Occurrence of Brain Metastasis and Treatment Patterns Among Patients with HER2-Positive Metastatic Breast Cancer

Gregory A. Vidal[1], Sonia Pulgar[2], Santosh Gautam[3], Anna Vlahiotis[3], Maxine D. Fisher[3], Kendra DeBusk[2]

[1] Breast Cancer Research, West Cancer Center and Research Institute, Germantown, TN USA; [2] Health Economics & Outcomes Research, Seattle Genetics, Bothell, WA USA, [3] Concerto HealthAI, Boston, MA USA

Background

- With the introduction of anti-HER2 targeted therapies, the outcomes among patients with HER2 positive (HER2+) breast cancer (about 15 to 22% of all breast cancer patients), have significantly improved.[1-3]
- Currently, pertuzumab plus trastuzumab and a taxane combination is considered standard of care first-line treatment for patients with HER2+ metastatic breast cancer (MBC) followed by T-DM1 as second-line therapy. However, there is no consensus about standard of care for patients who progress after second-line therapy.[4,5]
- Patients with HER2+ MBC are at higher risk of brain metastasis (BM), with up to 50% developing them, which adds complexity to treatment.[6,7]
- There is limited real-world data on development of BM in patients that are being treated with currently available systemic therapies, and the therapies used to treat patients with both systemic disease and BM.
- The objective of this observational study was to assess BM occurrence and subsequent treatment among women with HER2+ MBC.

Objectives

- To describe the demographic and clinical characteristics of patients diagnosed with HER2+ MBC.
- To assess incidence of BM and subsequent treatment among women diagnosed with HER2+ MBC.

Methods

Study Design

- Retrospective, observational study using electronic medical record (EMR) data

Data Source

- Data were abstracted from EMRs from a network of community oncology practices maintained in the Vector Oncology Data Warehouse

Eligibility

- Women ≥18 years old and diagnosed with HER2+ MBC between 6/1/2012 and 5/31/2018

Study Variables

- Age, race, stage at initial diagnosis, tumor grade at initial diagnosis, hormone receptor status at initial diagnosis, performance status (non-impaired: ECOG ≤1, impaired: ECOG >1), comorbidity

Outcomes

- Timing of brain metastasis
- Treatment patterns

Statistical Methods

- Descriptive analysis including means, standard deviations, medians, and minimum and maximum values for continuous variables (e.g., age at diagnosis) and frequencies and percentages for categorical variables (e.g., race, performance status)

Results

Demographic and clinical characteristics (Table 1)

- Of 372 study eligible patients, 165 (44.4%) had a record of BM.
- Demographic characteristics of BM patients were consistent with the overall sample.
- There were 89 patients (53.9%) who had hormone-receptor positive tumors among those who had BM.
- There were 82 patients (49.7%) with de novo MBC, and 89 (53.9%) had hormone-receptor positive tumors among those who had BM.

Table 1. Demographic and Clinical Characteristics

Variables	Overall (N=372)	Patients with BM (N=165)
Age (years) at Diagnosis of MBC		
Median	56	54
Min, Max	28, 88	29, 83
White Patients, n (%)	244 (65.6%)	99 (60.0%)
De novo MBC, n (%)	177 (47.6%)	82 (49.7%)
Hormone Receptor Positive, n (%)	218 (58.6%)	89 (53.9%)
Impaired Performance Status (ECOG≥2), n (%)	19 (5.1%)	9 (5.5%)
Number of Metastasis Sites at Index Date, n (%)		
0	152 (40.9%)	59 (35.8%)
1-2	110 (29.6%)	48 (29.1%)
3+	110 (29.6%)	58 (35.2%)
Weighted Comorbidity Index Score at Index Date, n (%)		
0	262 (70.4%)	121 (73.3%)
1-2	97 (26.1%)	40 (24.2%)
3+	13 (3.5%)	4 (2.4%)

Timing of brain metastasis (Table 2)

- There were 37 patients (22.4%) with a record of BM at the time of initial MBC diagnosis (baseline), 63 (38.2%) developed BM during the first-line treatment, 23 (13.9%) during the second-line, and 42 (25.5%) during the third-line or beyond.
- The median time to develop BM from initial MBC diagnosis was 12.5, 18.3, and 22.8 months for patients who developed BM during first-, second-, and third-line treatment or later, respectively.

