligible did not receive cisplatin-combination therapy, while 229 of the 1,836 (12.5%) patients who were cisplatin-ineligible received cisplatin-combination therapy (**Figure 1**).

Table 1. Baseline characteristics of patients treated in the 1L setting

Variable		Cisplatin-eligible (N=1,475)	Cisplatin-ineligible (N=1,836)
Sex	Female	315 (21.4)	593 (32.3)
	Male	1,159 (78.6)	1,243 (67.7)
Age at 1L therapy initiation	Mean ± SD (median)	69.0 ± 9.2 (70.0)	75.0 ± 7.7 (77.0)
	<75 years	1,002 (67.9)	706 (38.5)
	≥75 years	473 (32.1)	1,130 (61.5)
ECOG PS	0	498 (33.8)	340 (18.5)
	1	501 (34.0)	536 (29.2)
	≥2	0 (0)	536 (29.2)
	Missing/unknown	476 (32.3)	424 (23.1)
CrCl[a]	Mean ± SD (median)	86.9 ± 25.2 (80.7)	46.3 ± 19.2 (45.3)
	<45 mL/min	0 (0)	874 (47.6)
	45–60 mL/min	0 (0)	711 (38.7)
	≥60 mL/min	1,179 (79.9)	183 (10.0)
Total number of LOTs	1	708 (48.0)	1,132 (61.7)
	2	495 (33.6)	470 (25.6)
	3	158 (10.7)	153 (8.3)
	≥4	114 (7.7)	81 (4.4)

Data expressed as n (%) unless stated otherwise.
[a]Data missing for 364 patients (11.0%).
1L, first-line; CrCl, creatinine clearance; ECOG PS, Eastern Cooperative Oncology Group performance status; LOT, line of therapy; SD, standard deviation.

Figure 1. Treatment patterns among (A) patients who were cisplatin-eligible and (B) patients who were cisplatin-ineligible receiving 1L therapy

