HER2 Testing in the MOUNTAINEER Trial: Analysis of Treatment Response Based on Central HER2 Assessment Using IHC/ISH and NGS

John H Strickler,[1] Andrea Cercek,[2] Kimmie Ng,[3] Salvatore Siena,[4] Thierry Andre,[5] Eric Van Cutsem,[6] Christina Wu,[7] Andrew Scott Paulson,[8] Joleen M. Hubbard,[9] Andrew L. Coveler,[10] Christos Fountzilas,[11] Adel Kardosh,[12] Pashtoon Murtaza Kasi,[13] Heinz-Josef Lenz,[14] Kristen Keon Ciombor,[15] Elena Elez,[16] Michael Stecher,[17] Pauline Cronin,[17] Mark Bieda,[17] Tanios S. Bekaii-Saab[18]

[1]Duke University Medical Center, Durham, NC, USA; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Grande Ospedale Metropolitano Niguarda and Università degli Studi di Milano, Milan, Italy; [5]Sorbonne Université, Department of Medical Oncology, Saint-Antoine Hospital, AP-HP, Paris, France; [6]University Hospital Gasthuisberg and University of Leuven, Leuven, Belgium; [7]Mayo Clinic, Phoenix, AZ, USA; [8]Texas Oncology/The US Oncology Network, Dallas, TX, USA; [9]Mayo Clinic, Rochester, MN, USA; [10]Fred Hutchinson Cancer Center/University of Washington, Seattle, WA, USA; [11]Roswell Park Comprehensive Cancer Center, Buffalo, NY, USA; [12]OHSU Knight Cancer Institute, Portland, OR, USA; [13]Weill Cornell Medicine, New York City, NY, USA; [14]USC Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [15]Vanderbilt University Medical Center, Nashville, TN, USA; [16]Vall d'Hebron University Hospital, Barcelona, Catalonia, Spain; [17]Seagen Inc., Bothell, WA, USA; [18]Mayo Clinic, Scottsdale, AZ, USA

Objectives

- To explore correlations between tissue and blood-based biomarkers and clinical outcomes in the chemo-refractory HER2+ RAS WT mCRC patient population
- To evaluate HER2 testing methodologies (tissue and blood) that can be used to identify patients with HER2+ mCRC who may benefit from treatment with TUC + Tras

Conclusions

The percent agreement of HER2 status was high across the three different platforms (IHC/FISH, Tissue NGS, and Blood NGS), with higher percent agreement between tissue-based assays

Treatment response to TUC + Tras was predicted by a HER2+ result from any of the three testing platforms

- Patients in which HER2 amplification was not detected by ctDNA NGS may have HER2 amplification by a tissue-based assay and may benefit from treatment with a HER2-directed therapy
- cORR in IHC2+/ISH+ was numerically lower than IHC3+ but remained clinically relevant

These data support the use of both tissue and blood-based methods to identify HER2+ mCRC in patients that may benefit from treatment with TUC + Tras until an FDA-approved assay is available

Abbreviations

AE, adverse event; BICR, blinded independent central review; BID, twice a day; BRAF, proto-oncogene B-Raf; C1D1, cycle 1 day 1; CE, Conformité Européenne; CISH, chromogenic in situ hybridization; CLIA, Clinical Laboratory Improvement Amendments; cORR, confirmed objective response rate; CR, complete response; ctDNA, circulating tumor DNA; DOR, duration of response; FDA, United States Food and Drug Administration; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; ISH, in situ hybridization; ISO, International Organization for Standardization; mAb, monoclonal antibody; mCRC, metastatic colorectal cancer; mDOR, median duration of response; mOS, median overall survival; mPFS, median progression-free survival; NA, not available; NGS, next-generation sequencing; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PO, orally; PR, partial response; Q3W, every 3 weeks; QC, quality control; R, randomization; RAS, rat sarcoma virus; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease; TKI, tyrosine kinase inhibitor; Tras, trastuzumab; TUC, tucatinib; VEGF, vascular endothelial growth factor

Acknowledgements

This study was sponsored by Seagen Inc., Bothell, WA, USA, in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

The authors thank Sarah Canestaro, MS, of Populus Group, Troy, MI, supported by Seagen Inc. for providing medical writing and editorial support in accordance with Good Publication Practice guidelines.

References

1. Benson AB. J Natl Compr Canc Netw. 2021: 329-59.
2. Kang A. J Manag Care Spec Pharm. 2021: S20-1.
3. Patel JN. J Pers Med. 2019: 3.
4. Sartore-Bianchi A. The Oncologist. 2019: 1395-402.
5. Strickler J. J Clin Oncol. 2021: Abstract TPS153.
6. Cervantes A. Ann Oncol. 2023: 10-32.
7. Strickler JH. Lancet Oncol. 2023: 496-508.
8. Cercek A. J Clin Oncol. 2023: 198.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster,
John Strickler, john.strickler@duke.edu

American Society of Clinical Oncology; June 2-6, 2023; Abstract No. 3528

Background

- HER2 overexpression and/or amplification (HER2+) occurs in 3%-5% of patients with metastatic colorectal cancer (mCRC)[1-5]
 - Rates of HER2+ may be as high as ~10% in patients with RAS/BRAF wild-type mCRC tumors
- Established regional guidelines recommend HER2 testing and HER2-directed treatment options for mCRC[1,6]
- Tucatinib is a highly selective HER2-directed TKI approved by the FDA in combination with trastuzumab for treatment of patients with RAS wild-type HER2+ unresectable or mCRC that has progressed following treatment with fluoropyrimidine-, oxaliplatin-, and irinotecan-based chemotherapy
 - FDA approval was based on the primary analysis of the MOUNTAINEER trial (NCT03043313)

MOUNTAINEER Summary

- Results from the MOUNTAINEER primary analysis showed clinically meaningful activity and demonstrated tucatinib in combination with trastuzumab was well tolerated[7]

Efficacy (n=84)	
cORR per BICR	38.1%
mDOR	12.4 months
mPFS	8.2 months
mOS	24.1 months

Safety (n=86)
No deaths due to AEs
Diarrhea was most common AE • Grade 1 or 2: 60.5% • Grade 3: 3.5%
Low discontinuation rate due to AEs: 5.8%