Table 2. Incidence of Brain Metastasis by Line of Therapy

Parameter	Number of patients with BM (N = 165)	Median time to BM from MBC diagnosis (months)
Patients with BM, n (%)		
At the time of initial MBC diagnosis	37 (22.4%)	N/A
During first-line treatment	63 (38.2%)	12.5
During second-line treatment	23 (13.9%)	18.3
During third-line treatment or later	42 (25.5%)	22.8

Post-BM systemic treatment (Figure 1)

- Figure 1 shows the various treatment patients received in the line immediately after BM diagnosis.
- Among those with baseline BM (n=37), the most common regimen post-BM diagnosis was HER2-directed therapy combined with chemotherapy (n=15; 40.5%).
- In patients who developed BM during first- and second-line treatment, the most common regimen after BM diagnosis was HER2-directed therapy alone (47.6% and 43.5%, respectively).
- Among patients who developed BM during third- and fourth-line treatment, there was some variation in treatment they received after BM diagnosis. More importantly, 21.1% of patients who developed BM during third-line treatment and 17.6% of patients who developed BM during fourth-line treatment did not receive any systemic therapy after BM diagnosis.
- Capecitabine-based regimens were used by 2.7%, 15.9%, 17.4%, 31.6%, and 17.7% of patients who developed BM at baseline, during first-line, second-line, third-line, and fourth-line treatment, respectively.