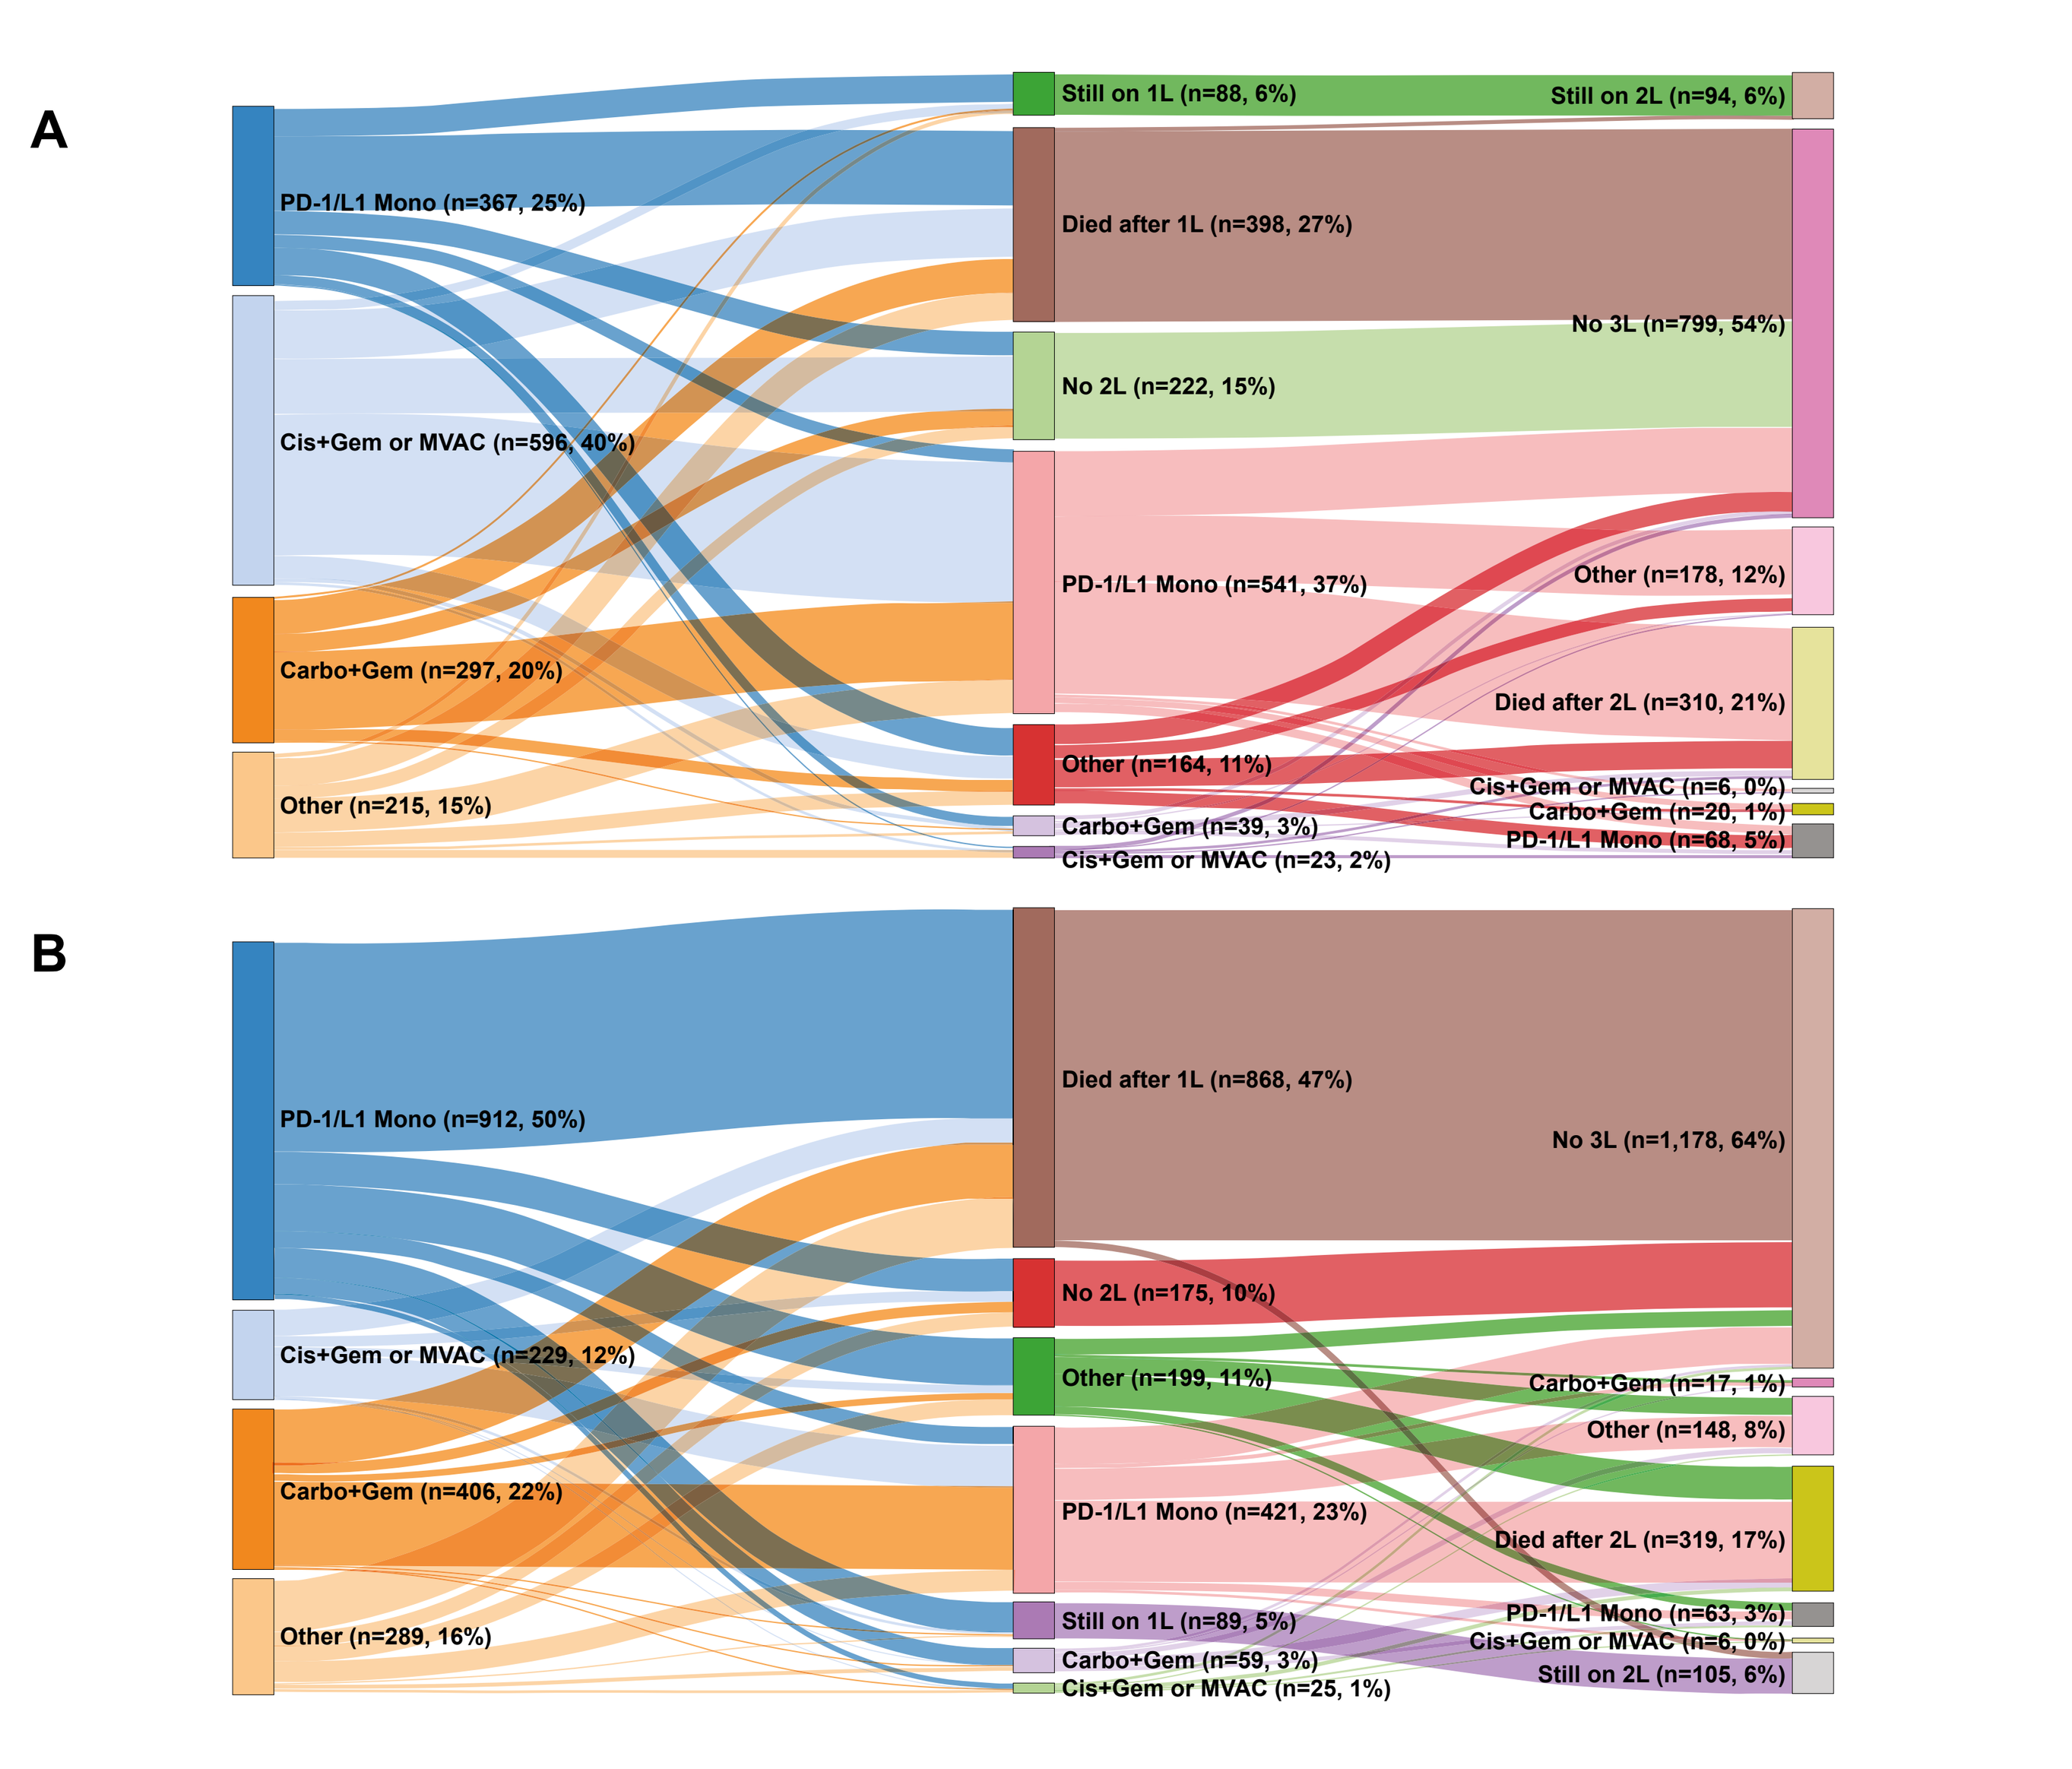
A
PD-1/L1 Mono (n=367, 25%)
Cis+Gem or MVAC (n=596, 40%)
Carbo+Gem (n=297, 20%)
Other (n=215, 15%)
Still on 1L (n=88, 6%)
Died after 1L (n=398, 27%)
No 2L (n=222, 15%)
PD-1/L1 Mono (n=541, 37%)
Other (n=164, 11%)
Carbo+Gem (n=39, 3%)
Cis+Gem or MVAC (n=23, 2%)
Still on 2L (n=94, 6%)
No 3L (n=799, 54%)
Other (n=178, 12%)
Died after 2L (n=310, 21%)
Cis+Gem or MVAC (n=6, 0%)
Carbo+Gem (n=20, 1%)
PD-1/L1 Mono (n=68, 5%)
B
PD-1/L1 Mono (n=912, 50%)
Cis+Gem or MVAC (n=229, 12%)
Carbo+Gem (n=406, 22%)
Other (n=289, 16%)
Died after 1L (n=868, 47%)
No 2L (n=175, 10%)
Other (n=199, 11%)
PD-1/L1 Mono (n=421, 23%)
Still on 1L (n=89, 5%)
Carbo+Gem (n=59, 3%)
Cis+Gem or MVAC (n=25, 1%)
No 3L (n=1,178, 64%)
Carbo+Gem (n=17, 1%)
Other (n=148, 8%)
Died after 2L (n=319, 17%)
PD-1/L1 Mono (n=63, 3%)
Cis+Gem or MVAC (n=6, 0%)
Still on 2L (n=105, 6%)