MOUNTAINEER: Global, Open-Label, Phase 2 Trial

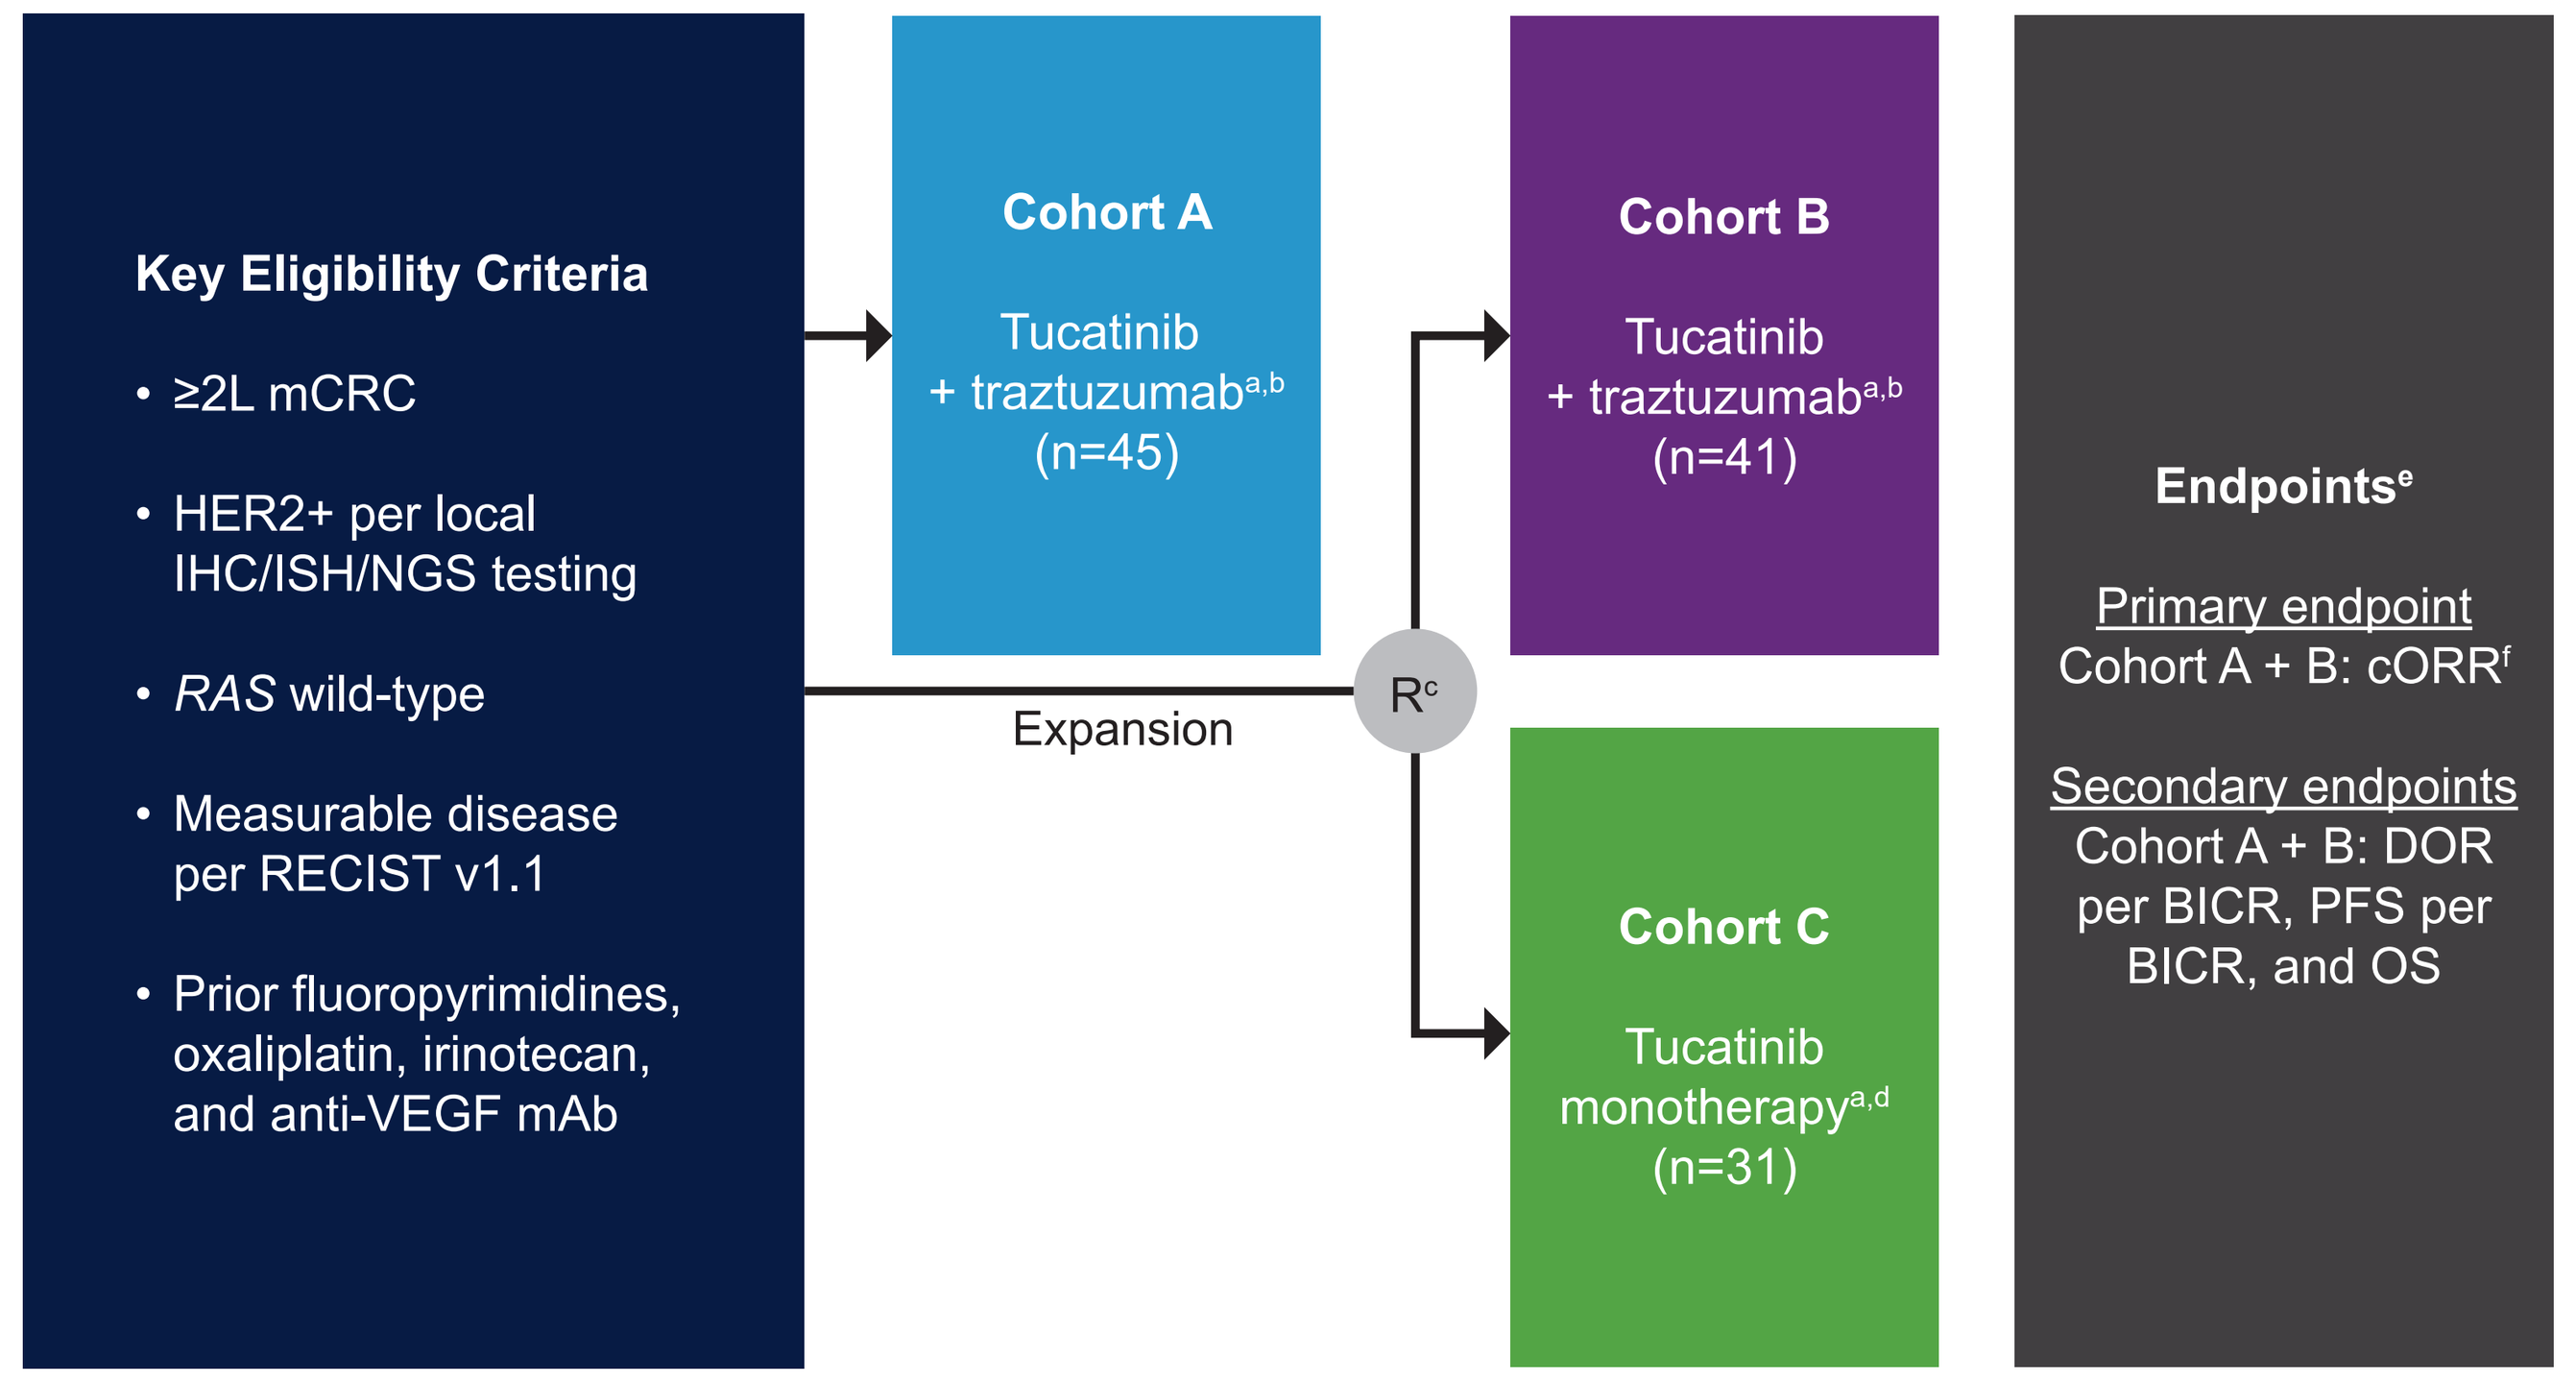
Key Eligibility Criteria
• ≥2L mCRC
• HER2+ per local IHC/ISH/NGS testing
• RAS wild-type
• Measurable disease per RECIST v1.1
• Prior fluoropyrimidines, oxaliplatin, irinotecan, and anti-VEGF mAb
Cohort A
Tucatinib + traztuzumab[a,b] (n=45)
Expansion
R[c]
Cohort B
Tucatinib + traztuzumab[a,b] (n=41)
Cohort C
Tucatinib monotherapy[a,d] (n=31)
Endpoints[e]
Primary endpoint
Cohort A + B: cORR[f]
Secondary endpoints
Cohort A + B: DOR per BICR, PFS per BICR, and OS