Figure 1. Treatment Received after Incidence of Brain Metastasis

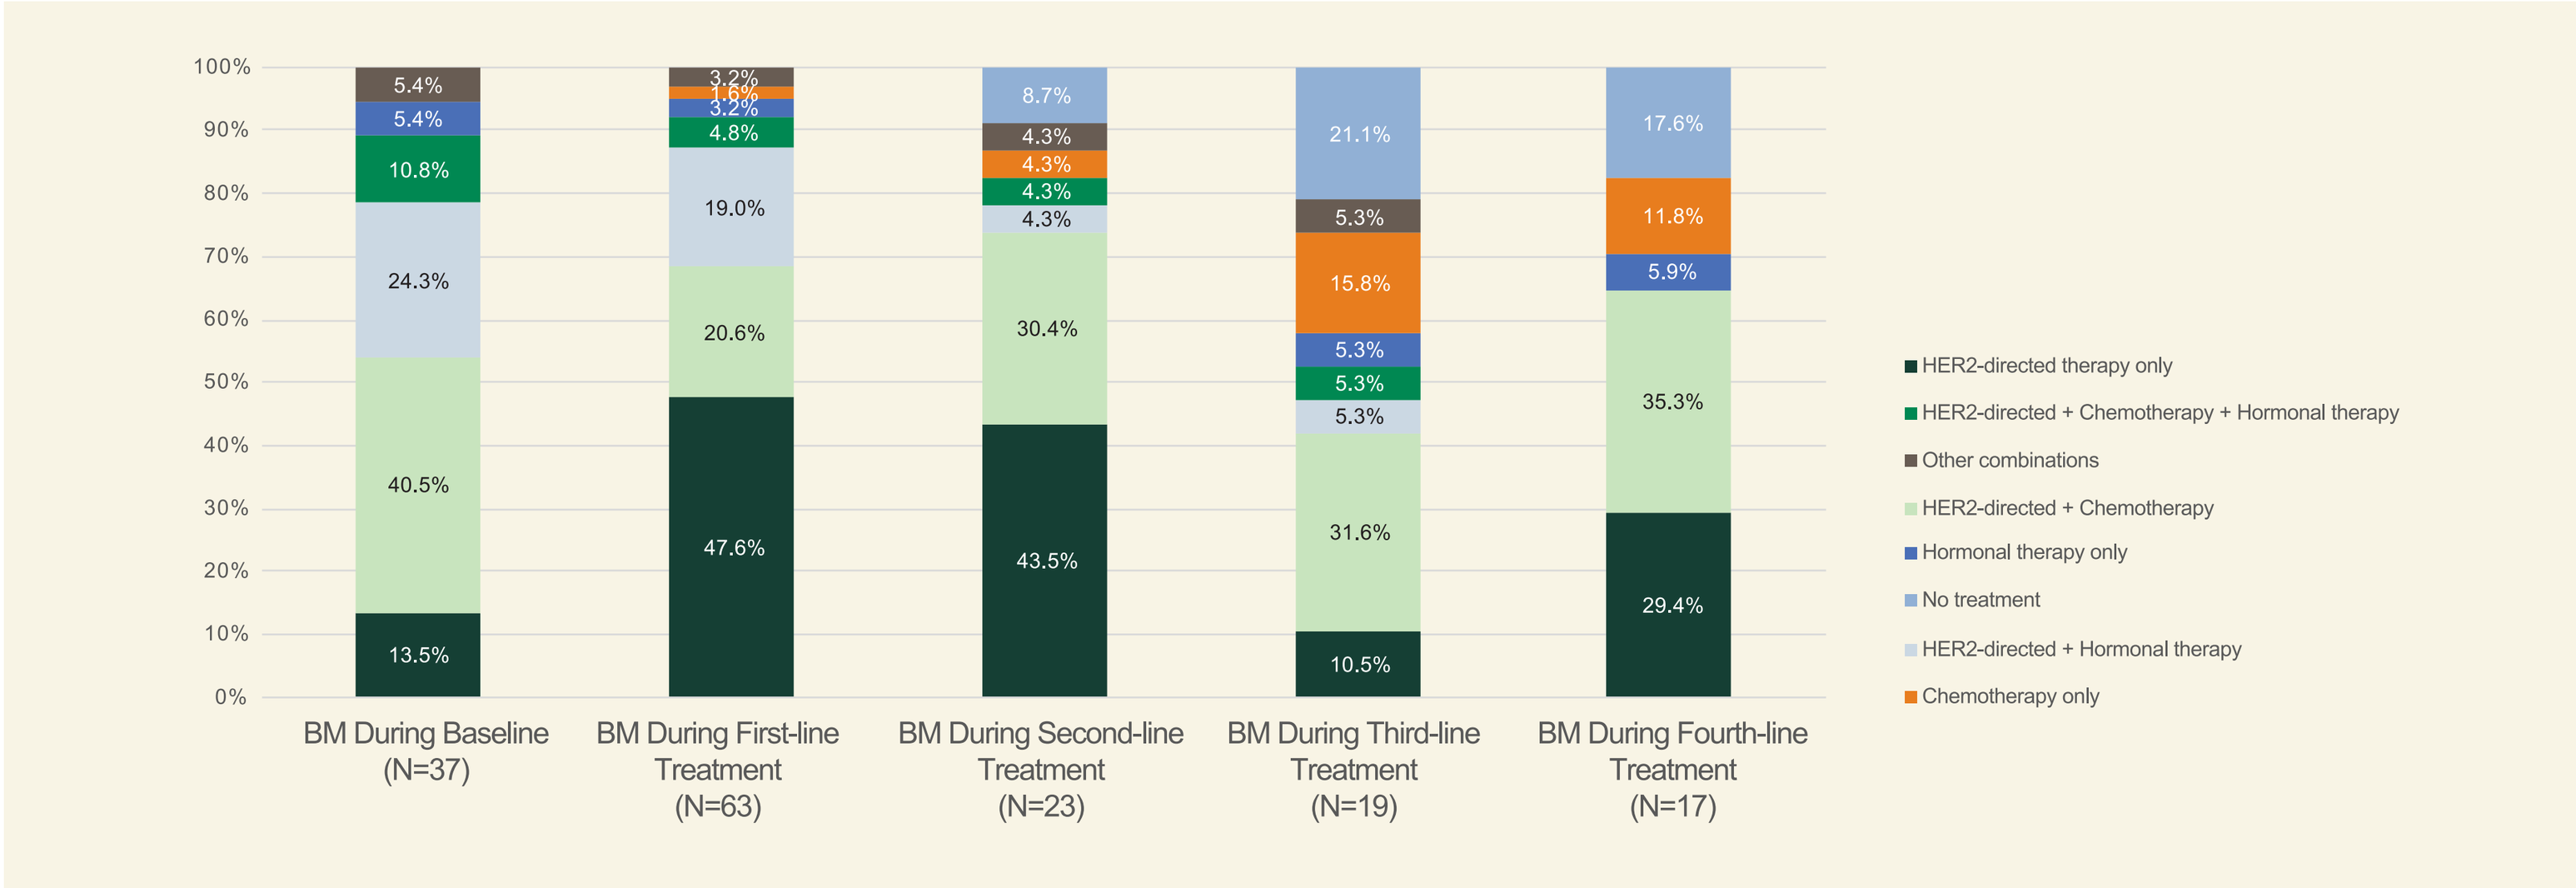
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
5.4%
5.4%
10.8%
24.3%
40.5%
13.5%
3.2%
1.6%
3.2%
4.8%
19.0%
20.6%
47.6%
8.7%
4.3%
4.3%
4.3%
4.3%
30.4%
43.5%
21.1%
5.3%
15.8%
5.3%
5.3%
5.3%
31.6%
10.5%
17.6%
11.8%
5.9%
35.3%
29.4%
BM During Baseline (N=37)
BM During First-line Treatment (N=63)
BM During Second-line Treatment (N=23)
BM During Third-line Treatment (N=19)
BM During Fourth-line Treatment (N=17)
HER2-directed therapy only
HER2-directed + Chemotherapy + Hormonal therapy
Other combinations
HER2-directed + Chemotherapy
Hormonal therapy only
No treatment
HER2-directed + Hormonal therapy
Chemotherapy only