Other therapies included PD-1/L1 combination therapy, mono-chemotherapy (taxanes, gemcitabine, cisplatin monotherapy, carboplatin monotherapy), and other off-label treatments.
1L/2L/3L, first-/second-/third-line; Carbo, carboplatin; Cis, cisplatin; Gem, gemcitabine; Mono, monotherapy; MVAC, methotrexate/vinblastine/doxorubicin/cisplatin; PD-1/L1, programmed death 1 or programmed death-ligand 1.

Overall survival with 1L therapy

- Median OS among patients receiving 1L therapy overall was 11.0 (95% confidence interval [CI]: 10.3–11.5) months.
- Median OS was longer in patients who were cisplatin-eligible than in patients who were cisplatin-ineligible (14.4 [95% CI: 13.4–16.4] vs 8.6 [95% CI: 8.1–9.2] months, respectively; HR 0.8 [95% CI: 0.7–1.1]; **Figure 2**).

Figure 2. Overall survival among patients receiving 1L therapy by cisplatin eligibility

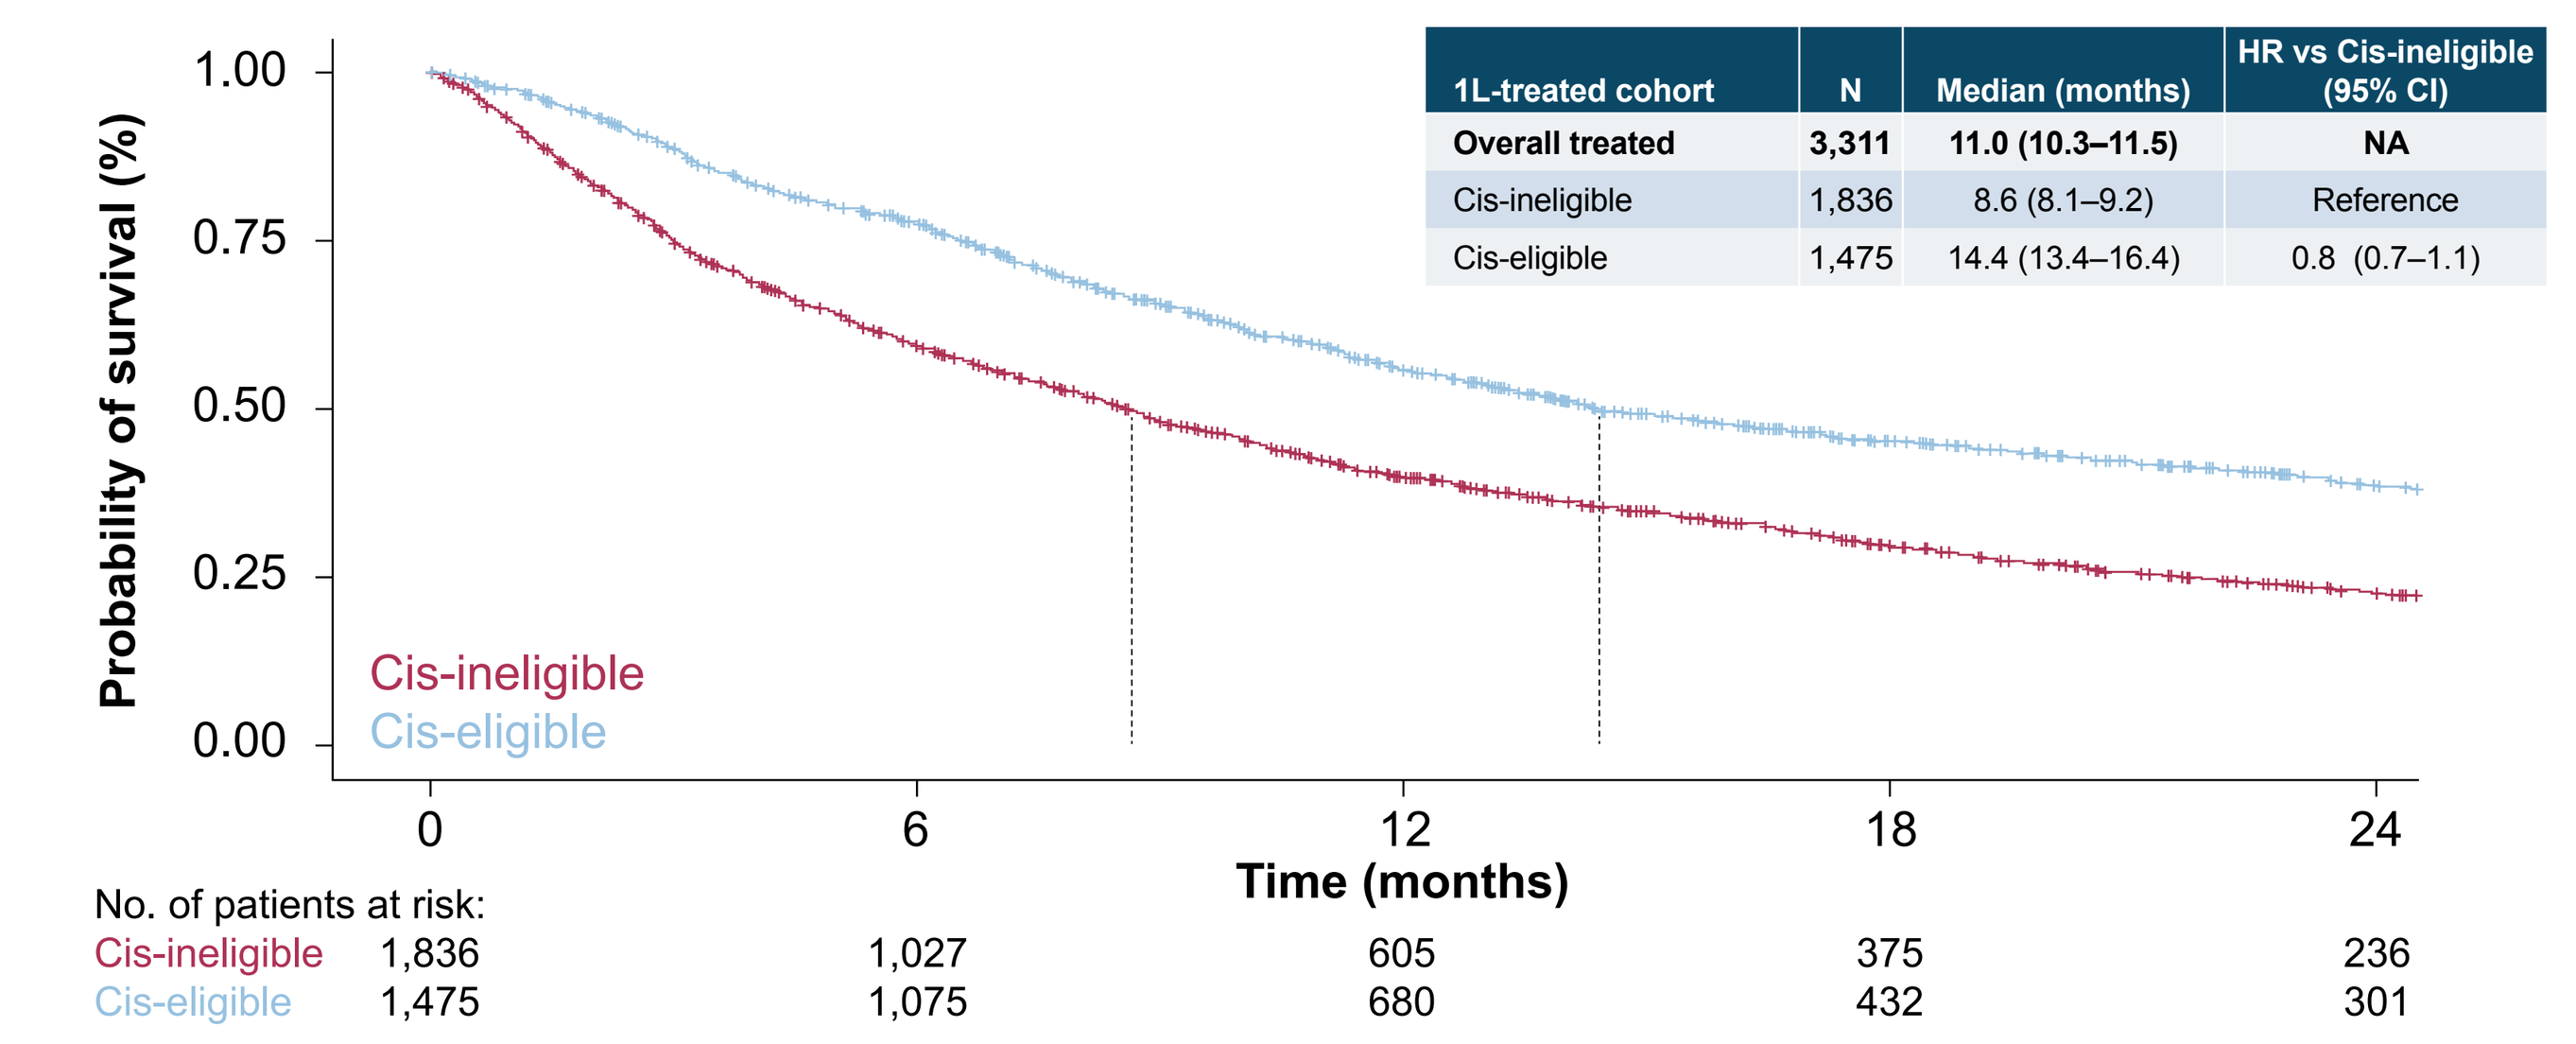
Probability of survival (%)
1.00
0.75
0.50
0.25
0.00
Cis-ineligible
Cis-eligible
0
6
12
18
24
Time (months)
No. of patients at risk:
Cis-ineligible 1,836 1,027 605 375 236
Cis-eligible 1,475 1,075 680 432 301