[a]Tucatinib dose: 300 mg PO BID, trastuzumab dose: 6 mg/kg Q3W (loading dose 8 mg/kg C1D1); each treatment cycle is 21 days
[b]Patients remained on therapy until evidence of radiographic or clinical progression, unacceptable toxicity, withdrawal of consent, or study closures
[c]Stratification: left sided tumor primary vs other
[d]Patients were allowed to cross over and receive tucatinib and trastuzumab if they experienced radiographic progression at any time point or if they had not achieved a PR or CR by week 12
[e]Efficacy assessed in patients who received any anount of study treatment and had HER2+ tumors
[f]RECIST v1.1 per BICR

Methods

Enrollment

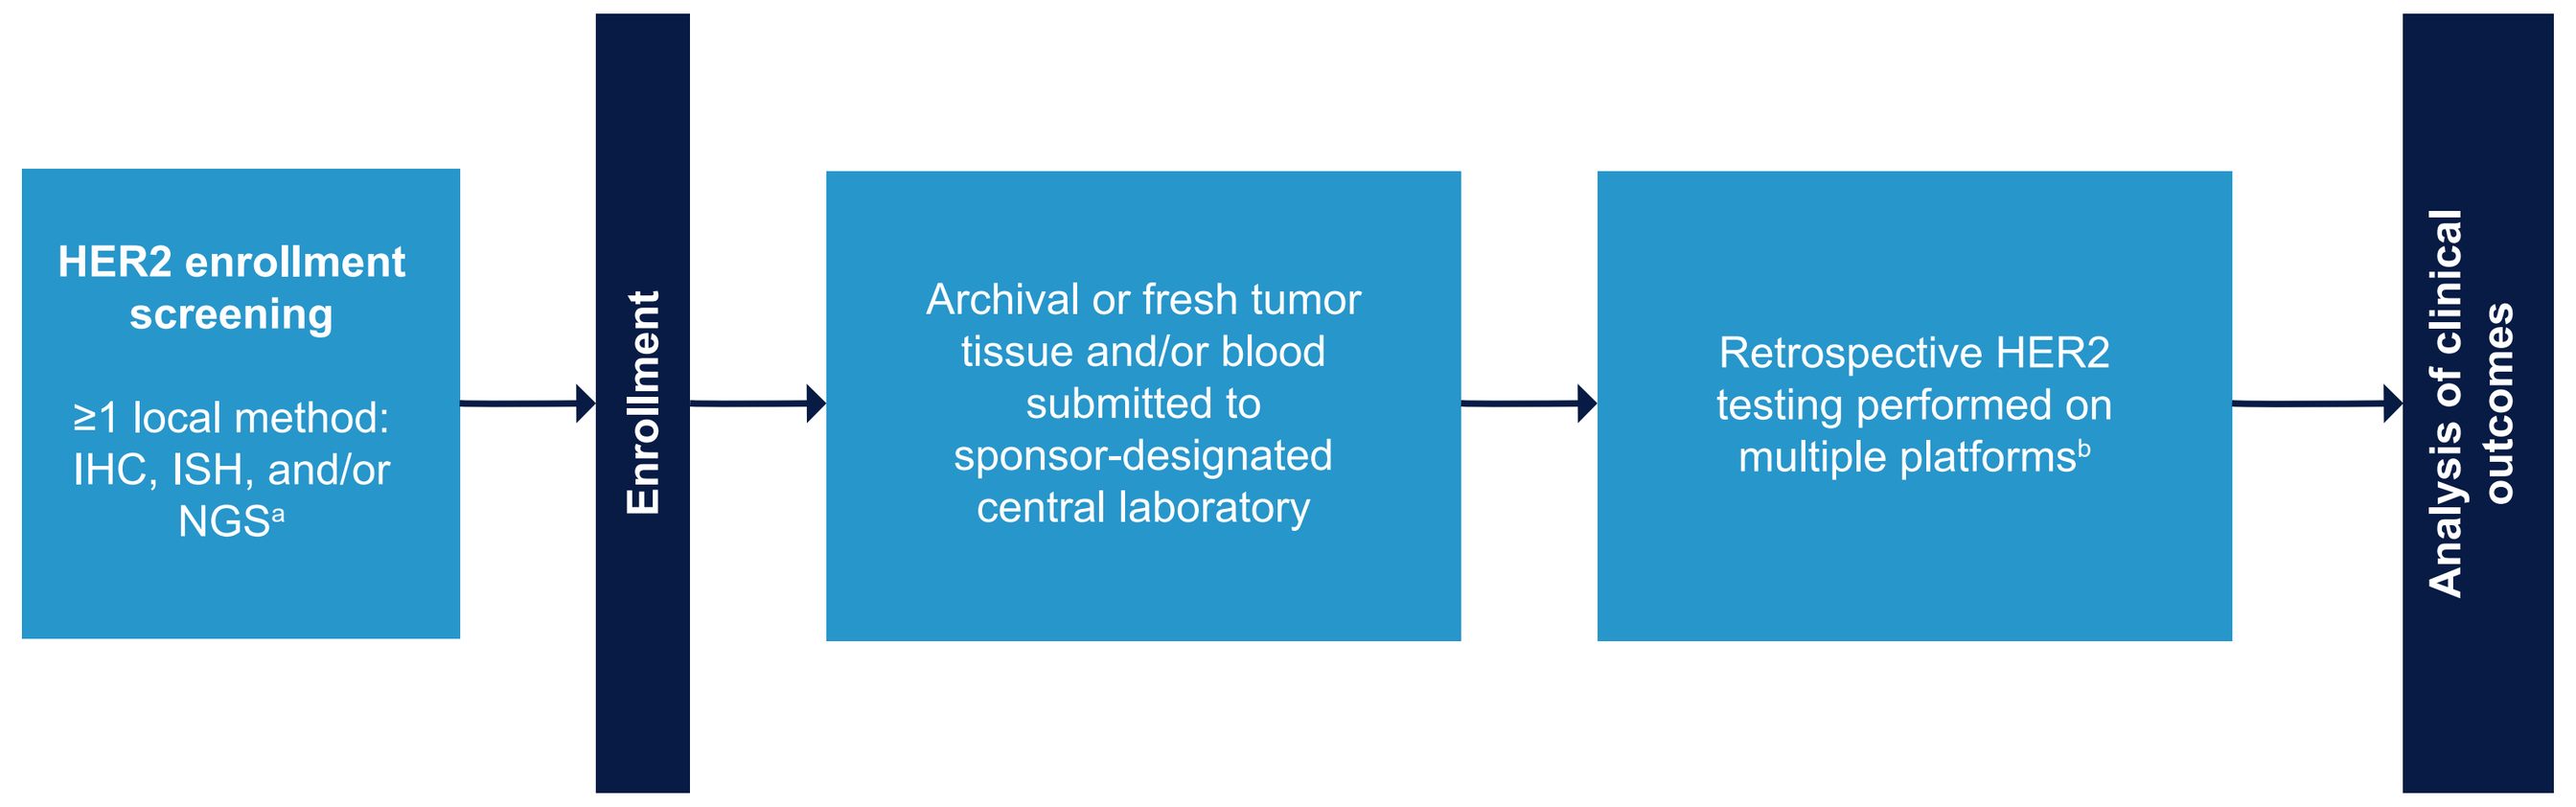
HER2 enrollment screening
≥1 local method: IHC, ISH, and/or NGS[a]
Enrollment
Archival or fresh tumor tissue and/or blood submitted to sponsor-designated central laboratory
Retrospective HER2 testing performed on multiple platforms[b]
Analysis of clinical outcomes

[a]Confirmed HER2-positive mCRC tested at CLIA-certified or ISO-accredited laboratory: IHC: HER2 3+ IHC by an FDA-approved or CE-marked HER2 IHC test following interpretational manual for breast cancer, ISH: HER2 2+ IHC with amplification by an FDA-approved or CE-marked HER2 in situ hybridization assay (FISH or CISH) following interpretational manual for breast cancer, NGS: HER2 amplification by CLIA-certified or ISO-accredited NGS sequencing assay
[b]HER2 status was evaluated in tissue and blood samples using multiple assays. Tissue samples were analyzed by IHC (Ventana HER2 (4B5) IHC Assay), FISH (Dako iQFISH pharmDx assay) per the interpretational manual for breast cancer. Tissue NGS was performed using the PGDx elio tissue complete assay. NGS analysis of ctDNA was done using the Guardant 360 assay