HER2-therapy type (Figure 2)

- Trastuzumab and pertuzumab combination based regimens were the most common post-BM HER2-directed therapy among patients who developed BM at baseline (54.1%) and during first-line treatment (57.1%) or second-line treatment (39.1%).
- Post-BM T-DM1-based regimens use was lowest among patients who had baseline BM (8.1%) and highest among patients who developed BM during third-line treatment (21.1%).
- Lapatinib-based regimens were used by 14.3%, 17.4%, 21.1%, and 11.8% of patients who developed BM during first-, second-, third- , and fourth-line treatment, respectively.

Figure 2. Type of HER2-therapy Post-BM Diagnosis

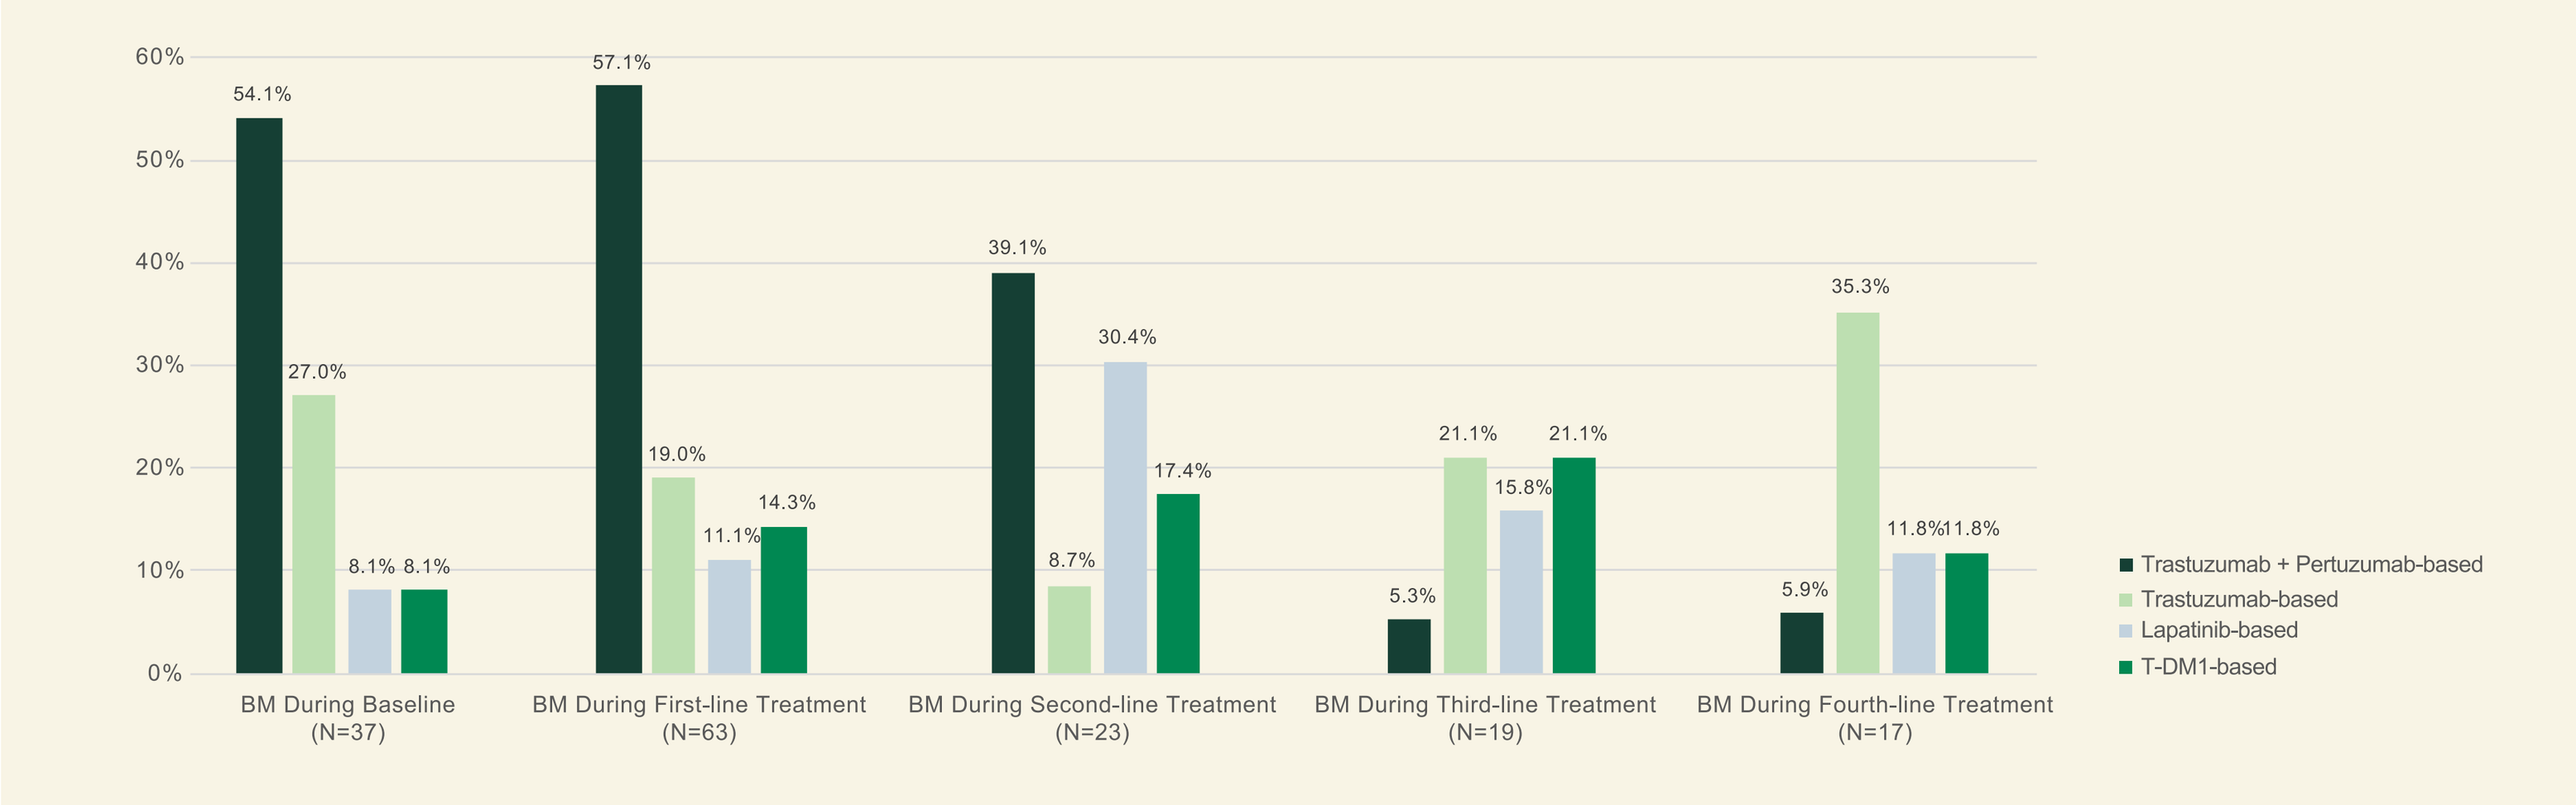
60%
50%
40%
30%
20%
10%
0%
54.1%
27.0%
8.1%
8.1%
57.1%
19.0%
11.1%
14.3%
39.1%
8.7%
30.4%
17.4%
5.3%
21.1%
15.8%
21.1%
5.9%
35.3%
11.8%
11.8%
BM During Baseline (N=37)
BM During First-line Treatment (N=63)
BM During Second-line Treatment (N=23)
BM During Third-line Treatment (N=19)
BM During Fourth-line Treatment (N=17)
Trastuzumab + Pertuzumab-based
Trastuzumab-based
Lapatinib-based
T-DM1-based