1L-treated cohort	N	Median (months)	HR vs Cis-ineligible (95% CI)
Overall treated	**3,311**	**11.0 (10.3–11.5)**	**NA**
Cis-ineligible	1,836	8.6 (8.1–9.2)	Reference
Cis-eligible	1,475	14.4 (13.4–16.4)	0.8 (0.7–1.1)

HR was adjusted for primary cancer site, age, sex, ECOG PS score, smoking status, PD-1/L1 status, and CrCl.
1L, first-line; CI, confidence interval; Cis, cisplatin; CrCl, creatinine clearance; ECOG PS, Eastern Cooperative Oncology Group performance status; HR, hazard ratio; NA, not applicable; PD-1/L1, programmed death 1 or programmed death-ligand 1.

- When stratified by 1L therapy received, cisplatin + gemcitabine or MVAC was associated with longer OS compared with other therapies, in both patients who were cisplatin-eligible (median OS 20.6 months vs range 11.7–13.0 months for other therapies) and patients who were cisplatin-ineligible (median OS 13.5 vs range 6.4–9.8 months; **Figure 3**).

Figure 3. Overall survival among patients receiving 1L therapy who were (A) cisplatin-eligible and (B) cisplatin-ineligible, according to treatments received

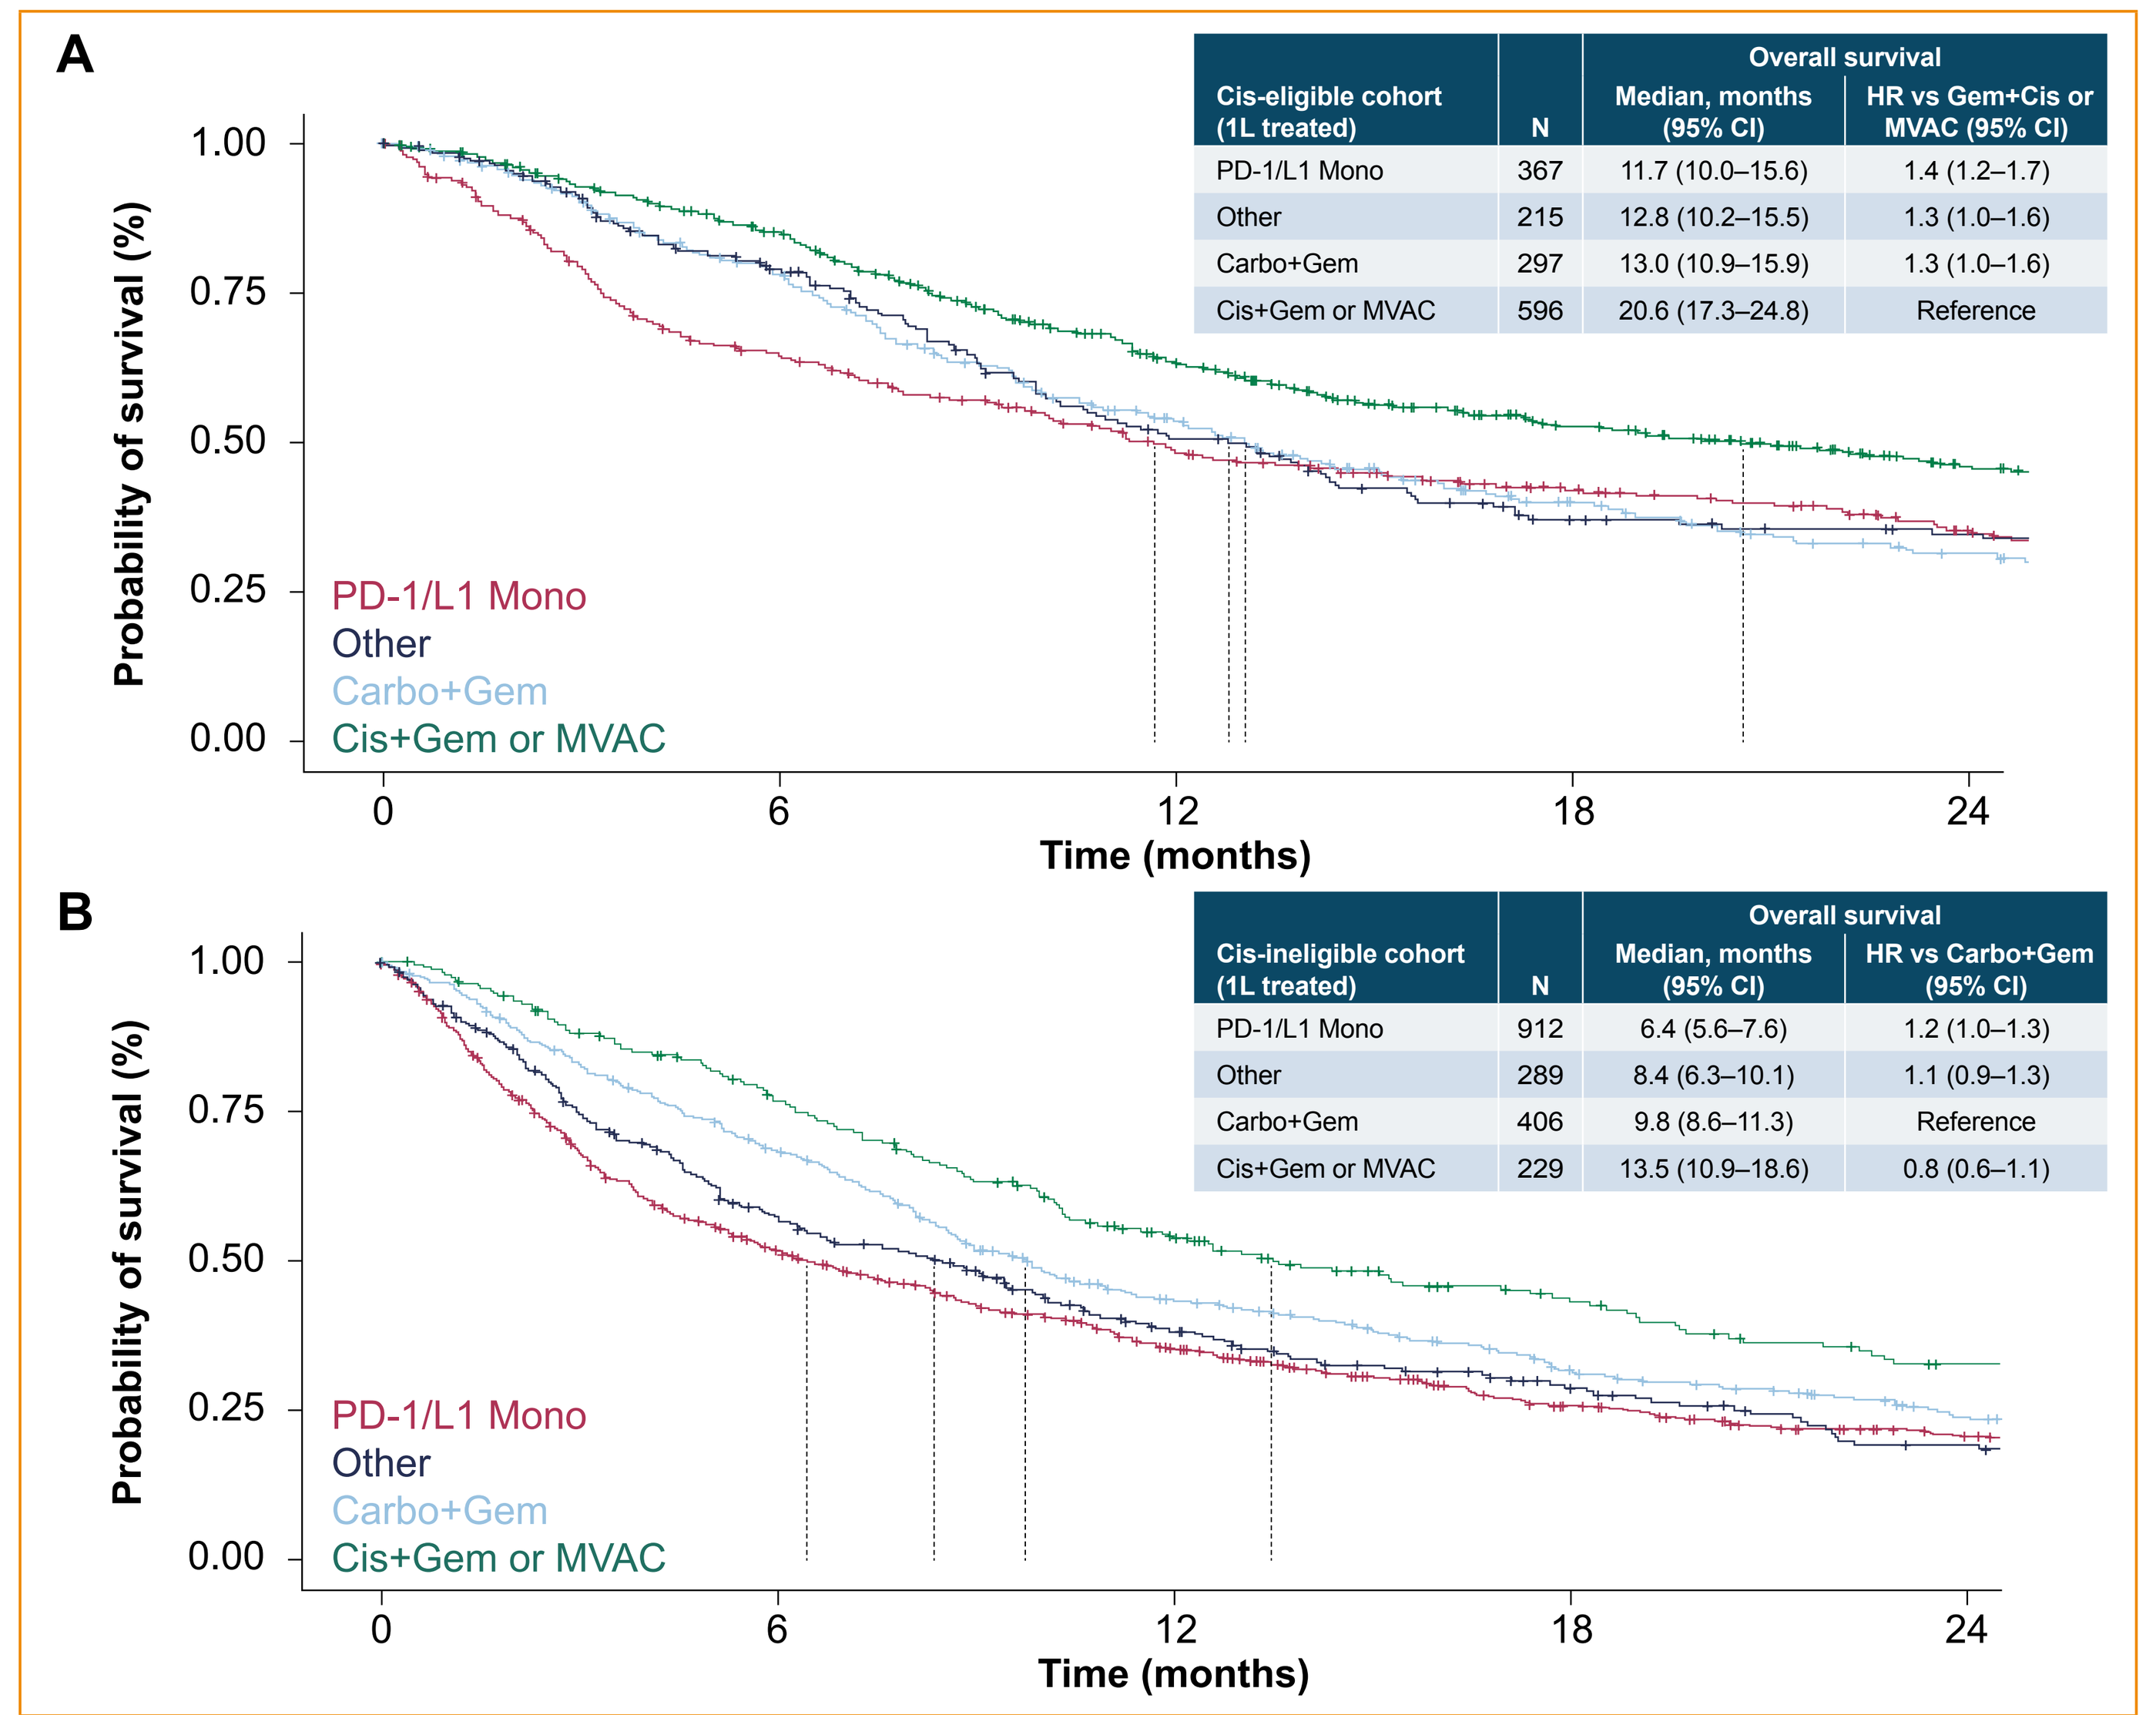
A
Probability of survival (%)
1.00
0.75
0.50
0.25
0.00
PD-1/L1 Mono
Other
Carbo+Gem
Cis+Gem or MVAC
0
6
12
18
24
Time (months)
B
Probability of survival (%)
PD-1/L1 Mono
Other
Carbo+Gem
Cis+Gem or MVAC
Time (months)