- There are currently no established best practices for HER2 testing and/or interpretation in mCRC
 - Previously presented analysis of MOUNTAINEER showed that there is a very high level of concordance between breast and gastric algorithms, suggesting either algorithm (with appropriate training) can be used to identify patients with HER2+ mCRC[8]
 - Here we present clinical outcomes stratified by HER2 testing method for patients with mCRC treated with TUC + TRAS

Results

Samples Available for Central HER2 Testing

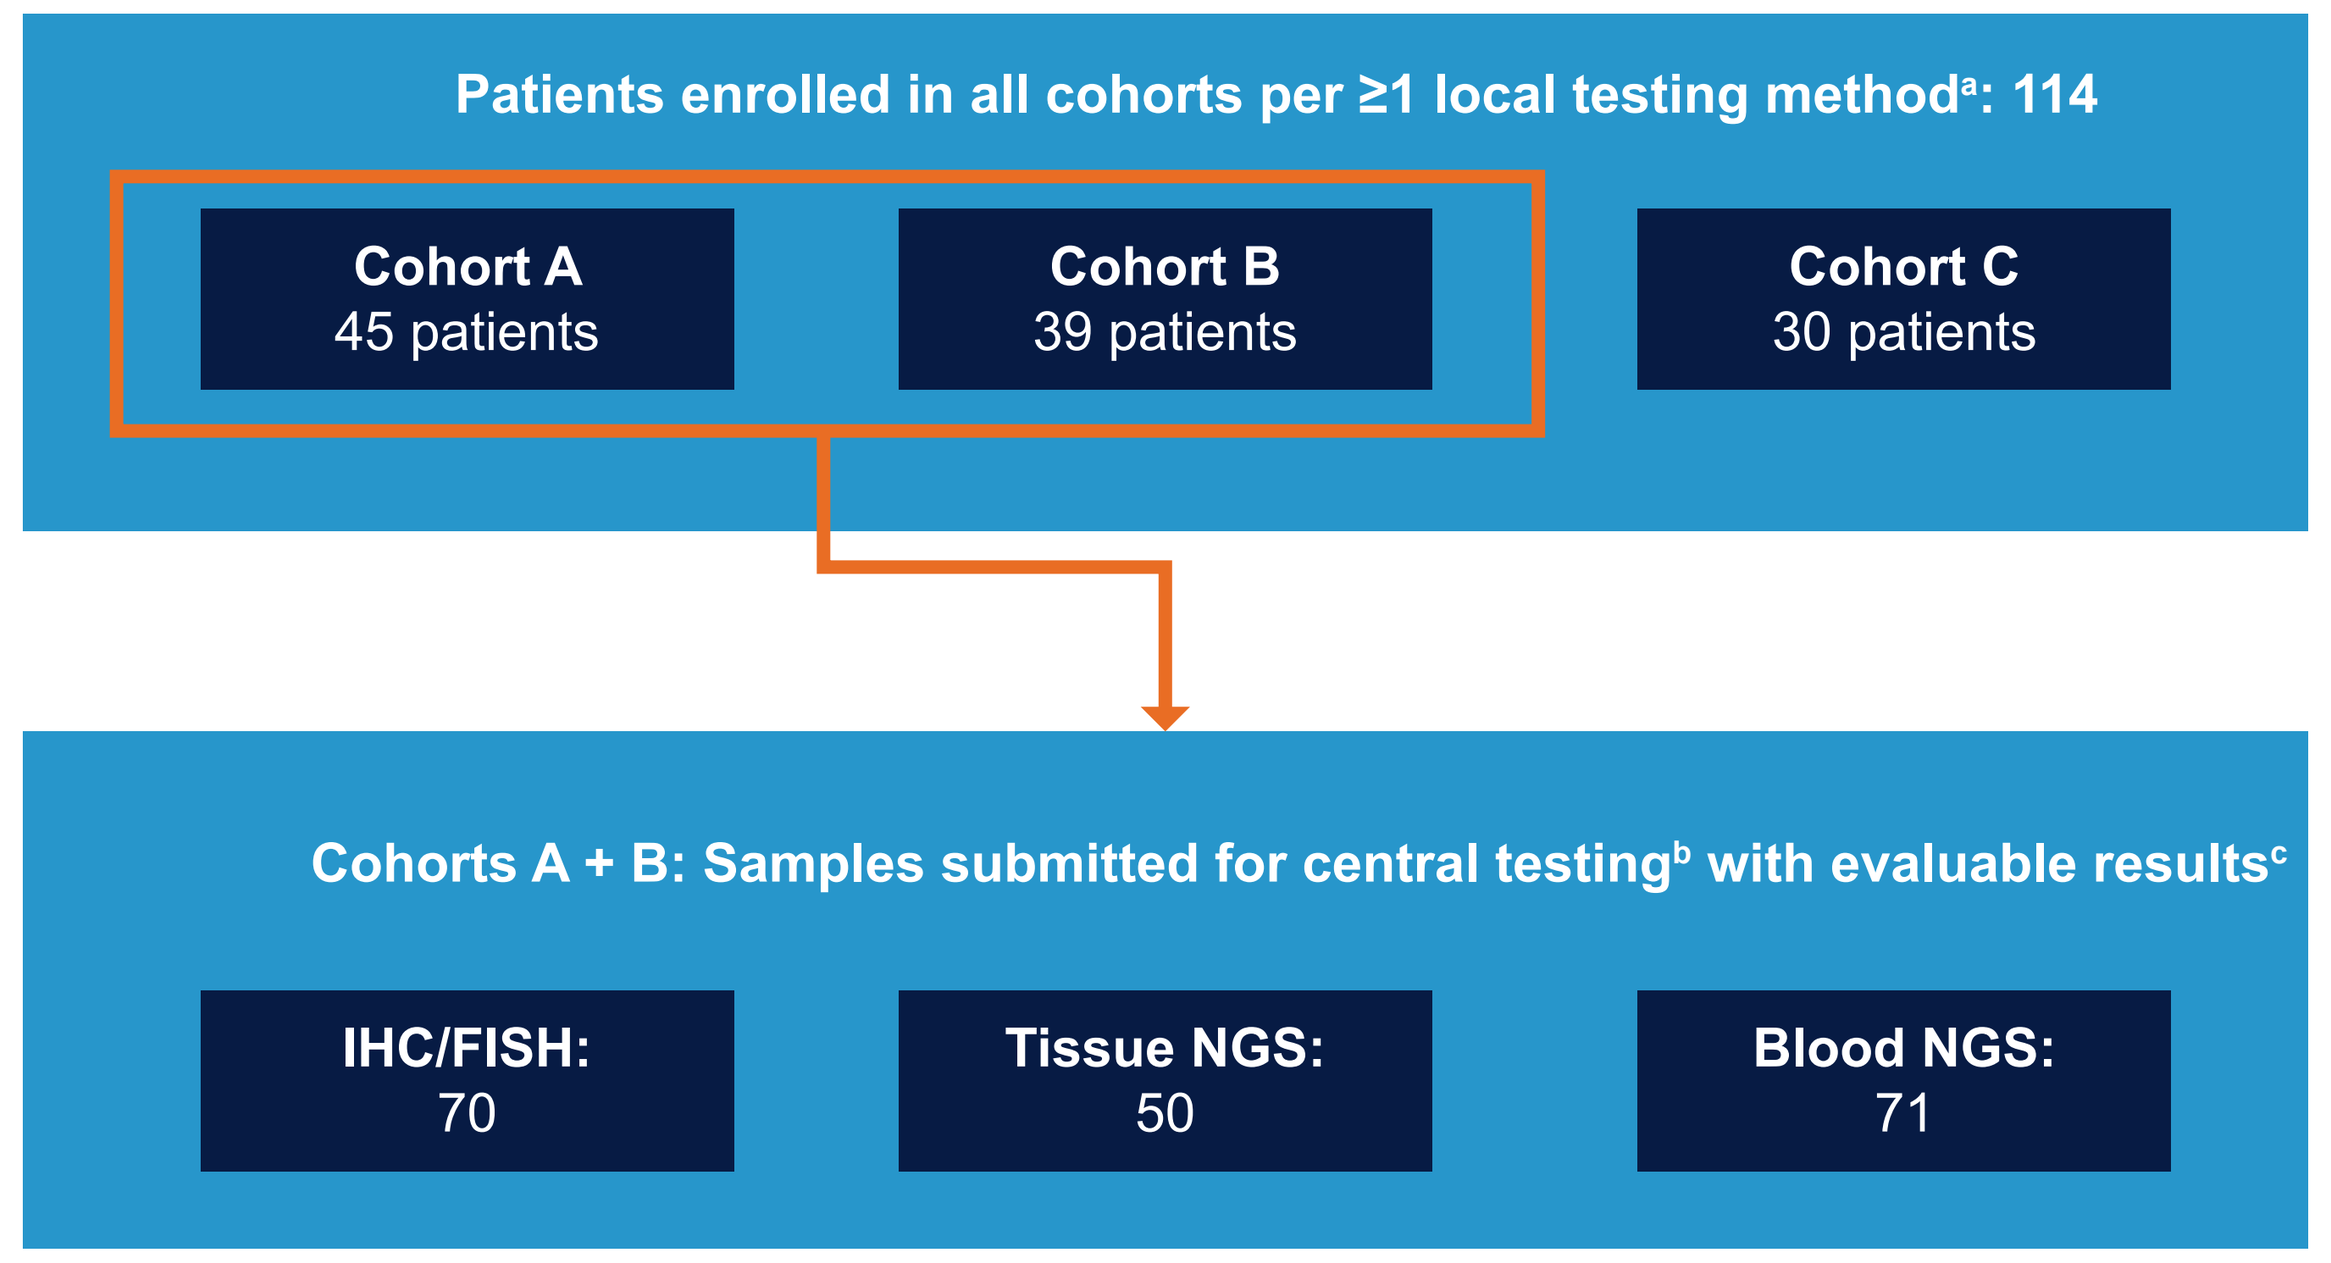
Patients enrolled in all cohorts per ≥1 local testing method[a]: 114
Cohort A
45 patients
Cohort B
39 patients
Cohort C
30 patients
Cohorts A + B: Samples submitted for central testing[b] with evaluable results[c]
IHC/FISH:
70
Tissue NGS:
50
Blood NGS:
71