Conclusions

- BM were common among patients with HER2+ MBC, and occurred at various points during the course of treatment.
- Post-BM systemic therapy varied widely, which may indicate a lack of standard of care for patients with HER2+ MBC after BM diagnosis and an unmet need in this patient population.
- Having BM does not seem to impact physicians' choice of potential central nervous system penetrating agents, as prescribers tended to follow current guideline indications for use of capecitabine and lapatinib.
- The variation in treatment, especially in the later lines, highlights the limitations of current standard of care and the need for further analysis on treatment efficacy.
- The results have to be interpreted with caution because with the advent of newer therapies in treating HER2+ MBC, it is expected that treatment patterns will change during the next few years.

References

1. Baselga J, Cortes J, Kim SB, et al. Pertuzumab plus trastuzumab plus docetaxel for metastatic breast cancer. *N Engl J Med*. 2012;366(2):109-119.
2. Geyer CE, Forster J, Lindquist D, et al. Lapatinib plus capecitabine for HER2-positive advanced breast cancer. *N Engl J Med*. 2006;355(26):2733-2743.
3. Verma S, Miles D, Gianni L, et al. Trastuzumab emtansine for HER2-positive advanced breast cancer. *N Engl J Med* 2012;367(19):1783-1791.
4. Giordano SH, Temin S, Chandarlapaty S, et al. Systemic therapy for patients with advanced human epidermal growth factor receptor 2–positive breast cancer: ASCO Clinical Practice Guideline update. *J Clin Oncol*. 2018;36(26):2736-2740.
5. Martinez-Janez N, Chacon I, de Juan A, et al. Anti-HER2 Therapy Beyond Second-Line for HER2-Positive Metastatic Breast Cancer: A Short Review and Recommendations for Several Clinical Scenarios from a Spanish Expert Panel. *Breast Care (Basel)*. 2016;11(2):133-138.
6. Santa-Maria CA, Nye L, Mutonga MB, Jain S, Gradishar WJ. Management of metastatic HER2-positive breast cancer: where are we and where do we go from here? Oncology *(Williston Park)*. 2016;30(2):148-155.
7. Kabraji S, Ni J, Lin NU, Xie S, Winer EP, Zhao JJ. Drug Resistance in HER2-Positive Breast Cancer Brain Metastases: Blame the Barrier or the Brain? *Clin Cancer Res*. 2018;24(8):1795-1804.

Disclosures

- This study was sponsored by Seattle Genetics.
- GAV is a paid consultant for Concerto HealthAI, Eli Lilly, Immunomedics, Novartis, Pfizer, Puma, and Roche/Genentech. He receives fees for non-CME services from AstraZeneca, Eli Lilly, Novartis, Pfizer, and Puma, and owns stock in Oncodisc. Dr. Vidal also reports contracted research with AstraZeneca, BMS, Celcuity, Eli Lilly, GTx, Inc., Halozyme Therapeutics, Immunomedics, Merck, Pfizer, Puma, Roche/Genentech, and Tesaro. SP is employed by Seattle Genetics and owns stock in Seattle Genetics, Johnson & Johnson, and Gilead. SG, AV, and MDF are employed by Concerto HealthAI, and they disclosure research funding to their institution from Seattle Genetics. KD is employed by Seattle Genetics and owns stock in Seattle Genetics and Roche.

Contact Information

Contact Gregory Vidal, MD, PhD at **gvidal@westclinic.com** for questions and comments

ESMO Breast Cancer Virtual Meeting, May 23-24, 2020