A

Cis-eligible cohort (1L treated)	N	Overall survival: Median, months (95% CI)	Overall survival: HR vs Gem+Cis or MVAC (95% CI)
PD-1/L1 Mono	367	11.7 (10.0–15.6)	1.4 (1.2–1.7)
Other	215	12.8 (10.2–15.5)	1.3 (1.0–1.6)
Carbo+Gem	297	13.0 (10.9–15.9)	1.3 (1.0–1.6)
Cis+Gem or MVAC	596	20.6 (17.3–24.8)	Reference

B

Cis-ineligible cohort (1L treated)	N	Overall survival: Median, months (95% CI)	Overall survival: HR vs Carbo+Gem (95% CI)
PD-1/L1 Mono	912	6.4 (5.6–7.6)	1.2 (1.0–1.3)
Other	289	8.4 (6.3–10.1)	1.1 (0.9–1.3)
Carbo+Gem	406	9.8 (8.6–11.3)	Reference
Cis+Gem or MVAC	229	13.5 (10.9–18.6)	0.8 (0.6–1.1)

HR adjusted for cisplatin eligibility, primary cancer site, age, sex, ECOG PS score, smoking status, PD-1/L1 status, and CrCl. Other therapies included PD-1/L1 combination therapy, mono-chemotherapy (taxanes, gemcitabine, cisplatin monotherapy, carboplatin monotherapy), and other off-label treatments.
1L, first-line; Carbo, carboplatin; CI, confidence interval; Cis, cisplatin; CrCl, creatinine clearance; ECOG PS, Eastern Cooperative Oncology Group performance status; Gem, gemcitabine; HR, hazard ratio; Mono, monotherapy; MVAC, methotrexate/vinblastine/doxorubicin/cisplatin; PD-1/L1, programmed death 1 or programmed death-ligand 1.

Limitations

- There was the potential for misclassification of cisplatin eligibility due to missing data; however, this was tested via sensitivity analysis (in which patients with missing ECOG PS and CrCl data were excluded), and the results remained consistent with the primary analysis.
- Comparisons should be interpreted with caution; there was the potential for residual confounding due to the Cox model's inability to adjust for all confounders, despite all available characteristics being included.
- This study did not assess the use of maintenance therapy; only a small number of patients (89 in total) were receiving avelumab therapy. As maintenance therapy continues to be part of standard of care, future analyses should explore the impact of maintenance therapy.

Conclusions

- Clinical outcomes among patients with la/mUC receiving 1L therapy were poor and may have been at least partly associated with the specific treatment regimen as well as cisplatin ineligibility.
- Many patients (23%) with la/mUC did not receive 1L therapy (although the proportion receiving 1L therapy is likely increasing with the availability of PD-1/L1 therapy[4]), and among those who did, fewer than half went on to receive 2L therapy.
 - Patients who were cisplatin-ineligible had shorter OS.
 - Regardless of cisplatin eligibility, OS was longer in patients treated with cisplatin-based therapy compared with all other treatments.
- Among patients who received 1L therapy, 12.5% of patients who were cisplatin-ineligible received cisplatin-based therapy while 59.6% of patients who were cisplatin-eligible did not.
 - This suggests there is some subjectivity in applying the cisplatin-eligibility criteria and that physicians are considering factors beyond conventional criteria to determine cisplatin eligibility.
- This study highlights the need for more effective and tolerable 1L therapy to improve outcomes for all patients with la/mUC, especially among patients who are cisplatin-ineligible.

References

1. Hepp Z, et al. Future Oncol. 2021;17:4343-53.
2. Hepp Z, et al. J Manag Care Spec Pharm. 2021;27:240-55.
3. Koufopoulou M, et al. Cancer Treat Rev. 2020;89:102072.
4. National Comprehensive Cancer Network. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Bladder Cancer V.5.2021. https://www.nccn.org/professionals/physician_gls/pdf/bladder.pdf.
5. Galsky MD, et al. J Clin Oncol. 2011;29:2432-8.

DISCLOSURES: This work was supported by funding from Seagen Inc. and Astellas, Inc.

GPS is a consultant for Amgen, Astellas/Agensys, AstraZeneca, Bayer, BMS, Eisai, EMD Serono, Exelixis, Genentech, Janssen, Merck, Novartis, Pfizer, Sanofi, and Seagen Inc.; has received research support to his institution from Amgen, AstraZeneca, Bayer, Boehringer Ingelheim, Janssen, Merck, Pfizer, and Sanofi; is an author for UpToDate; serves on the steering committee for Astellas, AstraZeneca, Bavarian Nordic, BMS, and Debiopharm; and is a speaker for OncLive, Research to Practice, and Physician Education Resource.

MG is a paid consultant for Aileron Therapeutics, Astellas, AstraZeneca, BioMotiv, BMS, Dendreon, Dracen Pharmaceutical, EMD Serono, Genentech, GSK, Incyte, Inovio Pharmaceuticals, Janssen, Lilly, Merck, Novartis, Numab, Pfizer, and Seagen Inc.; has equity ownership in Rappta Therapeutics; holds patents/royalties for "Methods and compositions for treating cancer and related methods (20120322792)"; and has received research funding from AstraZeneca, BMS, Dendreon, Genentech, Janssen, Merck, and Novartis.

PW, ZH, NC, and CLW are employees of, and own stock in, Seagen Inc.

SS is an employee of Astellas Pharma and owns stock in Seagen Inc. and Merck.

YL is an employee of Genesis Research and a paid consultant to Seagen Inc. in connection with this study.

AKM received consulting/honoraria from Advanced Accelerator Applications, Astellas, AstraZeneca, Bayer, Clovis, Dendreon, Genentech, Janssen, Lantheus, Myriad Genetics, Myovant, Novartis, Pfizer, Sanofi, Seagen Inc., and Telix; research support from Astellas, Bayer, Myovant, and Sanofi; and honoraria for medical education work for Medscape, OncLive, and UroToday.

ACKNOWLEDGMENTS: Medical writing support was provided by Joshua Fink, PhD, of Curo Consulting, a division of Envision Pharma Group, and funded by Seagen Inc.

Corresponding author: Guru P Sonpavde (GuruP_Sonpavde@DFCI.HARVARD.EDU)

Copies of this poster obtained through the Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, IL, June 3–7, 2022
© Copyright 2022