[a]Patients were enrolled after locally testing tissue by at least one of the approved testing methods (IHC, ISH, NGS)
[b]Tissue samples were retrospectively analyzed for HER2 status at the sponsor-designated central laboratory
[c]Variability observed in sample size for each assay is due to differences in QC/acceptance criteria of each assay and collection/availability of samples

Percent Agreement Between Three Central HER2 Testing Modalities

- High level of percent agreement of HER2 status observed across multiple central HER2 testing modalities (all cohorts included; pairwise comparisons)

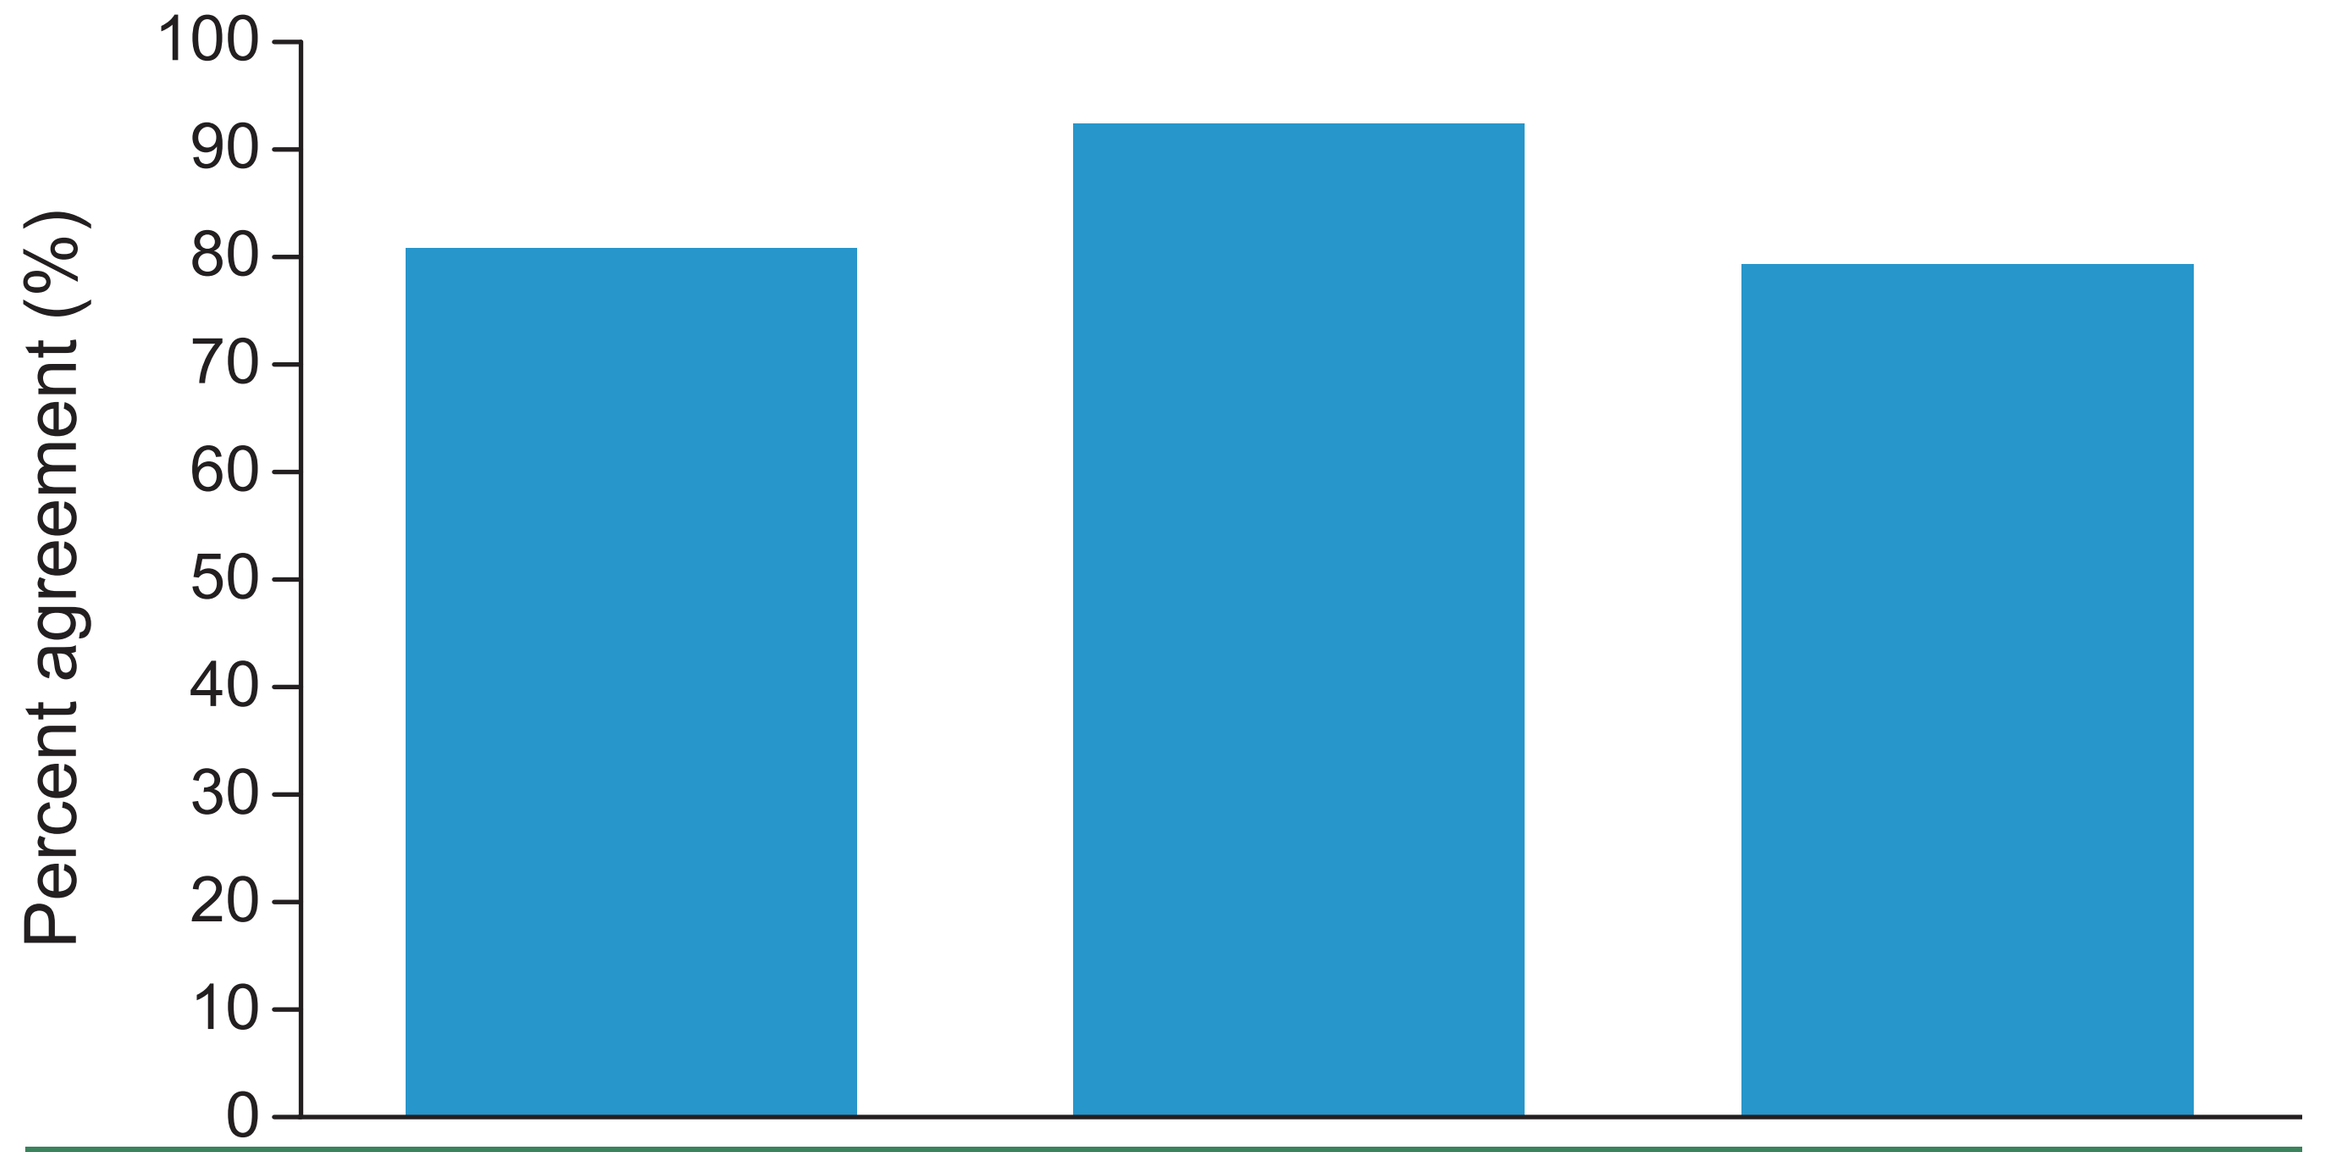
Percent agreement (%)
100
90
80
70
60
50
40
30
20
10
0

	Blood vs. tissue NGS	IHC/FISH vs. tissue NGS	IHC/FISH vs. blood NGS
n/N	47/58	63/68	66/83
%	81.0%	92.6%	79.5%
95% CI	68.6, 90.1	83.7, 97.6	69.2, 87.6

MOUNTAINEER Cohorts A+B: Response Assessment by Testing Platform

- cORR per BICR was 40.0% to 47.7% for patients identified as having HER2+ disease by any of the three assays
- DOR per BICR was 12.4-16.4 months for patients identified as having HER2+ disease by any of the three assays
- Tumors confirmed as HER2+ by central IHC/ISH had a mDOR of 16.4 months (95% CI: 10.6, 25.5) and mPFS of 10.1 months (95% CI: 4.2, 15.2)

Response	Central IHC + FISH (n=70)		
	Positive (IHC3+) (n=45)	Positive (IHC2+/ISH+) (n=15)	Negative (n=10)
CR	3	0	0
PR	18	3	1
SD[a]	17	5	4
PD	7	6	5
NA	0	1	0
cORR, n (%) (95% CI)	21 (46.7%) (31.7, 62.1)	3 (20.0%) (4.3, 48.1)	1 (10.0%) (0.3, 44.5)
mDOR, months (95% CI)	16.4 (10.6, 25.5)		-
mPFS, months (95% CI)	10.1 (4.2, 15.2)		2.8 (1.2, 6.3)

[a]Includes non-CR/non-PD

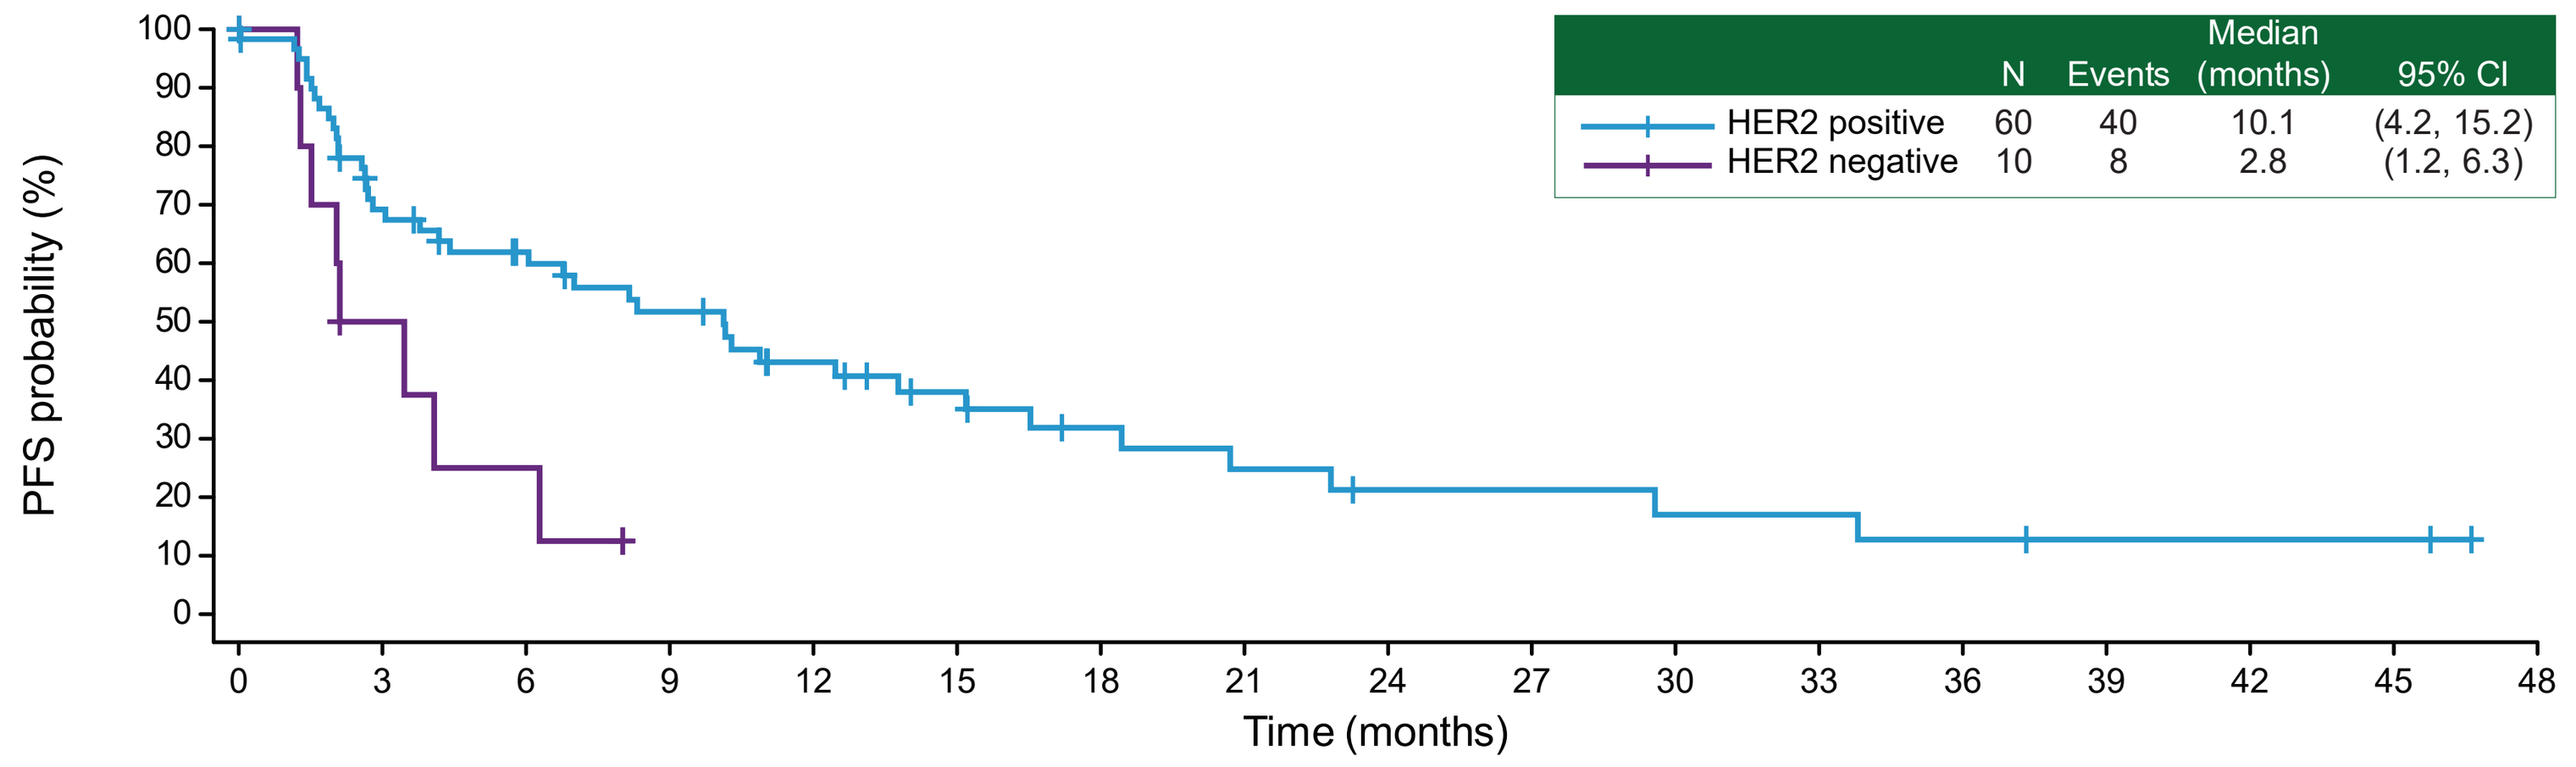
PFS probability (%)
Time (months)
N
Events
Median (months)
95% CI
HER2 positive 60 40 10.1 (4.2, 15.2)
HER2 negative 10 8 2.8 (1.2, 6.3)

- The one responder that was HER negative by central IHC/FISH was HER2 amplified by other central methods

Response	PGDx tissue NGS	
	HER amplification detected (n=44)	HER2 amplification not detected (n=6)
CR	1	0
PR	20	0
SD[a]	16	2
PD	7	4
NA	0	0
cORR, n (%) (95% CI)	21 (47.7%) (32.5, 63.3)	0 (0%) (0, 45.9)
mDOR, months (95% CI)	15.3 (8.9, 25.5)	-
mPFS, months (95% CI)	10.9 (7.0, 20.7)	2.1 (1.3, -)

[a]Includes non-CR/non-PD

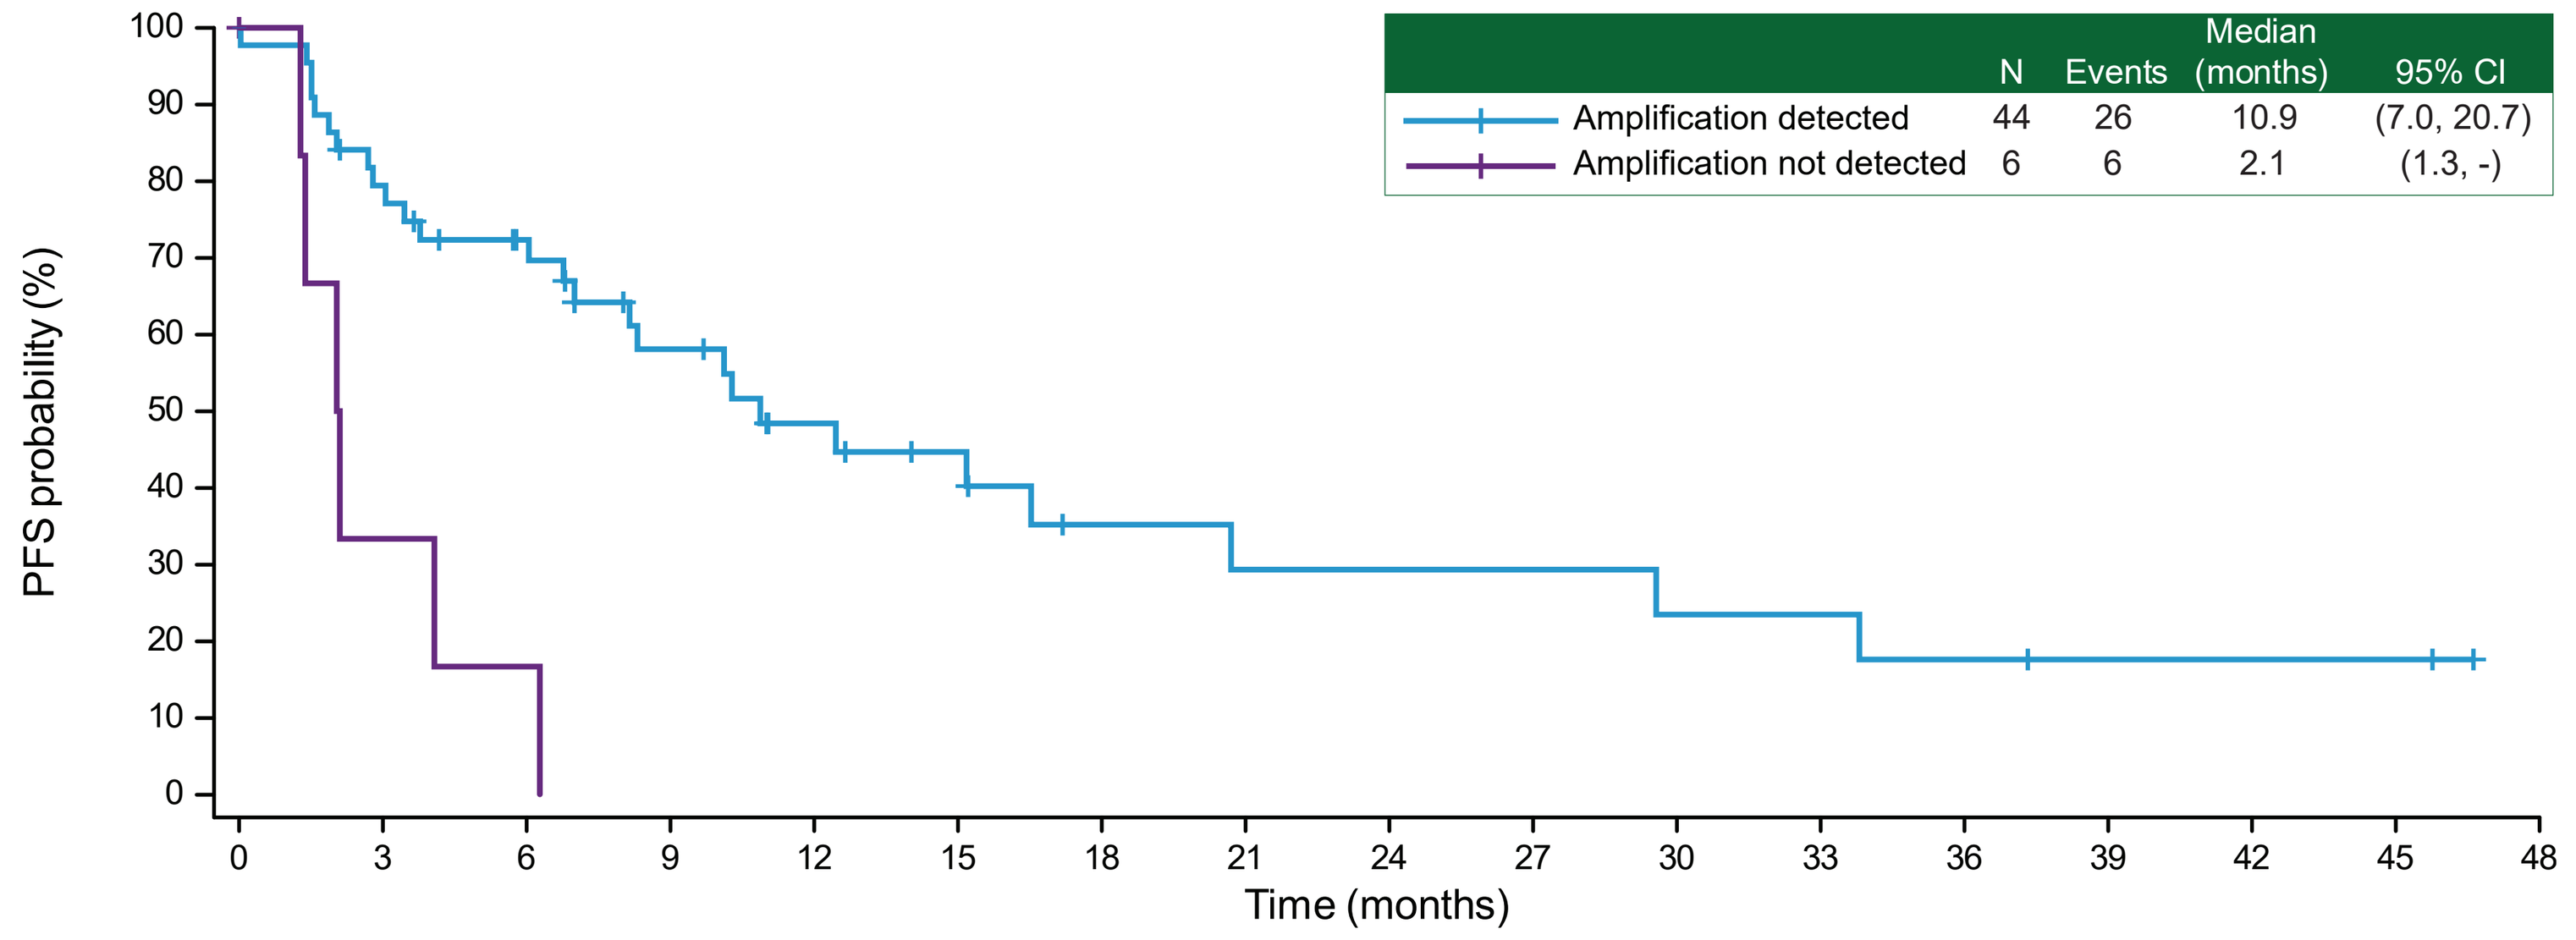
PFS probability (%)
Time (months)
N
Events
Median (months)
95% CI
Amplification detected 44 26 10.9 (7.0, 20.7)
Amplification not detected 6 6 2.1 (1.3, -)

Response	Guardant ctDNA	
	HER amplification detected (n=56)	HER2 amplification not detected (n=15)
CR	1	1
PR	22	2
SD[a]	18	7
PD	14	4
NA	1	1
cORR, n (%) (95% CI)	23 (41.1%) (28.1, 55)	3 (20.0%) (4.3, 48.1)
mDOR, months (95% CI)	12.4 (6.2, 38.3)	-
mPFS, months (95% CI)	8.1 (3.1, 10.2)	10.9 (2.0, 18.4)

[a]Includes non-CR/non-PD

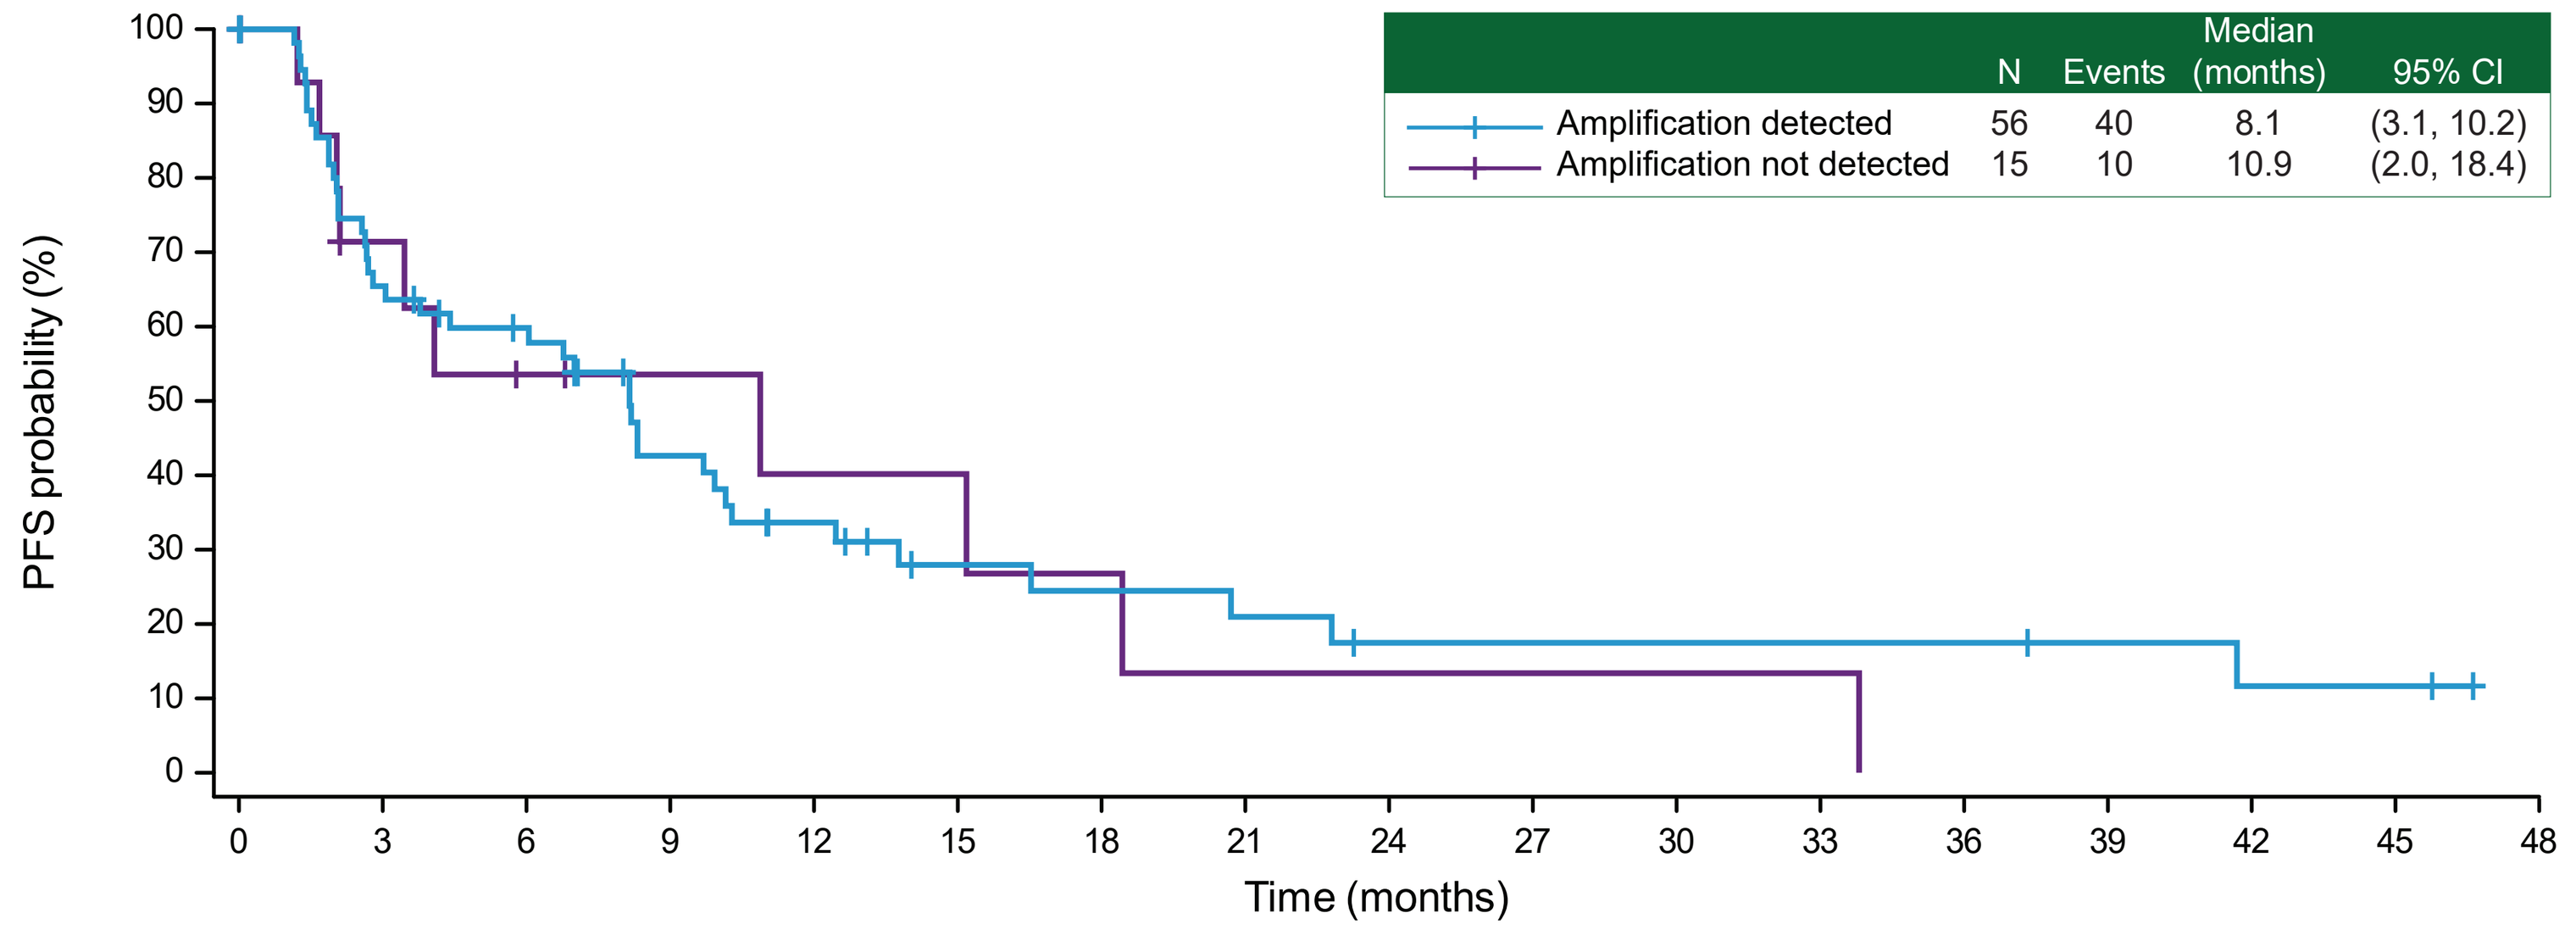
PFS probability (%)
Time (months)
N
Events
Median (months)
95% CI
Amplification detected 56 40 8.1 (3.1, 10.2)
Amplification not detected 15 10 10.9 (2.0, 18.4)